Duration of effect of abobotulinumtoxinA (Dysport®) in adult patients with lower limb spasticity post stroke or traumatic brain injury

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by TOXINS

Jean-Michel Gracies[1], Alberto Esquenazi[2], Allison Brashear[3], Michael O Dell[4], Thierry Deltombe[5], Senen Gonzalez[6], Francois Constant Boyer[7], Anne-Sophie Grandoulier[8], Claire Vilain[8], Philippe Picaut[8]

[1]EA 7377 BIOTN, Université Paris-Est, Hospital Albert Chenevier-Henri Mondor, Service de Rééducation Neurolocomotrice, Créteil, France; [2]MossRehab & Albert Einstein, Elkins Park, Pennsylvania, USA; [3]Wake Forest School of Medicine, Department of Neurology, Winston-Salem, NC, USA; [4]Weill Cornell Medicine, Department of Rehabilitation Medicine, New York, NY, USA; [5]Service de Médecine Physique et de Réadaptation, Centre Hospitalier Universitaire UCL Namur site Mont-Godinne, Yvoir, Belgium; [6]Royal Melbourne Hospital, Victoria, Australia; [7]Unités de Médecine Physique et de Réadaptation, Hôpital Sébastopol, CHU Reims Champagne Ardenne, Reims, France; [8]Ipsen, Les Ulis, France

INTRODUCTION

- Hemiparesis due to stroke or traumatic brain injury can impair gait related to abnormal passive and active antagonist muscle resistance.[1] Lower limb spasticity (LLS) has been associated with limited mobility and reduced quality of life.[2]
- Botulinum toxin type A (BoNT-A) is an effective treatment for muscle overactivity in spastic hemiparesis[3] and its safety and efficacy in LLS management has been demonstrated.[4,5] AbobotulinumtoxinA (aboBoNT-A; Dysport®) has not received FDA approval for use in adults with LLS.
- In a recent double-blind (DB), single-treatment study in patients with LLS, followed by a long-term open-label (OL) extension study, aboBoNT-A was found to be efficacious with a safety profile in line with previous findings.[3,6]
- Few studies have assessed the time to retreatment and duration of effect of BoNT-A between repeated injection cycles for LLS.
- An additional analysis of the recent DB[3] and OL extension[6] study data is reported here.

METHODS

Objective

- This analysis aimed to evaluate the duration of effect of aboBoNT-A injections over repeated treatment cycles (TCs) in hemiparetic patients with LLS.

Study design

- Phase 3, international, multicentre, prospective, DB, randomised, single-cycle study of aboBoNT A in adults with LLS, followed by a long-term OL multiple-cycle extension study (Figure 1).
- OL study patients received up to four additional TCs over a maximum of one year (number of injections over this fixed period varied from one patient to another according to patient needs).

Inclusion criteria

- DB study:
 - Adults with spastic hemiparesis causing gait deficiency.
 - 18 to 80 years of age, ≥6 months post-stroke or TBI.
 - Modified Ashworth Scale score ≥2 or 3 in the gastrocnemius–soleus complex (GSC) for toxin- and non-naïve patients, respectively.
 - Spasticity angle ≥5° in the GSC (Tardieu scale).
 - Comfortable barefoot walking speed between 0.1 and 0.8 m/s at baseline.
- OL extension:
 - Patients who completed the DB study with no major protocol deviations or ongoing treatment emergent adverse events (TEAEs).

Treatment

- DB study
 - Patients were randomised to receive a single TC of placebo, 1000 U aboBoNT-A or 1500 U aboBoNT-A into both soleus and gastrocnemius muscles and at least one other lower-limb muscle selected by the investigator (Figure 1).
- OL extension
 - At OL TC1, all patients received 1500 U aboBoNT-A.
 - At subsequent TC, patients were administered 1000 U or 1500 U aboBoNT-A at the investigator's discretion.
 - From TC 3, concomitant 500 U injection of aboBoNT-A into upper limb was allowed, (total body dose remained ≤1500 U).

Figure 1. Study flow.

