Baseline characteristics of patients from a pilot study to assess the effect of early use of abobotulinumtoxin-A (Dysport®) in spasticity progression post stroke

Raymond L Rosales[1] Khean Jin Goh[2], Witsanu Kumthornthip[3], Jovita Balcaitiene[4], Pascal Maisonobe[4], Keng He Kong[5]

[1]Centre for Neurodiagnostic and Therapeutic Services, Metropolitan Medical Centre, and Department of Neurology & Psychiatry, Faculty of Medicine and Surgery, University of Santo Tomas, Manila, Philippines; [2]Division of Neurology, Department of Medicine, University of Malaya Medical Centre, Kuala Lumpur, Malaysia; [3]Department of Rehabilitation Medicine, Siriraj Hospital, Mahidol University, Bangkok, Thailand; [4]Medical Affairs, Ipsen Group, Boulogne-Billancourt, France; [5]Department of Rehabilitation Medicine, Tan Tock Seng Hospital, Novena, Singapore

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by TOXINS

Introduction

- Prevalence and severity of spasticity continue to increase during the first year post stroke, with up to 40% of stroke survivors developing post-stroke spasticity.[1–3]
- Reducing the burden of post-stroke spasticity is important for both the patient and caregivers.[1]
- Botulinum toxin A (BoNT-A) is an effective focal intervention for reducing upper limb spasticity (ULS).[4]
- In most studies to date, BoNT-A injection has been performed more than 6 months after stroke.[4,5]
- However, patients at highest risk for permanent and severe spasticity, may profit from much earlier treatment.[6,7]
- Muscle tone changes in the affected limb may already be apparent within 2 weeks post stroke; however, studies on the time frame for presentation of post-stroke spasticity symptoms are scarce.[2,8–9]

Objective

- The ONTIME pilot study aims to evaluate whether early treatment delays development of disabling (symptomatic) spasticity as a result of post-stroke spasticity.

Methods

Study design

- The ONTIME study (NCT02321436) was a 28-week, phase IV, randomised (2:1; abobotulinumtoxinA [Dysport®, aboBoNT-A] 500 U:placebo), double-blind, placebo-controlled, exploratory trial to assess the impact of aboBoNT-A injections administered within 2 to 12 weeks post stroke in four countries (Malaysia, Thailand, Singapore and the Philippines; Figure 1).

Table 1. Symptomatic spasticity criteria

Functional domain and measure used	Assessment scale	
Pain Numeric Pain Rating Scale (NPRS)[10]	0 – No pain 1–3 – Mild pain 4–6 – Moderate pain 7–10 – Severe pain (disabling and impacts upon movement)	NPRS ≥4 indicates appearance or reappearance of active symptoms of spasticity
Passive function Likert scale	"In general, how much does spasticity have an impact on the following activities of daily living and/or your rehabilitation programme?" 0 – No impact 1 – Mild impact 2 – Moderate impact 3 – Severe impact	A score ≥1 indicates appearance or reappearance of passive symptoms of spasticity
Active function Likert scale	"In general, how much does spasticity have an impact on the following activities of daily living and/or your rehabilitation program: reaching, grasping, releasing, gripping, holding, bimanual function, manipulating objects, dexterity, fine motor skills, lifting and carrying?" 0 – No impact 1 – Mild impact 2 – Moderate impact 3 – Severe impact	A score ≥1 indicates appearance or reappearance of active symptoms of spasticity
Involuntary movements Likert scale	0 – No involuntary movements 0 – Presence of involuntary movements that have a mild impact on posture and ambulation 0 – Presence of involuntary movements that have a moderate impact on posture and ambulation 0 – Presence of involuntary movements that have a severe impact on posture and ambulation	A score ≥1 indicates appearance or reappearance of involuntary movements

NPRS, Numeric pain rating scale

Inclusion criteria

- National legal adult age and ≤80 years.
- Experienced first ever stroke 2 to 12 weeks prior to baseline.
- Computerised tomography/magnetic resonance imaging scan confirmed ischaemic/haemorrhagic stroke.
- Spasticity, either symptomatic or increased muscle tone (Modified Ashworth Scale [MAS] ≥2).
- MAS score of ≥2 in at least one of four muscle groups (elbow flexors or pronators, wrist flexors or finger flexors).

Figure 1. Study design.

