SPAsticity in PractiCE (SPACE) – an international, observational study of botulinum toxin type A in spasticity

Julian Harriss,[1] Jörg Wissel,[2] Olivier Simon,[3] Kati Sternberg,[3] Nicolas Roche,[4] Carlos Cantú-Brito,[5] Svetlana Khatkova,[6] Patrik Säterö[7]

[1]King's College London, St Thomas' Hospital, London, UK; [2]Vivantes Hospital Spandau, Berlin, Germany; [3]Merz Pharmaceuticals GmbH, Frankfurt, Germany; [4]Raymond-Poincaré Hospital, AP-HP, Garches, France; [5]Instituto Nacional de Ciencias Médicas y Nutrición Salvador Zubirán, Mexico City, Mexico; [6]Federal State Hospital for Treatment and Rehabilitation, Ministry of Health, Moscow, Russia; [7]Sahlgrenska Universitetssjukhuset, Göteborg, Sweden

Background

- Spasticity can be one of the consequences of common neurologic conditions such as stroke, multiple sclerosis, and injuries to the brain and spinal cord.[1]
- Individuals with spasticity may experience muscle stiffness, an impaired ability to perform functional tasks, painful muscle contractions and spasms, and a limited range of motion.[2]
- Botulinum neurotoxin type A (BoNT-A) injections form a key component of the multidisciplinary management of spasticity.[3–5] However, real-world data on the efficacy, safety, and effect on quality of life (QoL) of BoNT-A treatment in routine clinical practice are limited.

Objective

- To collect real-world routine clinical practice data on effectiveness, safety, and QoL for BoNT-A treatment of focal spasticity.

Methods

Study design

- SPACE was an international, observational study that followed BoNT-A-naïve adults with spasticity in 106 centers in Canada, France, Germany, Italy, Mexico, Russia, Spain, Sweden, and the UK.
- Patients received any BoNT-A formulation available in their country according to a treatment regimen (doses, dilutions, injection sites, treatment intervals) at the discretion of their physician.
- The physician's choice of BoNT treatment regimen for a patient was decided prior to and independently of study enrollment.
- Treatment goals, injection techniques, doses used, muscles treated, QoL (as assessed using the European Quality of Life-5 Dimensions [EQ-5D] questionnaire), and adverse drug reactions (ADRs) were documented.
- The follow-up period for each patient was ⩽2 years.

Eligibility criteria

- Patients were ⩾18 years of age with spasticity of any etiology and required BoNT-A treatment.
- Patients had not previously received BoNT-A or BoNT-B treatment for any reason.
- Sufficient understanding of the primary local language to be able to complete study questionnaires was a prerequisite.
- Patients were not participating in an interventional study at screening and nor did they plan to enroll in a study that included treatment with BoNT-A.

Data collected

- Baseline patient demographics and disease characteristics data, including duration, etiology, and pattern of spasticity.
- Data about participating physicians, including speciality, experience with BoNT injections, and dosing preferences.
- Details of BoNT-A injections, including the guidance used by physicians to administer treatment, the specific formulation (incobotulinumtoxinA [Xeomin®, Merz], onabotulinumtoxinA [Botox®, Allergan] or abobotulinumtoxinA [Dysport®, Ipsen]), dosage of BoNT-A used, muscles treated, and number of injection sites.
- The three most important treatment goals identified by each patient at each injection visit.
- QoL assessed using the EQ-5D.
- Incidence of ADRs.
- The tolerability of the previous treatment on a 1–4 rating scale (1=very good; 2=good; 3=moderate; 4=poor) assessed by physicians at the end of each treatment cycle.

Results

Patients and treatments

- A total of 701 patients were enrolled. Of these, data from 687 patients with a confirmed date of written consent and a reported BoNT-A formulation at the first injection were analyzed for effectiveness and health-related outcomes.
- The most frequent etiologies of spasticity were stroke (64.6%), multiple sclerosis (9.3%), and brain injury (6.0%) (**Table 1**).
- At baseline, the most common patterns of spasticity were flexed elbow (58.1%) and wrist (50.9%) in the upper limbs, and equinovarus (32.2%) and extended knee (21.0%) in the lower limbs (**Figure 1**).
- The most frequently treated muscles in the upper limbs were flexor digitorum superficialis and biceps brachii (**Figure 2a**). Data shown are for the first and fourth treatments; results after the other treatments were similar.
- In the lower limbs, the most frequently treated muscles were gastrocnemius medial and soleus (**Figure 2b**).
 - Notably, the quadriceps muscles were not injected frequently, suggesting that the pattern extended knee was not treated in many patients with this pattern.
- While electrostimulation (18.5%) was the most frequently used guidance technique for the administration of BoNT-A, many injections (40.2%) were given without use of a guidance technique (**Table 2**).
- The most common primary treatment goals were improvements in limb function (mobility and/or dexterity), pain relief, and improvements in self-care (**Figure 3**).
- Treatment with all BoNT-A formulations was associated with an improvement in health-related QoL for the duration of the study (**Figure 4**).

Table 1. Patient baseline characteristics

Parameter	IncobotulinumtoxinA N=465	OnabotulinumtoxinA N=142	AbobotulinumtoxinA N=80	All N=687
Male gender, n (%)	283 (60.9)	83 (58.5)	55 (68.8)	421 (61.3)
Age, years; mean (SD)	54.6 (15.6)	55.6 (15.3)	56.2 (15.3)	55.0 (15.5)
BMI, kg/m^2; mean SD	25.8 (5.1)	25.6 (5.7)	25.7 (4.6)	25.7 (5.1)
Time since spasticity-causing event, years; mean (SD)	7.2 (10.1)	6.4 (9.7)	7.7 (11.8)	7.1 (10.6)
Time since onset of spastic symptoms, years; mean (SD)	5.6 (9.2)	4.5 (8.0)	5.7 (12.0)	5.4 (9.3)
Etiology of spasticity, n (%)				
Stroke	310 (66.7)	85 (59.9)	49 (61.3)	444 (64.6)
Brain injury	33 (7.1)	4 (2.8)	4 (5.0)	41 (6.0)
Multiple sclerosis	36 (7.7)	19 (13.4)	9 (11.3)	64 (9.3)
Spinal cord injury	19 (4.1)	8 (5.6)	3 (3.8)	30 (4.4)
Cerebral palsy	10 (2.2)	4 (2.8)	4 (5.0)	18 (2.6)
Other/missing	57 (12.3)	22 (15.5)	11 (13.8)	90 (13.1
Distribution of paresis, n (%)				
Hemiplegia	377 (81.1)	106 (74.6)	58 (72.5)	541 (78.7)
Diplegia	44 (9.5)	23 (16.2)	12 (15.0)	79 (11.5)
Quadriplegia	41 (8.8)	9 (6.3)	8 (10.0)	58 (8.4)
Missing	3 (0.6)	4 (2.8)	2 (2.5)	9 (1.3)

BMI, body mass index; SD, standard deviation.

Physicians

- Overall, 171 physicians from nine countries participated, with a mean (standard deviation; SD) of 15.7 (8.7) years' experience in medical practice and 9.2 (5.8) years' experience with BoNT injections.
- The most common specialities of the physicians included neurologists (n=81, 47.4%) and physiatrists (n=48, 28.1%). Physicians could have more than one speciality.
- The majority of physicians (n=101, 59.1%) would have injected higher doses of BoNT if there had been no labeling restrictions.
- The mean (SD) doses that physicians wanted to inject were 651.8 (191.6) units for incobotulinumtoxinA, 640.3 (170.4) units for onabotulinumtoxinA, and 1751.9 (844.2) units for abobotulinumtoxinA.

