Improvement of active movement and function in adults with chronic spastic paresis following repeated treatment with abobotulinumtoxinA (Dysport®)

Jean-Michel Gracies[1], Allison Brashear[2], Alberto Esquenazi[3], Robert Jech[4], Marta Banach[5], Peter McAllister[6], Serdar Kocer[7], Anne-Sophie Grandoulier[8], Claire Vilain[8], Philippe Picaut[8]

[1]EA 7377 BIOTN, Université Paris-Est, Hospital Albert Chenevier-Henri Mondor, Service de Rééducation Neurolocomotrice, Créteil, France; [2]Wake Forest School of Medicine, Department of Neurology, Winston-Salem, NC, US; [3]MossRehab & Albert Einstein, Elkins Park, Pennsylvania, US; [4]Department of Neurology, First Faculty of Medicine, Charles University and General Faculty Hospital, Prague, Czech Republic; [5]Department of Neurology, Jagiellonian, University Medical College, Krakow, Poland; [6]New England Institute for Neurology and Headache, Stamford, CT, US; [7]Rehabilitation Center, Hopital du Jura, Porrentury, Switzerland; [8]Ipsen, Les Ulis, France.

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by TOXINS

INTRODUCTION

- Antagonistic muscle activation in patients with hemiparesis following stroke or traumatic brain injury (TBI) results in reduced active movement and impaired function in the affected limb.[1,2]
- AbobotulinumtoxinA (aboBoNT-A; Dysport®) is an effective treatment for muscle overactivity in spastic paresis.[3,4] However, limited data are available for improvements in active limb movement and active function following treatment with botulinum toxin A (BoNT-A).
- Few studies have assessed long-term safety and efficacy of repeat BoNT-A injection in upper and lower limb spasticity.
- This analysis reports the effect of repeat injection cycles of aboBoNT-A on active movement and active function in patients with chronic hemiparesis from two open-label (OL) trials in adults with spasticity post stroke or TBI. These are extensions to respective double-blind studies (DB), one in upper limb spasticity and the other in lower limb spasticity, in which adults received a single aboBoNT-A injection.[5,6]

METHODS

Study design

- Two phase 3, international, multicentre, prospective, randomised, OL studies of aboBoNT A in adults with upper (NCT01313312) and lower (NCT01251367) limb spasticity post stroke or TBI.
- Patients received up to four OL treatment cycles (TCs; injections ≥12 weeks apart) over a maximum of 1 year (5 TCs including DB injection).
 - Retreatment was per investigator's clinical judgement and was possible from Week 12.
- Upper limb study patients were administered 500 U, 1000 U or 1500 U aboBoNT-A, per investigator's judgement, into the upper limb muscles.
 - Total injection volume was 5 mL for 500 U and 1000 U doses, and 7.5 mL for 1500 U.
 - Primary targeted muscle group (PTMG); extrinsic finger, wrist or elbow flexors were selected by investigators, as the muscle group with the most severe increase in muscle tone. Injection volumes for PTMG were fixed (2 or 3 mL) and the remainder was distributed among other upper limb muscles, at the investigator's discretion (within a pre-established range). A 1500 U dose required 500 U to be injected into shoulder muscles.
- Lower limb study patients were administered 1000 or 1500 U into the lower limb muscles.
 - Total injection volume was 7.5 mL for both doses: 2.5 mL over at least three sites in the soleus muscle, 1.5 mL into the medial and/or lateral gastrocnemius muscle, and 3.5 mL into at least one other lower limb muscle at the investigator's discretion (within a pre-established range).

Study participants

- Adults (18–80 years) with spastic hemiparesis of the upper or lower limb, at least 6 months post stroke or TBI.
- Patients who completed the respective DB studies with no major protocol deviations or ongoing treatment emergent adverse events.
- The upper limb study included newly recruited patients, who were required to meet the same inclusion criteria as those enrolled in the DB study.[5]

Figure 1. Study flow.

