Efficacy of incobotulinumtoxinA (Xeomin®) in treatment of lower-limb spasticity, including pes equinovarus, in adults

Djamel Bensmail,[1] Alessio Baricich,[2] Franco Molteni,[3] Bruce Rubin,[4] Astrid Scheschonka,[5] Birgit Flatau-Baqué,[5] Olivier Simon,[5] Jörg Wissel[6]

[1]Raymond-Poincaré Hospital, AP-HP, University of Versailles Saint Quentin, Garches, France; [2]Maggiore della Carità University Hospital, Università del Piemonte Orientale, Novara, Italy; [3]Villa Beretta, Rehabilitation Center, Valduce Hospital, Como, Italy; [4]Design Neuroscience Center, Doral, Florida, USA; [5]Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany; [6]Vivantes Hospital Spandau, Berlin, Germany

Background

- Treatment guidelines recommend that botulinum neurotoxin (BoNT) injections should be offered as a treatment option for focal spasticity of the upper and lower limbs.[1-4]
- The efficacy and safety of BoNT type A formulations, including incobotulinumtoxinA (Xeomin®, Merz Pharmaceuticals GmbH), have been well documented for upper-limb spasticity. However, fewer data are available reporting the effectiveness of BoNT in adult patients with spasticity of the leg.[2,5,6]
- The TOWER study investigated the safety and efficacy of incobotulinumtoxinA for the treatment of spasticity in adults requiring multi-pattern treatment of the upper and lower limbs with total-body doses of up to 800 U per injection cycle.[7]
- In this post-hoc analysis we assessed the effectiveness of incobotulinumtoxinA for treating lower-limb spasticity, including pes equinovarus.

Methods

Study design and patients

- TOWER (clinicaltrials.gov NCT01603459) was a prospective, dose-escalation study investigating the safety and efficacy of incobotulinumtoxinA in patients (18–80 years of age) with chronic upper- and lower-limb spasticity who were deemed by the investigator to require total-body doses of up to 800 U.
- Patients received three injections with escalating, fixed total-body doses (400 U, 600 U, and up to 800 U) of incobotulinumtoxinA to be injected in the same body side, each followed by a 12–16-week observation period that included a follow-up (control) visit 4 weeks post-treatment in each cycle (**Figure 1**).
- In Cycles 1 and 2, patients could receive injections into the upper limb only, the lower limb only, or into both limbs. In Cycle 3 it was expected that patients would receive injections into both limbs.
- The maximum dose per limb in the first injection cycle was 400 U, increasing to 600 U in Cycles 2 and 3 (dose range for treatment of pes equinovarus, 100–400 U).

Outcome measures

- Muscle tone was assessed using the Ashworth Scale (AS).[8]
- All muscle groups on the side of the body selected for treatment were assessed to obtain the Resistance to Passive Movement Scale (REPAS) score.[9] REPAS comprises 16 items describing the condition of both upper limbs and 10 items describing both lower limbs, each rated from 0 to 4 using the AS. Here, evaluation of the five REPAS items for the lower limb of the treated body side resulted in a summed score from 0 to 20.
- AS and REPAS assessments were performed at each injection-cycle visit, at control visits 4 weeks after each injection, and at the end-of-study visit. Assessments were expected to be performed by the same investigator for any given patient.