Safety

- All 701 enrolled patients were included in the safety analyses.
 - Because in Mexico the incidence of non-treatment-related adverse events was collected in addition to the incidence of ADRs, non-Mexican (n=600) and Mexican patients (n=101) were analyzed separately. Here, data for non-Mexican patients are presented.
- The overall number of ADRs was low, with 16/600 (2.7%) non-Mexican patients experiencing 27 ADRs.
- The proportion of patients with ADRs was similar across the BoNT-A formulation groups (incobotulinumtoxinA, 3.0% [11/369]; onabotulinumtoxinA, 2.1% [3/142]; abobotulinumtoxinA, 2.7% [2/75]).
- Eight patients experienced serious ADRs, with a similar incidence across the BoNT-A formulation groups (incobotulinumtoxinA, 1.4% [5/369]; onabotulinumtoxinA, 1.4% [2/142]; abobotulinumtoxinA, 1.3% [1/75]).
- One patient died during the course of the study; the death was deemed unlikely to be related to treatment.
- The tolerability of BoNT-A formulations was rated 'very good' or 'good' for a large majority of patients throughout the study (**Figure 5**).

Table 2. Guidance technique used for administration of botulinum neurotoxin type A injection

Guidance used, n (%)	IncobotulinumtoxinA N=465	OnabotulinumtoxinA N=142	AbobotulinumtoxinA N=80	All N=687
None	219 (47.1)	27 (19.0)	30 (37.5)	276 (40.2)
EMG	57 (12.3)	25 (17.6)	19 (23.8)	101 (14.7)
Electrostimulation	80 (17.2)	34 (23.9)	13 (16.3)	127 (18.5)
Sonography	67 (14.4)	16 (11.3)	10 (12.5)	93 (13.5)
EMG/electrostimulation	23 (4.9)	28 (19.7)	5 (6.3)	56 (8.2)
EMG/sonography	5 (1.1)	0 (0.0)	0 (0.0)	5 (0.7)
Electrostimulation/sonography	1 (0.2)	4 (2.8)	0 (0.0)	5 (0.7)
EMG electrostimulation/sonography	0 (0.0)	1 (0.7)	0 (0.0)	1 (0.1)
Other	8 (1.7)	7 (4.9)	3 (3.8)	18 (2.6)
Missing	5 (1.1)	0 (0.0)	0 (0.0)	5 (0.7)

EMG, electromyography.

Figure 1. Most frequent spasticity patterns present at baseline, N=689

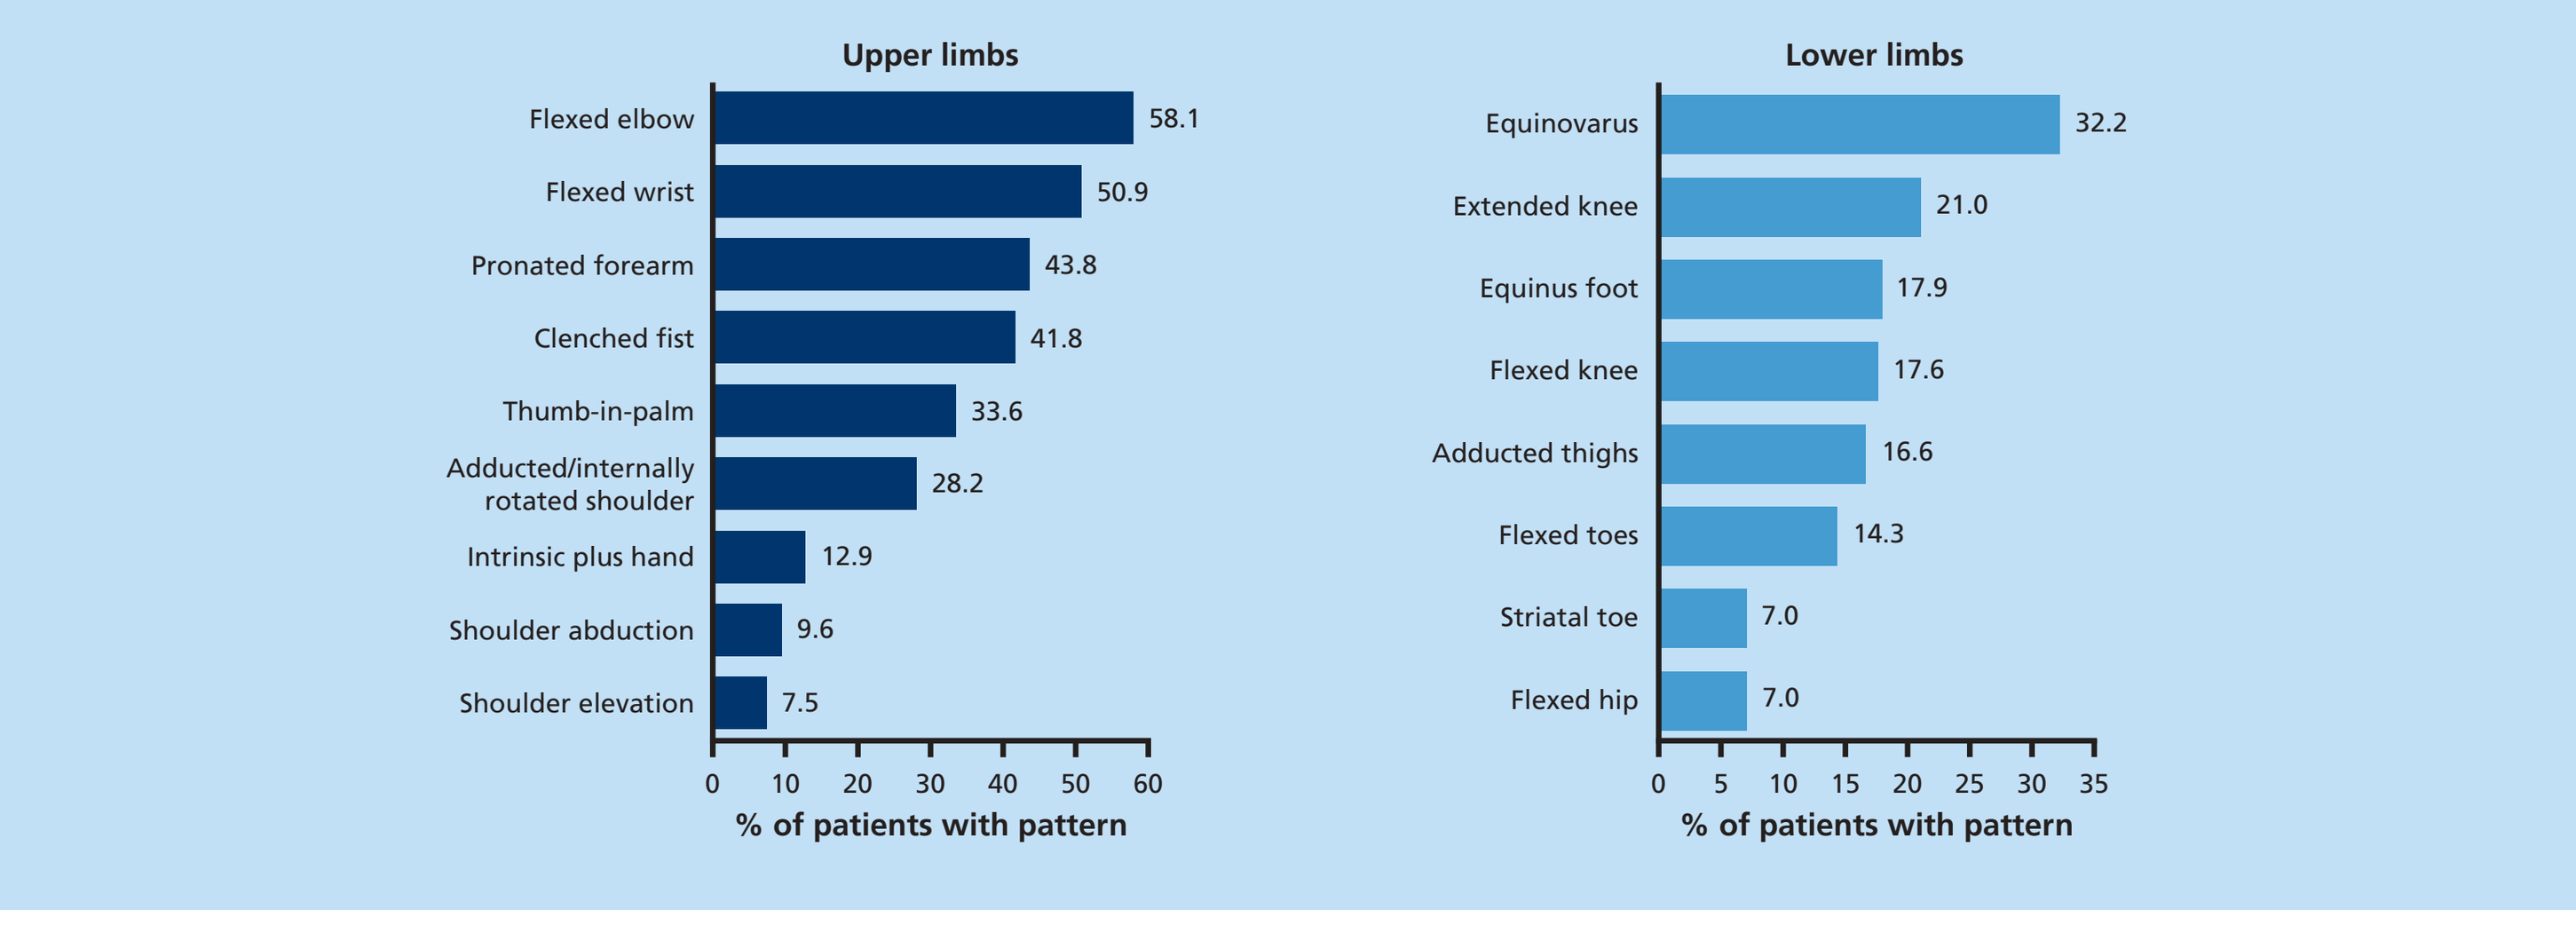
Upper limbs
Flexed elbow 58.1
Flexed wrist 50.9
Pronated forearm 43.8
Clenched fist 41.8
Thumb-in-palm 33.6
Adducted/internally rotated shoulder 28.2
Intrinsic plus hand 12.9
Shoulder abduction 9.6
Shoulder elevation 7.5
% of patients with pattern
Lower limbs
Equinovarus 32.2
Extended knee 21.0
Equinus foot 17.9
Flexed knee 17.6
Adducted thighs 16.6
Flexed toes 14.3
Striatal toe 7.0
Flexed hip 7.0
% of patients with pattern

