Increasing doses of incobotulinumtoxinA (Xeomin®; 400–800 U) improve the functional impact of upper-limb spasticity

David M Simpson,[1] Djamel Bensmail,[2] Franco Molteni,[3] Markus Ebke,[4] Montserrat Abenoza Guardiola,[5] Astrid Scheschonka,[6] Birgit Flatau-Baqué,[6] Olivier Simon,[6] Jörg Wissel[7]

[1]Icahn School of Medicine at Mount Sinai Medical Center, New York, NY, USA; [2]Raymond-Poincaré Hospital, AP-HP, University of Versailles Saint Quentin, Garches, France; [3]Villa Beretta, Rehabilitation Center, Valduce Hospital, Como, Italy; [4]Dr. Becker Rhein-Sieg-Klinik, Nümbrecht, Germany; [5]Xarxa Assistencial Universitària Althaia Manresa, Barcelona, Spain; [6]Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany; [7]Department of Neurology, Vivantes Hospital Spandau, Berlin, Germany

Background

- Current European and US guidelines recommend botulinum neurotoxin (BoNT) injections as a treatment for focal spasticity.[1–3]
- A key challenge in treating patients with multifocal spasticity is that the total dose of BoNT required to improve overall function may exceed the currently approved maximum dose of 400 U incobotulinumtoxinA (Xeomin®, Merz Pharmaceuticals GmbH), thereby forcing physicians to prioritize treating only those patterns with the highest potential impact on attaining the treatment goal.
- Dose escalation allows more muscles and clinical patterns to be treated, which may improve functional outcomes in patients with multifocal spasticity.
- IncobotulinumtoxinA is effective and well tolerated in patients with upper- and lower-limb spasticity at approved doses.[4–7] Higher-than-labeled doses are also well tolerated,[8–12] but their efficacy has not been assessed in large prospective trials.
- The favorable tolerability profile, lack of secondary non-response in clinical trials, and high purity of incobotulinumtoxinA[13] make it a good candidate for clinical evaluation of escalated doses in the treatment of multifocal spasticity.
- The TOWER study (NCT01603459) was a prospective, open-label, non-randomized, single-arm, multicenter, dose-titration study investigating the safety and efficacy of three injection cycles of escalating doses of incobotulinumtoxinA (400–800 U) for the treatment of upper- and lower-limb spasticity due to stroke and other cerebral causes.[14]

Objective

- The objective of this analysis was to evaluate the influence of incobotulinumtoxinA (400–800 U) on functional impairment in patients with upper-limb spasticity.

Methods

Study design

- This prospective dose-escalation study enrolled patients 18–80 years of age with spastic hemiparesis due to cerebral causes, deemed to require total body doses of incobotulinumtoxinA 800 U.
- Patients underwent three consecutive injection cycles with incobotulinumtoxinA at escalating fixed total body doses (400 U, 600 U, and 600–800 U, respectively; ⩽600 U per limb), injected into the same body side. Each injection was followed by 12–16 weeks of observation (**Figure 1**).