Figure 1. Study design

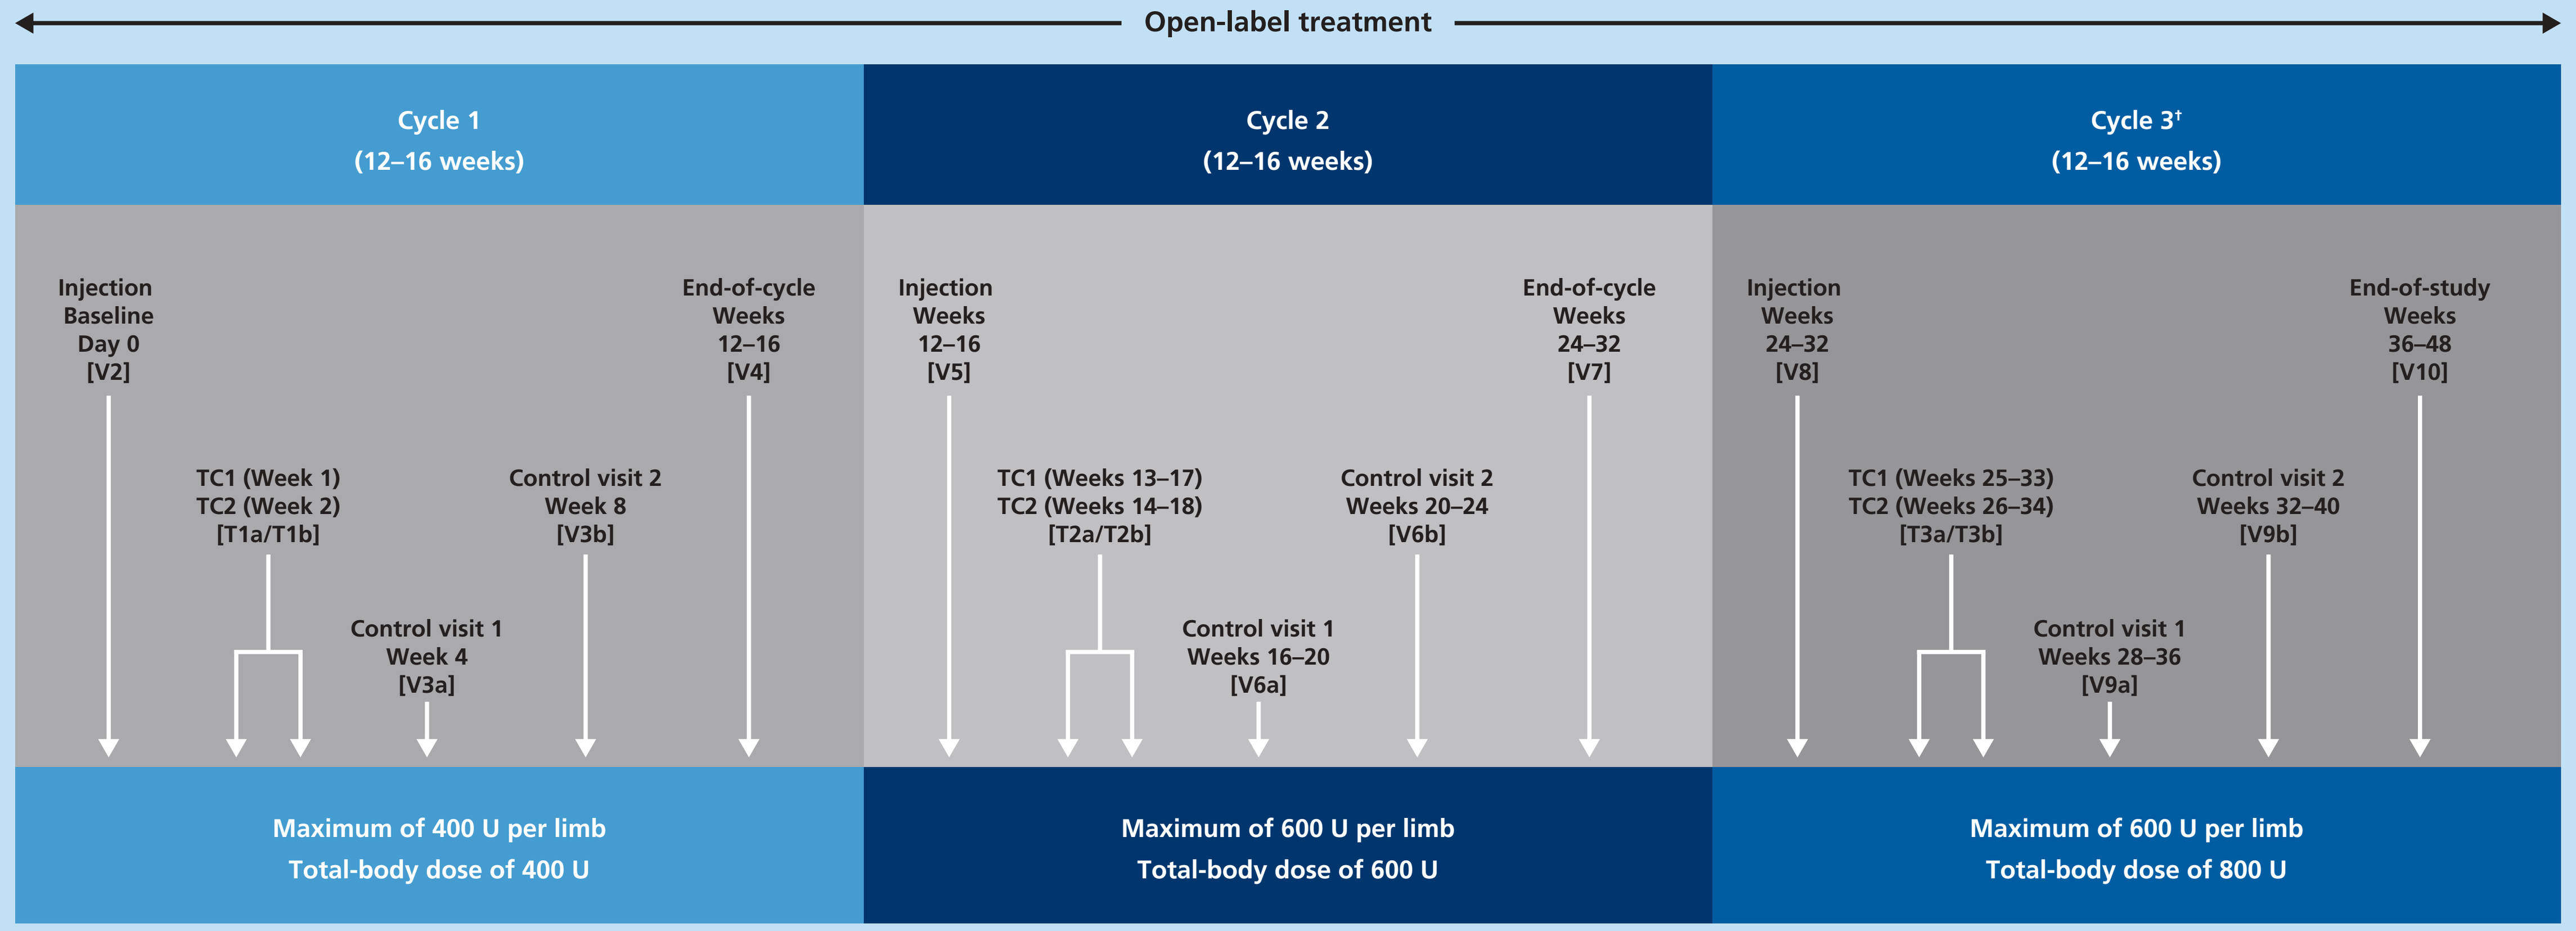
Open-label treatment
Cycle 1
(12–16 weeks)
Cycle 2
(12–16 weeks)
Cycle 3†
(12–16 weeks)
Injection
Baseline
Day 0
[V2]
TC1 (Week 1)
TC2 (Week 2)
[T1a/T1b]
Control visit 1
Week 4
[V3a]
Control visit 2
Week 8
[V3b]
End-of-cycle
Weeks
12–16
[V4]
Injection
Weeks
12–16
[V5]
TC1 (Weeks 13–17)
TC2 (Weeks 14–18)
[T2a/T2b]
Control visit 1
Weeks 16–20
[V6a]
Control visit 2
Weeks 20–24
[V6b]
End-of-cycle
Weeks
24–32
[V7]
Injection
Weeks
24–32
[V8]
TC1 (Weeks 25–33)
TC2 (Weeks 26–34)
[T3a/T3b]
Control visit 1
Weeks 28–36
[V9a]
Control visit 2
Weeks 32–40
[V9b]
End-of-study
Weeks
36–48
[V10]
Maximum of 400 U per limb
Total-body dose of 400 U
Maximum of 600 U per limb
Total-body dose of 600 U
Maximum of 600 U per limb
Total-body dose of 800 U

