Improvement of spasticity following abobotulinumtoxinA (Dysport®) injections in shoulder muscles in hemiparetic patients with upper limb spasticity – Sub-analysis of a prospective, long-term, open-label study with single and repeated injection cycles

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by TOXINS

Fatma Gul[1], Michael O'Dell[2], Robert Jech[3], Marta Banach[4], Claire Vilain[5], Anne-Sophie Grandoulier[5], Jean-Michel Germain[6], Jean-Michel Gracies[7]

[1]The University of Texas Southwestern Medical Center, Dallas, Texas, USA; [2]Weill Cornell Medical College, New York, New York, USA; [3]Department of Neurology, First Faculty of Medicine, Charles University and General Faculty Hospital, Prague, Czech Republic; [4]Department of Neurology, Jagiellonian University Medical College, Krakow, Poland; [5]Ipsen, Les Ulis, France; [6]Ipsen, Boulogne-Billancourt, France; [7]EA7377 BIOTN, Université Paris-Est Créteil, Service de Rééducation Neurolocomotrice, Hôpitaux Universitaires Henri Mondor, Créteil, France

Background

- Patients with significant upper limb spasticity (ULS) often have shoulder involvement, which can affect functional shoulder movements (e.g. reaching).[1,2]
- The benefits of abobotulinumtoxinA (aboBoNT-A, Dysport®) in ULS have been well established.[3] In particular, a recent double-blind study involving finger, wrist, elbow and shoulder injections with aboBoNT-A reported improvements in muscle tone, clinical benefit and perceived function following a single injection cycle.[4]
- Few studies have assessed long-term safety and efficacy of repeated aboBoNT-A injections in ULS.

Objective

- To descriptively analyze the effect of single and repeated injections of aboBoNT-A into shoulder extensors and internal rotators on spasticity in a subpopulation of hemiparetic adults.

Methods

- Sub-analysis of an international phase 3, multicenter, open-label, extension of a double-blind study[5] (Figure 1).
 - Patients were enrolled over 34 sites in Europe and the US.
- Key inclusion/exclusion criteria for the open-label study were:
 - Hemiparetic adults aged between 18 and 80 years, inclusive.
 - ≥6 months post-stroke or -traumatic brain injury (TBI).
 - Modified Ashworth Scale score ≥2 or 3 in primary target muscle group (PTMG) for toxin- and non-naïve patients, respectively.
 - Disability Assessment Scale score ≥2 in principal target of treatment.
 - Spasticity angle ≥10° in PTMG (difference between Tardieu Scale parameters passive range of motion [X_{V1}] and angle of catch [X_{V3}]).
 - Overall Modified Frenchay Scale (evaluating performance of 10 upper limb tasks) score of 1–8 (rated on a 10-point scale).
- Patients received 1000 or 1500 U of aboBoNT-A per cycle for up to 4/5 cycles with a minimum of 12 weeks between injection cycles.
 - This included single or repeated injection cycles into shoulder muscles at a maximum dose of 500 U per cycle.
- Post-treatment follow-up visits occurred at weeks 4 and 12, with discretionary visits at weeks 16, 20 and 24.

Main outcome measure

- Spasticity was assessed by the Tardieu Scale:
 - X_{V1}, X_{V3} and spasticity angle (X; $X=X_{V1}-X_{V3}$) in shoulder muscles.
- Descriptive statistics were used to characterise demographics, aboBoNT-A dosage and efficacy variables.
- Efficacy data are presented for week 4 of the last treatment cycle to include shoulder injections.

Figure 1. Study flow.

