Safety and Tolerability of OnabotulinumtoxinA in the Treatment of Facial Lines: Statistical Meta-analysis of Pooled Data From Global Registration Studies of Treatment of Crow's Feet Lines and Glabellar Lines in More Than 3900 Participants

Jean Carruthers,[1] Mitchell F. Brin,[2,3] Koen De Boulle,[4] Steven Liew,[5] Alastair Carruthers,[6] Alexander Rivkin,[7] Yan Wu,[8] Irina Yushmanova,[2] Terry I. Boodhoo,[2] Kathy Zhuang,[2] Conor J. Gallagher,[2] Elisabeth Lee[2]

[1]University of British Columbia, Vancouver, BC, Canada; [2]Allergan plc, Irvine, CA, USA; [3]University of California, Irvine, CA, USA; [4]Aalst Dermatology Clinic, Aalst, Belgium; [5]Private Practice, Sydney, NSW, Australia; [6]University of British Columbia, Vancouver, BC, Canada; [7]Private Practice, Los Angeles, CA, USA; [8]Peking University First Hospital, Beijing, China

Scan to obtain PDF of poster

ABSTRACT

Objective: A meta-analysis of pre-2009 registration studies (N=1678; Brin, *J Am Acad Dermatol* 2009;61:961-70) evaluated safety/tolerability of onabotulinumtoxinA for glabellar lines (GL) and crow's feet lines (CFL). We pooled safety data from pre- and post-2009 registration studies, focusing on those in CFL and CFL+GL combined.

Methods: Safety data were pooled for 3946 participants from 15 onabotulinumtoxinA studies. Of 3338 participants from 11 randomized, double-blind, placebo-controlled (DBPC) studies, 2394 received onabotulinumtoxinA for GL (n=665), CFL (n=1324), and CFL+GL (n=405); remaining participants received placebo. A total exposure (TE) population included onabotulinumtoxinA-treated participants (n=3189) from the DBPC studies plus 4 wherein all participants received onabotulinumtoxinA.

Results: Participants in the DBPC population were predominantly Caucasian (59.9%), Asian (33.6%), and female (85.1%); median age was 47.0 (21.8–85.0) years. Age and gender demographics were similar for DBPC studies in Asia (4) versus North America/Europe (7). Among CFL participants (7 DBPC studies), demographics were similar to the overall DBPC population. Among CFL+GL participants (2 DBPC studies), 87.4% were Caucasian. The GL subpopulation demographics were similar to Brin 2009. Adverse events (AEs) in the DBPC population are reported here. Overall AE incidence was similar between the onabotulinumtoxinA and placebo groups, respectively (41.2%; 37.1%) and generally lower in Asian studies (28.0%; 26.3%) versus North America or Europe (47.6%; 41.5%). Overall, most common AEs (>2%) were headache (onabotulinumtoxinA, 6.6%; placebo, 5.6%), nasopharyngitis (6.3%; 5.6%), and injection site bruising (2.5%; 1.8%). For CFL participants (onabotulinumtoxinA, n=1324; placebo, n=808), overall AE incidence with onabotulinumtoxinA and placebo, respectively, was 36.5% and 36.3%; for CFL+GL participants (onabotulinumtoxinA, n=405; placebo, n=306), 50.1% and 44.4%. Most common AEs were similar between subpopulations and the overall population. Treatment-related AEs (>1%) were, for onabotulinumtoxinA and placebo, respectively, headache (2.0%; 2.1%), injection site hematoma (1.7%; 1.0%), and injection site bruising (1.5%; 1.0%) for CFL participants; headache (5.7%; 3.6%), injection site hematoma (3.2%; 1.3%), injection site bruising (2.0%; 0.7%), and eyelid ptosis (1.2%; 0%) for CFL+GL participants. Serious AE incidence was similar between onabotulinumtoxinA-treated participants versus placebo in CFL (1.4% vs 1.5%) and CFL+GL participants (2.7% vs 2.3%). Analysis of AEs potentially related to distant spread of toxin in the DBPC studies showed an overall AE incidence of 1.7% and 0.7% for onabotulinumtoxinA and placebo, respectively; those AEs were known local effects. Treatment-related and serious AEs in the pooled GL population were similar to those reported in Brin.

Conclusion: This pooled safety analysis including >3900 participants confirmed the safety and tolerability of onabotulinumtoxinA for treatment of facial lines.

