Safety and Efficacy of OnabotulinumtoxinA in the Treatment of Crow's Feet Lines and Glabellar Lines in Japanese Subjects

Makoto Kawashima, MD[1]; Kiyonori Harii, MD[2]; Xiaofang Lei, PhD[3]; René Hopfinger, MS[3]

[1]Tokyo Women's Medical University School of Medicine, Tokyo, Japan; [2]Kyorin University School of Medicine, Tokyo, Japan; [3]Allergan plc, Irvine, CA, USA

Scan to obtain PDF of poster

ABSTRACT

Purpose: Evaluate the safety and efficacy of onabotulinumtoxinA treatment of moderate to severe crow's feet lines (CFL) in combination with glabellar lines (GL) in Japanese subjects.

Methods: This phase 3, multicenter, double-blind, randomized study included 2 treatment periods covering 13 months. Eligible subjects included Japanese adults ages 20–64 years with bilaterally symmetrical, moderate to severe CFL at maximum smile and moderate to severe GL at maximum frown, based on the investigator-assessed Facial Wrinkle Scale with Asian Photonumeric Guide (FWS-A; 0 = none, 1 = mild, 2 = moderate, 3 = severe). Exclusion criteria included previous or concurrent treatment with onabotulinumtoxinA. Subjects were randomized (1:1) to receive treatment with either onabotulinumtoxinA 44 U (CFL: 24 U, GL: 20 U) or onabotulinumtoxinA 32 U (CFL: 12 U, GL: 20 U). Subjects meeting re-treatment criteria, including the bilaterally symmetrical presence of CFL of at least moderate grade at maximum smile and GL of at least moderate grade at maximum frown per investigator-assessed FWS-A, could receive up to 5 treatments (initial + 1 additional treatment in period 1; 3 additional treatments in period 2). Efficacy endpoints included the proportion of responders, defined by subjects achieving a grade of none or mild at maximum smile for CFL and at maximum frown for GL at day 30 of period 1, based on the investigator-assessed FWS-A. Other efficacy endpoints assessed at day 30 of period 1 were the proportion of responders, based on various patient-reported outcomes, including Subject Assessment of Satisfaction with Appearance and the Facial Line Satisfaction Questionnaire–Overall Satisfaction item. Adverse events (AEs) were recorded.

Results: Of the 101 enrolled subjects (mean age [SD], 47.0 [8.3], 92.1% female), 92.1% completed the study. Most subjects received re-treatment with onabotulinumtoxinA. The majority of subjects in both treatment groups were responders (CFL: 89.6% [44 U] vs 84.9% [32 U]; GL: 93.8% [44 U] vs 98.1% [32 U]). The proportion of responders and patterns of responder rates over time during period 1 were similar to those during period 2. Most subjects receiving onabotulinumtoxinA were satisfied with their CFL appearance and with study drug on each patient-reported outcome measure (range, 71.7%–81.3%). Treatment-related AEs (10.6% [44 U], 11.3% [32 U]) were consistent with the known safety profile of onabotulinumtoxinA. Eyelid ptosis was the most common treatment-related AE (8.5% [4/47; 44 U], 3.8% [2/53; 32 U]). Except for 1 case of moderate eyelid ptosis in the 44 U group, all treatment-related AEs were mild in severity. No new safety signals were observed.

Conclusions: OnabotulinumtoxinA (44 U and 32 U doses) was effective in reducing the severity of CFL and GL over 13 months in Japanese subjects. Both doses were safe and well tolerated, with no new safety findings.

INTRODUCTION

- Simultaneous repeated treatment of multiple facial rhytids has become more frequent in clinical practice[1,2]
- OnabotulinumtoxinA (previously named botulinum toxin type A; Allergan plc, Dublin, Ireland) is approved for treatment of glabellar lines (GL) in more than 70 countries and for treatment of crow's feet lines (CFL) in more than 50 countries, including the United States and the European Union
- In a recent phase 3 study, onabotulinumtoxinA was shown to be safe and effective in the simultaneous treatment of rhytids in multiple areas of the face during a single treatment session[2]
- However, that study included predominantly Caucasian, non-Asian subjects who did not receive repeated treatments[2]
- In addition, the pivotal clinical trials of onabotulinumtoxinA in CFL included primarily Caucasian, non-Asian subjects[2,3]
- Assessment of CFL morphology in Asian subjects has shown a prevalence of different CFL patterns compared with those prevalent in Caucasian subjects[4,5]
 - This may be attributable to Asian skin having greater dermal thickness, collagen content, and melanin compared with Caucasian skin[6]

OBJECTIVE

- Evaluate the safety and efficacy of onabotulinumtoxinA for treatment of both CFL and GL in subjects with moderate to severe CFL and moderate to severe GL

METHODS

Subjects

- The study enrolled Japanese females and males aged 20 to 64 years who were botulinum toxin naive and had bilaterally symmetrical moderate to severe CFL at maximum smile on the investigator-assessed Facial Wrinkle Scale with Asian photonumeric guide (FWS-A)
- Key exclusion criteria included pregnancy, previous cosmetic treatments or surgical procedures at the treatment sites, eyebrow or eyelid ptosis, and eyelid hooding or other skin laxity unlikely to benefit from onabotulinumtoxinA treatment or likely to interfere with CFL assessments

Study Design and Treatments

- This phase 3, multicenter, double-blind, randomized, parallel group study was conducted at 6 sites in Japan from February 2013 through July 2014 (clinicaltrials.gov identifier: NCT01797094)
- The study comprised a 6-month treatment period (period 1) followed by a 7-month treatment period (period 2)
 - In total, the study duration for each subject was up to 13 months
- Eligible subjects were randomized 1:1 to receive either onabotulinumtoxinA 44 U or 32 U for both period 1 and period 2 (**Table 1**)
 - Investigators and subjects were blinded only to the dose of onabotulinumtoxinA, not to the treatment itself

Table 1. Treatment Groups

Treatment Group	Crow's Feet Lines	Glabellar Lines
OnabotulinumtoxinA 44 U	24 U (4 U per injection site × 6 sites)	20 U (4 U per injection site × 5 sites)
OnabotulinumtoxinA 32 U	12 U (2 U per injection site × 6 sites)	20 U (4 U per injection site × 5 sites)

