Efficacy of incobotulinumtoxinA (Xeomin®) in the treatment of shoulder spasticity due to cerebral causes

Jörg Wissel,[1] Djamel Bensmail,[2] Astrid Scheschonka,[3] Birgit Flatau-Baqué,[3] Olivier Simon,[3] David M Simpson[4]

[1]Department of Neurology, Vivantes Hospital Spandau, Berlin, Germany; [2]Raymond-Poincaré Hospital, AP-HP, University of Versailles Saint Quentin, Garches, France; [3]Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany; [4]Icahn School of Medicine at Mount Sinai Medical Center, New York, NY, USA

Background

- Current European and US guidelines recommend botulinum neurotoxin (BoNT) injections as a treatment for focal spasticity.[1–3]
- A key challenge in treating patients with multifocal upper- and lower-limb spasticity is that the total dose of BoNT needed clinically to effectively treat all spasticity patterns may exceed the doses approved currently.
- Escalating the total body dose of BoNT would reduce the need for physicians to prioritize certain clinical patterns, which may lead to better outcomes for the patient.
- The safe use of higher-than-labeled BoNT doses has been reported[4–10] but not evaluated in large prospective trials.
- IncobotulinumtoxinA (Xeomin®, Merz Pharmaceuticals GmbH), a BoNT-A formulation with a favorable tolerability profile,[4–13] lack of secondary non-response in clinical trials, and high purity,[14] was recently evaluated in a clinical study (the TOWER study: NCT01603459) at doses of up to 800 U.[15]
- Although BoNT is a well-established treatment for upper-limb spasticity, there is a lack of prospective studies that specifically assess efficacy in the shoulder. Previous studies have had low patient numbers or focused on pain management rather than assessment of muscle tone.[16–18]

Objective

- To employ a post-hoc analysis approach to evaluate the effectiveness of incobotulinumtoxinA doses (400–800 U) in treating patients with shoulder spasticity due to cerebral lesions.

Methods

Study design

- The TOWER study (NCT01603459) was a prospective, open-label, dose-escalation study investigating the safety and efficacy of incobotulinumtoxinA in patients (18–80 years of age) with chronic upper- and lower-limb spasticity due to stroke and other cerebral causes who were deemed by the investigator to require total body doses of incobotulinumtoxinA up to 800 U.
- Additional inclusion criteria were as follows:
 - ⩾12 weeks since the last event leading to spasticity on the body side of the selected target clinical pattern
 - focal spasticity with an Ashworth Scale (AS) score ⩾2 points in the joint associated with the target clinical pattern
 - Disability Assessment Scale (DAS) score ⩾2 points in the principal target domain.
- The study comprised three injection cycles (Cycles 1–3) with escalating fixed total body doses of incobotulinumtoxinA (400, 600, and 600–800 U) to be injected in the same body side. This was followed by a 12–16-week observation period that included a follow-up (control) visit 4 weeks post-treatment in each cycle (**Figure 1**).
- In Cycle 1 and Cycle 2, patients could receive injections into the upper limb only, the lower limb only, or into both limbs. In Cycle 3, it was expected that patients would receive injections into both the upper and lower limbs. The maximum dose per limb in Cycle 1 was 400 U, increasing to 600 U in Cycle 2 and Cycle 3 (**Figure 1**).
- Post-hoc analysis using data from the TOWER study assessed the change in muscle tone from baseline visit to control visit 1 in each injection cycle in patients who received treatment in the shoulder and those who did not.