Figure 2. Most frequently treated muscles in the a) upper and b) lower limbs; botulinum neurotoxin type A, Injection Cycle 1

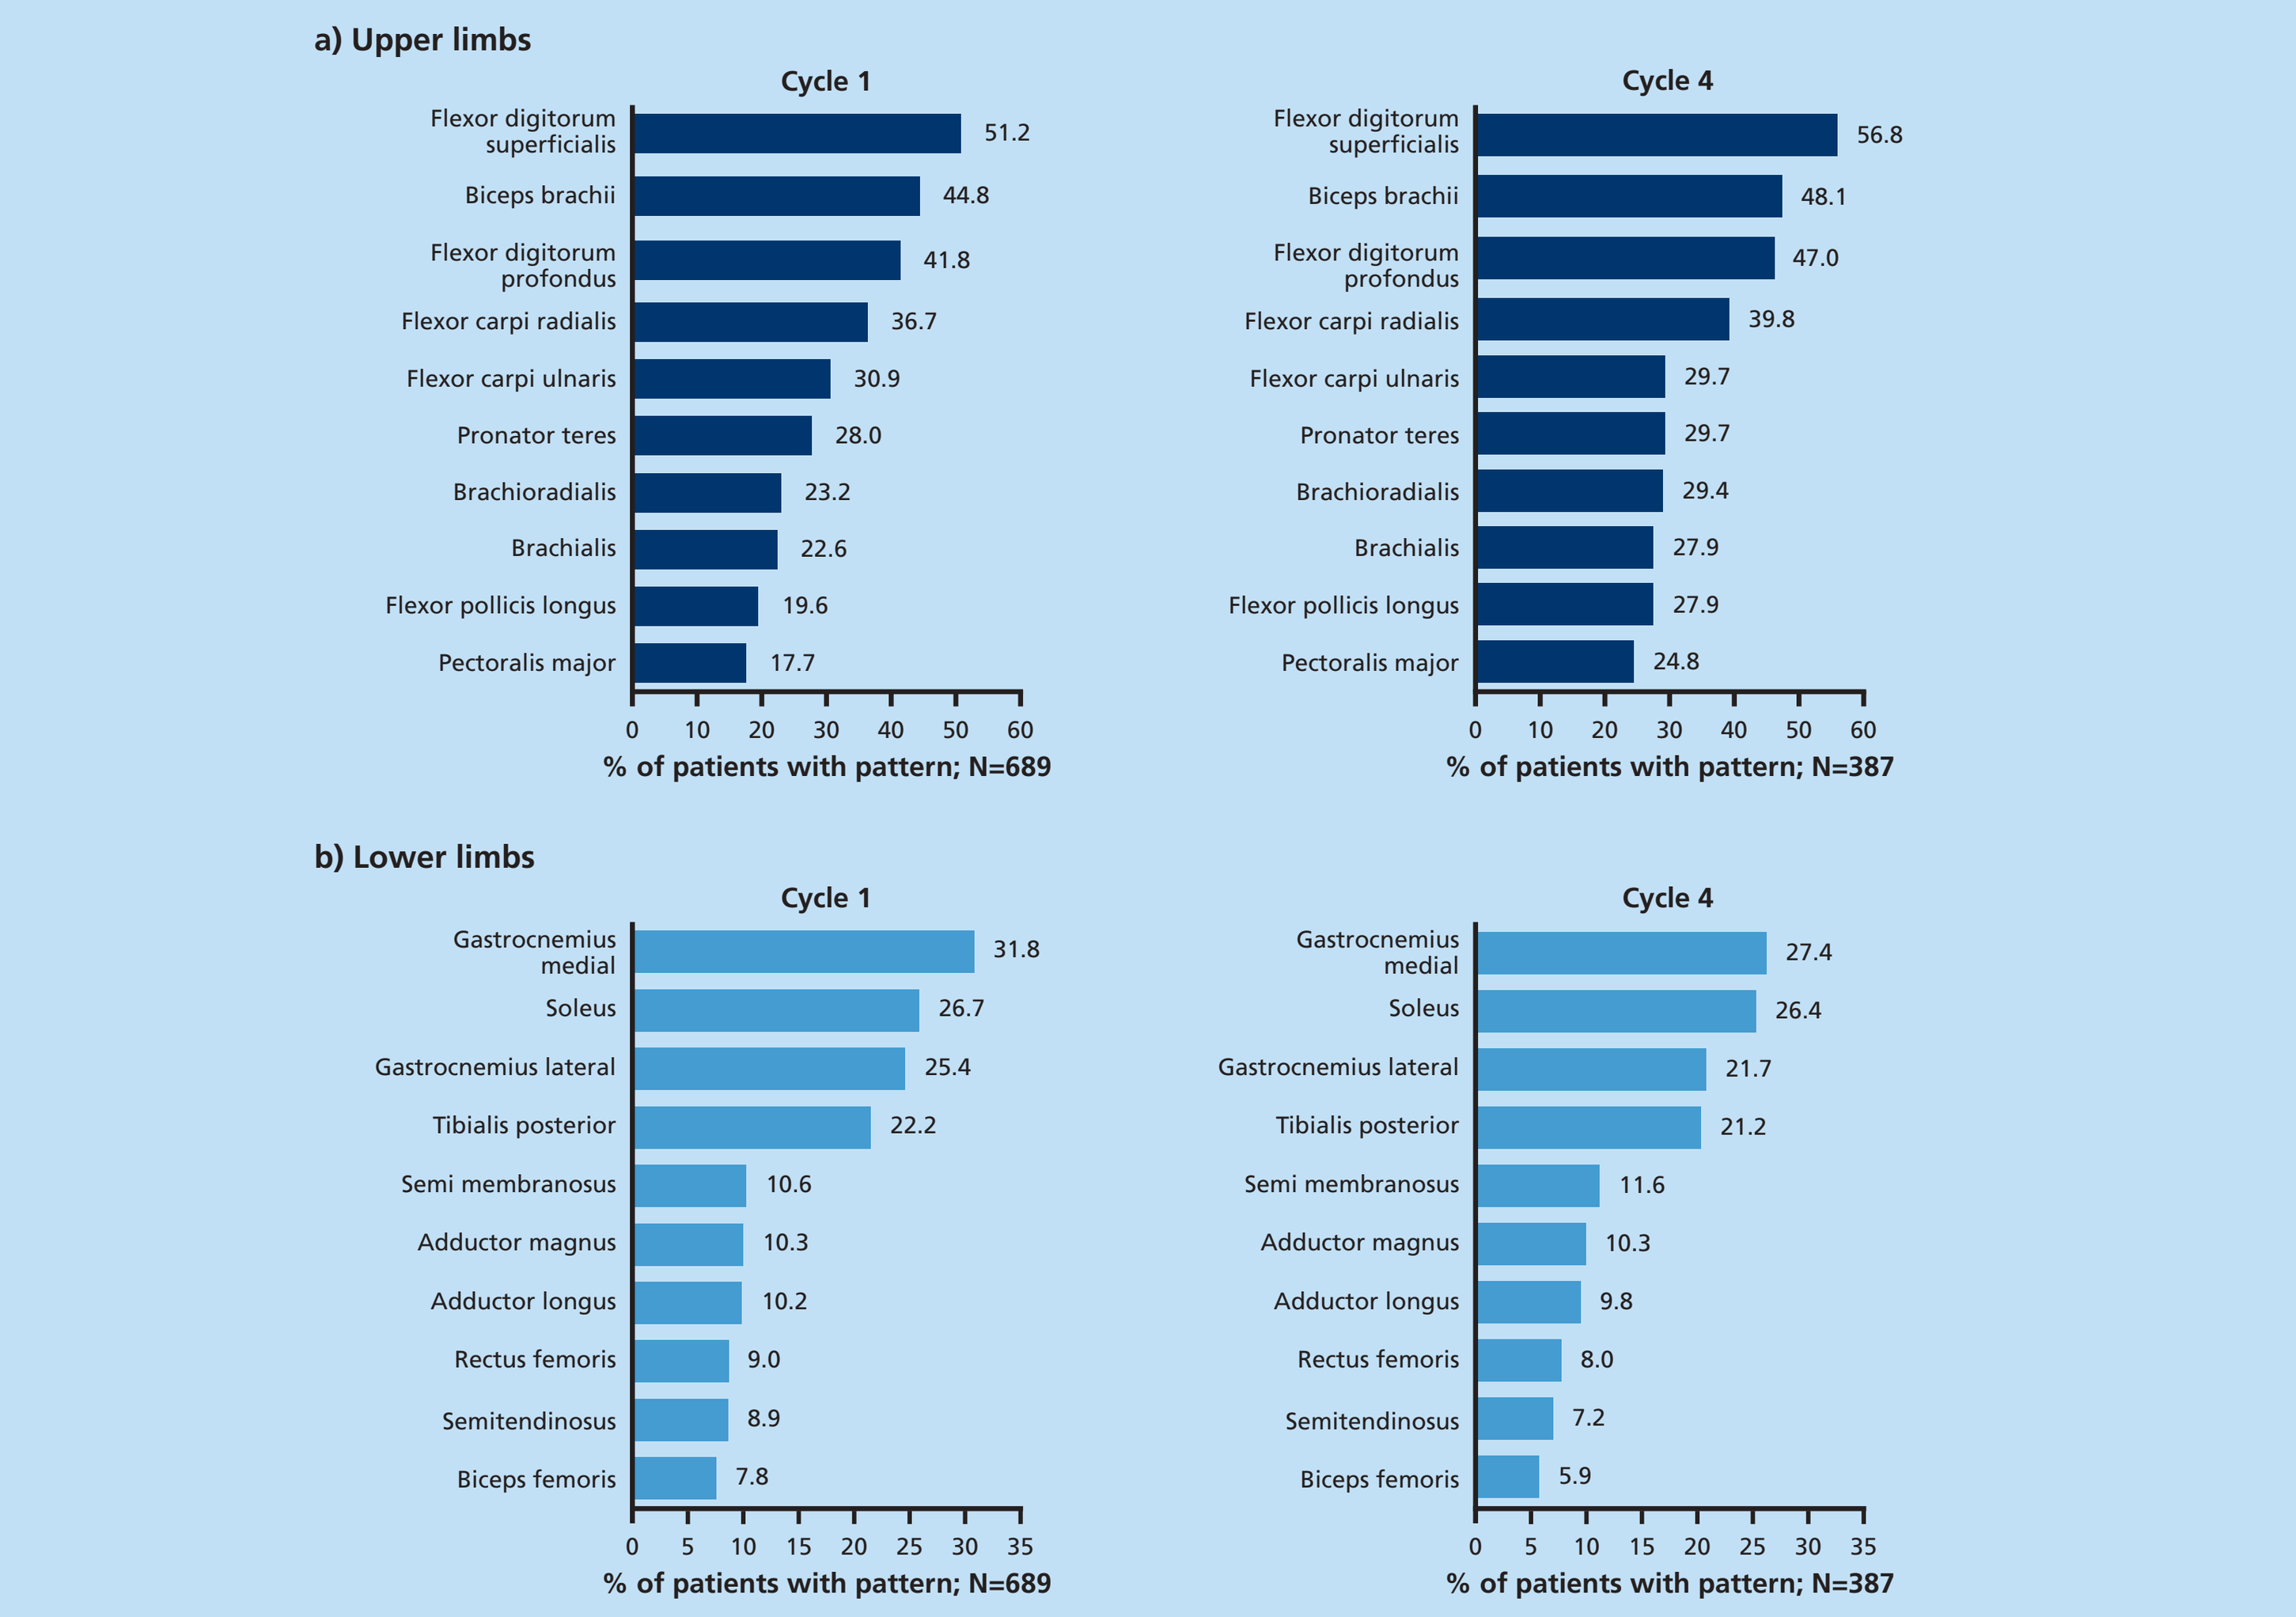
a) Upper limbs
Cycle 1
Flexor digitorum superficialis 51.2
Biceps brachii 44.8
Flexor digitorum profondus 41.8
Flexor carpi radialis 36.7
Flexor carpi ulnaris 30.9
Pronator teres 28.0
Brachioradialis 23.2
Brachialis 22.6
Flexor pollicis longus 19.6
Pectoralis major 17.7
% of patients with pattern; N=689
Cycle 4
Flexor digitorum superficialis 56.8
Biceps brachii 48.1
Flexor digitorum profondus 47.0
Flexor carpi radialis 39.8
Flexor carpi ulnaris 29.7
Pronator teres 29.7
Brachioradialis 29.4
Brachialis 27.9
Flexor pollicis longus 27.9
Pectoralis major 24.8
% of patients with pattern; N=387
b) Lower limbs
Cycle 1
Gastrocnemius medial 31.8
Soleus 26.7
Gastrocnemius lateral 25.4
Tibialis posterior 22.2
Semi membranosus 10.6
Adductor magnus 10.3
Adductor longus 10.2
Rectus femoris 9.0
Semitendinosus 8.9
Biceps femoris 7.8
% of patients with pattern; N=689
Cycle 4
Gastrocnemius medial 27.4
Soleus 26.4
Gastrocnemius lateral 21.7
Tibialis posterior 21.2
Semi membranosus 11.6
Adductor magnus 10.3
Adductor longus 9.8
Rectus femoris 8.0
Semitendinosus 7.2
Biceps femoris 5.9
% of patients with pattern; N=387

