Safety and Efficacy of OnabotulinumtoxinA for the Treatment of Crow's Feet Lines in Chinese Subjects

Yan Wu, MD, PhD[1]; Wang Gang, MD[2]; Li Chengxin, MD[3]; Cheri Mao, MS[4]; Xiaofang Lei, PhD[4]; Elisabeth Lee, MPH[4]

[1]Peking University First Hospital, Beijing, China; [2]The First Affiliated Hospital, The Fourth Military Medical University, Shaanxi Province, China; [3]Chinese PLA General Hospital, Beijing, China; [4]Allergan plc, Irvine, CA, USA

Scan to obtain PDF of poster

ABSTRACT

Introduction: This study evaluated the safety and efficacy of onabotulinumtoxinA for moderate to severe lateral canthal lines or crow's feet lines (CFL) in Chinese subjects.

Methods: This 5-month, multicenter, double-blind, randomized, placebo-controlled study was conducted in China. Subjects with moderate to severe CFL at maximum smile were randomized to a single treatment cycle of onabotulinumtoxinA 24 U (total) or placebo (6 injections, 3 in each CFL area). Primary efficacy variable was the percentage of subjects achieving a severity rating of none or mild for CFL at maximum smile, as assessed by the investigator at day 30 on the Facial Wrinkle Scale with Asian Photonumeric Guide (4-point scale: 0=none, 1=mild, 2=moderate, 3=severe).

Results: Of 417 subjects who received treatment (onabotulinumtoxinA, n=316; placebo, n=101), 98.3% completed the study. Mean [range] age was 46.4 [23–74] years; 86.3% were female. The primary endpoint was met: the proportion of subjects achieving none or mild CFL severity at maximum smile (responders) at day 30 was significantly (*P*<0.001) greater with onabotulinumtoxinA (63.9%) vs placebo (5.0%). At all other time points, the responder rate was significantly (*P*<0.001) greater with onabotulinumtoxinA (day 60, 63.9%; day 90, 59.5%; day 120, 48.7%; day 150, 30.7%) vs placebo (≤5% at all time points). At day 30, subject-perceived age was, on average, 2.7 years younger than baseline in the onabotulinumtoxinA group vs 0.5 years younger in the placebo group (*P*<0.001). Median duration of effect with onabotulinumtoxinA was ≥5 months using all responder definitions. There was a low occurrence of treatment-related adverse events.

Conclusions: Treatment with onabotulinumtoxinA 24 U was effective in reducing CFL severity in Chinese subjects, with responses maintained over 5 months. OnabotulinumtoxinA was safe and well tolerated, with no new safety findings.

INTRODUCTION

- The use of onabotulinumtoxinA (Botox; Allergan plc, Dublin, Ireland) in minimally invasive aesthetic procedures has become common worldwide[1]
 - OnabotulinumtoxinA is approved for the treatment of glabellar lines in more than 70 countries and for crow's feet lines (CFL) in more than 50 countries
- Most subjects in the pivotal clinical trials of onabotulinumtoxinA in CFL were non-Asian[2,3]
- CFL morphology in Asian subjects has shown different CFL pattern prevalence compared with Caucasian subjects[4,5]
- Harii and colleagues were the first to use the validated Facial Wrinkle Scale with Asian Photonumeric Guide (FWS-A) to assess the efficacy of onabotulinumtoxinA treatment of CFL in Japanese subjects[6]

OBJECTIVE

- Evaluate the safety and efficacy of onabotulinumtoxinA for moderate to severe CFL in Chinese subjects using the FWS-A

METHODS

Subjects

- This study enrolled Chinese females or males aged ≥18 years with moderate to severe bilateral CFL at maximum smile (same grade on both sides) as assessed by both the investigator and the subject using the FWS-A
- Key exclusion criteria: concurrent treatment with botulinum toxin or treatment within 1 year prior to enrollment; known immunization or hypersensitivity to botulinum toxin; any medical condition that may result in increased subject risk; pregnancy; previous cosmetic treatments or surgical procedures at the treatment sites; eyebrow or eyelid ptosis; and eyelid hooding or other skin laxity unlikely to benefit from onabotulinumtoxinA treatment or likely to interfere with CFL assessments

Study Design and Treatment

- This 5-month, multicenter, double-blind, randomized, parallel-group, placebo-controlled study was conducted at 9 sites in China from September 2014 through June 2015 (clinicaltrials.gov identifier NCT02195687)
- Subjects were randomized in a 3:1 ratio to a single treatment of onabotulinumtoxinA 24 U or placebo on day 1; study design is shown in **Figure 1**
- Investigators and subjects were blinded to the treatment administered
- Treatment consisted of 6 injections of 0.1 mL each (3 injections per side)
- Treatment pattern A (**Figure 2A**)
 - First injection (A) into orbicularis oculi at the level of the lateral canthus, ≥1.5 to 2.0 cm temporal to the lateral canthus and just temporal to the lateral orbital rim
 - Second injection (B): 1.0 to 1.5 cm above first injection site at ≈30° angle medially
 - Third injection (C): 1.0 to 1.5 cm below first injection at ≈30° angle medially
 - Repeated for contralateral side
- Treatment pattern B (**Figure 2B**)
 - If CFL were primarily below the lateral canthus, injections were given in a line that angled from anteroinferior to superposterior