Figure 1. Study design

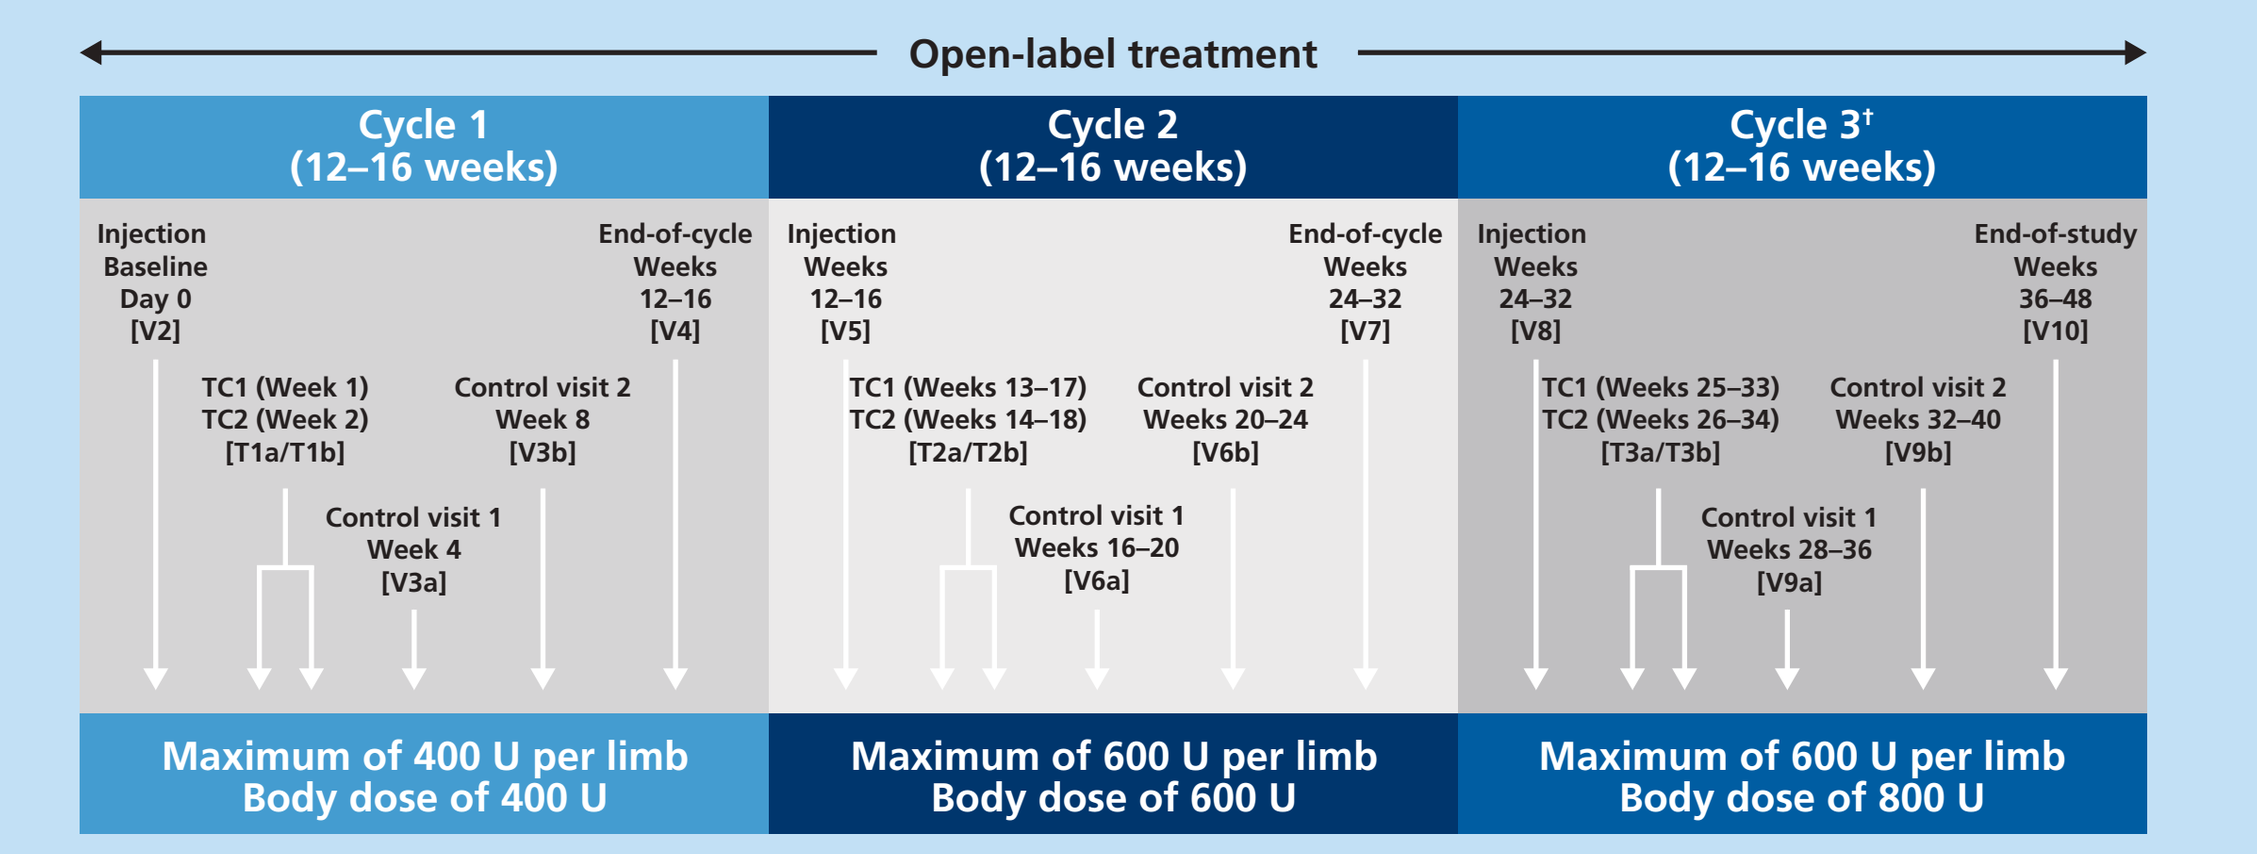
Open-label treatment
Cycle 1 (12–16 weeks)
Cycle 2 (12–16 weeks)
Cycle 3† (12–16 weeks)
Injection Baseline Day 0 [V2]
TC1 (Week 1) TC2 (Week 2) [T1a/T1b]
Control visit 1 Week 4 [V3a]
Control visit 2 Week 8 [V3b]
End-of-cycle Weeks 12–16 [V4]
Injection Weeks 12–16 [V5]
TC1 (Weeks 13–17) TC2 (Weeks 14–18) [T2a/T2b]
Control visit 1 Weeks 16–20 [V6a]
Control visit 2 Weeks 20–24 [V6b]
End-of-cycle Weeks 24–32 [V7]
Injection Weeks 24–32 [V8]
TC1 (Weeks 25–33) TC2 (Weeks 26–34) [T3a/T3b]
Control visit 1 Weeks 28–36 [V9a]
Control visit 2 Weeks 32–40 [V9b]
End-of-study Weeks 36–48 [V10]
Maximum of 400 U per limb Body dose of 400 U
Maximum of 600 U per limb Body dose of 600 U
Maximum of 600 U per limb Body dose of 800 U

†If a dose of 800 U was not justified for clinical or safety reasons, a lower dose of 600–800 U could be administered as an exception.
TC, telephone contact; V, visit.