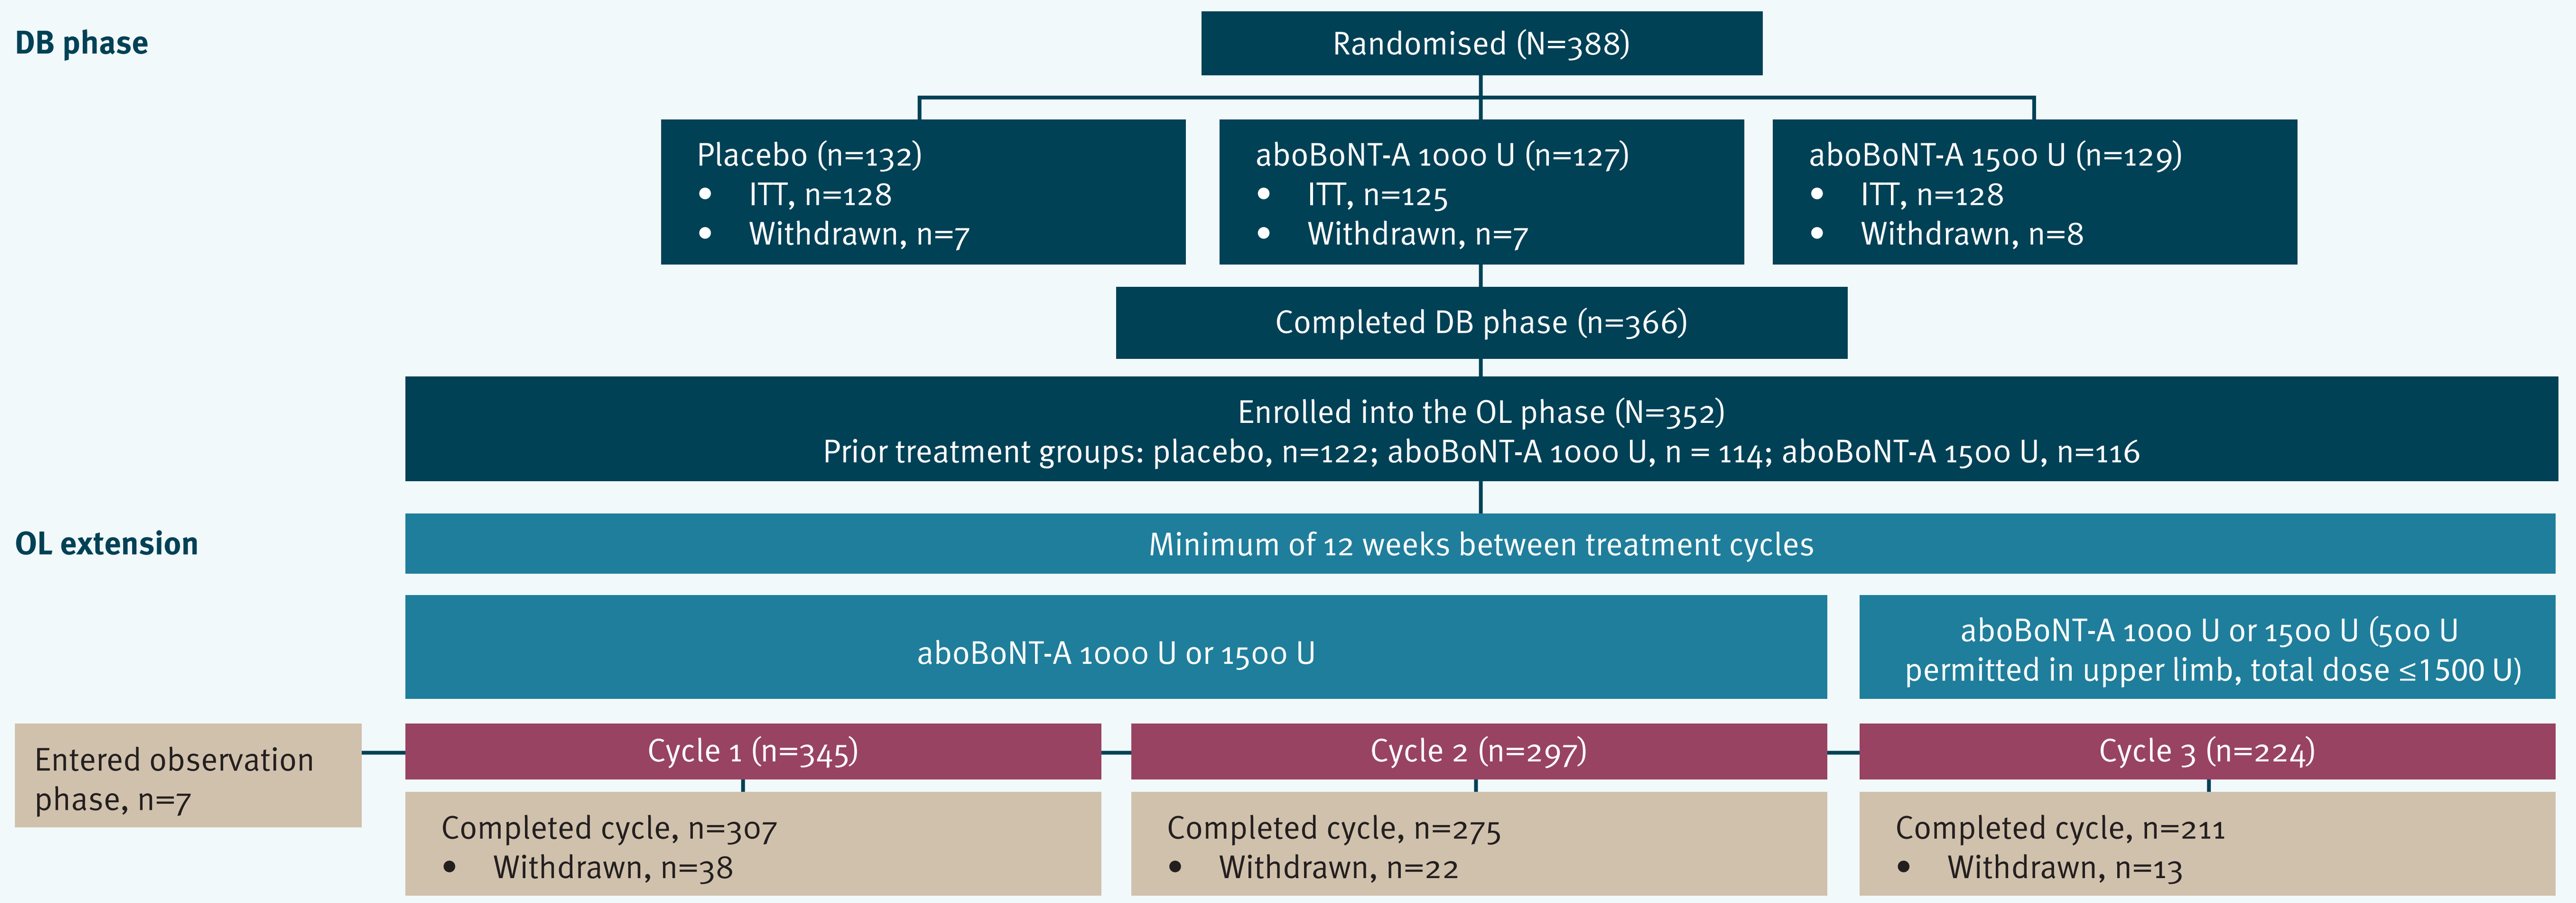

DB phase
Randomised (N=388)
Placebo (n=132)
ITT, n=128
Withdrawn, n=7
aboBoNT-A 1000 U (n=127)
ITT, n=125
Withdrawn, n=7
aboBoNT-A 1500 U (n=129)
ITT, n=128
Withdrawn, n=8
Completed DB phase (n=366)
Enrolled into the OL phase (N=352)
Prior treatment groups: placebo, n=122; aboBoNT-A 1000 U, n = 114; aboBoNT-A 1500 U, n=116
OL extension
Minimum of 12 weeks between treatment cycles
aboBoNT-A 1000 U or 1500 U
aboBoNT-A 1000 U or 1500 U (500 U permitted in upper limb, total dose ≤1500 U)
Entered observation phase, n=7
Cycle 1 (n=345)
Completed cycle, n=307
Withdrawn, n=38
Cycle 2 (n=297)
Completed cycle, n=275
Withdrawn, n=22
Cycle 3 (n=224)
Completed cycle, n=211
Withdrawn, n=13

AboBoNT-A, abobotulinumtoxinA; DB, double-blind; ITT, intention-to-treat; OL, open-label. Patient numbers decreased across OL cycles due to injection intervals of >12 weeks, with a maximum treatment duration of one year per patient. Cycle 4 data not shown due to the limited number of patients who could receive five treatment cycles due to the maximum treatment duration allowed in the protocol.