Figure 1. Study Design

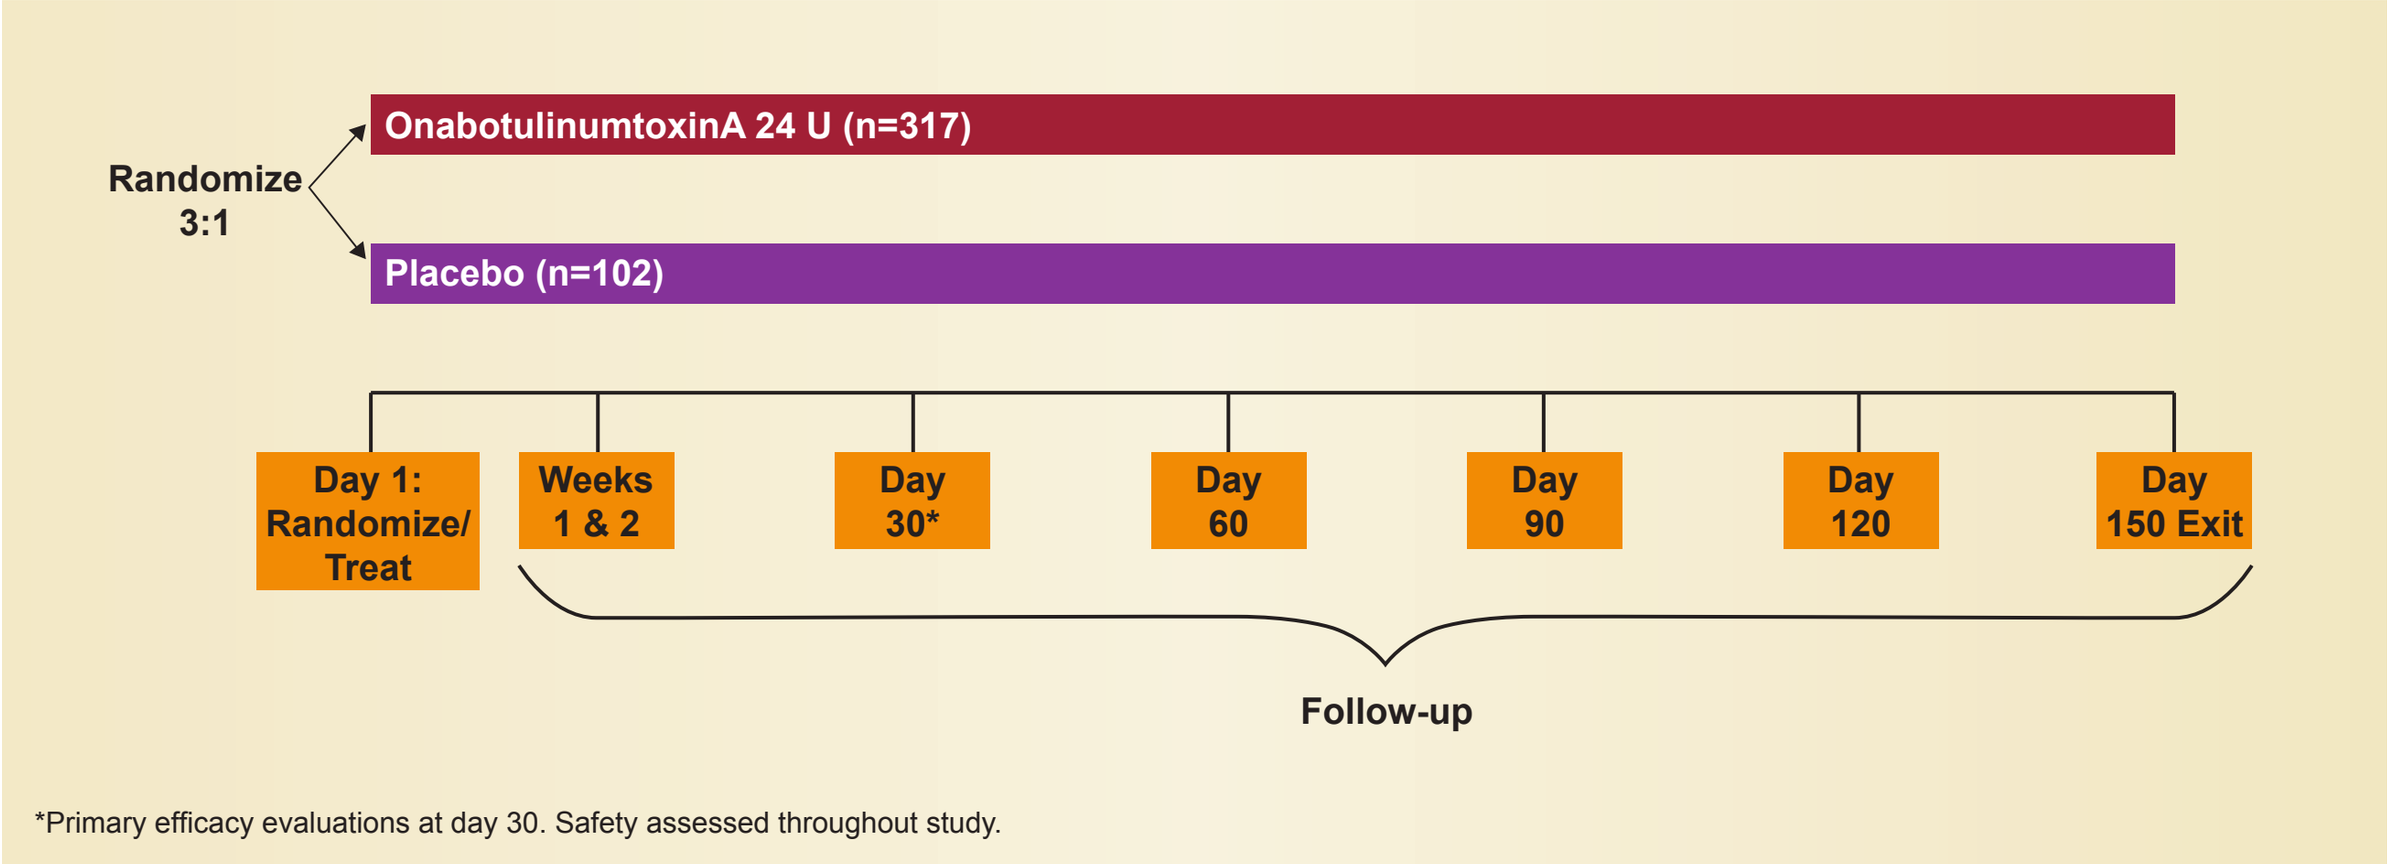

Randomize 3:1
OnabotulinumtoxinA 24 U (n=317)
Placebo (n=102)
Day 1: Randomize/ Treat
Weeks 1 & 2
Day 30*
Day 60
Day 90
Day 120
Day 150 Exit
Follow-up

*Primary efficacy evaluations at day 30. Safety assessed throughout study.

Figure 2. Injection Pattern (A) and Allowed Modification (B) for the Treatment of Crow's Feet Lines