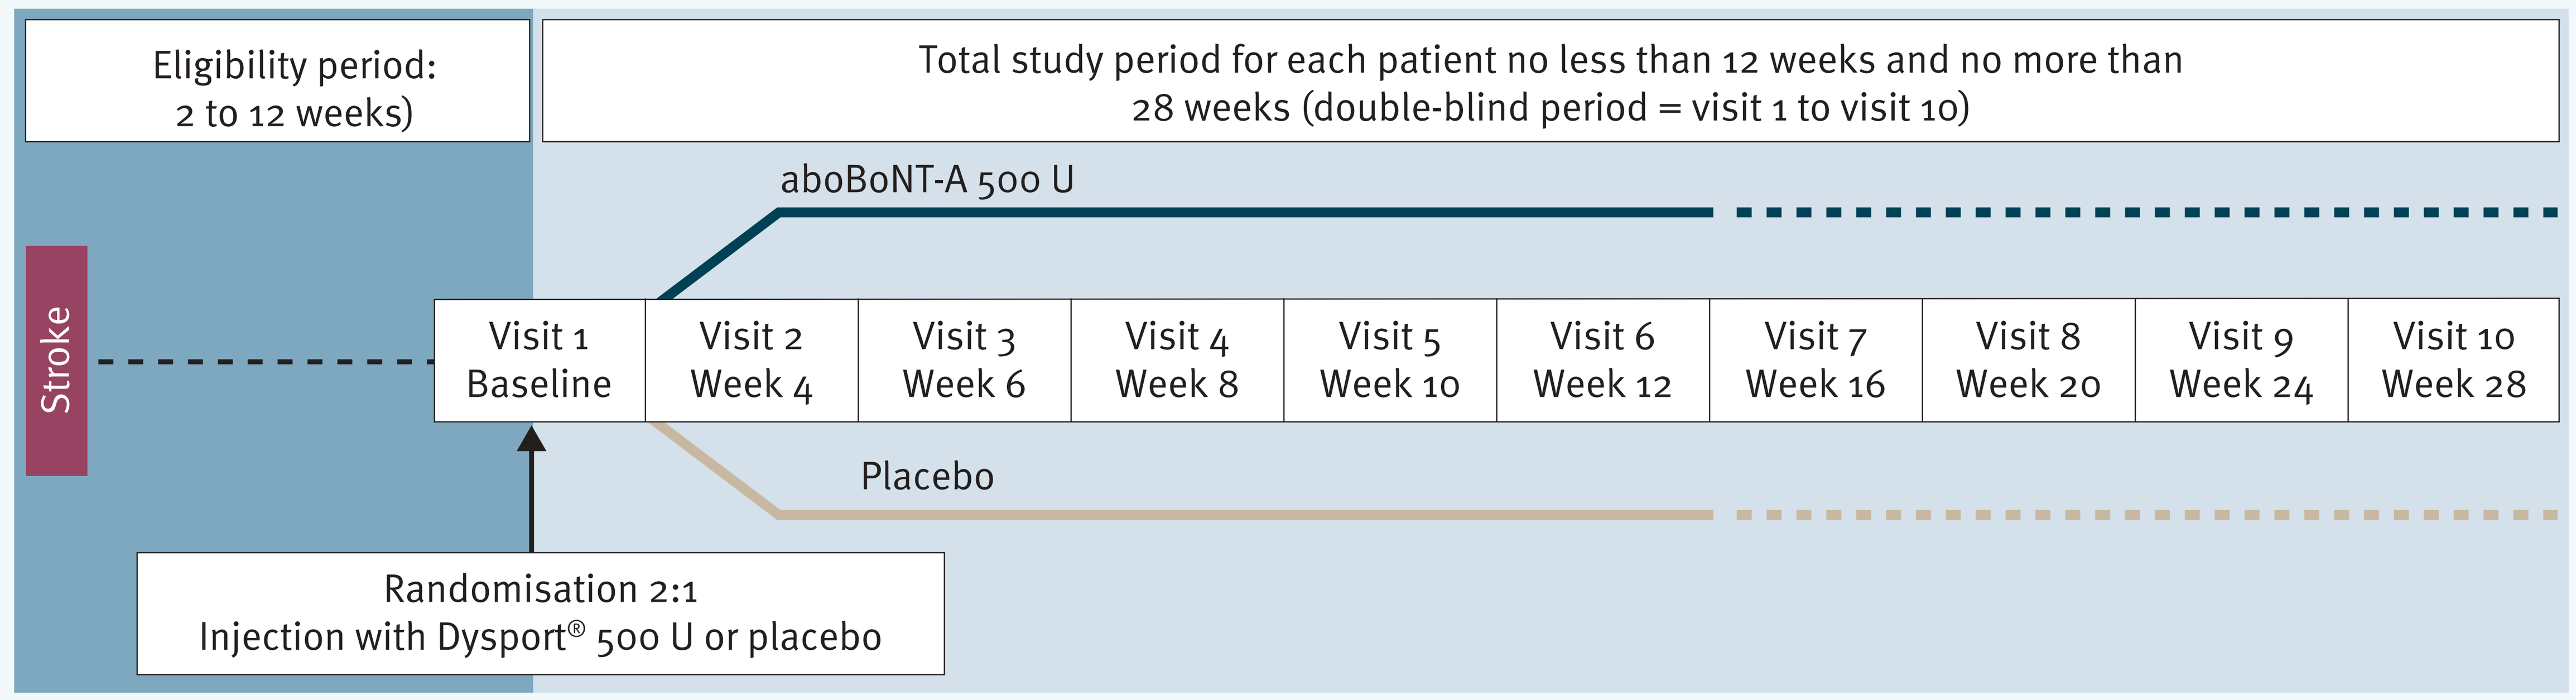
Eligibility period:
2 to 12 weeks)
Total study period for each patient no less than 12 weeks and no more than 28 weeks (double-blind period = visit 1 to visit 10)
aboBoNT-A 500 U
Stroke
Visit 1 Baseline
Visit 2 Week 4
Visit 3 Week 6
Visit 4 Week 8
Visit 5 Week 10
Visit 6 Week 12
Visit 7 Week 16
Visit 8 Week 20
Visit 9 Week 24
Visit 10 Week 28
Placebo
Randomisation 2:1
Injection with Dysport® 500 U or placebo