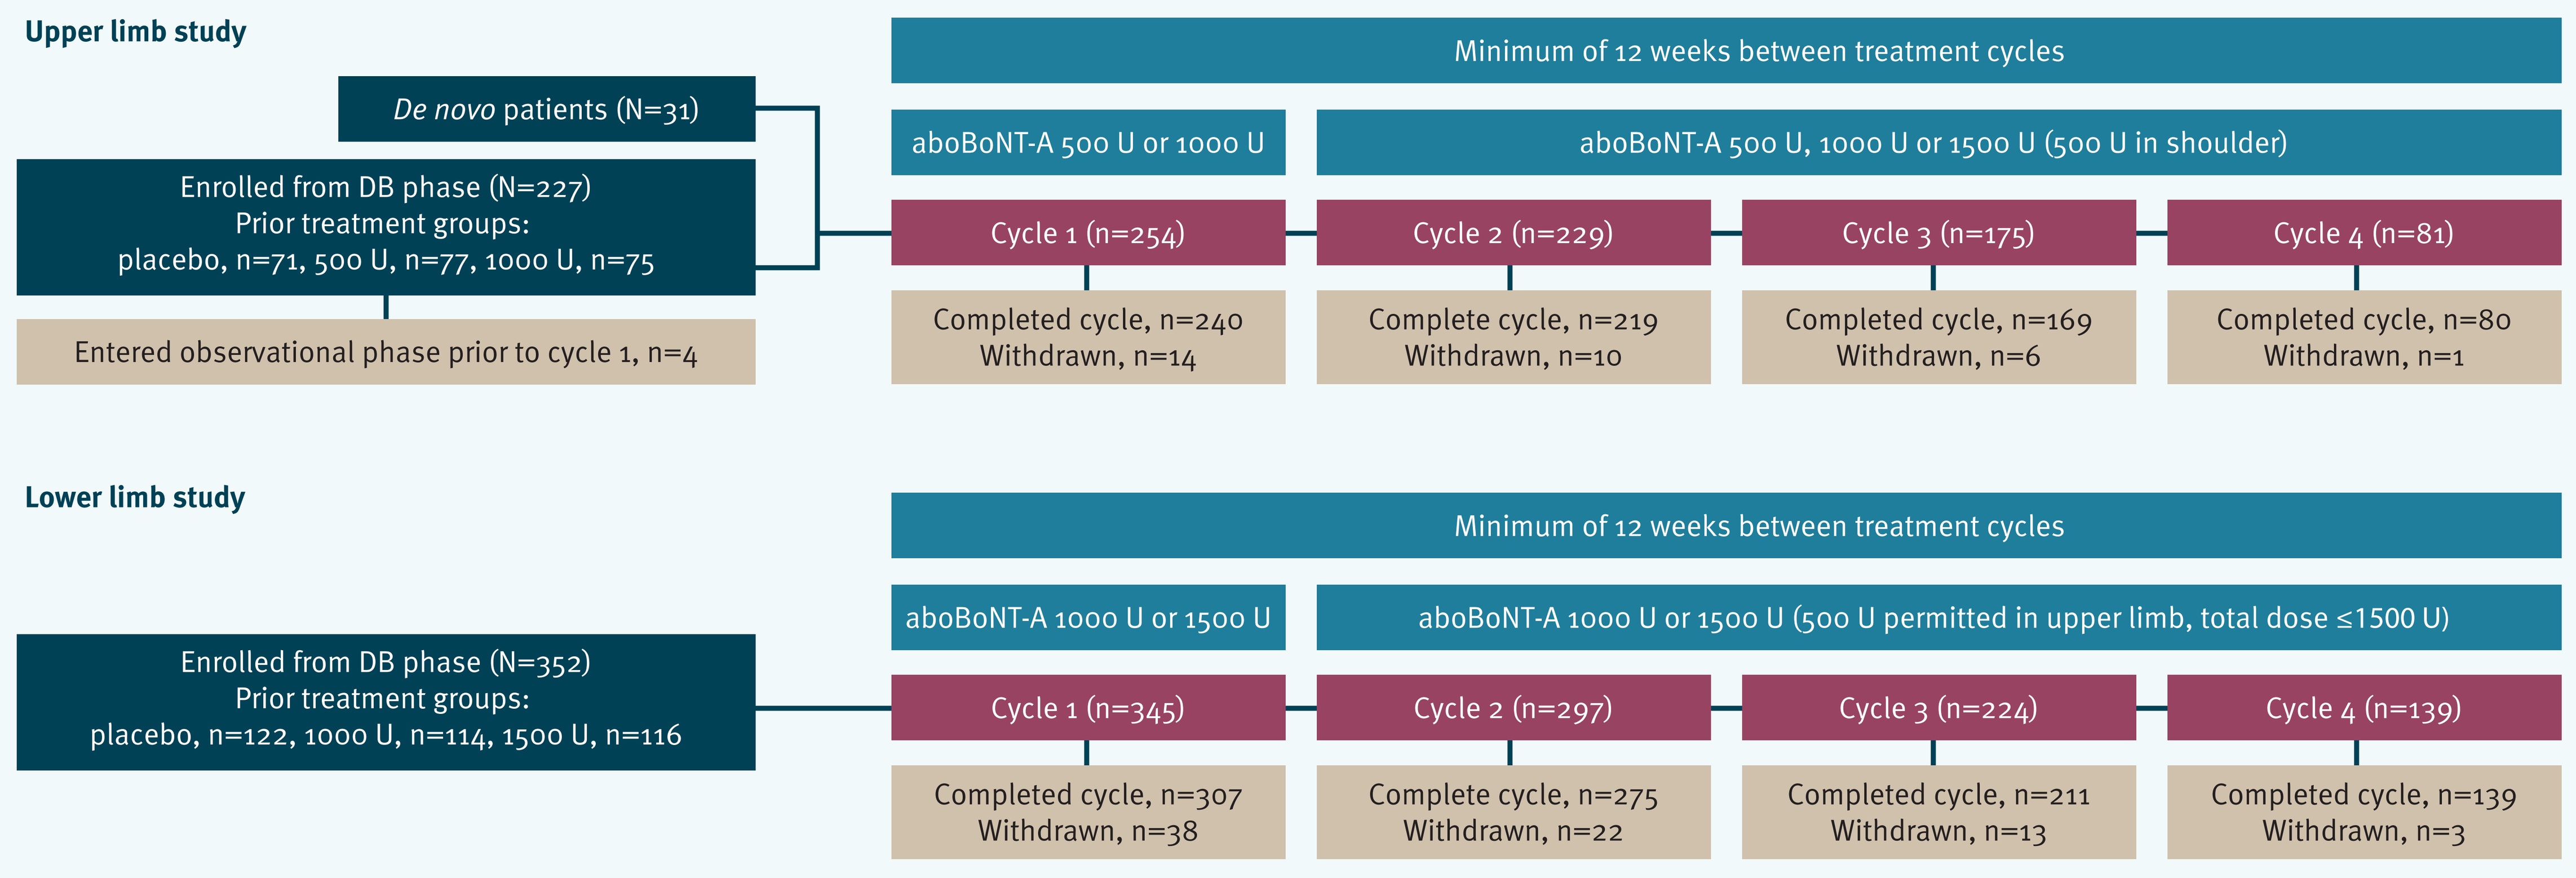
Upper limb study
De novo patients (N=31)
Enrolled from DB phase (N=227)
Prior treatment groups:
placebo, n=71, 500 U, n=77, 1000 U, n=75
Entered observational phase prior to cycle 1, n=4
Minimum of 12 weeks between treatment cycles
aboBoNT-A 500 U or 1000 U
aboBoNT-A 500 U, 1000 U or 1500 U (500 U in shoulder)
Cycle 1 (n=254)
Cycle 2 (n=229)
Cycle 3 (n=175)
Cycle 4 (n=81)
Completed cycle, n=240
Withdrawn, n=14
Complete cycle, n=219
Withdrawn, n=10
Completed cycle, n=169
Withdrawn, n=6
Completed cycle, n=80
Withdrawn, n=1
Lower limb study
Enrolled from DB phase (N=352)
Prior treatment groups:
placebo, n=122, 1000 U, n=114, 1500 U, n=116
Minimum of 12 weeks between treatment cycles
aboBoNT-A 1000 U or 1500 U
aboBoNT-A 1000 U or 1500 U (500 U permitted in upper limb, total dose ≤1500 U)
Cycle 1 (n=345)
Cycle 2 (n=297)
Cycle 3 (n=224)
Cycle 4 (n=139)
Completed cycle, n=307
Withdrawn, n=38
Complete cycle, n=275
Withdrawn, n=22
Completed cycle, n=211
Withdrawn, n=13
Completed cycle, n=139
Withdrawn, n=3