Figure 1. Study design

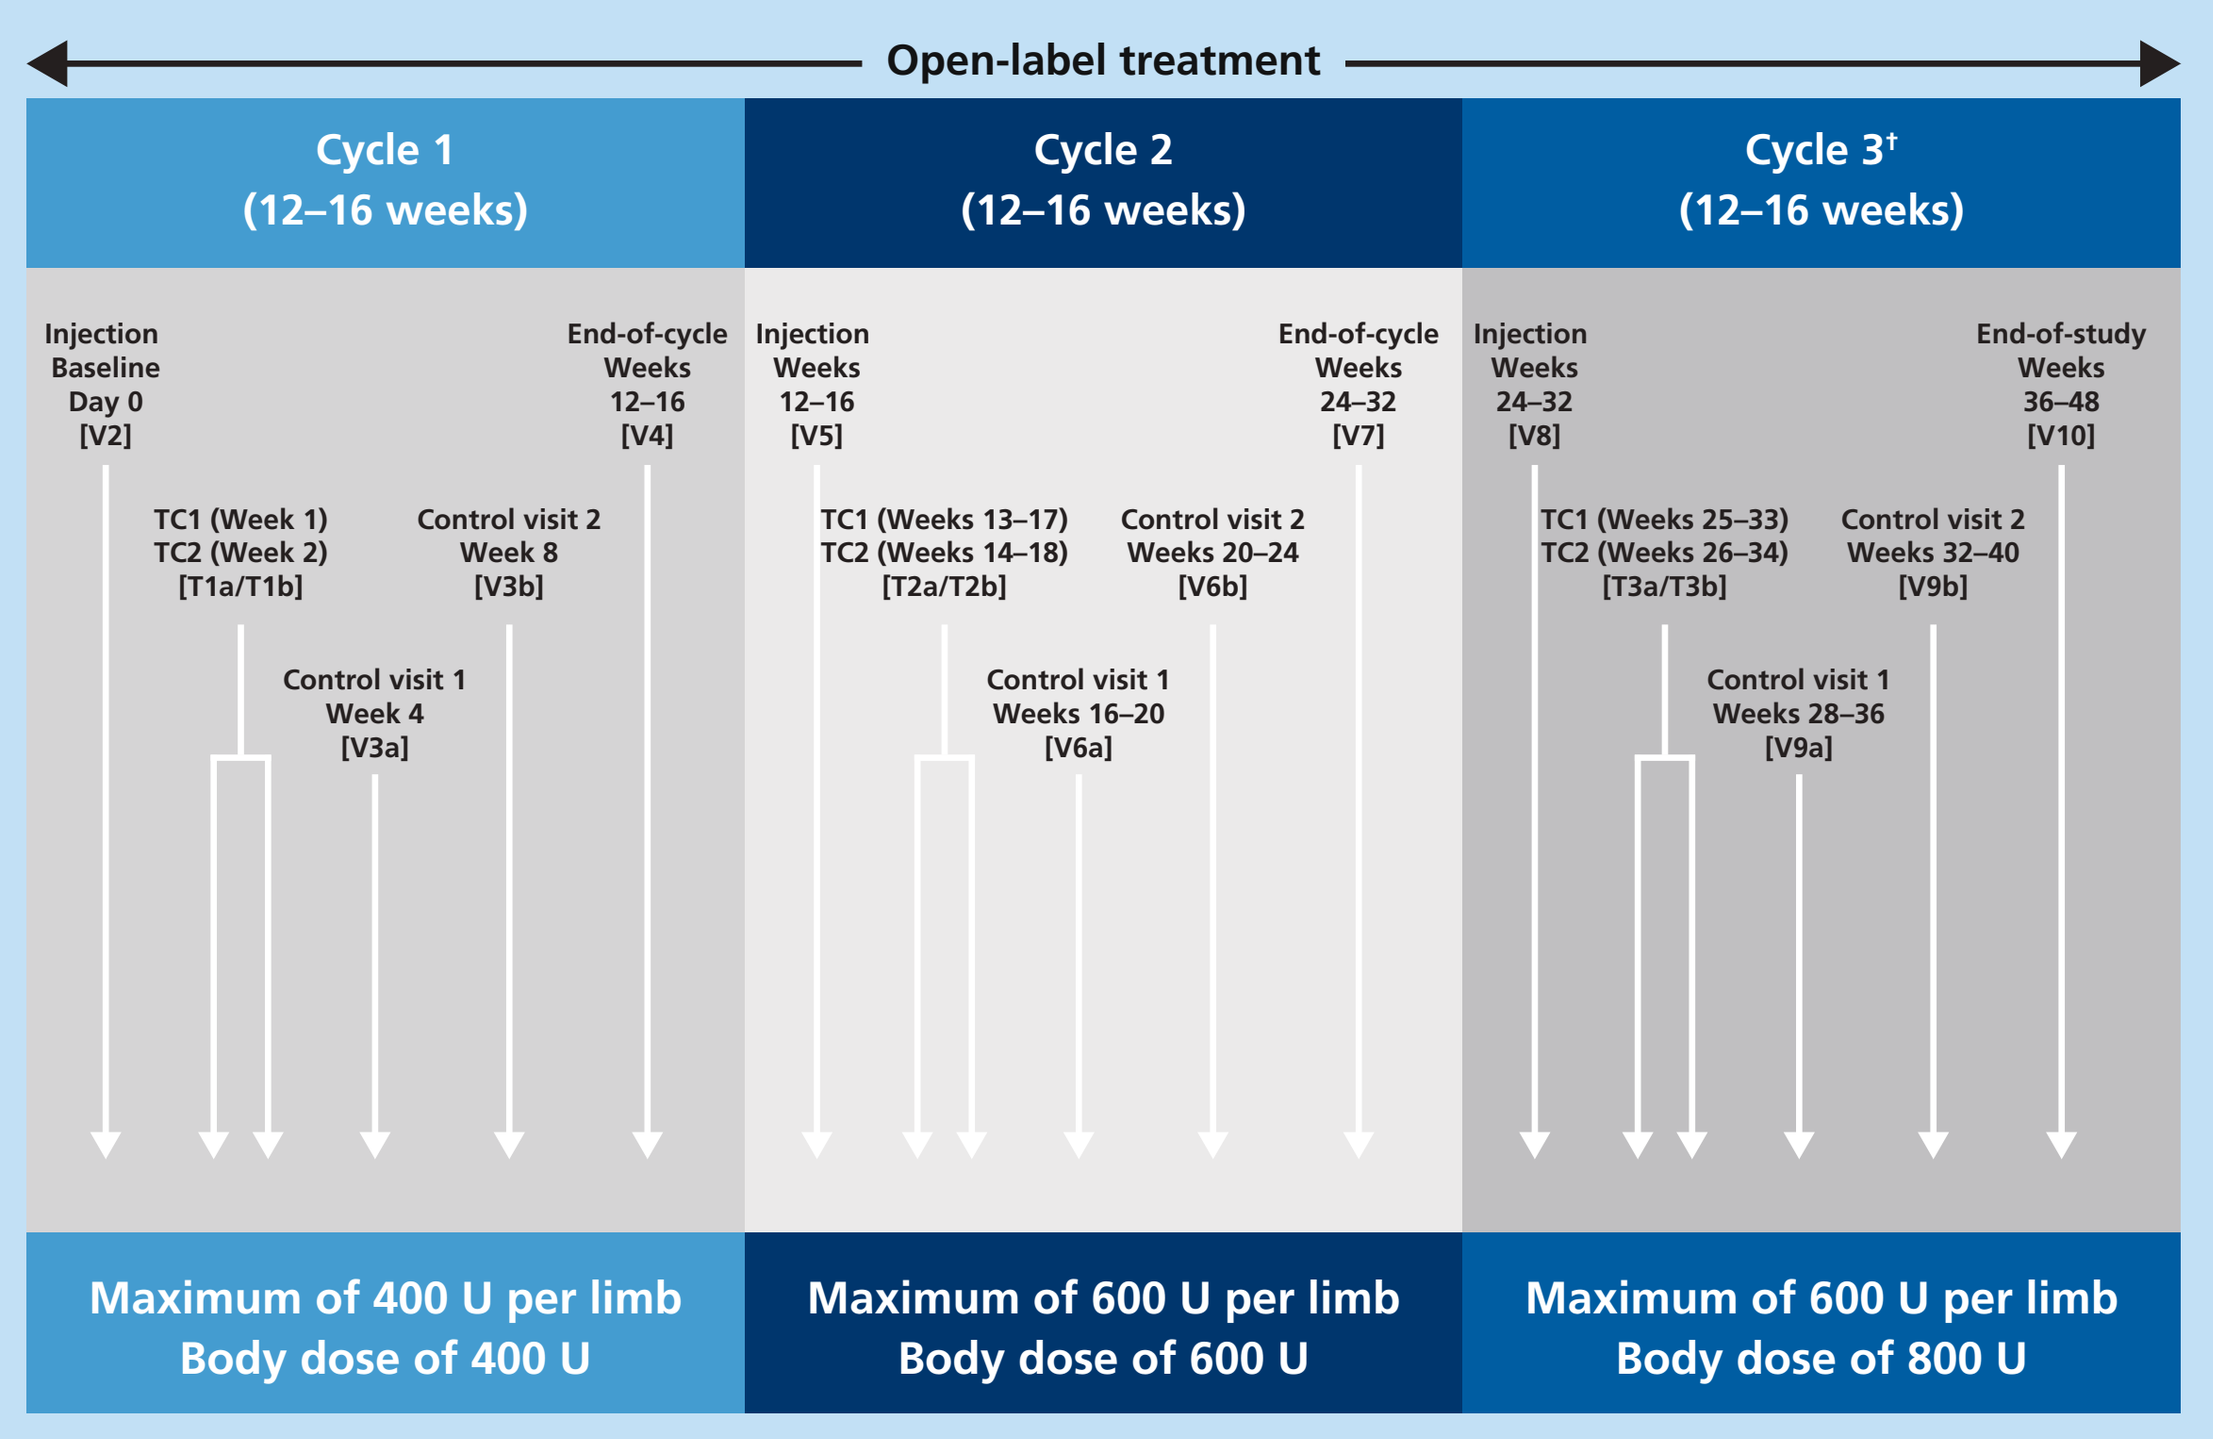
Open-label treatment
Cycle 1 (12–16 weeks)
Cycle 2 (12–16 weeks)
Cycle 3† (12–16 weeks)
Injection Baseline Day 0 [V2]
TC1 (Week 1) TC2 (Week 2) [T1a/T1b]
Control visit 1 Week 4 [V3a]
Control visit 2 Week 8 [V3b]
End-of-cycle Weeks 12–16 [V4]
Injection Weeks 12–16 [V5]
TC1 (Weeks 13–17) TC2 (Weeks 14–18) [T2a/T2b]
Control visit 1 Weeks 16–20 [V6a]
Control visit 2 Weeks 20–24 [V6b]
End-of-cycle Weeks 24–32 [V7]
Injection Weeks 24–32 [V8]
TC1 (Weeks 25–33) TC2 (Weeks 26–34) [T3a/T3b]
Control visit 1 Weeks 28–36 [V9a]
Control visit 2 Weeks 32–40 [V9b]
End-of-study Weeks 36–48 [V10]
Maximum of 400 U per limb Body dose of 400 U
Maximum of 600 U per limb Body dose of 600 U
Maximum of 600 U per limb Body dose of 800 U

