Prospective, single-arm, dose-titration study of incobotulinumtoxinA (Xeomin®; up to 800 U) for treatment of upper- and lower-limb spasticity

Jörg Wissel,[1] Djamel Bensmail,[2] Astrid Scheschonka,[3] Birgit Flatau-Baqué,[3] Olivier Simon,[3] Dirk Dressler,[4*] David M Simpson[5*]

[1]Vivantes Hospital Spandau, Berlin, Germany; [2]Raymond-Poincaré Hospital, AP-HP, University of Versailles Saint Quentin, Garches, France; [3]Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany; [4]Hannover Medical School, Hannover, Germany; [5]Icahn School of Medicine at Mount Sinai Medical Center, New York, NY, USA

*DD and DS contributed equally to this work

Background

- Current European and US guidelines recommend botulinum neurotoxin (BoNT) injections as a treatment for focal spasticity.[1–3]
- A key challenge in treating patients with multifocal upper- and lower-limb spasticity is that the total dose of BoNT needed clinically may exceed currently approved doses.
- Physicians may therefore prioritize treating only certain clinical patterns, which can impact outcomes.
- The ability to escalate the total body dose of BoNT (400–800 U) would reduce the need for physicians to prioritize certain clinical patterns, and may lead to better outcomes for the patient.
- The safe use of higher-than-labeled BoNT doses has been reported,[4–9] but has not been evaluated in any large, prospective trials.
- IncobotulinumtoxinA (Xeomin®, Merz Pharmaceuticals GmbH) has been shown to be well tolerated by patients with upper-limb spasticity.[4–12]
- Due to its favorable tolerability profile, the lack of secondary non-response in clinical trials, and high purity,[13] incobotulinumtoxinA may be considered for clinical evaluation of doses up to 800 U.

Objective

- To assess the safety and efficacy of escalating incobotulinumtoxinA doses (400–800 U) in patients with spasticity due to cerebral lesions.

Methods

Study design

- The TOWER study (NCT01603459) was a prospective, open-label, dose-escalation study investigating the safety and efficacy of incobotulinumtoxinA in patients (18–80 years of age) with chronic upper- and lower-limb spasticity, due to stroke and other cerebral causes, who were deemed by the investigator to require total body doses of up to 800 U.[14]
- Additional inclusion criteria were as follows:
 - ⩾12 weeks since the last event leading to spasticity on the body side of the selected target clinical pattern
 - focal spasticity with an Ashworth Scale (AS) score ⩾2 in the joint associated with the target clinical pattern
 - Disability Assessment Scale score ⩾2 in the principal target domain.
- The study comprised three injection cycles (Cycles 1–3), with escalating fixed total body doses of incobotulinumtoxinA (400, 600, and 600–800 U) to be injected in the same body side. This was followed by a 12–16-week observation period that included a follow-up (control) visit 4 weeks post-treatment in each cycle (**Figure 1**).
- In Cycle 1 and Cycle 2, patients could receive injections into the upper limb only, the lower limb only, or into both limbs. In Cycle 3, it was expected that patients would receive injections into both the upper and the lower limbs. The maximum dose per limb in Cycle 1 was 400 U, increasing to 600 U in Cycle 2 and Cycle 3 (**Figure 1**).

