Duration of treatment effect of incobotulinumtoxinA (Xeomin®) in upper-limb spasticity

Petr Kaňovský,[1] Michael Althaus,[2] Angelika Hanschmann,[2] Irena Pulte[2]

[1]Faculty of Medicine and Dentistry and University Hospital, Palacky University Olomouc, Olomouc, Czech Republic; [2]Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany

Background

- Repeated injections of botulinum neurotoxin type A are a recommended treatment option for the management of focal spasticity.[1-3]
- Two Phase III studies (Study 3001[4] and Study 0410[5,6]) confirmed the efficacy and safety of incobotulinumtoxinA (Xeomin®; Merz Pharmaceuticals GmbH) in upper-limb post-stroke spasticity.
- Both studies comprised a randomized, placebo-controlled main period (MP), in which patients received one treatment with incobotulinumtoxinA or placebo, and an open-label extension (OLEX).
 - In Study 0410, the OLEX comprised up to five incobotulinumtoxinA treatments with flexible doses of ⩽400 U at flexible intervals of ⩾12 weeks.[5,6] In contrast, the OLEX in Study 3001 comprised three injections with fixed doses of incobotulinumtoxinA at fixed intervals.[4]

Objective

- To evaluate the duration of treatment effect of incobotulinumtoxinA in upper-limb post-stroke spasticity by post-hoc analysis of incobotulinumtoxinA treatment intervals in Study 0410. Flexible treatment intervals in this study allowed the evaluation of treatment effect, in contrast to fixed, 12-week treatment intervals in Study 3001.[4]

Methods

Study design

- During the double-blind, placebo-controlled MP of Study 0410, patients were randomized 1:1 to receive either placebo or ⩽400 U incobotulinumtoxinA, with a follow-up period of 12–20 weeks.[5]
- Patients who completed the MP were then invited to participate in the OLEX, which allowed up to five treatments with flexible doses of ⩽400 U incobotulinumtoxinA at flexible intervals of ⩾12 weeks (**Figure 1**).[5,6]
 - The need for re-injection was mutually agreed between investigators and patients based on pre-specified clinical criteria (patients' expression of need for a new injection, Ashworth Scale scores, investigator's clinical impression).[6]