†If a dose of 800 U was not justified for clinical or safety reasons, a lower dose of 600–800 U could be administered as an exception.
TC, telephone contact; V, visit

Statistical analysis

- Patients treated in any of the following muscles were classed as "treated for pes equinovarus": M. gastrocnemius caput mediale; M. gastrocnemius caput laterale; M. soleus; M. tibialis posterior; M. tibialis anterior; M. flexor hallucis longus; and M. flexor digitorum longus. All patients not treated in any of these muscles were classed as "not treated for pes equinovarus", which included patients treated in the upper limb only and/or other lower-limb patterns.
- For each cycle, changes in AS scores for the ankle joint were compared for patients who were treated for pes equinovarus and those who were not by:
 - mean and standard deviation (SD)
 - analysis of variance (ANOVA) adjusting for the corresponding AS cycle baseline value
 - response rate (AS improvement of ⩾1 point)
 - Cochran–Mantel–Haenszel test adjusting for the AS cycle baseline value.
- For each cycle, changes in lower-limb REPAS scores were compared for patients treated in the lower limb (with or without treatment in the upper limb) and those treated in the upper limb only by:
 - mean and SD
 - analysis of covariance (ANCOVA) with the corresponding REPAS cycle baseline value as covariate.

Results

Patients

- Key patient demographics and baseline characteristics are summarized in **Table 1**.

Table 1. Patient demographics and baseline characteristics

Characteristic	Total patient population; N=155
Sex, n (%)	
Female	51 (32.9)
Male	104 (67.1)
Age, years; mean (SD)	53.7 (13.1)
Cause of spasticity, n (%)	
Stroke	132 (85.2)
Traumatic brain injury	11 (7.1)
Brain tumor	4 (2.6)
Other	8 (5.2)

SD, standard deviation.

- Of 155 patients who participated in the TOWER study, spasticity was due to stroke in 85.2% and other neurologic causes in 14.8%.
- The distribution of limbs treated at each injection cycle is summarized in **Table 2**. The proportion of patients receiving treatment in both limbs increased from 62.6% in Cycle 1 to 89.5% in Cycle 2 and 97.2% in Cycle 3.
- In Cycle 1, 109 patients were treated in the lower limb with a mean (SD) dose of 191.7 (97.6) U incobotulinumtoxinA.
- In Cycles 2 and 3, 137 patients were treated in the lower limb with mean (SD) doses of 254.0 (111.6) U and 320.9 (127.6) U incobotulinumtoxinA, respectively (**Table 3**).
- Of these patients, 100 were treated for pes equinovarus in Cycle 1 and 129 in each of Cycles 2 and 3. In Cycles 2 and 3, only 23 and 12 patients did not receive treatment for pes equinovarus, respectively (**Table 2**). Pes equinovarus was chosen as a target clinical pattern for treatment in 33 patients (i.e. this pattern was treated throughout the study).
- IncobotulinumtoxinA doses administered for pes equinovarus at each cycle are summarized in **Table 3**. Mean (SD) doses increased from 166.3 (94.9) U in Cycle 1 to 203.0 (102.0) U and 241.6 (102.8) U in Cycles 2 and 3, respectively.

Table 2. Distribution of limbs treated by injection cycle

Distribution of limbs treated	Patients treated, n (%)
Injection Cycle 1; N=155	
Upper limb only	46 (29.7)
Both upper and lower limb	97 (62.6)
Lower limb only	12 (7.7)
Treated for pes equinovarus	100 (64.5)
Not treated for pes equinovarus	55 (35.5)
Injection Cycle 2; N=152	
Upper limb only	15 (9.9)
Both upper and lower limb	136 (89.5)
Lower limb only	1 (0.7)
Treated for pes equinovarus	129 (84.9)
Not treated for pes equinovarus	23 (15.1)
Injection Cycle 3; N=140*	
Upper limb only	3 (2.1)
Both upper and lower limb	137 (97.9)
Lower limb only	0
Treated for pes equinovarus	129 (91.5)
Not treated for pes equinovarus	12 (8.5)

*Ashworth Scale data was available for 141 patients at Cycle 3 baseline, including one patient who was not treated in this Cycle.

