A randomized, double-blind, placebo-controlled trial on the efficacy and safety of incobotulinumtoxinA (Inco/A) for essential tremor using kinematics-guided clinical decision support

Mandar Jog,[1] Jack Lee,[1] Michael Althaus,[2] Astrid Scheschonka,[2] Hanna Dersch,[2] **David M Simpson**,[3] Essential Tremor Study Team

[1]Lawson Health Research Institute, London, Ontario, Canada; [2]Merz Pharmaceuticals GMbH, Frankfurt am Main, Germany; [3]Icahn School of Medicine at Mount Sinai Medical Center, New York, NY, USA

Background

- Essential tremor (ET) affects 4.6% of people aged 65 years and older.[1] Propanolol and primidone have shown efficacy against ET, but 30–50% of patients do not respond to treatment.[2]
- Placebo-controlled studies with botulinum neurotoxin A (BoNT-A) have demonstrated significant reductions in tremor amplitude, but only modest clinical benefit, mainly as a result of functional impairment relating to hand and wrist weakness.[3,4]
- A recent open-label study with incobotulinumtoxinA (Inco/A; Xeomin®, Merz Pharmaceuticals GmbH), using a new kinematically guided, individualized injection approach, suggested that ET can be treated effectively with Inco/A.[5]

Objective

- To investigate the efficacy and safety of a single, kinematic-analysis-based (TremorTek™) intramuscular injection of Inco/A, compared with placebo, in moderate-to-marked ET of the upper limb.

Methods

Study design

- This study was a prospective, randomized, double-blind, placebo-controlled, multicenter study of 30 patients with definite ET (modified Tremor Investigation Group [TRIG] criteria).
- Patients received a single, unilateral, intramuscular injection of Inco/A or placebo into the wrist and, optionally, into the elbow and/or shoulder, of the upper limb that had the greater impact on their activities of daily living, and were monitored regularly over 24 weeks (**Figure 1**).

Figure 1. Study flow chart

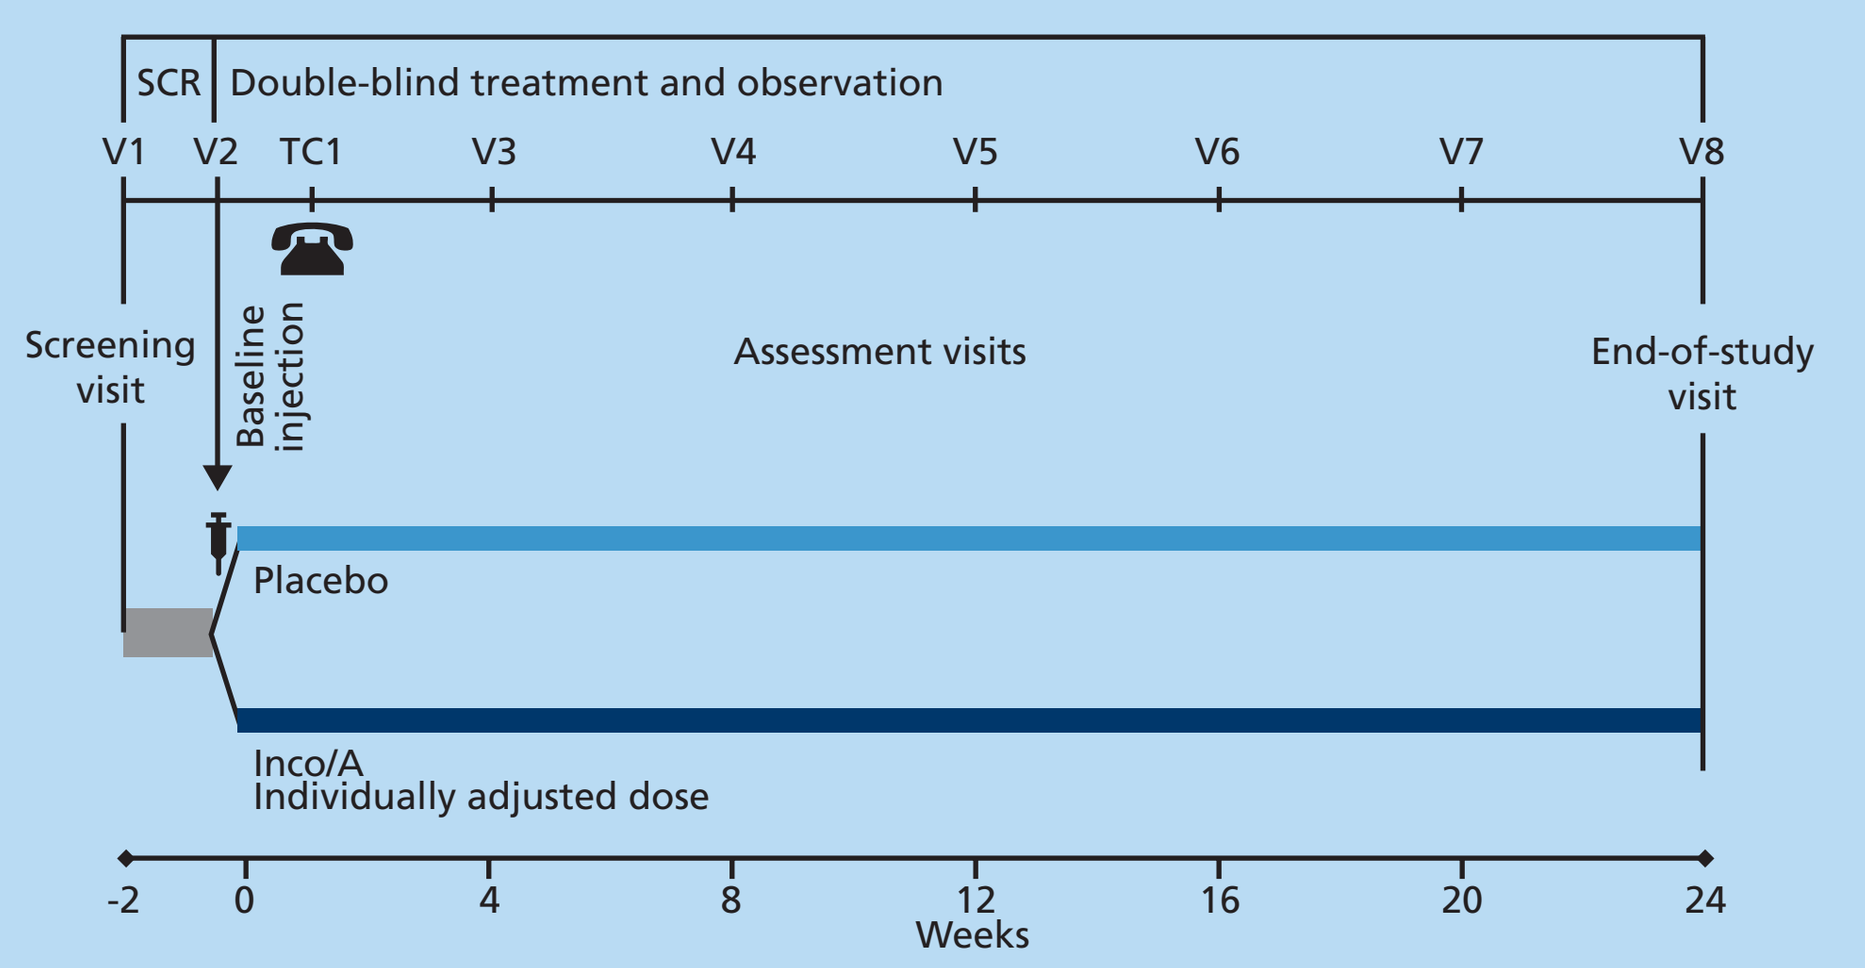
SCR
Double-blind treatment and observation
V1 V2 TC1 V3 V4 V5 V6 V7 V8
Screening visit
Baseline injection
Assessment visits
End-of-study visit
Placebo
Inco/A
Individually adjusted dose
-2 0 4 8 12 16 20 24
Weeks

