Sustained incobotulinumtoxinA (Xeomin®) efficacy in upper-limb post-stroke spasticity over 48 weeks in a Phase 3, placebo-controlled study with an open-label extension

Petr Kaňovský,[1] **Allison Brashear,**[2] Elie P. Elovic,[3] Michael C. Munin,[4] Angelika Hanschmann,[5] Reinhard Hiersemenzel,[5] Christina Marciniak[6]

[1]Palacký University Olomouc, Faculty of Medicine and Dentistry, and University Hospital, Olomouc, Czech Republic; [2]Department of Neurology, Wake Forest School of Medicine, Wake Forest Baptist Medical Center, Winston Salem, NC, USA; [3]Renown Rehabilitation Hospital, Reno, NV, USA; [4]Department of Physical Medicine and Rehabilitation, University of Pittsburgh School of Medicine, Pittsburgh, PA, USA; [5]Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany; [6]Department of Physical Medicine and Rehabilitation, Northwestern University Feinberg School of Medicine, and Rehabilitation Institute of Chicago, Chicago, IL, USA

Background

- Botulinum toxin injections are a recommended treatment for post-stroke spasticity of the upper limb.[1–3]
- In a previous randomized, placebo-controlled clinical trial with an open-label extension period (OLEX) conducted in Europe, incobotulinumtoxinA treatment led to significant improvements of spasticity-associated disability and muscle tone, and was well tolerated.[4,5]
- In the trial presented here, we investigated the long-term efficacy and safety of repeated incobotulinumtoxinA injections in a 36-week OLEX with up to three incobotulinumtoxinA injection cycles.
 - Results from the double-blind, placebo-controlled main period (MP) of this trial demonstrated the safety and efficacy of incobotulinumtoxinA vs placebo for a single 12-week injection cycle.[6]

Methods

Study design

- Randomized, double-blind, placebo-controlled, multicenter study (NCT01392300; EudraCT 2010-023043-15).
- Parallel-group MP (12-week duration) with a single treatment of incobotulinumtoxinA 400 U or placebo.
- Patients could continue to a 36-week OLEX.
 - Three additional treatments (400 U fixed dose) at 12-week intervals.
- All muscle groups (elbow flexors, wrist flexors, finger flexors, thumb flexors, and/or forearm pronators) with an Ashworth Scale (AS) score ⩾2 and the corresponding clinical pattern were required to be treated.
- Injection sites and doses per muscle were chosen by the investigator (within pre-defined dose ranges).

Figure 1. Study design

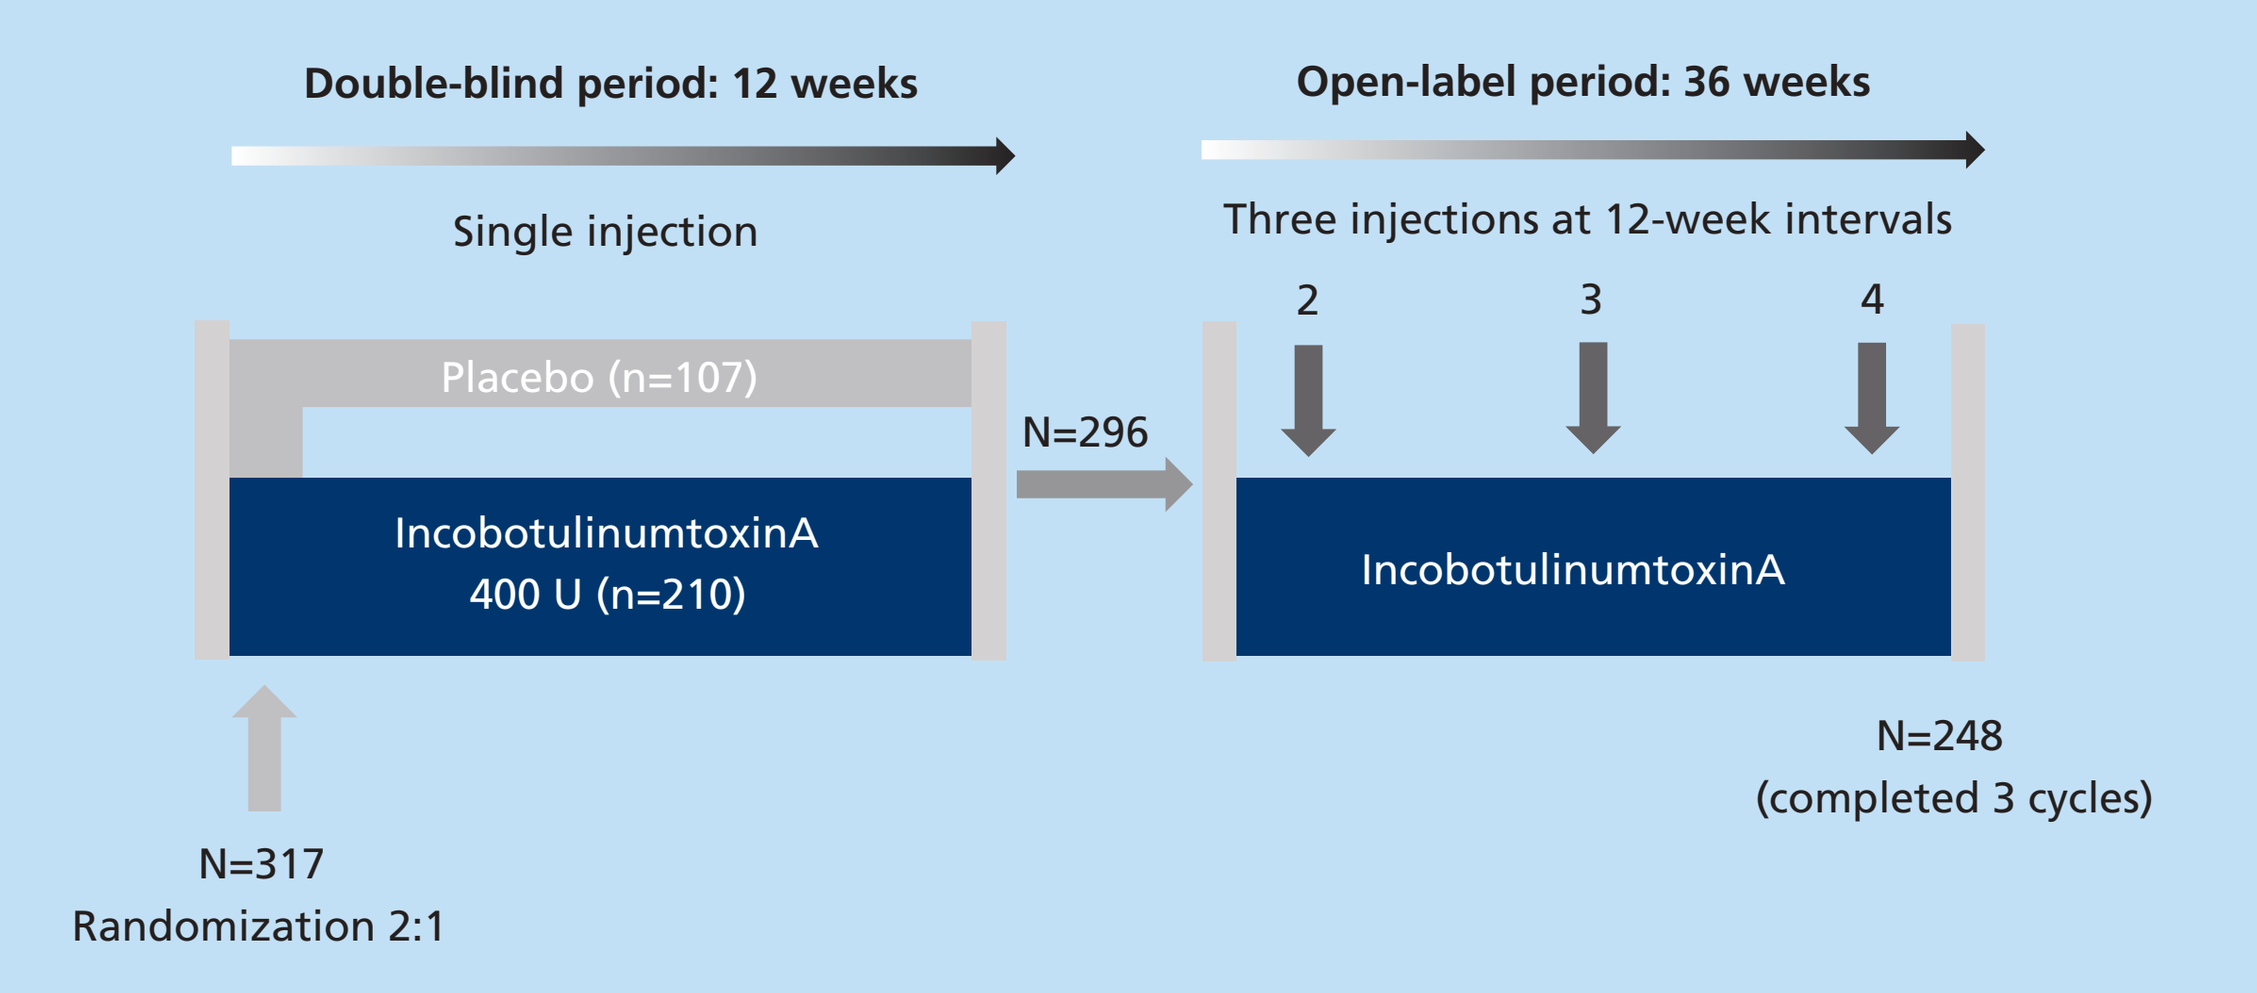
Double-blind period: 12 weeks
Single injection
Placebo (n=107)
IncobotulinumtoxinA
400 U (n=210)
N=296
Open-label period: 36 weeks
Three injections at 12-week intervals
2
3
4
IncobotulinumtoxinA
N=248
(completed 3 cycles)
N=317
Randomization 2:1

