OnabotulinumtoxinA in the Treatment of Crow's Feet Lines in Japanese Subjects

Kiyonori Harii, MD[1]; Makoto Kawashima, MD[2]; Xiaofang Lei, PhD[3]; René Hopfinger, MS[3]

[1]Kyorin University School of Medicine, Tokyo, Japan; [2]Tokyo Women's Medical University School of Medicine, Tokyo, Japan; [3]Allergan plc, Irvine, CA, USA

Scan to obtain PDF of poster

ABSTRACT

Purpose: Evaluate the safety and efficacy of onabotulinumtoxinA versus placebo in Japanese subjects with moderate to severe crow's feet lines (CFL).

Methods: This phase 3, multicenter, double-blind, randomized study comprised a 6-month, placebo-controlled treatment period followed by a 7-month, non–placebo-controlled treatment period. Eligible subjects included Japanese males and females aged 20–64 years with bilaterally symmetrical, moderate to severe CFL at maximum smile, based on the investigator-assessed Facial Wrinkle Scale with Asian Photonumeric Guide (FWS-A; 0 = none, 1 = mild, 2 = moderate, 3 = severe). Exclusion criteria included previous or concurrent treatment with botulinum toxin. Subjects were randomized (2:2:1:1) to receive treatment of onabotulinumtoxinA 24 U (both periods), onabotulinumtoxinA 12 U (both periods), placebo (period 1)/onabotulinumtoxinA 24 U (period 2), or placebo (period 1)/onabotulinumtoxinA 12 U (period 2). Subjects meeting re-treatment criteria, including the bilaterally symmetrical presence of CFL of at least moderate grade at maximum smile per FWS-A investigator assessments, could receive up to a total of 5 treatments (initial + 1 additional treatment in period 1; 3 additional treatments in period 2). The primary efficacy endpoint was the proportion of responders, defined by subjects achieving a CFL grade of none or mild at maximum smile on day 30 of period 1, based on the investigator's assessments using the FWS-A. Other efficacy endpoints included the proportion of subjects achieving at least a 1-grade improvement in CFL from baseline at maximum smile and at rest, based on the investigator-assessed FWS-A, and proportion of responders, based on various patient-reported outcomes, including Subject Assessment of Satisfaction with Appearance and Facial Line Satisfaction Questionnaire – Overall Satisfaction item. Adverse events (AEs) were recorded.

Results: Of the 300 enrolled subjects (mean age 49.7 ± 6.6 years, 25.3% male), 89.3% completed the study; most subjects received re-treatment. For the primary efficacy endpoint, the proportion of responders achieving a score of none or mild at maximum smile on the FWS-A with onabotulinumtoxinA (68.3% [24 U]; 56.6% [12 U]) was significantly greater versus placebo (8.2%; *P*<0.001). The proportion of responders and patterns of responder rates over time during treatment period 1 (placebo-controlled) were similar to those during treatment period 2 (non–placebo-controlled). The proportion of subjects achieving at least a 1-grade improvement in CFL at maximum smile on the FWS-A with onabotulinumtoxinA (80.8% [24 U]; 75.8% [12 U]) was significantly greater compared with placebo (17.5%; *P*<0.001). Similar results were observed for CFL at rest. Subjects receiving onabotulinumtoxinA were significantly more satisfied with their CFL appearance and with study drug than were subjects receiving placebo, per patient-reported outcome measures (*P*<0.001). Treatment-related AEs were uncommon (onabotulinumtoxinA 3.4% [10/294]; placebo 3.1% [3/97]) and were consistent with the known safety profile of onabotulinumtoxinA or were related to the injection procedure. No new safety signals were observed.

Conclusions: Treatment with onabotulinumtoxinA 24 U and 12 U for up to 5 treatment cycles over ≤13 months was effective in reducing the severity of moderate to severe CFL in Japanese subjects. Both doses were safe and well tolerated, with no new safety findings.

INTRODUCTION

- The use of botulinum toxin type A injections in minimally invasive aesthetic procedures has become common worldwide[1]
- OnabotulinumtoxinA (Allergan plc, Dublin, Ireland) is approved for the treatment of glabellar lines in more than 70 countries and crow's feet lines (CFL) in more than 50 countries, including the United States and countries in the European Union
- The majority of subjects in the pivotal clinical trials of onabotulinumtoxinA in CFL were non-Asian[2,3]
- In these trials, improvement in CFL severity with onabotulinumtoxinA treatment was evaluated using the Facial Wrinkle Scale (FWS)[2,3]
- Assessment of CFL morphology in Asian subjects has shown different CFL pattern prevalence compared with Caucasian subjects[4,5]
- Asian skin has increased dermal thickness, collagen content, and melanin compared with Caucasian skin[6]
- The FWS used in previous trials may not be sufficiently tailored for Asian subjects
- The current study is the first to use the validated FWS with Asian Photonumeric Guide (FWS-A) to assess the efficacy of onabotulinumtoxinA treatment of CFL in Japanese subjects

OBJECTIVE

- Evaluate the safety and efficacy of onabotulinumtoxinA compared with placebo in Japanese subjects with moderate to severe CFL

METHODS

Subjects

- Subjects were Japanese males and nonpregnant females aged 20 to 64 years, and were botulinum toxin naive, with bilaterally symmetrical moderate to severe CFL at maximum smile on the investigator-assessed FWS-A
- Key exclusion criteria included previous aesthetic treatments or surgical procedures at the treatment sites, eyebrow or eyelid ptosis, and eyelid hooding or other skin laxity that was unlikely to improve with onabotulinumtoxinA treatment or likely to interfere with CFL assessments

Study Design and Treatments

- This phase 3, multicenter, double-blind, randomized, placebo-controlled, parallel-group study was conducted at 21 centers in Japan from February 2013 through July 2014 (clinicaltrials.gov identifier: NCT01797081)
- The study comprised a 6-month placebo-controlled treatment period (period 1) followed by a 7-month non–placebo-controlled treatment period (period 2)
 - Subjects could participate for less than the total 13-month duration of the study
- Eligible subjects were randomized 2:2:1:1, respectively, to the following 4 treatment groups:
 - OnabotulinumtoxinA 24 U in treatment periods 1 and 2
 - OnabotulinumtoxinA 12 U in treatment periods 1 and 2
 - Placebo in period 1, onabotulinumtoxinA 24 U in treatment period 2
 - Placebo in period 1, onabotulinumtoxinA 12 U in treatment period 2
- Treatment consisted of 6 injections (0.1 mL each) of onabotulinumtoxinA or placebo administered bilaterally (3 on each side of the face) and intramuscularly to the orbicularis oculi
 - Two alternative injection patterns were used, based upon each subject's pattern of CFL and the investigator's clinical judgment (**Figure 1**)