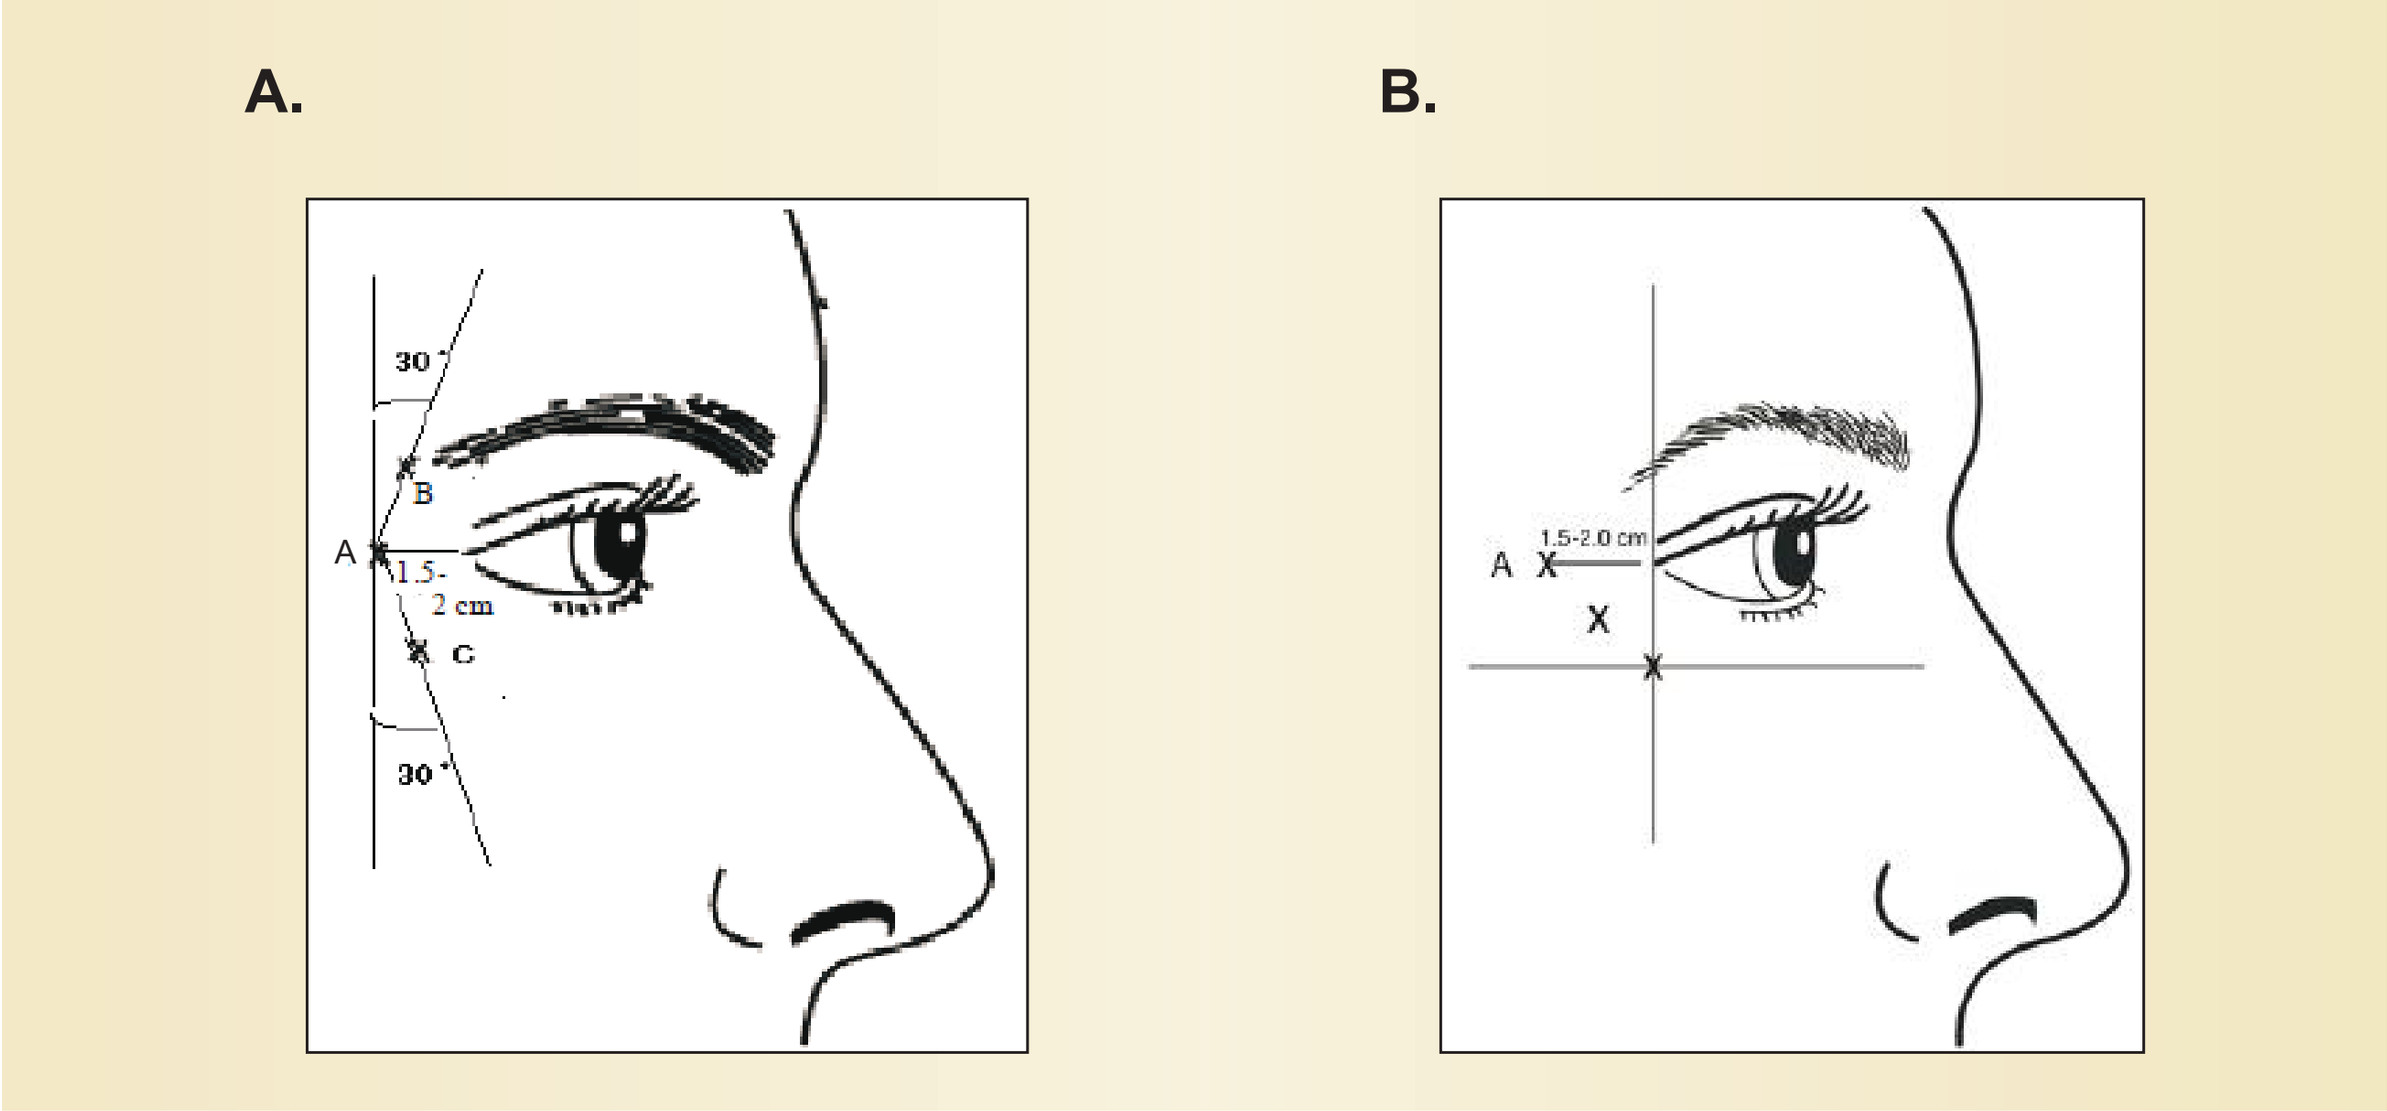

A.
B.
30°
B
A
1.5-2 cm
C
30°
A
1.5-2.0 cm
X

Efficacy Assessments

- The primary efficacy assessment was the percentage of FWS-A responders at day 30, defined as subjects achieving an FWS-A severity rating of none or mild for CFL at maximum smile as assessed by the investigator (FWS-A 4-grade scale: 0=none, 1=mild, 2=moderate, 3=severe)
- Non-primary efficacy assessments included:
 - Percentage of responders with ≥1-grade improvement on the FWS-A at maximum smile and at rest, as assessed by the investigator at day 30
 - Subject's Self-Perception of Age (SPA) responders, defined as subjects who rated themselves as "looking their current age or older" at baseline then rated themselves as being in a younger age category at day 30
 - The SPA is a single-item questionnaire asking: Do I look my current age, look older than my current age (by how many years?), or look younger than my current age (by how many years?)
 - Duration of effect, defined as the number of days to return to non-responder status (calculated for day 30 responders)
 - Subject Assessment of Satisfaction with Appearance responders, defined as subjects who reporting being "very satisfied" or "satisfied" with CFL appearance at day 30
 - Facial Line Satisfaction Questionnaire (FLSQ) Item 5 responders, defined as subjects who reported being "very satisfied" or "mostly satisfied" with treatment effect at day 30
 - Subject's Global Assessment of Change in CFL (SGA-CFL) responders, defined as subjects who rated themselves as "very much improved" or "much improved" in the appearance of their CFL at day 30

Safety

- Safety assessments included monitoring adverse events (AEs), neurologic assessments, vital signs, and urine pregnancy tests

Statistical Analyses

- All efficacy analyses were performed on the modified intent-to-treat (mITT) population, defined as all randomized subjects who received ≥1 injection of study drug
- Safety analyses were performed on the safety population, defined as all randomized subjects who received ≥1 injection of study drug and were analyzed by actual treatment regardless of randomization
- For all responder analyses, between-group *P* values were based on Cochran-Mantel-Haenszel tests stratified by baseline CFL severity at maximum smile using the investigator-rated FWS-A
- Kaplan-Meier survival method was calculated for the duration of effect analysis

RESULTS

Subjects

- Of 419 randomized subjects, 417 were included in the mITT population (n=316 onabotulinumtoxinA; n=101 placebo)
- Most subjects completed the study: onabotulinumtoxinA, 311 (98.4%); placebo, 99 (98.0%)
- Reasons for discontinuation in both groups were limited to "lost to follow-up" and "personal reasons"
- Overall, 360 subjects were female (86.3%) and the median age (range) was 47.0 (23–74) years
- Baseline FWS-A CFL severity at maximum smile was assessed by the investigator as moderate in 202 (48.4%) subjects and severe in 215 (51.6%) subjects

Efficacy

- Proportion of responders with FWS-A CFL severity of "none" or "mild" was significantly greater with onabotulinumtoxinA vs placebo at day 30 and all other time points (**Figure 3**)

Figure 3. Primary Efficacy Variable: Proportion of Responders* at Day 30 (per Investigator)

