Botulinum toxin treatment for spasticity: clinical experience with changing from abobotulinumtoxinA (Dysport®) to incobotulinumtoxinA (Xeomin®)

Martin Cossar,[1] Alastair Cozens[1]

[1]NHS Grampian, Specialist Rehabilitation Service, Woodend Hospital, Aberdeen, UK

NHS Grampian

Introduction

- Botulinum toxin injections have become a common and recommended treatment for many patients with focal spasticity.[1–4]
- In Europe and the US, three botulinum toxin type A products are currently available:
 - onabotulinumtoxinA (Botox®, Allergan)[5]
 - abobotulinumtoxinA (Dysport®, Ipsen)[6]
 - incobotulinumtoxinA (Xeomin®, Merz Pharmaceuticals).[7]
- The potency of botulinum toxins is measured in units that are proprietary to each commercial formulation. Hence, botulinum toxin preparations have different dose schedules that are not interchangeable with each other.[2]
 - As a result, clinicians may prefer not to switch between botulinum toxin preparations due to concerns about efficacy and safety, but commonly stay with the one they are mostly familiar with.
- Our Posture and Movement Clinic provides botulinum toxin therapy for outpatients with spasticity of various etiologies. Until 2012, we exclusively used abobotulinumtoxinA.
 - Based on cost-effective alternatives, our hospital pharmacy suggested switching to the incobotulinumtoxinA formulation.
- After initially treating a small number of patients with incobotulinumtoxinA over several months with positive results, we changed formulation from abobotulinumtoxinA to incobotulinumtoxinA in 2013. Here, we review treatment outcomes with both formulations.

Objective

- To review outcomes for patients with spasticity who switched from abobotulinumtoxinA treatment to incobotulinumtoxinA in a spasticity outpatient clinic.

Methods

- Patients with spasticity of the upper and/or lower limbs who received botulinum toxin treatment with abobotulinumtoxinA were switched to incobotulinumtoxinA.
- Switching was generally initiated at a 4:1 unit ratio of abobotulinumtoxinA to incobotulinumtoxinA. However, dosing was adjusted based on previous treatment outcomes and individual clinical need at the time of re-treatment.
- 500 U vials of abobotulinumtoxinA were reconstituted in 2.5 mL of sterile saline and 100 U vials of incobotulinumtoxinA in 2.0 mL of sterile saline.
- Electrostimulation guidance was used routinely.
- Patient records captured details of treatment intervals, muscles injected, doses administered (total and per muscle), treatment goal attainment, and occurrence of adverse drug reactions.
 - Treatment goal attainment was recorded as *yes*, *partial,* or *no*.

Results

Patient characteristics

- From 1 September 2012 to 28 February 2013, we treated 36 consecutive outpatients with spasticity with abobotulinumtoxinA. Patients received a mean (standard deviation; SD) dose of 843 (379) U (range 150–1500 U).
- Patient characteristics are summarized in **Table 1**.

Table 1. Patients treated with abobotulinumtoxinA from 1 September 2012 to 28 February 2013

Characteristic	N=36
Age, years; mean (SD) [range]	61.3 (17.0) [26–91]
Male gender, n (%)	12 (33.3)
Cause of spasticity, n (%)	
Stroke	22 (61.1)
Multiple sclerosis	5 (13.9)
Spinal–cord injury	2 (5.6)
Other	7 (19.4)
AbobotulinumtoxinA dose, U; mean (SD) [range]	843 (379) [150–1500]

SD, standard deviation.

Switch to incobotulinumtoxinA treatment

- Of the 36 patients who had received abobotulinumtoxinA, 23 required further botulinum toxin injections and were switched to incobotulinumtoxinA therapy.
- The characteristics of the patients who were switched are summarized in **Table 2**.
- On average, patients required their first incobotulinumtoxinA treatment a mean (SD) 21.6 (7.2) weeks (range 8–38 weeks) after their last abobotulinumtoxinA injections.
- The mean (SD) incobotulinumtoxinA dose at the first treatment after switching was 196 (106) U (range 35–400 U).
- Re-treatment with incobotulinumtoxinA was required by 16 of the 23 patients after a mean (SD) of 24.3 (12.4) weeks (range 12–56 weeks).

Dosing per muscle

- The most frequently injected muscles were the *flexor digitorum superficialis* (finger flexor) and the *biceps brachii* (elbow flexor) in the upper limb, and the *soleus* muscle and the thigh adductors in the lower limb.