Inclusion criteria

- The inclusion and exclusion criteria for the MP have been reported;[6] for the OLEX, the main inclusion criteria were completion of the randomized, placebo-controlled MP and a clinical need for treatment with 400 U of incobotulinumtoxinA.

Outcome measures

- Muscle tone
 - Muscle tone was assessed using the AS at Week 4.
 - Patients with an improvement of ⩾1 point between each injection and the corresponding post-injection visit 4 weeks later were classed as responders.
- Global Impression of Change
 - Investigator's Global Impression of Change due to treatment at Week 4 using a scale ranging from –3 (very much worse) to +3 (very much improved).
 - Patients and carers also rated Global Impression of Change at the post-injection visit 4 weeks after each treatment using the same scale.
- Disability
 - The Disability Assessment Scale (DAS) assesses four functional domains (hygiene, dressing, limb position, and pain) on a scale from 0 (no disability) to 3 (severe disability). A principal therapeutic target was chosen by the subject before each treatment.
- Carer burden
 - The Carer Burden Scale assesses five care activities (see **Figure 4**) on a scale from 0 (no difficulty) to 4 (cannot do the task).
- Safety
 - Standard safety assessments were performed throughout with a specific questionnaire completed during each visit or telephone contact to detect adverse events (AEs). Assessments also included continuous testing for neutralizing antibodies.

Results

Patients

- In total, 299 patients completed the MP and entered the OLEX.
- Baseline characteristics of the OLEX population are shown in **Table 1**.
- During the OLEX period, 296 patients received one incobotulinumtoxinA treatment, 276 patients received two treatments, and 257 patients received three treatments.
 - 99 of the patients who enrolled in the OLEX period had received placebo only during the MP.
 - Three patients did not require treatment with 400 U incobotulinumtoxinA 12 weeks after the MP and discontinued participation in the study.
- A total of 248 patients (82.9%) completed the OLEX.