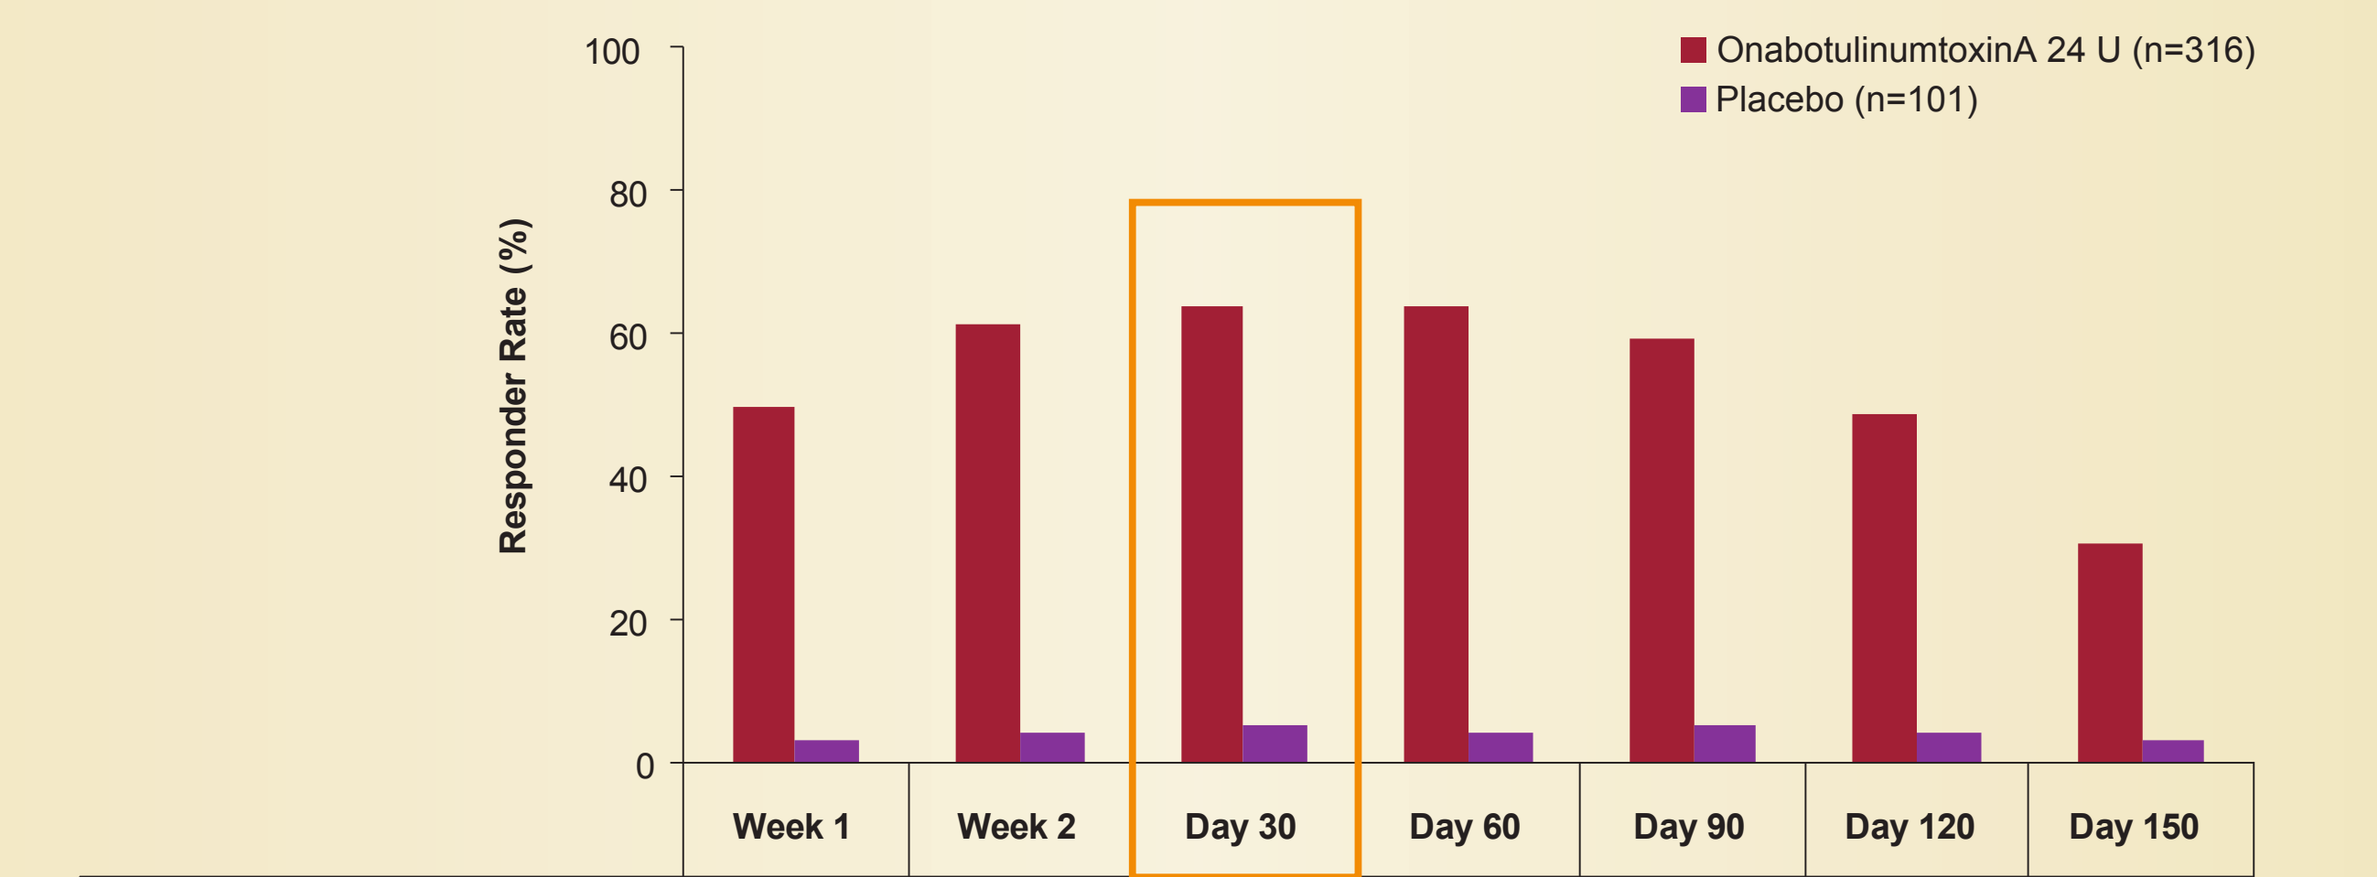

Responder Rate (%)
100
80
60
40
20
0
OnabotulinumtoxinA 24 U (n=316)
Placebo (n=101)

	Week 1	Week 2	Day 30	Day 60	Day 90	Day 120	Day 150
OnabotulinumtoxinA 24 U (n=316)	50.0	61.1	63.9	63.9	59.5	48.7	30.7
Placebo (n=101)	3.0	4.0	5.0	4.0	5.0	4.0	3.0

P<0.001, onabotulinumtoxinA vs placebo, all time points.

*Responder: subject achieving severity rating of none or mild at maximum smile on the Facial Wrinkle Scale with Asian Photonumeric Guide.

- The proportion of responders with ≥1-grade FWS-A improvement either at maximum smile or at rest was significantly greater in the onabotulinumtoxinA group than the placebo group at day 30 and all other time points (**Figure 4**)