Figure 1. Alternative Injection Patterns for Treatment of CFL

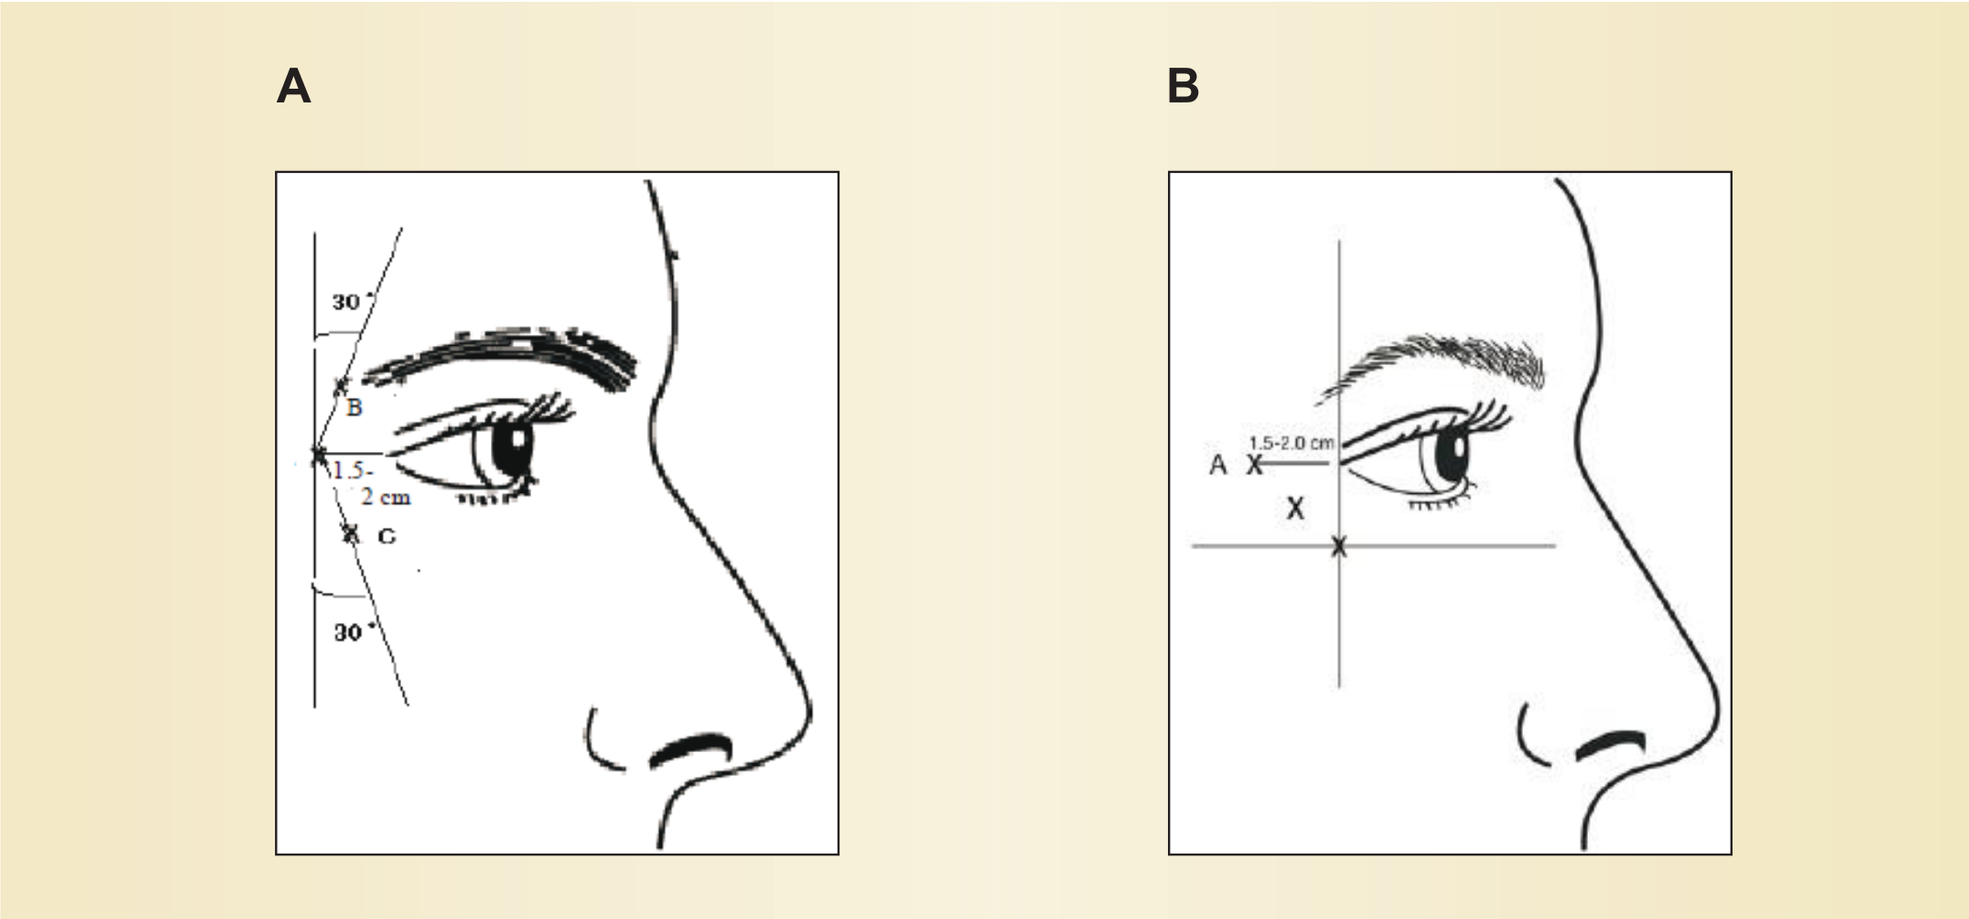
A
B
30°
B
1.5-2 cm
C
30°
A
1.5-2.0 cm
X
X

- Subjects were permitted up to 5 treatments (initial + 1 additional treatment in period 1 and up to 3 additional treatments in period 2)
 - A subject was eligible for re-treatment if he/she had bilaterally symmetrical CFL of at least moderate severity at maximum smile per the investigator-assessed FWS-A, at least 84 days since previous treatment, not at the day 390 visit, and was not pregnant
- Efficacy assessments were conducted on weeks 1 and 2 after each treatment, monthly beginning at month 1 through month 12, and at study exit (month 13 or early discontinuation)
- Differences in duration of response between subjects resulted in differences in timing to re-treatment eligibility; accordingly, the study design specified treatment/re-treatment intervals of differing potential durations (**Table 1**)

Table 1. Potential Maximum Duration of Treatment Intervals

Treatment	Duration (days)
1	≤390 (day 1 treatment in period 1)
2	≤300 (second treatment in period 1 or first treatment in period 2)
3	≤210 (first or second treatment in period 2)
4	≤120 (second or third treatment in period 2)
5	≤30 (third treatment in period 2)

Efficacy Measures

- The primary efficacy endpoint was the proportion of responders (defined as subjects achieving CFL severity of none or mild at maximum smile) at day 30 of treatment period 1 as assessed by the investigator using the FWS-A, where 0 = none, 1 = mild, 2 = moderate, and 3 = severe
 - CFL severity was based on the average of scores for the left and right sides
- Secondary efficacy measures included:
 - The proportion of responders (defined as subjects achieving at least a 1-grade improvement in CFL severity) at maximum smile and at rest at day 30 of treatment 1, as assessed by the investigator using the FWS-A
- Other efficacy measures included duration of effect, defined as time from achieving responder status to return to nonresponder status using the FWS-A
 - None or mild and ≥1-grade improvement responders were assessed

Patient-Reported Outcomes (PRO)

- Subject Assessment of Satisfaction with Appearance was a single-item measure for which subjects rated satisfaction with the appearance of their CFL on a scale of 1 ("very unsatisfied") to 5 ("very satisfied")
 - Responders were subjects who demonstrated improvement from "neutral" (3) or worse at baseline to "satisfied" (4) or "very satisfied" (5) on day 30
- Facial Line Satisfaction Questionnaire (FLSQ)–Overall Satisfaction item assessed each subject's satisfaction with treatment on a scale of −2 ("very dissatisfied") to +2 ("very satisfied")
 - Responders were subjects who were "mostly satisfied" or "very satisfied" on day 30

Safety

- Safety measurements included adverse events (AEs), vital sign measurements (pulse rate, respiratory rate, and blood pressure), physical examination, and neurologic assessment