Randomization: 2 (Inco/A):1 (placebo).
Inco/A, incobotulinumtoxinA; SCR, screening; TC, telephone contact; V, visit.

- Study treatment was administered into the muscles of the wrist and, optionally, into the elbow and/or shoulder, based on kinematic tremor analysis (**Figure 2**). Total dose was aided by the TremorTek decision support protocol and was at the discretion of the investigator within stipulated limits (**Table 1**). The actual dose per muscle was planned on the basis of kinematic tremor analysis.

Figure 2. Kinematic tremor assessment flow

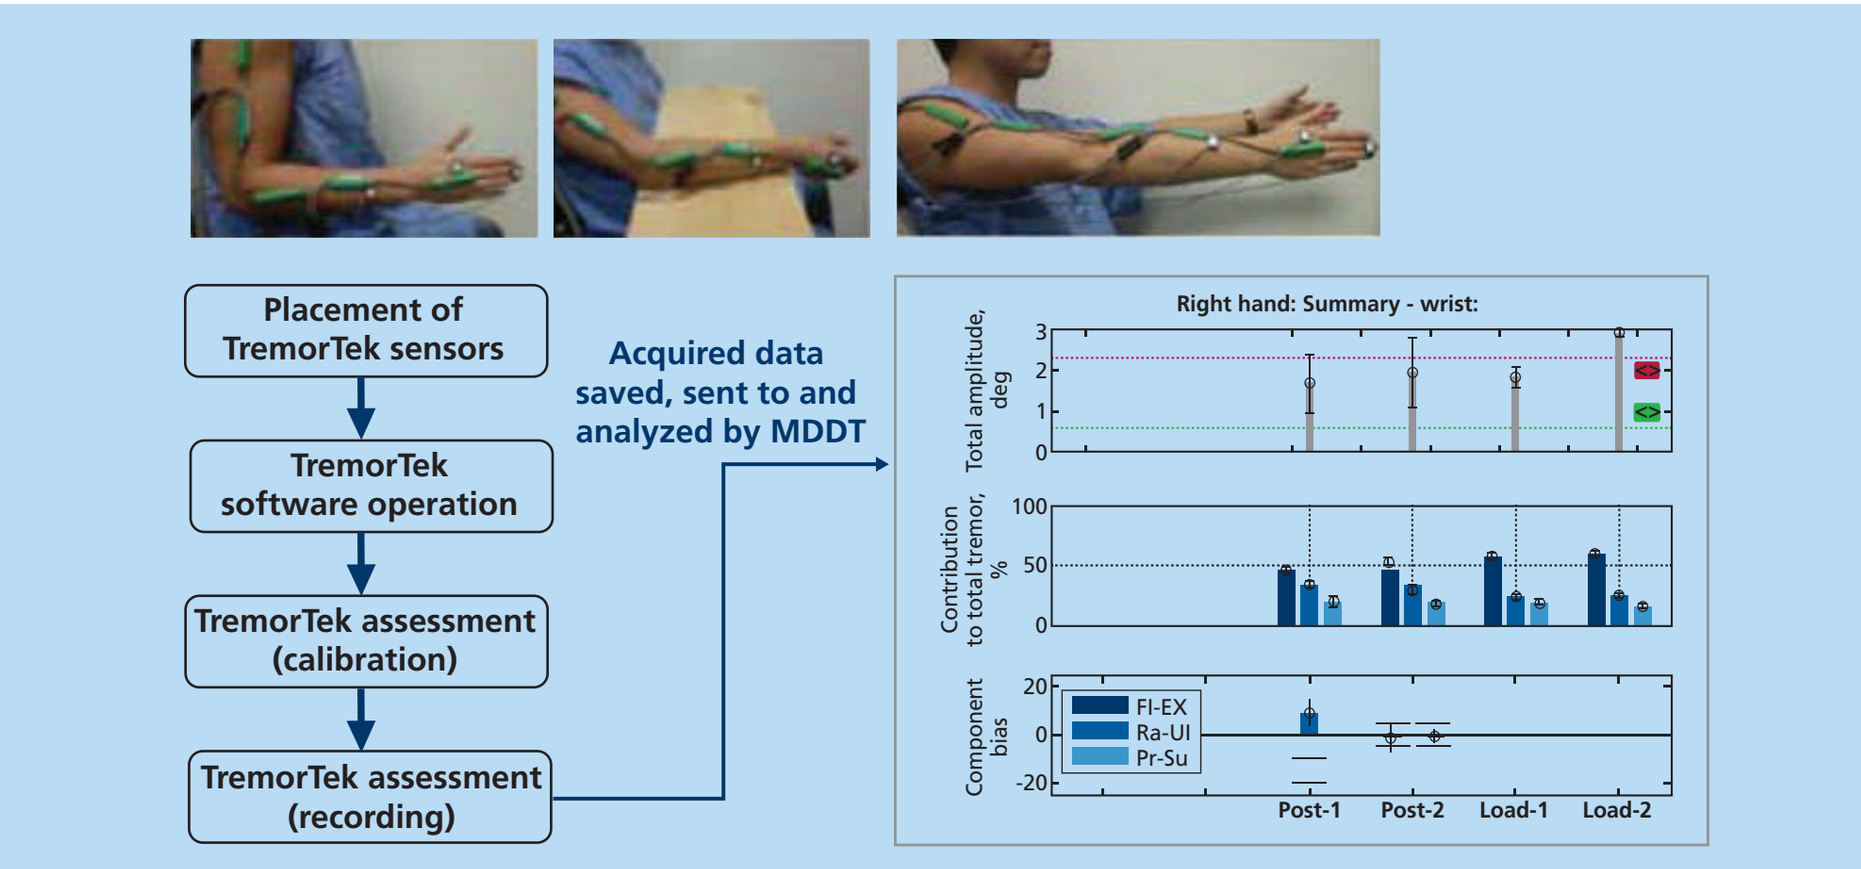
Placement of TremorTek sensors
TremorTek software operation
TremorTek assessment (calibration)
TremorTek assessment (recording)
Acquired data saved, sent to and analyzed by MDDT
Right hand: Summary - wrist:
Total amplitude, deg
Contribution to total tremor, %
Component bias
Fl-EX
Ra-Ul
Pr-Su
Post-1 Post-2 Load-1 Load-2