Figure 1. Design of Study 0410

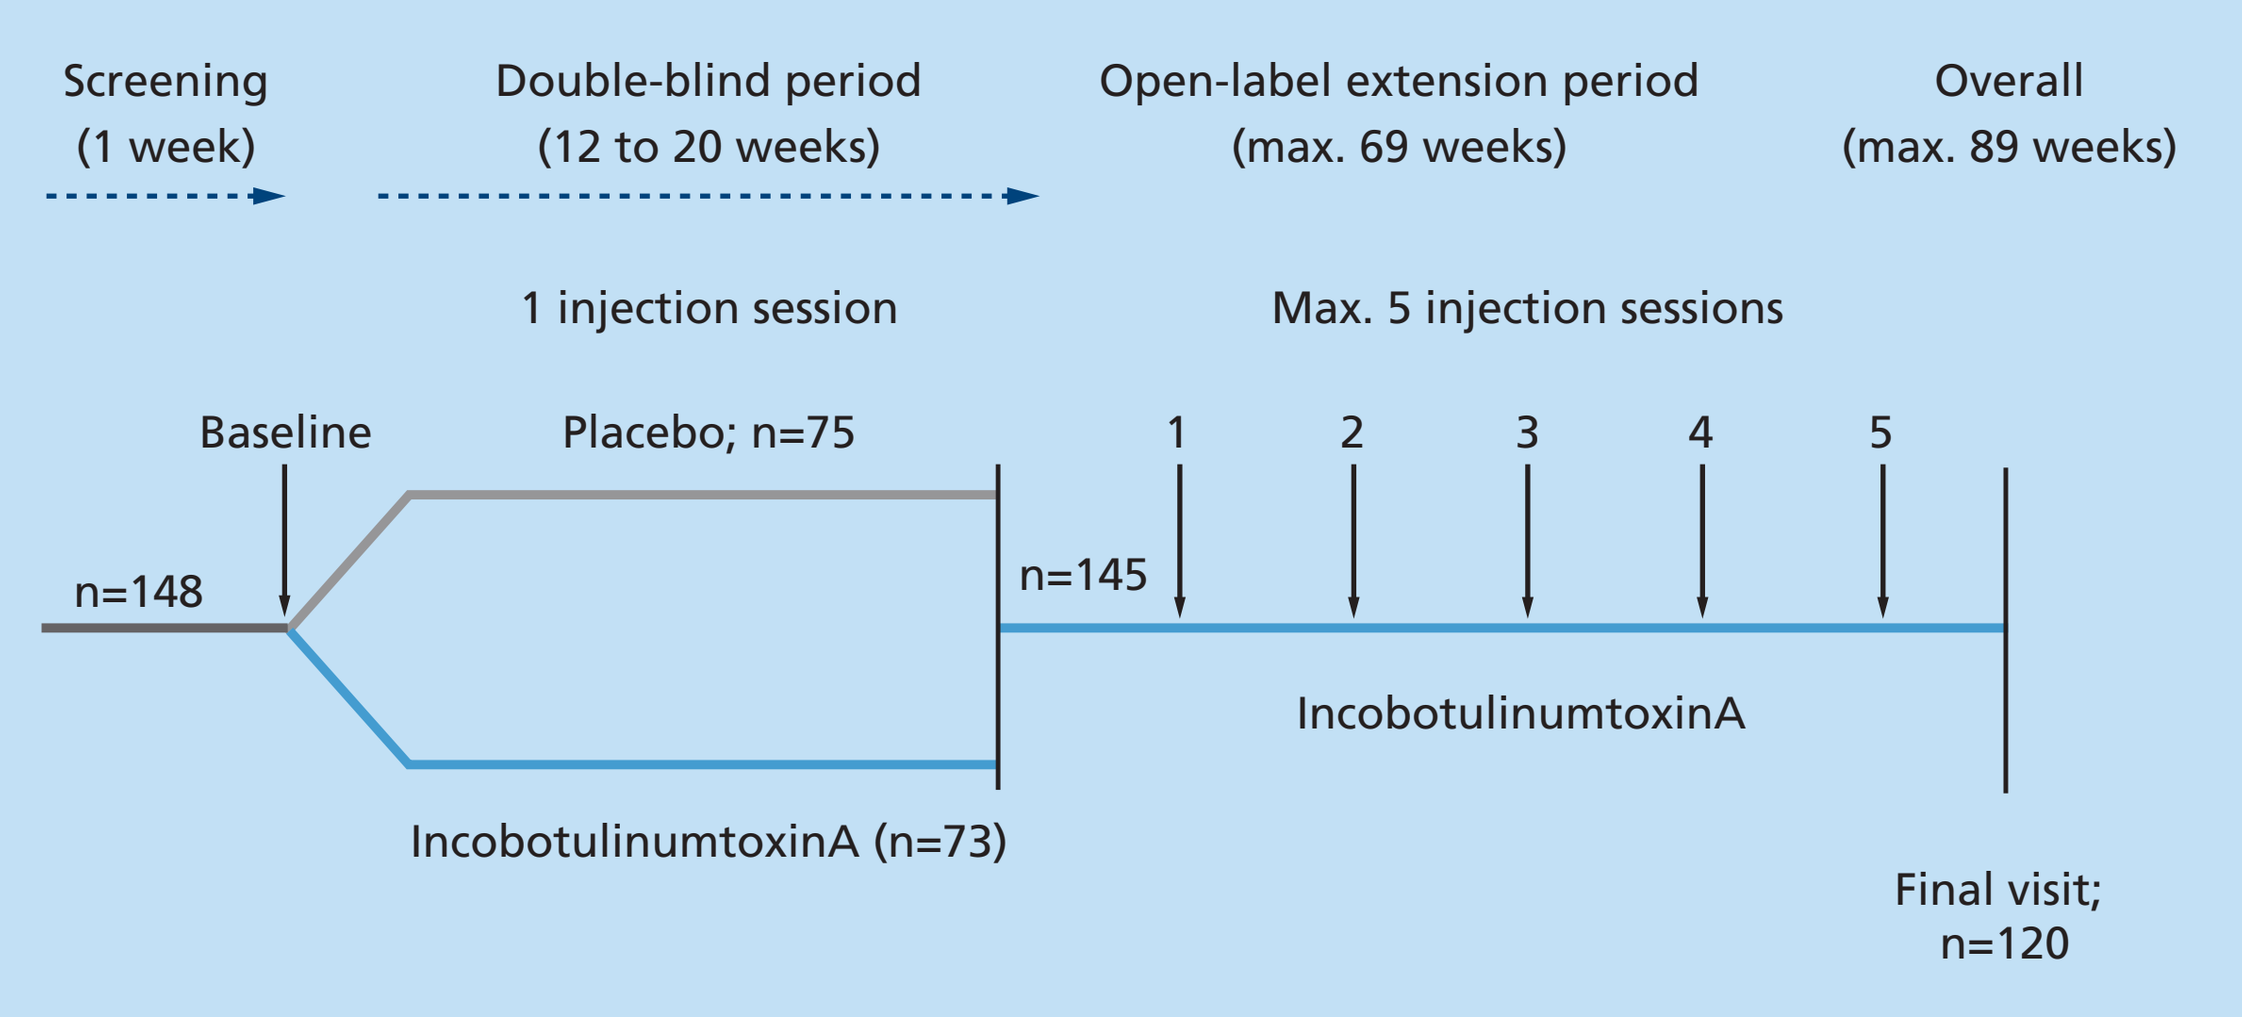
Screening
(1 week)
Double-blind period
(12 to 20 weeks)
Open-label extension period
(max. 69 weeks)
Overall
(max. 89 weeks)
1 injection session
Max. 5 injection sessions
Baseline
Placebo; n=75
1
2
3
4
5
n=148
n=145
IncobotulinumtoxinA
IncobotulinumtoxinA (n=73)
Final visit;
n=120

Outcome measures

- Key outcome measures included muscle tone (Ashworth Scale), Disability Assessment Scale, Global Assessments of Efficacy, and safety outcomes.