- At each TC, retreatment was per investigator's clinical judgement and possible at Weeks 12, 16, 20, and 24.
- TC 4 data not shown due to the limited number of patients who could receive five TCs due to the maximum treatment duration allowed in the protocol.

RESULTS

Baseline characteristics

- Patient demographics and baseline characteristics are presented in Table 1.

Time to retreatment

- Among patients who received aboBoNT-A in the DB study and were treated at TC1 of the OL extension (n=224), 20.1% were re-injected at Week 16 or later: 9.8% at Week 16, 4.9% at Week 20, 5.4% at Week 24 or later (Figure 2A).
- For patients retreated at TC2 of the OL phase (n=297), 32.0% were re-injected at Week 16 or later: 16.5% at Week 16, 8.8% at Week 20, 6.7% at Week 24 or later (Figure 2B).
- For those retreated at TC3 of the OL phase (n=224), 15.2% were re-injected at Week 16 or later: 13.0% at Week 16, 1.8% at Week 20 and 0.5% at Week 24 (Figure 2C).

Safety results

- A safety summary is provided in Table 2. This illustrates the percentage of TEAEs decreasing with each cycle.
- Overall, the most frequently reported TEAEs were muscular weakness, falls and pain in the treated extremity.

Table 1. Baseline demographics and patient characteristics.

Characteristic	Open-label study (N=352)
Mean age, years (SD)	53.2 (12.7)
Sex, n (%)	
Male	239 (67.9)
Mean weight, kg (SD)	79.9 (16.5)
Affected leg, n (%)	
Left	194 (55.1)
Cause, n (%)	
Stroke	309 (87.8)
TBI	43 (12.2)
Mean time since event, years (SD)	
Stroke	4.5 (4.8)
TBI	9.2 (10.1)
Treatment naïve,* n (%)	226 (64.2)

*Previous treatment defined as use of botulinum toxin A in the affected limb less than four months prior to study entry.
SD, standard deviation; TBI, traumatic brain injury.

Figure 2. Visit at which patients were re-injected at each cycle.

