Correlations between reduced spasticity, goal attainment and global assessment of benefits in patients with upper limb spasticity treated with botulinum toxin A

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by TOXINS

Khashayar Dashtipour[1], Jovita Balcaitiene[2], Stephen Ashford[3], Jorge Jacinto[4], Klemens Fheodoroff[5], Pascal Maisonobe[2], Lynne Turner-Stokes[3]

[1]Department of Neurology/Movement Disorders, School of Medicine, Loma Linda University, California, USA; [2]Medical Affairs, Ipsen Pharma, Boulogne-Billancourt, France; [3]King's College London School of Medicine, Palliative Care, Policy and Rehabilitation and Regional Rehabilitation Unit, Northwick Park Hospital, London, UK; [4]Centro de Medicina de Reabilitaçãode Alcoitão, Serviço de Reabilitação de Adultos, Estoril, Portugal; [5]Department of Neurorehabilitation, Gailtal-Klinik, Hermagor, Austria

Introduction

- Upper limb spasticity (ULS) is a chronic neurological condition that occurs frequently following stroke or traumatic brain injury.[1]
- Botulinum toxin type A (BoNT-A) is effective in reducing ULS at the level of impairment (reduction in muscle tone), but functional gains have been harder to demonstrate.[2]
- It is becoming increasingly important to evaluate the impact of treatment from the patient's personal perspective. The US Institute of Medicine has included patient-centred care as one of the top aims in care quality.[3]
 - Patient-centred goal setting, in which treatment goals are tailored to the individual patient, is an integral part of neurological rehabilitation.[4,5]
 - Goal Attainment Scaling (GAS) is an effective and well-established method for quantifying achievement of patient-centred goals.[6–8]

The Upper Limb International Spasticity (ULIS) programme

- The ULIS programme is a series of large international observational cohort studies evaluating BoNT-A for post-stroke ULS management in everyday clinical practice.[9,10]
- The second ULIS study (ULIS-II) evaluated patient-centred goal attainment as the primary outcome measure following a single BoNT-A injection.
 - The most common goal areas selected were pain reduction, active and passive function; 80% of patients achieved or over-achieved their primary goal.
 - Both patients and investigators reported global benefits.

Objective

- In this secondary analysis of data from ULIS-II, we examine the relationship between reduction in muscle tone following BoNT-A treatment, the investigator and patient global assessments of benefits, and overall goal attainment.

Methods

Design

- ULIS-II was an 18-month, prospective, observational cohort study conducted at 84 centres in 22 countries.[9,10] Patients with post-stroke ULS were treated with a single BoNT-A injection according to routine clinical practice.

Inclusion criteria and patients

- Adults (≥18 years) with post-stroke ULS in whom a decision to inject BoNT-A has been made prior to enrolment.
- No treatment with BoNT-A or BoNT-B in previous 12 weeks.
- A total of 468 patients were enrolled in the study; 456 received BoNT-A and attended follow-up evaluation, which included GAS (the 'efficacy population').

Treatment

- Investigators made all treatment decisions according to their usual clinical practice, including target muscles, type of BoNT-A preparation, injected doses, number of injection sites and volume for each site.

Outcome measures

- Follow-up evaluation was generally 3 to 5 months post-injection, at the investigators discretion.
- Muscle tone was recorded using the Modified Ashworth Scale (MAS). MAS scores across the shoulder, elbow, wrist, and fingers were summed to create a 'Composite MAS score'. Reduction in muscle tone was evaluated by change in composite score.
- GAS was applied using the GAS-Light method.[11]
 - GAS goals were rated on a six-point verbal rating scale, then transcribed (using computer software) to a five-point numerical scale (range −2 to +2) and combined into a single GAS T-score using the formula developed by Kiresuk and Sherman.[7]
- Global assessment of benefit was rated according to five categories: great benefit (+2), some benefit (+1), same (0), worse (−1) or much worse (−2).

Statistical analysis

- All analyses were conducted on the efficacy population (n=456).
- The relationships between GAS T-scores and the other outcome measures (MAS total scores and global assessment of benefit) were examined using Spearman's rank correlation coefficients.