INTRODUCTION

- The efficacy and safety of onabotulinumtoxinA has been established in >60 randomized, placebo-controlled clinical trials over >20 years
- In a previously published meta-analysis, Brin et al. examined safety and tolerability data for >1600 subjects treated with onabotulinumtoxinA for glabellar lines (GL) and crow's feet lines (CFL) in studies conducted prior to 2009[1]
- Adverse events (AEs) associated with onabotulinumtoxinA were generally mild to moderate, and most treatment-related AEs were attributable to local pharmacologic effects or to the physical injection of the product
- In the current meta-analysis, we analyzed the safety profile of onabotulinumtoxinA utilizing data pooled from 15 placebo-controlled and single-agent studies of onabotulinumtoxinA for treatment of GL, CFL, and concurrent CFL and GL (CFL/GL), including studies completed since 2009

METHODS

Study Design

- This analysis included double-blind, placebo controlled (DBPC) and open-label (OL) clinical trials of onabotulinumtoxinA used for treatment of GL, CFL, and combined CFL/GL
- All studies were sponsored by Allergan plc or its business partners and were completed, with databases locked and study reports available
- In addition to the studies previously analyzed in Brin et al., which included 9 unique studies (4 DBPC GL studies, 3 OL GL studies, and 2 DBPC CFL studies), the current meta-analysis includes 3 new DBPC CFL studies, 2 new DBPC CFL/GL studies, and 1 new OL CFL study
- Eleven DBPC studies and 4 OL studies were included in this meta-analysis (**Table 1**)
- Data from the majority of these studies have been published[1-10]

Table 1. Characteristics of Studies Included in Meta-Analysis

Indication	Type of Study (Identifier)	Geographic Region	Duration, Weeks	DBPC Population, OnabotA	DBPC Population, Placebo	Total Exposure Population
GL	DBPC (010)	NA	16	203	60	203
	DBPC (023)	NA	16	202	70	202
	OL (018)	NA	16	0	0	96
	OL (907)	Japan	16	0	0	125
	DBPC (908)	Japan	16	90	49	90
	OL (909)	Japan	64	0	0	363
	DBPC (107457)	China	16	170	57	190
CFL	DBPC (512)	EU	24	130	32	130
	DBPC (514)	EU	12	149	48	149
	DBPC (098)	NA/EU	20	220	224	220
	DBPC (113)	China	20	316	101	316
	DBPC (114)	Japan	56	203	97	294
CFL and CFL/GL	DBPC (099)	NA/EU	32	611	306	611
	DBPC (104)*	NA/EU	20	100	0	100
CFL/GL	OL (122)	Japan	56	0	0	100
TOTAL				2394	1044	3189

*Extension of Study 099, N=684 (onabotulinumtoxinA, n=588; placebo, n=96; however, new exposure to onabotulinumtoxinA, n=101).

CFL, crow's feet lines; CFL/GL, crow's feet lines and glabellar lines combined; DBPC, double-blind placebo-controlled; EU, European Union; GL, glabellar lines; NA, North America; OL, open-label; onabotA, onabotulinumtoxinA.

Eligible Study Subjects

- All subjects enrolled in the individual studies were required to have moderate to severe, bilaterally symmetrical GL, CFL, or CFL/GL at maximum contraction (smiling for CFL; frowning for GL), based on the validated, 4-grade Facial Wrinkle Scale (FWS; 0=none, 1=mild, 2=moderate, and 3=severe)

Assessments

- All analyses were based on safety populations, defined as all subjects who received ≥1 injection of onabotulinumtoxinA or placebo in each study
 - Analyses were stratified by study treatment received
- Demographic data and disposition were analyzed by treatment group
- Safety data were analyzed by treatment group, treatment cycle (where applicable), and geographic region
- All AEs classified as one of 40 Medical Dictionary for Regulatory Activities (MedDRA) preferred terms (**Table 2**) were evaluated relative to the potential distant spread of toxin (PDSOT) beyond the site of administration
 - AEs were excluded as indicative of PDSOT if they were a local effect, inconsistent with onabotulinumtoxinA pharmacology, a nonspecific symptom, or attributable to another cause

Table 2. MedDRA Preferred Terms Used to Evaluate Potential Distant Spread of Toxin