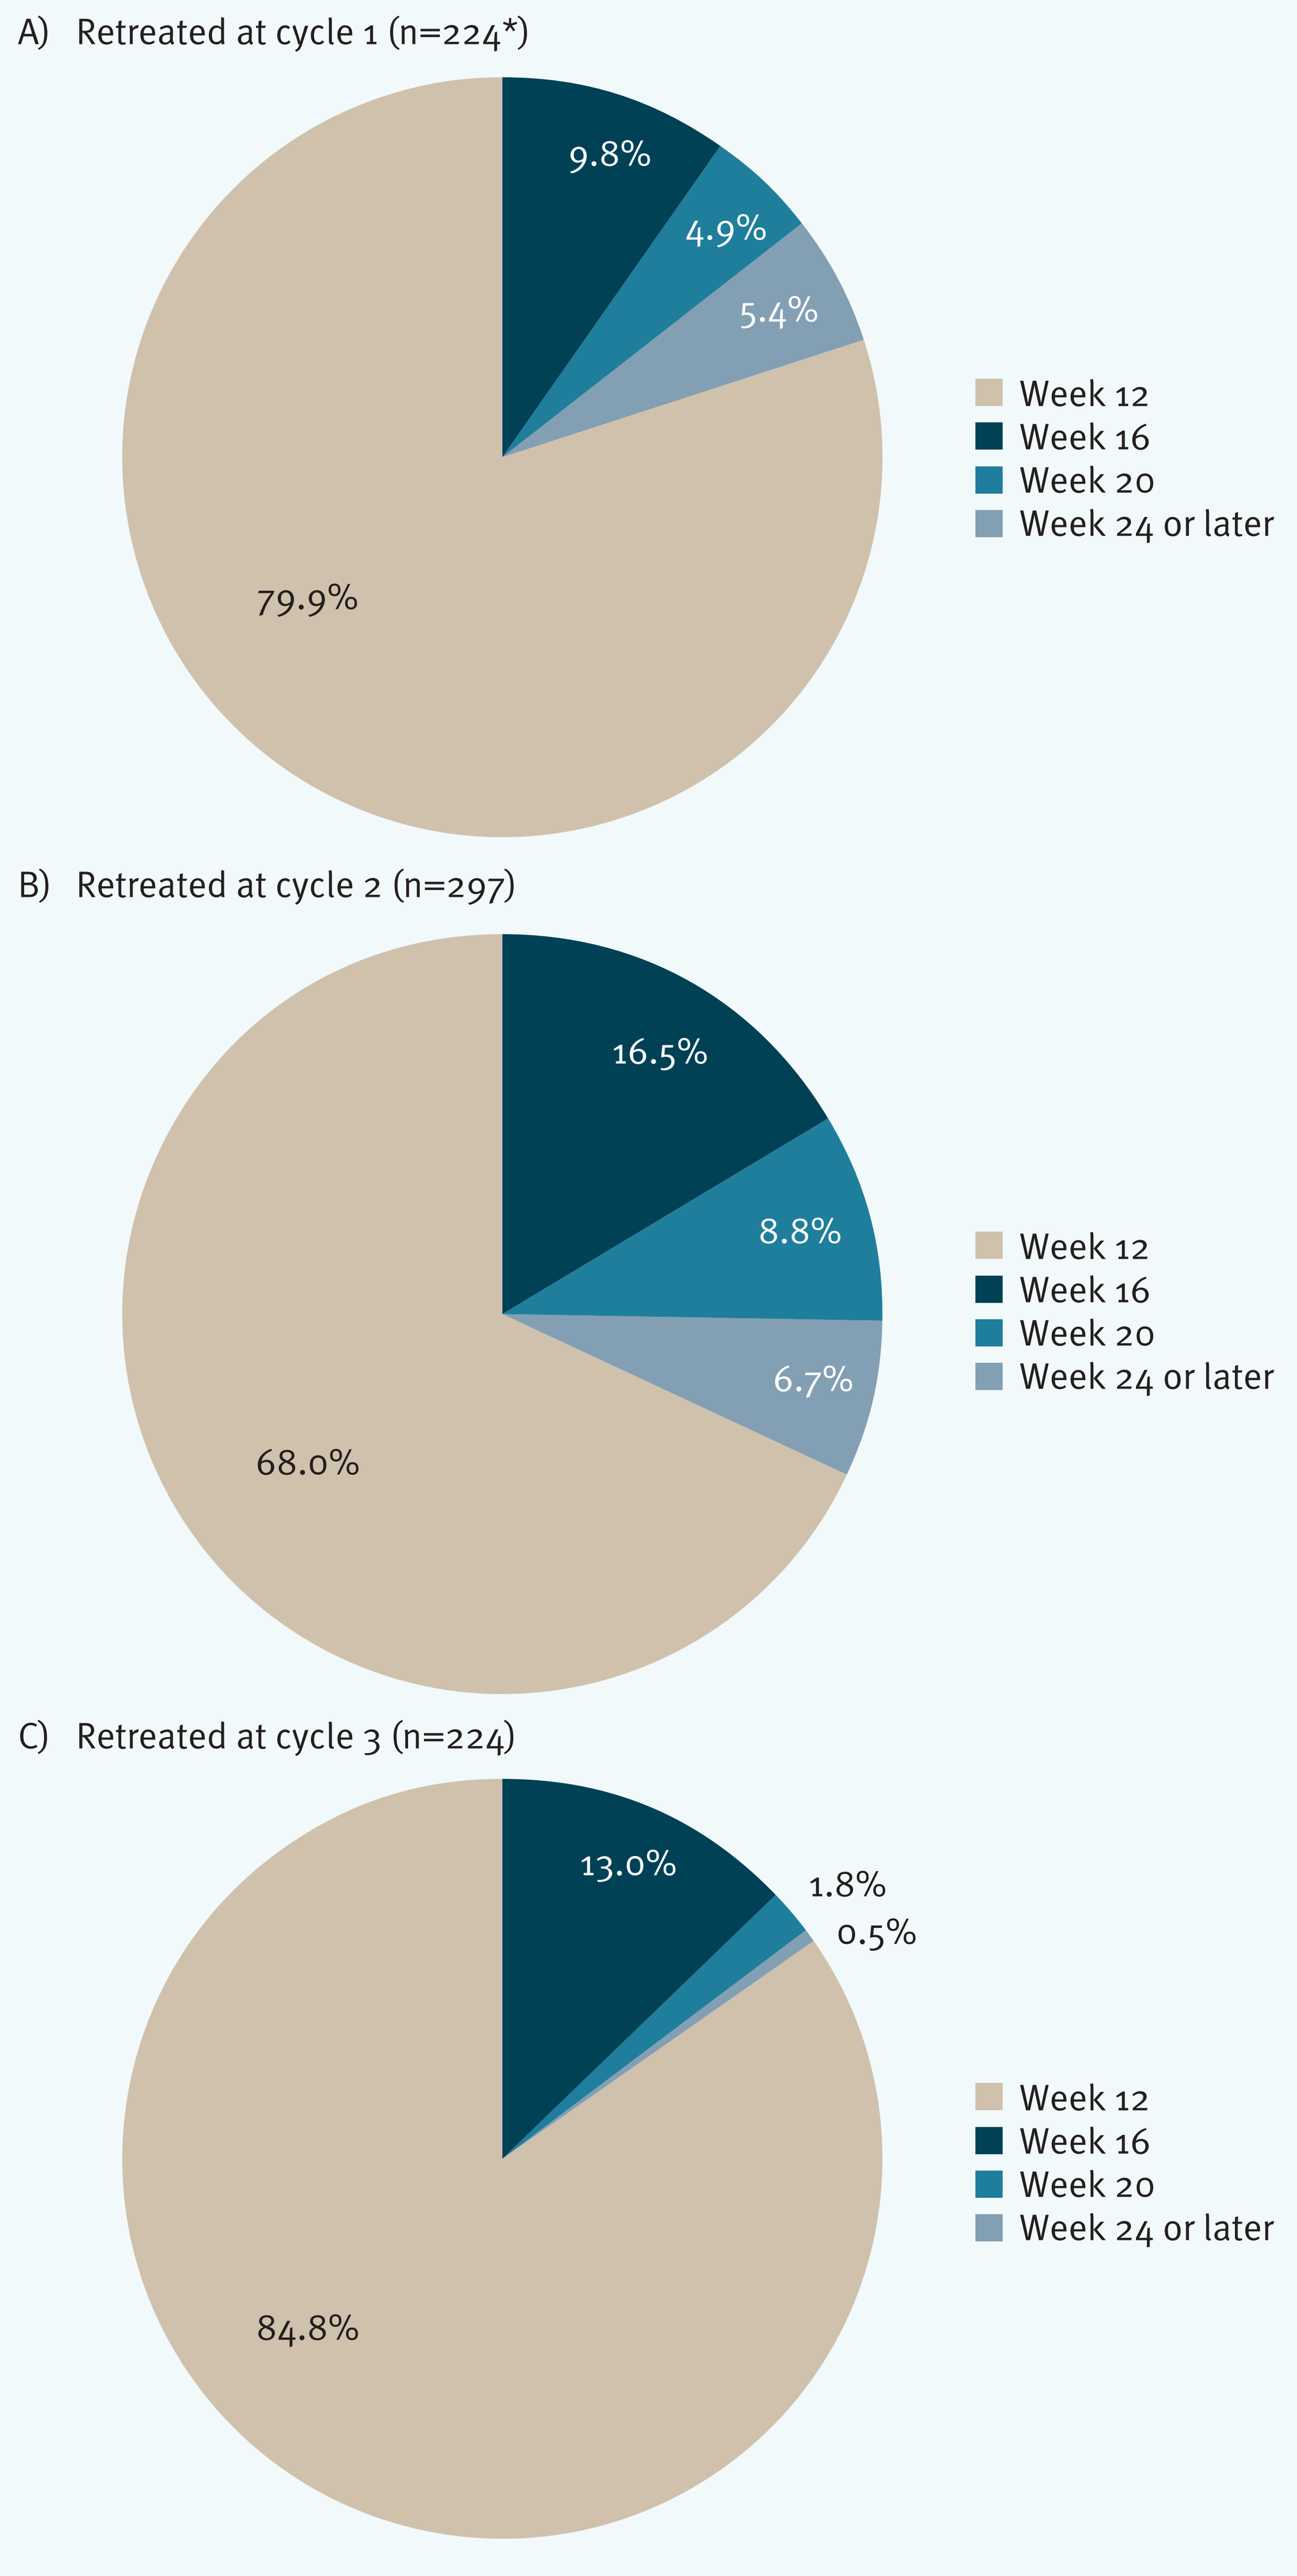

A) Retreated at cycle 1 (n=224*)
9.8%
4.9%
5.4%
79.9%
Week 12
Week 16
Week 20
Week 24 or later
B) Retreated at cycle 2 (n=297)
16.5%
8.8%
6.7%
68.0%
Week 12
Week 16
Week 20
Week 24 or later
C) Retreated at cycle 3 (n=224)
13.0%
1.8%
0.5%
84.8%
Week 12
Week 16
Week 20
Week 24 or later

*Data for patients who received placebo in the DB cycle are not included for retreatment at cycle 1

Table 2. Summary of TEAEs by treatment cycle and TEAEs displayed by ≥2% of patients at any cycle.

	Cycle 1 (n=345)	Cycle 2 (n=297)	Cycle 3 (n=224)
All doses			
TEAEs	140 (40.6) [290]	97 (32.7) [204]	47 (21.0) [97]
Related TEAEs	43 (12.5) [62]	23 (7.7) [39]	7 (3.1) [10]
TEAEs			
Muscle weakness	22 (6.4)	12 (4.0)	6 (2.7)
Fall	17 (4.8)	17 (5.7)	9 (4.0)
Pain in extremity	11 (3.2)	4 (1.3)	2 (0.9)
Arthralgia	6 (1.7)	6 (2.0)	1 (0.4)
Dysphagia	1 (0.3)	6 (2.0)	1 (0.4)
Asthenia	9 (2.6)	6 (2.0)	4 (1.8)

TEAE, treatment-emergent adverse event. Values are presented as n (percentage) [number of events].

Conclusions