Table 3. IncobotulinumtoxinA doses administered for lower limb and pes equinovarus

Cycle	Patients treated, n	Dose, U; mean (SD)	Observed dose range, U
Lower limb			
Cycle 1	109	191.7 (97.6)	40–400
Cycle 2	137	254.0 (111.6)	92–600
Cycle 3	137	320.9 (127.6)	48–600
Pes equinovarus			
Cycle 1	100	166.3 (94.9)	30–400
Cycle 2	129	203.3 (102.0)	30–450
Cycle 3	129	241.6 (102.8)	49–500

Doses >400 U for pes equinovarus treatment were injected in one patient in Cycle 2 and six patients in Cycle 3.
SD, standard deviation.

Muscle tone

- In all three cycles, mean changes in AS score for the ankle joint were greater in patients treated for pes equinovarus compared with those who were not: –0.63 versus –0.16 in Cycle 1; –0.68 versus –0.36 in Cycle 2; –0.80 versus –0.73 in Cycle 3. For the difference in Cycle 1, the ANOVA revealed a p-value of 0.0003. In Cycles 2 and 3, only 23 and 12 patients were not treated for pes equinovarus, respectively (**Table 4**).

Table 4. Change in AS score for patients treated for pes equinovarus compared with those who were not

Cycle	Mean (SD) [median] AS score at cycle baseline	Mean (SD) [median] AS score at 4 weeks post-injection	Mean (SD) change in AS score	p-value
Cycle 1				
Treated for pes equinovarus (n=100)	2.71 (0.80) [3]	2.08 (0.98) [2]	–0.63 (0.76)	0.0003
Not treated for pes equinovarus (n=55)	2.58 (0.69) [3]	2.42 (0.88) [3]	–0.16 (0.63)	
Cycle 2				
Treated for pes equinovarus (n=129)	2.60 (0.84) [3]	1.91 (0.95) [2][a]	–0.68 (0.86)[a]	0.1362
Not treated for pes equinovarus (n=23)	2.48 (0.79) [3]	2.09 (0.81) [2][b]	–0.36 (0.58)[b]	
Cycle 3				
Treated for pes equinovarus (n=129)	2.43 (0.87) [3]	1.63 (0.94) [2][a]	–0.80 (0.76)[a]	0.7570
Not treated for pes equinovarus (n=12)	2.08 (0.79) [2]	1.27 (0.47) [1][c]	–0.73 (0.79)[c]	

[a]n=127; [b]n=22; [c]n=11 based on number of patients assessed 4-weeks post-treatment; AS score for the ankle joint relates to patients treated for pes equinovarus (with or without treatment in the upper limb) and those not treated for pes equinovarus (including those treated in the upper limb and/or other lower limb patterns); p-value based on ANOVA comparing treated and untreated patients, adjusting for the corresponding AS cycle baseline value.
ANOVA, analysis of variance; AS, Ashworth Scale; SD, standard deviation.

- In all three cycles, the AS responder rate was higher in patients treated for pes equinovarus compared to those who were not: 55/100 (55%) versus 7/55 patients (12.7%) in Cycle 1; 69/127 (54.3%) versus 7/22 patients (31.8%) in Cycle 2; 83/127 (65.4%) versus 6/11 patients (54.5%) in Cycle 3. For the difference in Cycle 1, p<0.0001 (Cochran–Mantel–Haenszel) (**Figure 2**).
 - Increased AS response rate in those not treated for pes equinovarus across the three treatment cycles may be an indirect effect of the treatment in the surrounding muscles or of the injection of increased overall incobotulinumtoxinA doses (**Figure 2**).

Figure 2. AS responder rates in patients either treated or not treated for pes equinovarus during each cycle. Response was defined as ⩾1 point reduction in AS score based on the change between baseline and 4 weeks post-treatment