- Treatment for CFL consisted of 6 intramuscular injections (0.1 mL each) of onabotulinumtoxinA administered bilaterally (3 on each side) to the orbicularis oculi muscle
 - Two injection patterns were available for treating CFL, depending on the investigator's assessment and on varying patterns/locations of the CFL[7]
- Treatment for GL consisted of 5 intramuscular injections (0.1 mL each) of onabotulinumtoxinA administered bilaterally (2 on each side) to the corrugator muscle and medially into the procerus muscle
- Subjects were permitted to receive up to 5 treatments in total, according to the randomly assigned dosage: 1 initial treatment (treatment 1) in period 1, an additional treatment (treatment 2) in period 1, and 3 additional treatments (treatments 3–5) in period 2
 - Subjects were eligible for re-treatment if they had bilaterally symmetrical CFL that were of at least moderate severity on each side of the face at maximum smile, based on the investigator-assessed FWS-A; at least 84 days since previous treatment; not at the day 390 visit; and were not pregnant
- Assessments were to be conducted at weeks 1 and 2 following each treatment, monthly from months 1 through 12, and at study exit (month 13 or upon early discontinuation)
- The duration of treatment/re-treatment intervals varied due to the differences between subjects in duration of response; this resulted in differences in time to eligibility for re-treatment. Accordingly, the study design specified treatments of differing potential durations (**Table 2**)

Table 2. Maximum Potential Duration of Treatment Intervals

Treatment	Duration (days)
1	≤390 (treatment 1 in period 1, on day 1)
2	≤300 (treatment 2 in period 1 or treatment 1 in period 2)
3	≤210 (treatment 1 or 2 in period 2)
4	≤120 (treatment 2 or 3 in period 2)
5	≤30 days (treatment 3 or 4 in period 2)

- The total duration of study participation was up to 13 months, regardless of the number of treatments received

Efficacy Measures

- Efficacy measures were not designated as primary or secondary; the primary time point was day 30 of treatment 1
- For treatment of CFL, the efficacy endpoint was the proportion of responders (defined as subjects achieving a grade of none or mild at maximum smile) as assessed by the investigator using the FWS-A, wherein 0 = none, 1 = mild, 2 = moderate, and 3 = severe
 - CFL severity was based on the average of scores on the left and right sides of the face
- For treatment of GL, the efficacy endpoint was the proportion of responders (defined as subjects achieving a grade of none or mild at maximum frown), based on the investigator's FWS-A assessment
- Other efficacy FWS-A measures included:
 - The proportion of responders (defined as subjects achieving ≥1-grade improvement in severity) at maximum smile and at rest for CFL, or at maximum frown and at rest for GL
 - Responders of ≥1-grade improvement at rest applied only to subjects with at least mild severity at baseline

Patient-Reported Outcomes

- Subject Assessment of Satisfaction with Appearance was a single-item measure wherein subjects rated satisfaction with the appearance of their CFL on a scale of 1 (very unsatisfied) to 5 (very satisfied)
 - Subjects who demonstrated improvement from neutral (3) or worse at baseline to satisfied (4) or very satisfied (5) on day 30 were considered to be responders
- The Facial Line Satisfaction Questionnaire (FLSQ) – Overall Satisfaction item was used only for CFL to assess each subject's satisfaction with treatment at that particular moment on a scale of -2 ("very dissatisfied") to +2 ("very satisfied")
 - Subjects who were "mostly satisfied" or "very satisfied" with treatment on day 30 were considered responders

Safety

- Safety measurements included adverse events (AEs), vital signs, physical examination, and neurologic assessments
- AEs and neurologic events from neurologic assessments were evaluated for possible spread of botulinum toxin

Statistical Analysis

- Efficacy analyses were performed for the intent-to-treat population (all enrolled subjects), and safety analyses were performed for the safety population (all subjects who received ≥1 study drug injection)
- For FWS-A and patient-reported outcome measures, comparison of the proportion of responders vs nonresponders was performed using the Cochran-Mantel-Haenszel tests stratified by baseline CFL severity at maximum smile

RESULTS

Subject Disposition and Demographics

- Of 102 subjects screened, 101 were enrolled and randomized (**Figure 1**)
- Most of the enrolled subjects completed the study: 92.1% (93/101) overall, 87.5% (42/48) in the 44 U group, and 96.2% (51/53) in the 32 U group
- Demographic and baseline characteristics were similar between the treatment groups (**Table 3**)
 - The majority of subjects were aged ≤50 years (68.3% [69/101]) and most were female (92.1% [93/101])

Table 3. Demographic and Baseline Characteristics

Parameter	OnabotulinumtoxinA 44 U (n=48)	OnabotulinumtoxinA 32 U (n=53)	Total (N=101)
Age, years			
Mean	48.4	45.8	47.0
SD	7.78	8.71	8.34
Median	49.0	44.0	47.0
Min, max	30, 64	31, 63	30, 64
≤50, n (%)	29 (60.4)	40 (75.5)	69 (68.3)
>50, n (%)	19 (39.6)	13 (24.5)	32 (31.7)
Gender			
Female, n (%)	45 (93.8)	48 (90.6)	93 (92.1)
Investigator's FWS-A assessment of CFL severity at maximum smile			
Moderate, n (%)	32 (66.7)	35 (66.0)	67 (66.3)
Severe, n (%)	16 (33.3)	18 (34.0)	34 (33.7)
Investigator's FWS-A assessment of GL at maximum frown			
Moderate	26 (54.2)	34 (64.2)	60 (59.4)
Severe	22 (45.8)	19 (35.8)	41 (40.6)
Subject assessment of satisfaction with appearance			
Mean	2.1	2.0	2.1
SD	0.67	0.66	0.67
Median	2.0	2.0	2.0
Min, max	1, 3	1, 3	1, 3
Neutral, n (%)	14 (29.2)	12 (22.6)	26 (25.7)
Unsatisfied, n (%)	26 (54.2)	30 (56.6)	56 (55.4)
Very unsatisfied, n (%)	8 (16.7)	11 (20.8)	19 (18.8)

CFL, crow's feet lines; FWS-A, Facial Wrinkle Scale with Asian photonumeric guide; GL, glabellar lines; max, maximum; min, minimum; SD, standard deviation.