Figure 1. Study design

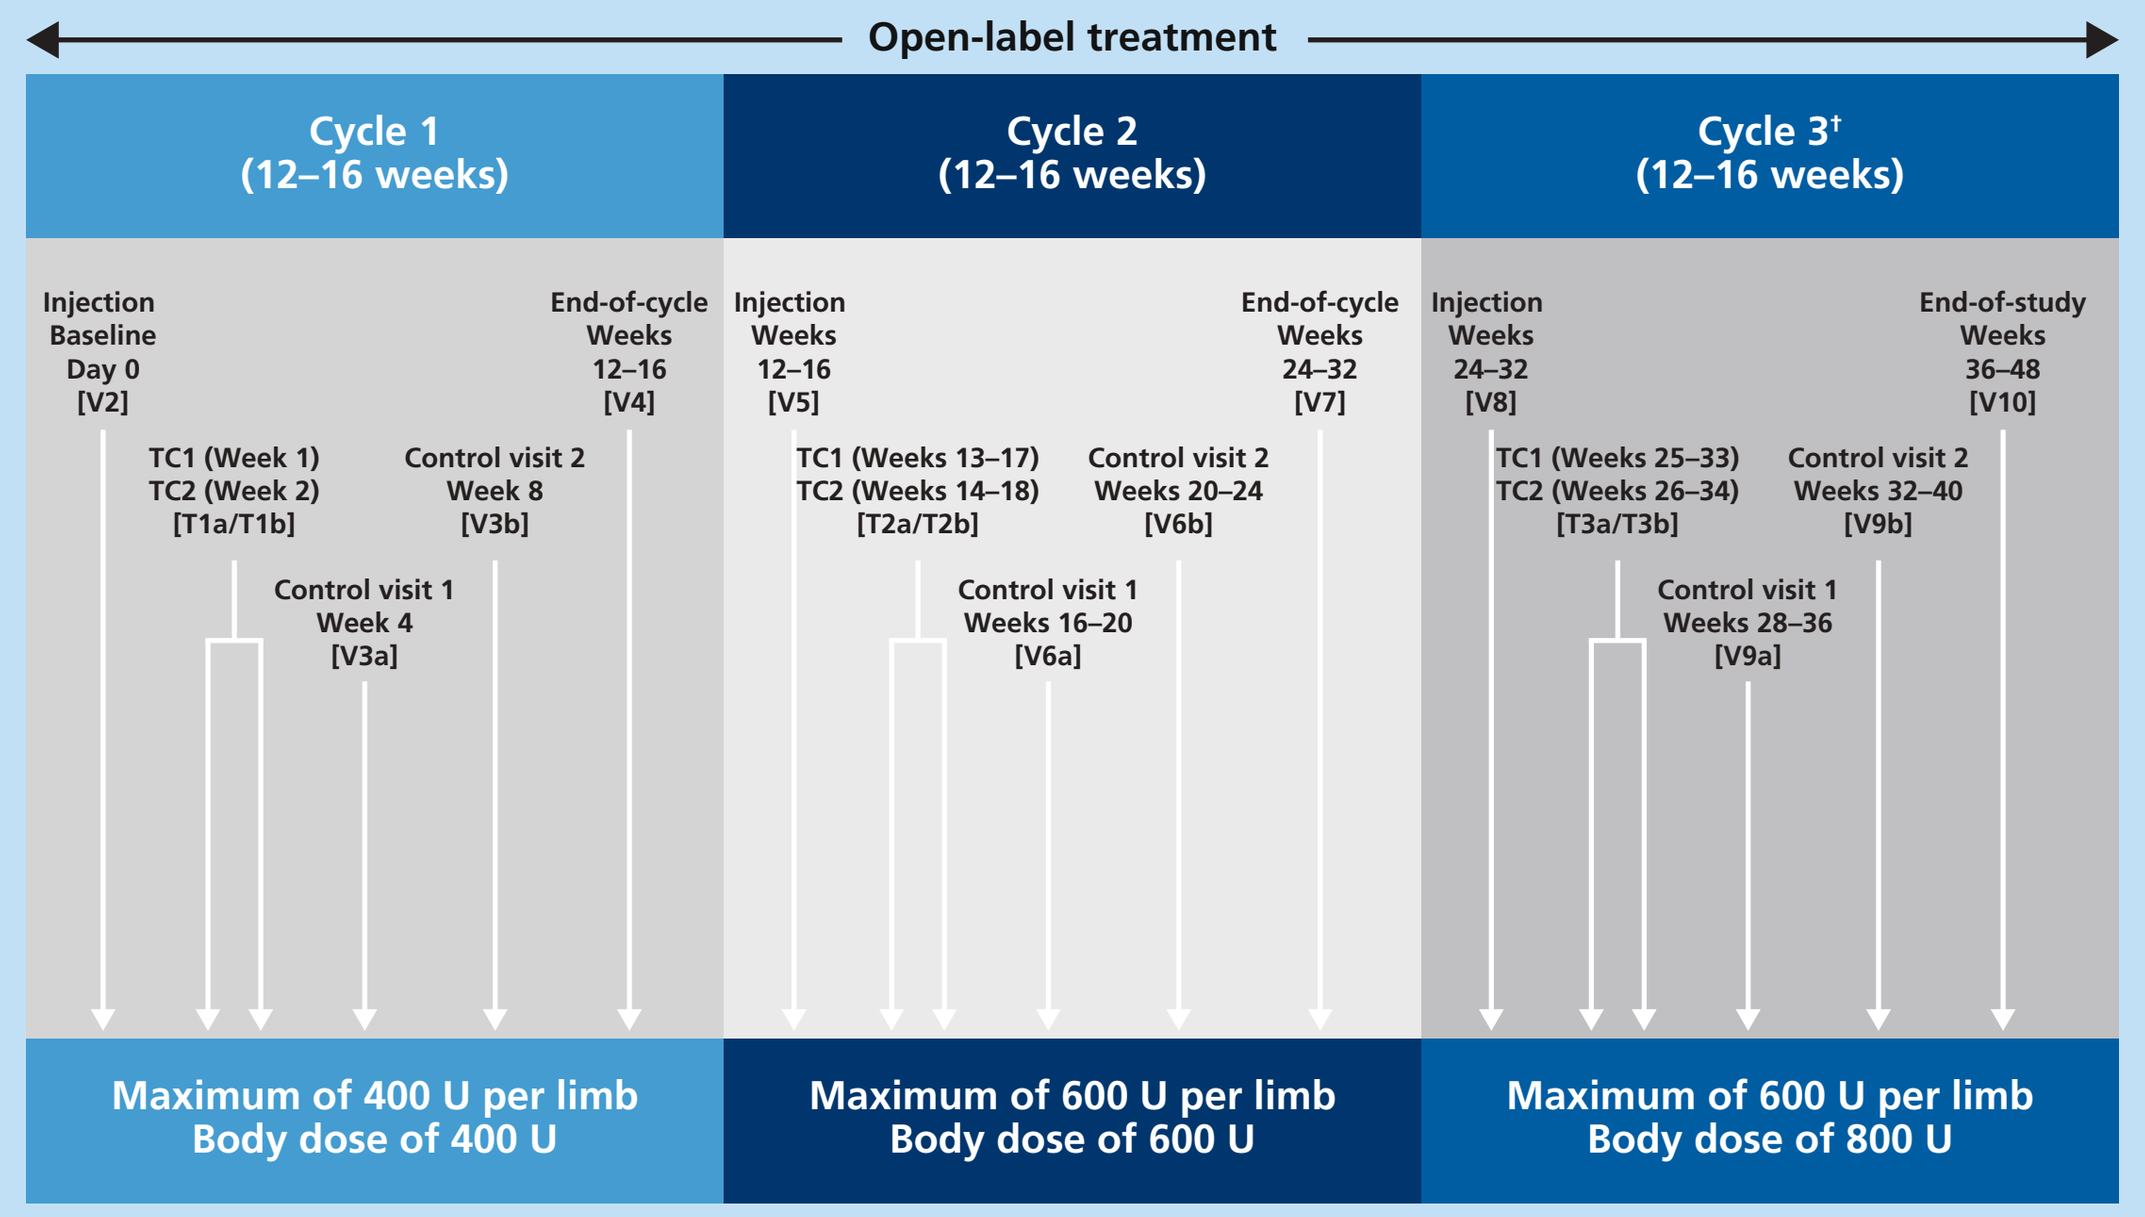
Open-label treatment
Cycle 1 (12–16 weeks)
Cycle 2 (12–16 weeks)
Cycle 3† (12–16 weeks)
Injection Baseline Day 0 [V2]
TC1 (Week 1) TC2 (Week 2) [T1a/T1b]
Control visit 1 Week 4 [V3a]
Control visit 2 Week 8 [V3b]
End-of-cycle Weeks 12–16 [V4]
Injection Weeks 12–16 [V5]
TC1 (Weeks 13–17) TC2 (Weeks 14–18) [T2a/T2b]
Control visit 1 Weeks 16–20 [V6a]
Control visit 2 Weeks 20–24 [V6b]
End-of-cycle Weeks 24–32 [V7]
Injection Weeks 24–32 [V8]
TC1 (Weeks 25–33) TC2 (Weeks 26–34) [T3a/T3b]
Control visit 1 Weeks 28–36 [V9a]
Control visit 2 Weeks 32–40 [V9b]
End-of-study Weeks 36–48 [V10]
Maximum of 400 U per limb Body dose of 400 U
Maximum of 600 U per limb Body dose of 600 U
Maximum of 600 U per limb Body dose of 800 U

†If a dose of 800 U was not justified for clinical or safety reasons, a lower dose of 600–800 U could be administered as an exception.
TC, telephone contact; V, visit.