AboBoNT-A, abobotulinumtoxina; DB, double-blind. Patient numbers decreased across cycles due to injection intervals of >12 weeks, with a maximum treatment duration of one year per patient.

Outcome measures

- Active movement assessed by:
 - Active range of motion (X_A) against primary targeted muscle groups (extrinsic finger, wrist or elbow flexors).
 - Active ankle dorsiflexion with knee flexed (90°) and extended.
- Active function assessed by:
 - Modified Frenchay Scale (MFS) score in upper limb.
 - Patients were videotaped performing 10 upper limb tasks and scored by an independent observer (10-point scale).
 - 10-metre comfortable barefoot walking speed without aids.
- All outcome measures are presented as change from baseline at Week 4 of each TC.

RESULTS

- Overall, in the upper limb study 227 patients were treated in the DB phase, 31 additional patients were enrolled in the first OL cycle and 81 patients received the maximum of five injections. In the lower limb study 352 patients were treated in the DB phase and 139 patients received five injections (Figure 1).
- Baseline demographics and patient characteristics were comparable between upper and lower limb OL extension studies (Table 1).

Upper limb study

- There was evidence of a marked progressive increase in active range of motion against the extrinsic finger flexors (most frequently chosen PTMG) with repeated injections:
 - Mean increase (standard deviation [SD]) of +38.0 (53.4)° by TC 4 of the OL phase (Table 2).
- The increase in the active range of motion seen at Week 4 of the DB cycle for elbow and wrist flexors was maintained across OL TCs.
 - The limited improvement observed at TC 4 against wrist flexors might be explained by the high baseline active range of motion (81.5°) versus 68 to 74° for patients with fewer treatment cycles.
- Overall increase of active function assessed by the MFS was +0.40 (0.75) at the fifth TC (Figure 2).
 - Improvement in active function was more pronounced with 1500 U compared with 1000 U (+0.62 [0.48] vs. +0.30 [0.83], respectively) at TC 4 of the OL phase.

Table 1. Baseline demographics and patients characteristics.

Characteristic	Upper limb study (N=258)	Lower limb study (N=352)
Mean age, years (SD)	52.4 (13.9)	53.2 (12.7)
Sex, n (%)		
Male	166 (64.3)	239 (67.9)
Mean weight, kg (SD)	81.3 (18.6)	79.9 (16.5)
Cause, n (%)		
Stroke	230 (89.1)	309 (87.8)
TBI	28 (10.9)	43 (12.2)
Mean time since event, years (SD)		
Stroke	5.1 (4.2)	4.5 (4.8)
TBI	9.9 (8.0)	9.2 (10.1)
Treatment naïve,* n (%)	116 (45.0)	226 (64.2)
Primary target muscle group, n (%)		
Extrinsic finger flexors	146 (56.6)	n/a
Wrist flexors	43 (16.7)	
Elbow flexors	69 (26.7)	

*Patients who never received botulinum toxin A in the affected limb prior to study entry. SD, standard deviation; TBI, traumatic brain injury.

Table 2. Upper Limb Study – change from baseline in active range of motion at Week 4 of each TC for each of the PTMGs (all doses combined).

Muscle group	DB cycle (N=152)	Cycle 1 (N=254)	Cycle 2 (N=229)	Cycle 3 (N=175)	Cycle 4 (N=81)
Extrinsic finger flexors	n=88	n=144	n=133	n=98	n=56
Mean change [SD]	19.8 [28.8]	31.3 [40.9]	34.4 [45.0]	33.5 [47.1]	38.0 [53.4]
Wrist flexors	n=21	n=39	n=34	n=23	n=10
Mean change [SD]	21.8 [24.6]	17.3 [28.6]	23.0 [32.7]	22.0 [30.9]	3.0 [25.1]
Elbow flexors	n=43	n=69	n=59	n=42	n=14
Mean change [SD]	14.3 [22.3]	10.7 [25.5]	17.6 [23.3]	14.5 [24.1]	13.6 [20.6]

DB, double-blind; N, number of subjects receiving dose; n, number of subjects with data; PTMG, primary targeted muscle group; SD, standard deviation.

Figure 2. Upper Limb Study – change from baseline in active function (MFS score) at Week 4 of each TC.