System Organ Class	Preferred Terms
Cardiac Disorders	Bradycardia
Eye Disorders	Accommodation disorder; Diplopia; Extraocular muscles paresis; Eyelid function disorder; Eyelid ptosis; Pupillary reflex impaired; Vision blurred
Gastrointestinal Disorders	Constipation; Dry mouth; Dysphagia; Ileus paralytic
Respiratory, Thoracic, and Mediastinal Disorders	Aspiration; Diaphragmatic paralysis; Dysphonia; Dyspnea; Pneumonia aspiration; Respiratory arrest; Respiratory depression; Respiratory failure
Nervous System Disorders	Bulbar palsy; Cranial nerve palsies, multiple; Cranial nerve paralysis; Dysarthria; VII nerve paralysis; Facial paresis; Hyporeflexia; Hypotonia; Paralysis; Paralysis flaccid; Paresis cranial nerve; Peripheral nerve palsy; Peripheral paralysis; Speech disorder; Vocal cord paralysis; Vocal cord paresis
Renal and Urinary Disorders	Urinary retention
Reproductive System and Breast Disorders	Pelvic floor muscle weakness
Infections and Infestations	Botulism
Musculoskeletal and Connective Tissue Disorders	Muscular weakness

Statistical Analysis

- For each AE, incidence data were fitted to fixed-effects models (which assumed a common odds ratio across studies) using both the Peto method and the Mantel-Haenszel method, with a continuity correction proportional to the reciprocal of the relative size of the opposite treatment group
- Q statistic was employed to assess heterogeneity, which was further evaluated with the I^2 index[11]
- Tests for homogeneity between treated areas (ie, CFL/GL vs CFL alone) were performed using the Breslow-Day test at the 0.10 significance level
- No adjustments were made for multiple testing; a conservative approach with no control of type I error is generally deemed appropriate for safety analyses
- All tests other than those for homogeneity were 2-sided and were conducted at the 0.05 significance level

RESULTS

- This presentation focuses on data pooled from subjects in the 11 DBPC studies (DBPC population), data from all subjects who had any exposure to onabotulinumtoxinA in the 15 included studies (total exposure population), and data from the studies of CFL and concurrent CFL/GL (CFL and CFL/GL subpopulations)
 - Detailed presentation of data from studies that focused on the treatment of GL alone (GL subpopulation) were previously reported[1] and are not reported here

Subjects

- All participants in the DBPC population (n=3338) were predominantly Caucasian (59.9%) or Asian (33.6%) and female (85.1%), with a median (range) age of 47.0 (21.8–85.0) years
- Demographic data for the total exposure population (n=3189) were similar to those of the DBPC population; the majority were female and Caucasian or Asian (**Table 3**)
- Demographics in the CFL and CFL/GL subpopulations were similar to that for the total exposure population
- Four of the DBPC studies were conducted in Asia, 7 in North America/Europe
 - Median age was 47.0 years for both; the proportion of females was 83.5% and 85.8%, respectively
- Demographic data in the GL subpopulation were similar to those reported in Brin 2009[1]

Table 3. Baseline Demographics and Characteristics

Parameter	CFL DBPC Population		CFL/GL DBPC Population		Total Exposure Population
	OnabotA (n=1324)	Placebo (n=808)	OnabotA (n=405)	Placebo (n=306)	(n=3189)
Age, year					
Median	48.0	47.0	50.0	49.0	47.0
Range	22–75	22–74	24–85	25–73	22–85
Gender, n (%)					
Male	193 (14.6)	127 (15.7)	50 (12.3)	43 (14.1)	435 (13.6)
Female	1131 (85.4)	681 (84.3)	355 (87.7)	263 (85.9)	2754 (86.4)
Race, n (%)					
Asian	529 (40.0)	209 (25.9)	6 (1.5)	6 (2.0)	1507 (47.3)
Black	12 (0.9)	10 (1.2)	4 (1.0)	2 (0.7)	41 (1.3)
Caucasian	740 (55.9)	543 (67.2)	358 (88.4)	265 (86.6)	1517 (47.6)
Hispanic	16 (1.2)	17 (2.1)	3 (0.7)	6 (2.0)	58 (1.8)
Other	27 (2.0)	29 (3.6)	34 (8.4)	27 (8.8)	66 (2.1)

CFL, crow's feet lines; CFL/GL, crow's feet lines and glabellar lines combined; DBPC, double-blind placebo-controlled; onabotA, onabotulinumtoxinA.

Overall Incidence of Adverse Events

- Overall incidence of AEs with onabotulinumtoxinA from all 15 studies was 51.4% (1640/3189)
- For all DBPC studies, the overall incidence of AEs was similar between the onabotulinumtoxinA and placebo groups, 41.2% and 37.1%, respectively
- Overall incidence of AEs with onabotulinumtoxinA and placebo, respectively, was 36.5% and 36.3% in the DBPC CFL studies and 50.1% and 44.4% in the DBPC CFL/GL studies
- Overall incidence of AEs with onabotulinumtoxinA and placebo, respectively, was generally lower in the studies conducted in Asia (28.0% and 26.3%) versus North America or Europe (47.6% and 41.5%)
- AEs were generally mild to moderate in severity