Post-hoc analysis

- The post-hoc analysis evaluated all treatment intervals between two consecutive incobotulinumtoxinA injections, excluding treatment intervals prior to the end-of-study visit and with doses <300 U.
- To account for outliers and factors other than a medical need for re-injection (e.g. visit scheduling), duration thresholds were applied and the number of patients with at least one treatment interval above the threshold(s) was calculated.

Results

Patients

- 98.0% of patients (145/148) completed the MP and entered the OLEX; 120 patients completed the OLEX. Patient baseline characteristics are summarized in **Table 1**.

Table 1. Baseline characteristics of the patient population at the OLEX baseline

Characteristic	N=145
Sex male, n (%)	93 (64.1)
Age, years; mean (SD)	55.7 (12.1)
Time since first diagnosis of spasticity,[a] months; mean (SD)	55.0 (48.7)

[a]At baseline of preceding double-blind main period.
OLEX, open-label extension; SD, standard deviation.

- Most patients (111/145; 76.6%) received at least three incobotulinumtoxinA treatments during the OLEX.
- The median dose of incobotulinumtoxinA received during the OLEX ranged from 385 U to 400 U.
- All doses administered during the MP and the OLEX are summarized in **Table 2**.

Table 2. Doses of incobotulinumtoxinA administered and frequency of administration during Study 0410

IncobotulinumtoxinA dose, U	Number of injections	Percentage of injections
300–400	344	99.1
>400	3	0.9

Key efficacy and safety outcomes

- IncobotulinumtoxinA significantly improved muscle tone from each of the first four OLEX injection sessions to their respective control visits 4 weeks later in all upper-limb muscle groups that were treated ($p<0.0001$). The Ashworth Scale responder rates were up to 80.6% at the control visits 4 weeks after Injection Sessions 1–4 (**Figure 2**).
 - Most patients did not show a clinical need for a fifth re-injection in the OLEX. Furthermore, as calculation of this interval was from injection until end of study visit, rather than from injection to injection, a meaningful analysis of this interval was not appropriate.

Figure 2. Ashworth Scale responder rates 4 weeks after each injection in the OLEX