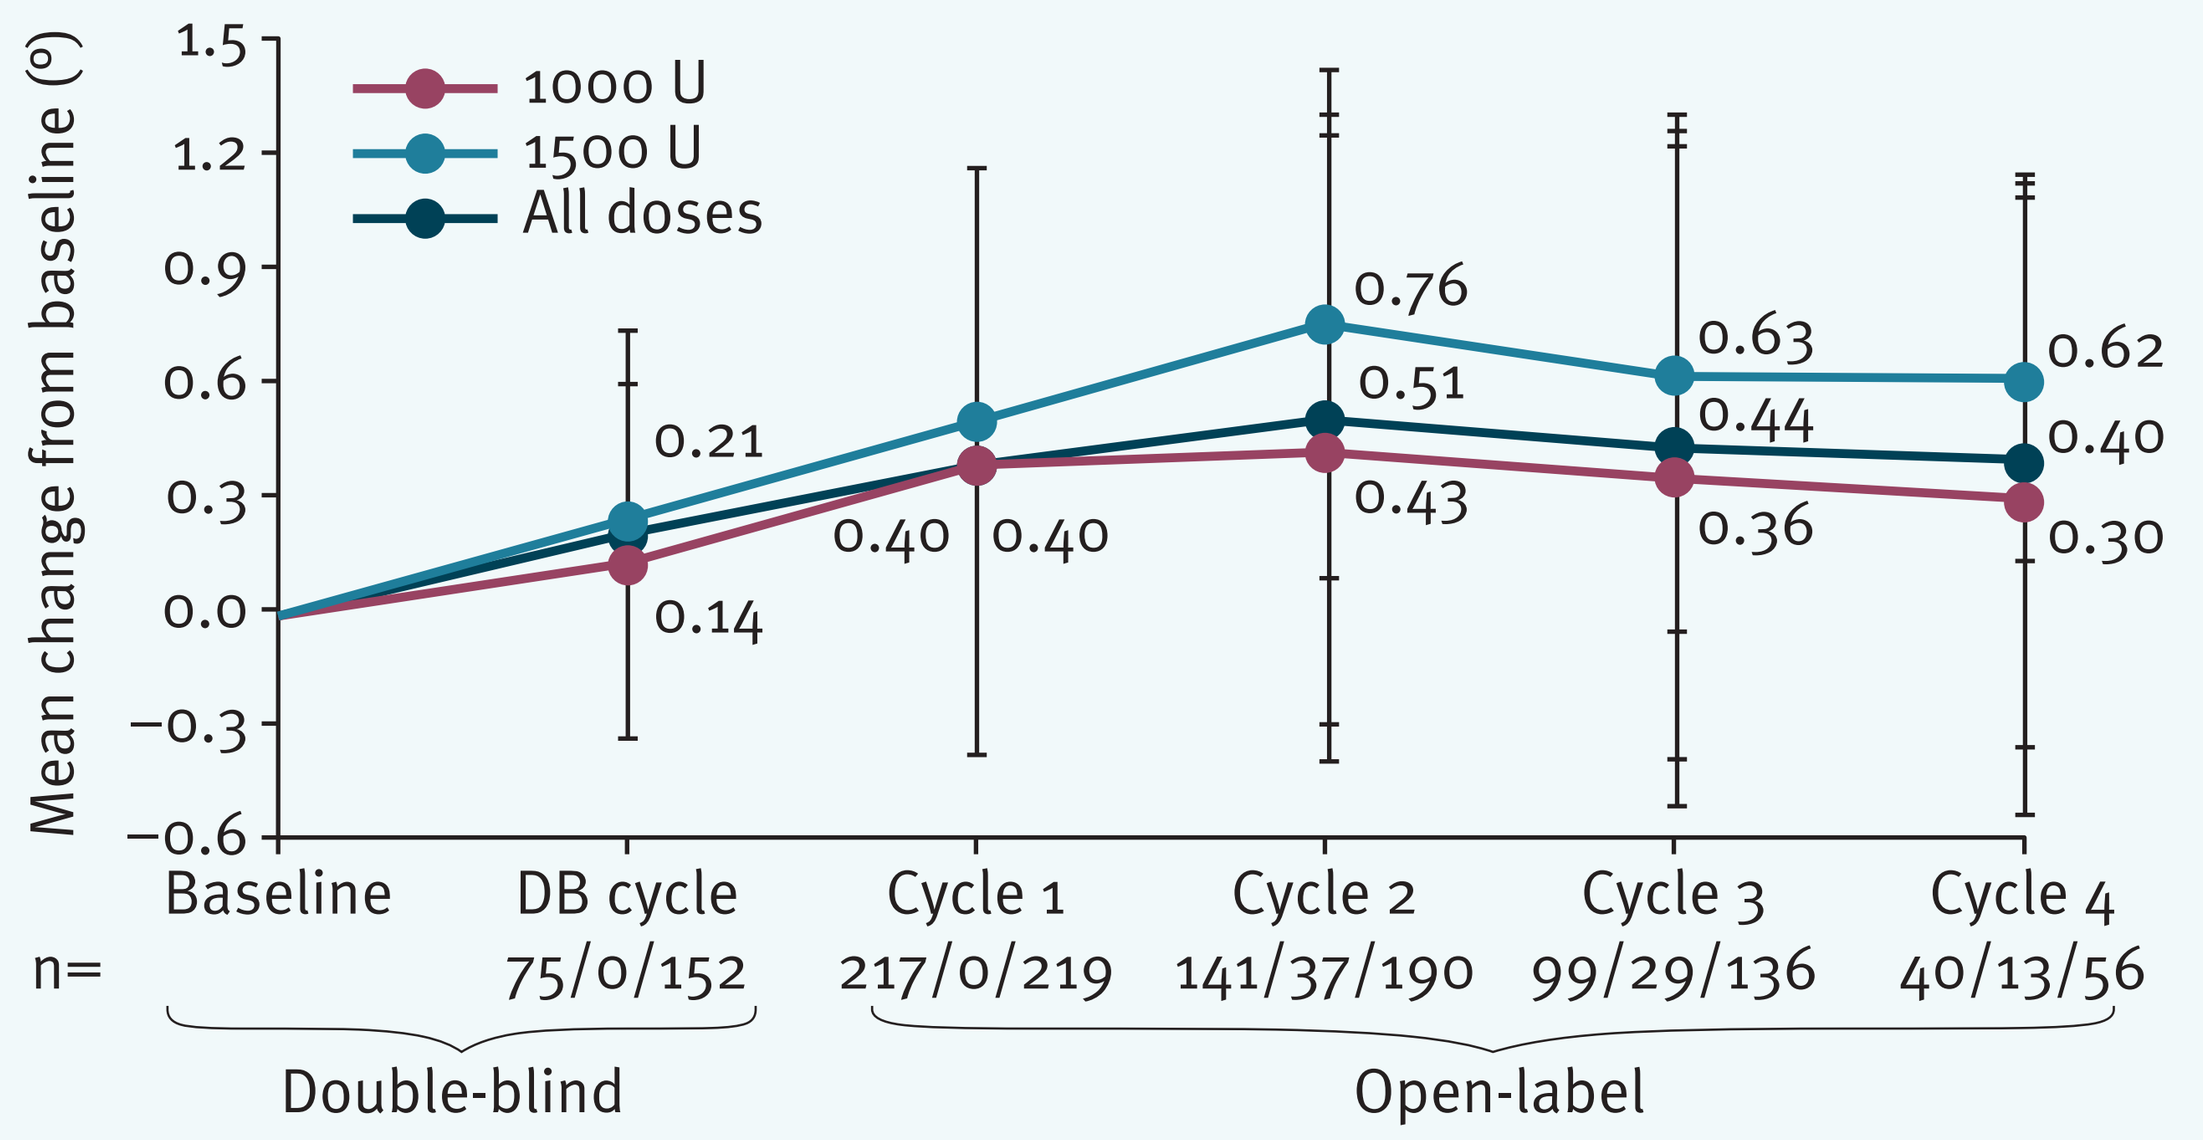
Mean change from baseline (°)
1.5
1.2
0.9
0.6
0.3
0.0
−0.3
−0.6
1000 U
1500 U
All doses
0.21
0.14
0.40
0.40
0.76
0.51
0.43
0.63
0.44
0.36
0.62
0.40
0.30
Baseline
DB cycle
Cycle 1
Cycle 2
Cycle 3
Cycle 4
n=
75/0/152
217/0/219
141/37/190
99/29/136
40/13/56
Double-blind
Open-label