Figure 4. Proportion of Responders With ≥1-Grade Improvement on the FWS-A

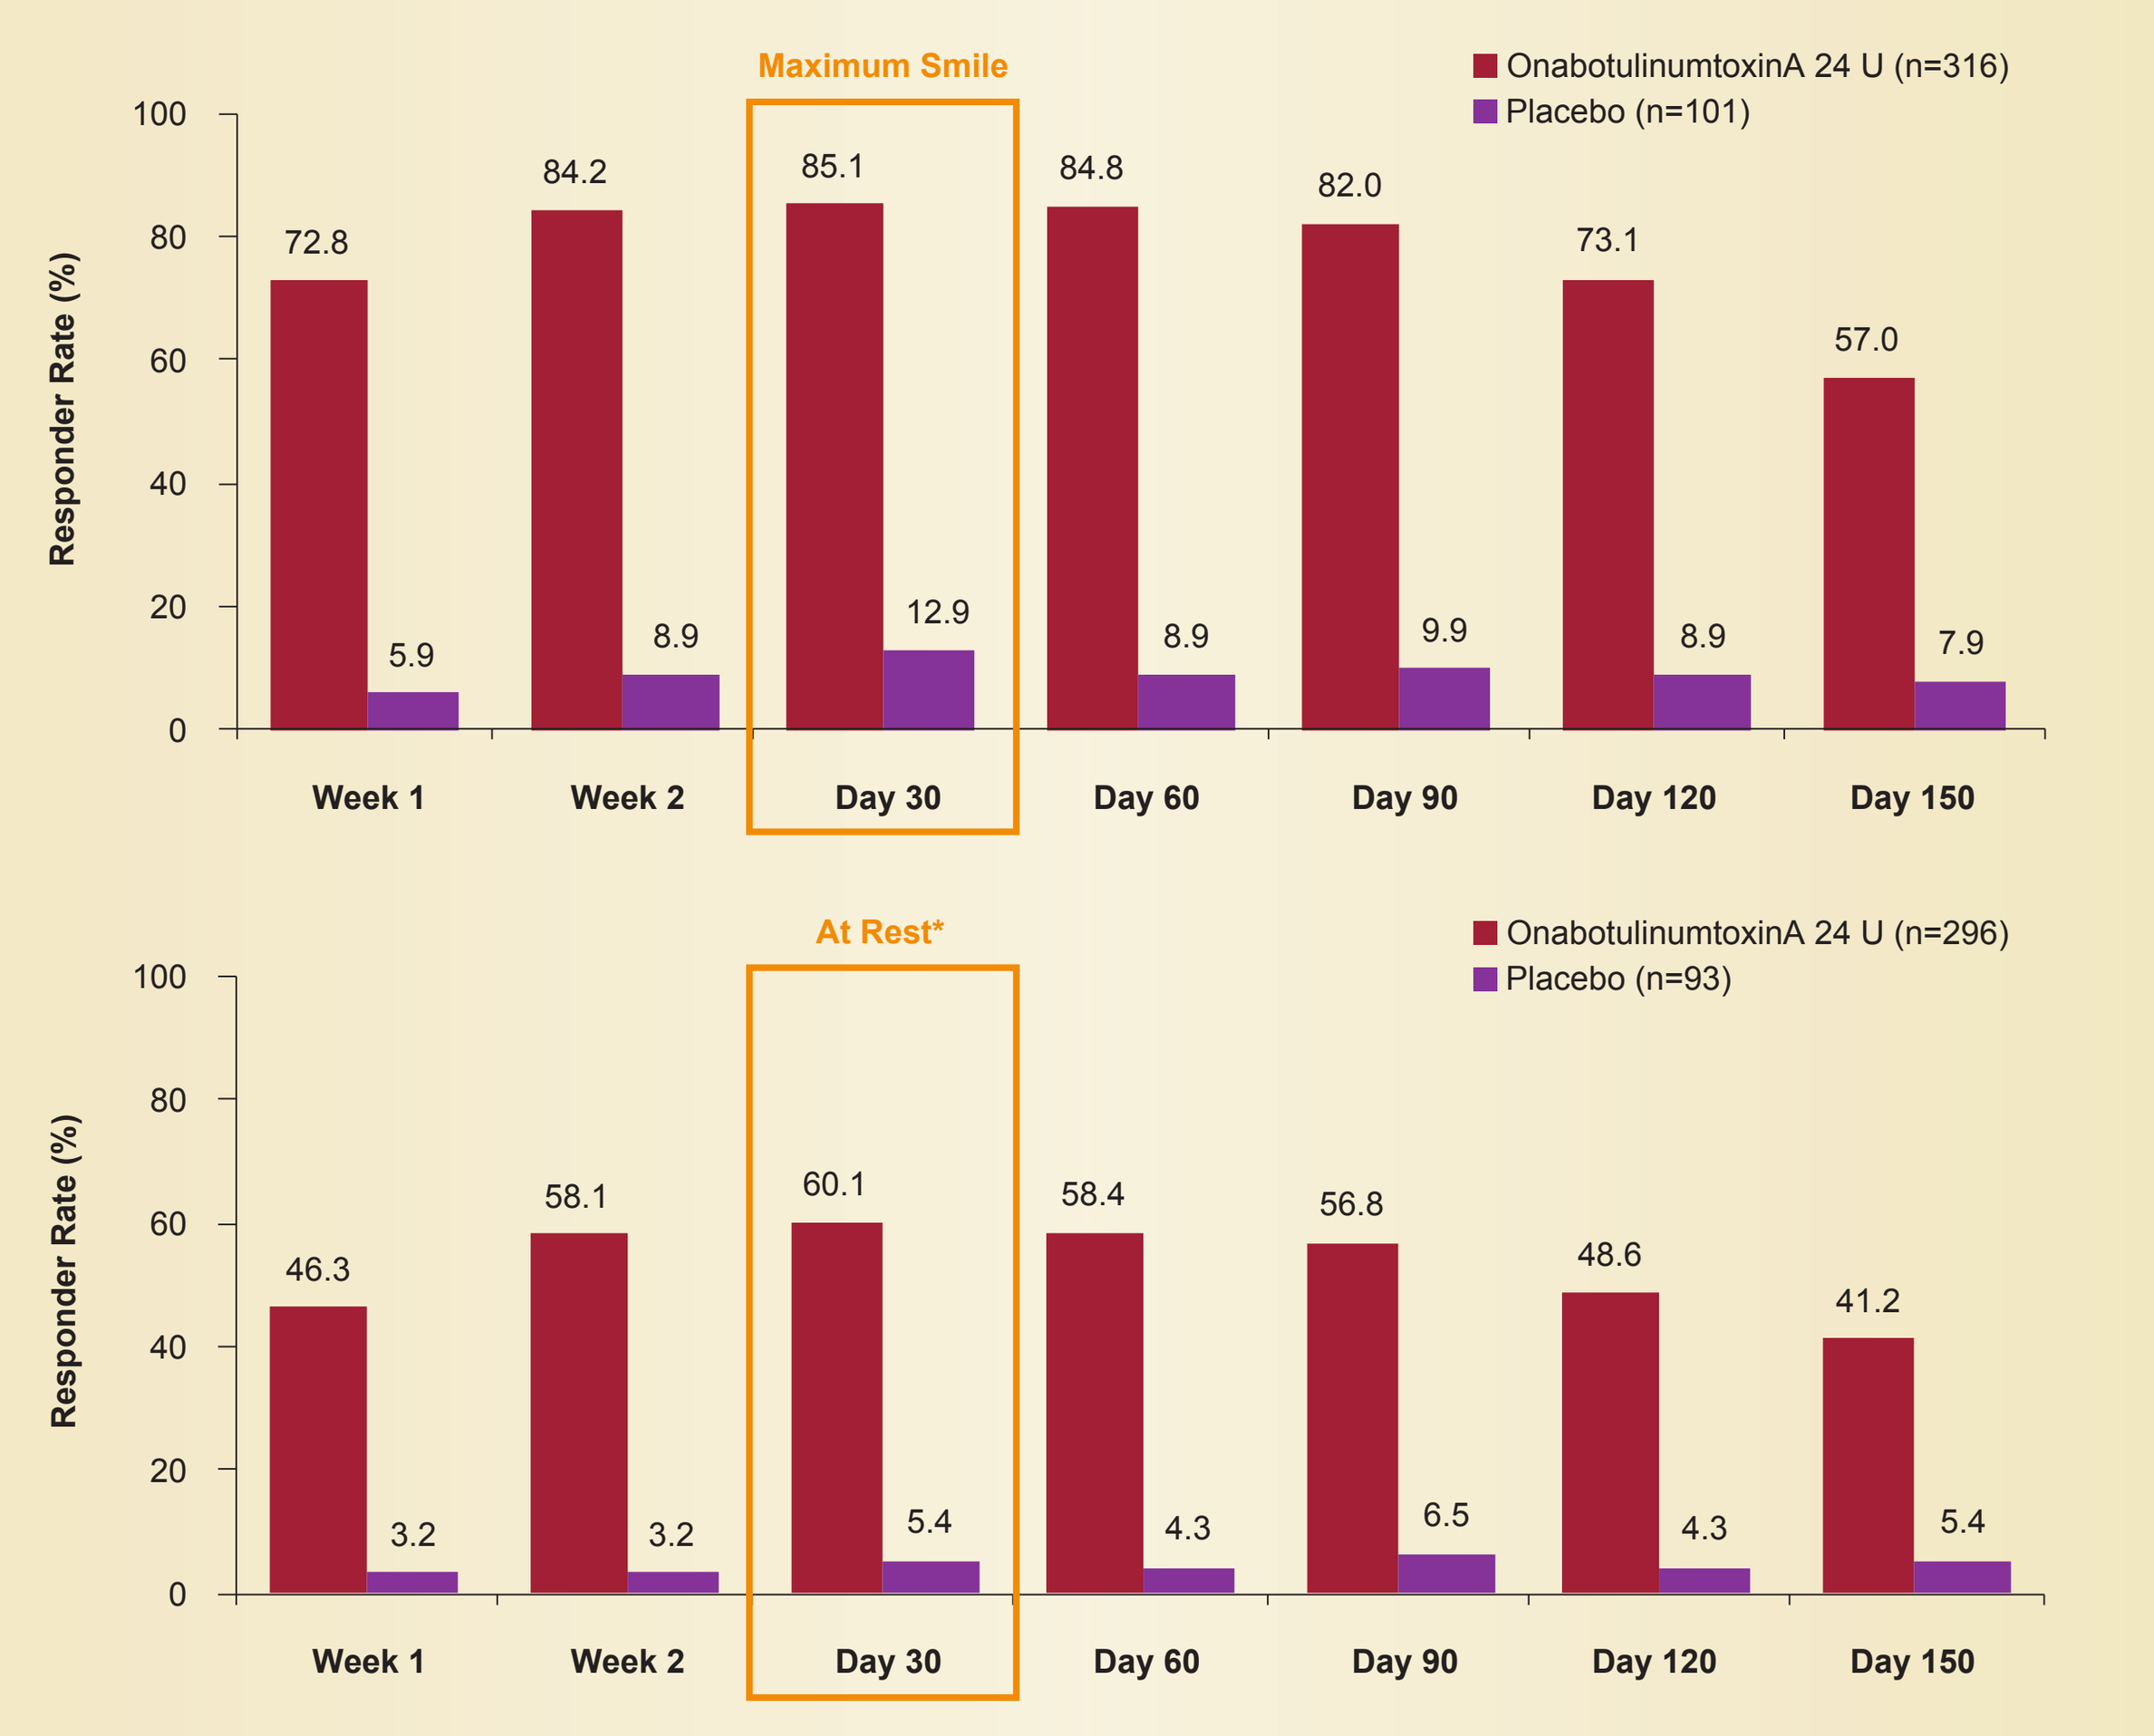

Maximum Smile
OnabotulinumtoxinA 24 U (n=316)
Placebo (n=101)
Responder Rate (%)
100
80
60
40
20
0
72.8
5.9
84.2
8.9
85.1
12.9
84.8
8.9
82.0
9.9
73.1
8.9
57.0
7.9
Week 1
Week 2
Day 30
Day 60
Day 90
Day 120
Day 150
At Rest*
OnabotulinumtoxinA 24 U (n=296)
Placebo (n=93)
Responder Rate (%)
100
80
60
40
20
0
46.3
3.2
58.1
3.2
60.1
5.4
58.4
4.3
56.8
6.5
48.6
4.3
41.2
5.4
Week 1
Week 2
Day 30
Day 60
Day 90
Day 120
Day 150

P<0.001, onabotulinumtoxinA vs placebo, all time points.