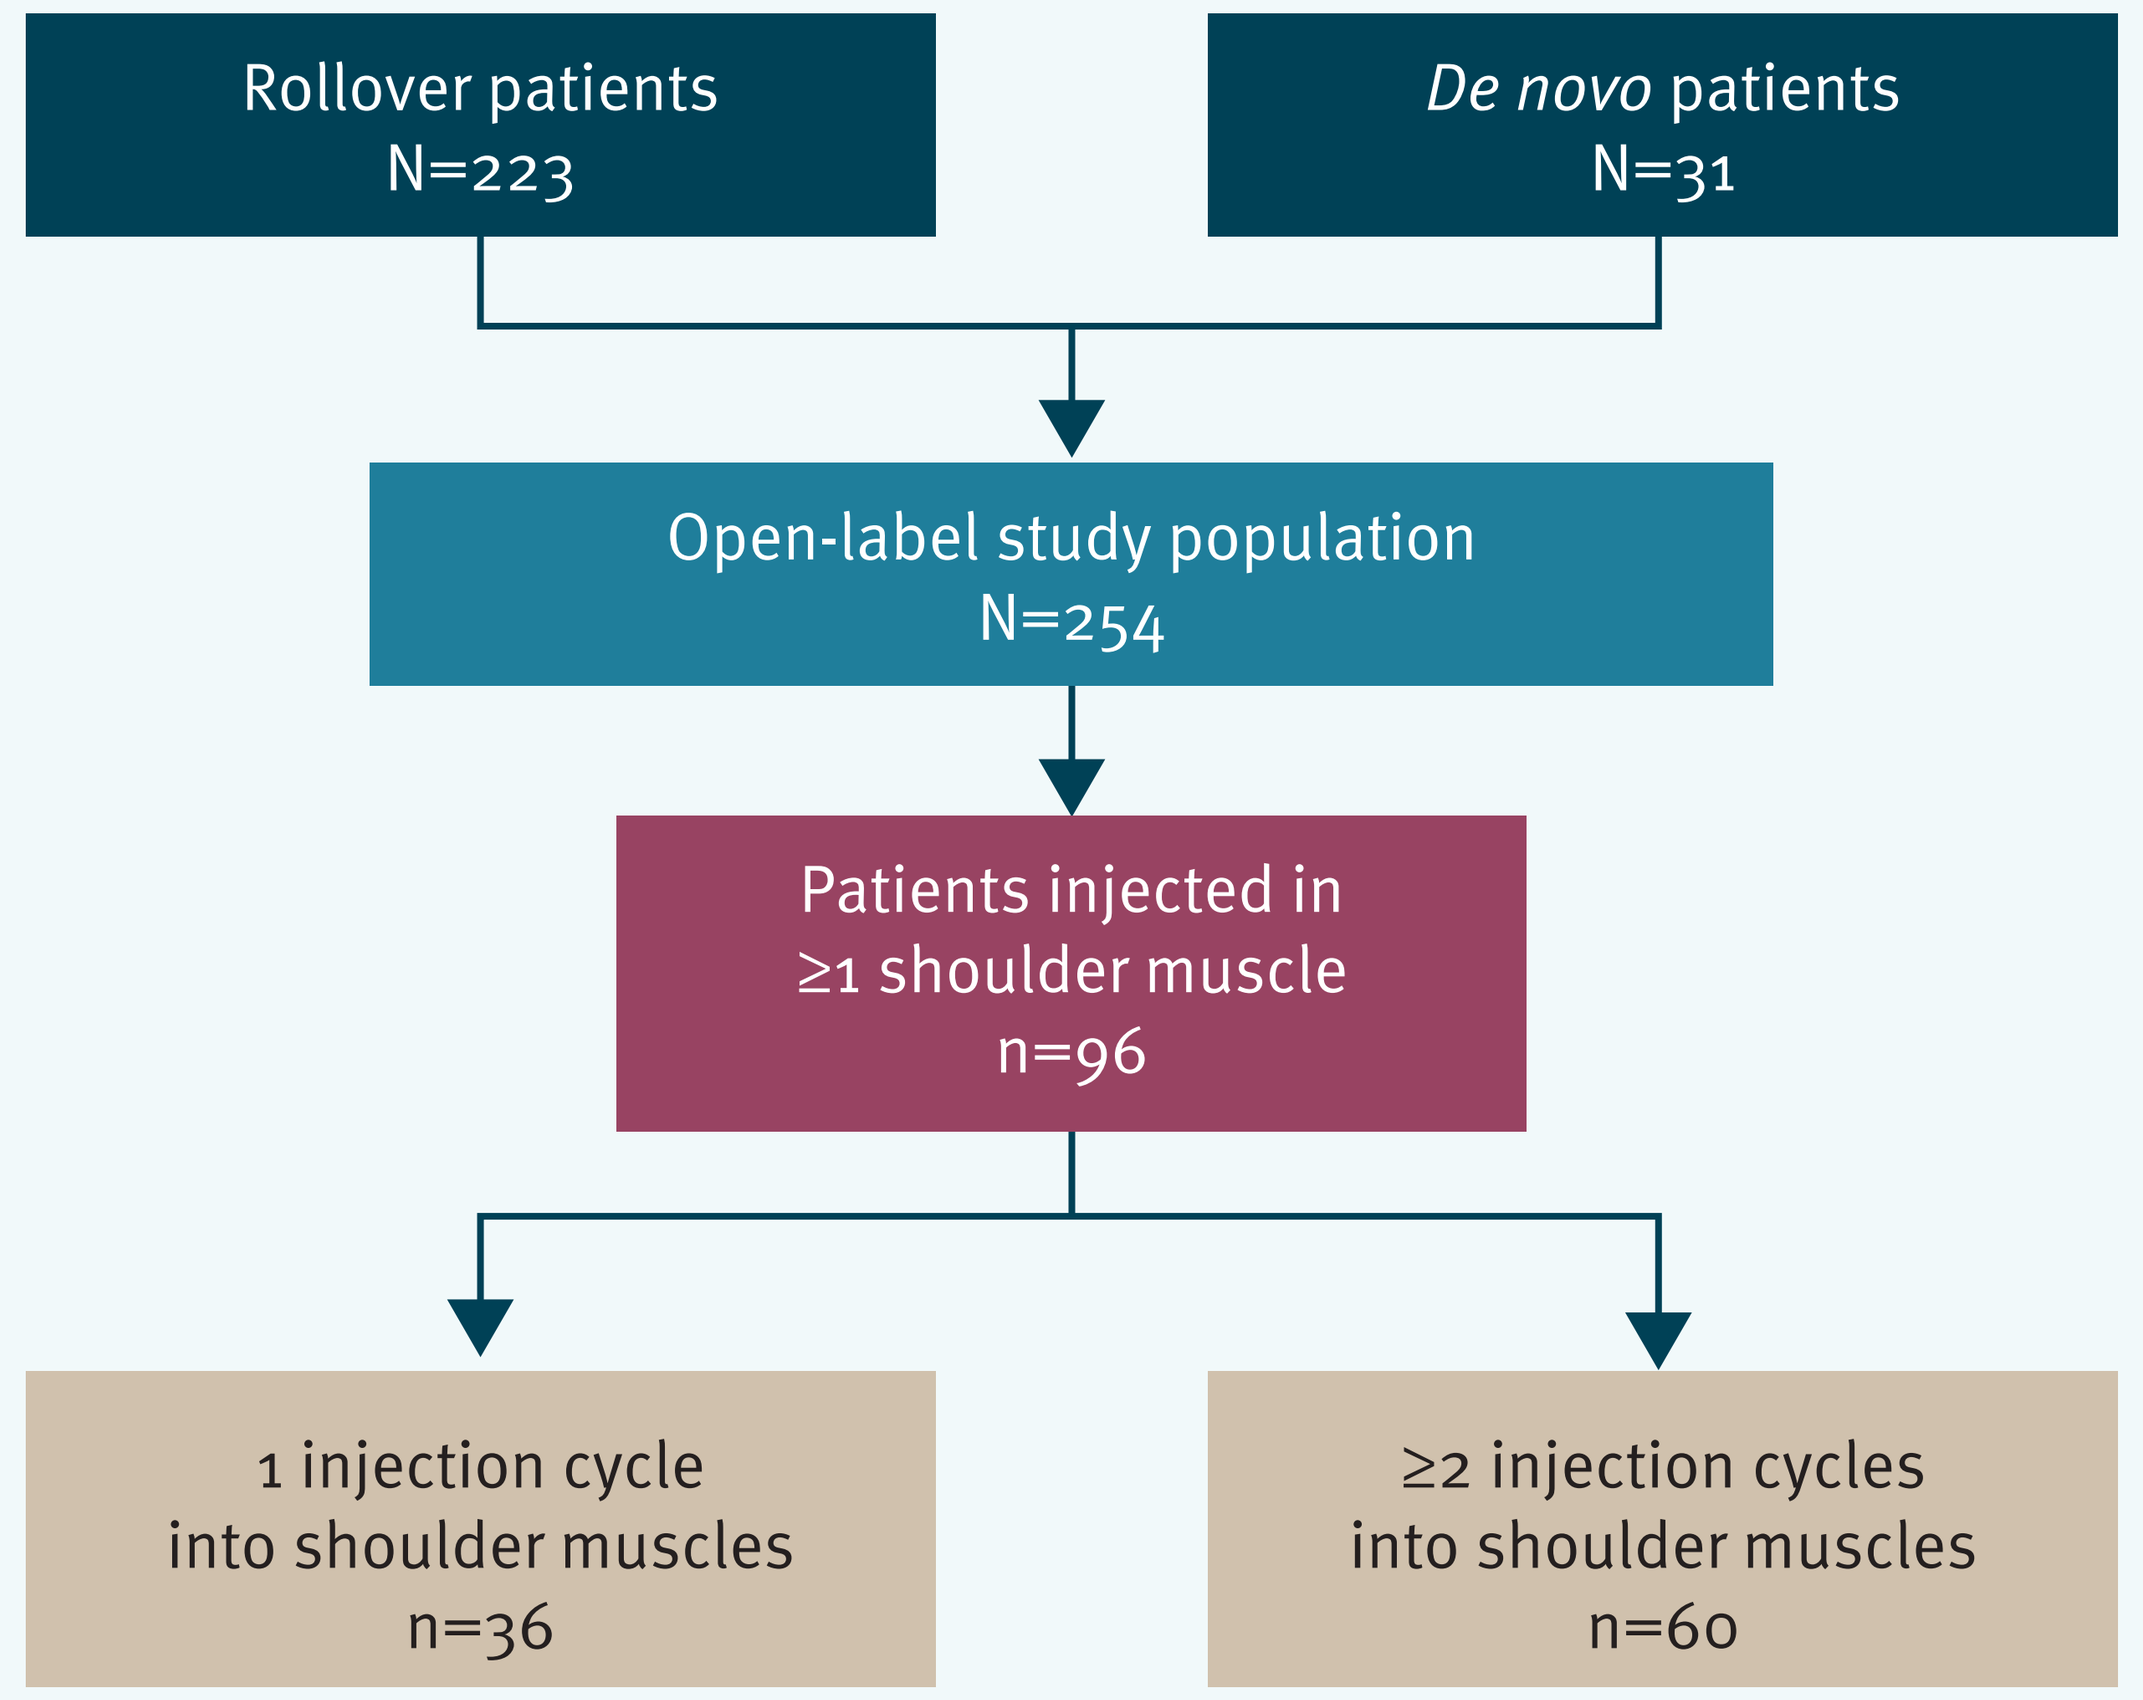
Rollover patients
N=223
De novo patients
N=31
Open-label study population
N=254
Patients injected in
≥1 shoulder muscle
n=96
1 injection cycle
into shoulder muscles
n=36
≥2 injection cycles
into shoulder muscles
n=60