Table 2. Patients switched from abobotulinumtoxinA to incobotulinumtoxinA

Characteristic	N=23
Age, years; mean (SD) [range]	61.5 (16.8) [26–86]
Male gender, n (%)	9 (39.1)
Cause of spasticity, n (%)	
Stroke	16 (69.6)
Multiple sclerosis	4 (17.4)
Spinal–cord injury	1 (4.3)
Other	2 (8.7)
Last abobotulinumtoxinA dose, U; mean (SD) [range]	819 (411) [150–1500]
Time to first incobotulinumtoxinA treatment, weeks; mean (SD) [range]	21.6 (7.1) [8–38]
First incobotulinumtoxinA dose, U; mean (SD) [range]	196 (106) [35–400]
Time to incobotulinumtoxinA re-treatment, weeks; mean (SD) [range]	24.3 (12.4) [12–56]

SD, standard deviation.

- Doses per muscle injected at patients' last abobotulinumtoxinA treatment and at their first incobotulinumtoxinA treatment are summarized in **Table 3**.

Goal attainment

- For the 23 patients who were switched from abobotulinumtoxinA to incobotulinumtoxinA, the most frequent treatment goal was to facilitate or to support the use of wrist-hand and/or ankle-foot orthoses (n=14; 60.9%).
- AbobotulinumtoxinA and incobotulinumtoxinA injections were similarly effective with respect to goal attainment (**Figure 1**).
 - Three of the five patients who partially achieved their treatment goals with incobotulinumtoxinA had also only achieved partial goal attainment after abobotulinumtoxinA treatment.

Tolerability

- Based on the experience in our outpatient clinic, adverse drug reactions after botulinum toxin treatment are very rare.
- In the period reviewed here, only one adverse treatment outcome occurred.
 - This was in a patient who received abobotulinumtoxinA 300 U to treat wrist flexor spasticity. One week post-treatment, the patient lost all active movement across the shoulder, elbow and fingers. Pre-treatment levels of muscle power and movement were regained after approximately four months. The patient did not receive further botulinum toxin treatments.

Table 3. AbobotulinumtoxinA and incobotulinumtoxinA dosing per muscle, N=23

	AbobotulinumtoxinA[a]		IncobotulinumtoxinA	
	Dose, U; mean (range)	Number of limbs injected[b]	Dose, U; mean (range)	Number of limbs injected[b]
Upper limb				
Flexor digitorum superficialis	204 (150–250)	12	48 (25–75)	14
Biceps brachii	278 (200–350)	9	72 (50–100)	8
Flexor digitorum profundus	192 (100–250)	6	55 (50–75)	5
Flexor carpi radialis	158 (150–200)	6	37 (25–50)	6
Brachialis	170 (150–200)	5	56 (50–75)	6
Pectoralis major	300 (300–300)	5	68 (50–75)	4
Flexor pollicis longus	81 (50–100)	4	23 (20–25)	4
Flexor carpi ulnaris	150 (150–150)	3	25 (20–30)	3
Opponens pollicis	50	1	25	1
Pronator teres	150	1	38 (25–50)	2
Flexor pollicis brevis	50	1	24 (20–25)	4
Brachioradialis	–	0	50 (50–50)	2
Lower limb				
Soleus	350 (300–400)	4	96 (75–100)	6
Gastrocnemius lateral	250 (200–300)	3	60 (20–100)	2
Gastrocnemius medial	225 (200–250)	2	100	1
Adductor magnus	200 (200–200)	2	67 (62.5–75)	3
Adductor longus	300 (300–300)	2	58 (50–75)	3
Adductor brevis	150 (150–150)	2	58 (50–75)	3
Tibialis anterior	200	1	50 (50–50)	3
Flexor digitorum longus	200	1	50 (50–50)	3
Tibialis posterior	–	0	50 (50–50)	3
Flexor digitorum brevis	–	0	50 (50–50)	2

[a]Dosing per muscle data were available for 19 patients.
[b]Patients may have been injected bilaterally; each limb was counted separately.