Outcome measures

- Outcomes included:
 - AS responses (⩾1-point reduction in AS score)
 - resistance to passive movement scale (REPAS; based on AS assessments)
 - global assessment of efficacy
 - adverse events (AEs)
 - antibody testing.
- The AS is a well-known and validated scale (scored from 0 to 4, where 0 indicates no increase in tone and 4 indicates that the limb is rigid in flexion or extension) used to categorize the severity of spasticity by judging resistance to passive movement.[15] In this study, assessment of the AS was carried out over approximately 1 second.
- The REPAS scale is a summary 26-item test,[16] consisting of 16 items describing the condition of both upper limbs and 10 items describing the condition of both lower limbs; these were scored individually using the AS.
- The assessment of muscle tone was restricted to the treated body side, hence resulting in a REPAS score from 0 to 52.

Statistical analysis

- Safety analyses were performed on the safety evaluation set (SES; all patients who received ⩾1 dose of study drug). AEs were coded according to the Medical Dictionary for Regulatory Activities version 15.0. Only treatment-emergent AEs were analyzed, i.e. AEs with onset/worsening after the first study drug administration up to and including 16 weeks after the last incobotulinumtoxinA injection or the end-of-study visit, whichever was later.
- Efficacy variables were analyzed in the full analysis set, identical to the SES in this study, using descriptive summary statistics. Continuous variables were summarized by number of non-missing observations, mean, standard deviation (SD), median, quartiles, minimum, and maximum. For qualitative variables, absolute and percent frequencies were calculated.

Results

- Of 193 patients screened, 155 were eligible to enroll and received treatment.
- Key patient demographics and baseline characteristics are summarized in **Table 1**.

Table 1. Patient demographics and baseline characteristics

Characteristic	Patients N=155
Sex, n (%)	
Male	104 (67.1)
Female	51 (32.9)
Age, years; mean (SD)	53.7 (13.1)
Race, n (%)	
Caucasian	129 (83.2)
Black/African-American	4 (2.6)
Other	3 (1.9)
Missing	19 (12.3)
Causes of spasticity, n (%)	
Stroke	132 (85.2)
Ischemic	87 (56.1)
Hemorrhagic	45 (29.0)
Other causes	23 (14.8)
Traumatic brain injury	11 (7.1)
Other cerebral vascular disorder	6 (3.9)
Brain tumor	4 (2.6)
Cerebral palsy	2 (1.3)
Time since diagnosis of event leading to spasticity, months; median (range)	
Right body side, n=68	46.5 (3.7–372.8)
Left body side, n=81	61.4 (2.8–428.9)

SD, standard deviation.

- Of 155 patients who participated in the TOWER study, spasticity was due to stroke in 85.2% and due to other neurologic causes in 14.8%.
- 137 patients completed the study. No patients withdrew due to lack of efficacy. Reasons for discontinuation of 18 patients included:
 - adverse events (n=5)
 - withdrawn consent (n=7)
 - predefined discontinuation criteria met (n=3)
 - loss to follow-up (n=3)
 - non-compliance (n=1)
 - other administrative reasons (n=1).
- Multiple discontinuation factors were entered for some patients.
- The majority of patients (>80%) received the planned maximum dose per injection cycle (**Table 2**).

Table 2. Doses administered per injection cycle

IncobotulinumtoxinA dose	Patients, n (%)
Cycle 1	N=155
375–<400 U	14 (9.0)
400 U	**141 (91.0)**
Cycle 2	N=152
560–<600 U	13 (8.6)
600 U	**138 (90.8)**
625–<700 U	1 (0.7)
Cycle 3	N=140
500–<600 U	1 (0.7)
600–<700 U	8 (5.7)
700–<800 U	15 (10.7)
800 U	**116 (82.9)**

- A total of 608 clinical patterns were treated in Cycle 1, 743 were treated in Cycle 2, and 811 were treated in Cycle 3; the mean number of patterns treated per patient increased from 3.9 (Cycle 1) to 5.8 (Cycle 3).
- In all injection cycles, the overall number of clinical patterns treated in the upper limb was higher than in the lower limb (Cycle 1: upper limb, 450; lower limb, 158; Cycle 2: upper limb, 517; lower limb, 219; Cycle 3: upper limb, 541; lower limb, 270).

Muscle tone

- AS responses (⩾1-point improvement) were seen in 364/608 (59.9%) patterns treated in Cycle 1 (155 patients), 431/743 (58.0%) patterns treated in Cycle 2 (152 patients), and 537/811 (66.2%) patterns treated in Cycle 3 (140 patients) (**Figure 2**).

Figure 2. The majority of patients showed improvement in AS score across all injection cycles