Efficacy outcomes

Muscle tone

Figure 2. AS responder rates 4 weeks after each injection

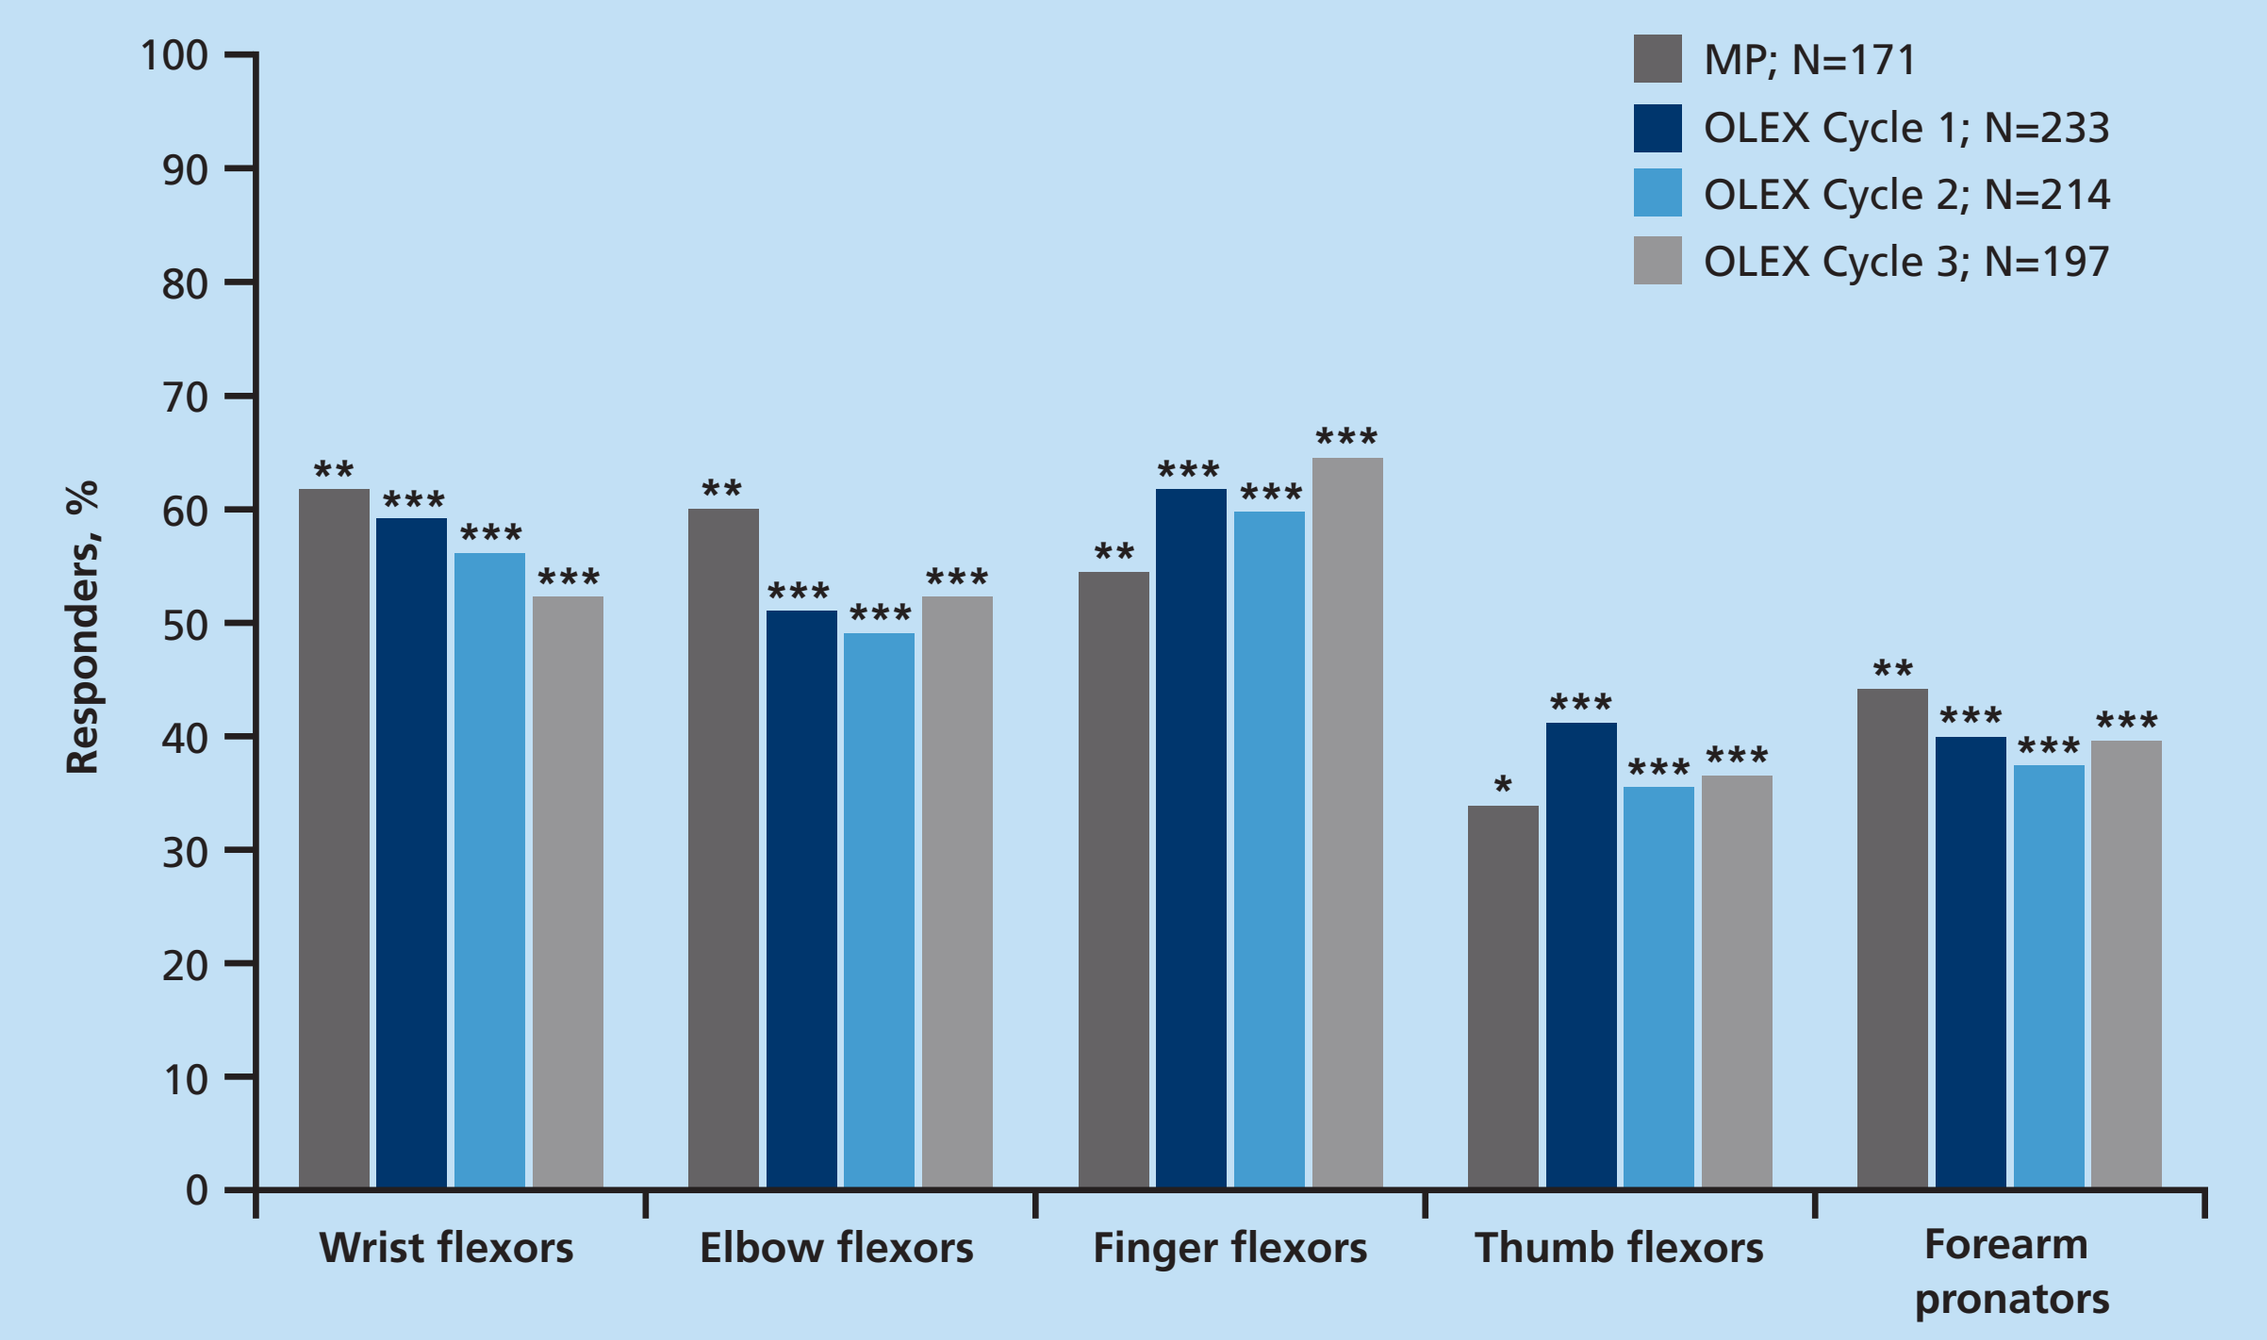
MP; N=171
OLEX Cycle 1; N=233
OLEX Cycle 2; N=214
OLEX Cycle 3; N=197
Responders, %
100
90
80
70
60
50
40
30
20
10
0
Wrist flexors
Elbow flexors
Finger flexors
Thumb flexors
Forearm pronators

MP: *P=0.028 vs placebo; **P<0.001 vs placebo by logistic regression; OLEX: ***P<0.0001 Wilcoxon signed-rank test.
Patients with a ⩾1-point improvement (reduction) in AS scores from each injection to the respective 4-week control visit were classified as responders.
AS, Ashworth Scale; MP, main period; OLEX, open-label extension period.