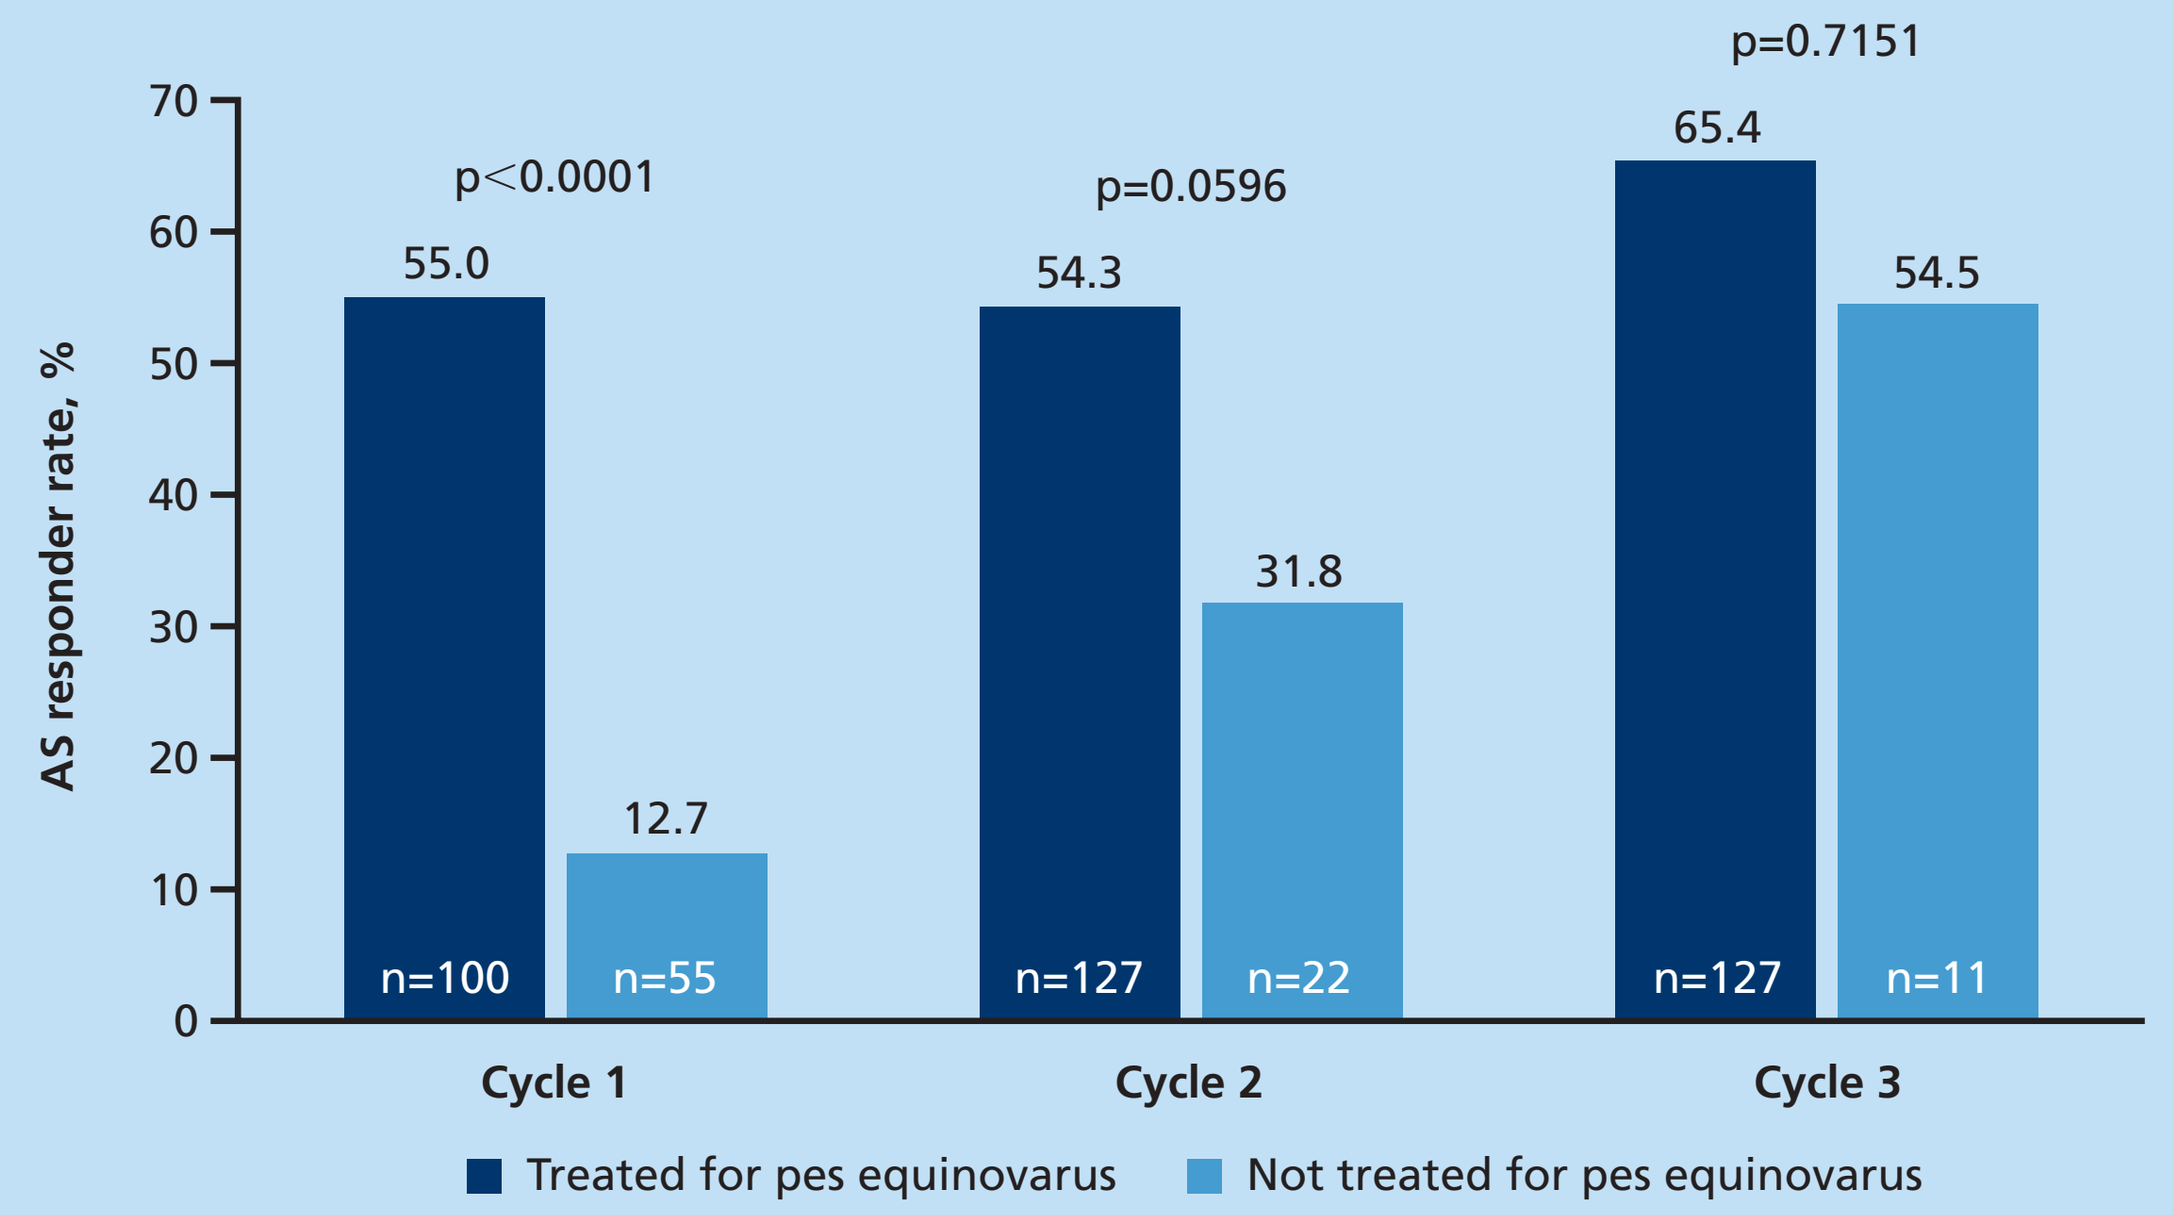
AS responder rate, %
70
60
50
40
30
20
10
0
p<0.0001
55.0
12.7
n=100
n=55
Cycle 1
p=0.0596
54.3
31.8
n=127
n=22
Cycle 2
p=0.7151
65.4
54.5
n=127
n=11
Cycle 3
Treated for pes equinovarus
Not treated for pes equinovarus