Inclusion criteria

- Adults with spasticity of the upper and lower limb of the same body side due to cerebral lesions, or adults with bilateral symptoms – due to cerebral or brain stem lesions – who agreed to be treated on one side of the body only during the study.
- Deemed by the investigator to require total body doses of 800 U incobotulinumtoxinA.
- ⩾12 weeks since the last event leading to spasticity on the body side of the selected target clinical pattern.
- Focal spasticity with an Ashworth Scale score ⩾2 points in the joint associated with the selected target clinical pattern.
- Disability Assessment Scale (DAS) score ⩾2 points in the principal target domain.

Outcome measures

Disability Assessment Scale

- The functional impact of upper-limb spasticity was evaluated using DAS, which assesses four functional domains (‘hygiene’, ‘dressing’, ‘limb position’, and ‘pain’) on a 4-point scale as follows:
 - 0 = no disability
 - 1 = mild disability (does not interfere significantly with normal activities)
 - 2 = moderate disability (normal activities require increased effort and/or assistance)
 - 3 = severe disability (normal activities limited).
- At each injection visit, patients and/or their caregivers selected one of the four domains as their principal target of treatment. The extent of functional impairment in the principal domain was assessed at each injection visit, control visits 4 weeks post-injection, and at the end-of-Cycle 3 visit.

Functional Ambulation Classification

- The independence (i.e. the level of function with regards to supervision or physical assistance needed from another person) and the ambulation abilities of patients were assessed using the 6-point functional ambulation classification (FAC) scale, where 0 = no functional ambulation and 5 = ambulator-independent.

Goal attainment

- At each injection visit, patients and their healthcare team identified two personal, realistic goals (one active and one passive) for each treated limb (up to four goals in total).
- Investigators rated the attainment of each goal at the following injection visit (or at the end-of-Cycle 3 visit) according to a 5-point scale ranging from –2 (a lot less than expected) to +2 (a lot better than expected), where 0 indicates the expected level of achievement.

Results

Patients

- Of 193 patients screened, 155 were eligible to enroll and received treatment.
- 137 patients completed the study. No patients withdrew due to lack of efficacy. Reasons for the discontinuation of 18 patients included:
 - adverse events (n=5)
 - withdrawn consent (n=7)
 - predefined discontinuation criteria met (n=3)
 - loss to follow-up (n=3)
 - non-compliance (n=1)
 - other administrative reasons (n=1).
- Multiple discontinuation factors were entered for some patients.
- Patient demographics and baseline characteristics are summarized in **Table 1**.
- Most patients received the planned maximum dose per injection cycle (**Table 2**).
- A total of 608 clinical patterns were treated in Cycle 1, 743 were treated in Cycle 2, and 811 were treated in Cycle 3; the mean number of patterns treated per patient increased from 3.9 (Cycle 1) to 5.8 (Cycle 3).

Disability of the upper limb

- In each injection cycle, most patients (62–65%) selected ‘limb position’ as their principal target domain (**Table 3**).
- Mean (standard deviation, SD) DAS score for the principal target domain at the study baseline was 2.6 (0.5), and was reduced to 1.6 (0.8) at the end of Cycle 3, with 60.0% of patients achieving an improvement of ⩾1 point overall.
- For each injection cycle, scores improved between the injection cycle baseline and 4 weeks post-injection, with mean (SD) improvements of –0.6 (0.7), –0.7 (0.7), and –0.7 (0.8) (Cycles 1, 2, and 3, respectively; **Figure 2**).
- The proportion of patients with ‘no disability’ or ‘mild disability’ (DAS score of 0 or 1) increased at each injection cycle, from 0.7% (Cycle 1 baseline) to 45.7% (Cycle 3 control visit) (**Figure 3**).

Functional ambulation

- The proportion of ambulator-independent patients (FAC score of 5) increased from 24.5% (Cycle 1 baseline) to 41.4% (end of Cycle 3) (**Figure 4**).

Goal attainment

- The proportion of patients achieving at least three of four possible treatment goals by the next injection visit (or end-of-Cycle 3 visit) increased after each injection, with 25.2%, 50.7%, and 68.6% of patients achieving ⩾3 goals in Cycle 1, Cycle 2, and Cycle 3, respectively (**Figure 5**).

Table 1. Patient demographics and baseline characteristics

Characteristic	Patients N=155
Sex, n (%)	
Males	104 (67.1)
Females	51 (32.9)
Age, years; mean (SD)	53.7 (13.1)
Race, n (%)	
Caucasian	129 (83.2)
Black/African American	4 (2.6)
Other	3 (1.9)
Missing	19 (12.3)
Causes of spasticity, n (%)	
Stroke	132 (85.2)
Ischemic	87 (56.1)
Hemorrhagic	45 (29.0)
Other causes	23 (14.8)
Traumatic brain injury	11 (7.1)
Other cerebral vascular disorder	6 (3.9)
Brain tumor	4 (2.6)
Cerebral palsy	2 (1.3)
Median time since diagnosis of event leading to spasticity, months (range)	
Right body side; n=68	46.5 (3.7–372.8)
Left body side; n=81	61.4 (2.8–428.9)

SD, standard deviation.