Statistical Analysis

- Analyses were performed for the intent-to-treat population (all enrolled subjects) for the efficacy analyses, and safety analyses were performed for all subjects who received ≥1 study drug injection (safety population)
- For FWS-A and PRO measures, comparison of the proportion of responders was performed using the Cochran-Mantel-Haenszel test, stratified by baseline CFL severity at maximum smile
- For period 1, both placebo groups were combined as one for comparison with the onabotulinumtoxinA 24 U and onabotulinumtoxinA 12 U groups

RESULTS

Subject Disposition and Demographics

- Of the 305 subjects screened, 300 were enrolled and randomized (**Table 2; Figure 2**)
- The majority (89.3%, 268/300) of the enrolled subjects completed the study
- Demographic and baseline characteristics between the treatment groups were similar
- The majority of subjects (56.3%, 169/300) were aged ≤50 years and were female (74.7%, 224/300)

Table 2. Demographic and Baseline Characteristics

Parameter	OnabotulinumtoxinA 24 U[a]/ OnabotulinumtoxinA 24 U[b] (n=104)	OnabotulinumtoxinA 12 U[a]/ OnabotulinumtoxinA 12 U[b] (n=99)	Placebo[a]/ OnabotulinumtoxinA 24 U[b] (n=48)	Placebo[a]/ OnabotulinumtoxinA 12 U[b] (n=49)	Total (N=300)	P value
Age, years						
Mean	50.2	50.0	49.3	48.3	49.7	
SD	6.05	6.11	7.24	8.10	6.64	
Median	50.0	49.0	49.0	48.0	49.0	
Minimum	36	36	26	25	25	
Maximum	63	64	64	64	64	
≤50, n (%)	53 (51.0)	58 (58.6)	28 (58.3)	30 (61.2)	169 (56.3)	0.578
>50, n (%)	51 (49.0)	41 (41.4)	20 (41.7)	19 (38.8)	131 (43.7)	
Gender, n (%)						
Female	84 (80.8)	70 (70.7)	36 (75.0)	34 (69.4)	224 (74.7)	0.309
Male	20 (19.2)	29 (29.3)	12 (25.0)	15 (30.6)	76 (25.3)	
Investigator's FWS-A assessment of CFL severity at maximum smile						
Moderate	51 (49.0)	49 (49.5)	23 (47.9)	24 (49.0)	147 (49.0)	0.998
Severe	53 (51.0)	50 (50.5)	25 (52.1)	25 (51.0)	153 (51.0)	
Investigator's FWS-A assessment of CFL severity at rest						
None	5 (4.8)	5 (5.1)	8 (16.7)	8 (16.3)	26 (8.7)	0.594
Mild	61 (58.7)	56 (56.6)	23 (47.9)	24 (49.0)	164 (54.7)	
Moderate	32 (30.8)	33 (33.3)	15 (31.3)	12 (24.5)	92 (30.7)	
Severe	6 (5.8)	5 (5.1)	2 (4.2)	5 (10.2)	18 (6.0)	
Subject assessment of satisfaction with appearance						
Mean	1.8	2.0	1.8	1.8	1.9	0.154
SD	0.55	0.65	0.60	0.63	0.61	
Median	2.0	2.0	2.0	2.0	2.0	
Minimum	1	1	1	1	1	
Maximum	3	3	3	3	3	

[a]Randomized treatment assignment in treatment period 1.
[b]Randomized treatment assignment in treatment period 2.
CFL, crow's feet lines; FWS-A, Facial Wrinkle Scale with Asian Photonumeric Guide; SD, standard deviation.

Subject Re-treatment

- The majority of subjects were re-treated (**Figure 2**)

Figure 2. Study Flow Diagram

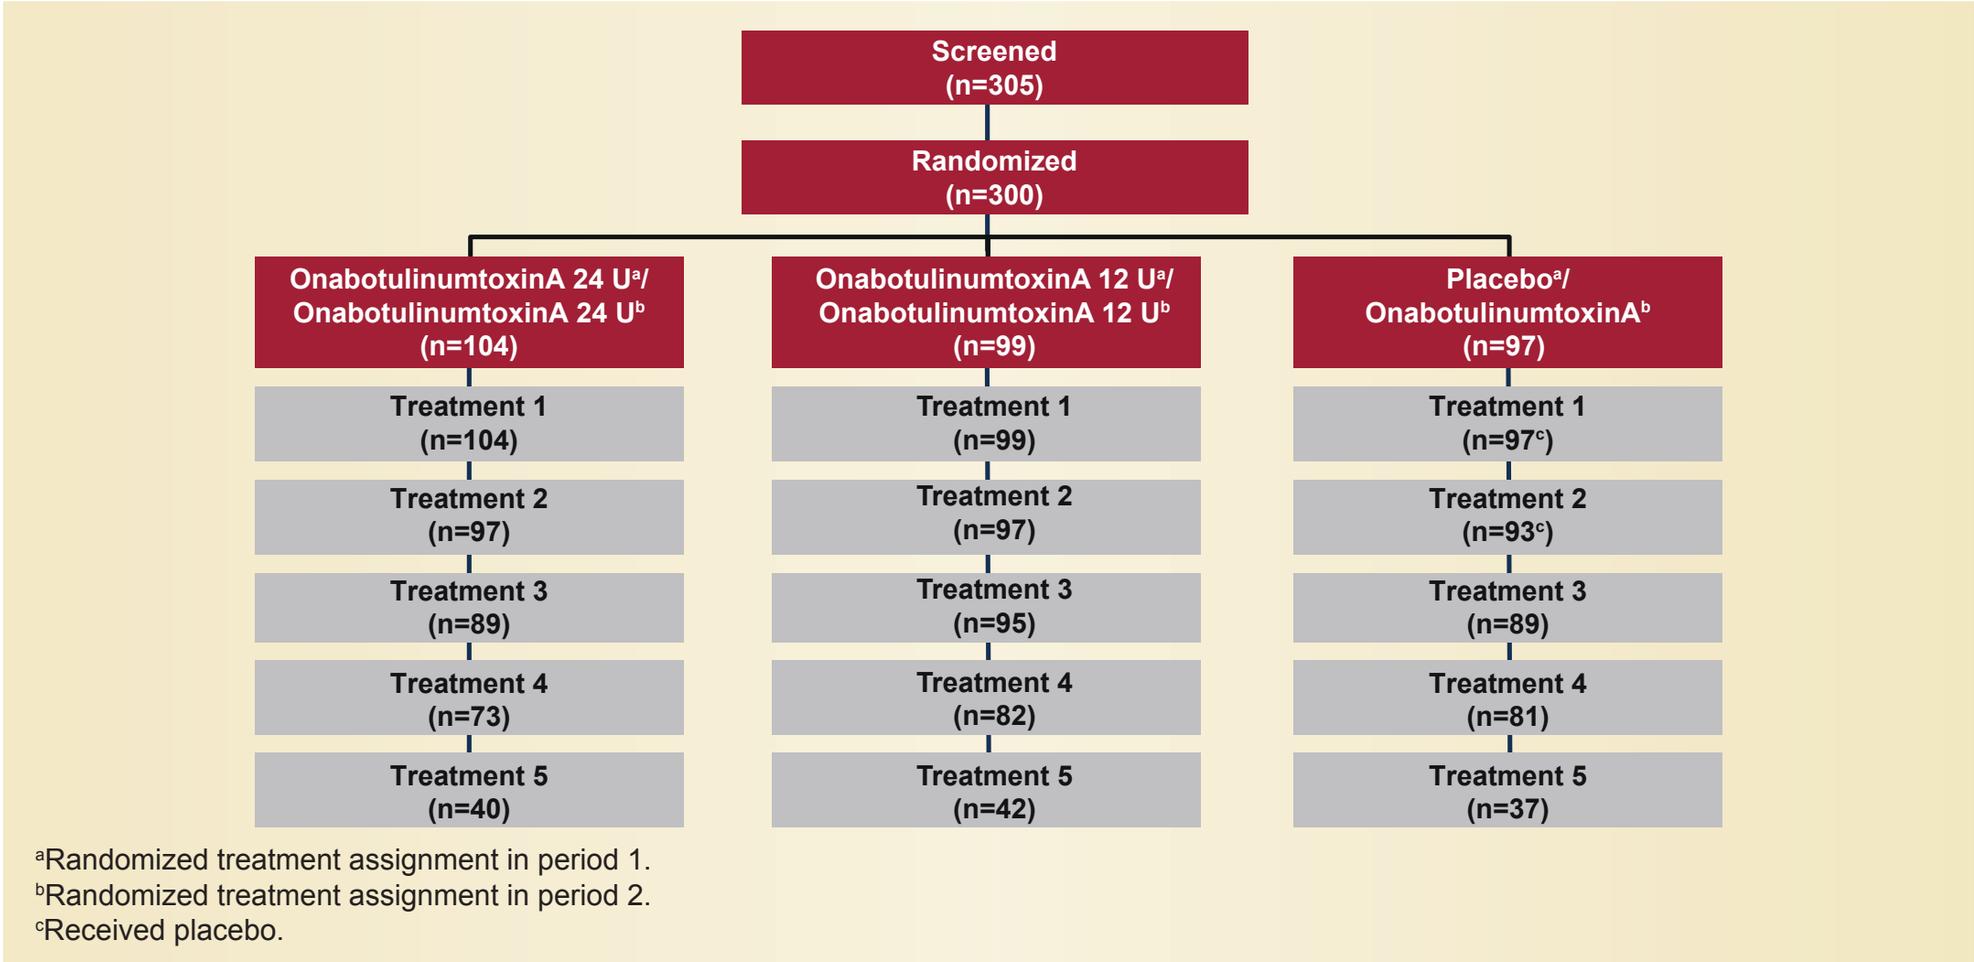
Screened (n=305)
Randomized (n=300)
OnabotulinumtoxinA 24 U[a]/ OnabotulinumtoxinA 24 U[b] (n=104)
OnabotulinumtoxinA 12 U[a]/ OnabotulinumtoxinA 12 U[b] (n=99)
Placebo[a]/ OnabotulinumtoxinA[b] (n=97)
Treatment 1 (n=104)
Treatment 2 (n=97)
Treatment 3 (n=89)
Treatment 4 (n=73)
Treatment 5 (n=40)
Treatment 1 (n=99)
Treatment 2 (n=97)
Treatment 3 (n=95)
Treatment 4 (n=82)
Treatment 5 (n=42)
Treatment 1 (n=97[c])
Treatment 2 (n=93[c])
Treatment 3 (n=89)
Treatment 4 (n=81)
Treatment 5 (n=37)