Most Common Adverse Events

- The most common AEs (in ≥2% of subjects) with onabotulinumtoxinA in the total exposure population are shown in **Figure 1**
- For all DBPC studies, the most common AEs (≥2%) were headache (onabotulinumtoxinA, 6.6%; placebo, 5.6%), nasopharyngitis (6.3%; 5.6%), and injection site bruising (2.5%; 1.8%)
- The most common AEs (≥2%) in DBPC CFL and CFL/GL studies were similar to those seen in the total exposure population

Figure 1. Most Common Adverse Events (≥2%) in Double-Blind Placebo-Controlled Studies of Crow's Feet Lines and Crow's Feet/Glabellar Lines and for Total OnabotulinumtoxinA Exposures

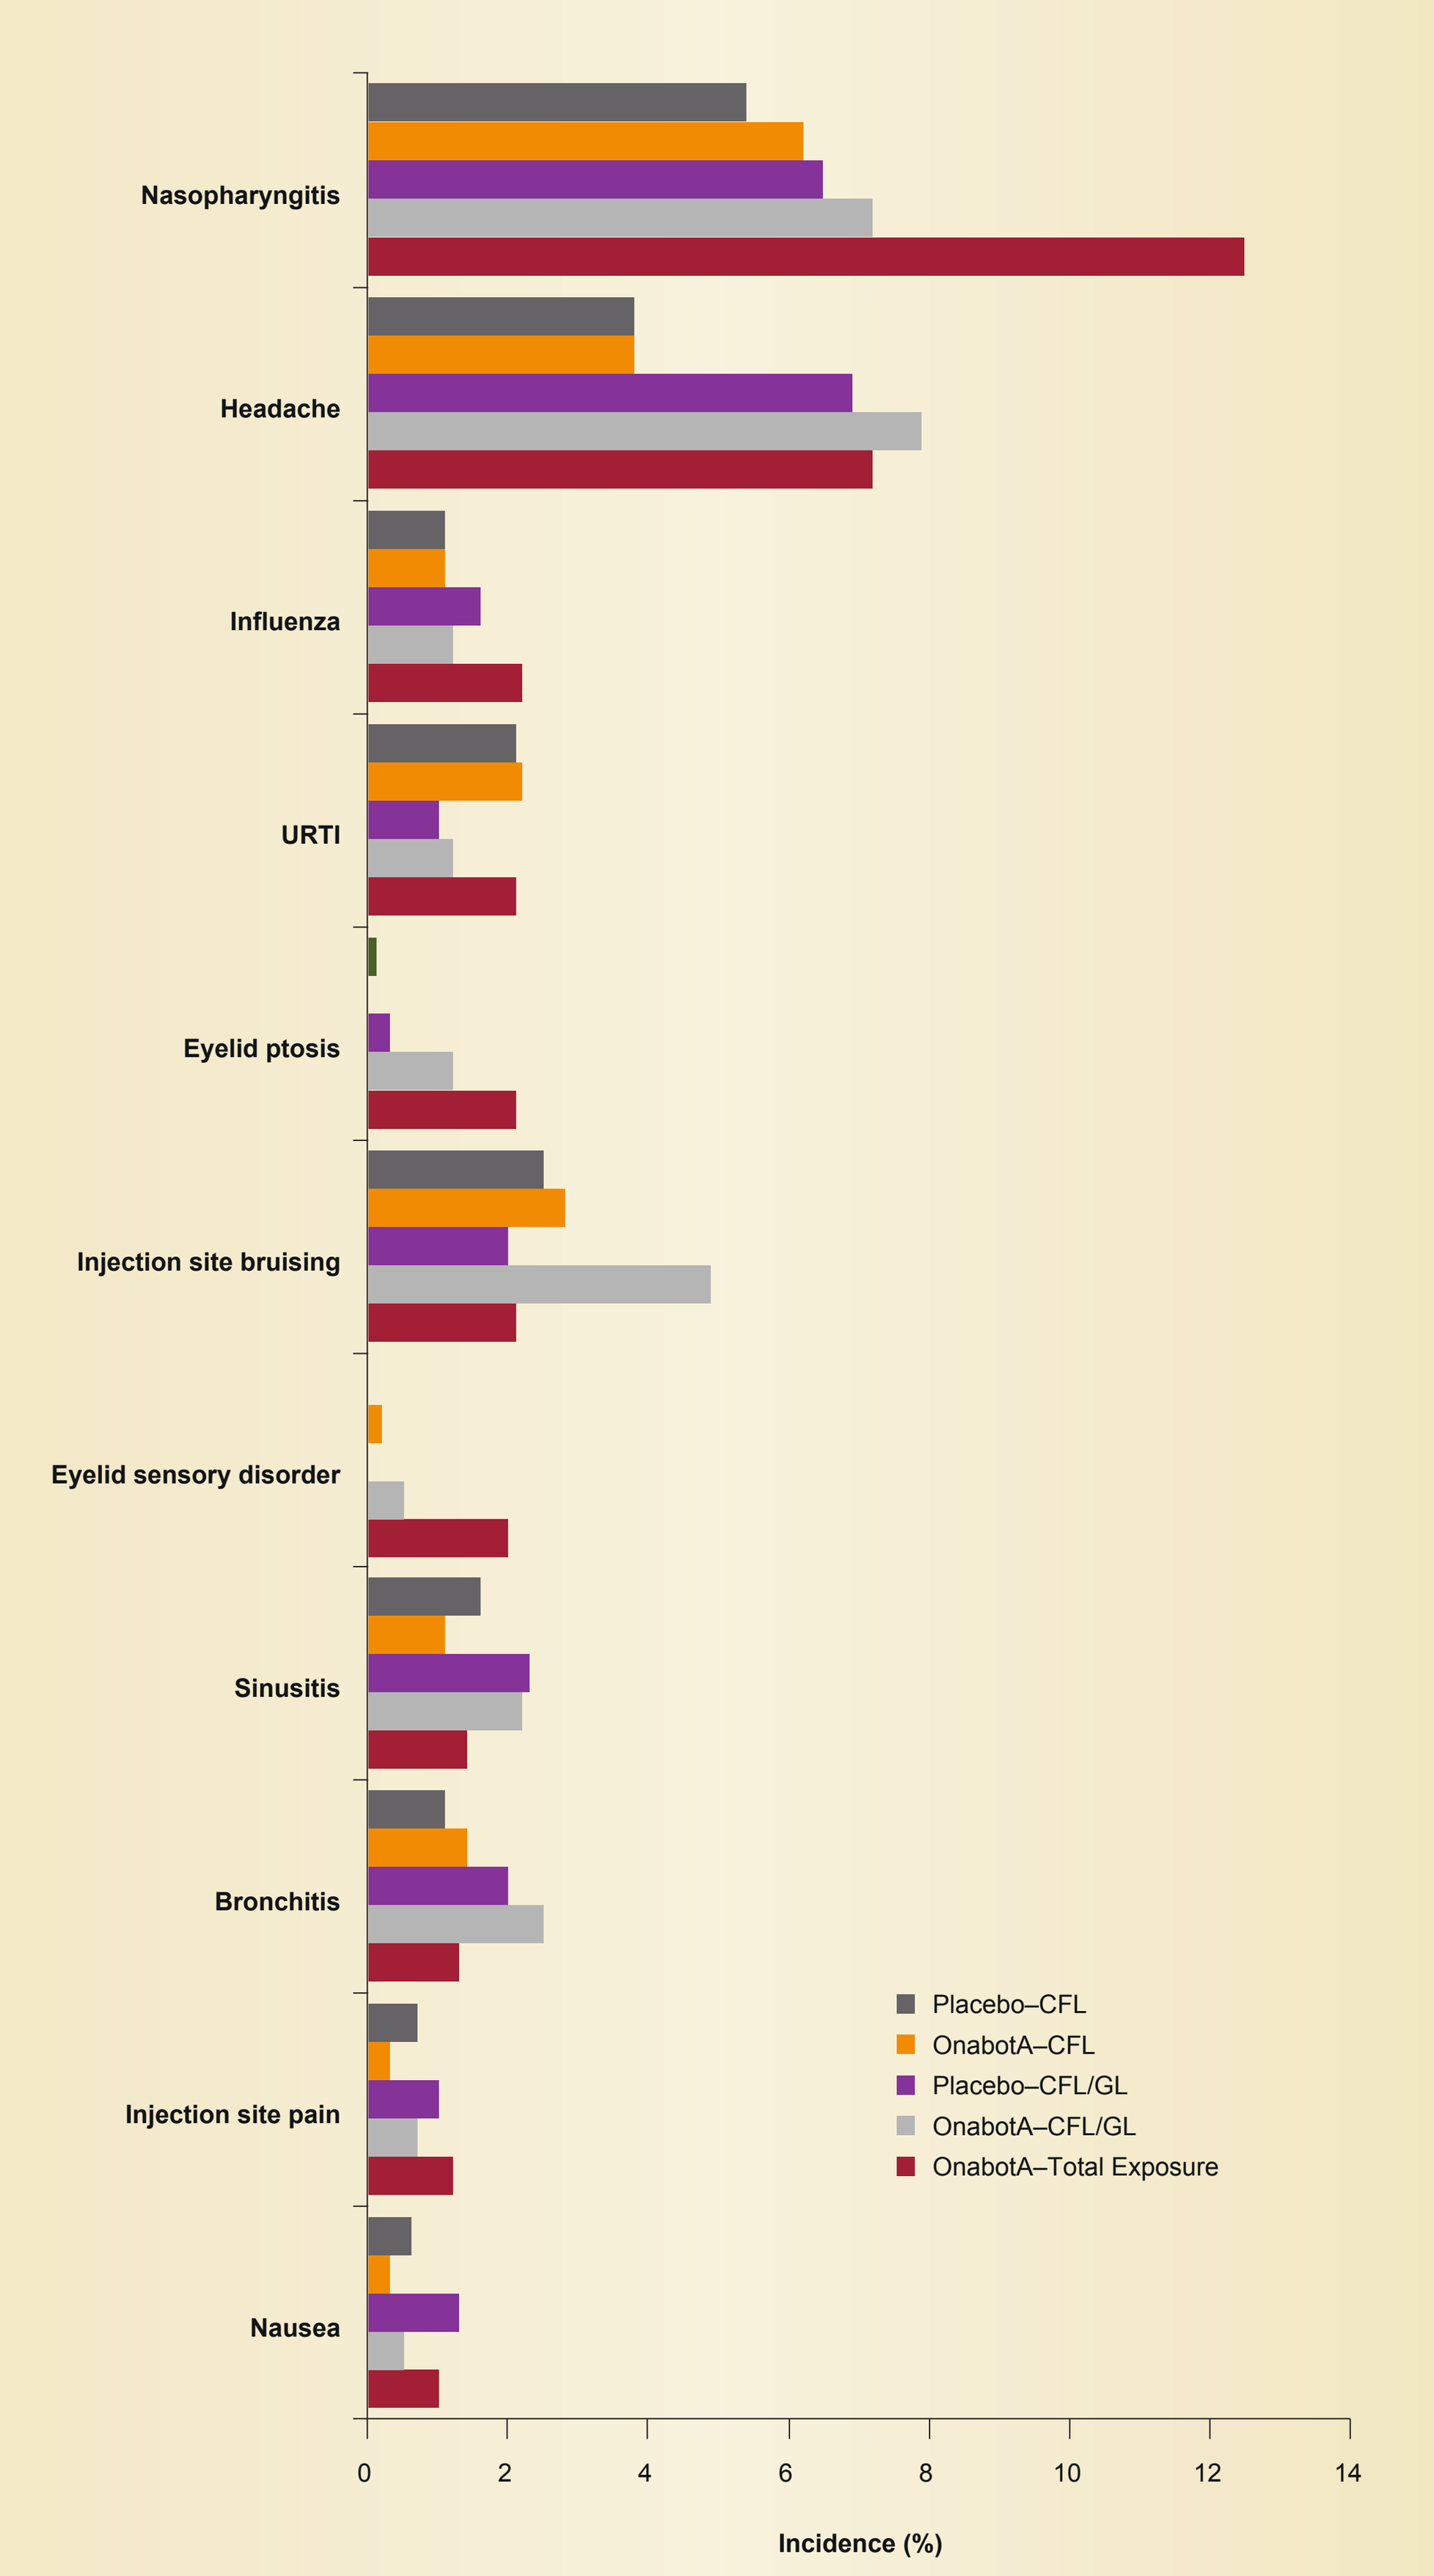

Nasopharyngitis
Headache
Influenza
URTI
Eyelid ptosis
Injection site bruising
Eyelid sensory disorder
Sinusitis
Bronchitis
Injection site pain
Nausea
0
2
4
6
8
10
12
14
Incidence (%)
Placebo–CFL
OnabotA–CFL
Placebo–CFL/GL
OnabotA–CFL/GL
OnabotA–Total Exposure

CFL, crow's feet lines; CFL/GL, crow's feet lines and glabellar lines combined; DBPC, double-blind placebo-controlled; URTI, upper respiratory tract infection; onabotA, onabotulinumtoxinA.

