Duration of effect of abobotulinumtoxinA (Dysport®) in adult patients with upper limb spasticity (ULS) post stroke or traumatic brain injury

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by TOXINS

Jean-Michel Gracies[1], Allison Brashear[2], Christina Marciniak[3], Robert Jech[4], Marta Banach[5], Philippe Marque[6], Anne-Sophie Grandoulier[7], Claire Vilain[7], Philippe Picaut[7]

[1]EA 7377 BIOTN, Université Paris-Est, Hospital Albert Chenevier-Henri Mondor, Service de Rééducation Neurolocomotrice, Créteil, France; [2]Wake Forest School of Medicine, Department of Neurology, Winston-Salem, NC, USA; [3]Northwestern University and Rehabilitation Institute of Chicago, Chicago, IL, US; [4]Department of Neurology, First Faculty of Medicine, Charles University and General Faculty Hospital, Prague, Czech Republic; [5]Department of Neurology, Jagiellonian, University Medical College, Krakow, Poland; [6]Rangueil Hospital, Physical Medicine Rehabilitation, Toulouse, France; [7]Ipsen, Les Ulis, France

INTRODUCTION

- The benefits of a single injection cycle of abobotulinumtoxinA (aboBoNT-A; Dysport®) on upper limb spasticity (ULS) post stroke or traumatic brain injury (TBI) have been well established,[1] but there are limited data on the long-term efficacy of repeat botulinum toxin A (BoNT-A) injection in ULS.
- Current BoNT-A prescribing Information indicates administration should not be performed in intervals of less than 12 weeks,[2,3] but few studies have assessed the time to retreatment during repeat injections of BoNT-A.
- In a recent double-blind (DB) study, followed by an open-label (OL) extension, single[4] and repeat injections[5,6] of aboBoNT-A demonstrated efficacy and a favourable safety profile in adult patients with ULS.

METHODS

Objective

- This analysis aimed to evaluate the duration of effect of aboBoNT-A injections over repeated treatment cycles (TCs) in hemiparetic patients with ULS.

Study design

- Phase 3, international, multicentre, DB, randomised, single-cycle study of aboBoNT-A in adults with ULS, followed by a long-term OL, multiple-cycle extension study (Figure 1).
- OL study patients received up to four additional TCs over a maximum of 1 year (number of injections over this fixed period varied from one patient to another according to patient needs).

Inclusion criteria

- DB criteria as previously described:[4]
 - Hemiparetic adults aged between 18 and 80 years, ≥6 months post-stroke or -TBI.
 - Modified Ashworth Scale score ≥2 or 3 in primary target muscle group (PTMG) for toxin-naïve and non-naïve subjects, respectively.
 - Disability Assessment Scale score ≥2 in principal target of treatment.
 - Spasticity angle ≥10° in PTMG.
 - Overall Modified Frenchay Scale (evaluating performance of ten upper limb tasks) score of between 1 and 8 (rated on a 10-point scale).
- OL extension:
 - Rollover patients who completed the DB study with no major protocol deviations or ongoing treatment emergent adverse events (TEAEs).
 - Newly recruited patients (ie, patients not participating in DB study) met the same criteria defined for the DB study.

Treatment

- DB study
 - Patients were randomised to receive a single TC of placebo, 500 U aboBoNT-A or 1000 U aboBoNT-A into the PTMG (elbow, wrist or extrinsic finger flexors) and at least two other upper limb muscles, as selected by the investigator (Figure 1).
- OL extension
 - At OL TC 1, patients were administered 1000 U aboBoNT-A. Patients who experienced adverse events in the DB study were administered 500 U or 1000 U aboBoNT-A at the investigator's discretion.
 - At subsequent TC, patients could receive 500 U aboBoNT-A into the shoulder muscles (total body dose ≤1500 U).
 - From TC 3, concomitant 500 U injection of aboBoNT-A into lower limbs was permitted (total body dose ≤1500 U)
- At each TC, retreatment was per investigator's clinical judgement and possible at Weeks 12, 16, 20, and 24.
- TC 4 data not shown due to the limited number of patients who could receive five TCs due to the maximum treatment duration allowed in the protocol.

Figure 1. Study flow.