Re-treatment

- The majority of subjects in both treatment groups were re-treated
- No subject received the maximum allowance of 5 treatments

Figure 1. Study Flow Diagram

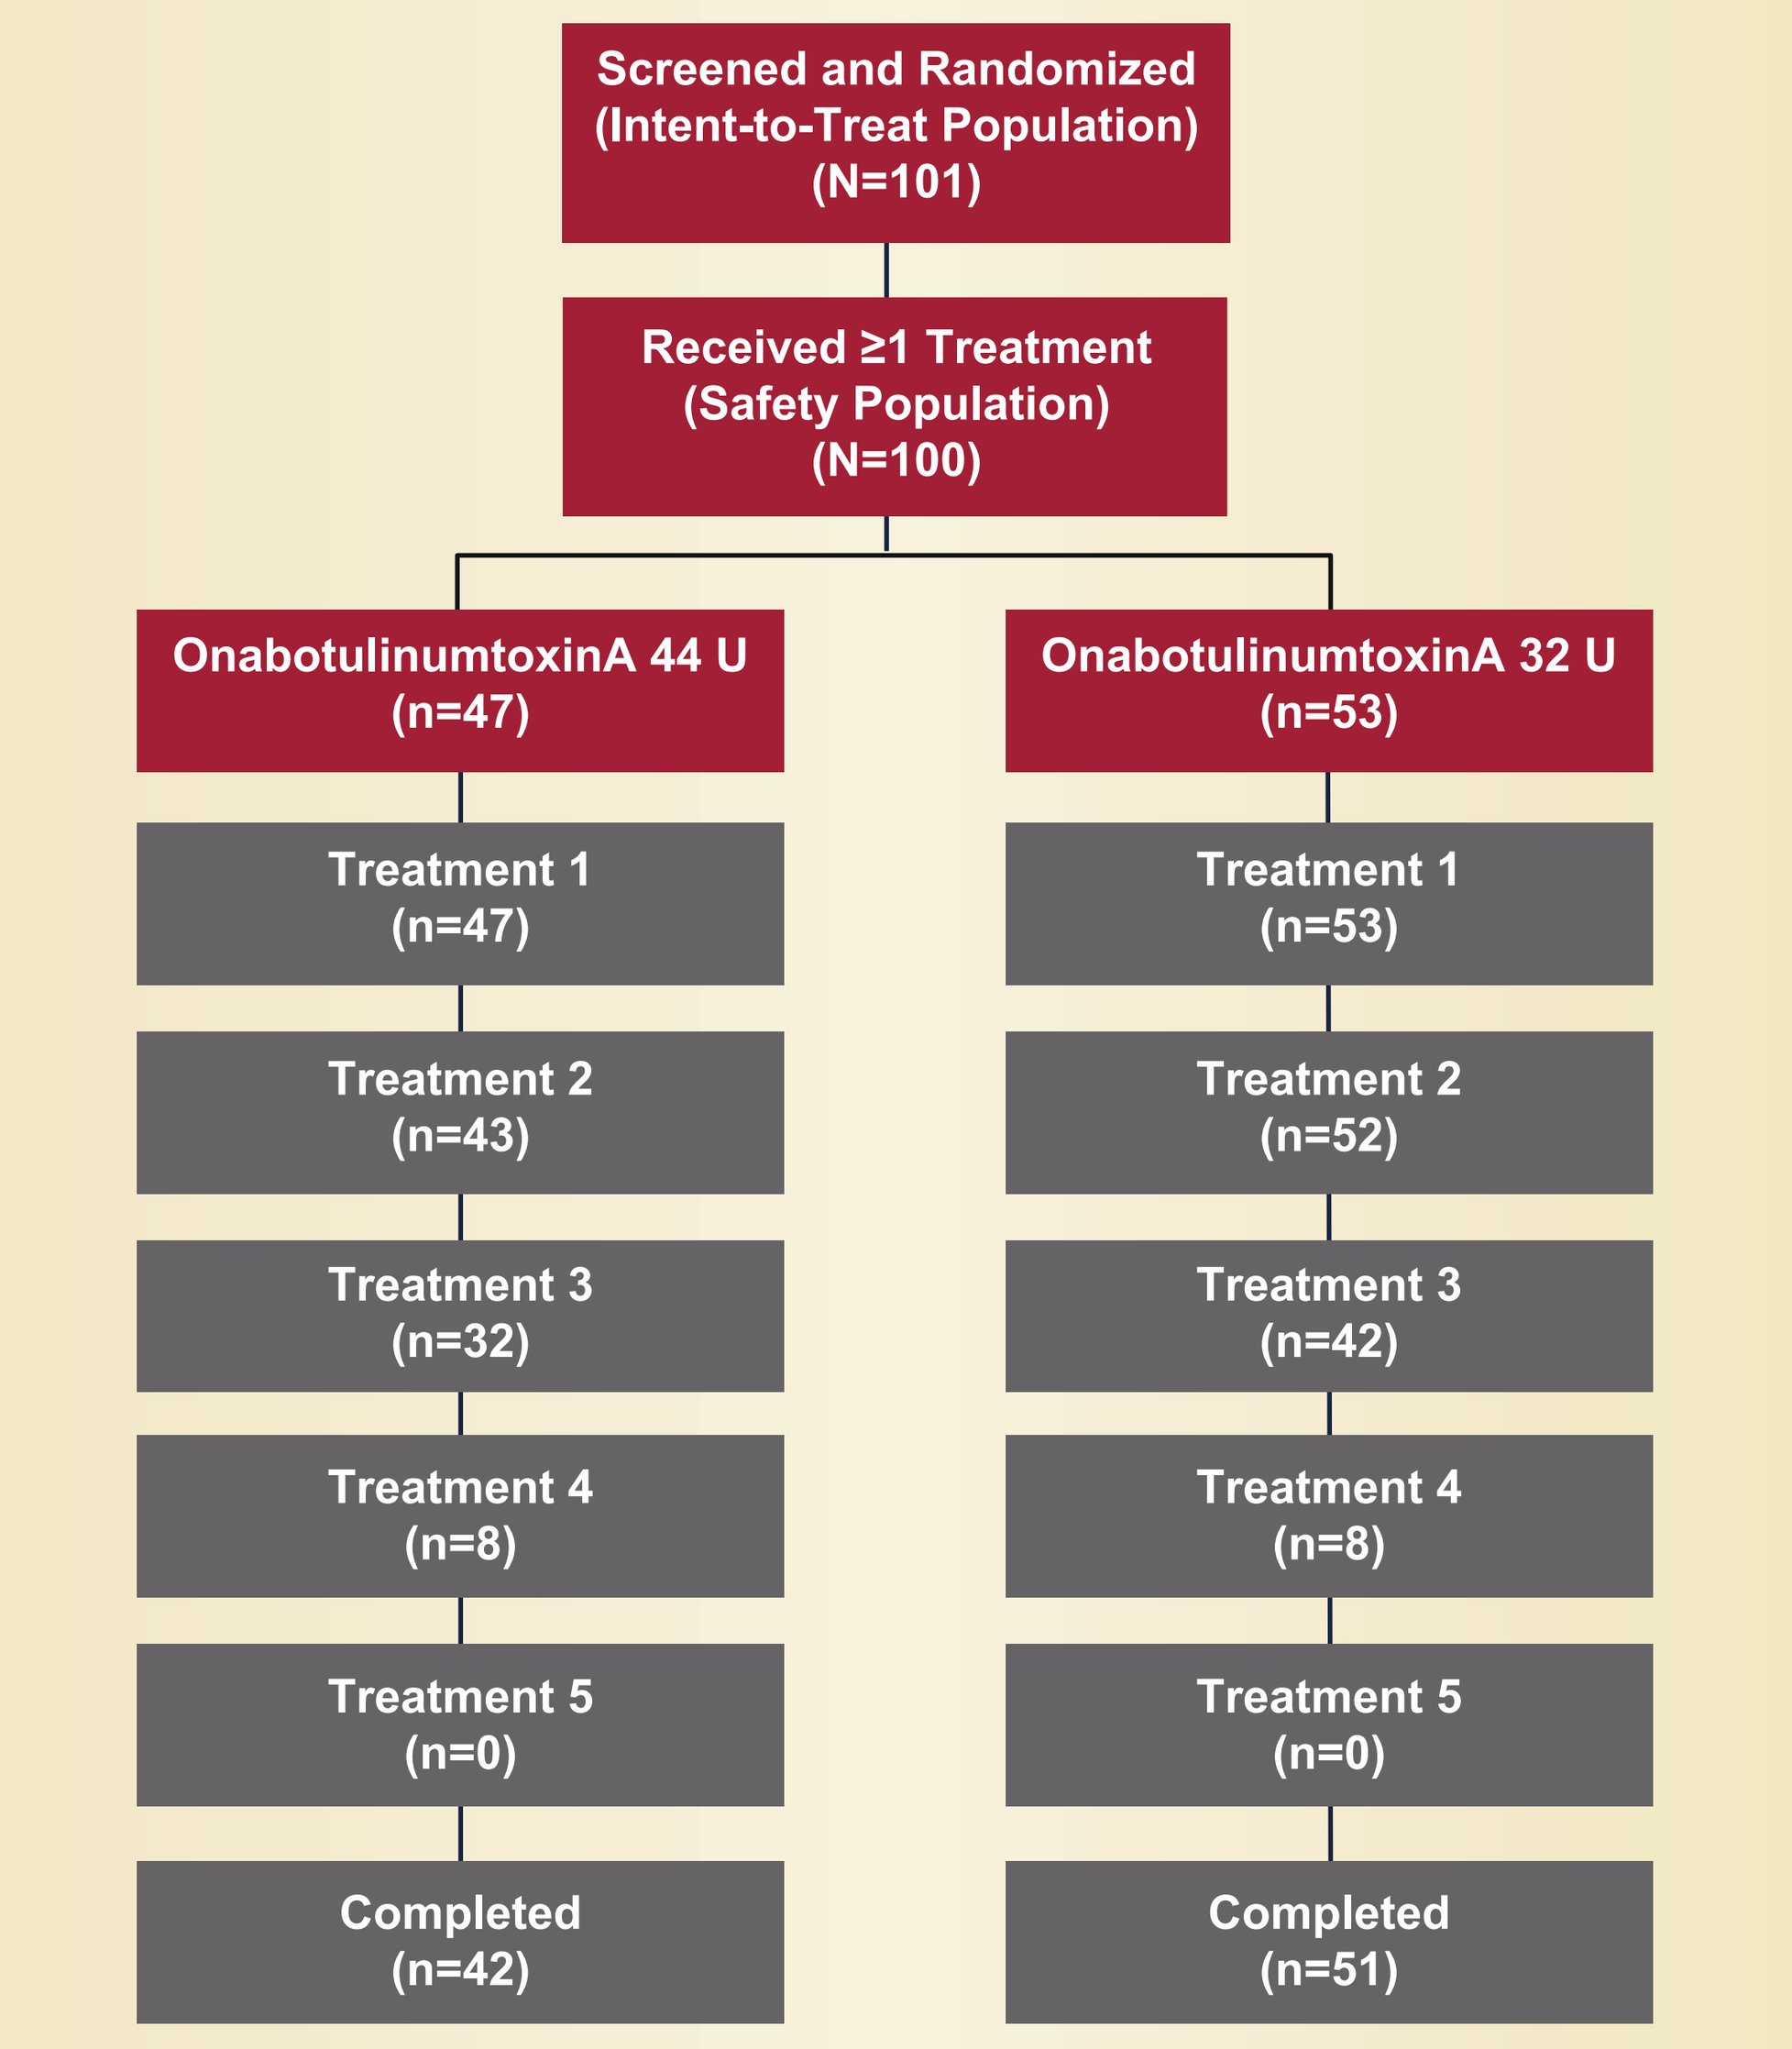
Screened and Randomized (Intent-to-Treat Population) (N=101)
Received ≥1 Treatment (Safety Population) (N=100)
OnabotulinumtoxinA 44 U (n=47)
OnabotulinumtoxinA 32 U (n=53)
Treatment 1 (n=47)
Treatment 1 (n=53)
Treatment 2 (n=43)
Treatment 2 (n=52)
Treatment 3 (n=32)
Treatment 3 (n=42)
Treatment 4 (n=8)
Treatment 4 (n=8)
Treatment 5 (n=0)
Treatment 5 (n=0)
Completed (n=42)
Completed (n=51)