Table 2. Doses administered by injection cycle

IncobotulinumtoxinA	Patients, n (%)
Cycle 1	**N=155**
375–<400 U	14 (9)
400 U	**141 (91.0)**
Cycle 2	**N=152**
560–<600 U	13 (8.6)
600 U	**138 (90.8)**
625–<700 U	1 (0.7)
Cycle 3	**N=140**
500–<600 U	1 (0.7)
600–<700 U	8 (5.7)
700–<800 U	15 (10.7)
800 U	**116 (82.9)**

Table 3. Principal therapeutic target domains selected by patients and healthcare teams for assessment of functional disability of the upper limb in each injection cycle

Patients, n (%)	Cycle 1 N=155	Cycle 2 N=152	Cycle 3 N=140
Limb position	89 (62.2)	98 (64.9)	87 (62.1)
Dressing	25 (17.5)	25 (16.6)	24 (17.1)
Hygiene	18 (12.6)	15 (9.9)	16 (11.4)
Pain	11 (7.7)	13 (8.6)	13 (9.3)
None selected	12 (7.7)	1 (0.7)	0 (0)

Figure 2. Change in DAS score for the principal therapeutic target domain 4 weeks after each injection

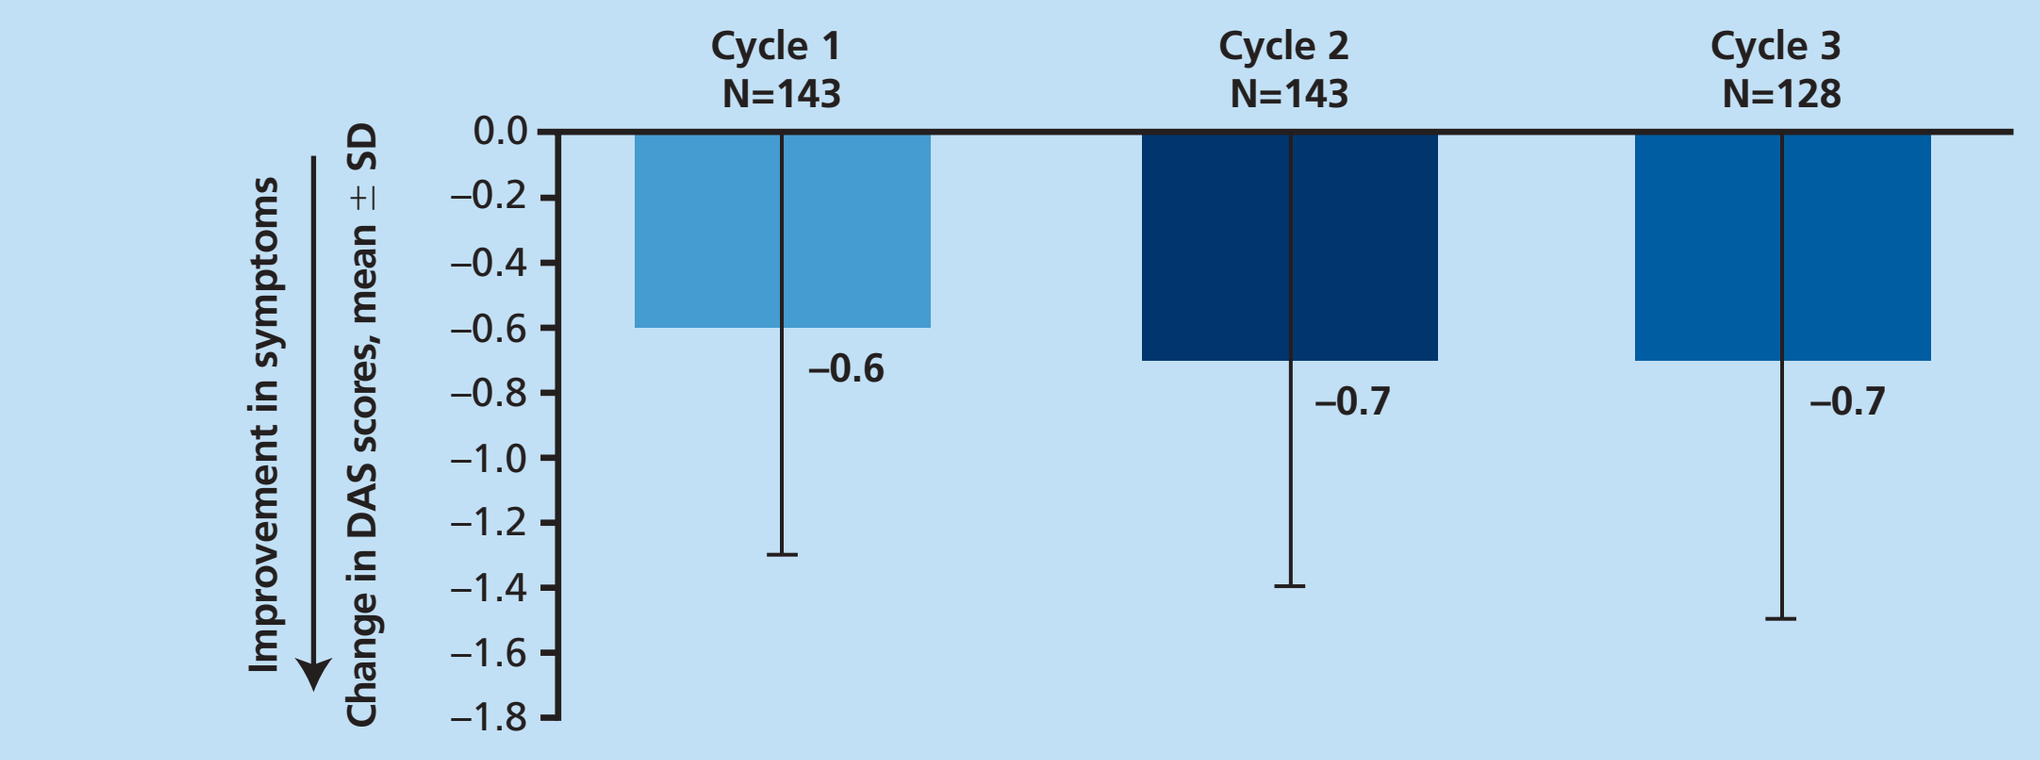
Cycle 1 N=143
Cycle 2 N=143
Cycle 3 N=128
Improvement in symptoms
Change in DAS scores, mean ± SD
0.0
–0.2
–0.4
–0.6
–0.8
–1.0
–1.2
–1.4
–1.6
–1.8
–0.6
–0.7
–0.7