†If a dose of 800 U was not justified for clinical or safety reasons, a lower dose of 600–800 U could be administered as an exception.
TC, telephone contact; V, visit.

Outcome measures

- The AS is a well-known and validated scale (scored from 0 to 4; where 0 indicates no increase in tone and 4 indicates that the limb is rigid in flexion or extension) to assess muscle tone and the severity of spasticity by judging resistance to passive movement.[19] In this study, assessment of the AS was carried out over ~1 second at the baseline and control visit of each injection cycle.
- AS scores for shoulder adductors, extensors, and internal rotators were summed to give an AS shoulder sum-score (AS-SSS).
- The assessment of muscle tone using AS was restricted to the treated body side.

Statistical analysis

- Efficacy variables were analyzed in the full analysis set (all patients who received ⩾1 dose of study drug) using descriptive summary statistics.
- Post-hoc analysis included multiple regression of AS-SSS change on shoulder dose, adjusting for the AS-SSS at the respective injection cycle baseline visit in those that received treatment in the shoulder.

Results

Patients

- Of 193 patients screened, 155 were eligible to enroll and received treatment.
- Key patient demographics and baseline characteristics are summarized in **Table 1**.
- Of 155 patients who participated in the TOWER study, spasticity was due to stroke in 85.2% and other neurologic causes in 14.8%.
- The number of patients treated in the shoulder increased at each injection cycle. Of the total number of patients treated in the TOWER study: 33.5% (52/155) were treated in the shoulder in Cycle 1, 45.4% (69/152) in Cycle 2, and 60% (84/140) in Cycle 3 (**Table 2**).
- IncobotulinumtoxinA doses administered to the shoulder in each cycle are summarized in **Table 3.** Mean (SD) doses increased from 71.5 (32.7) U in Cycle 1 to 103.5 (49.8) U and 118.4 (60.2) U in Cycle 2 and Cycle 3, respectively.

Table 1. Patient demographics and baseline characteristics

Characteristic	Patients N=155
Sex, n (%)	
Male	104 (67.1)
Female	51 (32.9)
Age, years; mean (SD)	53.7 (13.1)
Race, n (%)	
Caucasian	129 (83.2)
Black/African-American	4 (2.6)
Other	3 (1.9)
Missing	19 (12.3)
Causes of spasticity, n (%)	
Stroke	132 (85.2)
Ischemic	87 (56.1)
Hemorrhagic	45 (29.0)
Other causes	23 (14.8)
Traumatic brain injury	11 (7.1)
Other cerebral vascular disorder	6 (3.9)
Brain tumor	4 (2.6)
Cerebral palsy	2 (1.3)
Time since diagnosis of event leading to spasticity, months; median (range)	
Right body side, n=68	46.5 (3.7–372.8)
Left body side, n=81	61.4 (2.8–428.9)

SD, standard deviation.