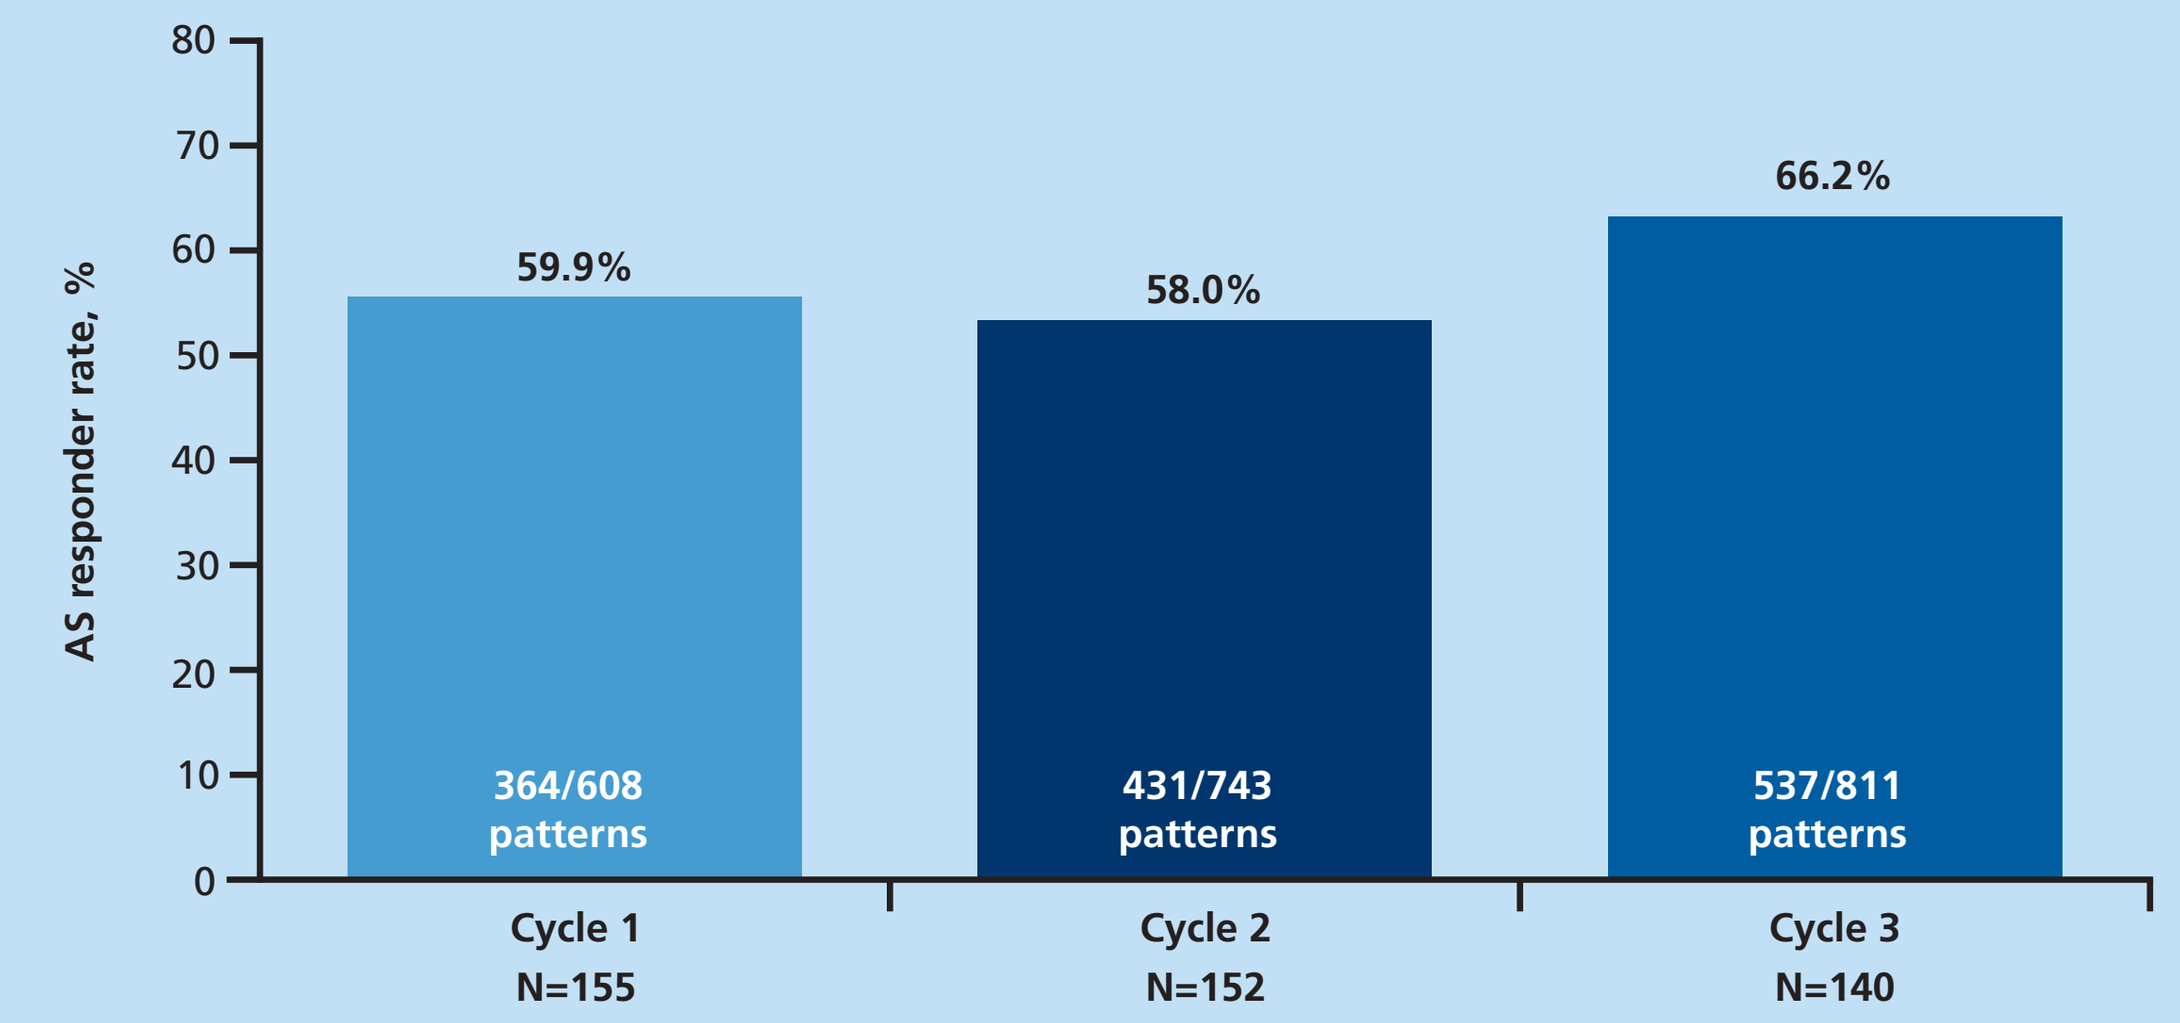
AS responder rate, %
80
70
60
50
40
30
20
10
0
59.9%
364/608 patterns
58.0%
431/743 patterns
66.2%
537/811 patterns
Cycle 1 N=155
Cycle 2 N=152
Cycle 3 N=140

Response was defined as ⩾1-point reduction of the AS score based on the change in AS between the baseline visit and Control Visit 1 (4 weeks post-injection) of each injection cycle; N=number of patients assessed at Control Visit 1 (4 weeks post-injection).
AS, Ashworth Scale.

- From injection to 4 weeks post-injection, mean ± SD REPAS score improved (decreased) significantly by 4.6 ± 3.9 in Cycle 1, 5.9 ± 4.2 in Cycle 2, and 7.1 ± 4.8 in Cycle 3 (p<0001; paired student's t-test) (**Figure 3**).