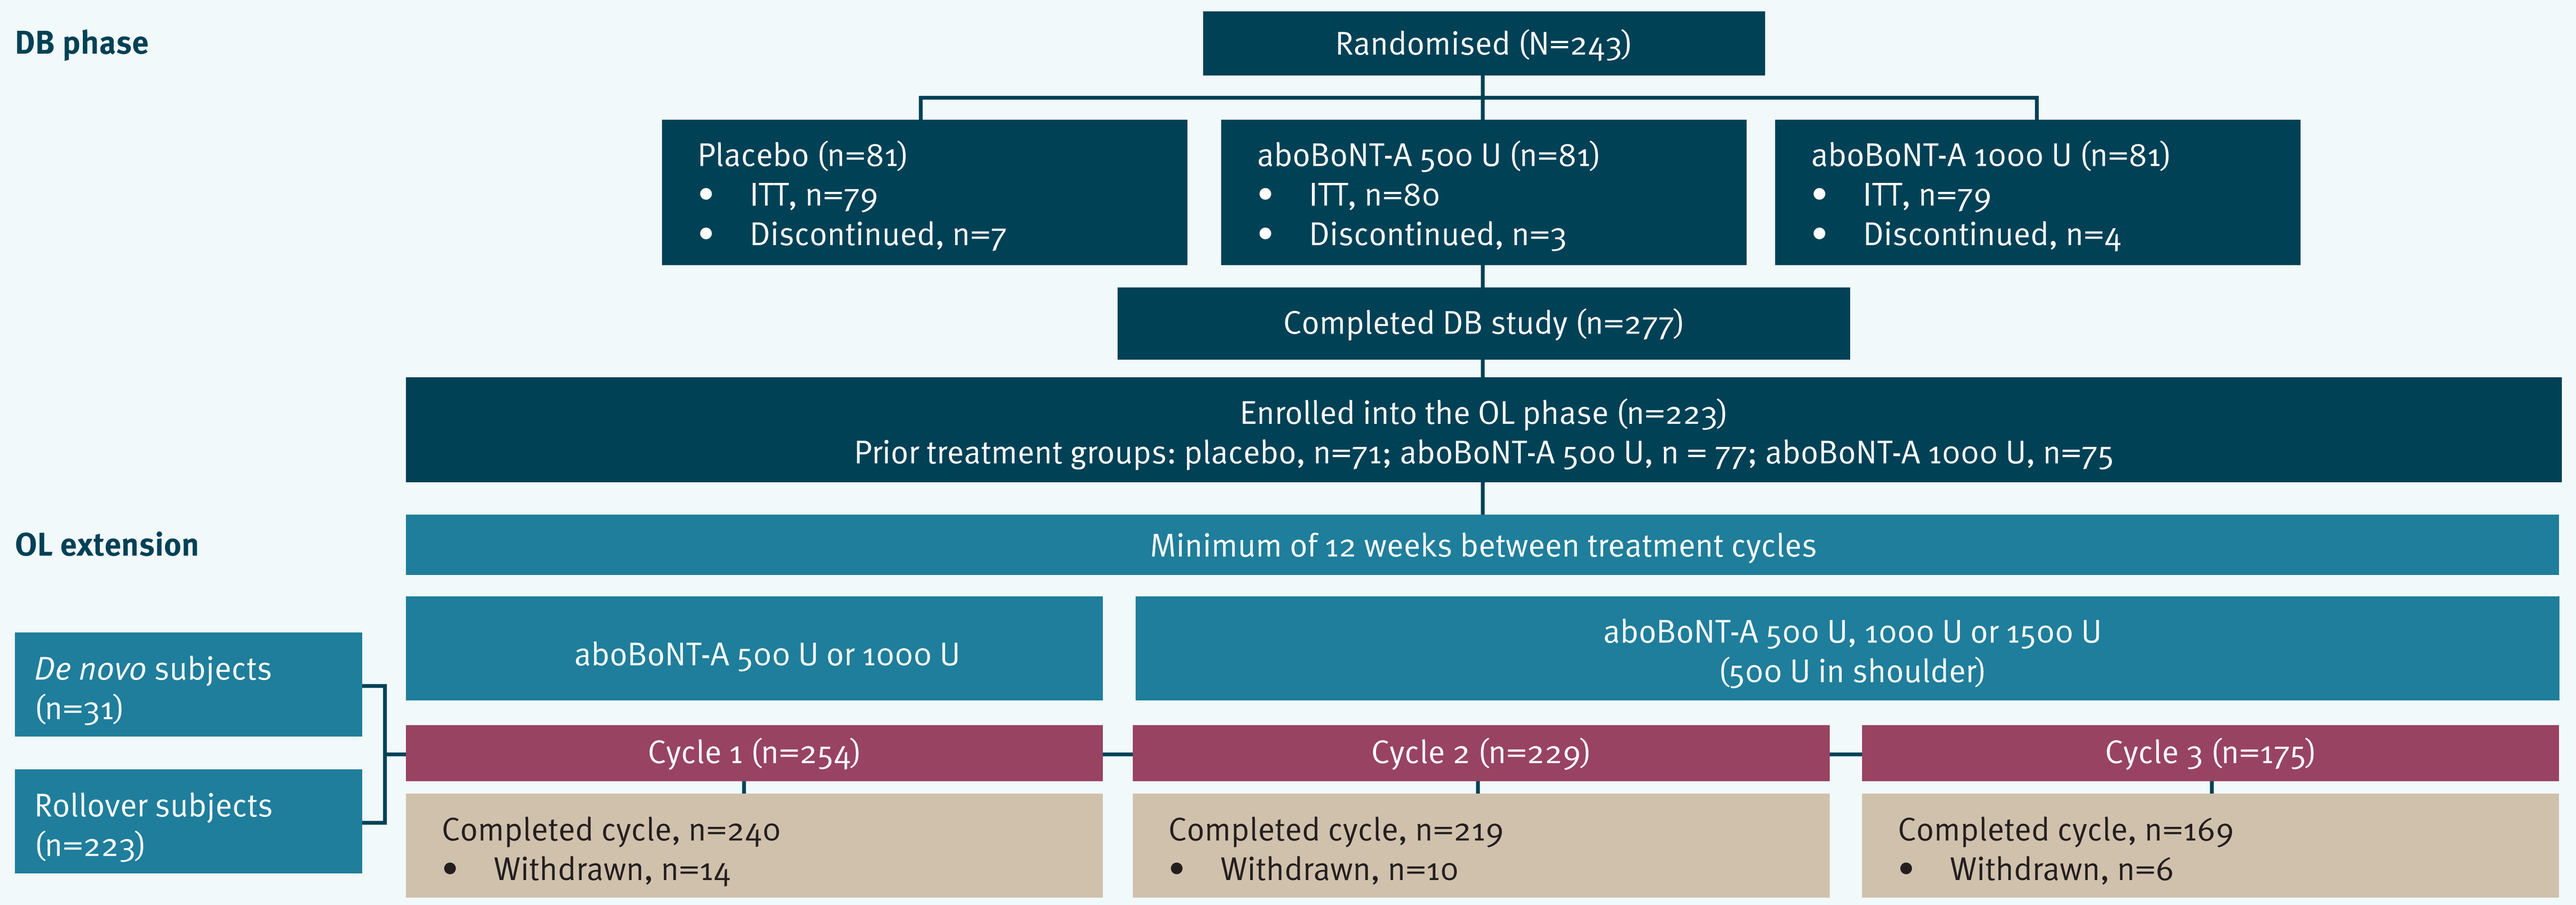
DB phase
Randomised (N=243)
Placebo (n=81)
ITT, n=79
Discontinued, n=7
aboBoNT-A 500 U (n=81)
ITT, n=80
Discontinued, n=3
aboBoNT-A 1000 U (n=81)
ITT, n=79
Discontinued, n=4
Completed DB study (n=277)
Enrolled into the OL phase (n=223)
Prior treatment groups: placebo, n=71; aboBoNT-A 500 U, n = 77; aboBoNT-A 1000 U, n=75
OL extension
Minimum of 12 weeks between treatment cycles
De novo subjects (n=31)
Rollover subjects (n=223)
aboBoNT-A 500 U or 1000 U
aboBoNT-A 500 U, 1000 U or 1500 U (500 U in shoulder)
Cycle 1 (n=254)
Cycle 2 (n=229)
Cycle 3 (n=175)
Completed cycle, n=240
Withdrawn, n=14
Completed cycle, n=219
Withdrawn, n=10
Completed cycle, n=169
Withdrawn, n=6