Table 2. Number of patients treated per upper limb clinical pattern

	Cycle 1 N=155		Cycle 2 N=152		Cycle 3 N=140		Any cycle N=155	
	n	(%)	n	(%)	n	(%)	n	(%)
Upper limb								
Internally rotated or extended or adducted shoulder	**52**	**(33.5)**	**69**	**(45.4)**	**84**	**(60.0)**	**96**	**(61.9)**
Flexed elbow	117	(75.5)	122	(80.3)	124	(88.6)	142	(91.6)
Extended elbow	11	(7.1)	16	(10.5)	19	(13.6)	23	(14.8)
Pronated forearm	37	(23.9)	50	(32.9)	48	(34.3)	71	(45.8)
Flexed wrist	84	(54.2)	87	(57.2)	91	(65.0)	117	(75.5)
Clenched fist	96	(61.9)	110	(72.4)	110	(78.6)	128	(82.6)
Thumb-in-palm	53	(34.2)	63	(41.4)	65	(46.4)	85	(54.8)

Table 3. IncobotulinumtoxinA doses administered to the shoulder in each injection cycle

Cycle	Patients treated, n	Dose, U; mean (SD)	Dose, U; range
Shoulder			
Cycle 1	52	71.5 (32.7)	23–190
Cycle 2	69	103.5 (49.8)	22–300
Cycle 3	84	118.4 (60.2)	25–300

SD, standard deviation.

Muscle tone

- In patients treated with incobotulinumtoxinA in the shoulder, the mean [SD] AS-SSS was improved at each injection cycle (Cycle 1: 6.3 [1.9] at baseline to 5.3 [2.2] at 4-week follow-up; Cycle 2: 6.2 [2.3] at baseline to 4.5 [2.3] at 4-week follow-up; Cycle 3: 5.8 [2.2] at baseline to 4.2 [2.3] at 4-week follow-up) (Figure 2a).
- In those patients that were not treated in the shoulder, the mean (SD) AS-SSS was:
 - Cycle 1: 4.5 (2.5) at baseline and 4.0 (2.3) at 4-week follow-up.
 - Cycle 2: 4.5 (2.4) at baseline and 3.7 (2.6) at 4-week follow-up.
 - Cycle 3: 3.7 (2.4) at baseline and 2.8 (2.0) at 4-week follow-up (Figure 2b).
- In patients treated in the shoulder, the mean (SD) change in AS-SSS between baseline and 4-week follow-up was: Cycle 1 –1.1 (1.9), Cycle 2 –1.7 (1.8), and Cycle 3 –1.7 (1.8) (Figure 3).
- In those patients not treated in the shoulder, the mean (SD) change between baseline and 4-week follow up was: Cycle 1 –0.5 (1.3), Cycle 2 –0.8 (1.6), and Cycle 3 –0.9 (1.4) (Figure 3).
- In treated patients, multiple regression analysis adjusting for AS-SSS injection cycle baseline revealed a significant effect of incobotulinumtoxinA shoulder dose on AS-SSS in Cycle 3 (p=0.0081).

Figure 2. a) AS-SSS score in patients treated with incobotulinumtoxinA in the shoulder across injection cycles and b) AS-SSS score in patients that were not treated with incobotulinumtoxinA in the shoulder across injection cycles