Results

- Baseline data for patients (n=96) receiving at least one shoulder muscle injection cycle are shown in Table 1.
- The most commonly injected shoulder muscle was pectoralis major followed by latissimus dorsi, triceps brachii and subscapularis.
 - Mean injection doses are shown in Table 2.
- In patients with only one shoulder injection, both X_{V1} and X_{V3}, improved with a mean change from baseline (standard deviation [SD]) of +8.1° (16.9) and +15.4° (26.1) for X_{V1} and X_{V3}, respectively. Consequently the spasticity angle (X) was reduced by −7.3° (25.1; Figure 2).
- In patients receiving shoulder injections repeatedly, no further improvements were observed for X_{V1} compared with one injection cycle, with a mean change from baseline (SD) of +7.9 (25.7) compared with +8.1° (16.9; Figure 2).
- Nevertheless greater improvements were observed for X_{V3} and X for repeated injections compared with a single injection cycle, mean change from baseline (SD) was 19.1 (25.63) vs. +15.4° (26.1) for X_{V3} and −11.2° (28.3) vs. −7.3° (25.1) for X (Figure 2).

Safety

- Safety analysis did not generate any unexpected safety findings.
- Consistent with the overall study population, the most common adverse events were fall, muscular weakness and pain in the extremity (Table 3).

Table 1. Baseline characteristics.