AboBoNT-A, abobotulinumtoxinA; DB, double-blind; ITT, intention-to-treat; OL, open-label. Patient numbers decreased across OL cycles due to injection intervals of >12 weeks, with a maximum treatment duration of one year per patient. Cycle 4 data not shown due to the limited number of patients who could receive five treatment cycles due to the maximum treatment duration allowed in the protocol.

RESULTS

Baseline characteristics

- Patient demographics and baseline characteristics are presented in Table 1.

Time to retreatment

- Among patients who received aboBoNT-A in the DB study and were treated at TC 1 of the OL extension (n=152), 36.9% were re-injected at Week 16 or later:
 - 17.1% at Week 16, 9.9% at Week 20 and 9.9% at Week 24 or later (Figure 2A).
- For patients retreated at TC 2 of the OL phase (n=229), 34.9% were re-injected at Week 16 or later:
 - 20.1% at Week 16, 7.0% at Week 20 and 7.9% at Week 24 or later (Figure 2B).
- For those retreated at TC 3 of the OL phase (n=175), 24.0% were re-injected at Week 16 or later:
 - 18.9% at Week 16, 3.4% at Week 20 and 1.7% at Week 24 or later (Figure 2C).

Safety results

- A safety summary is provided in Table 2. This shows the percentage of TEAEs decreasing over repeated cycles.
- Overall the most frequently reported TEAEs were fall, muscle weakness and pain in extremity.

Table 1. Baseline demographics and characteristics.

Characteristic	Open-label study (N=258)
Mean age, years (SD)	52.4 (13.9)
Sex, n (%)	
Male	166 (64.3)
Mean weight, kg (SD)	81.3 (18.6)
Affected arm, n (%)	
Left	128 (49.6)
Cause, n (%)	
Stroke	230 (89.1)
TBI	28 (10.9)
Mean time since event, years (SD)	
Stroke	5.1 (4.2)
TBI	9.9 (8.0)
Treatment naïve,* n (%)	116 (45.0)
Primary target muscle group, n (%)	
Extrinsic finger flexors	146 (56.6)
Wrist flexors	43 (16.7)
Elbow flexors	69 (26.7)

*Previous treatment defined as use of botulinum toxin A in the affected limb less than four months prior to study entry.
SD, standard deviation; TBI, traumatic brain injury.

Figure 2. Visit at which patients were re-injected at each cycle.