Mean changes in DAS scores were calculated 4 weeks after the injection cycle baseline visit.
DAS, Disability Assessment Scale; SD, standard deviation.

Figure 3. Proportion of patients with ‘no disability’ or ‘mild disability’ (DAS score = 0 or 1) in the principal target domain at injection cycle baseline and 4 weeks after each injection

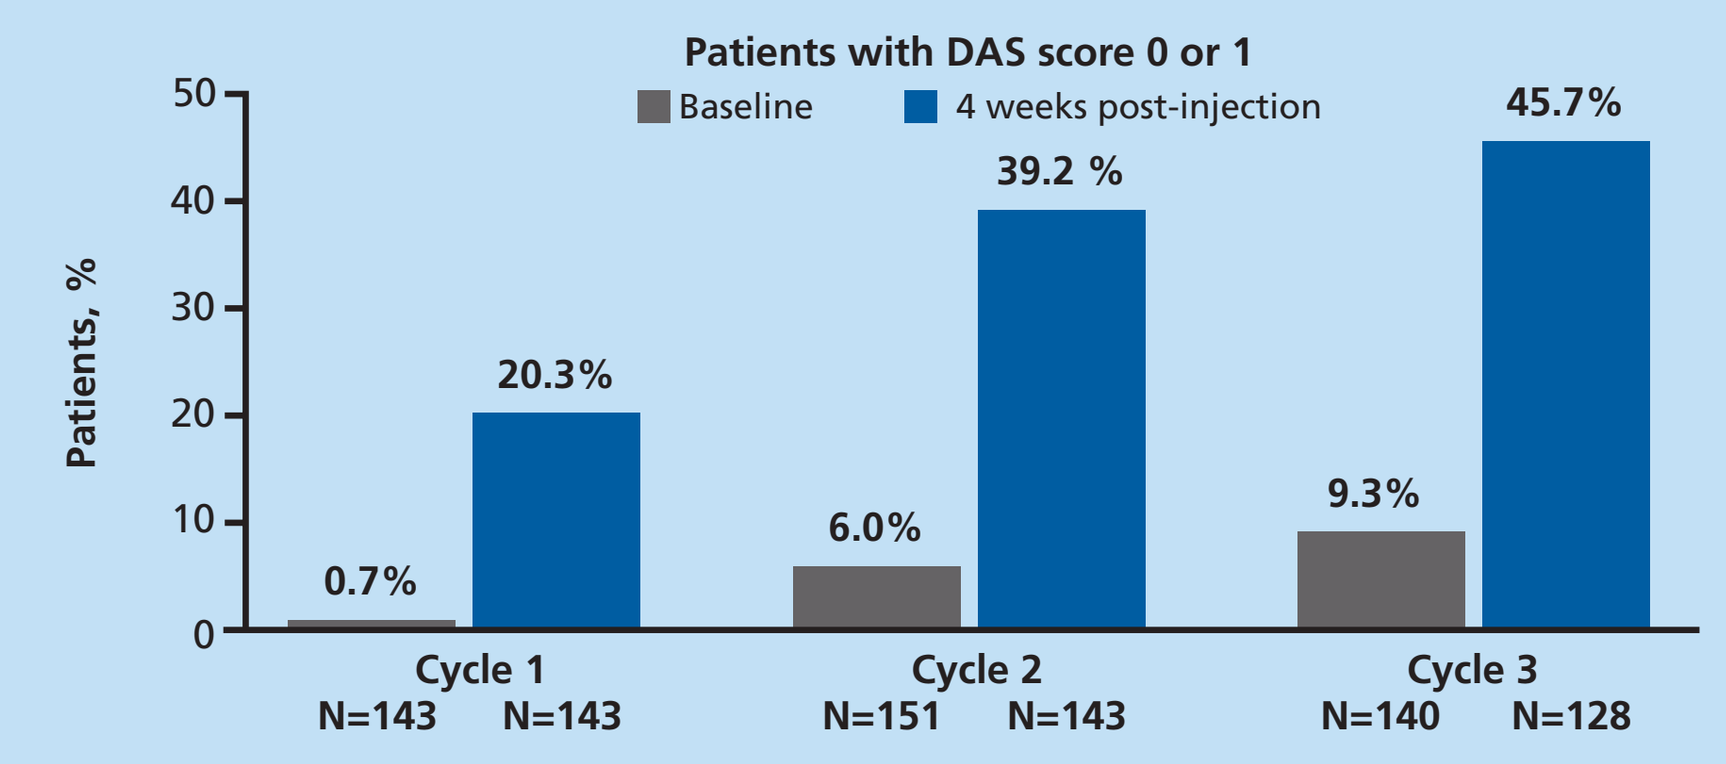
Patients with DAS score 0 or 1
Baseline
4 weeks post-injection
Patients, %
50
40
30
20
10
0
0.7%
20.3%
6.0%
39.2 %
9.3%
45.7%
Cycle 1
N=143 N=143
Cycle 2
N=151 N=143
Cycle 3
N=140 N=128