MDDT, Movement Disorders Diagnostic Technologies.

Table 1. Dosing guidance

Level	Target muscles	Range of injection sites	Dose range per muscle overall	Maximum total dose
Shoulder (treatment of at least 2 out of 4 muscles)	M. pectoralis major (adduction, flexion)	1–2	10–40 U	60 U
	M. latissimus dorsi (extension)	1–2	10–40 U	
	M. deltoid (abduction)	1–3	10–40 U	
	M. supraspinatus (abduction)	1–3	10–40 U	
Elbow	M. brachialis (flexion, supination)	2	15–40 U	60 U
	M. triceps brachii (extension)	2	15–40 U	
Wrist	M. flexor carpi radialis (flexion, radial deviation)	1	5–20 U	80 U
	M. flexor carpi ulnaris (flexion, ulnar deviation)	1	5–20 U	
	M. extensor carpi radialis (extension, radial deviation)	1	5–20 U	
	M. extensor carpi ulnaris (extension, ulnar deviation)	1	5–20 U	
	M. pronator teres (pronation)	1	5–20 U	
	M. supinator (supination)	1	5–20 U	

Table 2. Adverse events of special interest

MedDRA preferred term (MedDRA version 18.0)		
Accommodation disorder	Eyelid function disorder	Paresis cranial nerve
Areflexia	Eyelid ptosis	Peripheral nerve palsy
Aspiration	Facial paresis	Peripheral paralysis
Botulism	Hemiparesis	Pelvic floor muscle weakness
Bradycardia	Hypoglossal nerve paresis	Pneumonia aspiration
Bulbar palsy	Hyporeflexia	Pupillary reflex impaired
Constipation	Hypotonia	Quadriparesis
Cranial nerve palsies, multiple	Ileus paralytic	Respiratory arrest
Cranial nerve paralysis	Monoparesis	Respiratory depression
Diaphragmatic paralysis	Muscular weakness	Respiratory failure
Diplopia	IIIrd nerve paresis	Speech disorder
Dry mouth	IVth nerve paresis	Trigeminal nerve paresis
Dysarthria	VIIth nerve paralysis	Urinary retention
Dysphagia	Paralysis	Vision blurred
Dysphonia	Paralysis flaccid	Vocal cord paralysis
Dyspnea	Paraparesis	Vocal cord paresis
Extraocular muscle paresis	Paresis	

MedDRA, Medical Dictionary for Regulatory Activities.

- Efficacy outcomes included a change from baseline to Week 4 in maximum log-transformed accelerometric tremor amplitude at wrist level (kinematic analysis), Fahn-Tolosa Marin (FTM) Part B motor performance score, Quality of Life in Essential Tremor (QUEST), and Global Impression of Change Scale (GICS).
- The incidence of adverse events of special interest (AESI) as a possible indication of toxin spread was monitored by active questioning as part of the safety analysis (**Table 2**).