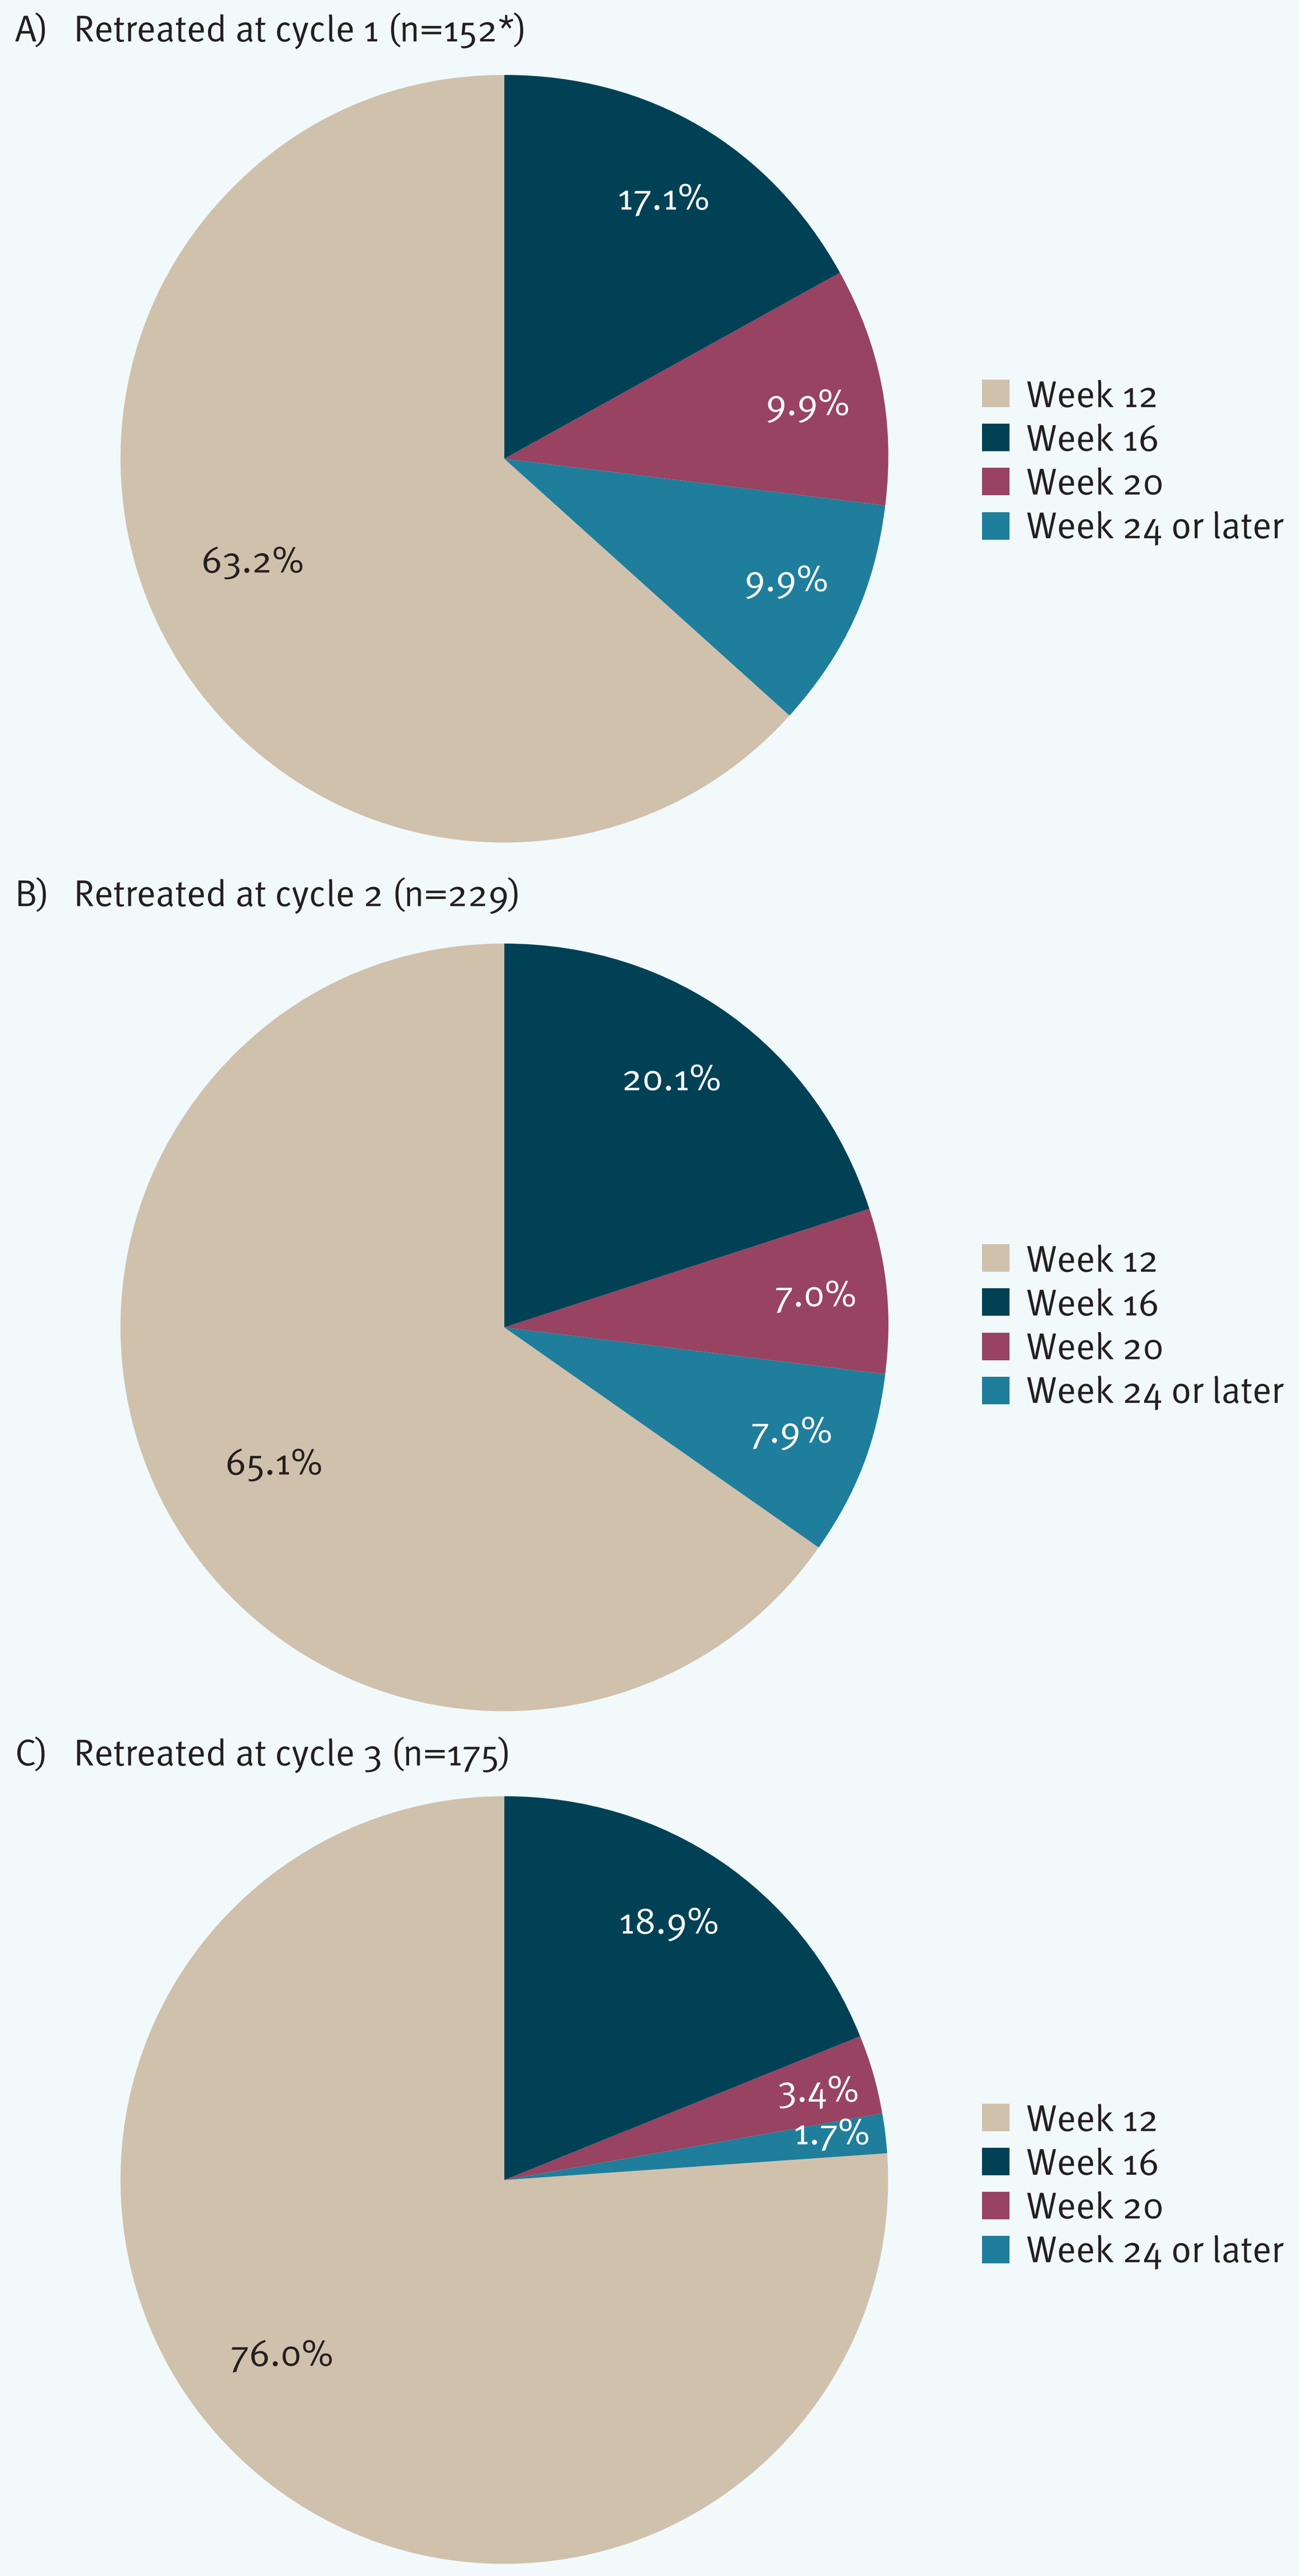
A) Retreated at cycle 1 (n=152*)
17.1%
9.9%
9.9%
63.2%
Week 12
Week 16
Week 20
Week 24 or later
B) Retreated at cycle 2 (n=229)
20.1%
7.0%
7.9%
65.1%
Week 12
Week 16
Week 20
Week 24 or later
C) Retreated at cycle 3 (n=175)
18.9%
3.4%
1.7%
76.0%
Week 12
Week 16
Week 20
Week 24 or later