Figure 3. Most frequent primary goals defined

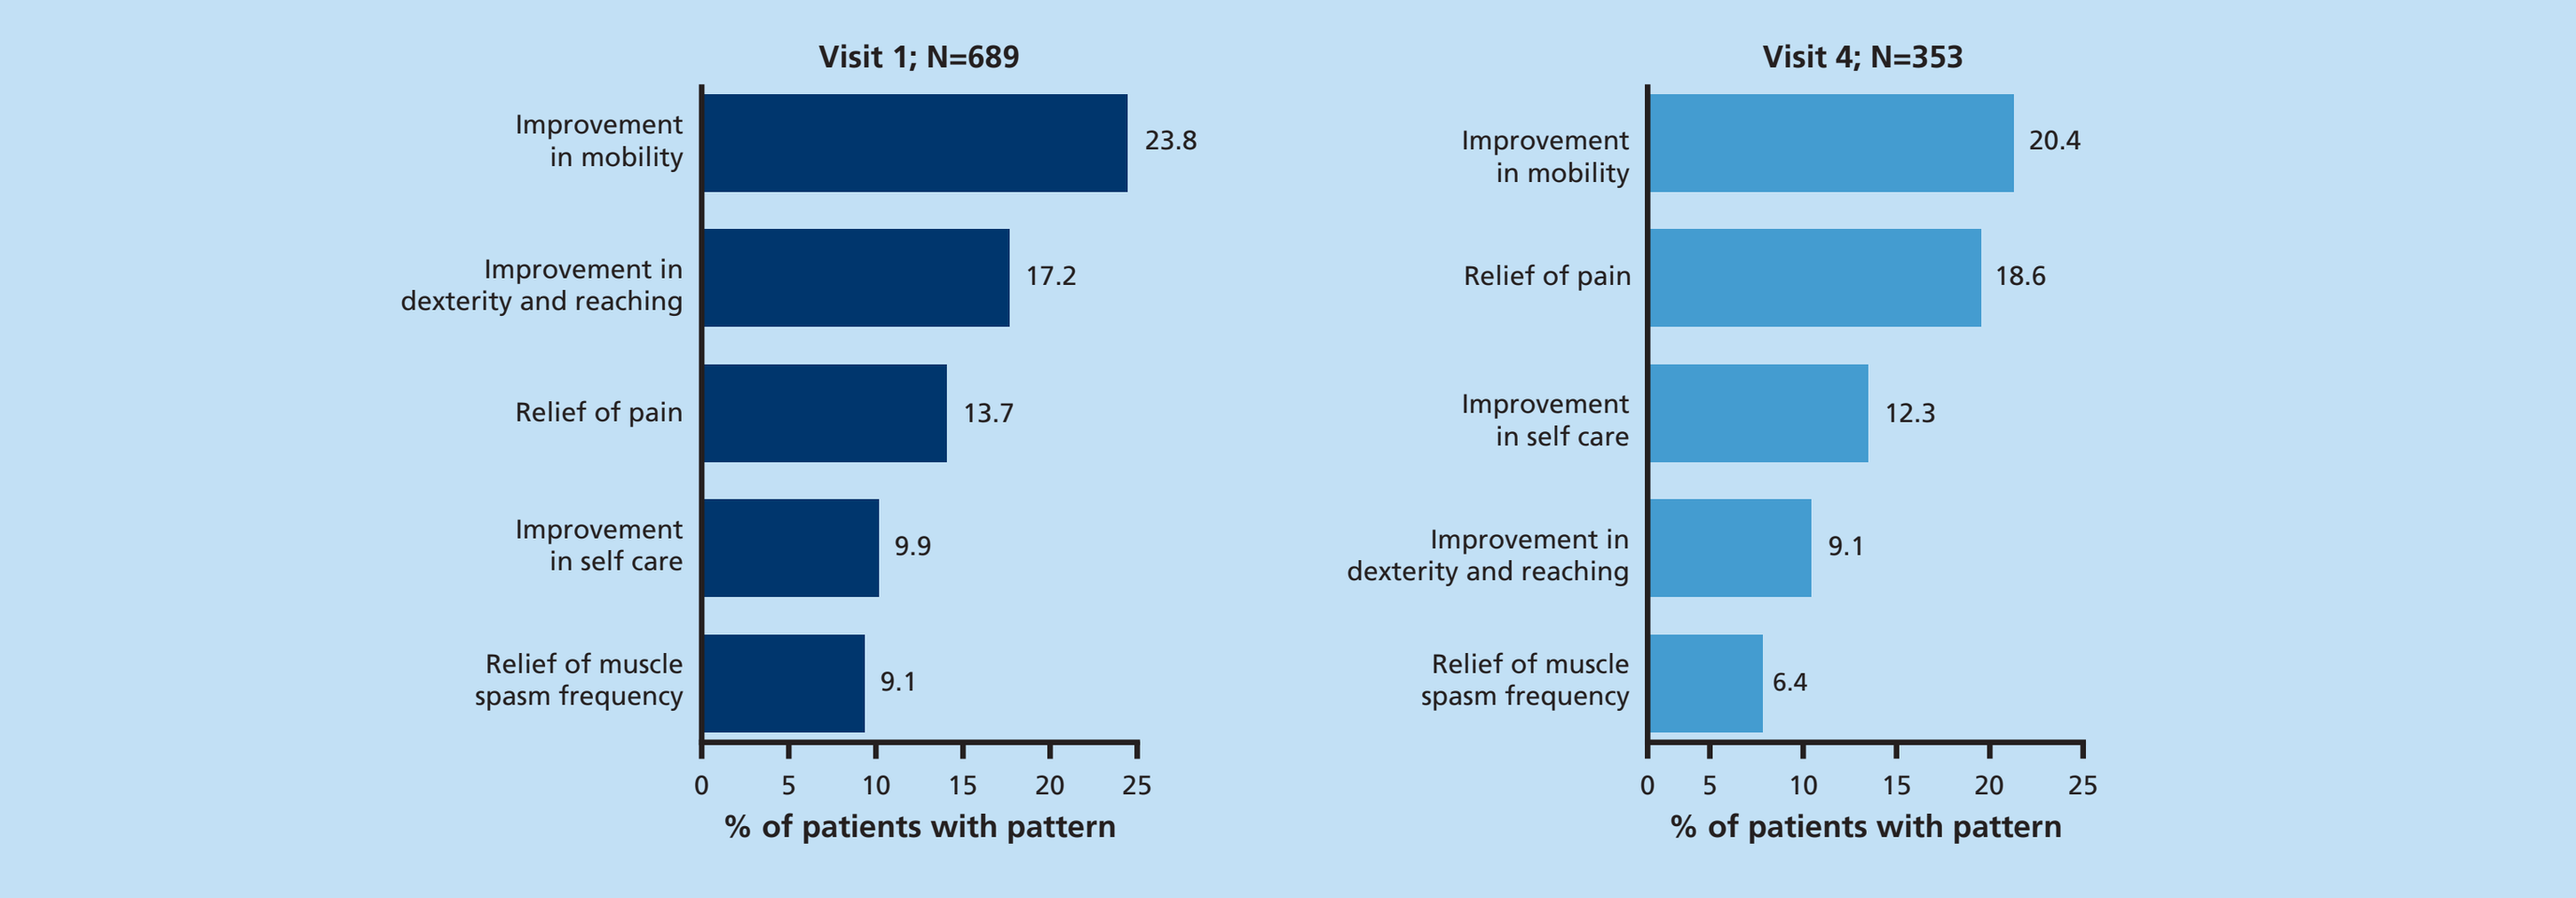
Visit 1; N=689
Improvement in mobility 23.8
Improvement in dexterity and reaching 17.2
Relief of pain 13.7
Improvement in self care 9.9
Relief of muscle spasm frequency 9.1
% of patients with pattern
Visit 4; N=353
Improvement in mobility 20.4
Relief of pain 18.6
Improvement in self care 12.3
Improvement in dexterity and reaching 9.1
Relief of muscle spasm frequency 6.4
% of patients with pattern