Figure 1. Goal attainment with botulinum toxin treatment, N=23

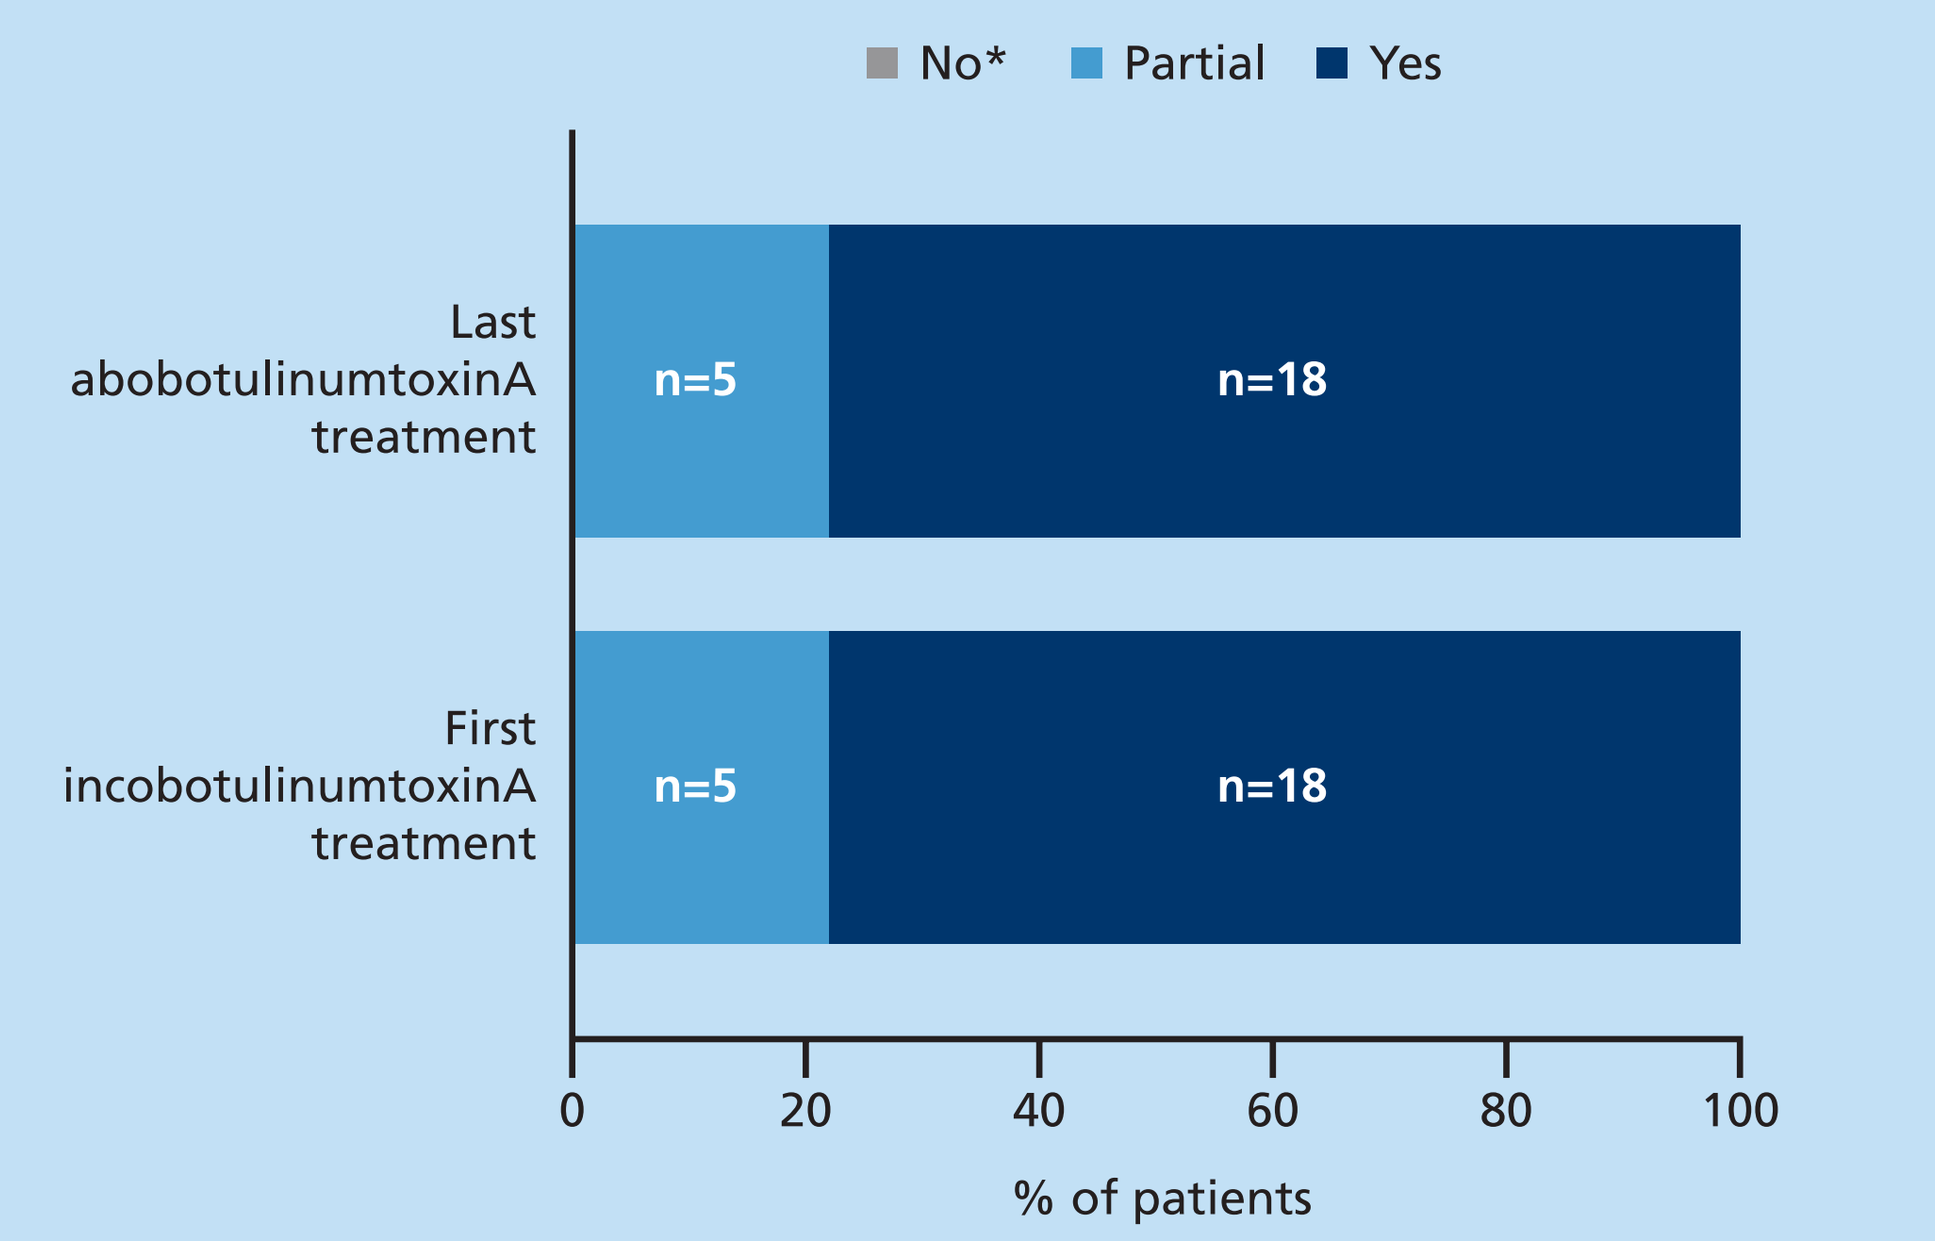
No*
Partial
Yes
Last abobotulinumtoxinA treatment
First incobotulinumtoxinA treatment
n=5
n=18
n=5
n=18
0
20
40
60
80
100
% of patients