Global efficacy assessment

- In each injection cycle, a majority in each group (investigators and patients) globally rated efficacy as either 'good' or 'very good' (**Figure 4**).
- There was a shift to better outcomes over the course of the study for both groups (**Figure 4**).

Safety

- IncobotulinumtoxinA dose escalation did not lead to an increased incidence of AEs (Cycle 1: 36.1%; Cycle 2: 37.5%; Cycle 3: 25.7%) or treatment-related AEs (Cycle 1: 4.5 %; Cycle 2: 5.3%; Cycle 3: 2.9%). No treatment-related serious AEs occurred (**Table 3**).
- Overall, the most common AEs were falls (7.7 %), arthralgia, diarrhea, and nasopharyngitis (each 6.5%) (**Table 4**).
- Overall, discontinuations due to AEs were low (3.2%).
- There were no cases of clinical non-responsiveness due to antibodies.

Figure 3. Mean REPAS score decreased significantly (p<0.0001) across all injection cycles

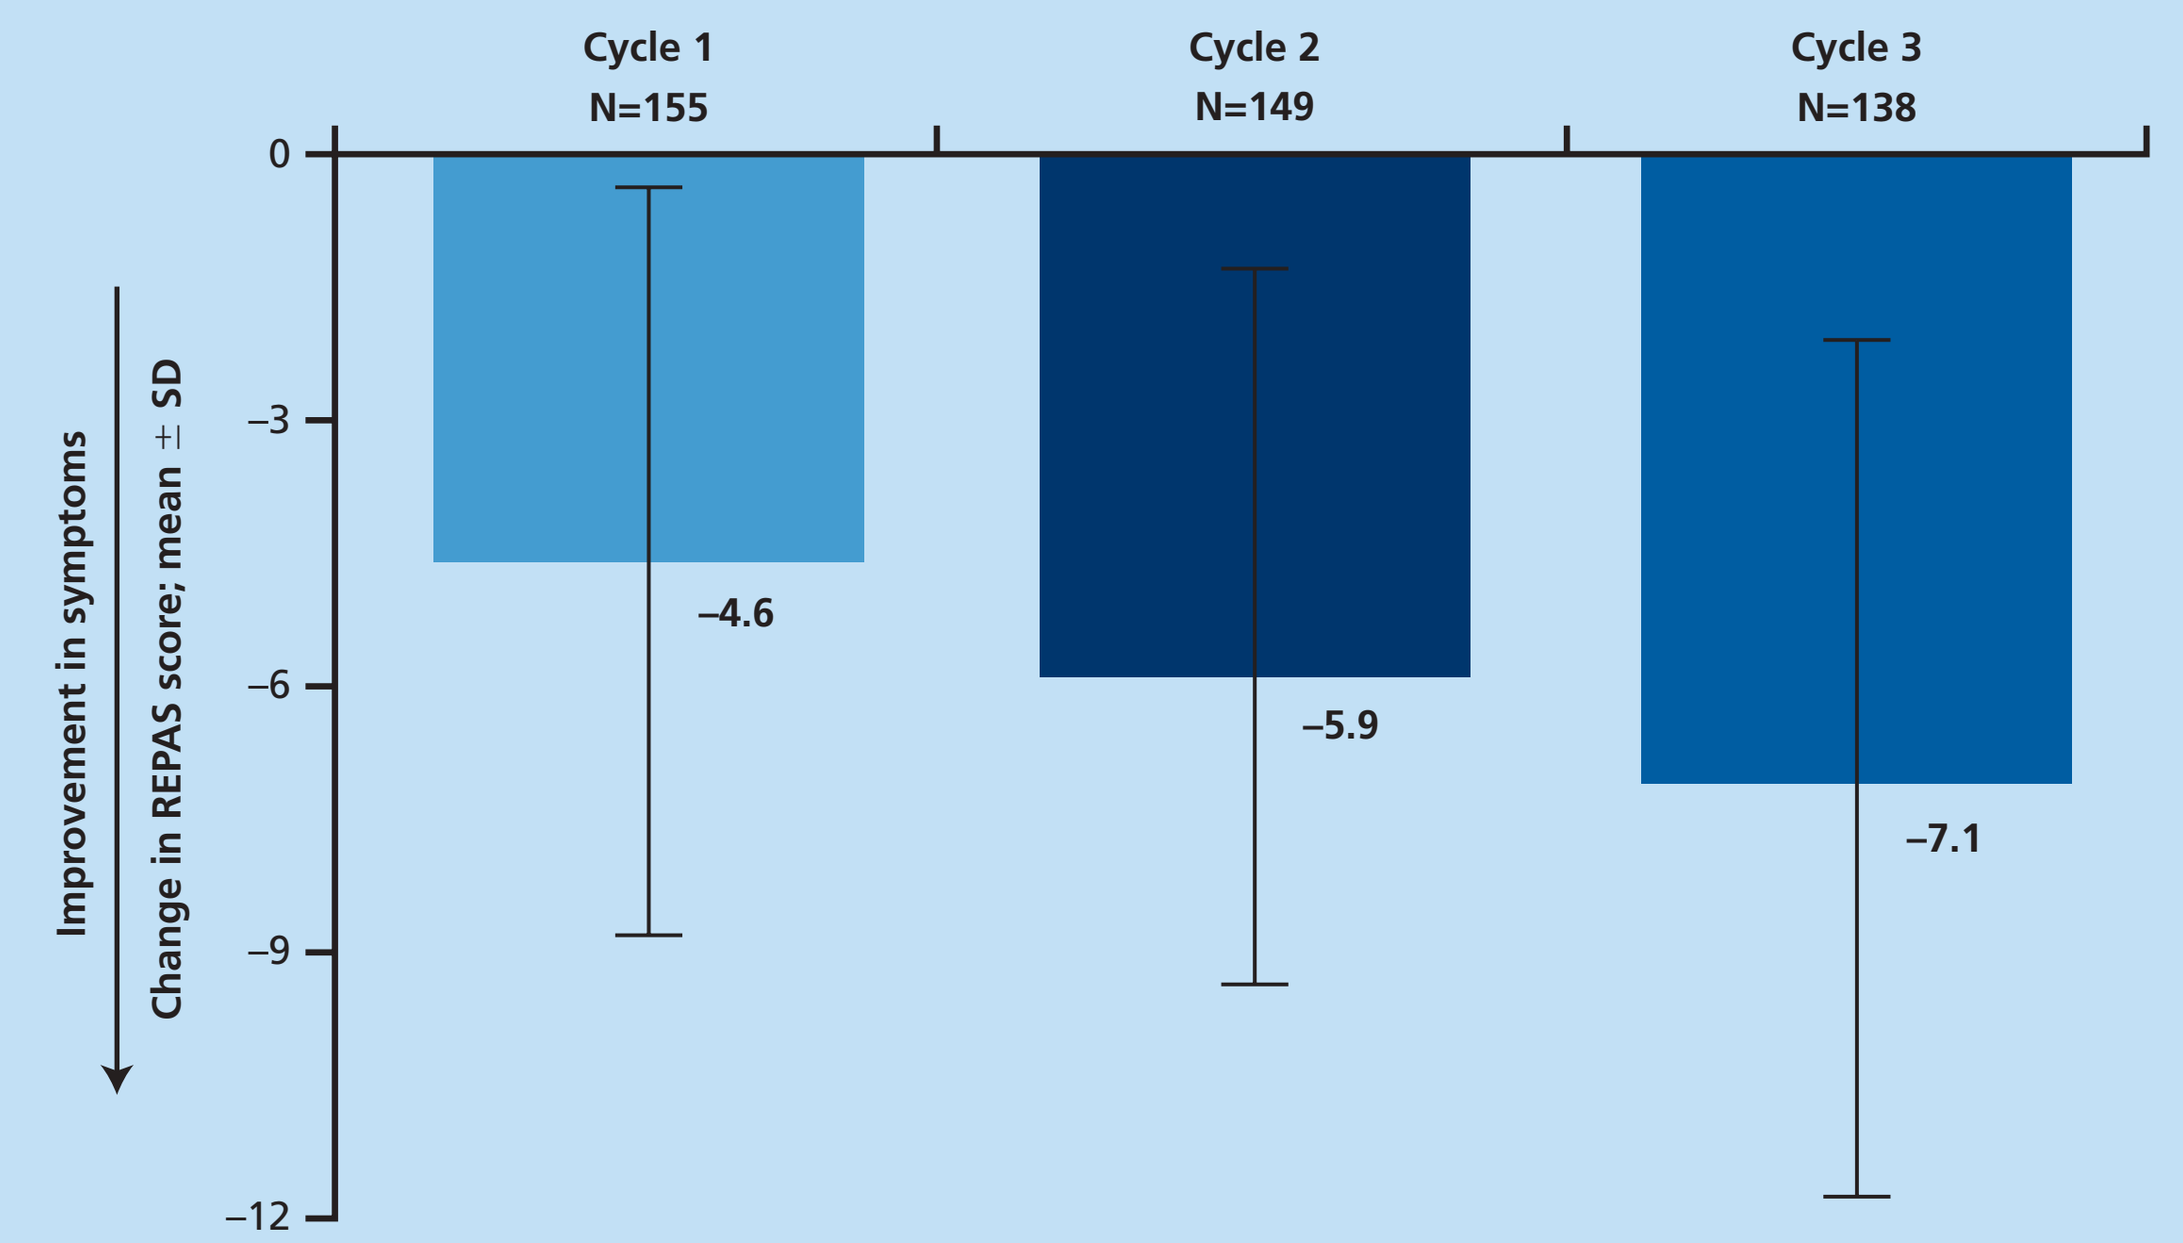
Cycle 1 N=155
Cycle 2 N=149
Cycle 3 N=138
Improvement in symptoms
Change in REPAS score; mean ± SD
0
–3
–6
–9
–12
–4.6
–5.9
–7.1