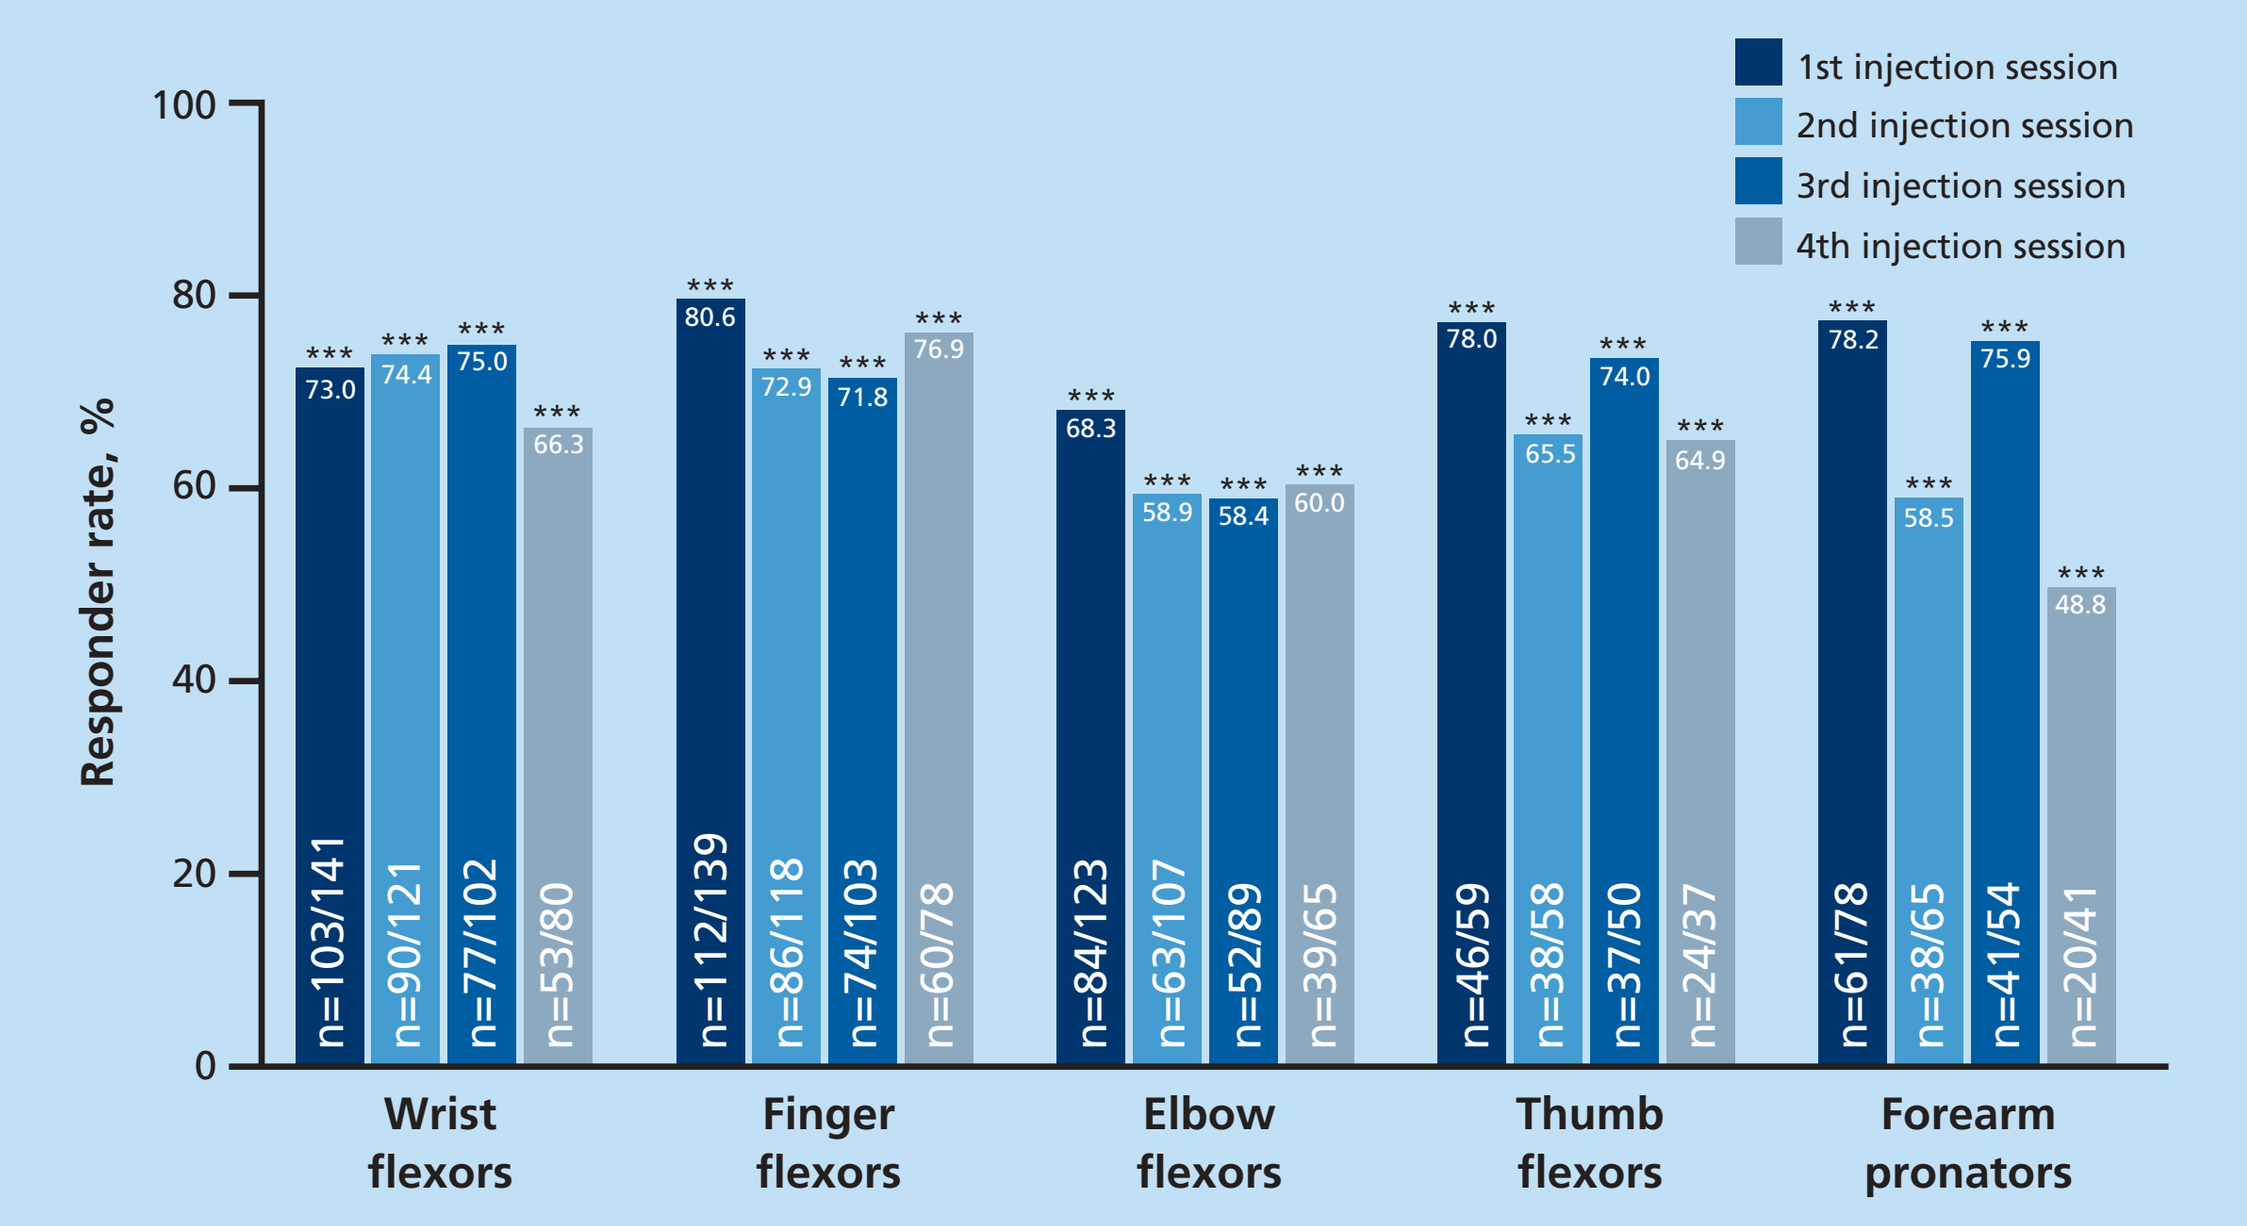
1st injection session
2nd injection session
3rd injection session
4th injection session
Responder rate, %
0
20
40
60
80
100
Wrist flexors: 73.0 (n=103/141), 74.4 (n=90/121), 75.0 (n=77/102), 66.3 (n=53/80)
Finger flexors: 80.6 (n=112/139), 72.9 (n=86/118), 71.8 (n=74/103), 76.9 (n=60/78)
Elbow flexors: 68.3 (n=84/123), 58.9 (n=63/107), 58.4 (n=52/89), 60.0 (n=39/65)
Thumb flexors: 78.0 (n=46/59), 65.5 (n=38/58), 74.0 (n=37/50), 64.9 (n=24/37)
Forearm pronators: 78.2 (n=61/78), 58.5 (n=38/65), 75.9 (n=41/54), 48.8 (n=20/41)