Characteristic	Patients (N=96)
Age, mean (SD)	52.1 (13.1)
Sex, male n (%)	64 (66.7)
Treatment naïve, n (%)	45 (46.9)
Cause of spasticity, n (%)	
Stroke	85 (88.5)
TBI	11 (11.5)
PTMG, n (%)	
Extrinsic finger flexors	56 (58.3)
Wrist flexors	13 (13.5)
Elbow flexors	27 (28.1)

PTMG, primary target muscle group TBI, traumatic brain injury.

Figure 2. Mean change from baseline to Week 4 of the last injection cycle to include shoulder muscles in passive range of motion (X_{V1}), angle of catch (X_{V3}) and spasticity angle (X).

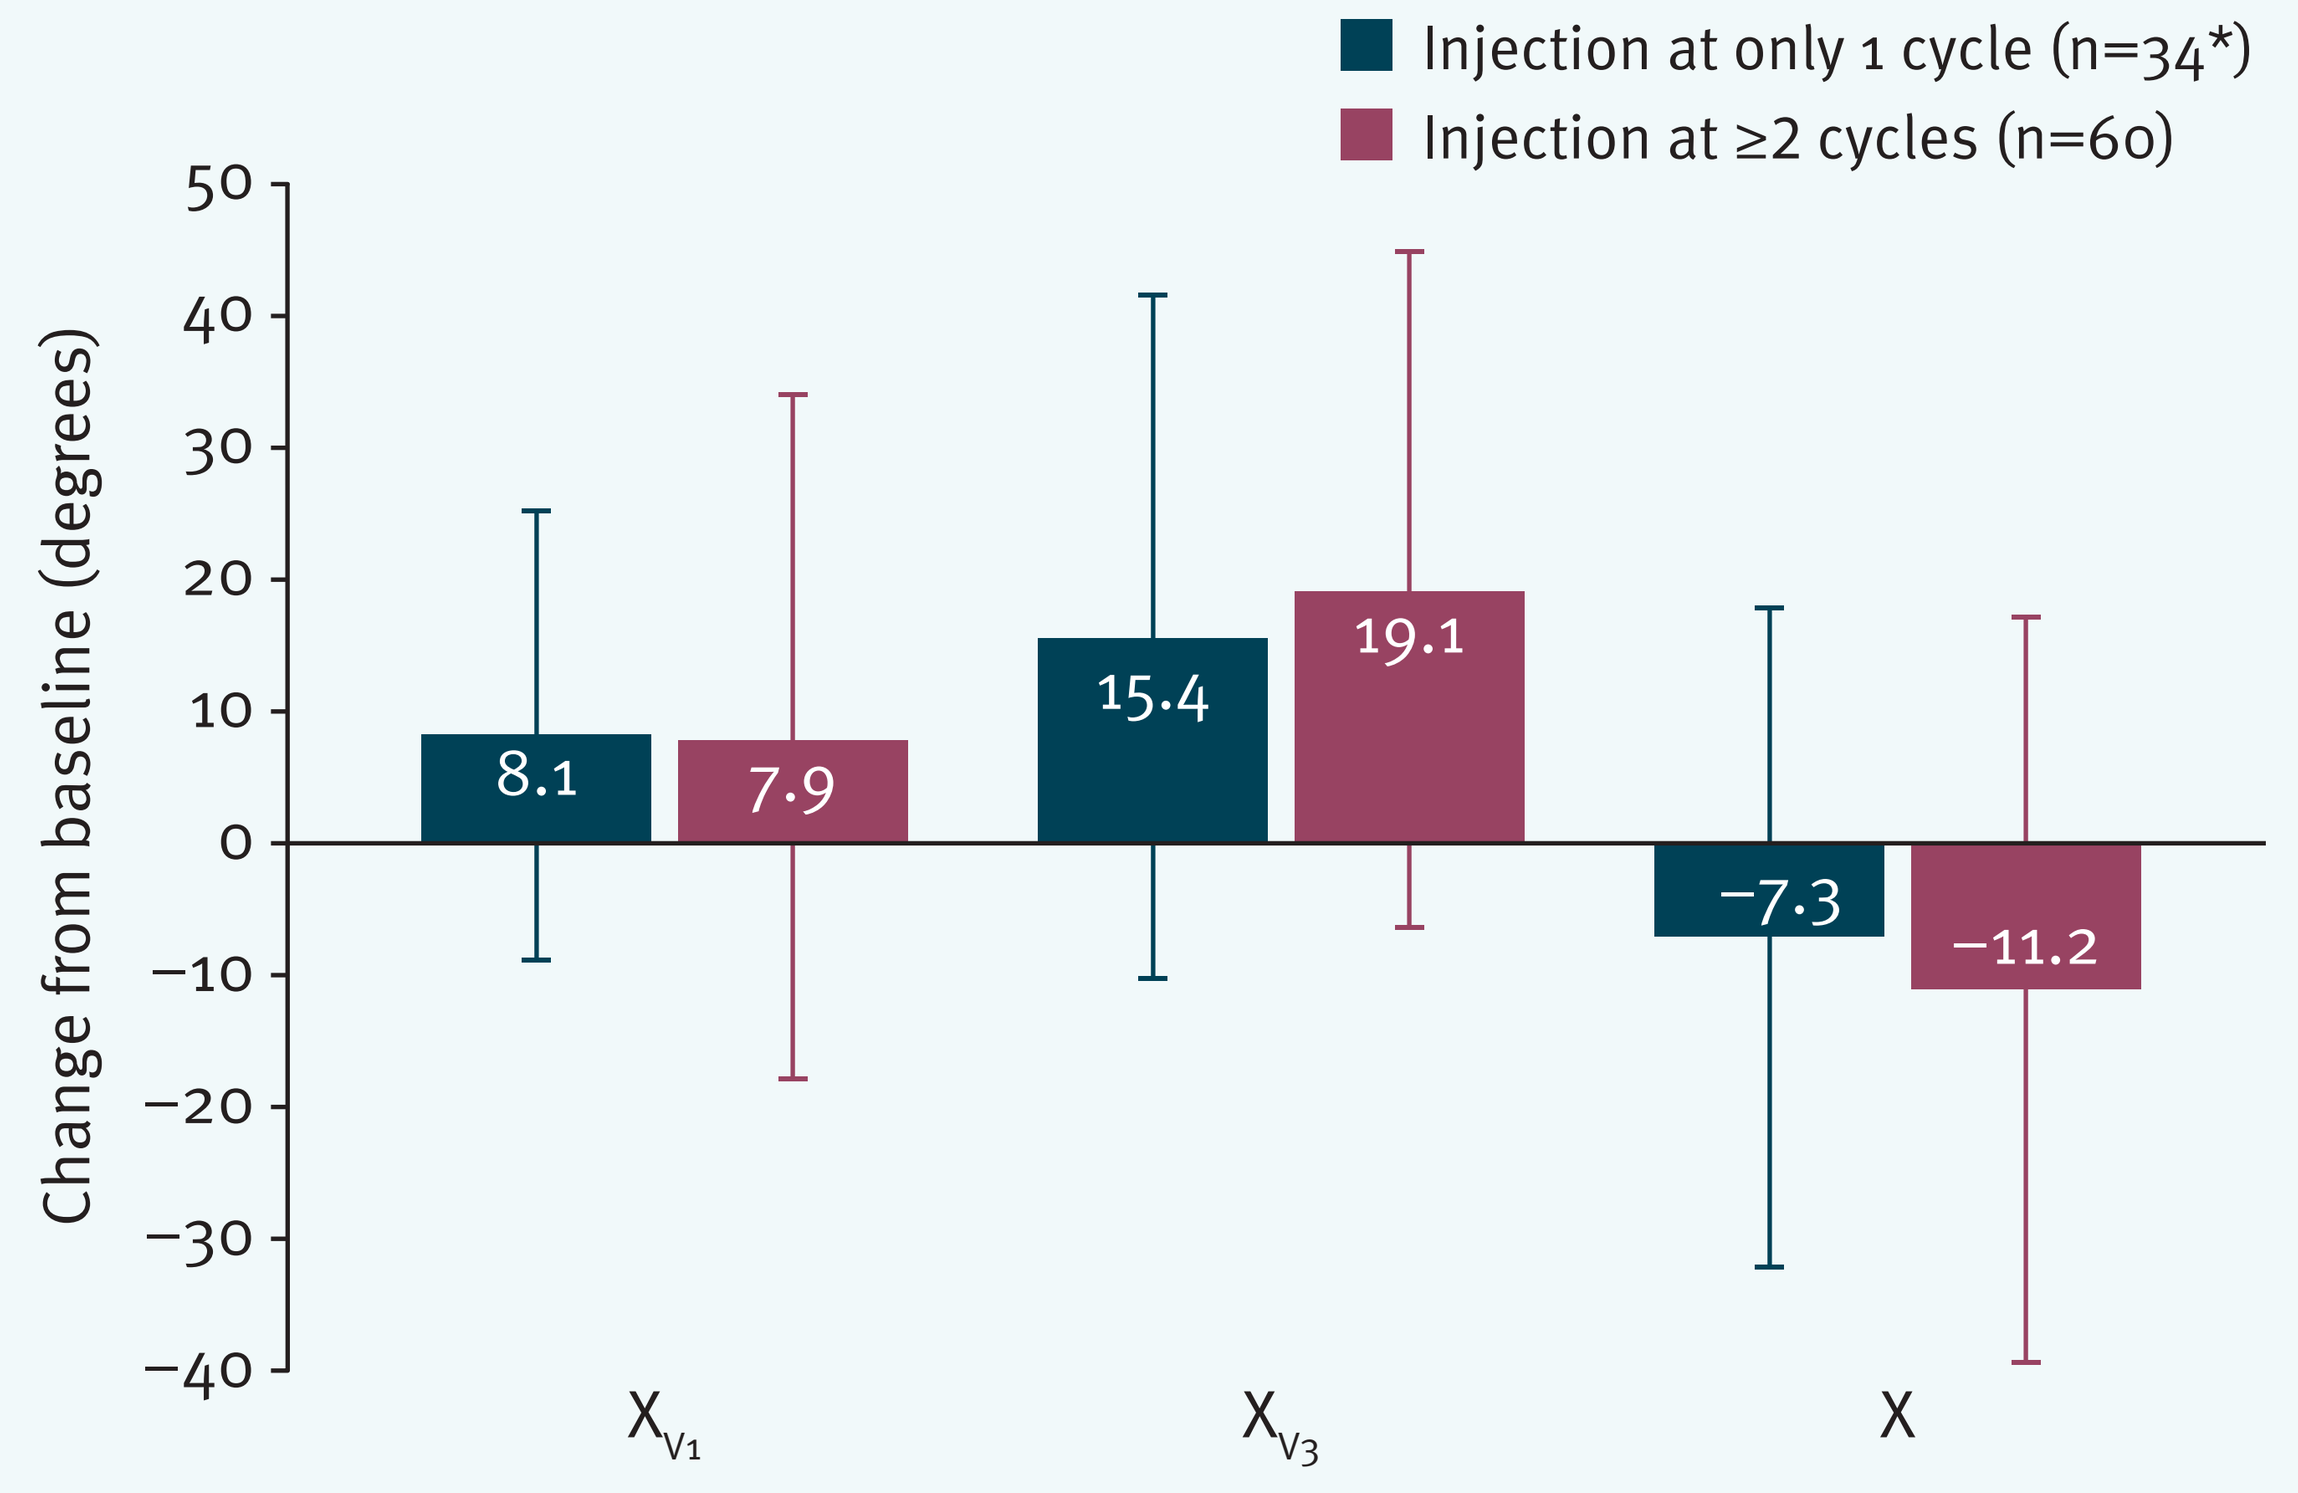
Injection at only 1 cycle (n=34*)
Injection at ≥2 cycles (n=60)
Change from baseline (degrees)
50
40
30
20
10
0
−10
−20
−30
−40
8.1
7.9
15.4
19.1
−7.3
−11.2
X_{V1}
X_{V3}
X