Change between injection cycle baseline and control visits for each injection cycle is shown. The p-value is based on paired student's t-test comparing the baseline visit and Control Visit 1 of each injection cycle; N=number of patients assessed at Control Visit 1.
REPAS, resistance to passive movement scale; SD, standard deviation.

Figure 4. Investigator and patient global efficacy assessments increased across all injection cycles

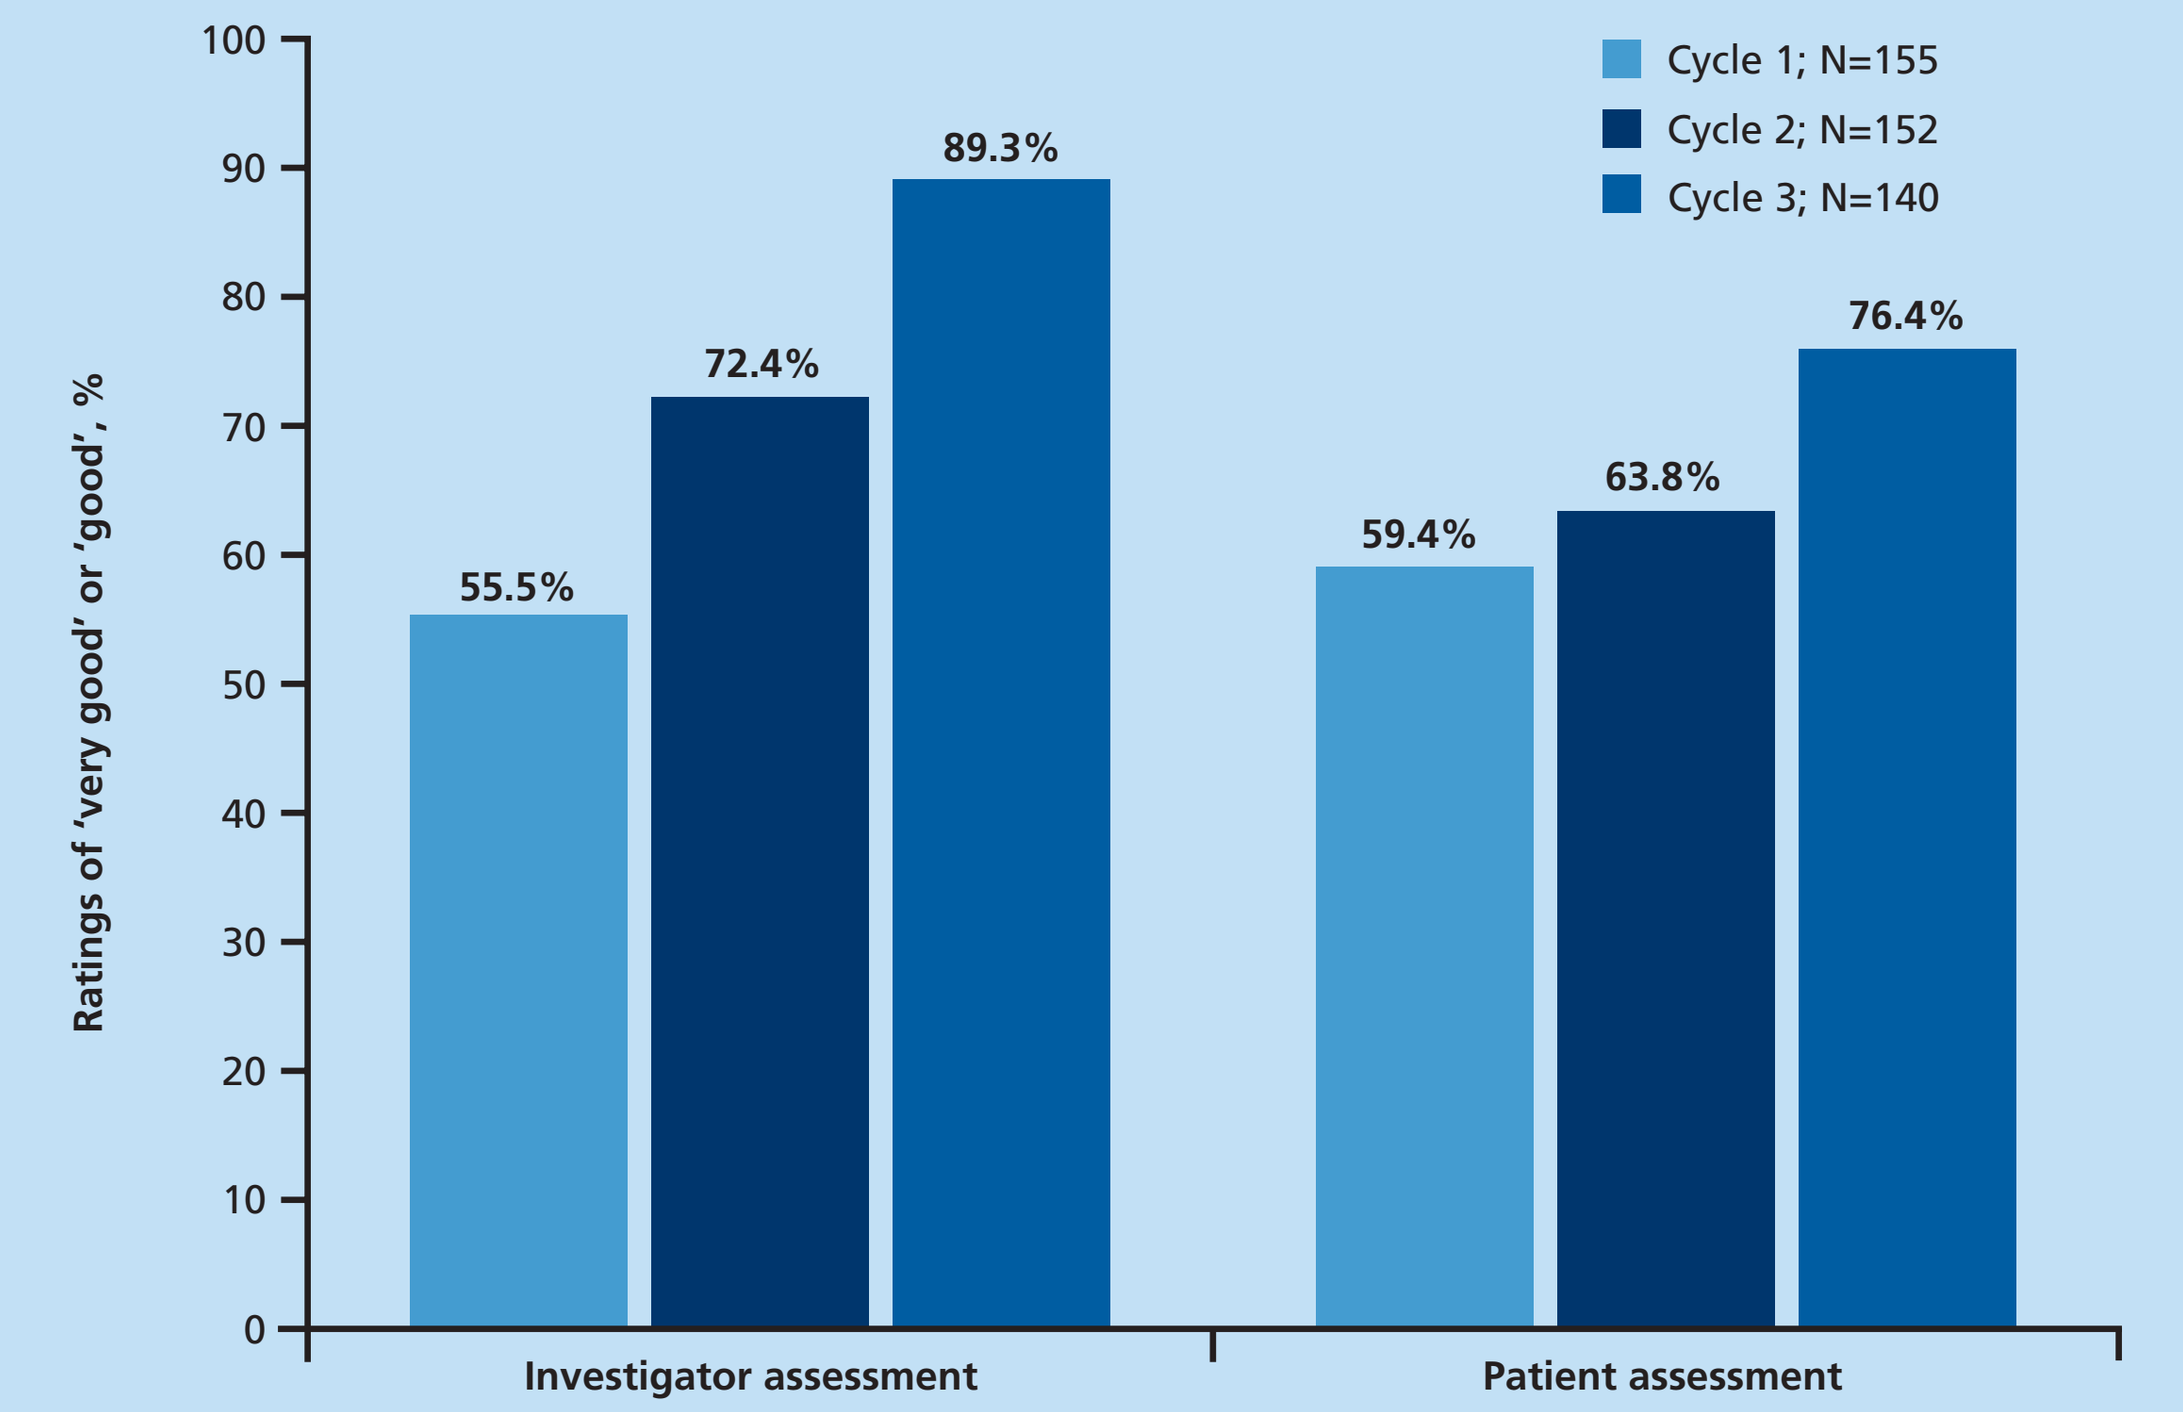
Ratings of 'very good' or 'good', %
100
90
80
70
60
50
40
30
20
10
0
Cycle 1; N=155
Cycle 2; N=152
Cycle 3; N=140
55.5%
72.4%
89.3%
59.4%
63.8%
76.4%
Investigator assessment
Patient assessment