***$p<0.0001$ (Wilcoxon signed-rank test for change from injection session to control visit).
The number of patients (n=16) who received 5 injection sessions during the OLEX was too low to allow a meaningful analysis for this interval.
OLEX, open-label extension.

- 11% of patients (n=16) experienced adverse events that were considered to be treatment-related.
 - The most common, related, treatment-emergent adverse events were muscular weakness (n=5), injection-site pain (n=4), dysphagia (n=2), and pain in the extremity (n=2).

Treatment intervals

- A total of 347/437 incobotulinumtoxinA treatment intervals were included in the post-hoc analysis, with the earliest re-injection at Week 9 and the latest at Week 49 (**Figure 3a**).
- More than half (54.8%) of incobotulinumtoxinA re-injections were given at Week ⩾14. 32.6% of incobotulinumtoxinA re-injections were administered at Week ⩾16, 19.6% at Week ⩾18, and 11.5% at Week ⩾20 (**Figure 3a**).
- For comparison, 30.7% of the intervals between a placebo injection in the MP and the first incobotulinumtoxinA treatment in the OLEX were at Week ⩾14, 17.3% at Week ⩾16, and 6.7% at Week ⩾18 (**Figure 3b**).

Figure 3. Distribution of treatment interval length in weeks for all injection cycles