Inclusion criteria, patients with DAS score ≥2 points (moderate/severe disability) in the principal target domain at study baseline.
Principal target domain was selected at each injection cycle baseline visit.
DAS, Disability Assessment Scale.

Figure 4. Proportion of ambulator-independent patients (FAC score of 5) at baseline and end of Cycle 3

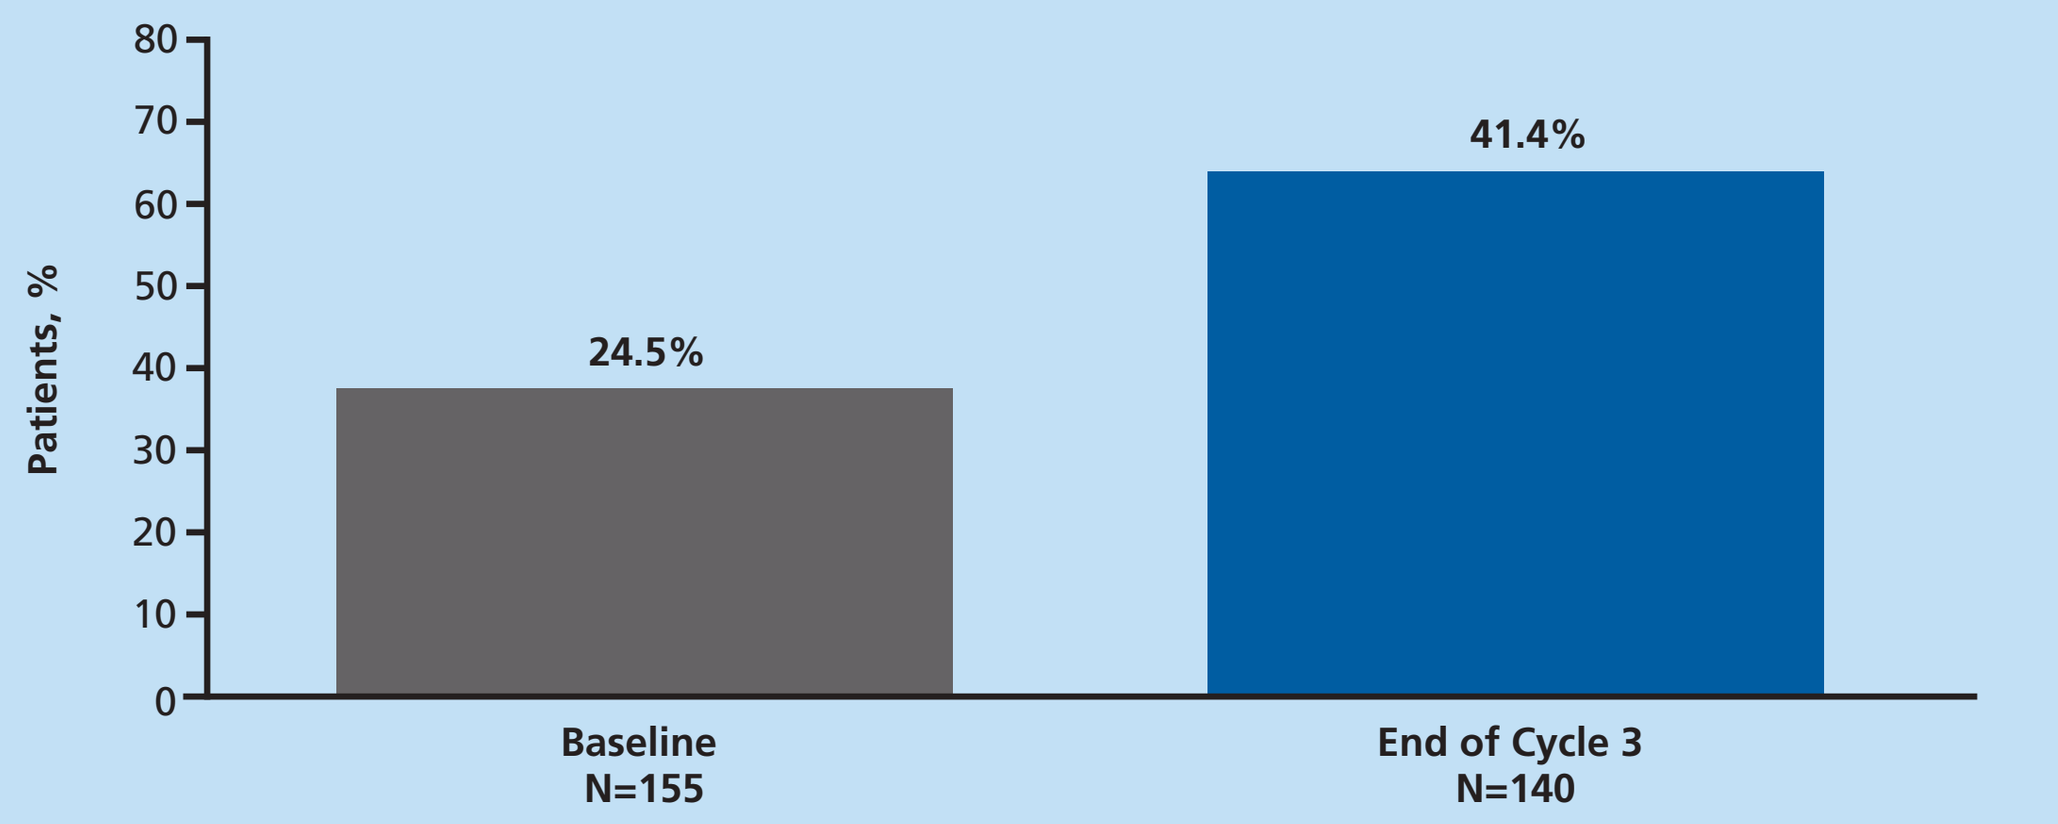
Patients, %
80
70
60
50
40
30
20
10
0
24.5%
41.4%
Baseline
N=155
End of Cycle 3
N=140