Disability

Figure 3. DAS responder rates for the principal target domain 4 weeks after each injection increased throughout the study compared with MP baseline

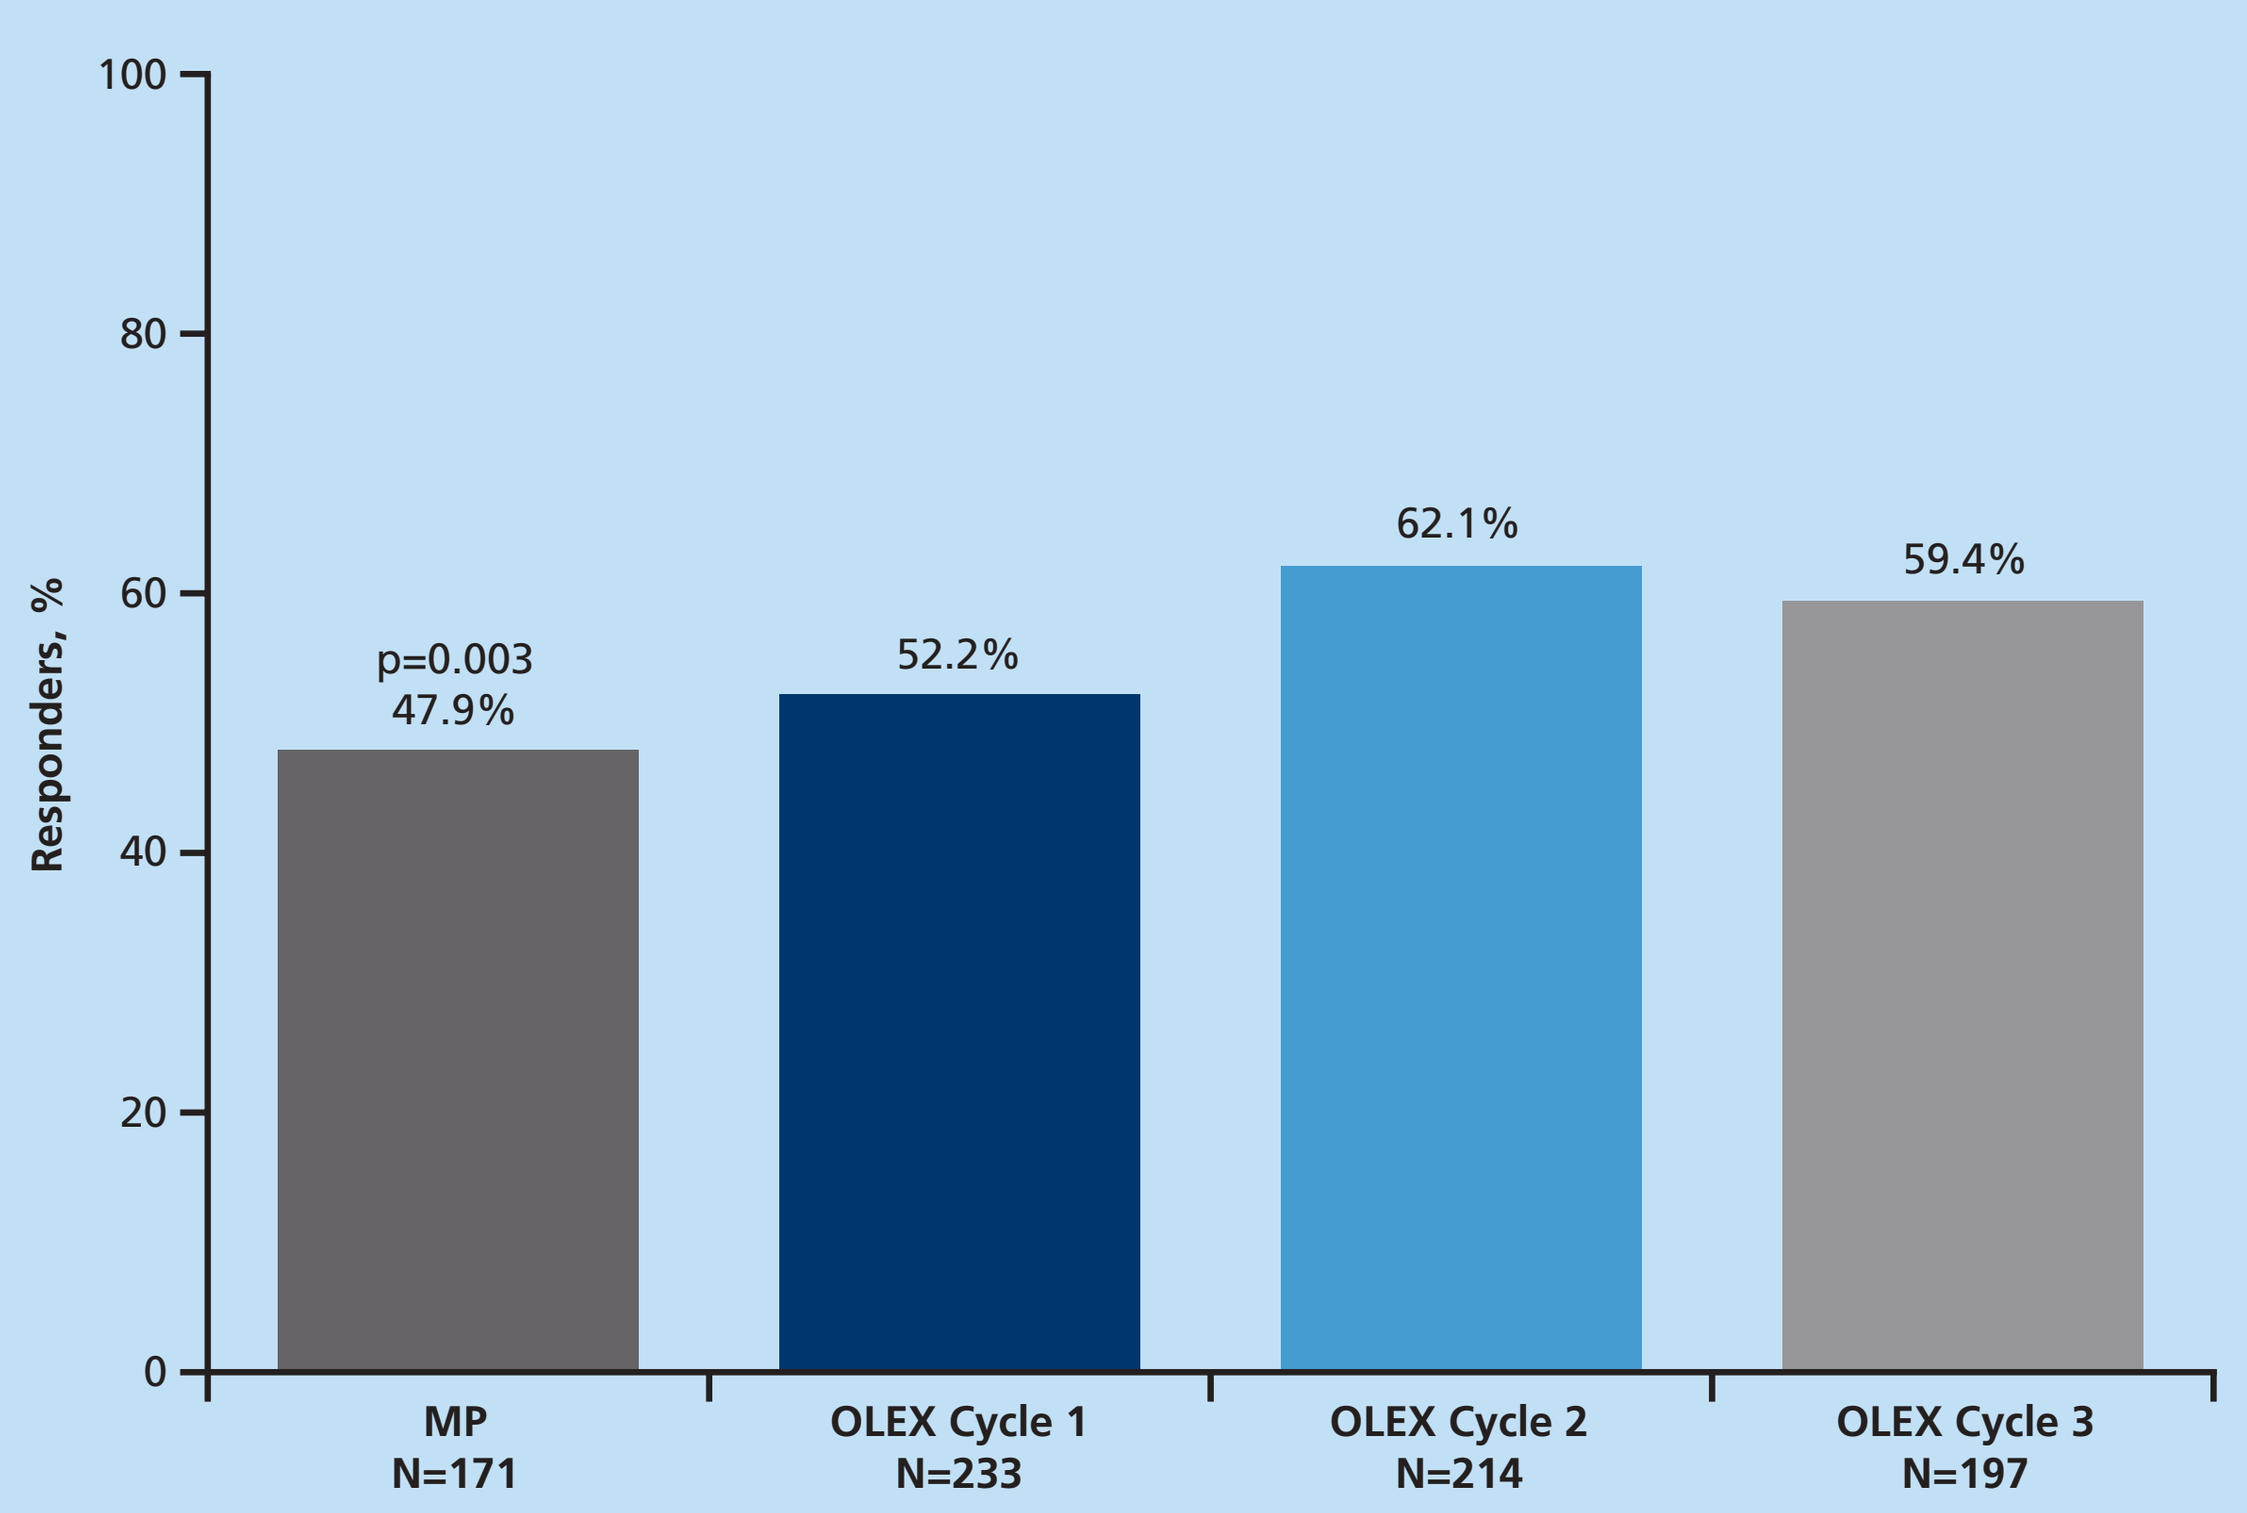
Responders, %
100
80
60
40
20
0
p=0.003
47.9%
52.2%
62.1%
59.4%
MP
N=171
OLEX Cycle 1
N=233
OLEX Cycle 2
N=214
OLEX Cycle 3
N=197