Investigator-Assessed FWS-A Endpoints: Improvement to None or Mild in CFL Severity

- At day 30 of treatment 1, the majority of subjects achieved severity ratings for CFL of none or mild at maximum smile, with no significant differences between the two groups (89.6% [44 U] vs 84.9% [32 U]; P=0.453) (**Figure 2**)

Figure 2. Proportion of Subjects Achieving CFL Severity Ratings of None or Mild at Maximum Smile up to Day 90 of Each Treatment, Based on the FWS-A (Intent-to-Treat Population)

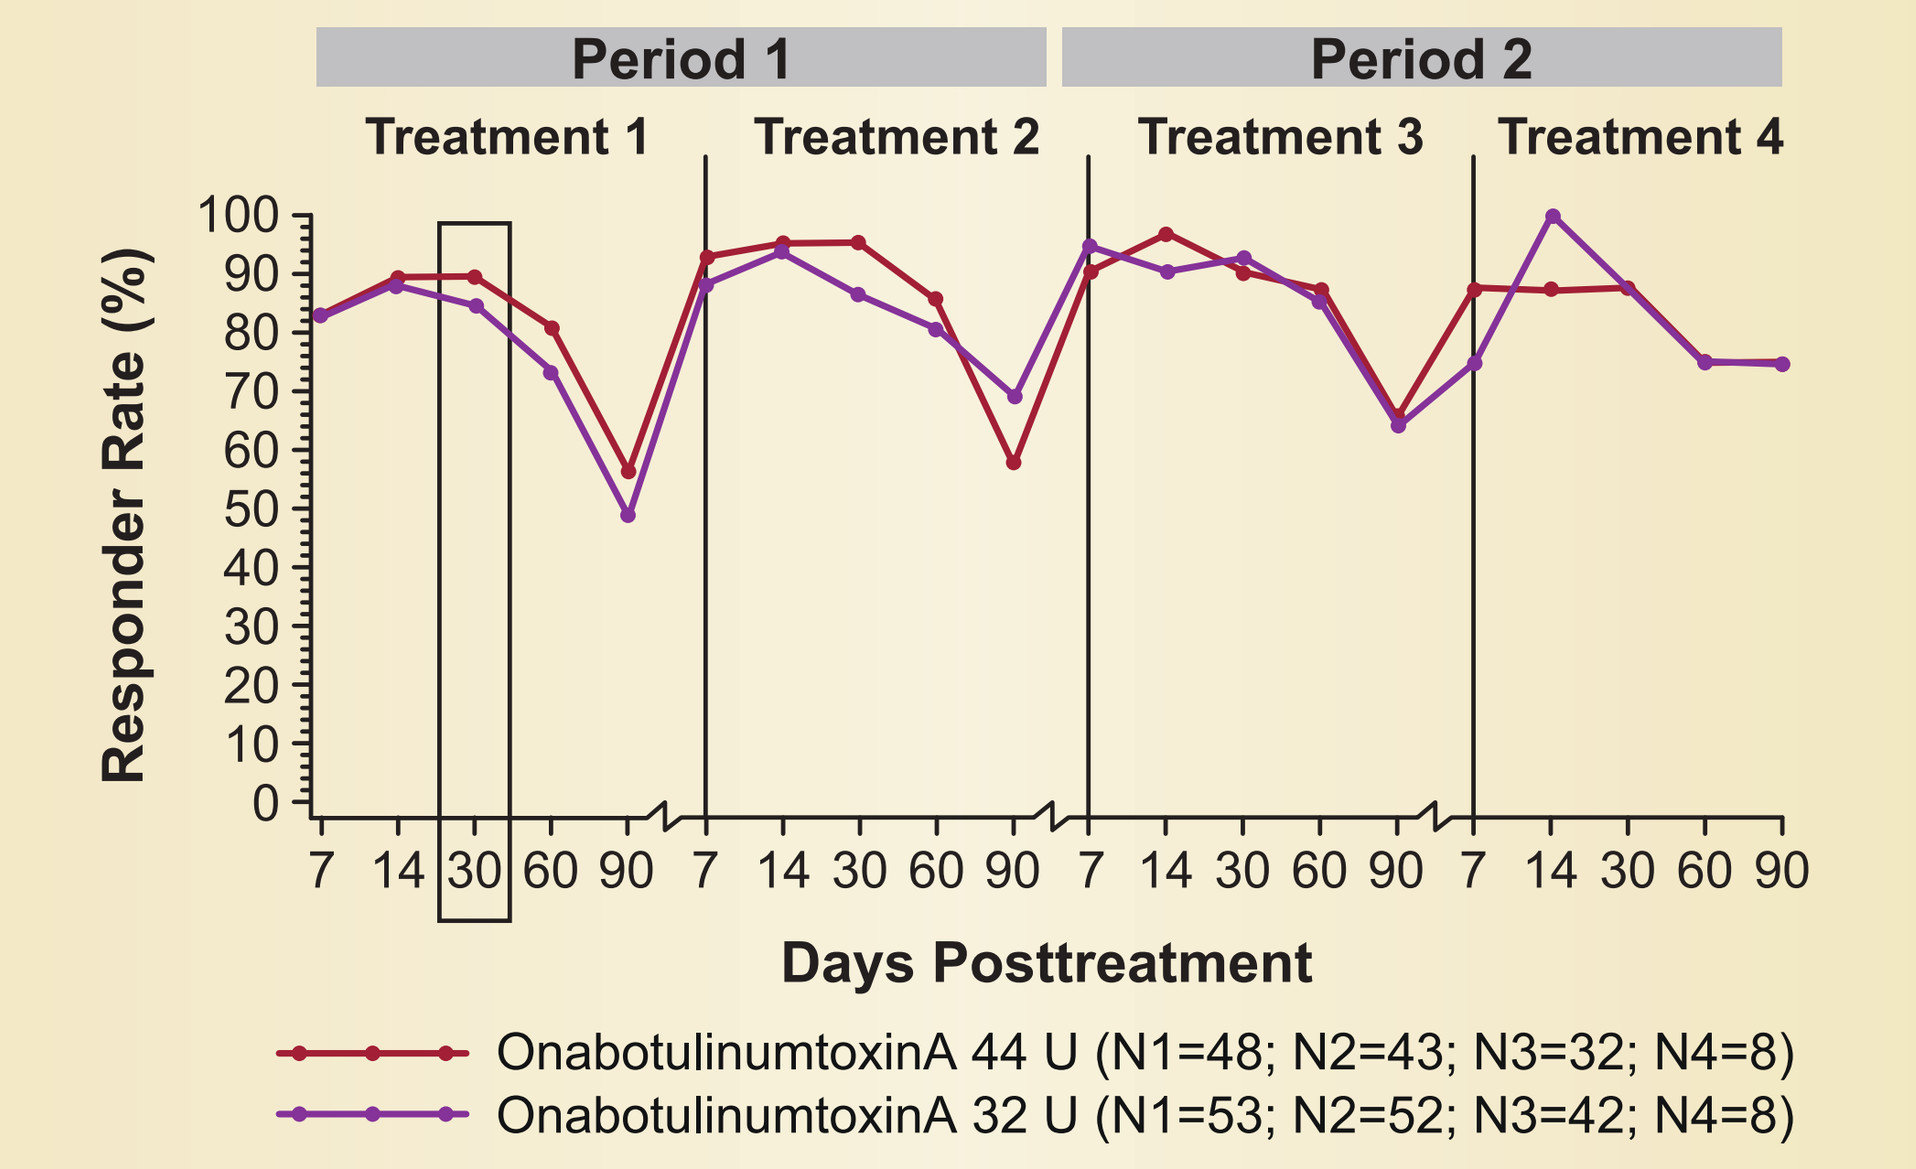
Period 1
Period 2
Treatment 1
Treatment 2
Treatment 3
Treatment 4
Responder Rate (%)
Days Posttreatment
OnabotulinumtoxinA 44 U (N1=48; N2=43; N3=32; N4=8)
OnabotulinumtoxinA 32 U (N1=53; N2=52; N3=42; N4=8)