Study population

- Patients were randomized in a 2:1 ratio to receive either Inco/A (n=19) or placebo (n=11).
- Patients were required to meet the following inclusion criteria:
 - a diagnosis of 'definite ET'
 - first onset of ET ⩾6 months before screening, with stability of symptoms over 4 weeks
 - moderate-to-marked upper-limb postural and/or kinetic tremor at wrist level, (FTM Part C, items 17–23) in the limb to be treated with a rating of ⩾2 in ⩾2 categories
 - tremor deemed by the investigator to require treatment with 30–200 U Inco/A for wrist (mandatory, minimum 30 U) and elbow or shoulder joints
 - concomitant anti-tremor medication on a stable dose.
- Patients meeting any of the following criteria were excluded from the study:
 - any abnormal neurologic signs other than tremor and Froment's maneuver
 - prior exposure to lithium, valproic acid, amiodarone, or neuroleptics
 - significant trauma to the central nervous system or the nerves of the target limb during the 3 months preceding onset of tremor
 - historic or clinical evidence of psychogenic origins of tremor.

Results

Study population

- Patient demographics were broadly similar at baseline (**Table 3**).
- Mean injected units of Inco/A are shown in **Table 4**. For most patients (78.9% and 72.7% for Inco/A and placebo groups, respectively), the actual dose of study treatment was equal to that planned following kinematic tremor analysis.

Table 3. Patient demographics; safety evaluation set

	Inco/A N=19	Placebo N=11	Total N=30
Female, n (%)	10 (52.6)	5 (45.5)	15 (50.0)
Age, years			
Mean ± SD	68.1 ± 10.6	68.2 ± 10.2	68.1 ± 10.3
Min, max	41, 88	49, 85	41, 88

Inco/A, incobotulinumtoxinA; SD, standard deviation.

Efficacy outcomes

- Treatment with Inco/A significantly improved accelerometric wrist tremor amplitude versus placebo at Weeks 4 and 8 (sensitivity analysis by analysis of covariance [ANCOVA]: Week 4, p=0.004; Week 8, p<0.001) (**Figure 3**).

Table 4. Mean injected units of Inco/A; safety evaluation set

Clinical pattern	Inco/A N=19	
Muscle	**n**	**Dose, U; mean ± SD**
Total	19	116.3 ± 53.0
Wrist (mandatory treatment)		
Total	19	47.4 ± 18.1
Extensor carpi radialis	14	9.6 ± 6.0
Extensor carpi ulnaris	11	8.6 ± 3.9
Flexor carpi radialis	16	10.6 ± 5.1
Flexor carpi ulnaris	16	7.2 ± 2.6
Pronator quadratus	13	8.1 ± 4.8
Pronator teres	14	8.9 ± 4.5
Supinator	15	10.3 ± 6.4
Shoulder (optional treatment)		
Total	14	45.0 ± 7.6
Deltoideus	5	9.0 ± 2.2
Latissimus dorsi	12	11.3 ± 3.8
Pectoralis major	14	21.1 ± 4.5
Supraspinatus	14	11.1 ± 4.0
Teres major	0	0.0
Elbow (optional treatment)		
Total	16	42.5 ± 13.9
Biceps brachii	2	30.0 ± 0.0
Brachialis	14	20.0 ± 6.5
Triceps brachii	16	21.3 ± 7.0

Inco/A, incobotulinumtoxinA; N, number of patients; n, number of observations; SD, standard deviation.

Figure 3. Maximum log-transformed accelerometric tremor amplitude at wrist (injected limb); full analysis set, observed cases