The global assessment of efficacy was performed using the 4-point Likert scale where: 1=very good, 2=good, 3=moderate, and 4=poor.
Global assessment of efficacy was performed for Cycle 1 and Cycle 2 at the baseline visit of the following injection cycle (visits 5 and 8; Figure 1).
For Cycle 3, it was performed at the end of the Cycle 3 visit (visit 10; Figure 1).

Table 3. Summary of adverse events

Patients, n (%)	Overall N=155	Cycle 1 400 U N=155	Cycle 2 600 U N=152	Cycle 3	
				All doses N=140	800 U N=116
Any AE	97 (62.6)	56 (36.1)	57 (37.5)	36 (25.7)	33 (28.4)
Any treatment-related AE	17 (11.0)	7 (4.5)	8 (5.3)	4 (2.9)	3 (2.6)
Any treatment-related AESI*	8 (5.2)	2 (1.3)	4 (2.6)	3 (2.1)	3 (2.6)
Any serious AE	17 (11.0)	4 (2.6)	11 (7.2)	3 (2.1)	3 (2.6)
Any treatment-related serious AE	0	0	0	0	0

*AEs were classified as AESIs on the basis of a predefined list of AEs that could potentially indicate toxin spread, regardless of whether an AE was regarded as treatment related by the investigator.
AE, adverse event; AESI, adverse events of special interest.

Table 4. Most frequent adverse events

Patients, n (%)	Overall N=155	Cycle 1 400 U N=155	Cycle 2 600 U N=152	Cycle 3	
				All doses N=140	800 U N=116
Fall	12 (7.7)	5 (3.2)	2 (1.3)	8 (5.7)	8 (6.9)
Arthralgia	10 (6.5)	4 (2.6)	2 (1.3)	5 (3.6)	5 (4.3)
Diarrhea	10 (6.5)	1 (0.6)	5 (3.3)	6 (4.3)	5 (4.3)
Nasopharyngitis	10 (6.5)	4 (2.6)	5 (3.3)	3 (2.1)	3 (2.6)
Musculoskeletal pain	8 (5.2)	2 (1.3)	2 (1.3)	4 (2.9)	4 (3.4)
Headache	7 (4.5)	4 (2.6)	3 (2.0)	1 (0.7)	1 (0.9)
Fatigue	6 (3.9)	3 (1.9)	1 (0.7)	3 (2.1)	2 (1.7)
Contusion	5 (3.2)	3 (1.9)	0	2 (1.4)	2 (1.7)
Convulsion	5 (3.2)	2 (1.3)	3 (2.0)	0	0
Dysphagia	5 (3.2)	2 (1.3)	1 (0.7)	2 (1.4)	2 (1.7)
Edema peripheral	5 (3.2)	5 (3.2)	0	0	0
Hyperpyrexia	5 (3.2)	0	3 (2.0)	2 (1.4)	2 (1.7)

Conclusions

- TOWER was the first prospective study to examine the safety and efficacy of increasing incobotulinumtoxinA doses for patients with upper- and lower-limb spasticity.
- Dose escalation (400–800 U) resulted in improvements in muscle tone and global assessments of efficacy.
- Increasing doses and repeated injection did not increase the incidence of AEs, and did not compromise safety or tolerability.
- The findings from this study support the conclusion that escalating incobotulinumtoxinA doses (400–800 U) allow treatment of an increasing number of spasticity patterns without compromising safety or tolerability, leading to enhanced efficacy based on improved muscle tone and physician global assessments.

References

1. Esquenazi A et al. Eur J Neurol 2010; 17 (Suppl 2): 1–8.
2. Simpson DM et al. Neurology 2016; 86: 1818–1826.
3. Wissel J et al. J Rehabil Med 2009; 41: 13–25.
4. Baricich A et al. J Neural Transm 2015; 122: 1283–1287.
5. Dressler D. Eur J Neurol 2009; 16 (Suppl 2): 2–5.
6. Dressler D et al. J Neural Transm 2015; 122: 327–333.
7. Intiso D et al. NeuroRehabilitation 2014; 34: 515–522.
8. Santamato A et al. J Neural Transm 2013; 120: 469–476.
9. Santamato A et al. J Neurol Sci 2015; 350: 1–6.
10. Elovic EP et al. Muscle Nerve 2016; 53: 415–421.
11. Kaňovský P et al. Clin Neuropharmacol 2009; 32: 259–265.
12. Kaňovský P et al. J Rehabil Med 2011; 43: 486–492.
13. Dressler D. Eur J Neurol 2012; 19: 385–389.
14. Wissel J et al. PM&R 2013; 5 (Suppl): S142–S143 (primary analysis data from this study have been submitted for publication in Neurology).
15. Ashworth B. Practitioner 1964; 192: 540–542.
16. Platz T et al. Disabil Rehabil 2008; 30: 44–53.

Acknowledgments

This study was sponsored by Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Medical writing support was provided by Heather Rafferty, PhD, of Complete Medical Communications, funded by Merz Pharmaceuticals GmbH.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017