Goal attainment was recorded as *yes*, *no*, or *partial*. Patients may have had more than one treatment goal. For the overall rating shown, patients with more than one goal were classed as *yes* if all goals were classed as *yes*. Patients who were classed as *yes* for some goals but as *partial* for others were classed as *partial* overall.
*A rating of *no* did not occur.

Conclusions

- In our outpatient clinic, abobotulinumtoxinA and incobotulinumtoxinA, administered at a ~4:1 unit ratio, were similarly effective in enabling patients with spasticity to achieve their treatment goals.
- Treatment intervals remained similar after switching from abobotulinumtoxinA to incobotulinumtoxinA.
- The formulations had similarly favorable safety profiles. Only one adverse treatment outcome occurred in a patient who had a single treatment with abobotulinumtoxinA and was not switched to incobotulinumtoxinA.

References

1. Esquenazi A et al. Toxicon 2013; 67: 115–128.
2. Royal College of Physicians et al. 2009. Available at: https://www.rcplondon.ac.uk/sites/default/files/documents/spasticity-in-adults-management-botulinum-toxin.pdf.
3. Simpson DM et al. Neurology 2016; 86: 1818–1826.
4. Wissel J et al. J Rehabil Med 2009; 41: 13–25.
5. Allergan. 2014. Available at: http://www.medicines.org.uk/EMC/medicine/112/SPC/.
6. Ipsen. 2013. Available at: http://www.medicines.org.uk/EMC/medicine/870/SPC/.
7. Merz Pharma UK Ltd. 2014. Available at: http://www.medicines.org.uk/emc/medicine/20666.

Acknowledgments

The authors did not receive financial support for this study. Editorial support was provided by Simone Boldt, PhD, of Complete Medical Communications and funded by Merz Pharmaceuticals GmbH.

Presented at TOXINS 2017, Madrid, Spain, 18–21 January 2017

MRZ82220j_TOXINS_Cossar.indd 1
11/01/2017 13:39