MP: P=0.003 vs placebo by Wilcoxon rank-sum test.
Patients with a >1 point improvement (reduction) in DAS scores for the principal target domain from the mainperiod baseline to the 4-week post-injection visit after each injection were classified as responders.

Carer burden

Figure 4. Significant and sustained improvements in Carer Burden Scale scores were observed from the MP baseline to the end of the study

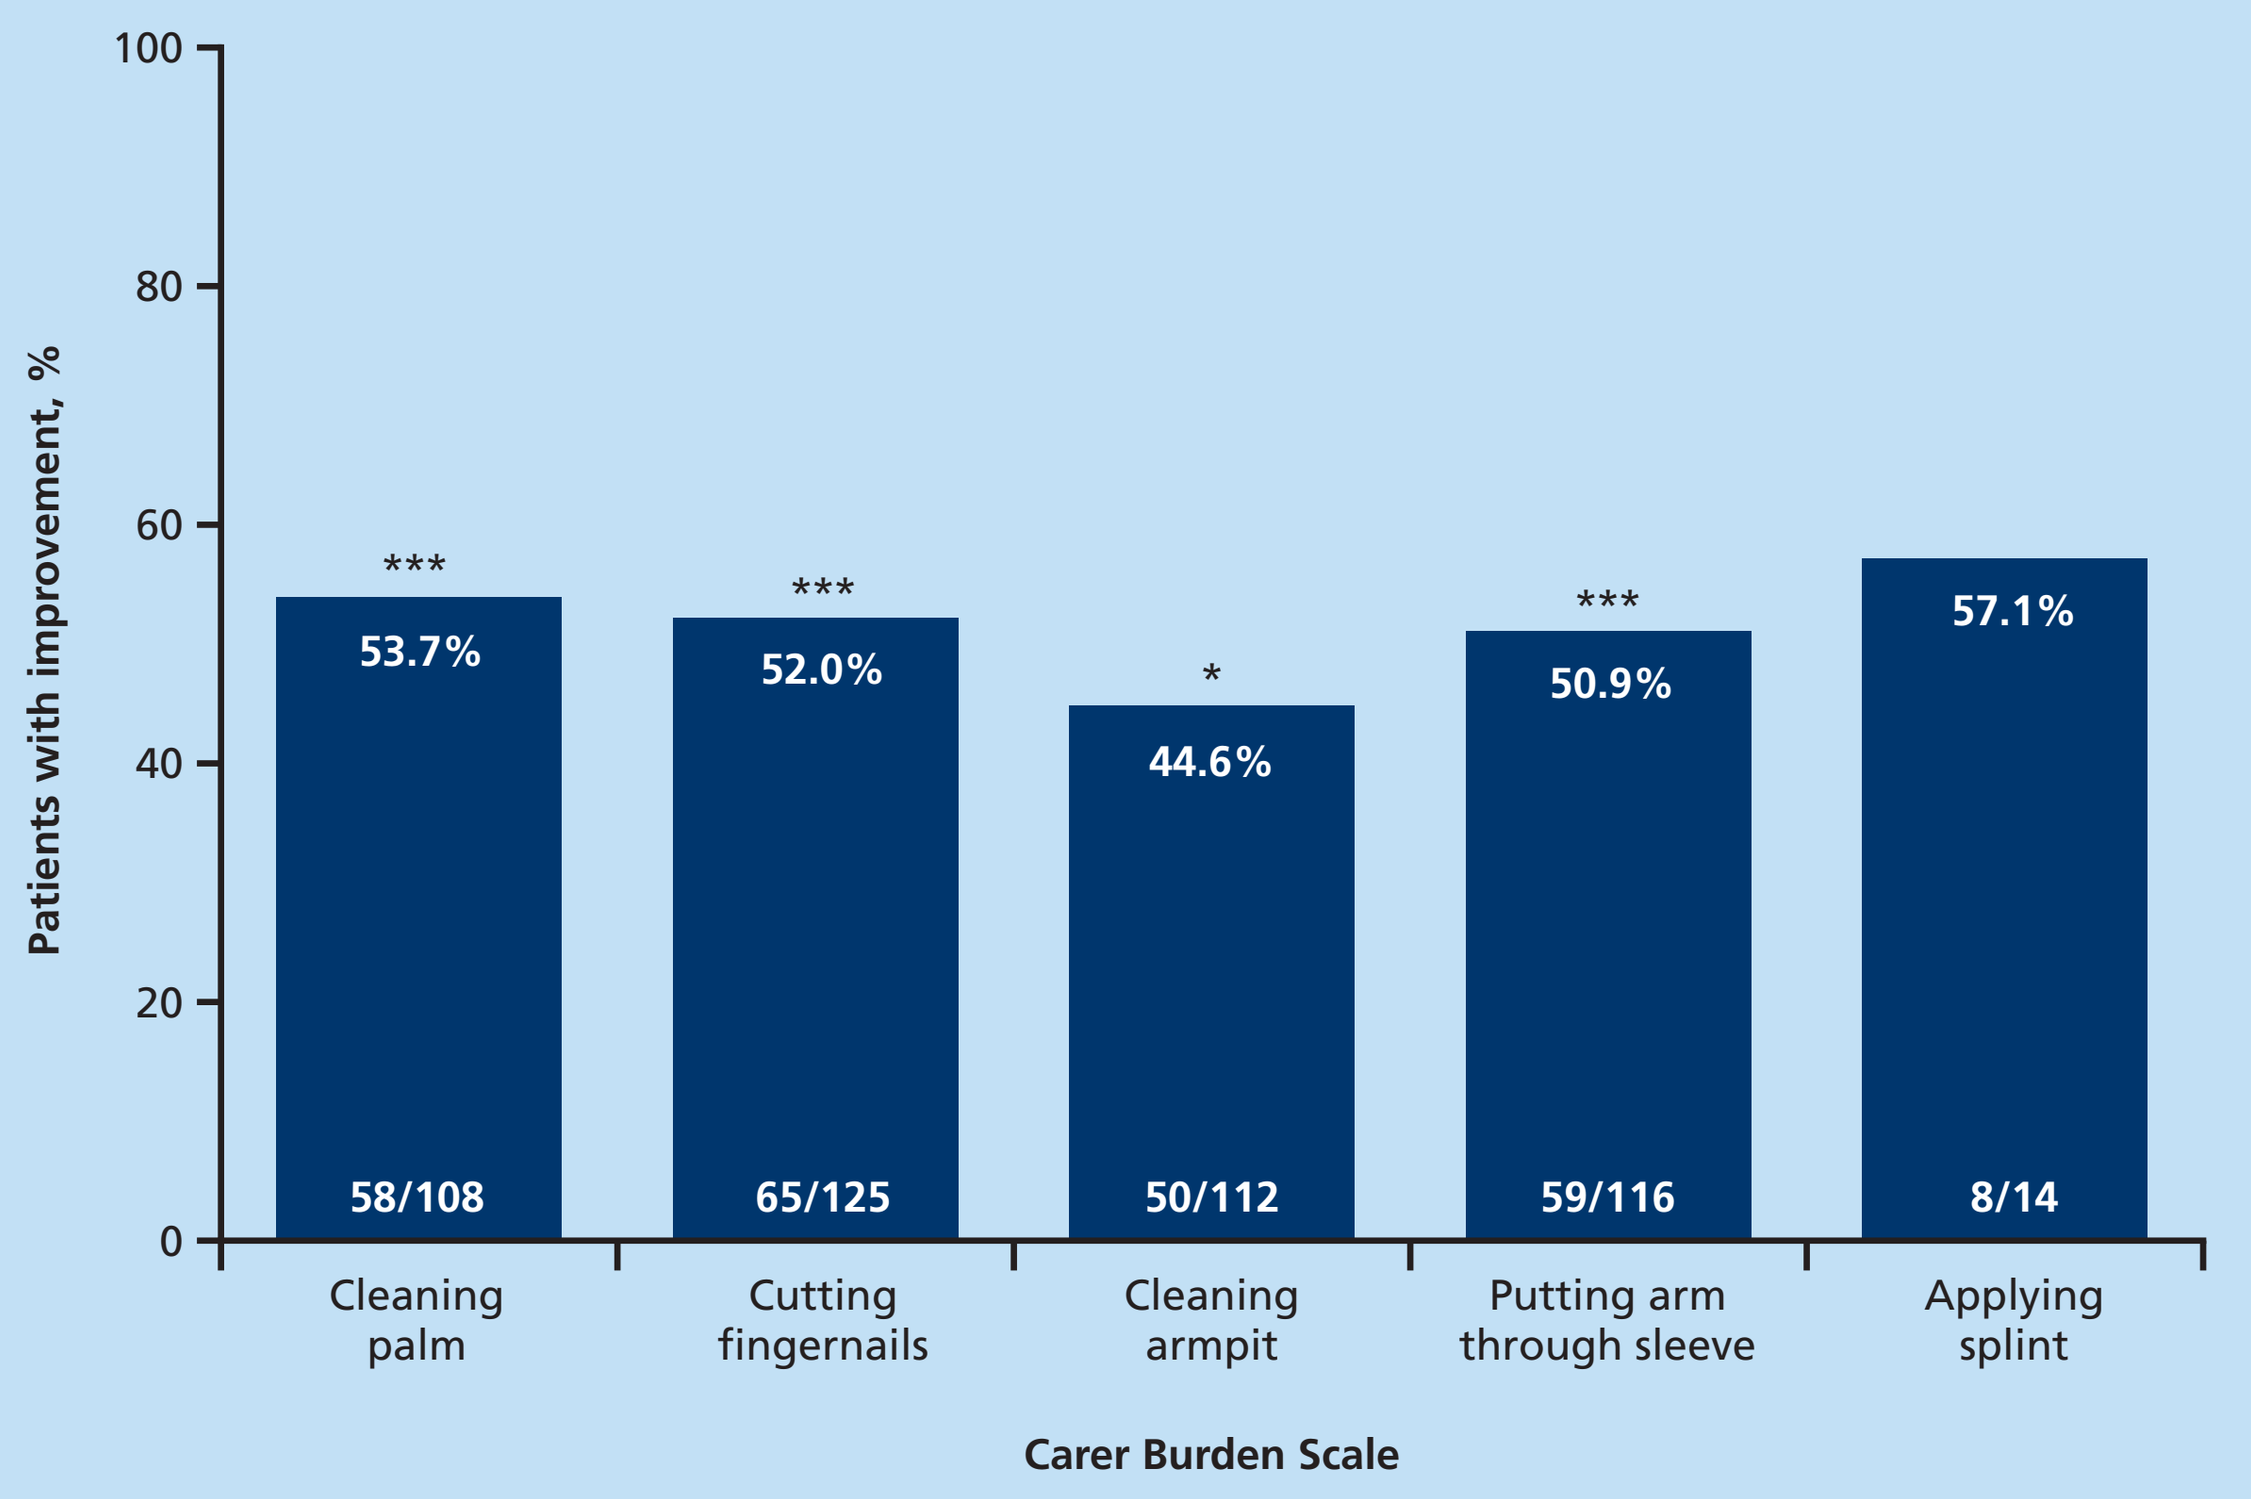
Patients with improvement, %
100
80
60
40
20
0
53.7%
58/108
52.0%
65/125
44.6%
50/112
50.9%
59/116
57.1%
8/14
Cleaning palm
Cutting fingernails
Cleaning armpit
Putting arm through sleeve
Applying splint
Carer Burden Scale

*P<0.01 and ***P<0.0001 for improvement from MP baseline to end of study visit (Wilcoxon signed-rank test). Improvement in 'Applying splint' did not reach statistical significance; however, the number of patients for whom this was applicable was low (N=14).
MP, main period.