DB, double-blind; MFS, Modified Frenchay Scale. Patient numbers reported as n= 1000 U/1500 U/all doses combined (including 500 U).

Lower limb study

- Active ankle dorsiflexion with knee extended improved by +6.5 (10.9)° at TCs 4 of the OL phase, while improvement in ankle dorsiflexion with knee flexed was maintained across TCs (Table 3).
- Comfortable barefoot walking speed progressively increased across TCs, reaching +25.35% (95% confidence interval [CI] 17.48; 33.21) at TC 4 of the OL phase.
 - Improvements reached +0.088 (0.14) m/s at this TC (Figure 3).

Table 3. Lower limb study – range of active ankle dorsiflexion at Week 4 of each TC (doses combined).

	DB cycle (N=224)	Cycle 1 (N=341)	Cycle 2 (N=290)	Cycle 3 (N=218)	Cycle 4 (N=135)
Knee extended	n=224	n=341	n=290	n=218	n=135
Mean change [SD]	3.3 [10.2]	4.1 [10.6]	4.4 [10.6]	6.0 [11.4]	6.5 [10.9]
Knee flexed	n=224	n=341	n=290	n=218	n=135
Mean change [SD]	3.5 [9.0]	4.1 [10.7]	5.0 [10.3]	5.2 [10.9]	3.8 [9.8]

DB, double-blind; N, number of subjects receiving dose; n, number of subjects with data; SD, standard deviation.

Figure 3. Lower limb study – mean change in comfortable barefoot walking speed at Week 4 of each TC (doses combined).