*Only subjects who were rated at least mild at baseline were included in analysis "at rest."
FWS-A, Facial Wrinkle Scale with Asian Photonumeric Guide.

SPA Responders

- The proportion of responders who rated themselves as "looking their current age or older" at baseline and in a younger age category after treatment was significantly greater in the onabotulinumtoxinA group vs the placebo group at day 30 and all other time points (**Figure 5**)

Figure 5. Subject Self-Perception of Age*

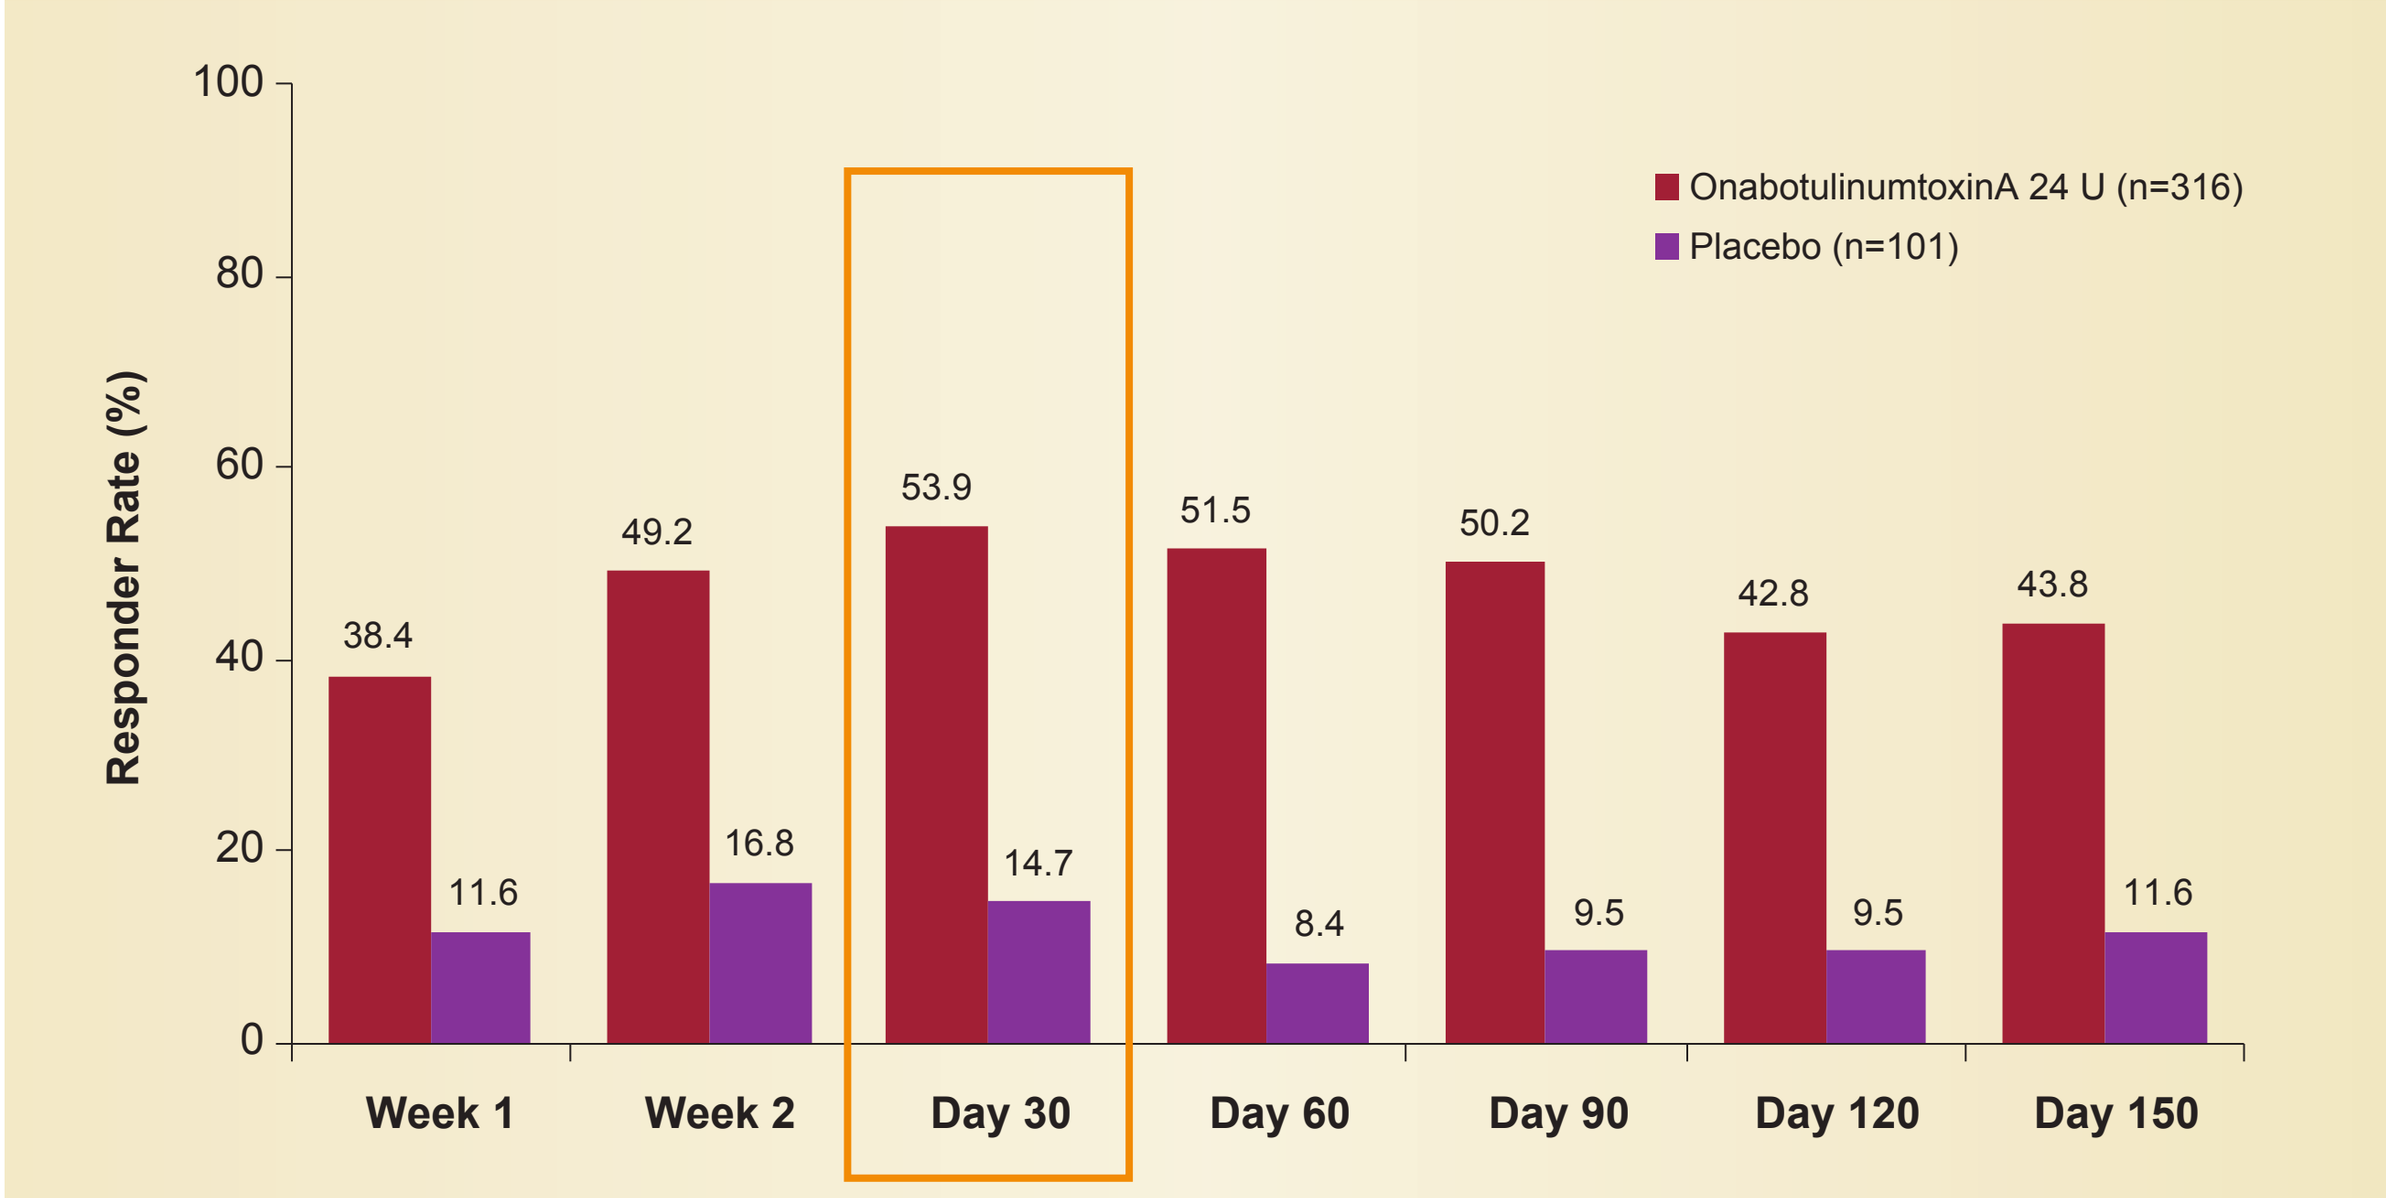

Responder Rate (%)
100
80
60
40
20
0
OnabotulinumtoxinA 24 U (n=316)
Placebo (n=101)
38.4
11.6
49.2
16.8
53.9
14.7
51.5
8.4
50.2
9.5
42.8
9.5
43.8
11.6
Week 1
Week 2
Day 30
Day 60
Day 90
Day 120
Day 150

P<0.001, onabotulinumtoxinA vs placebo, all time points.

*Subject who rated themselves as "looking their current age or older" at baseline and in a younger agecategory post-baseline.

Duration of Effect

- The median time to loss of efficacy (from responder to non-responder) was estimated for day 30 responders using the responder definitions shown in **Figure 6**
- The median duration of effect was at ≥5 months (ie, through end of study)