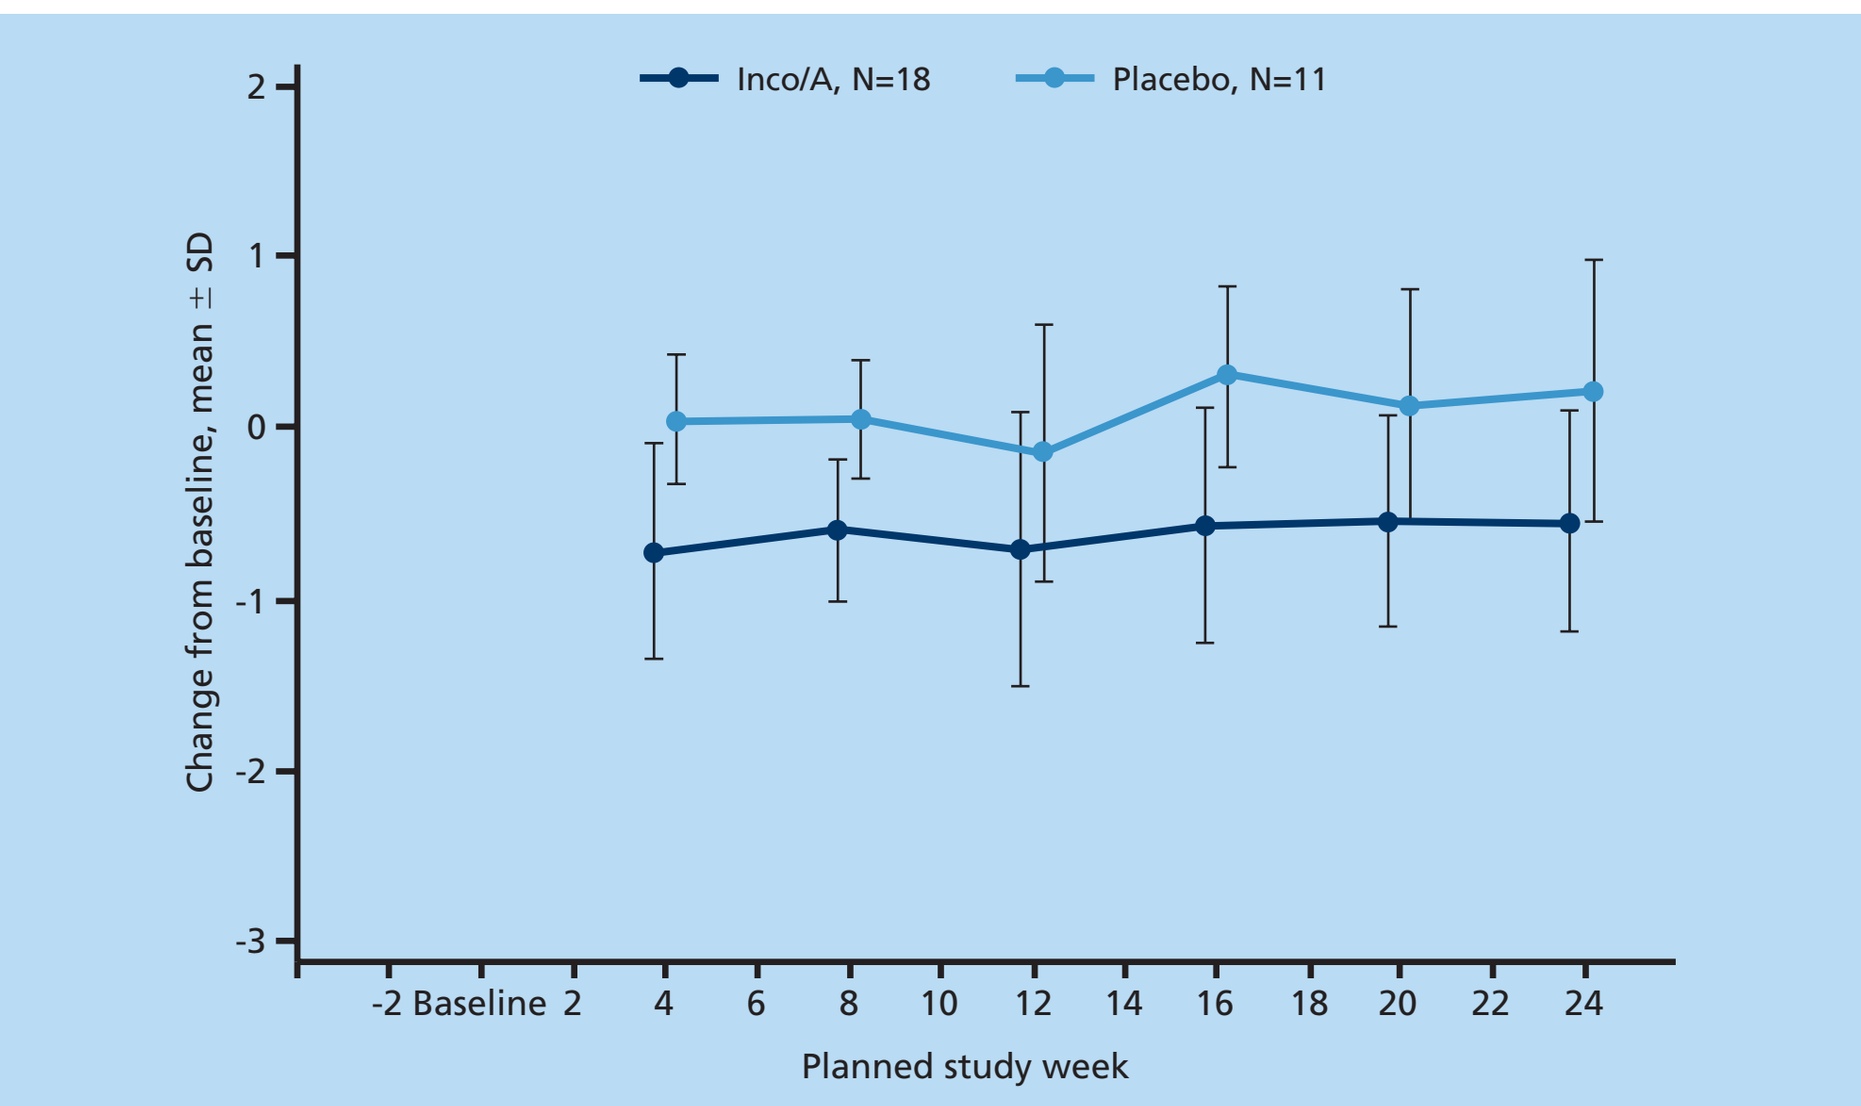
Inco/A, N=18
Placebo, N=11
Change from baseline, mean ± SD
2 1 0 -1 -2 -3
-2 Baseline 2 4 6 8 10 12 14 16 18 20 22 24
Planned study week

Survey		Inco/A N=18	Placebo N=11
Week 4 change	n	18	11
	Mean ± SD	–0.73 ± 0.63	0.05 ± 0.38
	p-value (ANCOVA)	0.004*	
	95% CI (t-test)	–1.21, –0.34*	
Week 8 change	n	18	11
	Mean ± SD	–0.60 ± 0.42	0.05 ± 0.36
	p-value (ANCOVA)	<0.001*	
	95% CI (t-test)	–0.95, –0.33*	

*p-value <0.05 for Inco/A versus placebo.
ANCOVA, analysis of covariance; CI, confidence interval; Inco/A, incobotulinumtoxinA; SD, standard deviation.

- The change from baseline in FTM Part B motor performance score showed significant improvement in the Inco/A group versus placebo at Week 4 and Week 8 (sensitivity analysis by ANCOVA: Week 4, p=0.003; Week 8, p=0.031) (**Figure 4**).
- No significant differences were observed between treatment groups for total QUEST score or the QUEST category sub-scores 'communication', 'work and finance', 'hobbies and leisure', 'physical', and 'psychosocial' at any visit (data not shown).
- Physician-rated GICS indicated a significant improvement in the Inco/A group versus placebo (95% confidence interval 0.2, 1.3) at Week 4. At Week 8 physician-rated GICS, and for both Week 4 and Week 8 patient-rated GICS, there was a non-significant trend towards improvement for Inco/A versus placebo (data not shown).