- These data support the long duration of effect of abobotulinumtoxinA in patients with spastic paresis injected in the lower limb, with between 15 and 32% of patients re-injected at Week 16 or later across repeated cycles.
- A long duration of effect, leading to longer intervals between injections, would potentially reduce the burden associated with frequency of injections for patients and their caregiver/families.
- These results highlight the need for a tailored approach to treatment of patients with spastic paresis.

References

1. Gracies JM. *Muscle Nerve* 2005;31:535–51.
2. Martin A, *et al. Neuropsychiatr Dis Treat* 2014;10:111–22.
3. Gracies JM, *et al. Lancet Neurol* 2015;14:992–1001.
4. Dashtipour K, *et al. Medicine* 2016;95:e2468.
5. Olver J, *et al. Eur J Neurol* 2010;17(Suppl 2):57–73.
6. Gracies JM, *et al. PM&R* 2016;8(9S):S254.

Disclosures

Jean-Michel Gracies has served as a consultant and received research grant support from Allergan, Ipsen, and Merz; he is also an investigator on an Ipsen trial. Allison Brashear has consulted with Ipsen, Allergan, Concert, Revance and WorldMeds. Research is supported by NINDS, Allergan, Ipsen, Merz and Revance. Dr Brashear's conflict of interest is managed by Wake Forest School of Medicine. Alberto Esquenazi has received research funding from Ipsen and Allergan. Mike O'Dell has been a consultant for, and has received research grants from Ipsen. Dr O'Dell is an investigator in an Ipsen trial. Thierry Deltombe has served on scientific advisory boards and as a speaker for Ipsen, Allergan and Merz. Senen Gonzalez has received hospitality from Allergan, received research funding from Ipsen and served as a consultant for Merz. Francois Constant Boyer has served on scientific advisory boards for Ipsen, Allergan, Merz and Medtronic. Anne-Sophie Grandoulier, Claire Vilain and Philippe Picaut are employees of Ipsen.

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield Company, for medical writing support (funded by Ipsen) in preparing this poster.

Presented at TOXINS, 18–21 January 2017, Madrid, Spain.

This study was sponsored by Ipsen