[a]Randomized treatment assignment in period 1.
[b]Randomized treatment assignment in period 2.
[c]Received placebo.

Investigator-Assessed FWS-A Endpoint: Improvement to None or Mild Severity

- The primary efficacy endpoint was met
- At day 30 of treatment 1, a greater proportion of subjects had CFL severity ratings of none or mild at maximum smile in the onabotulinumtoxinA 24 U (68.3% [71/104]) and onabotulinumtoxinA 12 U (56.6% [56/99]) groups compared with the placebo group (8.2% [8/97]) (*P*<0.001, both comparisons) (**Figure 3**)
- The proportion of responders and patterns of responder rates over time during treatment period 1 (placebo-controlled) were similar to those during treatment period 2 (non–placebo-controlled) (**Figure 3**)

Figure 3. Proportion of Subjects Achieving CFL Severity of None or Mild at Maximum Smile up to Day 90 of Each Treatment Using the FWS-A (Intent-to-Treat Population)

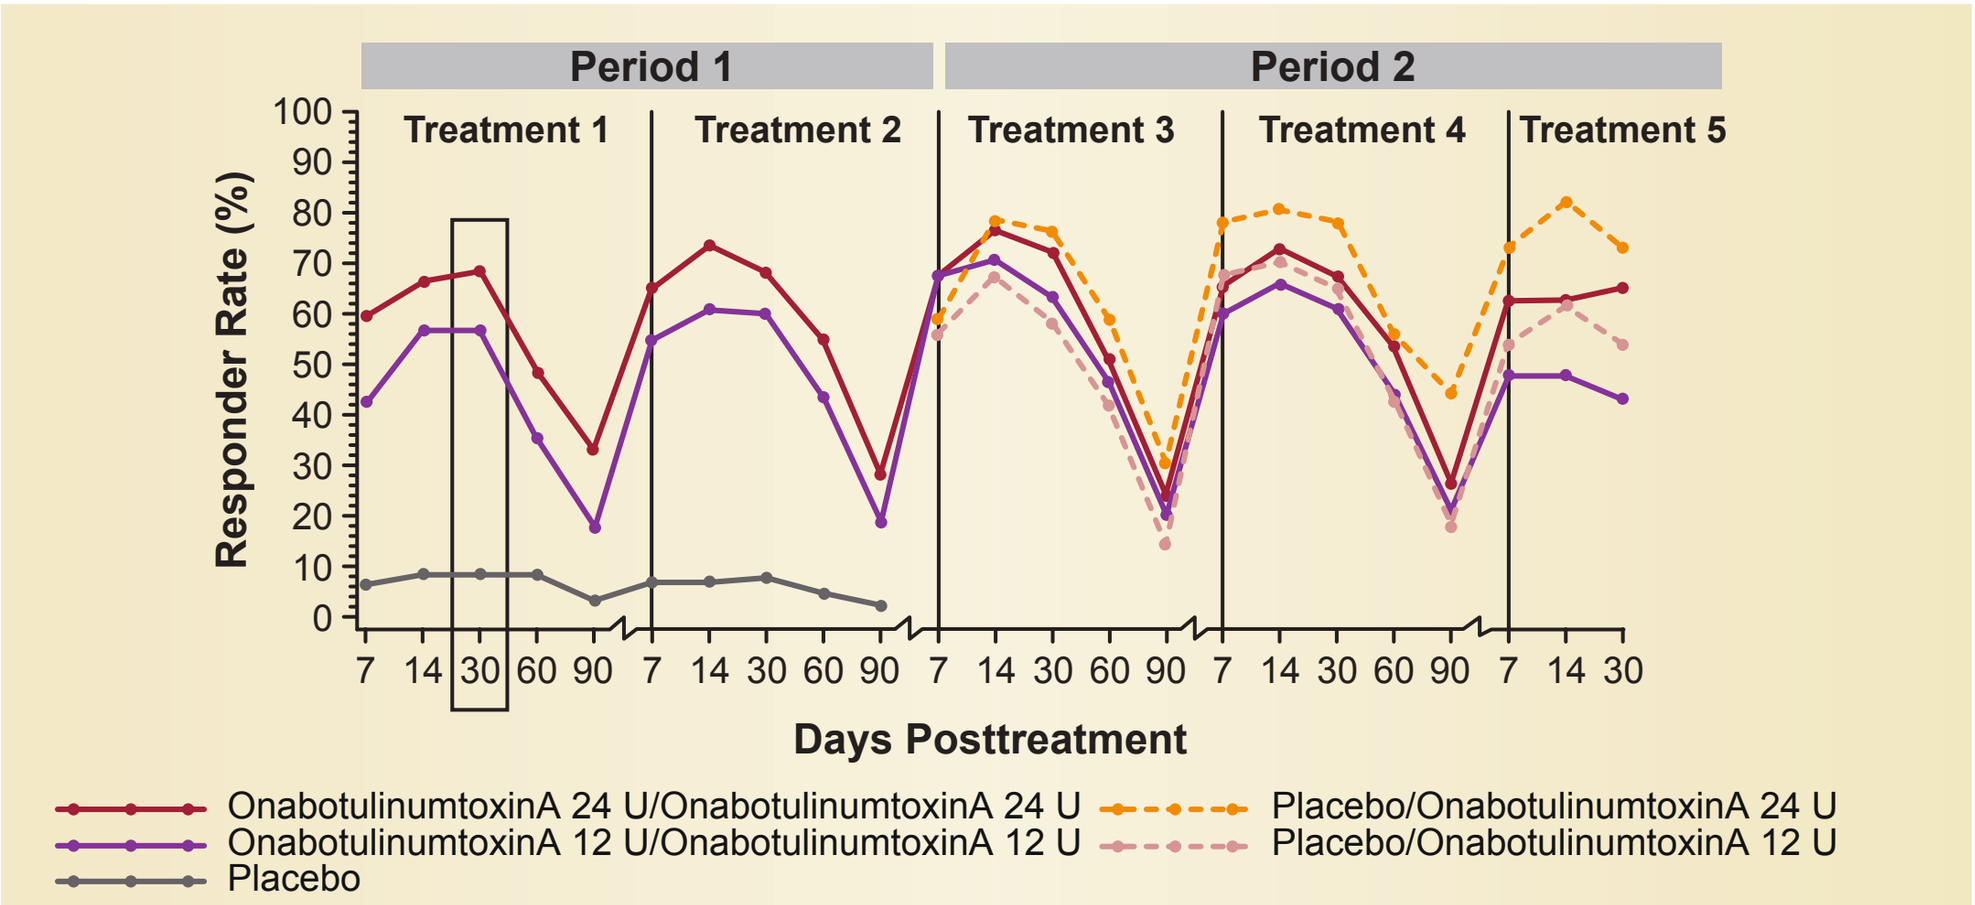
Period 1
Period 2
Treatment 1
Treatment 2
Treatment 3
Treatment 4
Treatment 5
Responder Rate (%)
Days Posttreatment
OnabotulinumtoxinA 24 U/OnabotulinumtoxinA 24 U
OnabotulinumtoxinA 12 U/OnabotulinumtoxinA 12 U
Placebo
Placebo/OnabotulinumtoxinA 24 U
Placebo/OnabotulinumtoxinA 12 U