*Data for patients who received placebo in the DB cycle are not included for retreatment at TC 1.

Table 2. Summary of TEAEs by treatment cycle and TEAEs displayed by ≥2 patients at any cycle.

Events	Cycle 1 (n=254)	Cycle 2 (n=229)	Cycle 3 (n=175)
All doses			
TEAEs	102 (40.2) [194]	62 (27.1) [120]	47 (26.9) [107]
Related TEAEs	18 (7.1) [26]	8 (3.5) [9]	5 (2.9) [6]
TEAE			
Fall	9 (3.5) [9]	7 (3.1) [7]	6 (3.4) [7]
Muscle weakness	9 (3.5) [9]	2 (0.9) [2]	2 (1.1) [2]
Pain in extremity	6 (2.4) [6]	6 (2.6) [8]	2 (1.1) [2]
Nasopharyngitis	4 (1.6) [5]	0	2 (1.1) [2]
Bronchitis	0	1 (0.4) [1]	4 (2.3) [4]
Blood triglycerides increased	0	1 (0.4) [1]	0

TEAE, treatment-emergent adverse event. Values are presented as n (percentage) [number of events].

Conclusions

- These data demonstrate the long duration of effect of abobotulinumtoxinA in patients with spastic paresis injected in the upper limb, with between 24 and 37% of patients re-injected at Week 16 or later across repeated cycles.
- This long duration of clinical effect, leading to longer intervals between injections, would potentially reduce the burden associated with frequency of injections for patients and their caregivers/families.
- These results highlight the need for a tailored approach to treatment of patients with spastic paresis.

References

1. Dashtipour K, *et al. Am J Phys Med Rehabil* 2015;94:229–38.
2. Sheean G, *et al. Eur J Neurol* 2010;17(Suppl 2):74–93.
3. Ipsen Biopharm Ltd. Dysport Full Prescribing Information. 2016; https://www.dysport.com/pdfs/Dysport_Full_Prescribing_Information.pdf.
4. Gracies JM, *et al. Lancet Neurol* 2015;14:992–1001.
5. Brashear A, *et al. Neurology* 2016;86(Suppl P3) abst 307.
6. Marciniak C, *et al. PM&R* 2016;8(9S):S260.

Disclosures

Jean-Michel Gracies has served as a consultant and received research grant support from Allergan, Ipsen, and Merz; he is also an investigator on an Ipsen trial. Allison Brashear has consulted with Ipsen, Allergan, Concert, Revance and WorldMeds. Research is supported by NINDS, Allergan, Ipsen, Merz and Revance. Dr Brashear's conflict of interest is managed by Wake Forest School of Medicine. Christina Marciniak has served on advisory boards for Ipsen and Merz. Dr Marciniak's institution received research support for activities from Ipsen, Allergan, and Merz. Dr Marciniak is an investigator in an Ipsen trial. Robert Jech has received grants from the Czech Science Foundation, Czech Ministry of Health, Czech Ministry of Education and Charles University, Prague; and honoraria from Ipsen for consultations and lectures. Marta Banach has received honoraria from Ipsen for consultations and lectures. Dr Banach is an investigator in an Ipsen trial. Philippe Marque has served on scientific advisory boards and as a speaker for Ipsen, Allergan and Merz, and as a speaker for Almirall. Dr Marque received research support from Ipsen and Merz. Anne-Sophie Grandoulier, Claire Vilain and Philippe Picaut are employees of Ipsen.

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield Company, for medical writing support (funded by Ipsen) in preparing this poster.

Presented at TOXINS, 18–21 January 2017, Madrid, Spain.

This study was sponsored by Ipsen