A comparison of goal selection and achievement between botulinum toxin A (BoNT-A) naïve and non-naïve patients in the Upper Limb International Spasticity (ULIS)-II study

Christina Marciniak[1], Jovita Balcaitiene[2], Jorge Jacinto[3], Klemens Fheodoroff[4], Stephen Ashford[5], Pascal Maisonobe[2], Lynne Turner-Stokes[5]

[1]Northwestern University and Rehabilitation Institute of Chicago, Chicago, IL, USA; [2]Ipsen Pharma, Boulogne-Billancourt, France; [3]Centro de Medicina de Reabilitação de Alcoitão, Serviço de Reabilitação de Adultos, Estoril, Portugal; [4]Department of Neurorehabilitation, Gailtal-Klinik, Hermagor, Austria; [5]King's College London School of Medicine, Palliative Care, Policy and Rehabilitation and Regional Hyper-acute Rehabilitation Unit, Northwick Park Hospital, London, UK

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by TOXINS

Introduction

- Intramuscularly injected botulinum toxin A (BoNT-A) is effective in decreasing muscle tone (resistance to passive movement) in patients with upper limb spasticity (ULS).[1]
- Patient involvement is central in making treatment decisions.[2]
- Goal attainment scaling (GAS) is an effective method for facilitating patient goal setting and quantifying goal achievement.[3]
- Differences in goal selection between patients naïve and non-naïve to BoNT-A treatmentis still unknown.

Objective

- This *post hoc* analysis aimed to evaluate differences in goal selection and achievement between BoNT-A naïve and non-naïve patients in the ULIS-II study.

Methods

- A population of 456 patients were included in this sub analysis.
- Details of the ULIS-II methodology and results have been previously published.[4,5]

Outcome measures

- Goals were assigned to goal areas: active function, impairment, involuntary movement, mobility, pain and passive function.
- Goals were rated using GAS, which was applied using a six-point verbal scale.[3,6]

Box 1. The Upper Limb International Spasticity (ULIS) programme

- The ULIS programme evaluates patient-centred outcomes in adults receiving BoNT-A for post-stroke ULS management in real-life clinical practice.[4]
- The ULIS-II study was an 18 month, international, non-interventional study in 456 adults conducted at 84 centres in 22 countries.
- This study demonstrated a clinically significant impact of treatment with BoNT-A on attainment of patient-centred goals, measured by GAS.[3]
 - The most common primary goals were passive function, active function and impairment.[4]
 - 80% of patients achieved their primary goals.

Results

- The mean age of non-naïve patients was 59.1 years and naïve, 55.5 years (p=0.0081).
- Non-naïve patients had a significantly longer time since stroke than naïve patients (51.0 months vs. 18.4 months, p<0.0001; Table 1).
- Motor impairment, composite contracture and Modified Ashworth Scale scores were not different between groups (Table 1).

Table 1. Baseline characteristics

Parameter	Naïve (N=149)	Non-naïve (N=307)	Difference	95% CI	*p*-value
Sex (males), %	67.8	53.8	−14.0	−0.23; −0.05	0.0046
Mean age, years (SD)	59.1 (12.4)	55.5 (14.0)	−3.6 (13.5)	−6.2; −0.9	0.0081
Median time since onset of stroke (months)	18.4	51.0	−27.3	−35.1; −20.1	<0.0001
Mean motor impairment score (SD)	4.2 (1.3)	4.4 (1.3)	0.2 (1.26)	−0.1; 0.4	0.1265
Mean arm raising score (SD)	1.88 (0.75)	1.90 (0.78)	0.02 (0.77)	−0.13; 0.17	0.7857
Mean hand function score (SD)	2.37 (0.70)	2.53 (0.64)	0.16 (0.66)	0.03; 0.29	0.0155
Mean composite contracture score (SD)	4.4 (3.4)	4.8 (3.7)	0.4 (3.6)	−0.3; 1.1	0.3853
Mean proximal score (SD)	2.0 (1.6)	2.1 (1.8)	0.2 (1.8)	−0.2; 0.5	0.4603
Mean distal score (SD)	2.5 (2.1)	2.7 (2.1)	0.2 (2.1)	−0.2; 0.6	0.2735
Mean MAS score (SD)	10.9 (3.2)	11.1 (3.3)	0.25 (3.3)	−0.4; 0.9	0.4307

CI, confidence interval; MAS, Modified Ashworth Scale; SD, standard deviation.