Outcome measures

- Primary endpoint was the time between the first study injection and the visit at which composite reinjection criteria were met.
- A novel composite index, developed specifically for this study, was used to determine reinjection and defined:
 - MAS score increased to ≥2 in the primary targeted muscle group after first injection.
 - Appearance or reappearance of one of the following signs of symptomatic spasticity: pain, involuntary movements, impaired active or passive function (Table 1).
- Assessment of the impact on patients' global functioning.
- Safety endpoints included the incidence of treatment-emergent adverse events and changes in the patients' physical condition and vital signs, including blood pressure, heart rate, body temperature.

Baseline characteristics

- A cohort of 42 patients (28 aboBoNT-A 500 U, 14 placebo) from Malaysia, the Philippines, Singapore and Thailand were included.
- Mean [SD] age was 59.8 [12.3] years and 78.6% of patients were male (Table 2).
- Symptomatic spasticity was present in 32 patients (76.2%; Table 2).

Table 2. Baseline characteristics and demographics.

Characteristic	aboBoNT-A n=28	Placebo n=14	All subjects N=42
Age, years (SD)	61.5 (13.3)	56.5 (9.7)	59.8 (12.3)
Male, %	82.1	71.4	78.6
Country, n (%)			
Malaysia	3 (10.7)	1 (7.1)	4 (9.5)
Philippines	11 (39.3)	4 (28.6)	15 (35.7)
Singapore	9 (32.1)	5 (35.7)	14 (33.3)
Thailand	5 (17.9)	4 (28.6)	9 (21.4)
Spasticity at baseline (%)			
Symptomatic*	78.6	71.4	76.2
Asymptomatic†	21.4	28.6	23.8

*Status of subject is symptomatic if at least one of the following items was met: passive/active or involuntary movements (Likert score >0) and NPRS score ≥4. †Status of subject is asymptomatic if score is equal to 0 for passive/active functions and involuntary movements, NPRS score <4. aboBoNT-A, abobotulinumtoxinA; SD, standard deviation