CFL, crow's feet lines; FWS-A, Facial Wrinkle Scale with Asian Photonumeric Guide.

Investigator-Assessed FWS-A Endpoints: ≥1-Grade Improvement

- At day 30 of treatment 1, a greater proportion of ≥1-grade improvement responders in CFL severity at maximum smile was observed with onabotulinumtoxinA 24 U (80.8% [84/104]) and onabotulinumtoxinA 12 U (75.8% [75/99]) compared with placebo (17.5% [17/97]) (*P*<0.001, both comparisons) (**Figure 4A**)
- At day 30 of treatment 1, a greater proportion of ≥1-grade responders in CFL severity at rest was observed with onabotulinumtoxinA 24 U (61.6% [61/99]) and onabotulinumtoxinA 12 U (43.6% [41/94]) compared with placebo (17.3% [14/81]) (*P*<0.001, both comparisons) (**Figure 4B**)
- Significantly greater proportions of ≥1-grade improvement responders at maximum smile (**Figure 4A**) and at rest (**Figure 4B**) with onabotulinumtoxinA 24 U and onabotulinumtoxinA 12 U compared with placebo (*P*<0.003, all comparisons) were observed at all time points up to 90 days during treatments 1 and 2 (**Figure 4A**)
- The proportion of responders and patterns of responder rates over time during period 1 (placebo-controlled) were similar to those during period 2 (non–placebo-controlled)

Figure 4. Proportion of Subjects Achieving CFL Severity of ≥1-Grade Improvement up to Day 90 of Each Treatment Using the FWS-A at Maximum Smile (A) and at Rest (B) (Intent-to-Treat Population)