Figure 4. Health-related quality of life (EQ-5D visual analog scale)

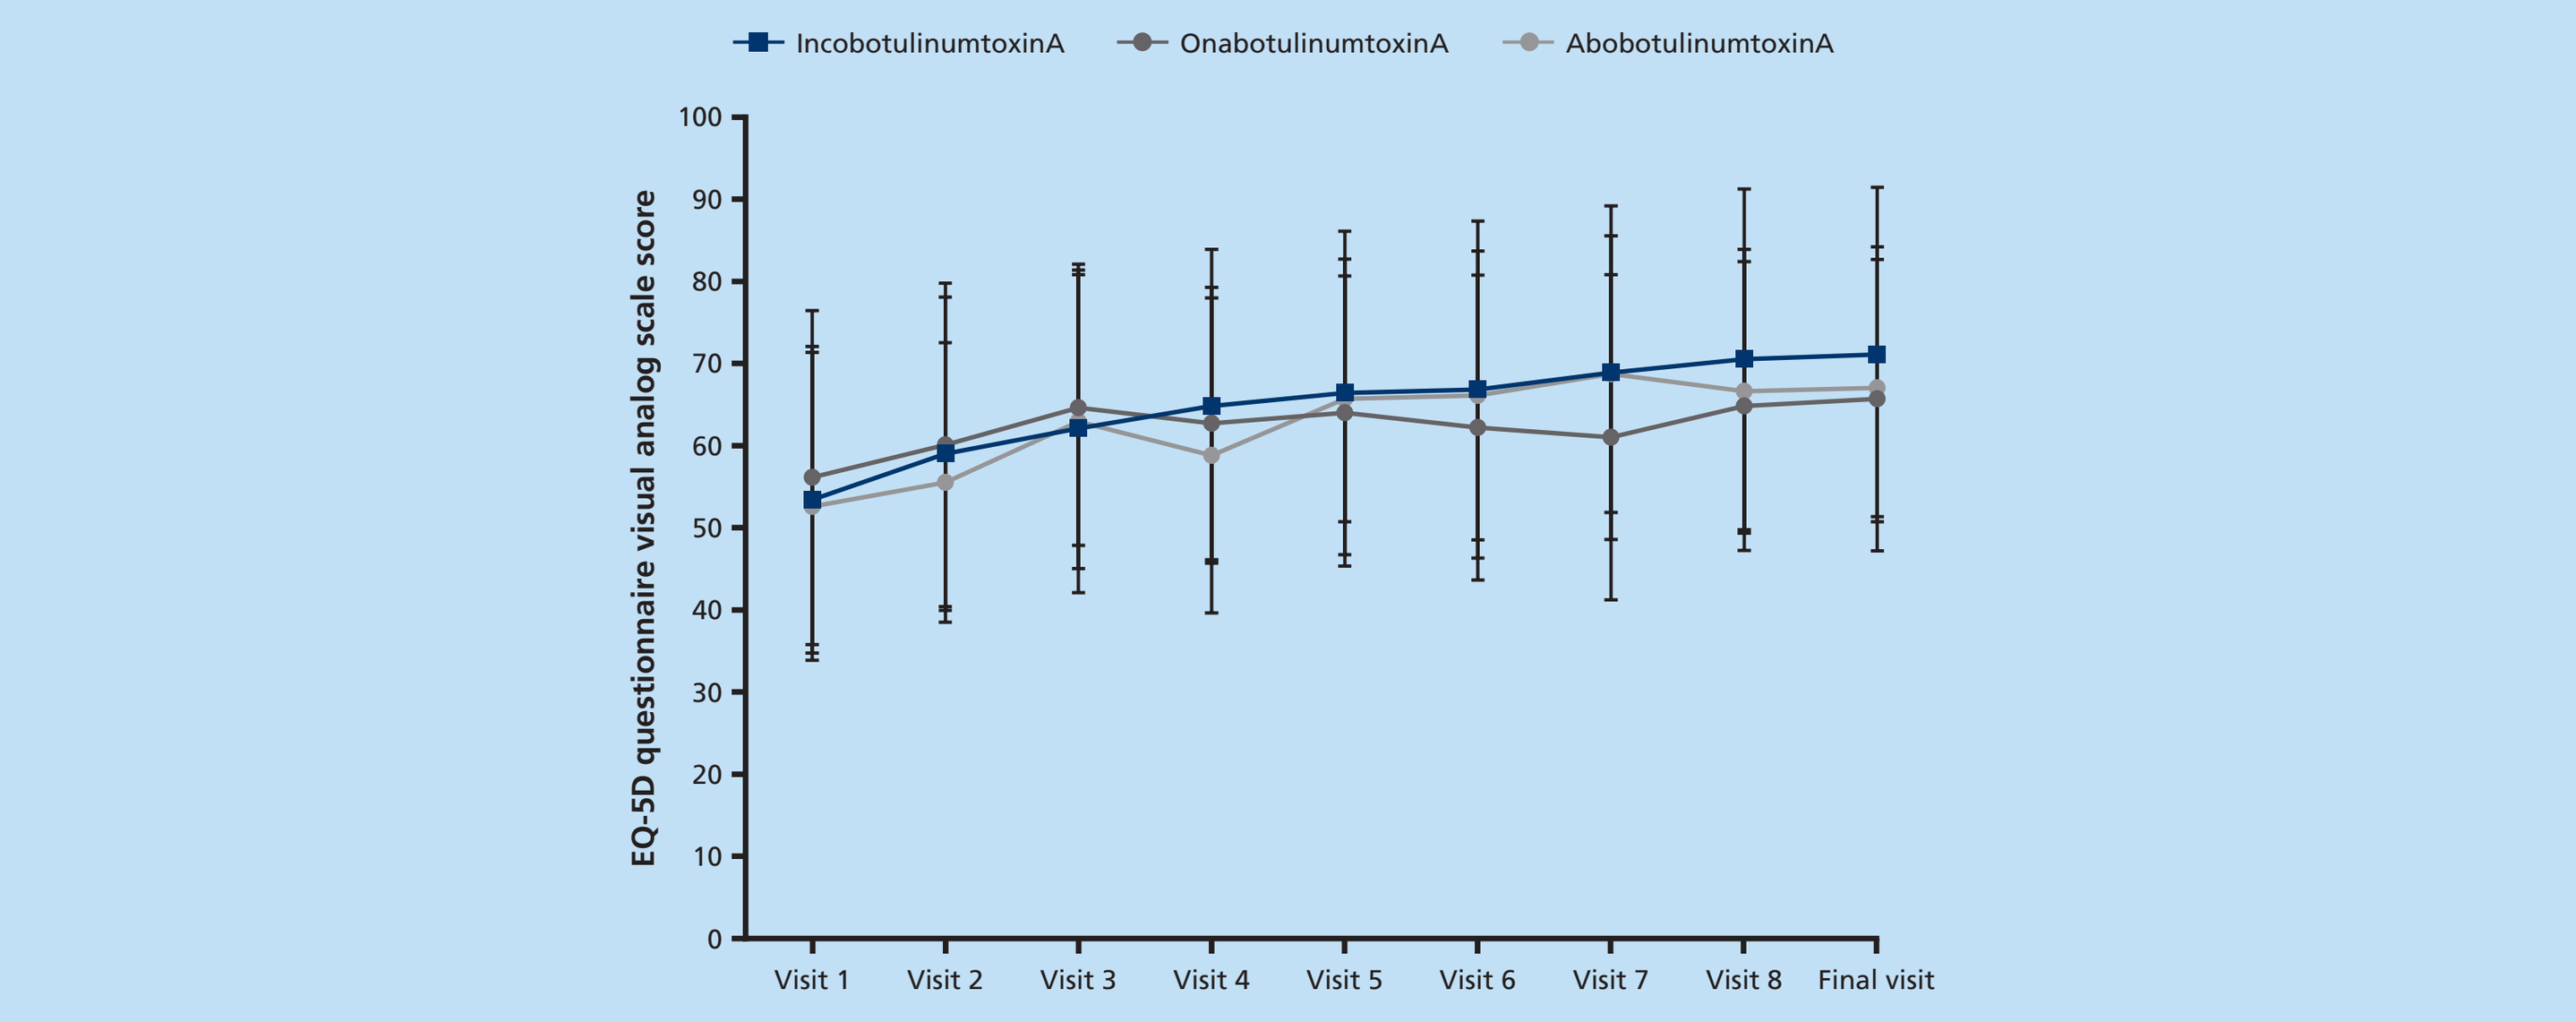
IncobotulinumtoxinA
OnabotulinumtoxinA
AbobotulinumtoxinA
EQ-5D questionnaire visual analog scale score
Visit 1
Visit 2
Visit 3
Visit 4
Visit 5
Visit 6
Visit 7
Visit 8
Final visit