Table 3. Disease history

	aboBoNT-A n=28	Placebo n=14	All subjects N=42
Type of stroke, n (%)			
Ischaemic	20 (71.4)	10 (71.4)	30 (71.4)
Haemorrhagic	8 (28.6)	4 (28.6)	12 (28.6)
Mean time since stroke diagnosis (weeks)	6.18	6.52	6.29
Arm affected			
Left, n (%)	16 (57.1)	8 (57.1)	24 (57.1)
Right, n (%)	12 (42.9)	6 (42.9)	18 (42.9)
Both, n (%)	0	0	0

aboBoNT-A, abobotulinumtoxinA

- Mean [SD] time since stroke was 6.29 [2.73] weeks and the most common type of stroke was ischaemic (71.4%) (Table 3).
- Elbow flexors were selected as the primary target muscle group in 73.8% of cases, followed by wrist flexors (14.3%) and finger flexors (2.4%; Table 4).
- At baseline, a Likert scale score of 2 (moderate impact) for passive and active function was more common in aboBoNT-A patients compared with placebo (passive function 39.3 vs. 7.1%, active function 35.7 vs. 21.4%, respectively; Table 4).
- A Likert scale of 3 (severe impact) was recorded more often in the placebo group than the aboBoNT-A group for passive function (14.3 vs. 10.7%), active function (21.4 vs. 14.3%) and involuntary movements (7.1 vs 3.6%), respectively.
- Mean NPRS rating was 3.1 in both groups (Table 4).

Table 4. Presence of symptomatic spasticity at baseline

	aboBoNT-A n=28	Placebo n=14	All subjects N=42
Primary targeted muscle group (%)			
Elbow flexors	71.4	78.6	73.8
Elbow pronators	14.3	0	9.5
Wrist flexors	14.3	14.3	14.3
Finger flexors	0	7.1	2.4
Modified Ashworth Scale score	2.11	2.14	2.12
Likert scale score			
Passive function, n (%)			
0 – No impact	8 (28.6)	7 (50.0)	15 (35.7)
1 – Mild impact	6 (21.4)	4 (28.6)	10 (23.8)
2 – Moderate impact	11 (39.3)	1 (7.1)	12 (28.6)
3 – Severe impact	3 (10.7)	2 (14.3)	5 (11.9)
Active function, n (%)			
0 – No impact	11 (39.3)	7 (50.0)	18 (42.9)
1 – Mild impact	3 (10.7)	1 (7.1)	4 (9.5)
2 – Moderate impact	10 (35.7)	3 (21.4)	13 (31.0)
3 – Severe impact	4 (14.3)	3 (21.4)	7 (16.7)
Involuntary movements, n (%)			
0 – No impact	15 (53.6)	7 (50.0)	22 (52.4)
1 – Mild impact	6 (21.4)	3 (21.4)	9 (21.4)
2 – Moderate impact	6 (21.4)	3 (21.4)	9 (21.4)
3 – Severe impact	1 (3.6)	1 (7.1)	2 (4.8)
NPRS score, n (SD)	3.1 (3.2)	3.1 (2.7)	3.1 (3.0)

aboBoNT-A, abobotulinumtoxinA; MAS, Modified Ashworth scale; NPRS, Numeric Pain Rating Scale; SD, standard deviation

Conclusions

- Symptomatic spasticity was present in 76.2 % of cases at 2 to 12 weeks after stroke.
- This indicates that current standard clinical practice to start spasticity treatment with botulinum toxin type A ≥3 months post stroke may not be optimal and there is rationale for earlier treatment.
- The ONTIME pilot study will investigate the impact of early post stroke treatment with abobotulinumtoxinA.
- Results from this study will provide insight into the optimal delivery of post stroke patient management.

References

1. Zorowitz RD, *et al. Neurology* 2013;80(3) Suppl 2:S45–52.
2. Kong KH, *et al. J Rehabil Med* 2010;42:453–7.
3. Opheim A, *et al. Am J Phys Med Rehabil* 2014;93(10):884–96
4. Gracies JM, *et al. Lancet Neurol* 2015;14:992–1001.
5. Ozcakir S, *et al. Clin Med Res* 2007;5:132–138.
6. Wissel J, *et al. J Rehabil* 2009;41(1):13-25.doi: 10.2340/16501977-0303.
7. Rosales R, *et al. J Neurol Sci* 2016;371:6–14.
8. Welmer AK, *et al. Eur J Neurol* 2010;17:720–5.
9. Wissel J, *et al. J Neurol* 2010;257:1067–72.
10. McCaffery M, *et al.* http://www.rehabmeasures.org/PDF%20Library/Numeric%20Pain%20Rating%20Scale%20Instructions.pdf.

Disclosures

RR, KKH, KJG and WK have received consultancy fees from Ipsen Pharma. JB and PM are full-time employees of Ipsen Pharma.

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield Company, for medical writing support in preparing this poster, funded by Ipsen.

Presented at TOXINS, 18–21 January 2017, Madrid, Spain.

This study was sponsored by Ipsen