Figure 6. Duration of Effect

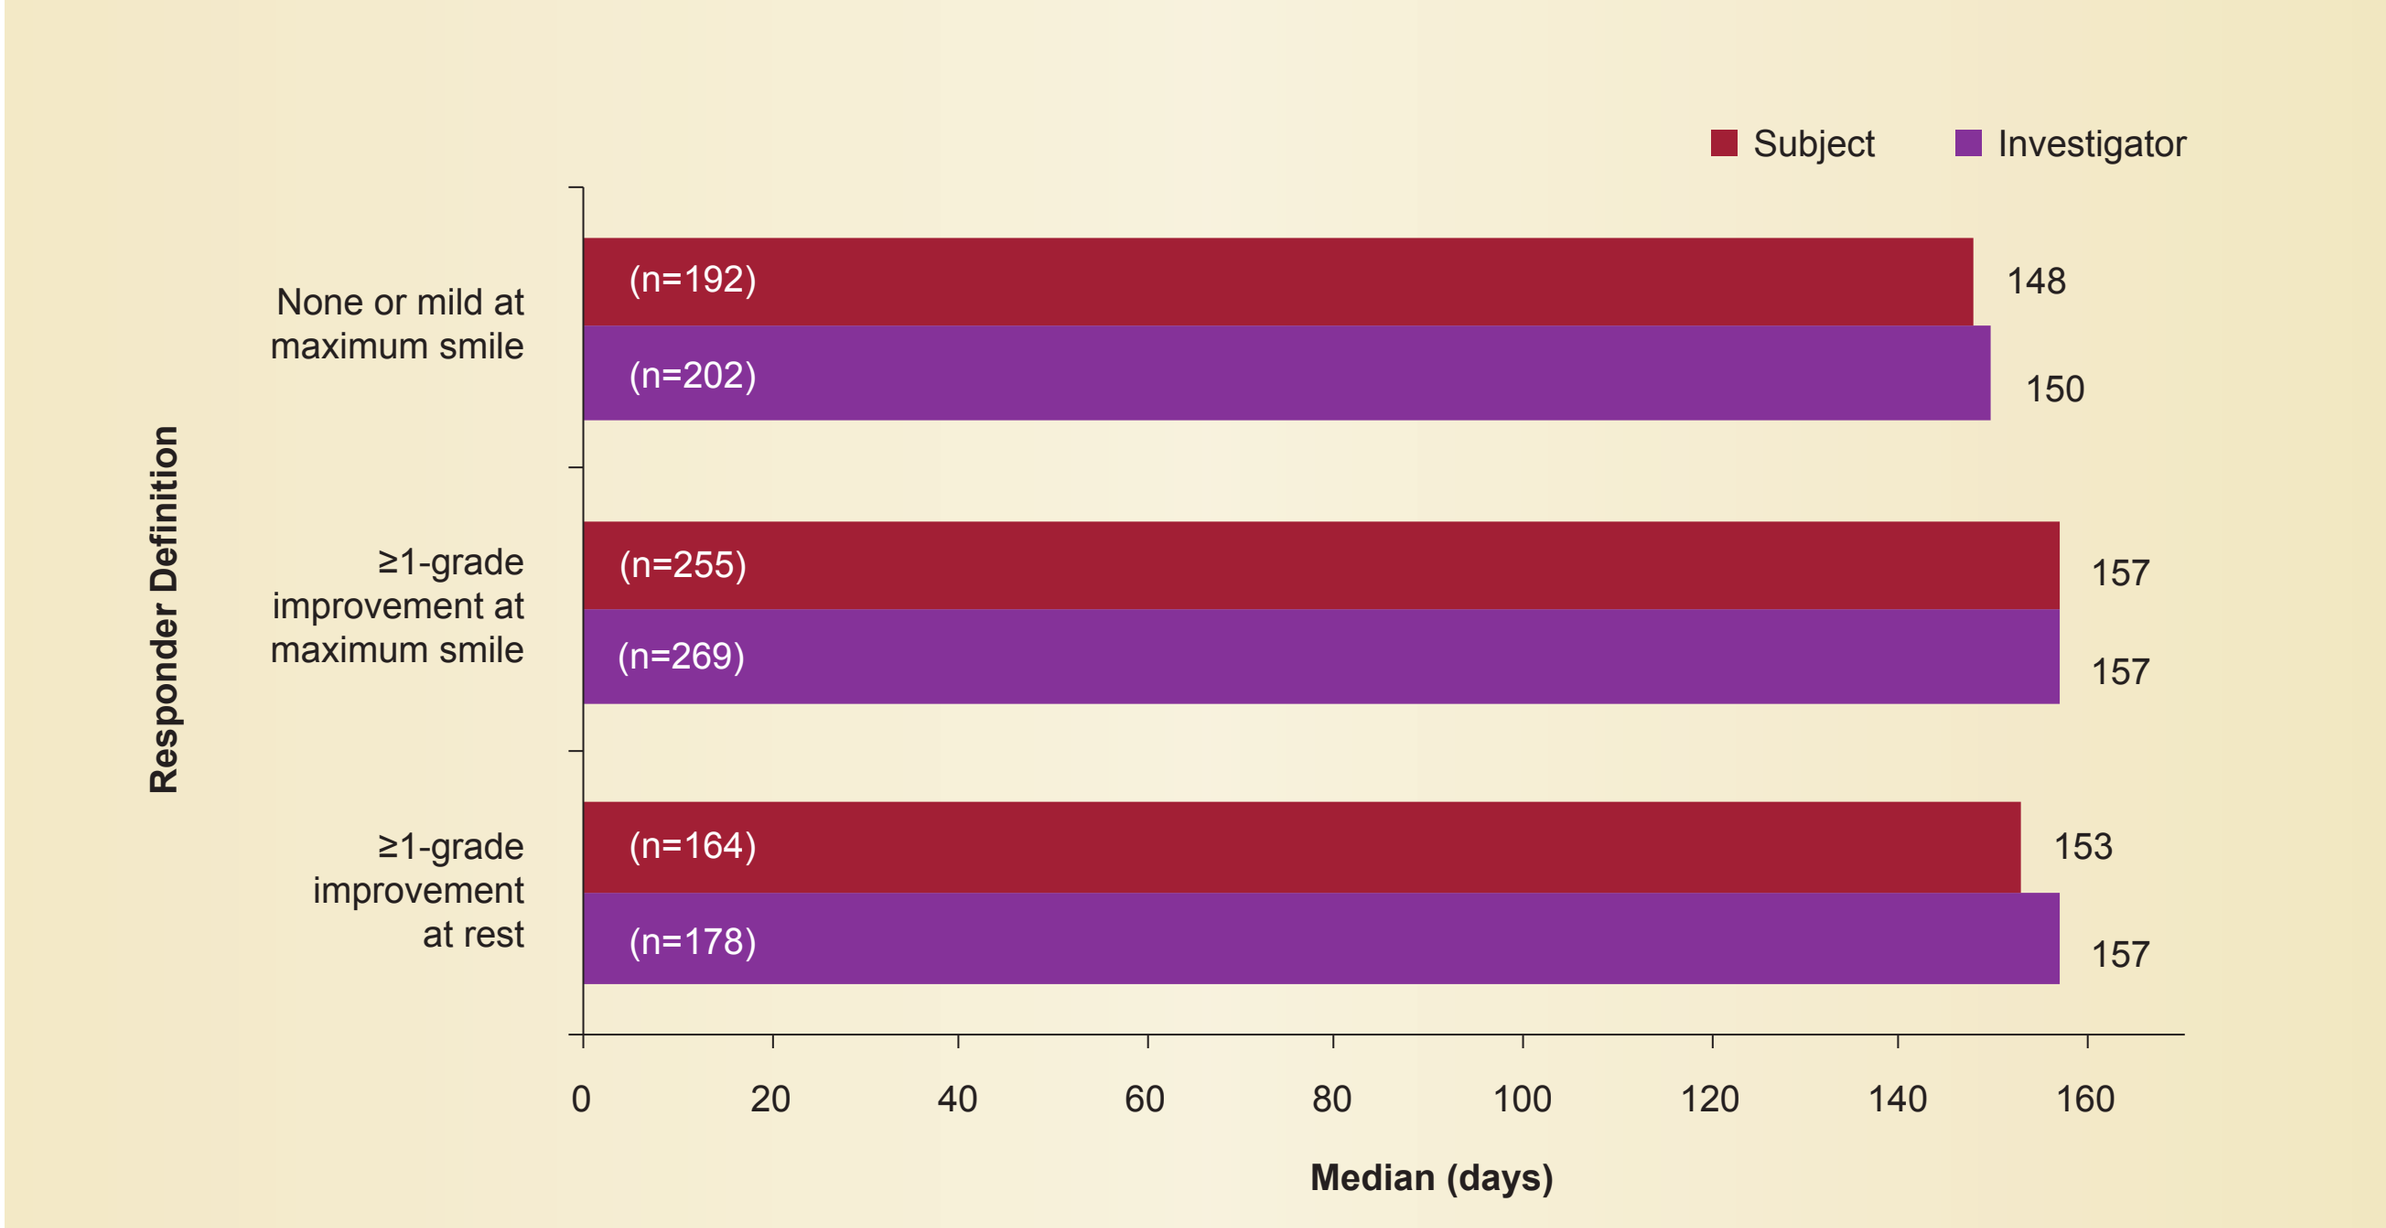

Subject
Investigator
Responder Definition
None or mild at maximum smile
(n=192)
148
(n=202)
150
≥1-grade improvement at maximum smile
(n=255)
157
(n=269)
157
≥1-grade improvement at rest
(n=164)
153
(n=178)
157
0
20
40
60
80
100
120
140
160
Median (days)

N values within bars are number of responders.

Subject Assessment of Satisfaction With Appearance

- Responders: subjects who reported being "very satisfied" or "satisfied" with CFL appearance
- Among subjects who were "neutral," "unsatisfied," or "very unsatisfied" with CFL appearance at baseline (98.6% [411/417]), the proportion of responders at day 30 was significantly greater in the onabotulinumtoxinA group (57.7% [180/312]) than in the placebo group (4.0% [4/99]) (*P*<0.001 for day 30 and all other time points)
- This indicates that subjects who were treated with onabotulinumtoxinA were more satisfied with their CFL appearance than those treated with placebo throughout the course of the study

FLSQ Item 5

- Responders: subjects who reported being "very satisfied" or "mostly satisfied" with treatment effect
- Proportions of responders were significantly greater for the onabotulinumtoxinA group (70.8% [221/312]) vs the placebo group (16.0% [16/100]) at day 30 and at all other time points (*P*<0.001)
- This indicates that subjects who were treated with onabotulinumtoxinA were more satisfied than subjects treated with placebo with the effect of their treatment on CFL appearance over the course of the study

SGA-CFL

- Responders: subjects who rated themselves as "very much improved" or "much improved" in the appearance of their CFL
- Proportions of responders were significantly greater for the onabotulinumtoxinA group (54.4% [172/316]) than the placebo group (3.0% [3/101]) at day 30 and at all other time points (*P*<0.001)

SAFETY

Incidence and Severity of Treatment-Emergent AEs

- The proportion of subjects with treatment-emergent AEs (TEAEs) was similar in the onabotulinumtoxinA (7.6% [24/316]) and placebo (5.0% [5/101]) groups
- The most frequently reported TEAEs in the onabotulinumtoxinA and placebo groups, respectively, were:
 - Upper respiratory tract infection (1.3% [4/316]; 0.0% [0/101])
 - Injection site bruising (1.3% [4/316]; 3.0% [3/101])
 - Nasopharyngitis (0.9% [3/316]; 0.0% [0/101])
 - Lip infection (0.6% [2/316]; 0.0% [0/101])
- All TEAEs were mild or moderate in severity, except for 1 event of eye injury that was severe and was considered unrelated to treatment