Figure 4. FTM motor Part B performance score (injected limb); full analysis set, observed cases

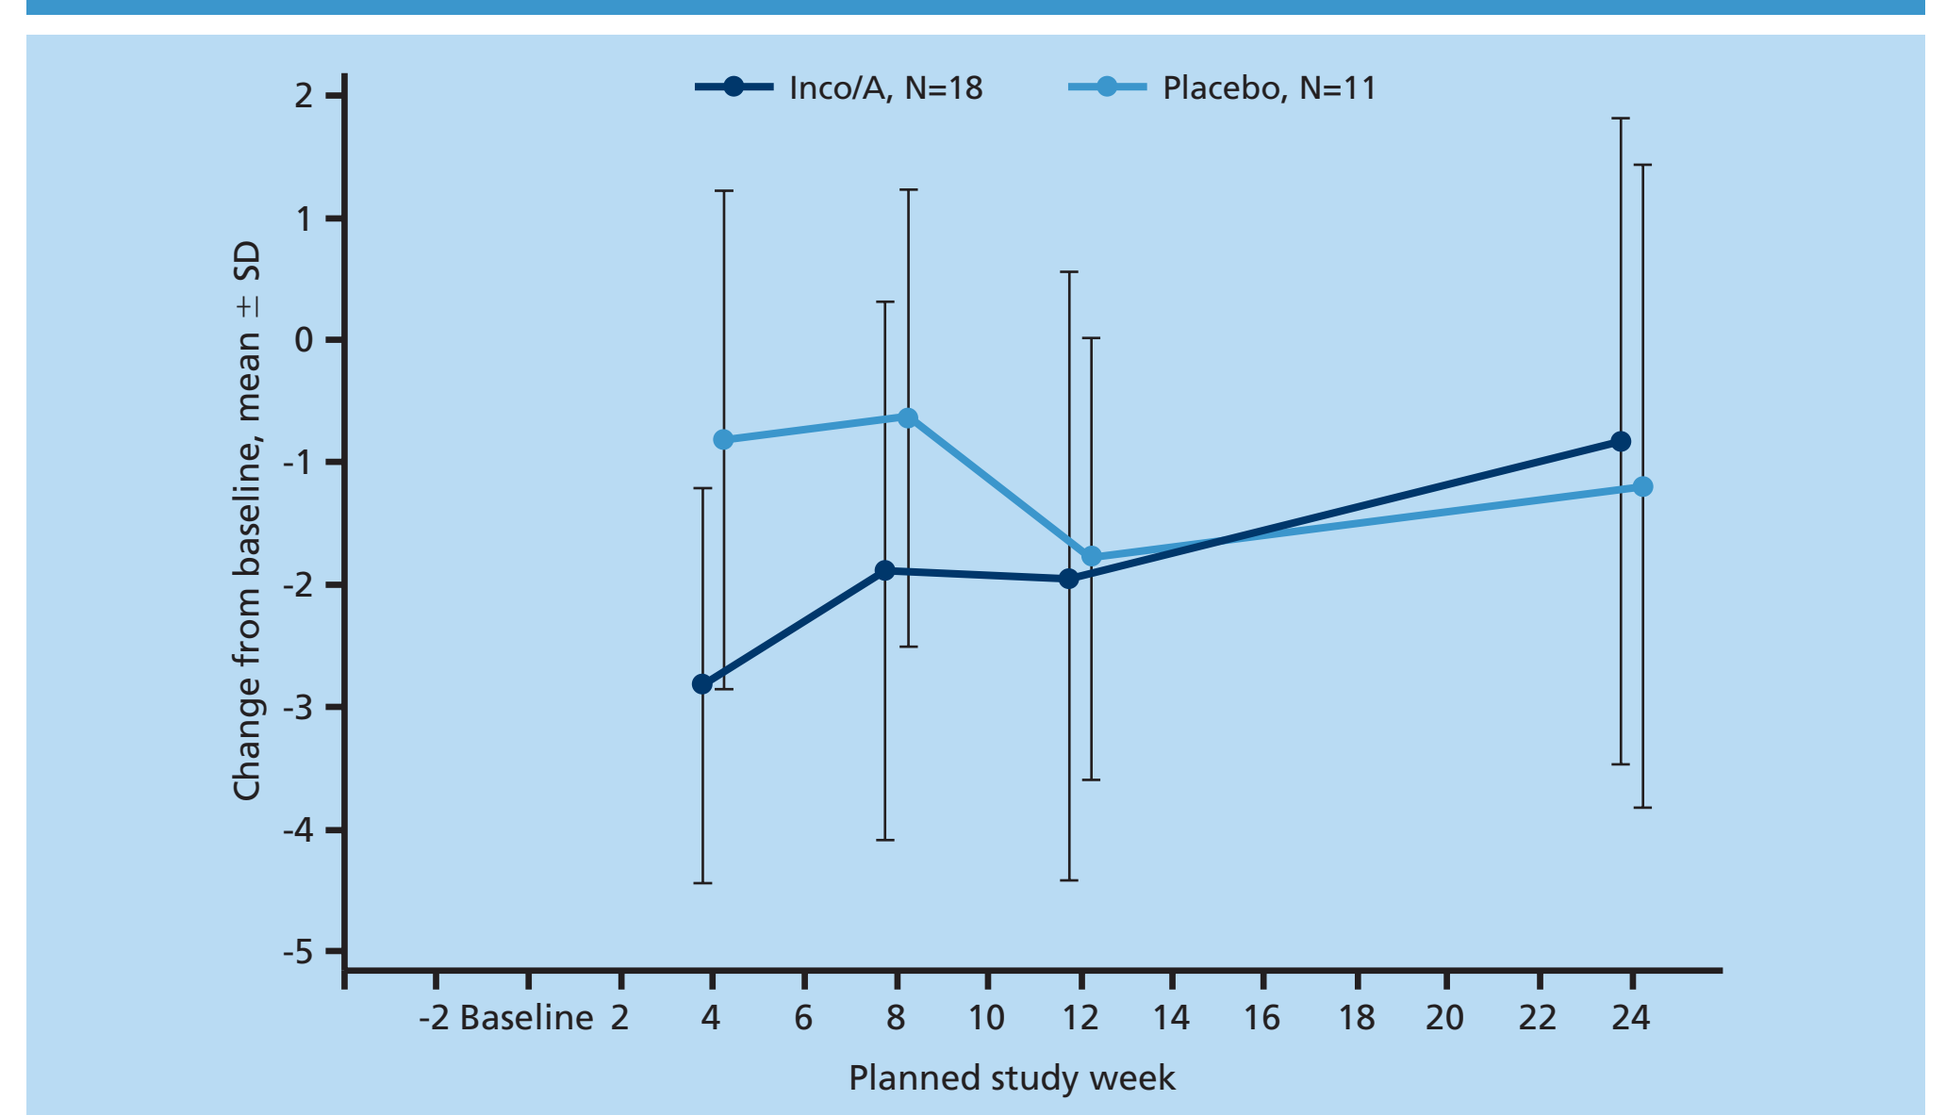
Inco/A, N=18
Placebo, N=11
Change from baseline, mean ± SD
2 1 0 -1 -2 -3 -4 -5
-2 Baseline 2 4 6 8 10 12 14 16 18 20 22 24
Planned study week