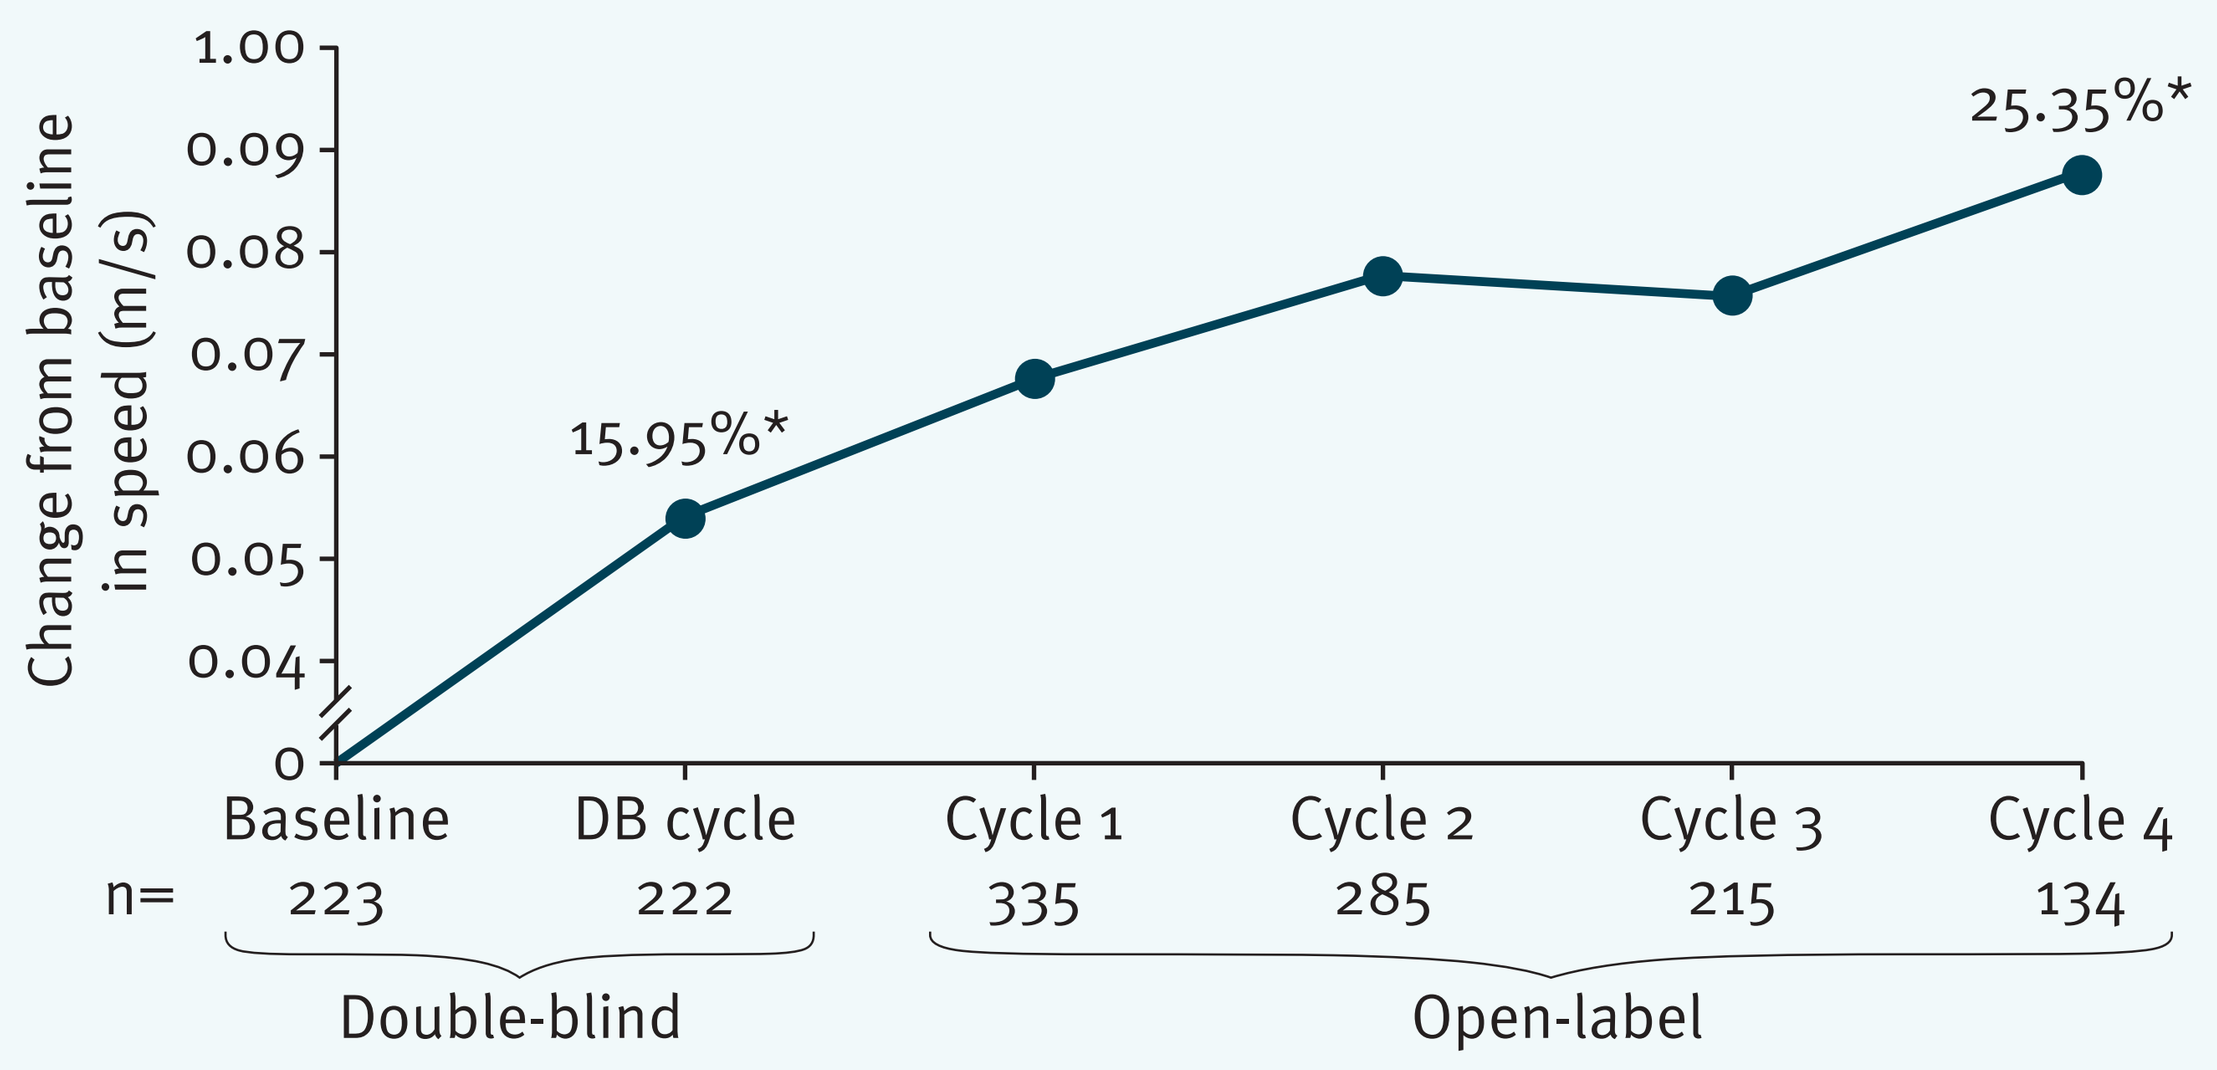
Change from baseline in speed (m/s)
1.00
0.09
0.08
0.07
0.06
0.05
0.04
0
15.95%*
25.35%*
Baseline
DB cycle
Cycle 1
Cycle 2
Cycle 3
Cycle 4
n=
223
222
335
285
215
134
Double-blind
Open-label