- Treatment-related AEs occurred in 3 onabotulinumtoxinA subjects (dizziness, 0.6% [2/316] and headache, (0.3% [1/316])
- No subject discontinued the study due to an AE
- No deaths, other serious TEAEs, possible spread of toxin events, neurologic events, or clinically meaningful changes in vital signs were reported

CONCLUSIONS

- All of the primary and secondary efficacy FWS-A endpoints were met
- At the day 30 primary time point following a single onabotulinumtoxinA treatment, statistically significant improvements in CFL severity favoring onabotulinumtoxinA vs placebo were observed across all responder definitions of CFL severity as assessed by the investigator
 - These results were consistent with subject-reported data, which demonstrated that subjects perceived the treatment benefits as meaningful
- Treatment of CFL with onabotulinumtoxinA 24 U was safe and well tolerated, with no new safety findings
- OnabotulinumtoxinA 24 U is an effective treatment for significantly reducing CFL severity in Chinese subjects

REFERENCES

1. ISAPS International Survey on Aesthetic/Cosmetic Procedures Performed in 2013. International Society of Aesthetic Plastic Surgeons. Available at: http://www.isaps.org/Media/Default/global-statistics/2014%20ISAPS%20Global%20Stat%20Results.pdf.
2. Carruthers A, et al. *Dermatol Surg.* 2014;40:1181-90.
3. Moers-Carpi M, et al. *Dermatol Surg.* 2015;41:102-12.
4. Park DH, et al. *Aesthetic Plast Surg.* 2012;36:1211-5.
5. Kane MAC, et al. *Dermatol Surg.* 2015;41:447-56.
6. Harii K, et al. Poster presented at: the 12th Annual Maui Derm for Dermatologists, January 25–29, 2016, Maui, HI, USA.

ACKNOWLEDGMENTS

This study was sponsored by Allergan Information Consulting (Shanghai) Co., Ltd., Shanghai, China. Writing and editorial assistance was provided to the authors by Peloton Advantage, Parsippany, NJ, USA, and was funded by Allergan plc, Dublin, Ireland. All authors met the ICMJE authorship criteria. Neither honoraria nor payments were made for authorship.

FINANCIAL DISCLOSURES

Y. Wu, W. Gang, and L. Chengxin have no conflicts to disclose. C. Mao, X. Lei, and E. Lee are employees of Allergan plc.

To obtain a PDF of this poster:
Scan the QR code
OR
Visit www.allergancongressposters.com/599695
Charges may apply. No personal information is stored.

Enter a valid e-mail to send this QR code or click to download.
Please ensure that qrcode@allergancongressposters.com has been added to your safe senders list or spam filter.

Presented at the TOXINS Meeting (International Neurotoxin Association), January 18-21, 2017, Madrid, Spain