Global Impression of Change

Figure 5. Improvement during OLEX on the Global Impression of Change Scale 4 weeks after each injection

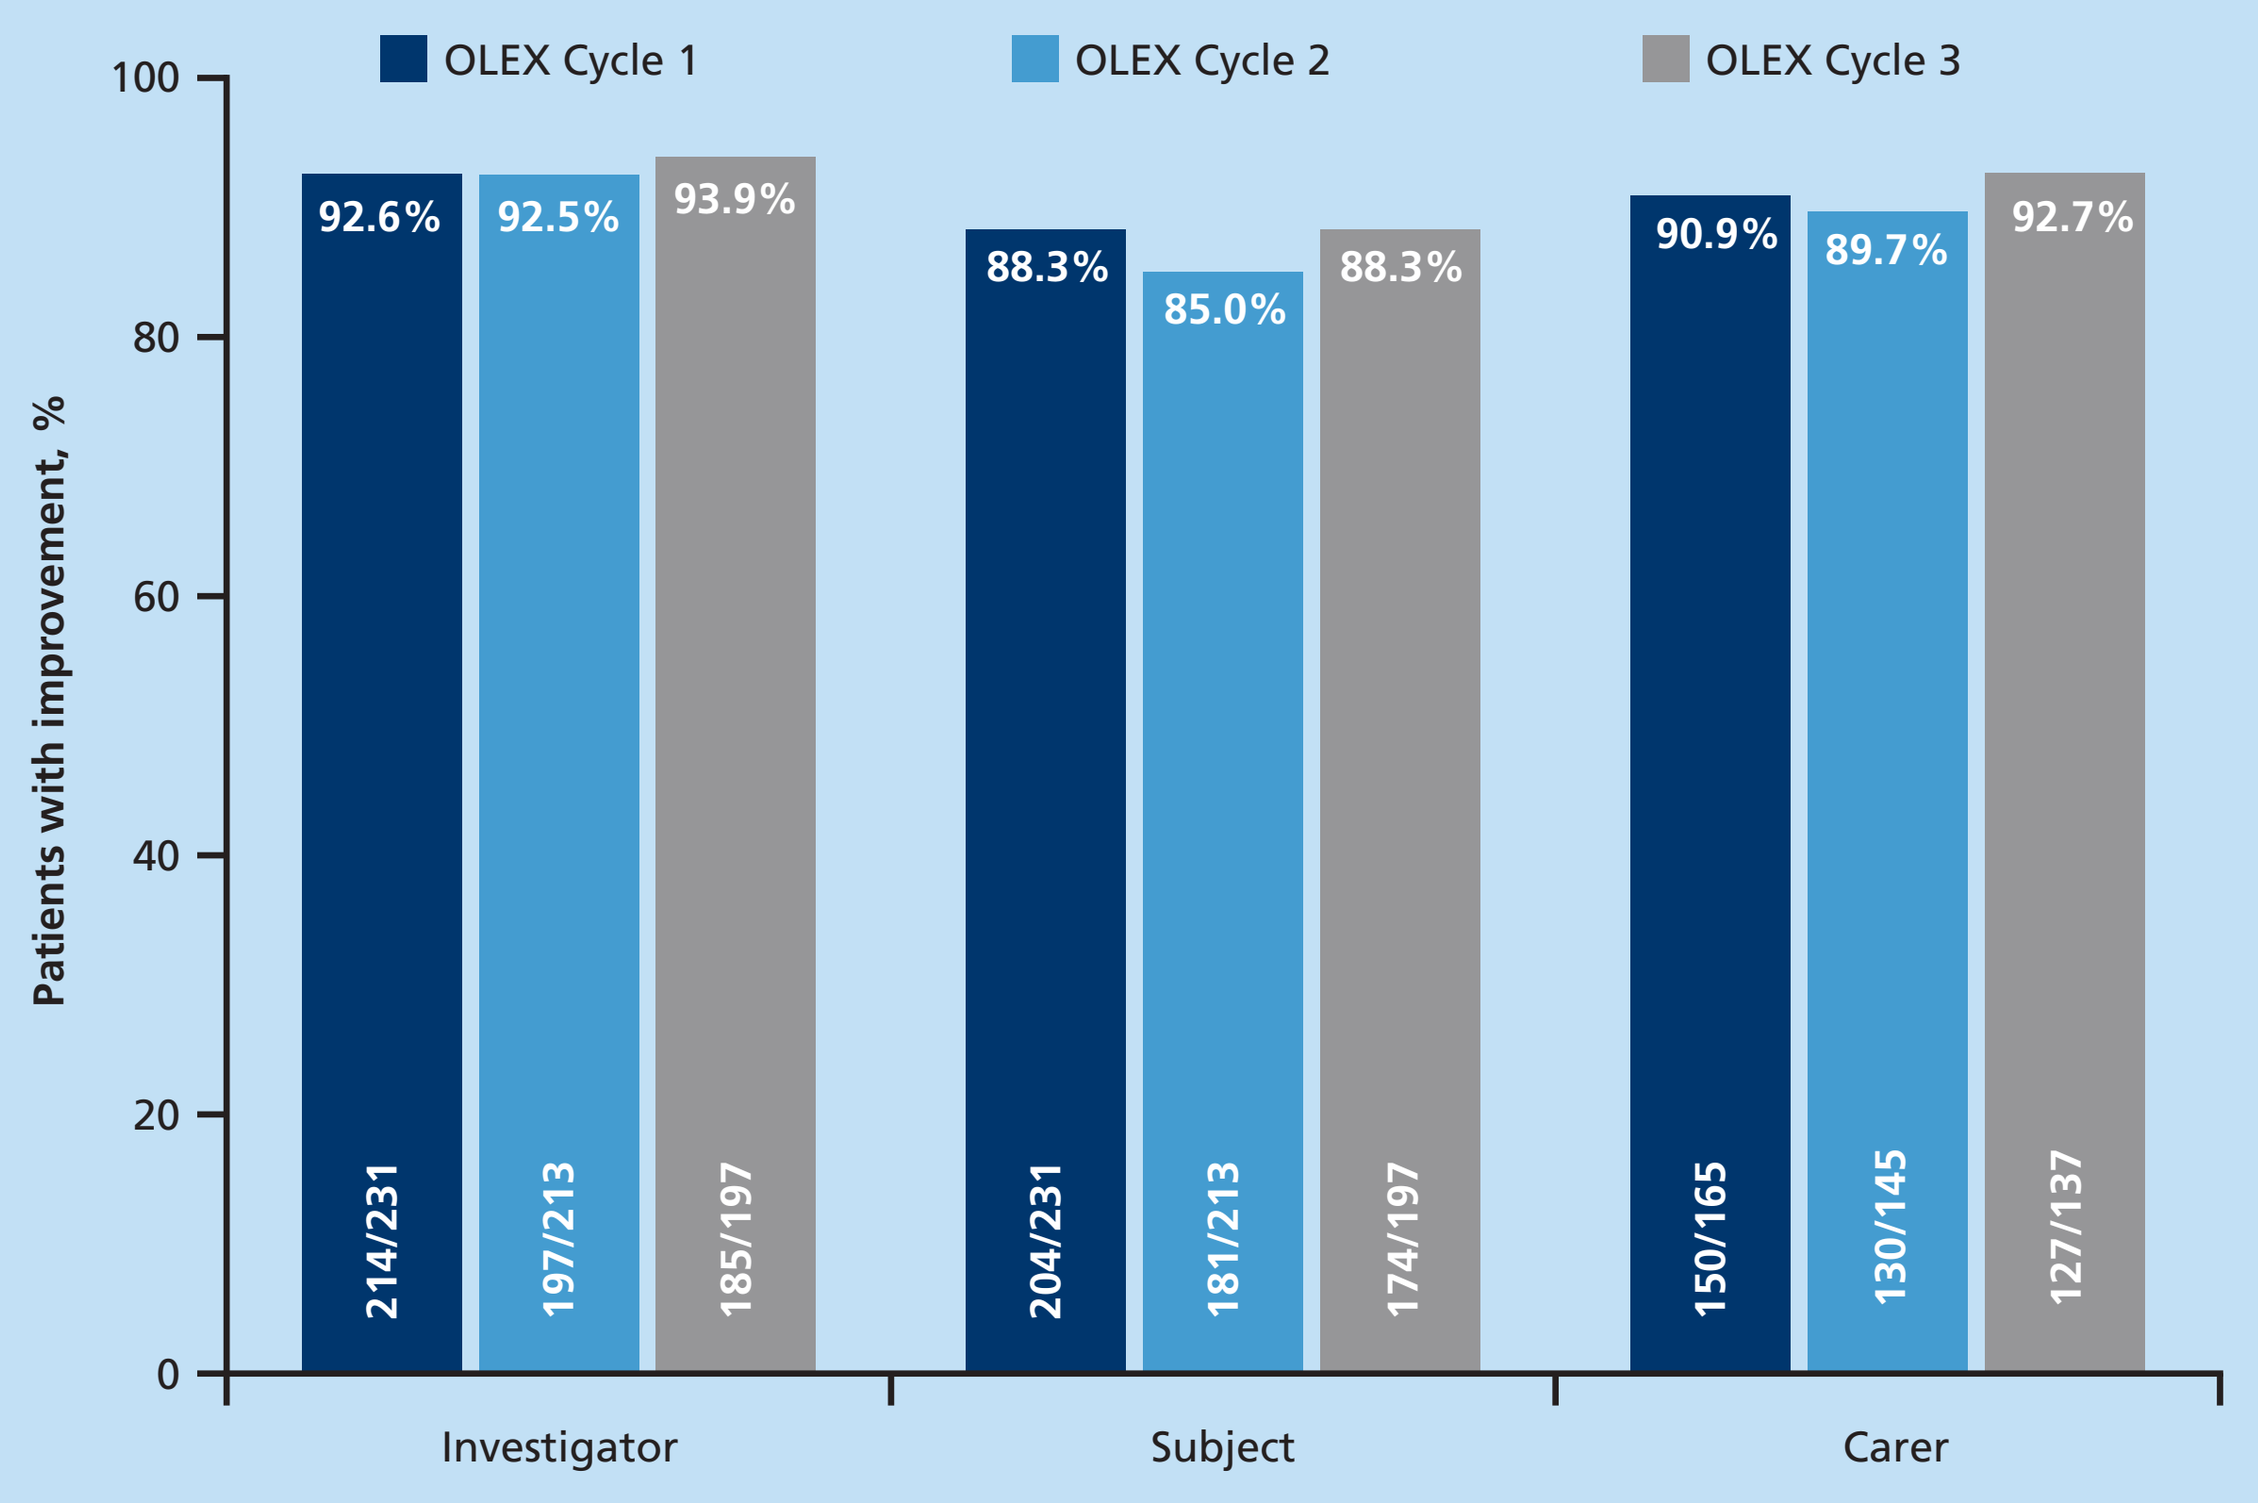
OLEX Cycle 1
OLEX Cycle 2
OLEX Cycle 3
Patients with improvement, %
100
80
60
40
20
0
92.6%
214/231
92.5%
197/213
93.9%
185/197
88.3%
204/231
85.0%
181/213
88.3%
174/197
90.9%
150/165
89.7%
130/145
92.7%
127/137
Investigator
Subject
Carer