EQ-5D, European Quality of Life-5 Dimensions.

Figure 5. Physicians' assessment of tolerability

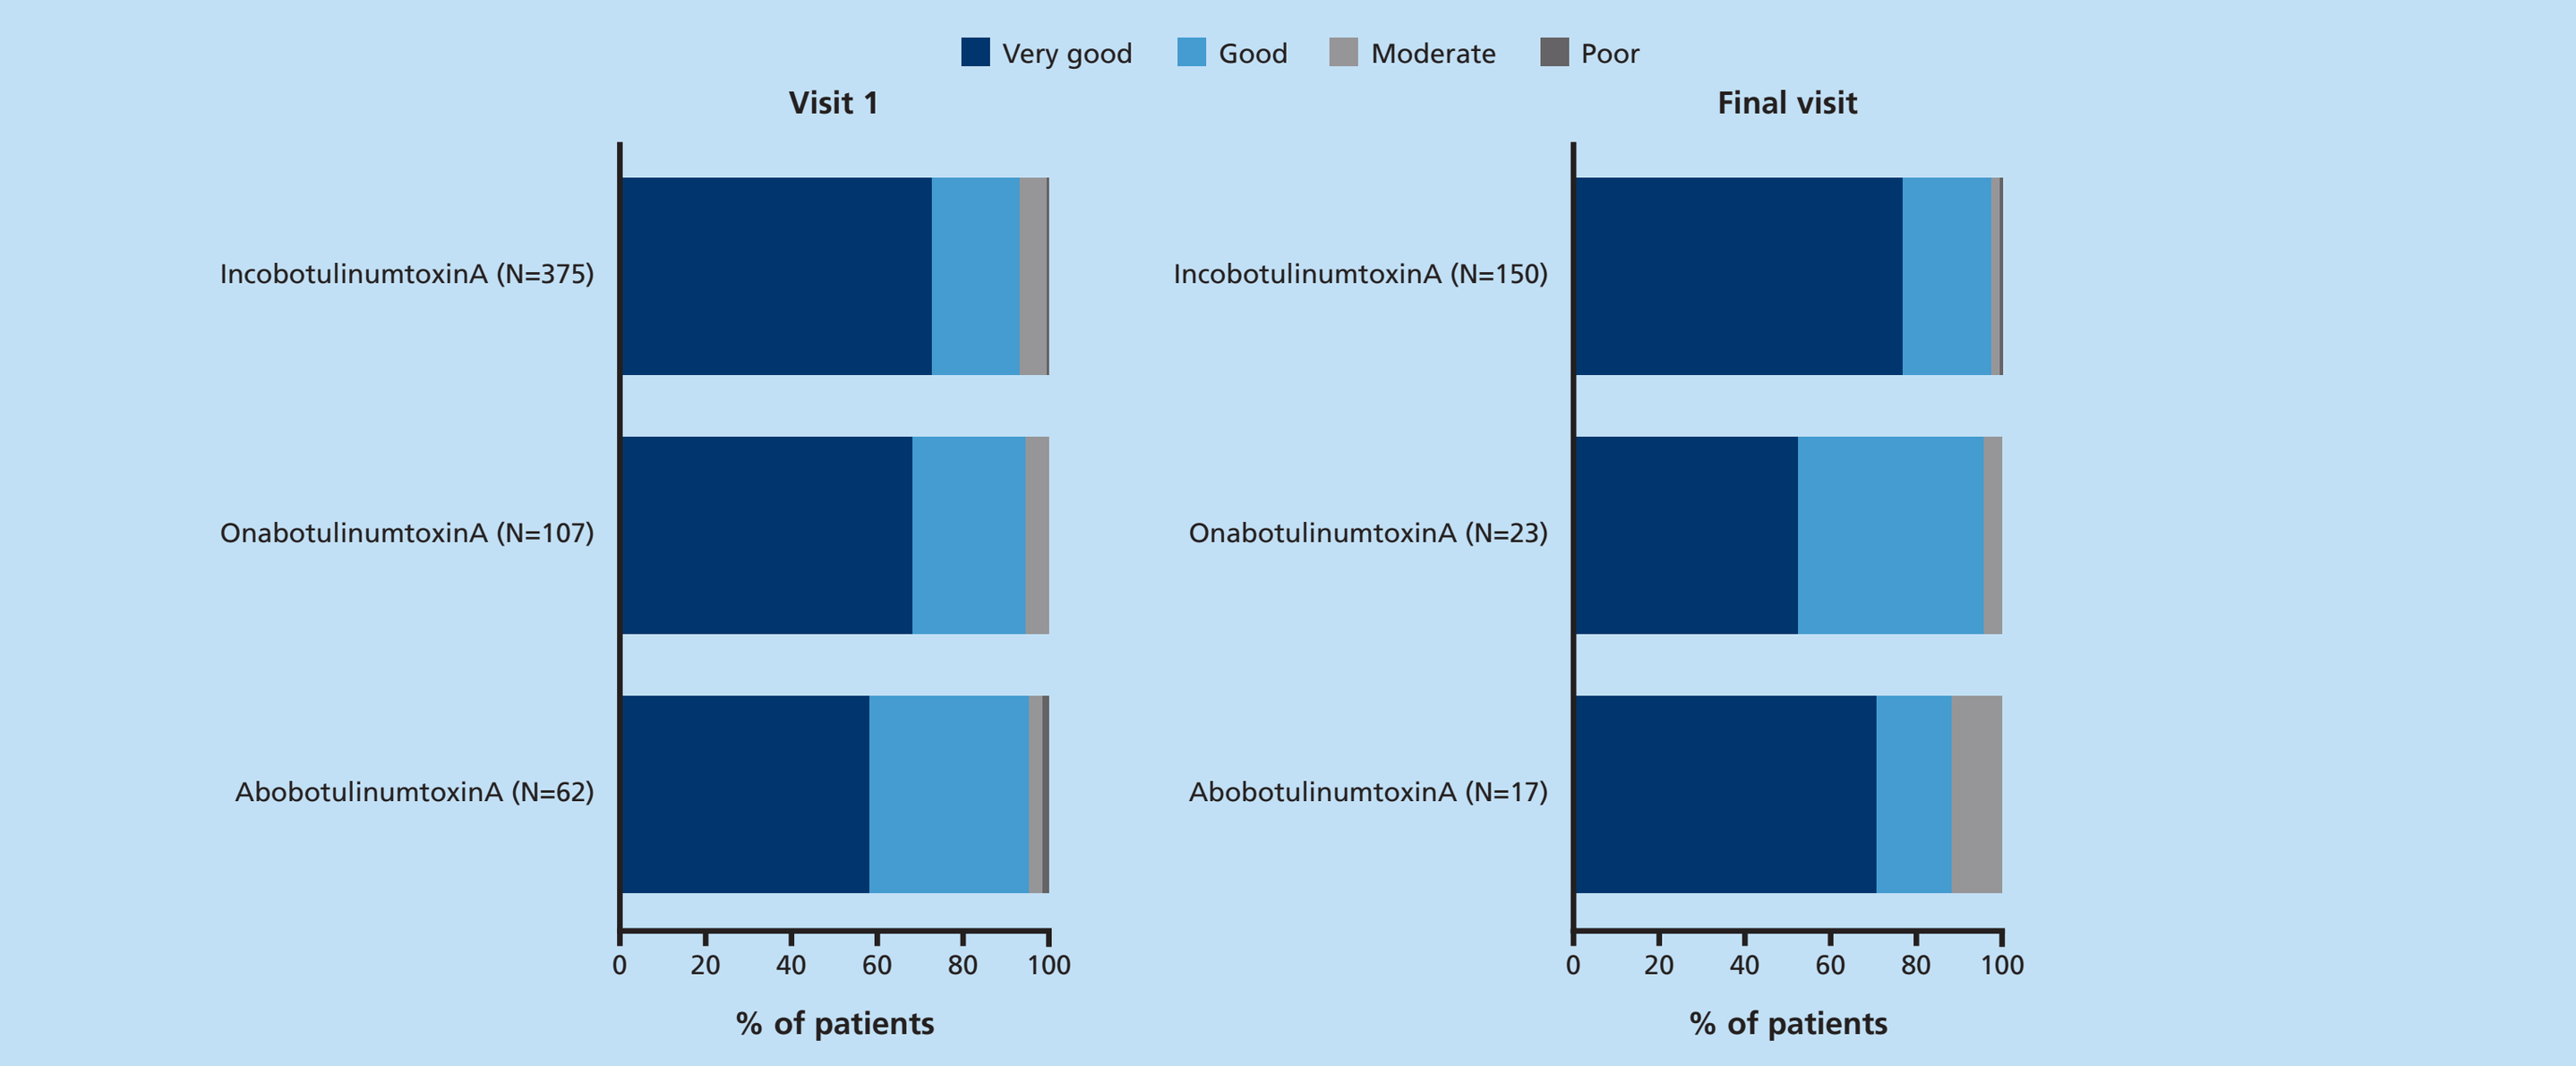
Very good
Good
Moderate
Poor
Visit 1
IncobotulinumtoxinA (N=375)
OnabotulinumtoxinA (N=107)
AbobotulinumtoxinA (N=62)
% of patients
Final visit
IncobotulinumtoxinA (N=150)
OnabotulinumtoxinA (N=23)
AbobotulinumtoxinA (N=17)
% of patients

% are based on non-missing values.

Conclusions

- Results from the SPACE study demonstrated how clinicians use BoNT-A therapy to manage spasticity in real-world clinical practice with regard to patterns treated, muscles injected, goals defined, and use of injection-guidance techniques.
- Patients with spasticity who received BoNT-A treatment in real-world clinical practice experienced an improvement in QoL throughout the 2-year study, and BoNT-A treatment was well tolerated.
- Many physicians expressed a desire to inject higher total doses of BoNT-A to enable them to treat more spasticity patterns in patients with complex multifocal spasticity.
- Further clinical studies are required to explore the safety and efficacy of higher doses of BoNT-A than those used currently.

References

1. Nair KP, Marsden J. BMJ 2014; 349: g4737.
2. Mayer NH. Muscle Nerve Suppl 1997; 6: S1–S13.
3. Wissel J et al. J Rehabil Med 2009; 41: 13–25.
4. Esquenazi A et al. Toxicon 2013; 67: 115–128.
5. Simpson DM et al. Neurology 2016; 86: 1818–1826.

Acknowledgments

This study was sponsored by Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Medical writing support was provided by Rhian Harper-Owen, PhD, on behalf of Complete Medical Communications and funded by Merz Pharmaceuticals GmbH.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017

MRZ82220G_Harriss.indd 1
11/01/2017 12:17