N1–N4 indicate the number of subjects receiving treatments 1–4. No patient received 5 treatments.
CFL, crow's feet lines; FWS-A, Facial Wrinkle Scale with Asian photonumeric guide.

- High rates of none or mild responders for CFL severity were observed at most time points through day 90 of treatments 1–4 (range, 49.1%–100%), with no statistically significant differences between the groups

- At day 30 of treatment 1, the majority of subjects achieved severity ratings for GL of none or mild at maximum frown, with no significant differences between the groups (93.8% [44 U] vs 98.1% [32 U]; P=0.255) (**Figure 3**)
- A high proportion of none or mild responders for GL severity was observed at all time points through day 90 (range, 78.1%–100.0%), with no statistically significant differences between the groups

Figure 3. Proportion of Subjects Achieving GL Severity of None or Mild at Maximum Frown up to Day 90 of Each Treatment, Based on the FWS-A (Intent-to-Treat Population)

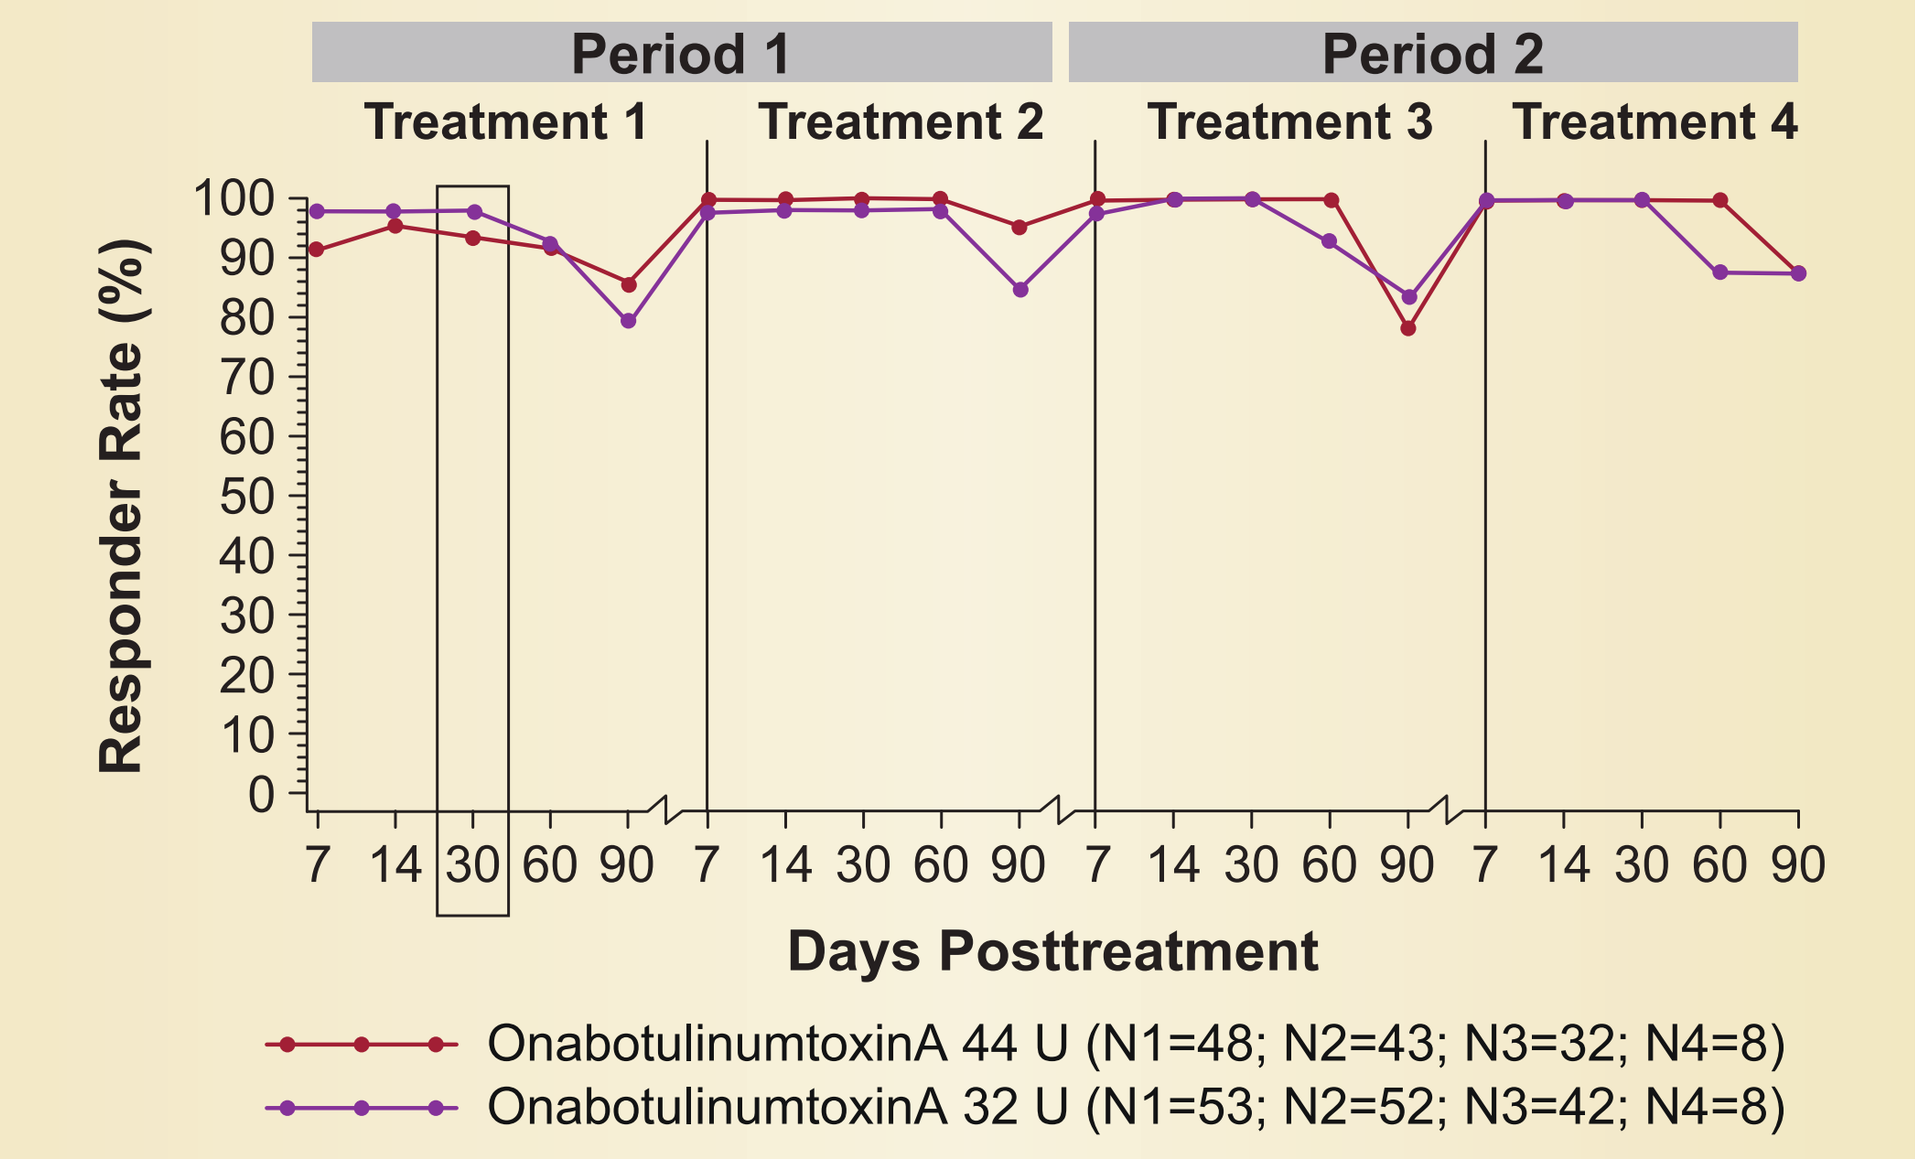
Period 1
Period 2
Treatment 1
Treatment 2
Treatment 3
Treatment 4
Responder Rate (%)