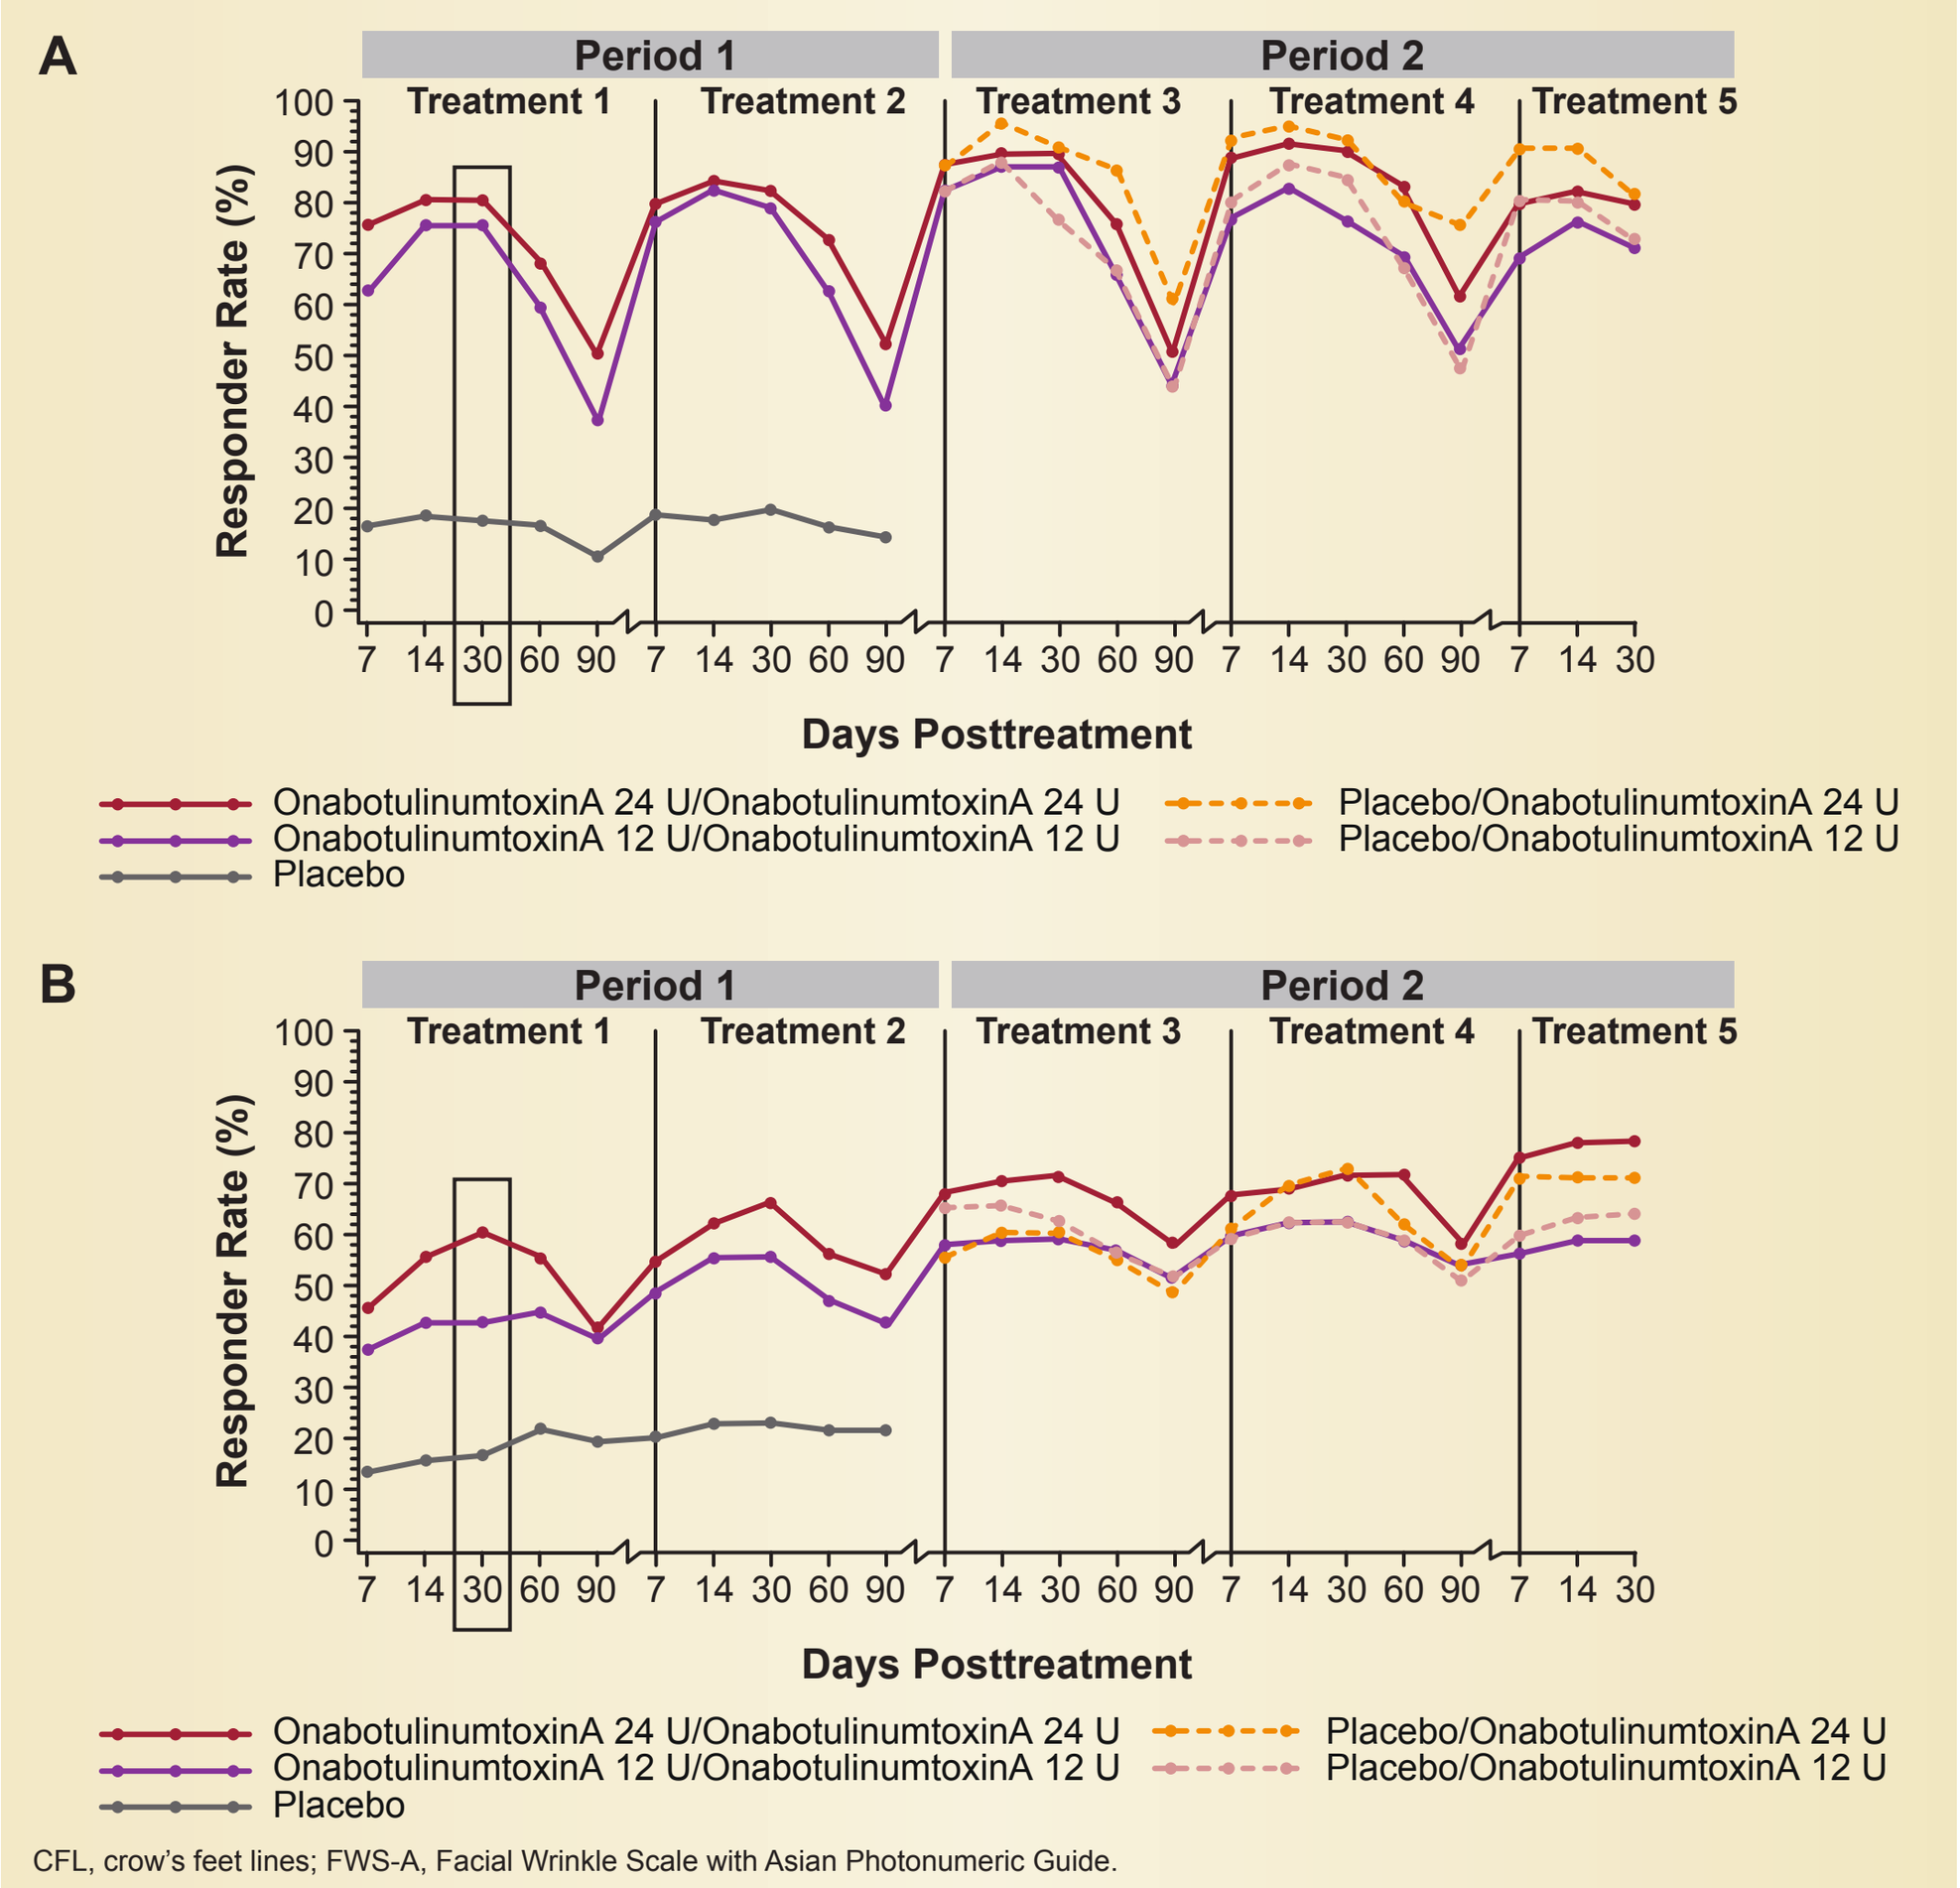
A
B
Period 1
Period 2
Treatment 1
Treatment 2
Treatment 3
Treatment 4
Treatment 5
Responder Rate (%)
Days Posttreatment
OnabotulinumtoxinA 24 U/OnabotulinumtoxinA 24 U
OnabotulinumtoxinA 12 U/OnabotulinumtoxinA 12 U
Placebo
Placebo/OnabotulinumtoxinA 24 U
Placebo/OnabotulinumtoxinA 12 U