Figure 1. Primary goal selection of naïve compared with non-naïve patients.

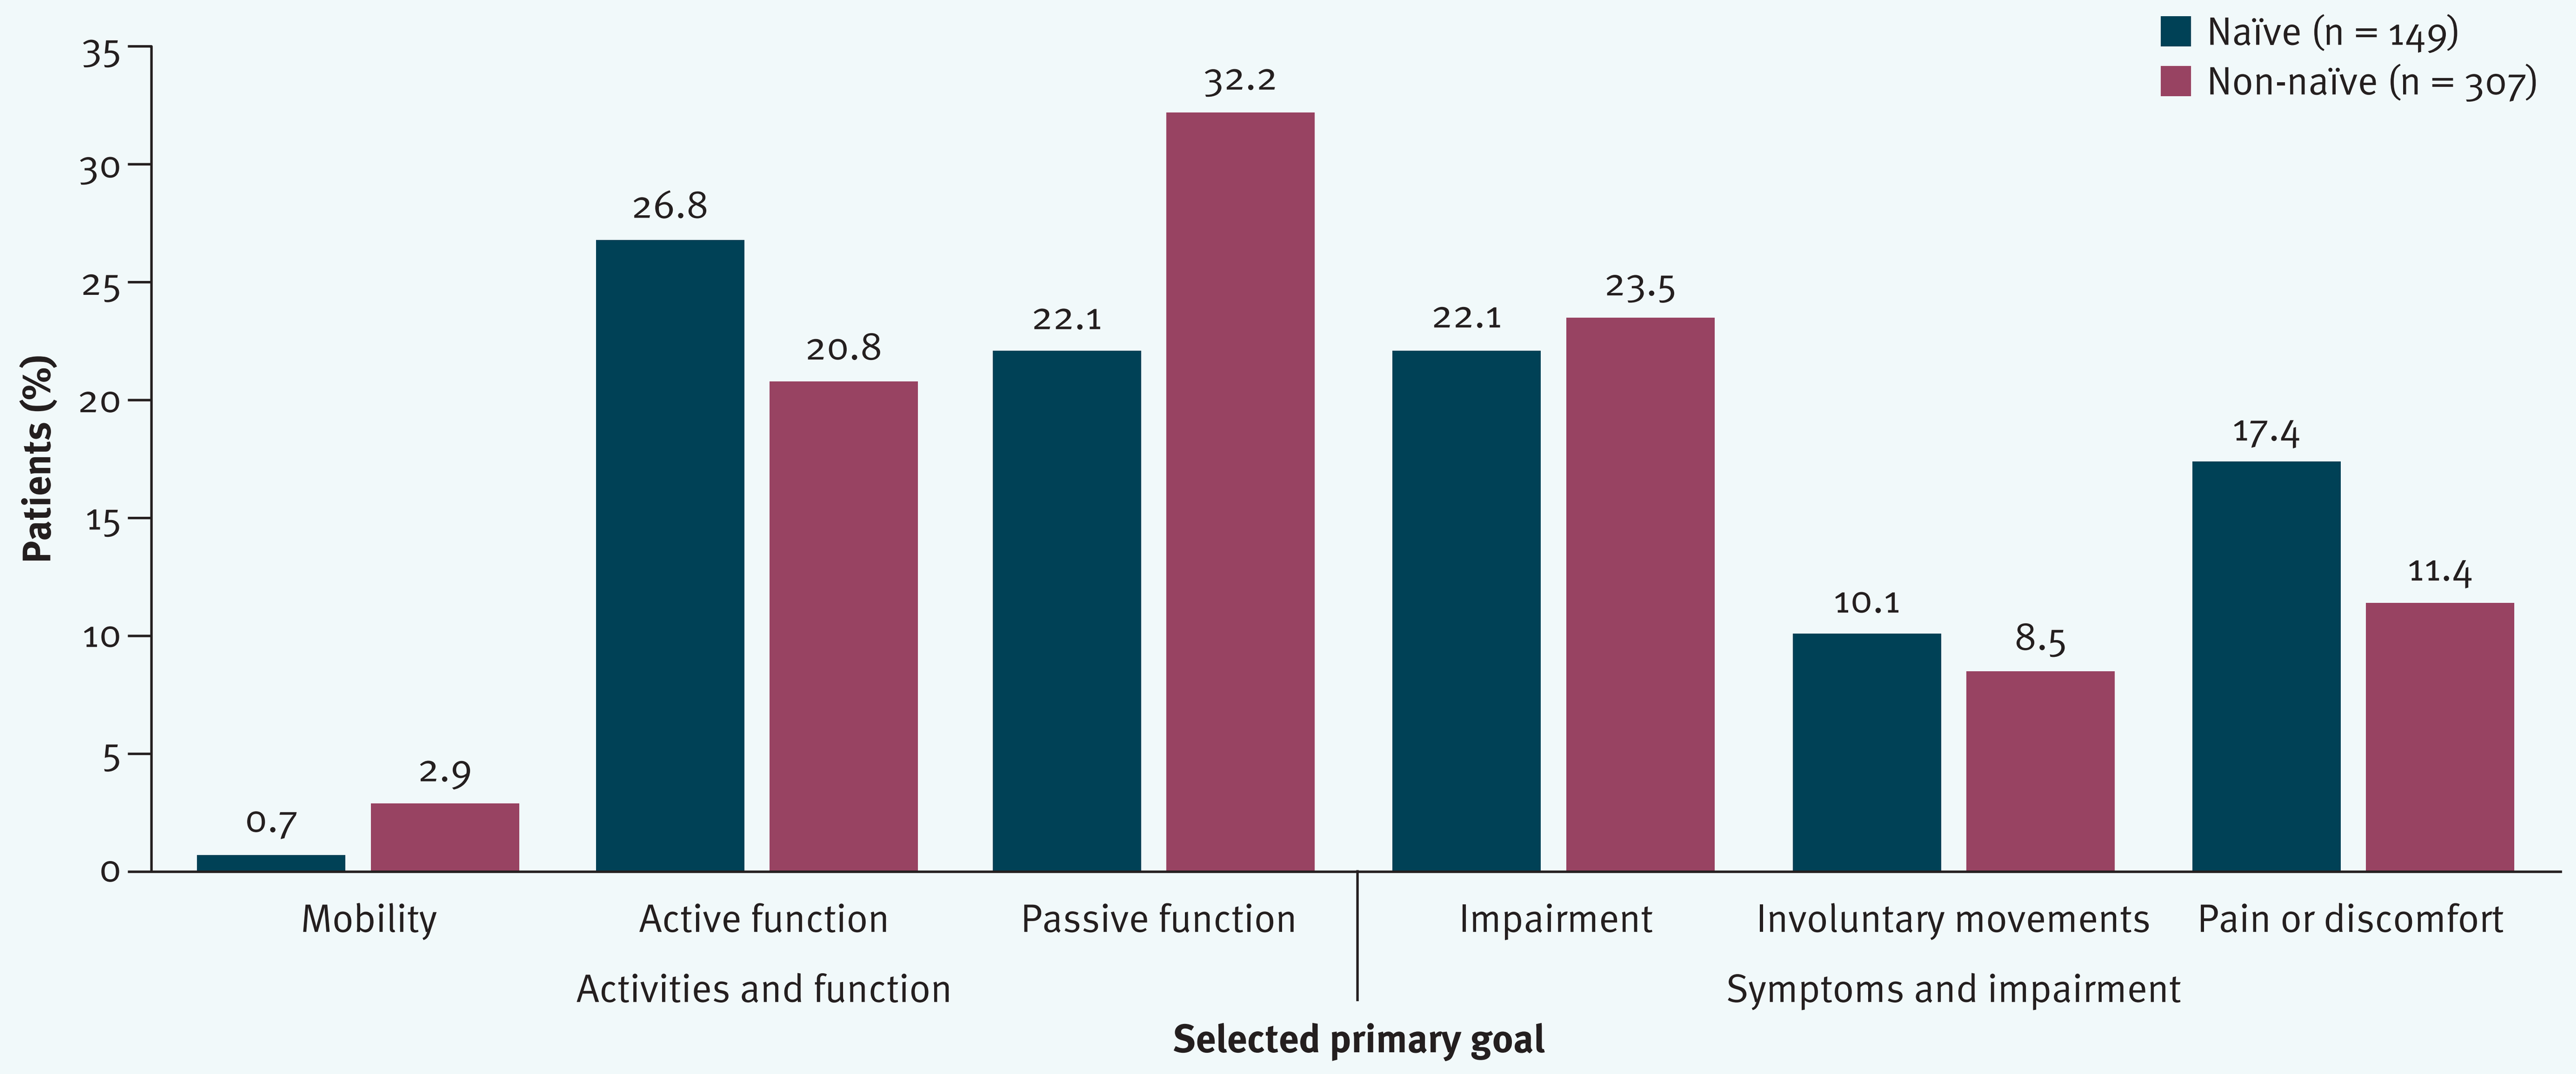
Naïve (n = 149)
Non-naïve (n = 307)
Patients (%)
0
5
10
15
20
25
30
35
0.7
2.9
26.8
20.8
22.1
32.2
22.1
23.5
10.1
8.5
17.4
11.4
Mobility
Active function
Passive function
Impairment
Involuntary movements
Pain or discomfort
Activities and function
Symptoms and impairment
Selected primary goal