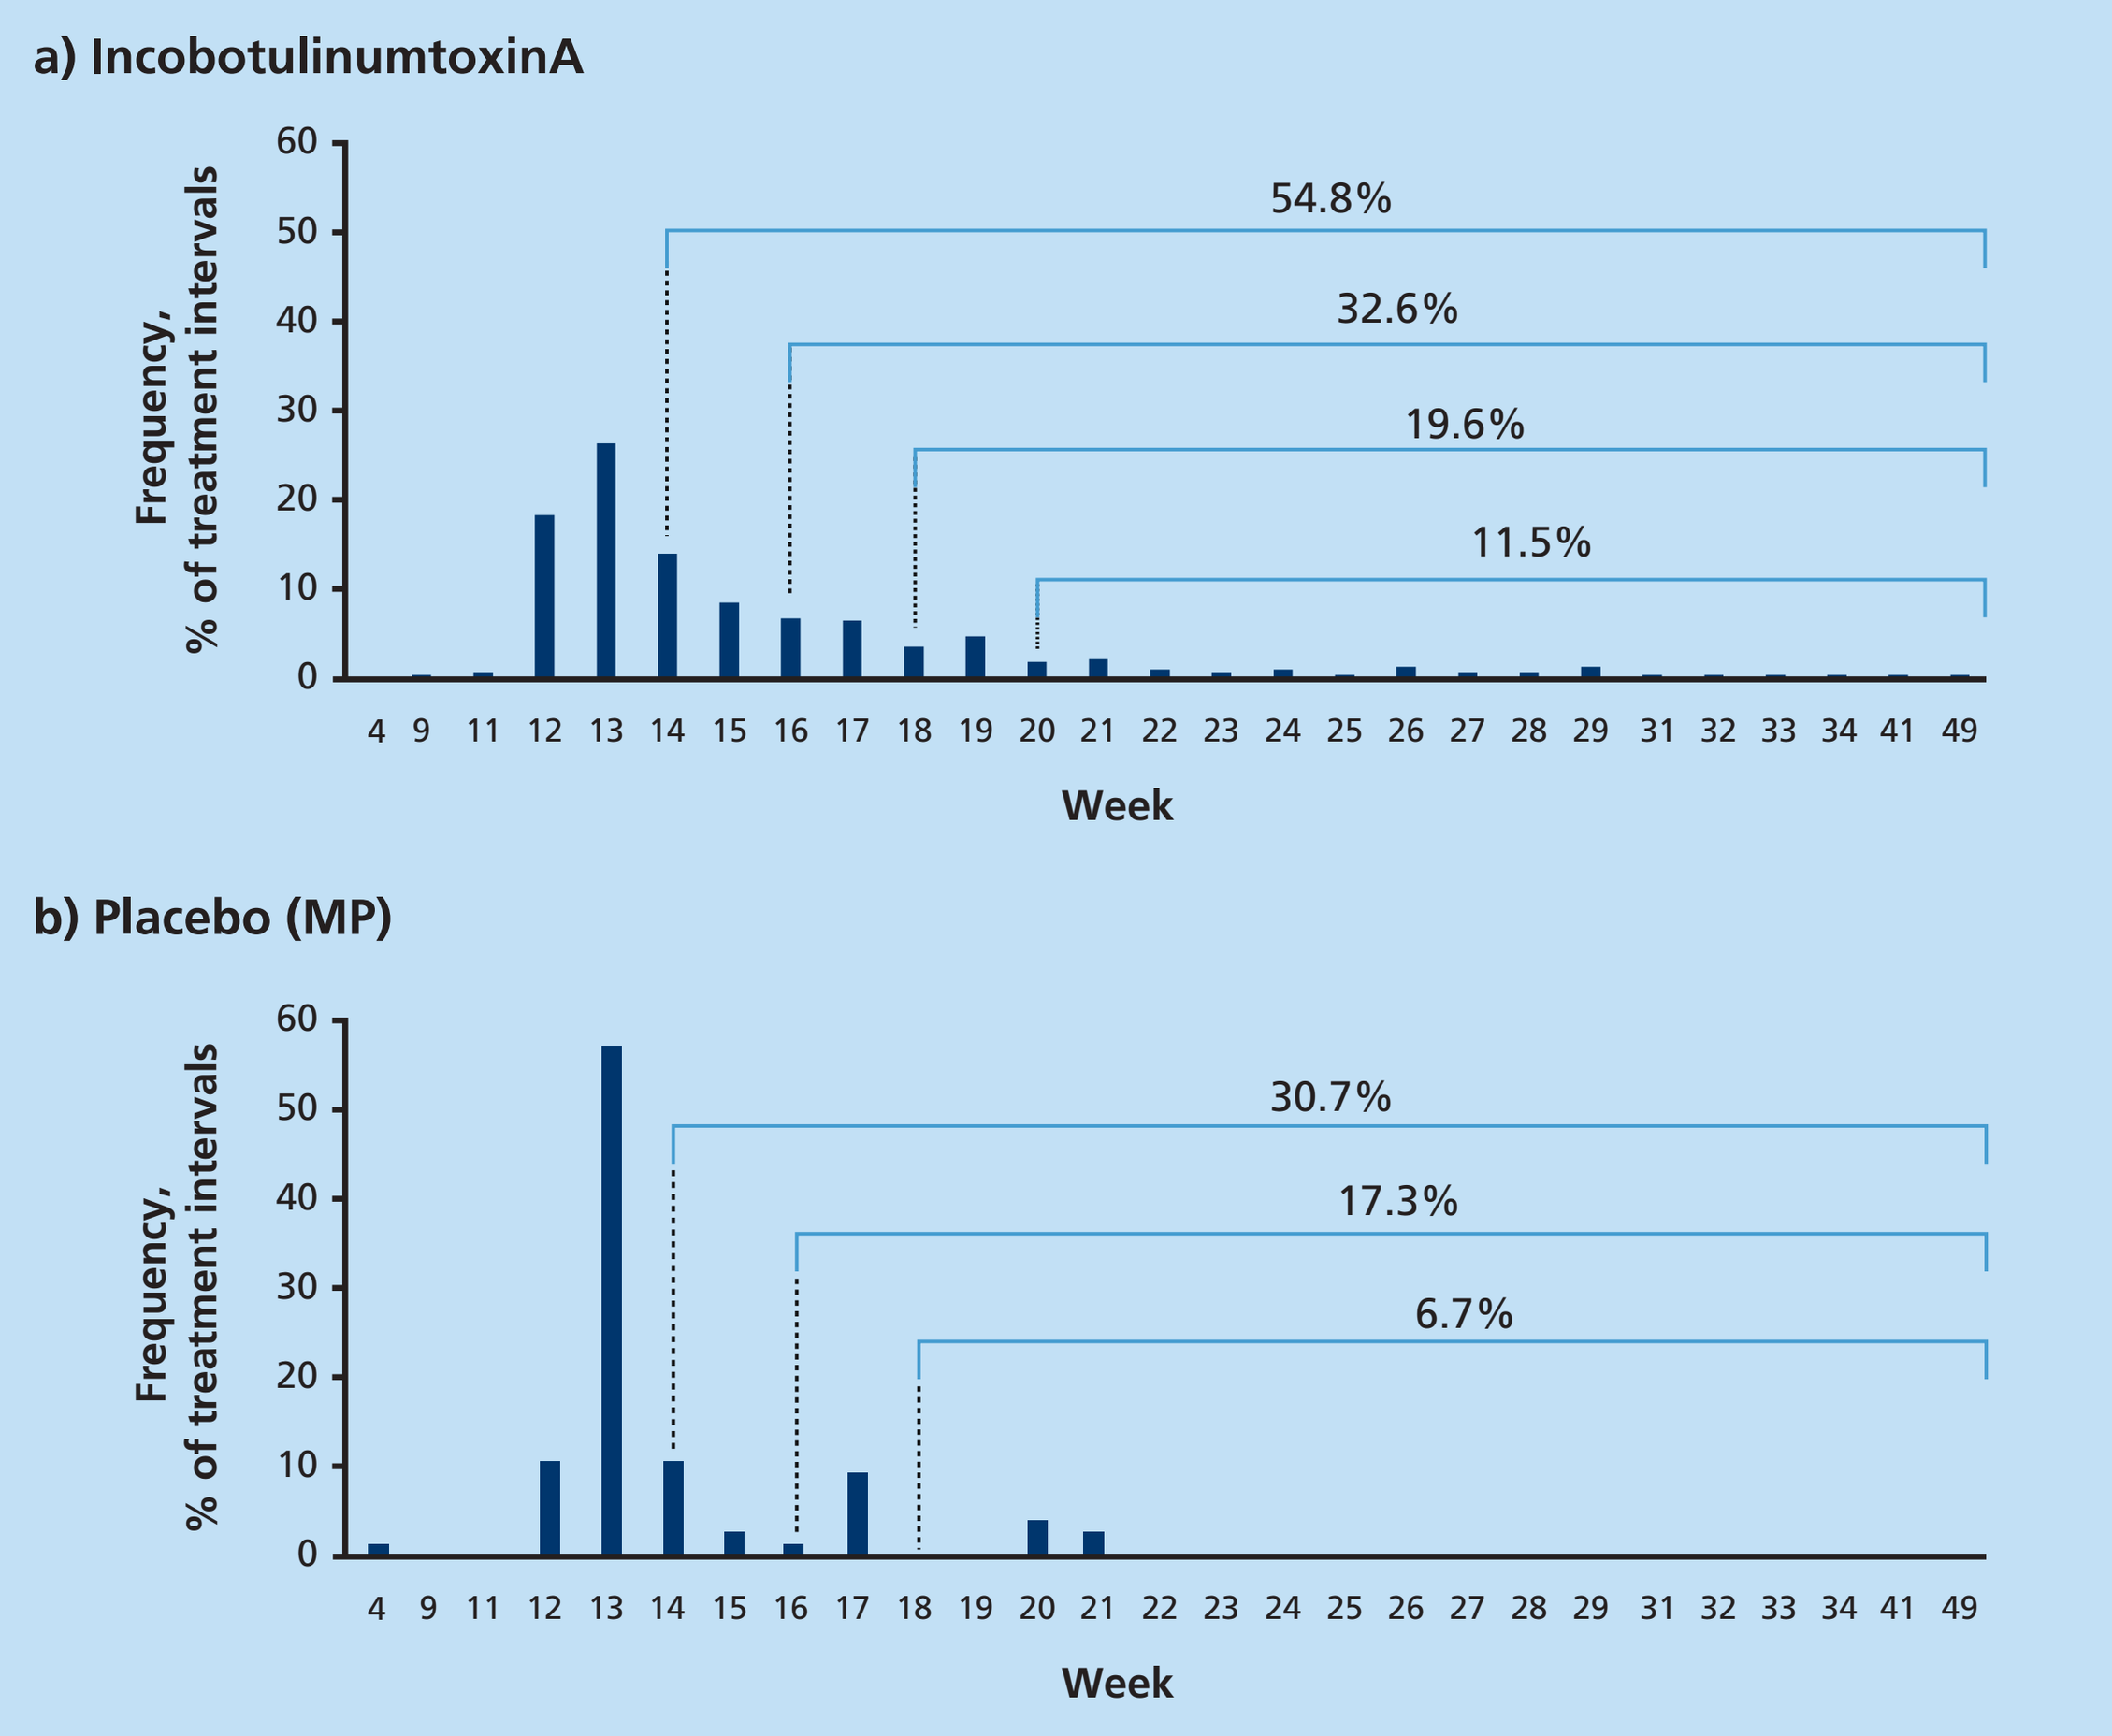
a) IncobotulinumtoxinA
Frequency, % of treatment intervals
54.8%
32.6%
19.6%
11.5%
4 9 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28 29 31 32 33 34 41 49
Week
b) Placebo (MP)
Frequency, % of treatment intervals
30.7%
17.3%
6.7%
4 9 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28 29 31 32 33 34 41 49
Week