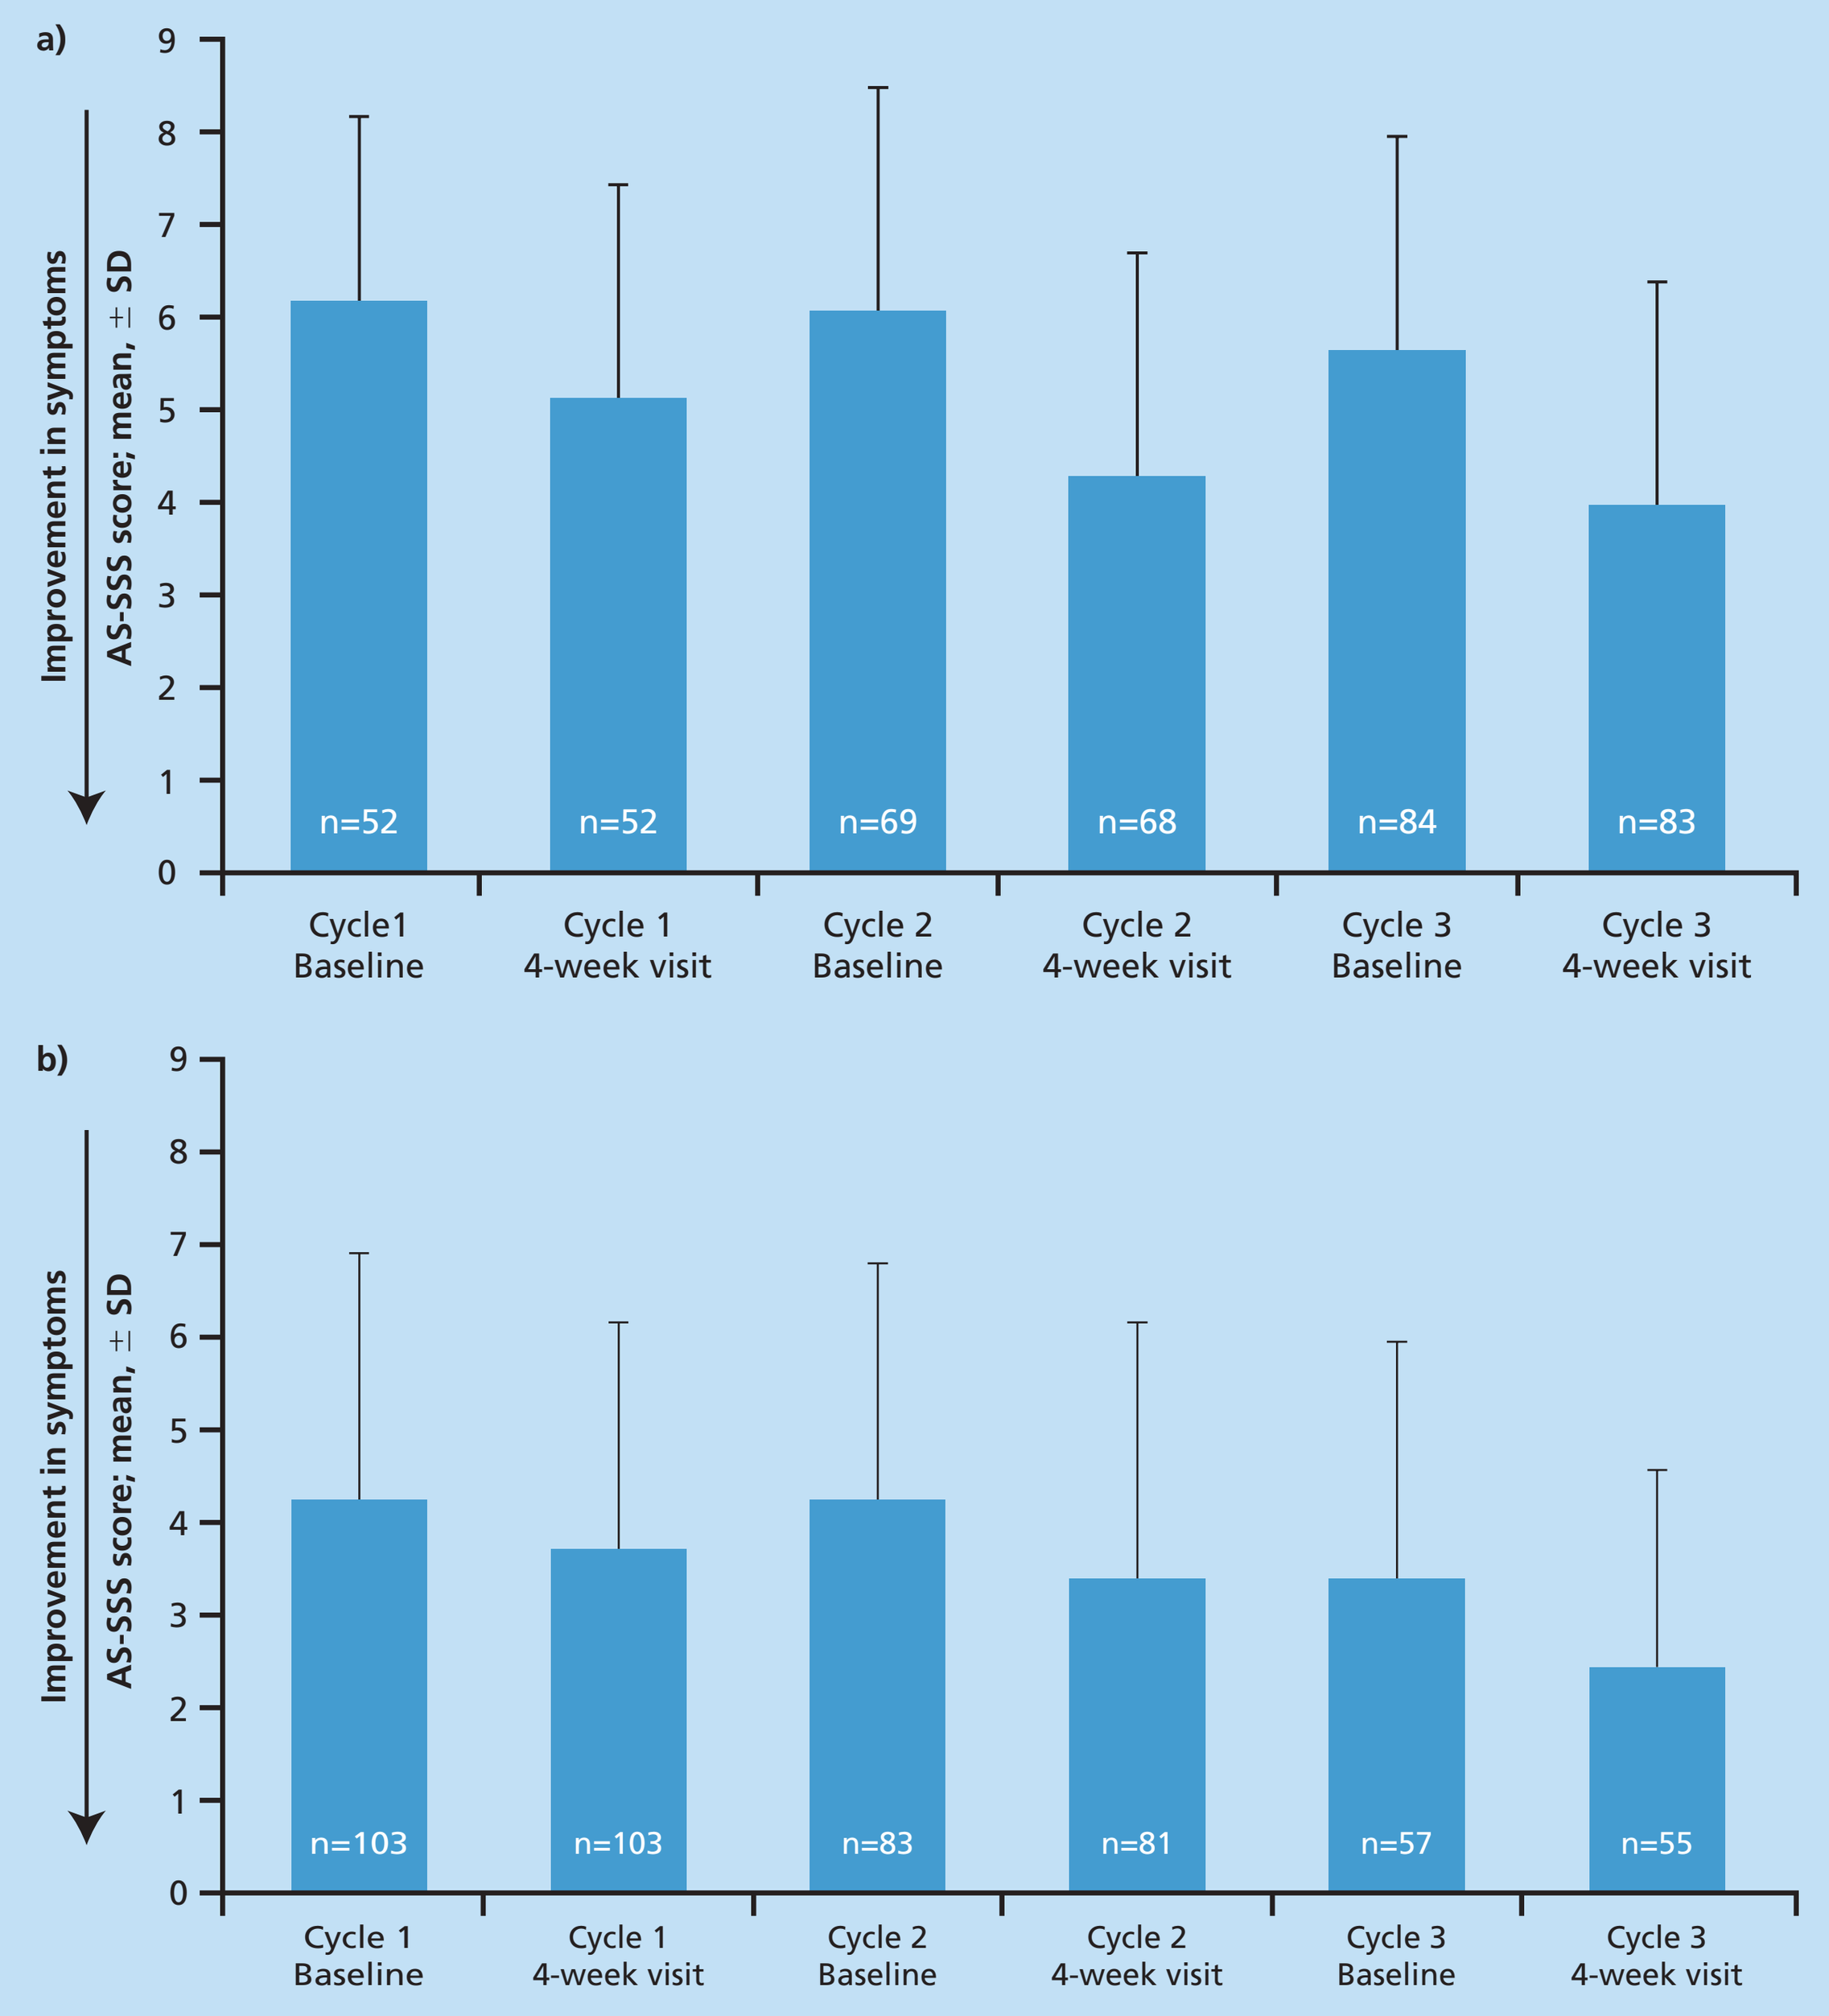
a)
Improvement in symptoms
AS-SSS score; mean, ± SD
0
1
2
3
4
5
6
7
8
9
n=52
n=52
n=69
n=68
n=84
n=83
Cycle1 Baseline
Cycle 1 4-week visit
Cycle 2 Baseline
Cycle 2 4-week visit
Cycle 3 Baseline
Cycle 3 4-week visit
b)
Improvement in symptoms
AS-SSS score; mean, ± SD
n=103
n=103
n=83
n=81
n=57
n=55
Cycle 1 Baseline
Cycle 1 4-week visit
Cycle 2 Baseline
Cycle 2 4-week visit
Cycle 3 Baseline
Cycle 3 4-week visit