Figure 2. Primary goals achieved by naïve compared with non-naïve patients.

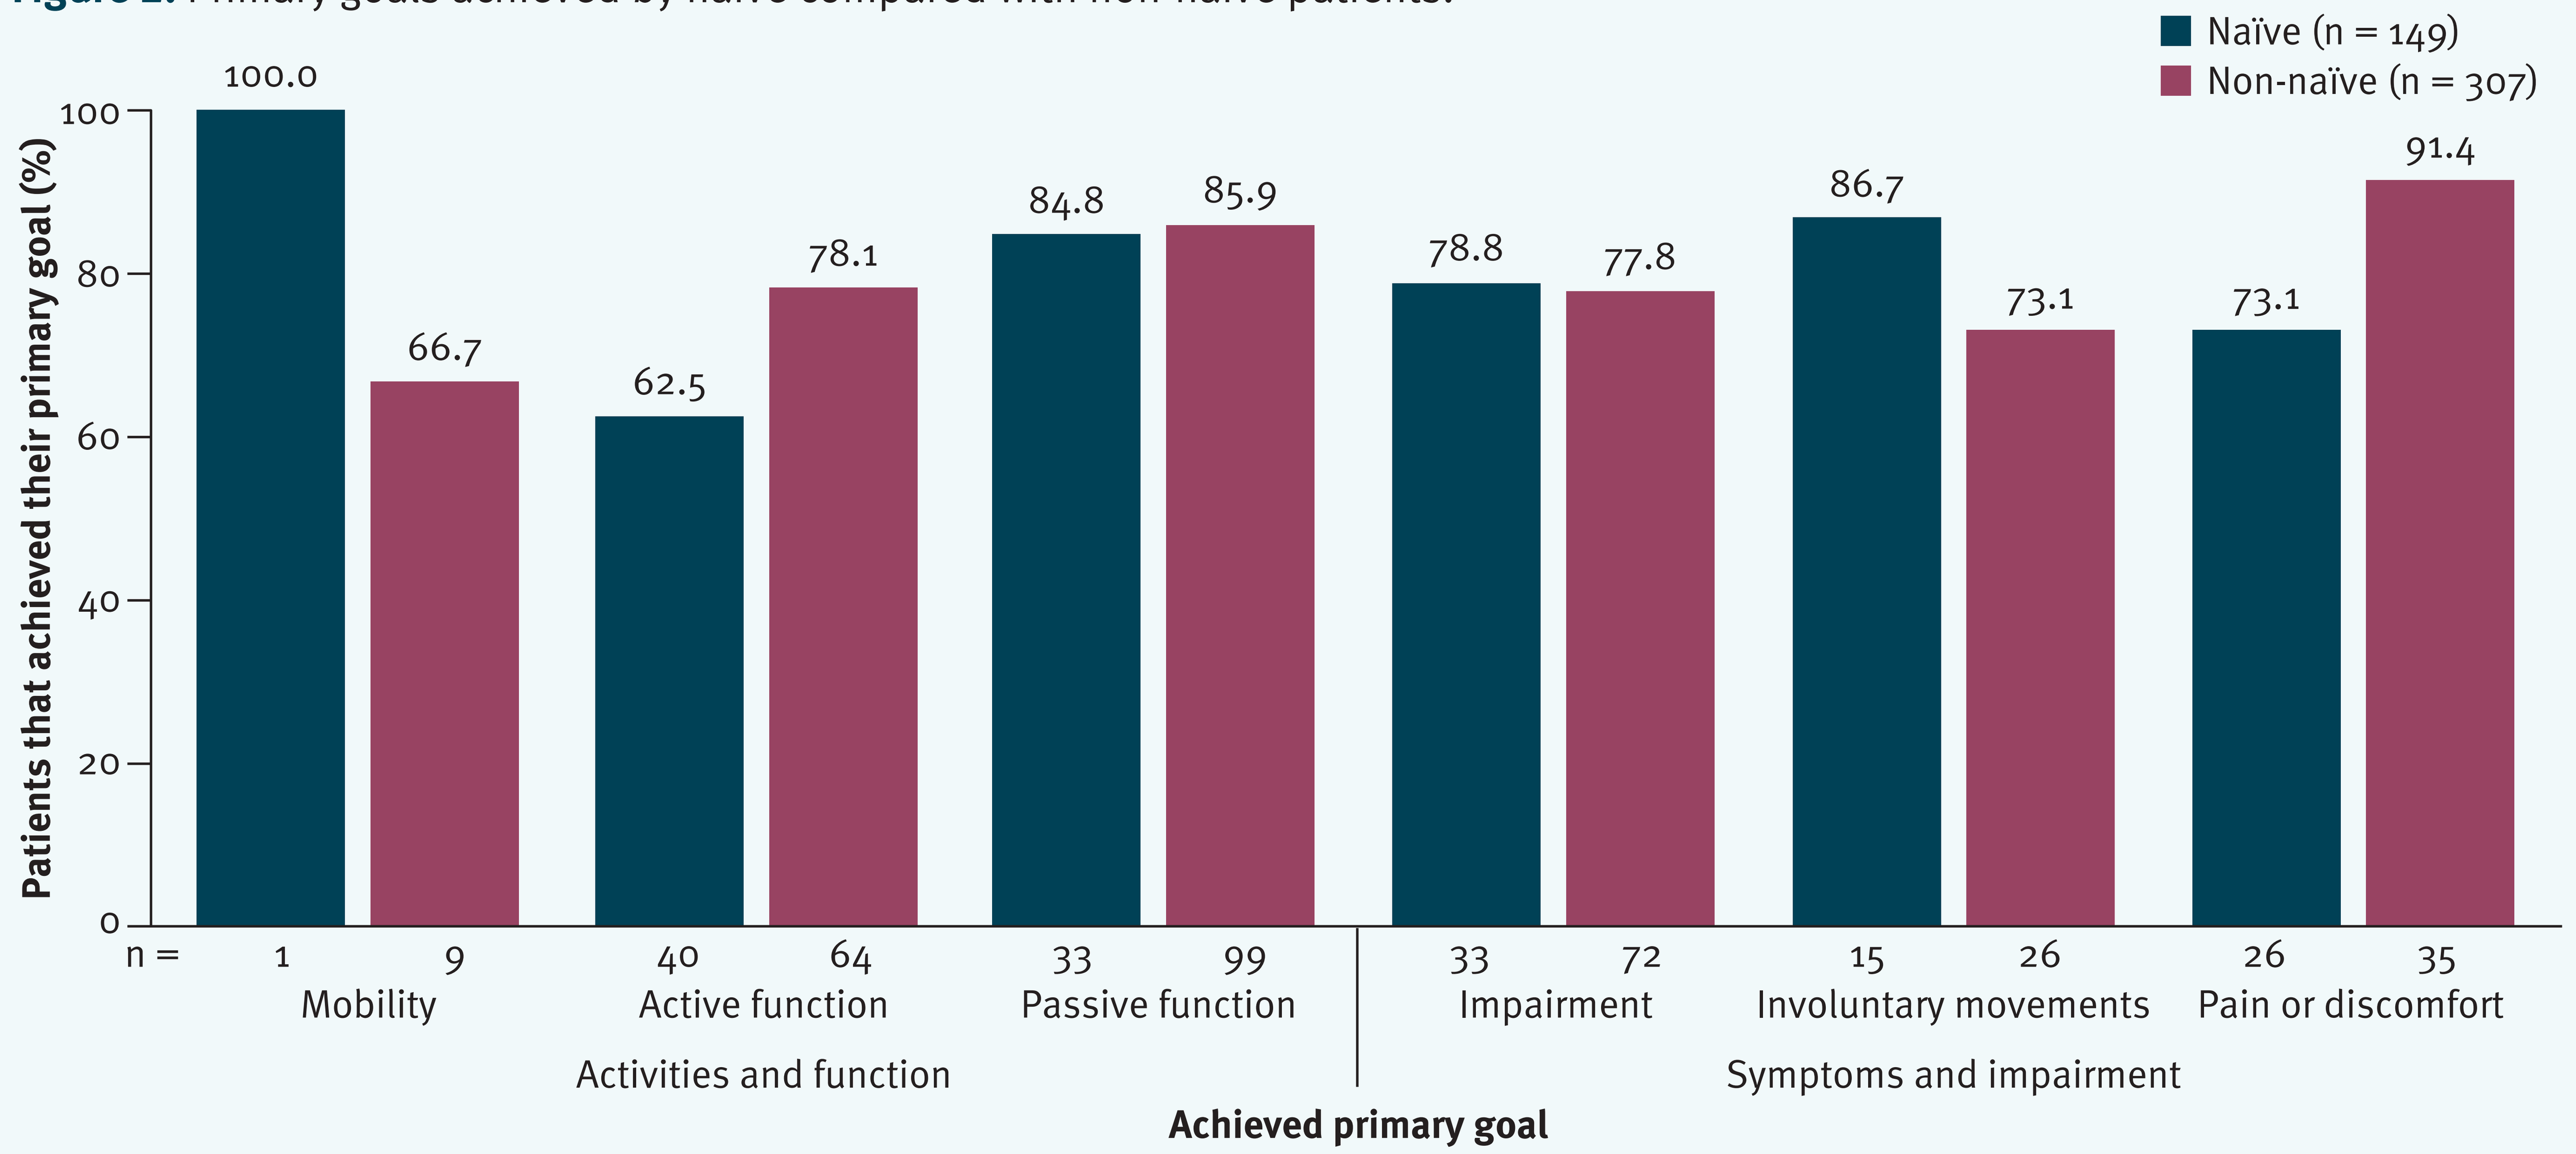
Naïve (n = 149)
Non-naïve (n = 307)
Patients that achieved their primary goal (%)
0
20
40
60
80
100
100.0
66.7
62.5
78.1
84.8
85.9
78.8
77.8
86.7
73.1
73.1
91.4
n =
1
9
40
64
33
99
33
72
15
26
26
35
Mobility
Active function
Passive function
Impairment
Involuntary movements
Pain or discomfort
Activities and function
Symptoms and impairment
Achieved primary goal