p-values based on Cochran–Mantel–Haenszel test (adjusting for the AS cycle baseline value) for change in AS responder rate in patients treated for pes equinovarus compared with those not treated; n=number of patients assessed 4-weeks post-treatment.
AS, Ashworth Scale

Resistance to Passive Movement

- Lower-limb REPAS scores improved significantly between the injection visit and 4 weeks post-treatment in patients treated in the lower limb, but not in those treated only in the upper limb: –1.6 versus –0.3 in Cycle 1; –1.9 versus –0.5 in Cycle 2; –2.1 versus –0.7 in Cycle 3 (**Figure 3**). For the difference in Cycle 1, the ANCOVA revealed a p-value <0.0001. In Cycles 2 and 3 only 15 and four patients, respectively, did not receive treatment in the lower limb.

Figure 3. Mean change in lower limb REPAS score from baseline to 4 weeks post-treatment for all cycles in patients treated in the lower limb and those treated in the upper limb only

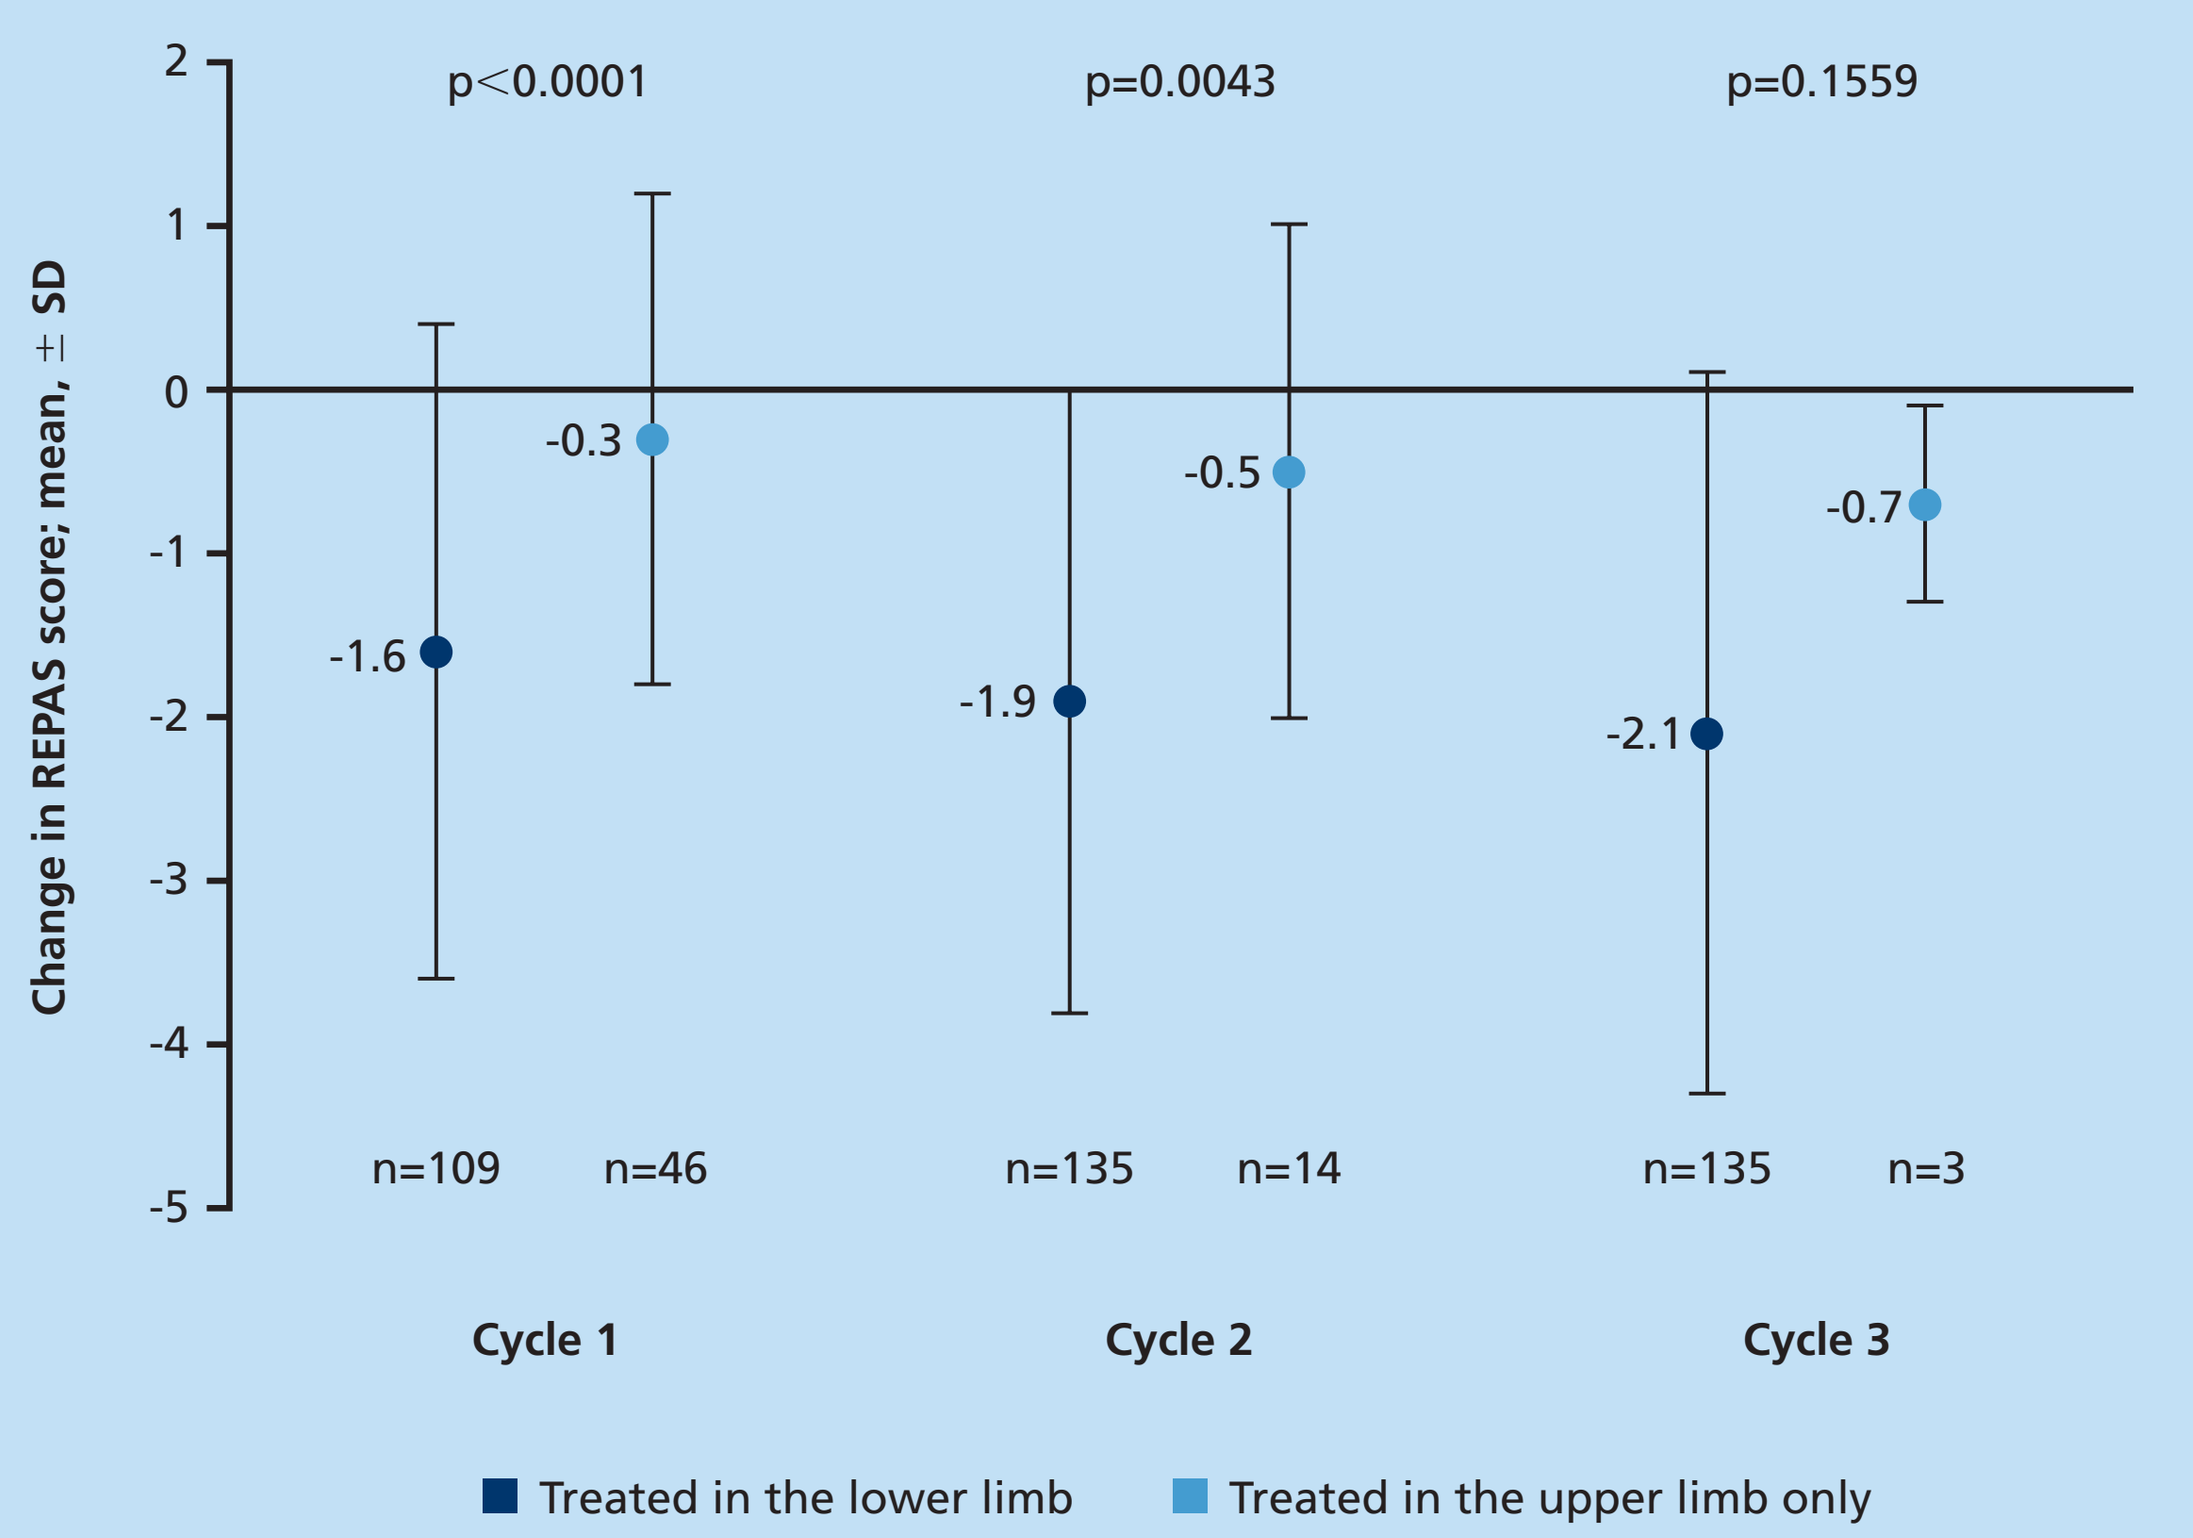
Change in REPAS score; mean, ± SD
2
1
0
-1
-2
-3
-4
-5
p<0.0001
-1.6
-0.3
n=109
n=46
Cycle 1
p=0.0043
-1.9
-0.5
n=135
n=14
Cycle 2
p=0.1559
-2.1
-0.7
n=135
n=3
Cycle 3
Treated in the lower limb
Treated in the upper limb only