Survey		Inco/A N=18	Placebo N=11
Week 4 change	n	18	11
	Mean ± SD	–2.8 ± 1.6	–0.8 ± 2.0
	p-value (ANCOVA)	0.003*	
	95% CI (t-test)	–3.4, –0.6*	
Week 8 change	n	18	11
	Mean ± SD	–19 ± 2.2	–0.6 ± 1.9
	p-value (ANCOVA)	<0.031*	
	95% CI (t-test)	–2.9, 0.4	

*p-value <0.05 for Inco/A versus placebo.
ANCOVA, analysis of covariance; CI, confidence interval; FTM, Fahn-Tolosa Marin; Inco/A, incobotulinumtoxinA; SD, standard deviation.

Table 5. Summary of AEs; safety evaluation set

	Inco/A N=19	Placebo N=11
Number (%) of patients with:		
Any AE	9 (47.4)	6 (54.5)
Any related AE	3 (15.8)	1 (9.1)
Any AESI	3 (15.8)	1 (9.1)
Any related AESI	2 (10.5)	0 (0.0)
Any serious AE	0 (0.0)	2 (18.2)
Any related serious AE	0 (0.0)	0 (0.0)
Any AE leading to discontinuation	0 (0.0)	1 (9.1)
Any related AE leading to discontinuation	0 (0.0)	0 (0.0)
Any fatal AE	0 (0.0)	0 (0.0)
Any related fatal AE	0 (0.0)	0 (0.0)

AEs defined as those with onset or worsening on or after date of the first dose of study treatment.
AE, adverse event; AESI, adverse event of special interest; Inco/A, incobotulinumtoxinA.

Safety outcomes

- The incidence of adverse events was generally similar between treatment groups (**Table 5**). Only localized finger muscle weakness (n=2 in the Inco/A group) and upper respiratory tract infection (n=2 in each group) were reported in >1 patient.
- Although there was some fluctuation in grip strength in the injected limb during the study, there was no significant difference from baseline in either group at the end of the study (Week 24; **Figure 5**).

Figure 5. Grip strength using handheld dynamometers (injected limb); safety evaluation set