CFL, crow's feet lines; FWS-A, Facial Wrinkle Scale with Asian Photonumeric Guide.

Duration of Effect

- The median duration of effect was approximately 3 months for onabotulinumtoxinA 24 U and 12 U using the responder definition of "none or mild" CFL severity at maximum smile and was slightly longer using the responder definition of ≥1-grade improvement in CFL severity at maximum smile (**Figure 5**)

Figure 5. Median Duration of Effect by Loss of Responder Status for None or Mild and for ≥1-Grade Improvement on the FWS-A at Maximum Smile (Intent-to-Treat Population)

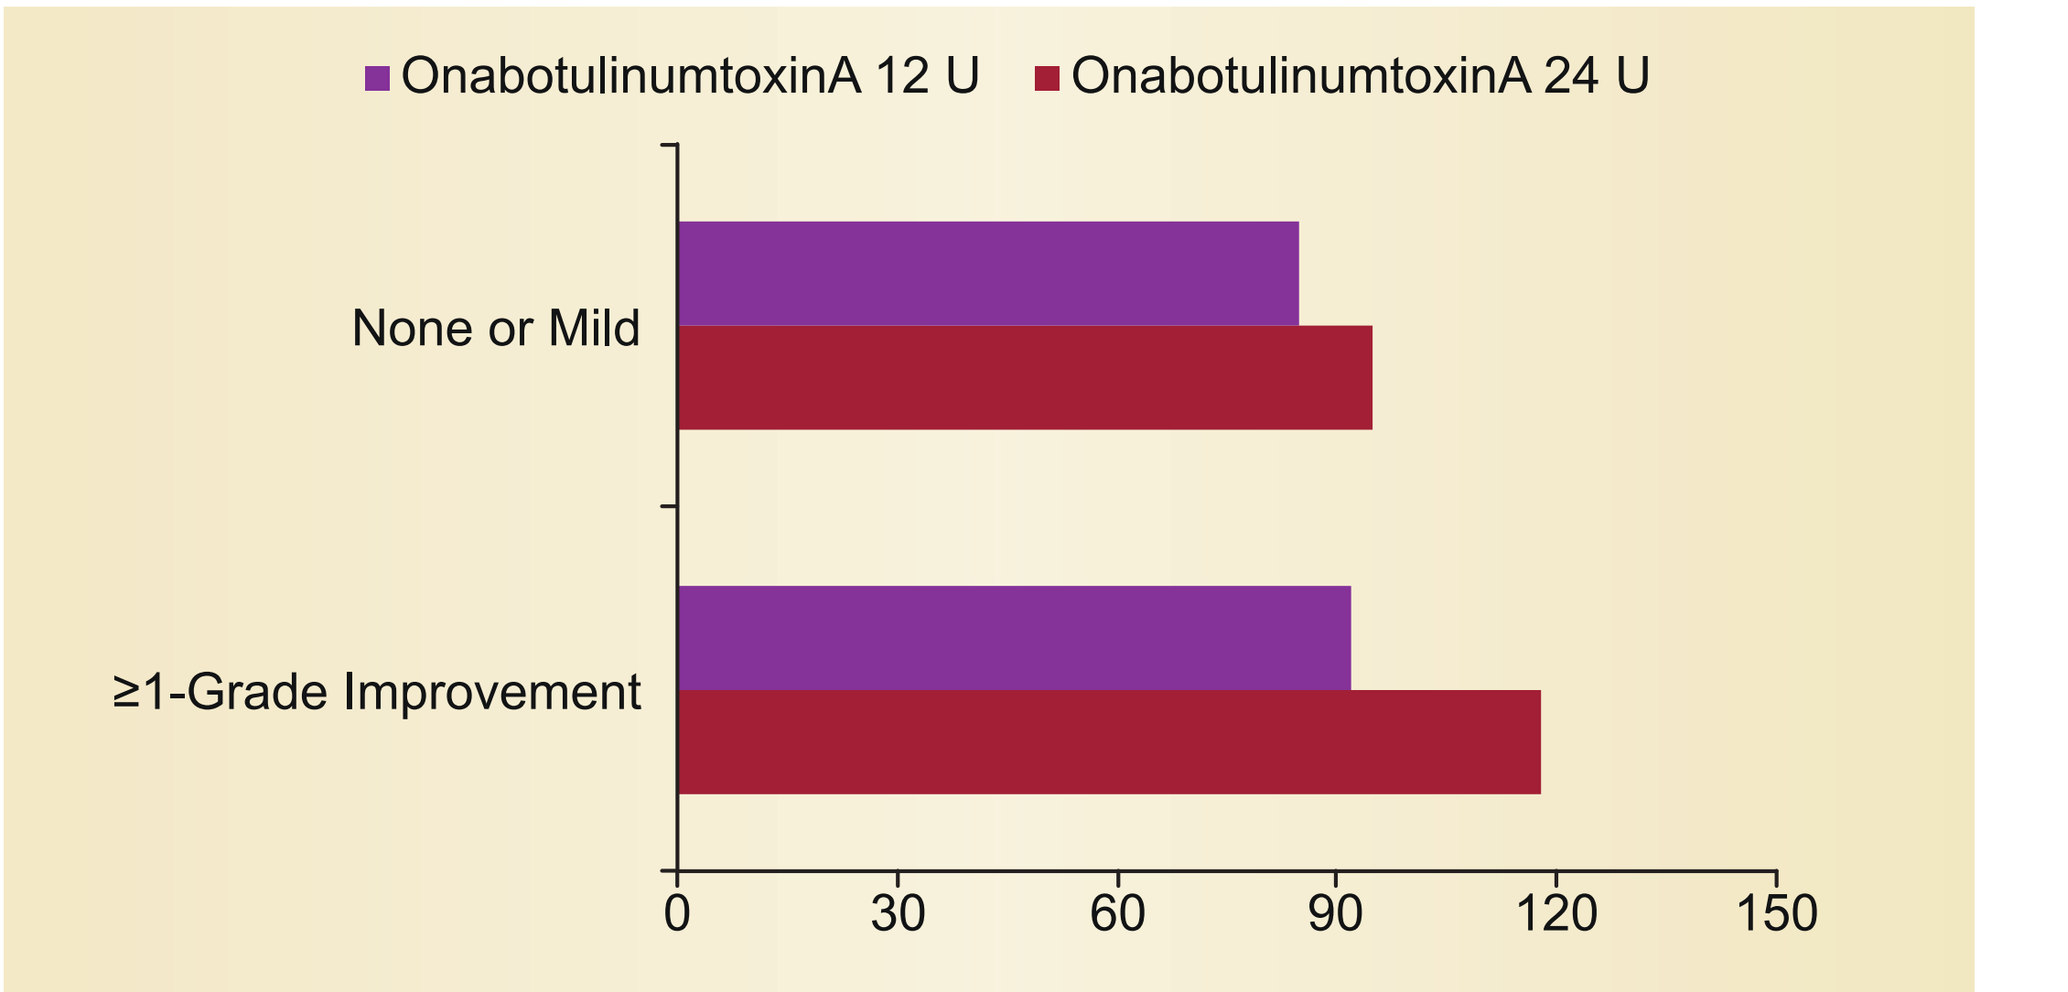
OnabotulinumtoxinA 12 U
OnabotulinumtoxinA 24 U
None or Mild
≥1-Grade Improvement
0
30
60
90
120
150

FWS-A, Facial Wrinkle Scale with Asian Photonumeric Guide.