The highlighted values represent the cumulative frequencies of incobotulinumtoxinA re-injections given at Weeks ⩾14, ⩾16, ⩾18 and ⩾20.
MP, main period.

- The mean ± standard deviation incobotulinumtoxinA treatment interval during the MP was 14.8 ± 2.76 weeks versus 13.8 ± 2.46 weeks in the placebo group ($p<0.05$, t-test).
- The mean of all incobotulinumtoxinA treatment cycle intervals was 15.5 ± 4.63 weeks resulting in $p\leqslant0.001$ (t-test) when compared to the MP placebo cycle.
- More than half of all patients (59.1%) had at least one incobotulinumtoxinA treatment interval at Week ⩾16.
 - This included 33.1% of patients who had one treatment interval, 22.1% who had two intervals, and 3.9% who had three intervals (all at Week ⩾16).
- 42.5% of patients had at least one incobotulinumtoxinA treatment interval at Week ⩾18.
 - This included 33.1% of patients who had one treatment interval, 7.9% who had two intervals, and 1.6% who had three intervals (all at Week ⩾18).
- 26.0% of patients had at least one incobotulinumtoxinA treatment interval at Week ⩾20.
 - This included 21.3% of patients who had one treatment interval, 3.9% who had two intervals, and 0.8% who had three intervals (all at Week ⩾20).

Conclusions

- Repeated injections of incobotulinumtoxinA at doses ⩽400 U, administered at flexible intervals of ⩾12 weeks, were well tolerated and provided sustained efficacy in reducing muscle tone.
- This post-hoc analysis demonstrated individual variability in the duration of incobotulinumtoxinA treatment effect; more than half of patients had at least one treatment interval of ⩾16 weeks.
- Treatment effects of ⩾20 weeks were observed for one in four of the patients.
- These results support individualized and flexible incobotulinumtoxinA dosing intervals for the treatment of upper-limb post-stroke spasticity.

References

1. Simpson DM, et al. Neurology 2016; 86: 1818-1826.
2. Esquenazi A, et al. Eur J Neurol 2010; 17 Suppl 2: 1–8.
3. Wissel J, et al. J Rehabil Med 2009; 41: 13–25.
4. Elovic EP, et al. Muscle Nerve 2016; 53: 415–421.
5. Kaňovský P, et al. Clin Neuropharmacol 2009; 32: 259–265.
6. Kaňovský P, et al. J Rehabil Med 2011; 43: 486–492.

Acknowledgments

This study was sponsored by Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Medical writing support was provided by A.G. McCluskey, PhD, of Complete Medical Communications and funded by Merz Pharmaceuticals GmbH.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017

MRZ82220k_Kanovsky.indd 1
11/01/2017 14:43