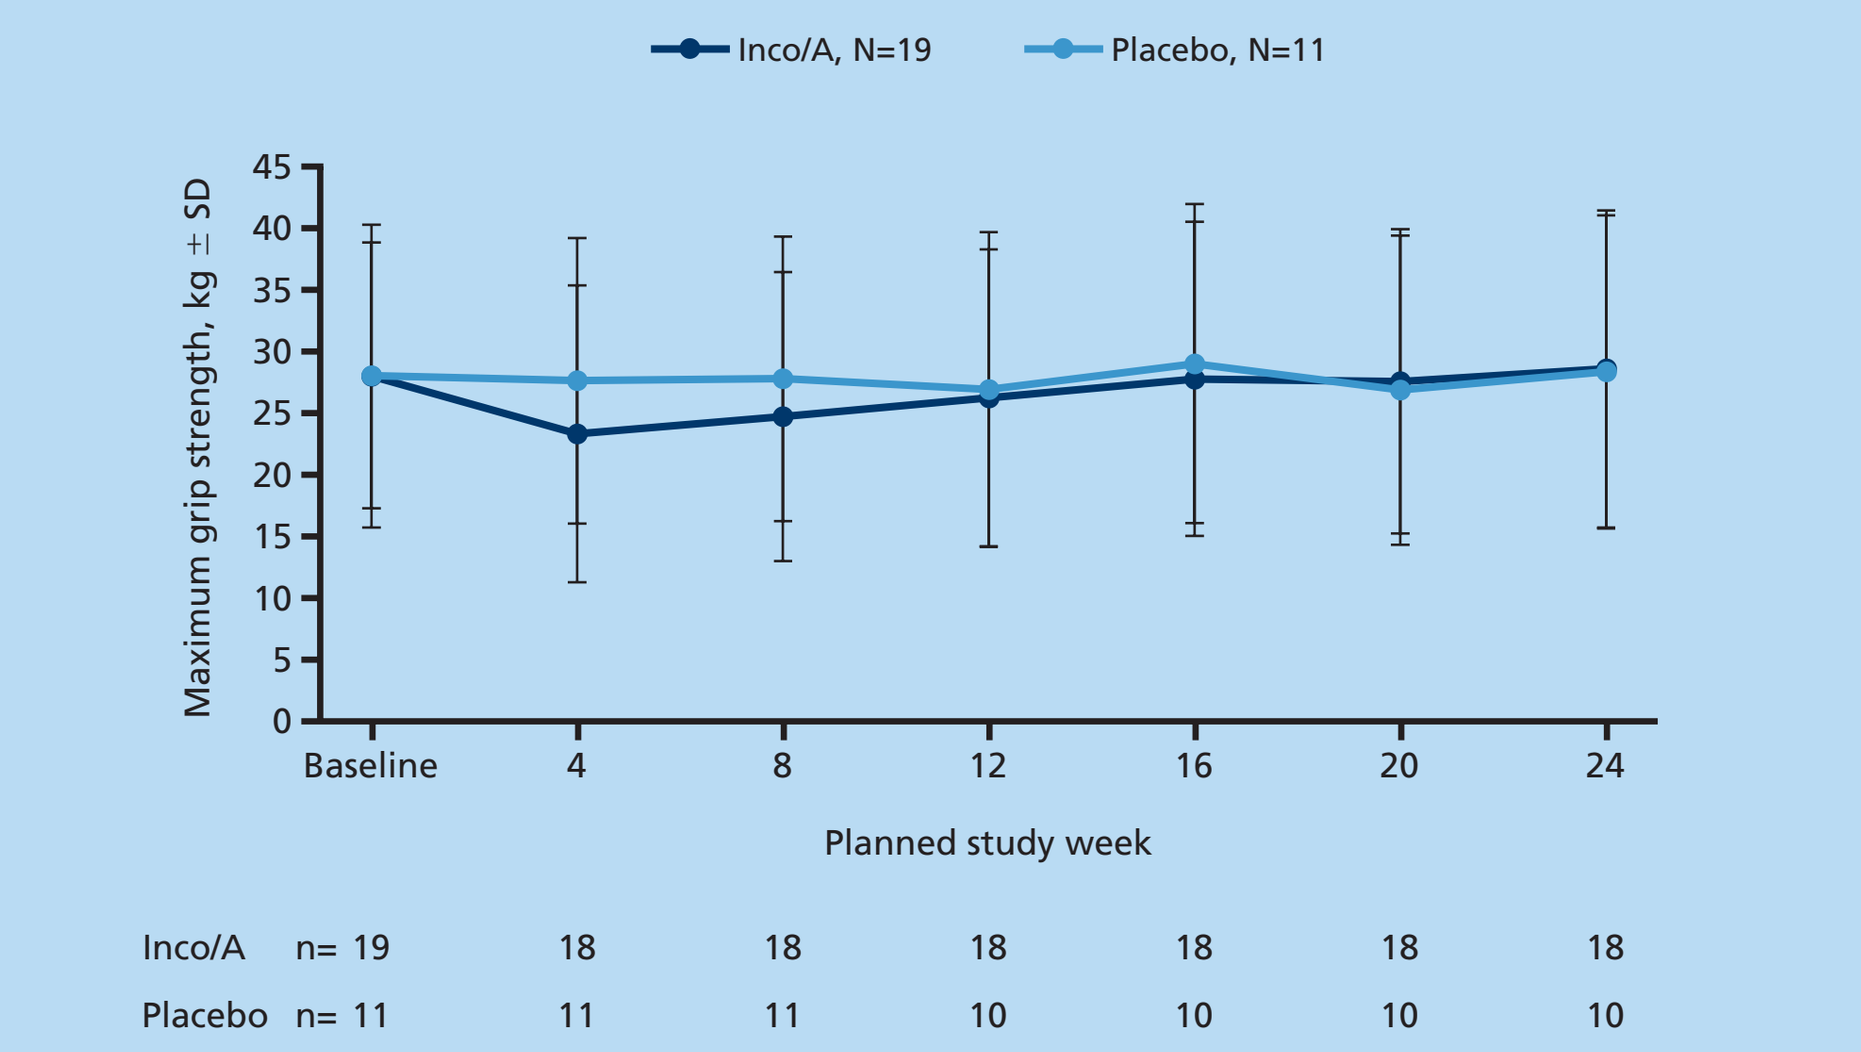
Inco/A, N=19
Placebo, N=11
Maximum grip strength, kg ± SD
45 40 35 30 25 20 15 10 5 0
Baseline 4 8 12 16 20 24
Planned study week
Inco/A n= 19 18 18 18 18 18 18
Placebo n= 11 11 11 10 10 10 10

Inco/A, incobotulinumtoxinA; SD, standard deviation.

Conclusions

- Kinematics-guided Inco/A therapy significantly decreased tremor severity and improved hand function versus placebo in patients with moderate-to-marked essential tremor of the upper limb.
- Individualized dosing based on kinematic tremor analysis was well tolerated, and no premature discontinuation due to muscle weakness was observed over the course of the study.

References

1. Deuschl G et al. Lancet Neurol 2011; 10: 148–161.
2. Koller WC et al. Neurology 1989; 39: 1587–1588.
3. Jankovic J et al. Mov Disord 1996; 11: 250–256.
4. Brin MF et al. Neurology 2001; 56: 1523–1528.
5. Samotus O et al. PLoS One 2016; 11: e0153739.

Acknowledgments

We would like to acknowledge the other investigators on the Essential Tremor Study Team who contributed to the study results: Dr Steven Frucht (NYC), Dr David King (Halifax), Dr Doug Hobson (Winnipeg), Dr Robert Chen (Toronto), Dr Farokh Ismail (Toronto), and Dr Chris Boulias (Toronto).

This study was sponsored by Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Medical writing support was provided by Ellie Finn, PhD, on behalf of Complete Medical Communications, funded by Merz Pharmaceuticals GmbH.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017