The investigator, patient, and carer were asked to evaluate the global change in spasticity of the upper limb at the 4-week post-injection visits. The proportion of patients with a score ⩾1 on the Global Impression of Change Scale is shown (ie minimally improved, much improved, or very much improved).
OLEX, open-label extension period.

Safety

- In the MP, treatment-related AEs were reported for 3.8% and 1.9% of patients for incobotulinumtoxinA and placebo, respectively.
- Throughout the three incobotulinumtoxinA OLEX treatment cycles, treatment-related AEs were reported by 9/296 patients (3.0%).
 - All treatment-related AEs were resolved by the end of the study except one case of increased γ-glutamyl transferase in a patient who also had high levels of γ-glutamyl transferase at study baseline.
- No serious AEs were considered related to treatment.
- There were no cases of clinical non-responsiveness due to antibodies.

Table 1. Patient characteristics at OLEX baseline (SES)

	All patients N=296
Age, years; mean (SD)	56.3 (11.5)
Male gender, n (%)	169 (57.1)
Time since first diagnosis of upper-limb spasticity, months; mean (SD)	38.2 (55.2)
Clinical pattern of upper-limb spasticity, n (%)	
Flexed wrist	296 (100.0)
Flexed elbow	296 (100.0)
Clenched fist	296 (100.0)
Thumb-in-palm	243 (82.1)
Pronated forearm	272 (91.9)

OLEX, open-label extension period; SD, standard deviation; SES, safety evaluation set.

Table 2. Summary of AEs during the OLEX (SES)

AEs	All patients N=296
Any AE	93 (31.4)
Any AE related to treatment	9 (3.0)
Any serious AE related to treatment	0
Any AE leading to drop-out related to treatment	0
Treatment-related AEs	
Constipation	2 (0.7)
Pain in extremity	2 (0.7)
Dermatitis, allergic	1 (0.3)
γ-glutamyl transferase increased	1 (0.3)
Injection site bruising	1 (0.3)
Muscular weakness	1 (0.3)
Pelvic floor muscle weakness	1 (0.3)
Presyncope	1 (0.3)
Uveitis	1 (0.3)

AE, adverse event; OLEX, open-label extension period; SES, safety evaluation set.

Conclusions

- During this 36-week OLEX following a double-blind, placebo-controlled study, repeated injections of incobotulinumtoxinA 400 U significantly reduced muscle tone in patients with post-stroke spasticity of the upper limb.
- Reduced muscle tone translated into sustained significant and clinically meaningful improvements in Disability Assessment Scale and Carer Burden Scale scores.
- Repeated treatment with incobotulinumtoxinA 400 U at 12-week intervals was well tolerated, without any treatment-related serious AEs.

References

1. Esquenazi A et al. Toxicon 2013; 67: 115–128.
2. Wissel J et al. J Rehabil Med 2009; 41: 13–25.
3. Simpson DM et al. Neurology 2016; 86: 1818–1826.
4. Kaňovský P et al. Clin Neuropharmacol 2009; 32: 259–265.
5. Kaňovský P et al. J Rehabil Med 2011; 43: 486–492.
6. Elovic EP et al. Muscle Nerve 2016; 53: 415-421.

Acknowledgments

This study was sponsored by Merz Pharmaceuticals GmbH, Frankfurt am Main, Germany. Editorial support with the layout of this poster was provided by Complete Medical Communications.

These data were previously presented at the American Academy of Neurology Annual Meeting 2016, the European Congress of Physical Medicine and Rehabilitation 2016, the International Congress of Parkinson's Disease and Movement Disorders 2016, and the American Congress of Rehabilitation Medicine 2016, and are in part published in Elovic EP et al. Muscle Nerve 2016; 53: 415–421.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017

MRZ82220f_TOXINS_Brashear.indd 1
11/01/2017 15:22