FAC, Functional Ambulation Category.

Figure 5. Proportion of patients achieving one, two, or at least three goals by the end of the injection cycle

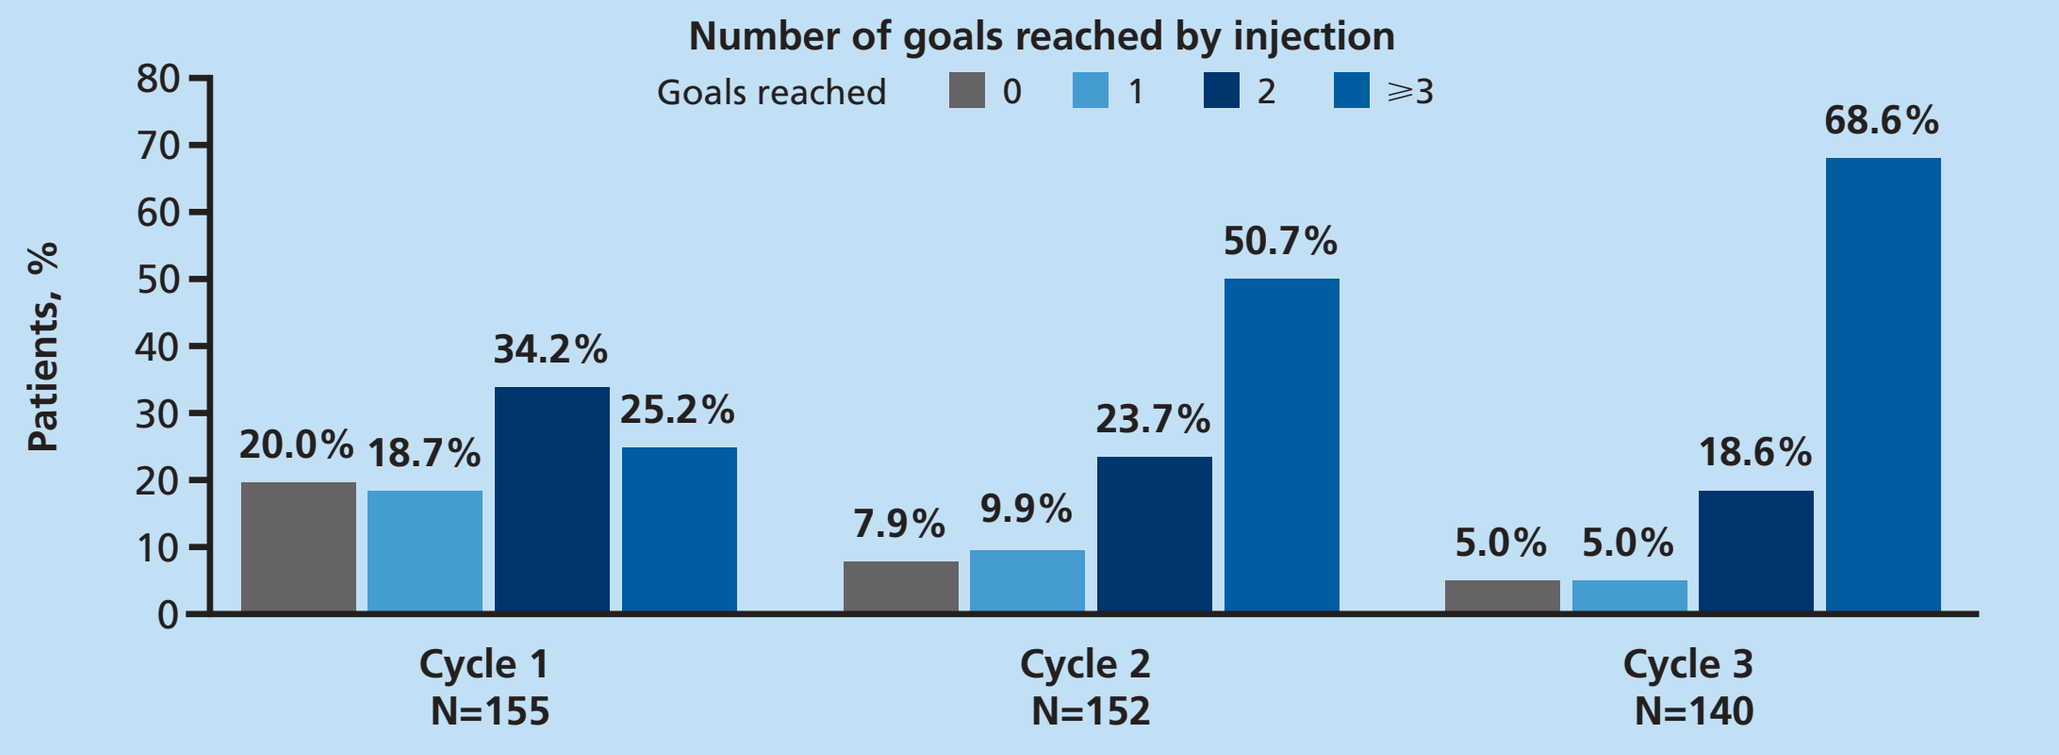
Number of goals reached by injection
Goals reached
0
1
2
⩾3
Patients, %
80
70
60
50
40
30
20
10
0
20.0%
18.7%
34.2%
25.2%
7.9%
9.9%
23.7%
50.7%
5.0%
5.0%
18.6%
68.6%
Cycle 1
N=155
Cycle 2
N=152
Cycle 3
N=140

Each patient and healthcare team identified two realistic treatment goals (one active and one passive) for each limb at each injection visit.
Goal attainment for each injection cycle was rated at the next injection visit or the study end visit.

Conclusions

- These data support findings of randomized, placebo-controlled studies showing that incobotulinumtoxinA treatment improves functional disability associated with upper-limb spasticity.
- Besides substantial improvements in functional disability of the upper limb, dose escalation of incobotulinumtoxinA led to improvements in functional ambulation and patient-specific goal attainment after treatment of both the upper and lower limbs on one body side.

References

1. Esquenazi A et al. Eur J Neurol 2010; 17 (Suppl 2): 1–8.
2. Simpson DM et al. Neurology 2016; 86: 1818–1826.
3. Wissel J et al. J Rehabil Med 2009; 41: 13–25.
4. Dressler D et al. BMJ Open 2015; 5: e009358.
5. Elovic EP et al. Muscle Nerve 2016; 53: 415–421.
6. Kaňovský P et al. Clin Neuropharmacol 2009; 32: 259–265.
7. Kaňovský P et al. J Rehabil Med 2011; 43: 486–492.
8. Baricich A et al. J Neural Transm (Vienna) 2015; 122: 1283–1287.
9. Intiso D et al. NeuroRehabilitation 2014; 34: 515–522.
10. Santamato A et al. J Neural Transm (Vienna) 2013; 120: 469–476.
11. Santamato A et al. J Neurol Sci 2015; 350: 1–6.
12. Dressler D et al. J Neural Transm 2015; 112: 427–431.
13. Jost WH et al. Drug Des Devel Ther 2015; 9: 1913–1926.
14. Wissel J et al. PM&R 2013; 5 (Suppl): S142–S143 (primary analysis data from this study have been submitted for publication in Neurology).

Acknowledgments

This study was sponsored by Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Medical writing support was provided by Nina Divorty of Complete Medical Communications, funded by Merz Pharmaceuticals GmbH.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017