n=number of patients assessed.
AS-SSS, Ashworth scale shoulder sum-score; SD, standard deviation.

Figure 3. Change in AS-SSS score in treated and untreated patients across injection cycles

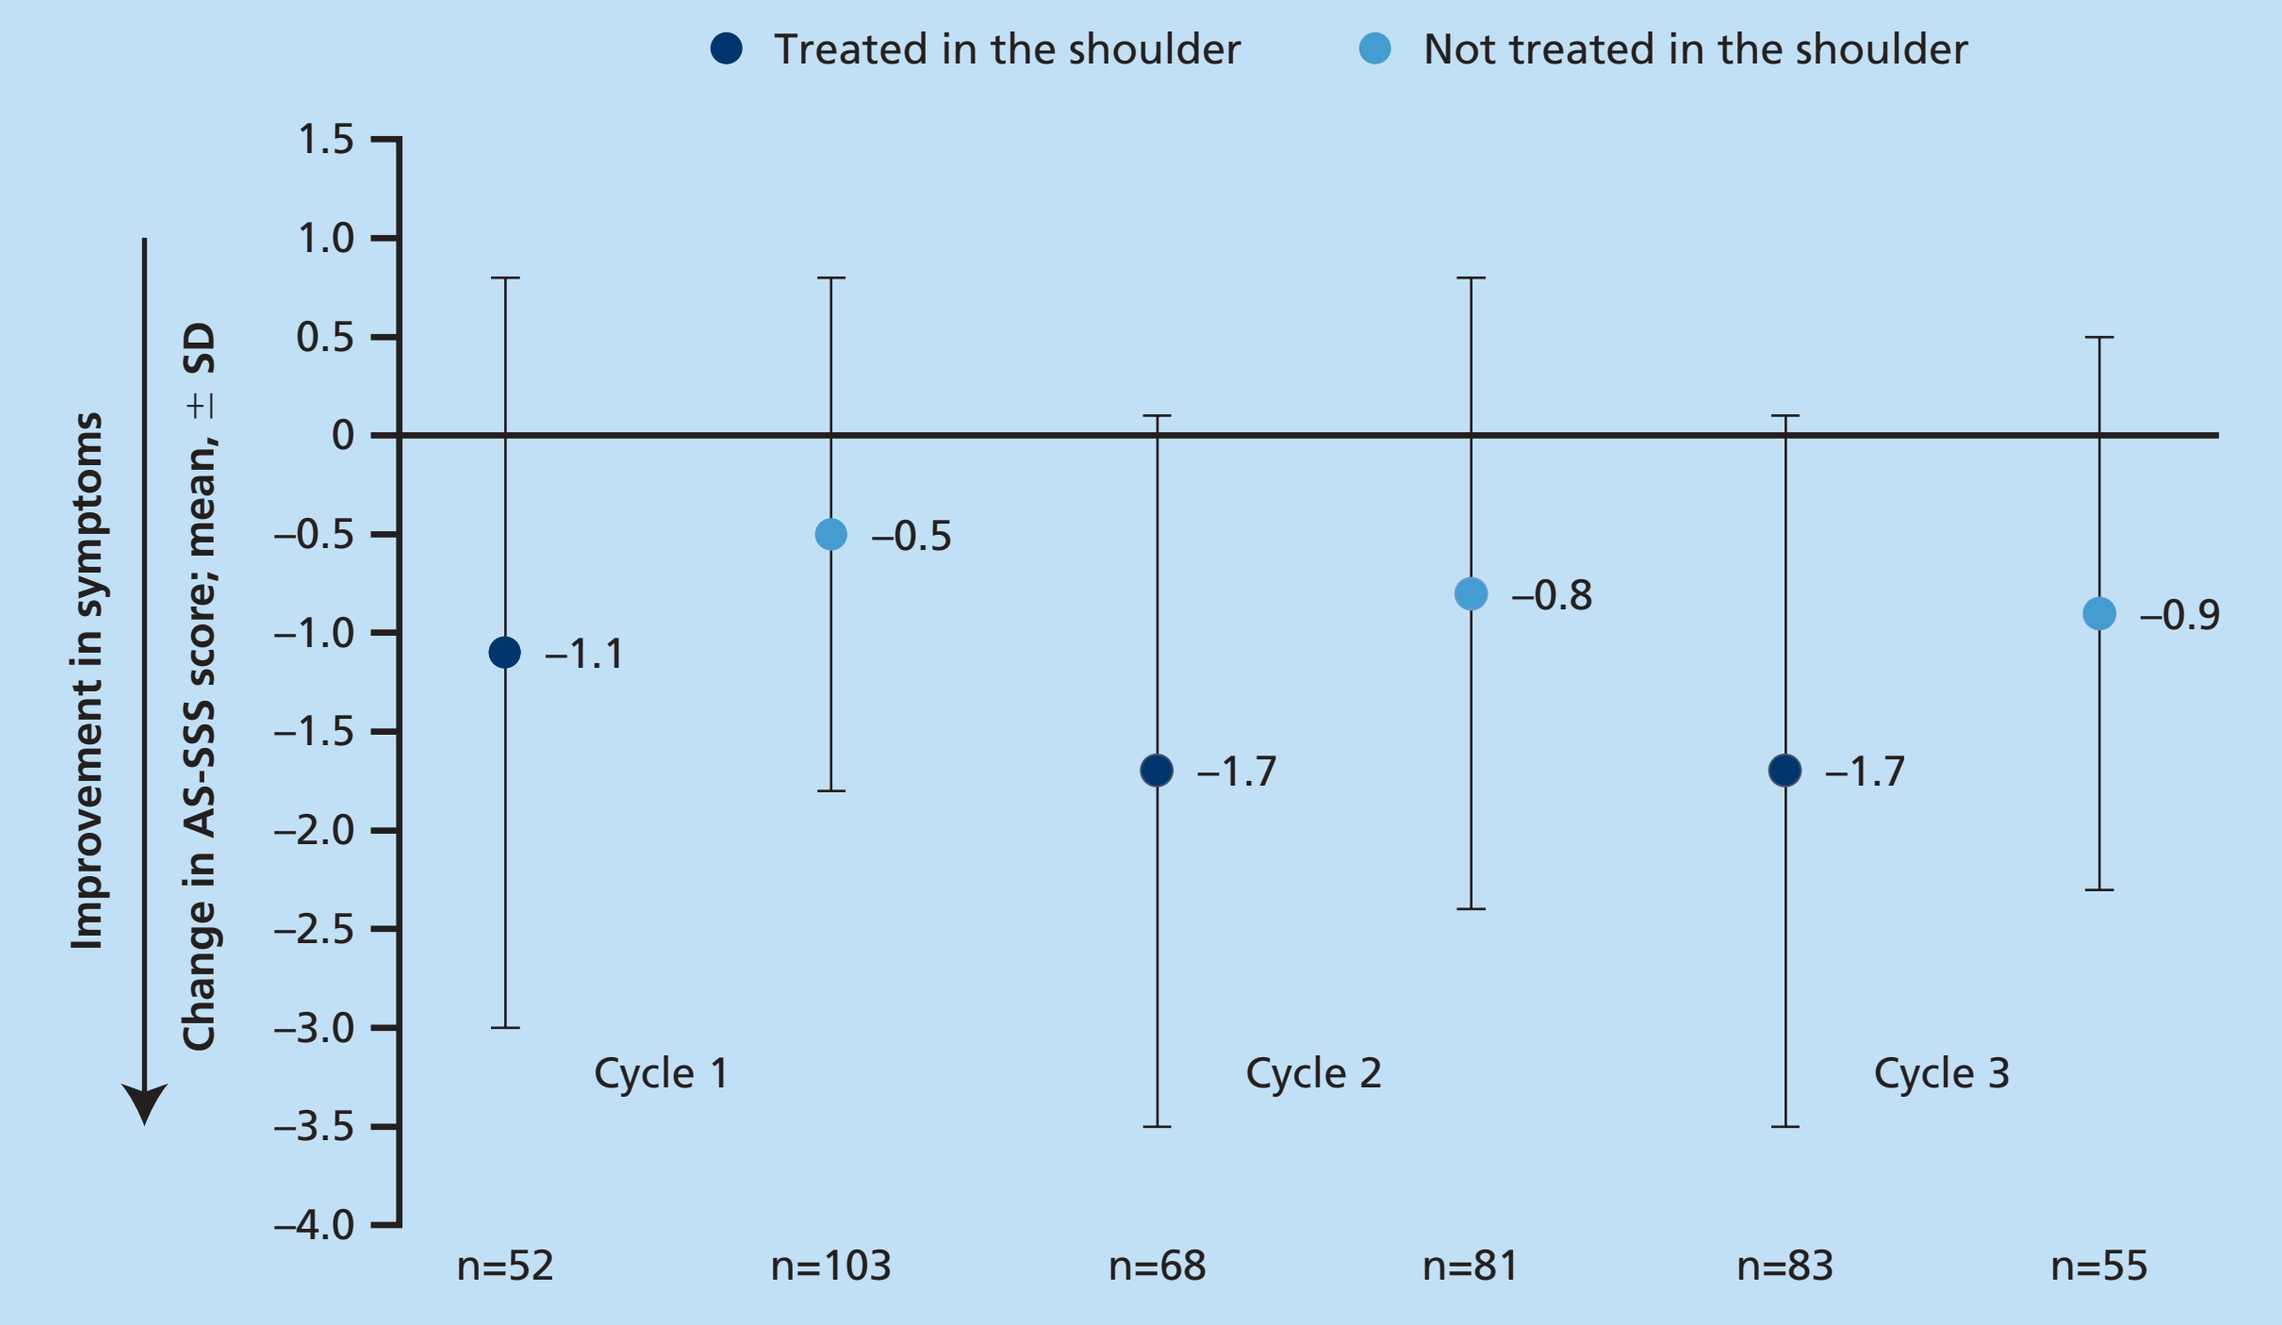
Treated in the shoulder
Not treated in the shoulder
Improvement in symptoms
Change in AS-SSS score; mean, ± SD
1.5
1.0
0.5
0
–0.5
–1.0
–1.5
–2.0
–2.5
–3.0
–3.5
–4.0
–1.1
–0.5
–1.7
–0.8
–1.7
–0.9
Cycle 1
Cycle 2
Cycle 3
n=52
n=103
n=68
n=81
n=83
n=55