*Percentage improvement from baseline. DB, double-blind.

Conclusions

- The results from these studies demonstrate that repeat injection of abobotulinumtoxinA improves active movement and active function in the affected upper or lower limb of patients with spastic paresis. Improvements were beyond those previously reported on muscle tone and spasticity.
- In patients with chronic upper limb spastic paresis, improvements in both active movement and active function were observed after repeat injection of abobotulinumtoxinA over one year.
- A more pronounced efficacy on active function was observed with 1500 U compared with 1000 U aboBoNT-A in the upper limb, underlining the importance of including shoulder muscles when medically appropriate.
- In patients with chronic lower limb spastic paresis, improvements in both active movement and active function (walking speed) were observed after repeated injections of 1000 and 1500 U abobotulinumtoxinA.

References

1. Gracies JM. *Muscle Nerve* 2005;31:535–51.
2. Gracies JM. *Muscle Nerve* 2005;31:552–71.
3. Dashtipour K, *et al. Am J Phys Med Rehabil* 2015;94:229–38.
4. Dashtipour K, *et al. Medicine* 2016;95:e2468.
5. Gracies JM, *et al. Lancet Neurol* 2015;14:992–1001.
6. Gracies JM, *et al. PM&R* 2016;8(9S):S254.

Disclosures

Jean-Michel Gracies has served as a consultant and received research grant support from Allergan, Ipsen, and Merz; he is also an investigator on an Ipsen trial. Allison Brashear has consulted with Ipsen, Allergan, Concert, Revance and WorldMeds. Research is supported by NINDS, Allergan, Ipsen, Merz and Revance. Dr Brashear's conflict of interest is managed by Wake Forest School of Medicine. Alberto Esquenazi has received research funding from Ipsen and Allergan. Robert Jech has received grants from the Czech Science Foundation, Czech Ministry of Health, Czech Ministry of Education and Charles University, Prague; and honoraria from Ipsen for consultations and lectures. Marta Banach has received honoraria from Ipsen for consultations and lectures. Dr Banach is an investigator in an Ipsen trial. Peter McAllister served on the speakers' bureau for Ipsen, Allergan and Merz, and received grant support from Allergan. Sedar Kocer has received honorarium for serving as a trainer and speaker for Ipsen, and as a trainer for Allergan and Merz. Anne-Sophie Grandoulier, Claire Vilain and Philippe Picaut are employees of Ipsen.

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield Company, for medical writing support (funded by Ipsen) in preparing this poster.

Presented at TOXINS, 18–21 January 2017, Madrid, Spain.

This study was sponsored by Ipsen