n=number of patients who selected a primary goal per goal area naïve/non-naïve

Efficacy

Primary goals

- Proportion of patients achieving their primary goal (95% confidence interval [CI]):
 - Naïve patients: 75.8% [68.2; 82.5].
 - Non-naïve patients: 81.4% [76.6; 85.6].
- Naïve patients more often selected active function (naïve: 26.8% vs. non-naïve: 20.8%) and pain-related (17.4 vs. 11.4%) primary goals (Figure 1).
 - In naïve compared with non-naïve patients, goal achievement rates were lower in the active function (62.5 vs. 78.1%) and pain reduction (73.1 vs. 91.4%) goal areas, respectively (Figure 2).
- Non-naïve patients more often selected passive function (naïve: 22.1 vs. non-naïve: 32.2%) primary goals (Figure 1).
- Goal achievement rates were comparable between naïve and non-naïve patients (Figure 2).

Secondary goals

- Patterns of goal setting and achievement were similar between primary and secondary goals.

Conclusions

- This was the first analysis to investigate differences in goal selection between botulinum toxin A naïve and non-naïve patients.
- Botulinum toxin A naïve patients more often selected active function and pain-related goals compared with non-naïve patients, suggesting botulinum toxin A naïve patients have higher expectations of changes in active function following treatment compared with non-naïve patients.
- Patient–clinician communication is important and can help to manage patients' expectations.

References

1. Gracies JM, *et al. Lancet Neurol* 2015;14:992–1001.
2. Brown M, *et al. Disabil Rehabil* 2014;36:1020–6.
3. Turner-Stokes L. *Clin Rehabil* 2009;23:362–70.
4. Turner-Stokes L, *et al. BMJ Open* 2013;3:e002771.
5. Turner-Stokes L, *et al. BMJ Open* 2013;3:e002230.
6. Williams RC and Stieg RL. *Clin J Pain* 1986;2:219–28.

Disclosures

CM, LTS, SA, JJ and KF have all received honoraria and conference attendance fees from Ipsen whilst undertaking this research, but have no financial interest in any of the materials mentioned in this poster. PM and JB are employees of Ipsen.

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield Company, for medical writing support in preparing this poster, funded by Ipsen.

Presented at TOXINS, 18–21 January 2017, Madrid, Spain.

This study was sponsored by Ipsen