Patient-Reported Outcomes

- The proportion of responders on the Subject Assessment of Satisfaction with Appearance and FLSQ–Overall Satisfaction item on day 30 of treatment 1 were significantly greater for onabotulinumtoxinA 24 U and 12 U compared with placebo (*P*<0.001, all comparisons)
- Responder rates for onabotulinumtoxinA treatment groups were similar for subsequent treatments in periods 1 and 2

Safety

- Treatment-emergent AEs (TEAEs) with onabotulinumtoxinA were reported by 27.2%, 20.4%, 29.4%, and 20.0% of subjects during the 90-day treatments 1, 2, 3, and 4, respectively
- An overview of TEAEs is shown in **Table 3**

Table 3. Overview of Treatment-Emergent Adverse Events (Total Population)

TEAE, n (%)	OnabotulinumtoxinA 24 U[a] (n=151)	OnabotulinumtoxinA 12 U[a] (n=143)	Placebo[a] (n=97)
All TEAEs	71 (47.0)	85 (59.4)	34 (35.1)
Treatment-related TEAEs	6 (4.0)	4 (2.8)	3 (3.1)
All serious TEAEs	3 (2.0)	3 (2.1)	1 (1.0)
Treatment-related serious TEAEs	0	0	0
AEs leading to discontinuation	2 (1.3)	0	0

[a]Subjects randomized to the placebo/onabotulinumtoxinA 24 U and 12 U groups are included in the onabotulinumtoxinA 24 U, 12 U, or placebo groups, respectively.
TEAE, treatment-emergent adverse event.

The incidence of treatment-related AEs was low and was similar across treatment groups (**Table 4**)

Table 4. Treatment-Related AEs (Safety Population)

Event, n (%)	OnabotulinumtoxinA 24 U (n=151)	OnabotulinumtoxinA 12 U (n=143)	OnabotulinumtoxinA Total (n=294)
Overall	6 (4.0)	4 (2.8)	10 (3.4)
Eye			
Abnormal sensation in eye	0 (0.0)	1 (0.7)	1 (0.3)
Eyelid edema	0 (0.0)	1 (0.7)	1 (0.3)
Eyelid pain	1 (0.7)	0 (0.0)	1 (0.3)
Eyelid ptosis	1 (0.7)	0 (0.0)	1 (0.3)
Gastrointestinal disorders			
Abdominal pain, upper	1 (0.7)	0 (0.0)	1 (0.3)
General			
Malaise	1 (0.7)	0 (0.0)	1 (0.3)
Thirst	1 (0.7)	0 (0.0)	1 (0.3)
Injection site pain	1 (0.7)	0 (0.0)	1 (0.3)
Injection site warmth	1 (0.7)	0 (0.0)	1 (0.3)
Injection site pruritus	0 (0.0)	1 (0.7)	1 (0.3)
Nervous system disorders			
Sensory disturbance	0 (0.0)	1 (0.7) 1 (0.3)	1 (0.7) 1 (0.3)
Psychiatric disorders			
Dysphoria	0 (0.0)	1 (0.7)	1 (0.3)
Skin			
Eyebrow ptosis	1 (0.7)	0 (0.0)	1 (0.3)
Skin tightness	1 (0.7)	0 (0.0)	1 (0.3)

AE, adverse event.

- No consistent differences in TEAEs were observed between onabotulinumtoxinA and placebo when examined by treatment over the 13-month study
- All TEAEs were mild or moderate in severity
- Nasopharyngitis was the most frequently reported TEAE in each group (15.2% [23/151], onabotulinumtoxinA 24 U; 27.3% [39/143], onabotulinumtoxinA 12 U; 5.2% [5/97], placebo)
- Eight TEAEs in 7 subjects (2.3% [7/300]) were identified as possible local spread of toxin events; none were considered related to distant spread of toxin
- Seven serious TEAEs were reported, none of which were considered to be related to treatment
- No changes in vital signs or other safety parameters were noted

CONCLUSIONS

- Treatment with onabotulinumtoxinA 24 U and 12 U for up to 5 treatments over 13 months was effective in reducing the severity of moderate to severe CFL in Japanese subjects
 - Improvements with onabotulinumtoxinA were observed as early as week 1 of treatment 1 and throughout both placebo-controlled treatment periods
 - The magnitude and time course of effects of onabotulinumtoxinA on responder rates for improvement in CFL severity as assessed at maximum smile were consistent over the 5 treatments
- Subjects reported significantly greater satisfaction with onabotulinumtoxinA treatment compared with placebo over the total course of 13 months
 - Overall, satisfaction scores were numerically higher for onabotulinumtoxinA 24 U than for 12 U
- Median duration of effect of onabotulinumtoxinA treatment was approximately 3 to 4 months, which is consistent with previous studies in CFL treatment with onabotulinumtoxinA[7]
- The safety profiles of both doses of onabotulinumtoxinA were similar; no new safety findings were identified

REFERENCES

1. International Society of Aesthetic Plastic Surgeons. Available at: http://www.isaps.org/Media/Default/global-statistics/2014%20ISAPS%20Global%20Stat%20Results.pdf.
2. Carruthers A, et al. *Dermatol Surg*. 2014;40:1181-90.
3. Moers-Carpi M, et al. *Dermatol Surg*. 2015;41:102-12.
4. Park DH, et al. *Aesthetic Plast Surg*. 2012;36:1211-5.
5. Kane MAC, et al. *Dermatol Surg*. 2015;41:447-56.
6. Kim MM, Byrne PJ. *Facial Plast Surg Clin North Am*. 2007;15:381-6.
7. Lowe NJ, et al. *Dermatol Surg*. 2005;31:257-62.

ACKNOWLEDGMENTS

This study was sponsored by Allergan plc, Dublin, Ireland. Medical writing and editorial assistance was provided to the authors by Peloton Advantage, Parsippany, NJ, USA, and was funded by Allergan plc. All authors met the ICMJE authorship criteria. Neither honoraria nor other form of payments were made for authorship.

FINANCIAL DISCLOSURES

K Harii and M Kawashima have no conflicts to disclose.
X Lei and R Hopfinger are employees of Allergan plc.

To obtain a PDF of this poster:
Scan the QR code
OR
Visit www.allergancongressposters.com/270193
Charges may apply. No personal information is stored.

Enter a valid e-mail to send this QR code or click to download.
Please ensure that qrcode@allergancongressposters.com has been added to your safe senders list or spam filter.

Presented at the TOXINS Meeting (International Neurotoxin Association), January 18-21, 2017, Madrid, Spain