*Two patients receiving shoulder injections at only one injection cycle had no Tardieu Scale results at Week 4.

Table 2. Mean doses injected in each shoulder muscle.

Muscle	Cycle 1 n=35	Cycle 2 n=67	Cycle 3 n=57	Cycle 4 n=33	Cycle 5 n=5
Triceps brachii	n=12	n=29	n=23	n=10	n=1
Mean (SD)	150.0 (67.4)	184.8 (69.2)	186.1 (58.9)	194.0 (92.9)	200 (–)
[range]	[100 to 300]	[80 to 300]	[100 to 300]	[80 to 300]	
Pectoralis major	n=23	n=52	n=48	n=30	n=4
Mean (SD)	168.3 (47.5)	226.0 (52.1)	227.7 (65.3)	224.0 (69.4)	285.0 (144.6)
[range]	[100 to 260]	[100 to 300]	[100 to 500]	[100 to 500]	[200 to 500]
Subscapularis	n=4	n=17	n=18	n=10	n=1
Mean (SD)	175.0 (50)	147.6 (59.5)	122.8 (60.6)	130.0 (48.3)	100 (–)
[range]	[100 to 200]	[100 to 300]	[0 to 260]	[100 to 200]	
Latissimus dorsi	n=11	n=31	n=32	n=13	n=2
Mean (SD)	200 (0)	194.5 (70.6)	193.4 (65.5)	175.4 (51.7)	150 (70.7)
[range]	[200 to 200]	[100 to 400]	[0 to 300]	[100 to 260]	[100 to 200]

SD, standard deviation.

Table 3. Treatment-emergent adverse events reported in at least two patients in each sub-group.

Adverse event	n (%) [Number of events]	
	Shoulder injection at only 1 cycle n=36	Shoulder injection at ≥2 cycles n=60
Fall	8 (22.2) [9]	3 (5.0) [3]
Muscular weakness	5 (13.9) [8]	3 (5.0) [3]
Pain in extremity	4 (11.1) [6]	4 (6.7) [5]
Arthralgia	4 (11.1) [5]	1 (1.7) [1]
Nasopharyngitis	3 (8.3) [3]	3 (5.0) [3]
Bronchitis	3 (8.3) [3]	2 (3.3) [2]
Musculoskeletal pain	3 (8.3) [4]	2 (3.3) [4]
Headache	2 (5.6) [2]	3 (5.0) [3]
Epilepsy	0	3 (5.0) [6]
Fatigue	2 (5.6) [2]	2 (3.3) [3]
Influenza	2 (5.6) [2]	2 (3.3) [3]
Pyrexia	2 (5.6) [4]	1 (1.7) [1]
Urinary tract infection	2 (5.6) [7]	1 (1.7) [1]
Cystitis	1 (2.8) [1]	2 (3.3) [2]
Joint swelling	1 (2.8) [1]	2 (3.3) [3]
Constipation	2 (5.6) [2]	0
Nasal congestion	2 (5.6) [2]	0
Peripheral swelling	2 (5.6) [2]	0
Vision blurred	2 (5.6) [3]	0
Contusion	0	2 (3.3) [2]
Herpes zoster	0	2 (3.3) [2]
Hyperhidrosis	0	2 (3.3) [2]

Conclusions

- These data suggest the potential benefit of abobotulinumtoxinA injections in shoulder extensors and internal rotators in patients with upper limb spasticity.
- Greater improvements were observed in angle of catch and spasticity angle with repeated abobotulinumtoxinA injections in shoulder muscles compared with single injection cycles.
- Safety data were in line with the known safety profile of abobotulinumtoxinA.

References

1. Yelnik AP, *et al. J Neurol Neurosurg Psychiatry* 2007; 78:845–8.
2. Marco E, *et al. J Rehabil Med* 2007; 39:440–7.
3. Dashtipour K, *et al. Am J Phys Med Rehabil* 2015;94:229–38.
4. Gracies JM, *et al. Lancet Neurol* 2015;14:992–1001.

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield Company, for medical writing support (funded by Ipsen) in preparing this poster.

Disclosures

FG has received compensation from Ipsen for being an advisory board member and support from Allergan for consultancy, speaking engagements, and preceptorship. FG is an investigator in an Ipsen trial. MO'D has been a consultant for, and has received research grants from Ipsen. MO'D is an investigator in an Ipsen trial. RJ has received grants from the Czech Science Foundation, Czech Ministry of Health, Czech Ministry of Education and Charles University, Prague; and honoraria from Ipsen for consultations and lectures. MB has received honoraria from Ipsen for consultations and lectures. MB is an investigator in an Ipsen trial. CV, A-SG and J-M Germain are employees of Ipsen. J-M Gracies has served as a consultant and received research grant support from Allergan, Ipsen, and Merz; he is an investigator on an Ipsen trial.

Presented at TOXINS, 18–21 January 2017, Madrid, Spain.

This study was sponsored by Ipsen