Days Posttreatment
OnabotulinumtoxinA 44 U (N1=48; N2=43; N3=32; N4=8)
OnabotulinumtoxinA 32 U (N1=53; N2=52; N3=42; N4=8)

N1–N4 indicate the number of subjects receiving treatments 1–4. No patient received 5 treatments.
FWS-A, Facial Wrinkle Scale with Asian photonumeric guide; GL, glabellar lines.

Investigator-Assessed FWS-A Endpoint: ≥1-Grade Improvement in CFL Severity

- The majority of subjects were responders, based on achieving ≥1-grade improvement as assessed with the FWS-A both at maximal contraction and at rest, with no significant differences between the groups (**Table 4**)

Table 4. Responders: Subjects Who Achieved ≥1-Grade Improvement From Baseline

Parameter	OnabotulinumtoxinA 44 U	OnabotulinumtoxinA 32 U	P value
≥1 Grade improvement from baseline in CFL severity (responders), % (n/N)			
Maximum smile	95.8 (46/48)	98.1 (52/53)	0.497
Rest	70.0 (28/40)	75.0 (33/44)	0.624
≥1 Grade improvement from baseline in GL severity (responders), n/N (%)			
Maximum frown	97.9 (47/48)	100.0 (53/53)	0.289
Rest	75.0 (24/32)	25/30 (83.3)	0.398

CFL, crow's feet lines; GL, glabellar lines.