p-values based on ANCOVA with corresponding REPAS cycle baseline value as covariate, regressing lower limb REPAS changes on lower limb treatment (yes/no) and corresponding REPAS cycle baseline; n, number of patients assessed 4-weeks post-treatment.
ANCOVA, analysis of covariance; REPAS, Resistance to Passive Movement; SD, standard deviation.

Conclusions

- Results of these post-hoc analyses of the TOWER study suggested that incobotulinumtoxinA could be effective in the treatment of pes equinovarus and other patterns of lower-limb spasticity in patients receiving combined treatment for upper and lower limb spasticity.

References

1. Esquenazi A et al. Eur J Neurol 2010; 17 (Suppl 2): 1–8.
2. Simpson DM et al. Neurology 2016; 86: 1818–1826.
3. Royal College of Physicians et al. 2009. Available at: https://www.rcplondon.ac.uk/sites/default/files/documents/spasticity-in-adults-management-botulinum-toxin.pdf.
4. Wissel J et al. J Rehabil Med 2009; 41: 13–25.
5. Jost WH et al. Drug Des Devel Ther 2015; 9: 1913–1926.
6. Kaku M, Simpson DM. Drug Des Devel Ther 2016; 10: 1085–1099.
7. Wissel J et al. PM&R 2013; 5 (Suppl): S142–S143 (primary analysis data from this study have been submitted for publication in Neurology).
8. Ashworth B. Practitioner 1964; 192: 540–542.
9. Platz T et al. Disabil Rehabil 2008; 30: 44–53.

Acknowledgements

The authors wish to thank the study patients and investigators. This study was sponsored by Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Editorial support was provided by Dr Claire Cairney of Complete Medical Communications, funded by Merz.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017

MRZ82220e_TOXINS_Simpson.indd 1
12/01/2017 13:27