AS-SSS, Ashworth scale shoulder sum-score; SD, standard deviation.

Conclusions

- In this post-hoc analysis of TOWER, the first prospective study to examine the safety and efficacy of increasing incobotulinumtoxinA doses for patients with upper- and lower-limb spasticity, incobotulinumtoxinA was shown to be efficacious for treating shoulder spasticity.
- Further evaluation of the efficacy of incobotulinumtoxinA in patients with shoulder spasticity is warranted.

References

1. Esquenazi A et al. Eur J Neurol 2010; 17 (Suppl 2): 1–8.
2. Simpson DM et al. Neurology 2016; 86: 1818–1826.
3. Wissel J et al. J Rehabil Med 2009; 41: 13–25.
4. Baricich A et al. J Neural Transm 2015; 122: 1283–1287.
5. Dressler D. Eur J Neurol 2009; 16 (Suppl 2): 2–5.
6. Dressler D et al. J Neural Transm 2015; 122: 327–333.
7. Intiso D et al. NeuroRehabilitation 2014; 34: 515–522.
8. Santamato A et al. J Neural Transm 2013; 120: 469–476.
9. Santamato A et al. J Neurol Sci 2015; 350: 1–6.
10. Barnes M et al. Acta Neurol Scand 2010; 122: 295–302.
11. Elovic EP et al. Muscle Nerve 2016; 53: 415–421.
12. Kanovský P et al. Clin Neuropharmacol 2009; 32: 259–265.
13. Kanovský P et al. J Rehabil Med 2011; 43: 486–492.
14. Dressler D. Eur J Neurol 2012; 19: 385–389.
15. Wissel J et al. PM&R 2013; 5 (Suppl): S142–S143 (primary analysis data from this study have been submitted for publication in Neurology).
16. Lim J et al. Stroke 2008; 39: 126–131.
17. Marciniak CM et al. Am J Phys Med Rehabil 2012; 91: 1007–1019.
18. Ashford S et al. Disabil Rehabil 2009; 31: 220–226.
19. Ashworth B. Practitioner 1964; 192: 540–542.

Acknowledgments

This study was sponsored by Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Medical writing support was provided by Heather Rafferty, PhD, of Complete Medical Communications and funded by Merz Pharmaceuticals GmbH.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017

MRZ82220c_TOXINS_Wisel.indd 1
12/01/2017 13:33