Treatment-Related Adverse Events

- The incidence of treatment-related AEs for all onabotulinumtoxinA exposures was 16.7% (533/3189)
- For onabotulinumtoxinA and placebo groups, respectively, in all DBPC studies, the incidence of treatment-related AEs was 13.8% (330/2394) and 8.2% (86/1044)
- The most common treatment-related AEs for all onabotulinumtoxinA exposures were similar to those in all DBPC studies for onabotulinumtoxinA and placebo (**Table 4**)

Table 4. Treatment-Related Adverse Events (≥0.5%) in the Total Exposure and Double-Blind Placebo-Controlled Populations

Preferred Term, n (%)	Total Exposure Population (n=3189)	DBPC Population (n=3338) OnabotA (n=2394)	DBPC Population (n=3338) Placebo (n=1044)
Overall	533 (16.7)	330 (13.8)	86 (8.2)
Headache	142 (4.5)	105 (4.4)	37 (3.5)
Eyelid ptosis	67 (2.1)	22 (0.9)	0 (0.0)
Eyelid sensory disorder	61 (1.9)	22 (0.9)	1 (0.1)
Injection site hematoma	36 (1.1)	36 (1.5)	8 (0.8)
Injection site pain	35 (1.1)	20 (0.8)	9 (0.9)
Injection site bruising	31 (1.0)	30 (1.3)	9 (0.9)
Eyelid edema	27 (0.8)	15 (0.6)	0 (0.0)
Injection site pruritus	22 (0.7)	5 (0.2)	4 (0.4)
Skin tightness	18 (0.6)	6 (0.3)	1 (0.1)
Injection site hemorrhage	16 (0.5)	11 (0.5)	2 (0.2)

DBPC, double-blind placebo-controlled; onabotA, onabotulinumtoxinA.

- For the CFL DBPC and CF/GL studies, the overall incidence of treatment-related AEs and the profile of the most common treatment-related AEs were similar (**Table 5**)

Table 5. Treatment-Related Adverse Events (≥2 subjects in any group)

Preferred Term, n (%)	CFL DBPC		CFL/GL DBPC	
	OnabotA (n=1324)	Placebo (n=808)	OnabotA (n=405)	Placebo (n=306)
Overall	116 (8.8)	48 (5.9)	53 (13.1)	23 (7.5)
Headache	27 (2.0)	17 (2.1)	23 (5.7)	11 (3.6)
Injection site hematoma	23 (1.7)	8 (1.0)	13 (3.2)	4 (1.3)
Injection site bruising	20 (1.5)	8 (1.0)	8 (2.0)	2 (0.7)
Injections site hemorrhage	7 (0.5)	2 (0.2)	4 (1.0)	2 (0.7)
Eyelid edema	5 (0.4)	0 (0.0)	1 (0.2)	0 (0.0)
Influenza-like illness	5 (0.4)	0 (0.0)	0 (0.0)	0 (0.0)
Injection site pain	3 (0.2)	5 (0.6)	2 (0.5)	2 (0.7)
Skin wrinkling	3 (0.2)	0 (0.0)	0 (0.0)	0 (0.0)
Eyelid ptosis	0 (0.0)	0 (0.0)	5 (1.2)	0 (0.0)
Eyelid sensory disorder	0 (0.0)	0 (0.0)	2 (0.5)	0 (0.0)
Injection site pruritus	1 (0.1)	0 (0.0)	2 (0.5)	0 (0.0)
Muscle twitching	2 (0.2)	1 (0.1)	0 (0.0)	1 (0.3)
Dizziness	2 (0.2)	0 (0.0)	0 (0.0)	0 (0.0)
Injection site reaction	2 (0.2)	0 (0.0)	0 (0.0)	0 (0.0)
Migraine	2 (0.2)	0 (0.0)	0 (0.0)	0 (0.0)
Nasopharyngitis	2 (0.2)	0 (0.0)	0 (0.0)	0 (0.0)
Tension headache	2 (0.2)	0 (0.0)	0 (0.0)	0 (0.0)

CFL, crow's feet lines; CFL/GL, crow's feet lines and glabellar lines combined; DBPC, double-blind placebo-controlled; onabotA, onabotulinumtoxinA.

Serious Adverse Events

- The incidence of serious AEs (SAEs) for the total onabotulinumtoxinA exposures was 1.8% (57/3189)
 - One SAE in a subject treated for CFL was deemed possibly related to treatment
 - SAEs that occurred in >1 subject (all, n=2) were myocardial infarction, herpes zoster, pneumonia, intervertebral disc protrusion, basal cell carcinoma, breast cancer, and invasive ductal breast carcinoma
- For the onabotulinumtoxinA and placebo groups, respectively, in all DBPC studies, the incidence of SAEs was 1.6% (39/2394) and 1.3% (14/1044)
- For the CFL DBPC studies, the overall incidence of SAEs was 1.4% (18/1324) for onabotulinumtoxinA and 1.5% (12/808) for placebo
- For the GL/CFL DBPC studies, the overall incidence of SAEs was 2.7% (11/405) for onabotulinumtoxinA and 2.3% (7/306) for placebo