Patient-Reported Outcomes Related to Satisfaction

- At day 30 of treatment 1, the majority of subjects in both groups were responders based on the Subject Assessment of Satisfaction with Appearance of CFL (81.3% [39/48] in the 44 U group and 71.7% [38/53] in the 32 U group; P=0.266)
 - No significant difference was observed between onabotulinumtoxinA 44 U and 32 U at most time points
- At day 30 of treatment 1, most subjects in both groups were responders, based on the FLSQ-Overall Satisfaction item (81.3% [39/48] in the 44 U group; 81.1% [43/53] in the 32 U group; P=0.990)
 - No significant difference was observed between the onabotulinumtoxinA 44 U and 32 U groups at any time point

Safety

- Treatment-emergent AEs (TEAEs) occurred in 59% (59/100) of subjects: 61.7% (29/47) in the 44 U group and 56.6% (30/53) in the 32 U group
 - The only injection site TEAE was hemorrhage (19.1% [9/47] in the 44 U group; 11.3% [6/53] in the 32 U group)
 - Treatment-related TEAEs occurred in 11.0% of subjects overall and were similar between treatment groups over the entire study period (**Table 5**)
 - Eyelid ptosis was the most commonly reported treatment-related AE in subjects in both the 44 U and 32 U groups

Table 5. Treatment-Related AEs (Safety Population)

Parameter	OnabotulinumtoxinA 44 U (n=47)	OnabotulinumtoxinA 32 U (n=53)	Total (N=100)
Overall	5 (10.6)	6 (11.3)	11 (11.0)
Eye disorders			
Eyelid ptosis	4 (8.5)	2 (3.8)	6 (6.0)
Eyelid sensory disorder	1 (2.1)	1 (1.9)	2 (2.0)
Eyelid edema	1 (2.1)	0 (0.0)	1 (1.0)
Vision blurred	0 (0.0)	1 (1.9)	1 (1.0)
General disorders and administration site conditions			
Malaise	1 (2.1)	0 (0.0)	1 (1.0)
Musculoskeletal and connective tissue disorders			
Muscle spasms	0 (0.0)	1 (1.9)	1 (1.0)
Nervous system disorders			
Headache	0 (0.0)	2 (3.8)	2 (2.0)

AEs, adverse events.

- All but one treatment-related AE occurred over the 90 days following treatment 1 and were mild in severity; 1 event of eyelid edema in the 44 U group occurred during treatment 2; no treatment-related AEs occurred following treatments 3 or 4
- Six serious TEAEs occurred in 5 subjects following treatments 1 or 2; none were considered related to treatment
 - Serious TEAEs were hepatitis A, Bartholin's cyst, supraventricular tachycardia, benign female reproductive tract neoplasm, and appendicitis and peritonitis in 1 subject
 - Except for 1 event of severe peritonitis during treatment 1, all serious AEs were considered mild or moderate in severity
 - No subject discontinued because of a serious AE
- Two study discontinuations were attributed to AEs: eyelid ptosis (in the 44 U group) that was considered to be related to treatment and collagen-vascular disease (in the 32 U group) that was not considered to be treatment related
- Overall, the tolerability profile of onabotulinumtoxinA improved with repeated treatments
- No clinically meaningful changes from baseline were found in vital signs or physical examinations
- No AE was associated with the distant spread of toxin
- No new safety signals emerged in subjects who received additional treatments over 13 months

CONCLUSIONS

- OnabotulinumtoxinA administered to CFL and GL in Japanese subjects at total doses of 44 U or 32 U was effective in reducing the severity of CFL and GL, based on investigator and subject assessments, for up to 13 months
 - Improvements in both CFL and GL were consistent over multiple onabotulinumtoxinA treatments
 - Improvements in both CFL and GL were seen at maximal contraction and at rest
- Repeated treatment with onabotulinumtoxinA was well tolerated and resulted in no cumulative deleterious effects
- The safety profile observed in this study population was consistent with the known safety profile of onabotulinumtoxinA
- OnabotulinumtoxinA was safe and effective across multiple assessments and multiple treatments, resulting in consistent positive treatment effects and meaningful improvement in the appearance of CFL and GL, according to investigators and subjects

REFERENCES

1. Carruthers A, Carruthers J. *J Am Acad Dermatol*. 2009;60:972-9.
2. Moers-Carpi M, et al. *Dermatol Surg*. 2015;41:102-12.
3. Carruthers A, et al. *Dermatol Surg*. 2014;40:1181-90.
4. Park DH, et al. *Aesthetic Plast Surg*. 2012;36:1211-5.
5. Kane MAC, et al. *Dermatol Surg*. 2015;41:447-56.
6. Kim MM, Byrne PJ. *Facial Plast Surg Clin North Am*. 2007;15:381-6, vii.
7. Botox Cosmetic [package insert]. Dublin, Ireland: Allergan plc; 2015.

ACKNOWLEDGMENTS

This study was sponsored by Allergan plc, Dublin, Ireland. Medical writing and editorial assistance was provided to the authors by Peloton Advantage, Parsippany, NJ, USA, and was funded by Allergan plc. All authors met the ICMJE authorship criteria. Neither honoraria nor other form of payments were made for authorship.

FINANCIAL DISCLOSURES

K Harii and M Kawashima have no conflicts to disclose. X Lei and R Hopfinger are employees of Allergan plc.

To obtain a PDF of this poster:
Scan the QR code
OR
Visit www.allergancongressposters.com/689681
Charges may apply. No personal information is stored.

Enter a valid e-mail to send this QR code or click to download.
Please ensure that qrcode@allergancongressposters.com has been added to your safe senders list or spam filter.

Presented at the TOXINS Meeting (International Neurotoxin Association), January 18-21, 2017, Madrid, Spain