Adverse Events by Treatment Cycle

- For all subjects who received onabotulinumtoxinA, the incidence of AEs decreased over repeated cycles (cycle 1: 37.7%; cycle 2: 30.9%; cycle 3: 30.8%; cycle 4: 24.4%; cycle 5: 15.5%)
- In CFL studies that included up to 5 onabotulinumtoxinA treatments over 13 months (12 U or 24 U) and CFL/GL studies that included up to 4 treatments over 12 months (32 U or 44 U), no onabotulinumtoxinA-treated subject (0/1717) developed neutralizing antibodies
- Possible manifestations of distant spread of toxin
 - An analysis of all 15 studies showed an overall PDSOT AE incidence of 3.4% (107/3189)
 - These PDSOT AEs were considered to be known local effects and not related to distant spread
 - PDSOT AEs included eyelid ptosis, blurred vision, accommodation disorder, diplopia, dry mouth, constipation, muscle weakness, facial paresis, urinary retention, and dyspnea
 - For DBPC studies, an analysis of AEs potentially related to distant spread of toxin (PDSOT) showed an overall incidence of 1.7% (41/2394) and 0.7% (7/1044) for onabotulinumtoxinA and placebo, respectively

CONCLUSIONS

- The current analysis expanded on the safety data previously reported for onabotulinumtoxinA for glabellar lines,[1] more than doubling the number of subjects and reporting on the additional indications of CFL and combined CFL/GL treatment
- The overall safety profile of onabotulinumtoxinA was unchanged, with a similar incidence and AE profile with CFL and CFL/GL treatment compared with GL treatment[1]
 - No new safety signals and no neutralizing antibodies were identified
- AEs were generally mild or moderate in severity, and the incidence decreased over repeated treatment cycles
- Analysis of AEs potentially related to distant spread of toxin found only known local effects, such as eyelid ptosis
- This updated safety meta-analysis of data from more than 3900 subjects confirmed the safety and tolerability of onabotulinumtoxinA for upper facial CFL and GL

REFERENCES

1. Brin MF, et al. *J Am Acad Dermatol.* 2009;61(6):961-70.
2. Carruthers JA, et al. *J Am Acad Dermatol.* 2002;46(6):840-9.
3. Carruthers JD, et al. *Plast Reconstr Surg.* 2003;112(4):1089-98.
4. Harii K, Kawashima M. *Aesthetic Plast Surg.* 2008;32(5):724-30.
5. Carruthers A, et al. *J Clin Res.* 2004;7:1-20.
6. Lowe NJ, et al. *Dermatol Surg.* 2005;31(3):257-62.
7. Kawashima M, *Harii K. Int J Dermatol.* 2009;48(7):768-76.
8. Moers-Carpi M, et al. *Dermatol Surg.* 2015;41(1):102-12.
9. Carruthers J, et al. *Dermatol Surg.* 2015;41(6):702-11.
10. Carruthers A, et al. *Dermatol Surg.* 2014;40(11):1181-90.
11. Huedo-Medina TB, et al. *Psychol Methods.* 2006;11(2):193-206.

ACKNOWLEDGMENTS

This analysis was sponsored by Allergan plc, Dublin, Ireland. Medical writing and editorial assistance was provided to the authors by Peloton Advantage, Parsippany, NJ, USA, and was funded by Allergan plc. All authors met the ICMJE authorship criteria. Neither honoraria nor other form of payments were made for authorship.

FINANCIAL DISCLOSURES

J Carruthers and A Carruthers serve as consultants and investigators for Allergan plc, Alphaeon, Merz, and Revance. K De Boulle serves as a consultant and investigator for Allergan plc. S Liew has no conflicts to disclose. A Rivkin serves as a consultant and investigator for Allergan plc and Merz Aesthetics USA. Y Wu has served as an investigator for Allergan plc. MF Brin, I Yushmanova, TI Boodhoo, K Zhuang, CJ Gallagher, and E Lee are employees of Allergan plc and may own stock/stock options in that company.

To obtain a PDF of this poster:
Scan the QR code
OR
Visit www.allergancongressposters.com/798356
Charges may apply. No personal information is stored.

Enter a valid e-mail to send this QR code or click to download.
Please ensure that qrcode@allergancongressposters.com has been added to your safe senders list or spam filter.

Presented at the TOXINS Meeting (International Neurotoxin Association), January 18-21, 2017, Madrid, Spain