Results

Demographics

- The majority of patients (58%) were male and the mean (standard deviation [SD]) age was 57 (13.5) years.
- Mean (SD) time since onset of stroke was 61.4 (69.1) months.
 - 70% had infarcts and 30% had haemorrhagic stroke.

Table 1. Clinical outcomes; GAS T-score (weighted mode 2) and MAS composite score at baseline and follow-up

Parameter	GAS-T Mean (SD)	MAS Mean (SD)
Baseline	34.4 (5.1)	11.0 (3.3)
Follow-up	52.0 (10.0)	8.4 (3.4)
Change score	17.6 (11.0)	−2.6 (2.6)
95% CI	16.6, 18.6	−2.9, −2.4
Significance (based on a paired T-test)	*p*<0.001	*p*<0.001

CI, confidence interval; GAS, Goal Attainment Scaling; SD, standard deviation.

Figure 1. Patient- and investigator-reported benefit

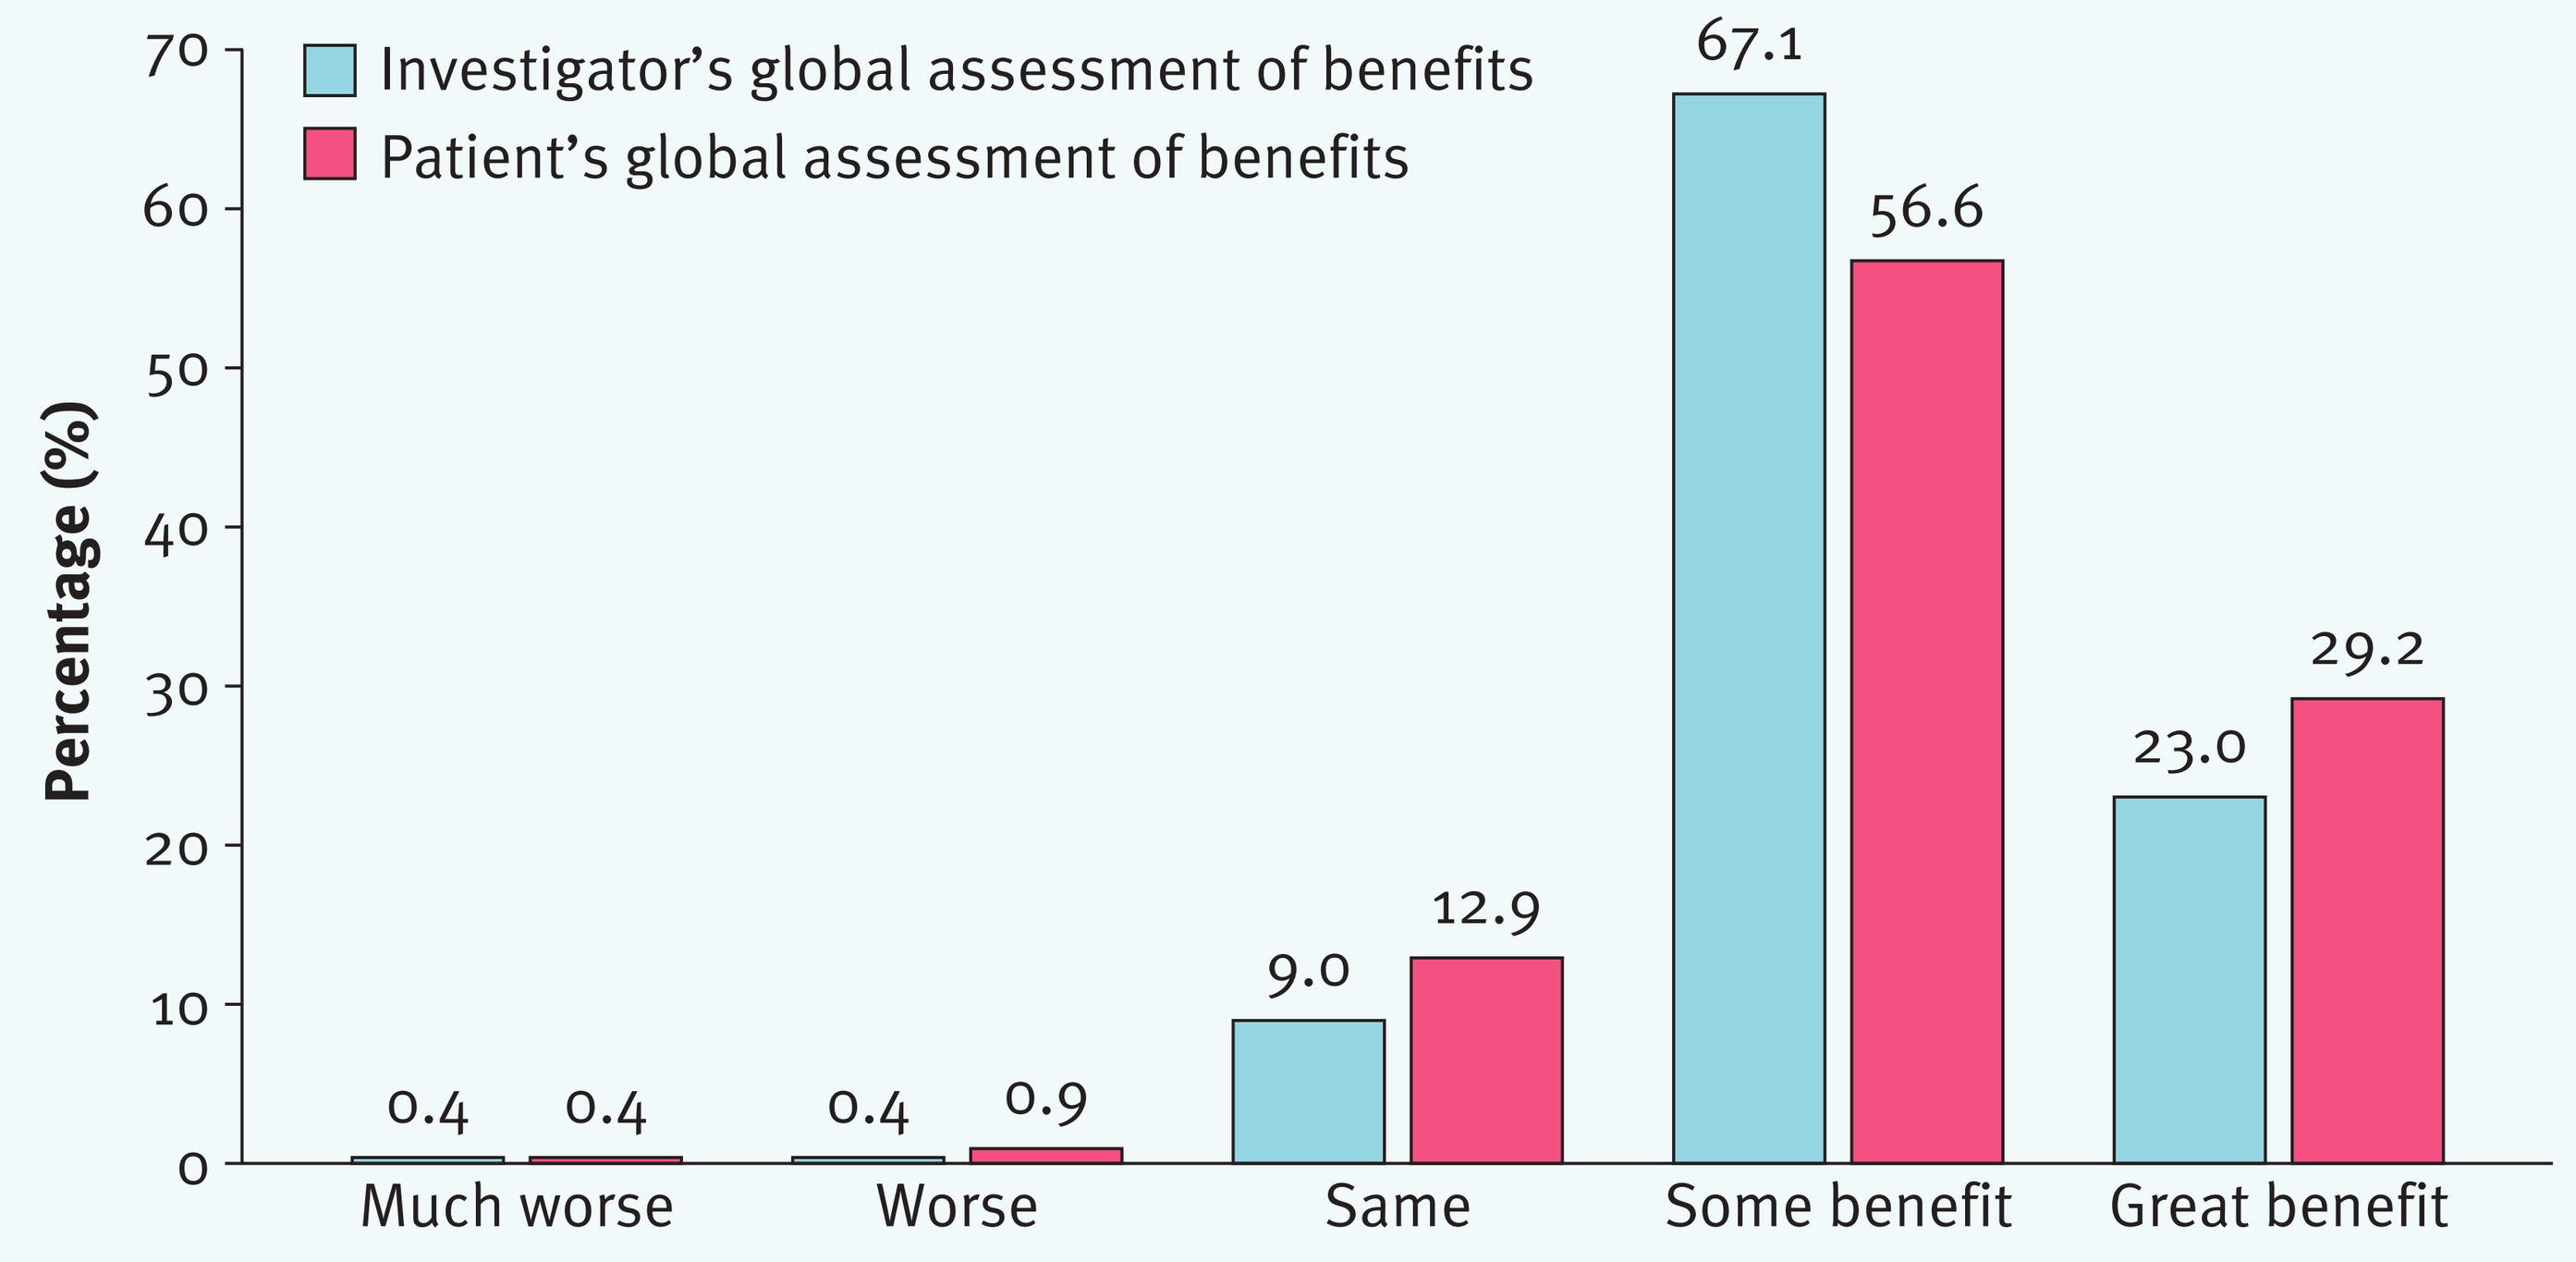
Investigator's global assessment of benefits
Patient's global assessment of benefits
Percentage (%)
70
60
50
40
30
20
10
0
0.4
0.4
0.4
0.9
9.0
12.9
67.1
56.6
23.0
29.2
Much worse
Worse
Same
Some benefit
Great benefit

Clinical outcomes

- There were statistically significant improvements in MAS composite score and GAS T-score between baseline and follow-up (Table 1).
 - Mean change in GAS T-score was 17.6 (change of 10 is considered clinically significant).[8]
 - Mean (95% confidence interval [CI]) change in MAS composite score was −2.6 (−2.9, −2.4).
- Overall, 90.1% of investigators and 85.8% of patients considered BoNT-A treatment to be of benefit (Figure 1).

Correlations between outcome measures

- Changes in MAS composite scores were weakly correlated with changes in GAS T-scores (rho=−0.28; *p*<0.001; Figure 2).
 - Statistical significance may be due to large patient numbers.
- Substantially stronger correlations were observed between GAS T-scores at visit 2 and both investigator- and patient-reported benefits (rho=0.39 and 0.47, respectively; *p*<0.001; Table 2).

Figure 2. Correlations in clinical outcomes; change from baseline in MAS composite score and weighted GAS T-score (mode 2)

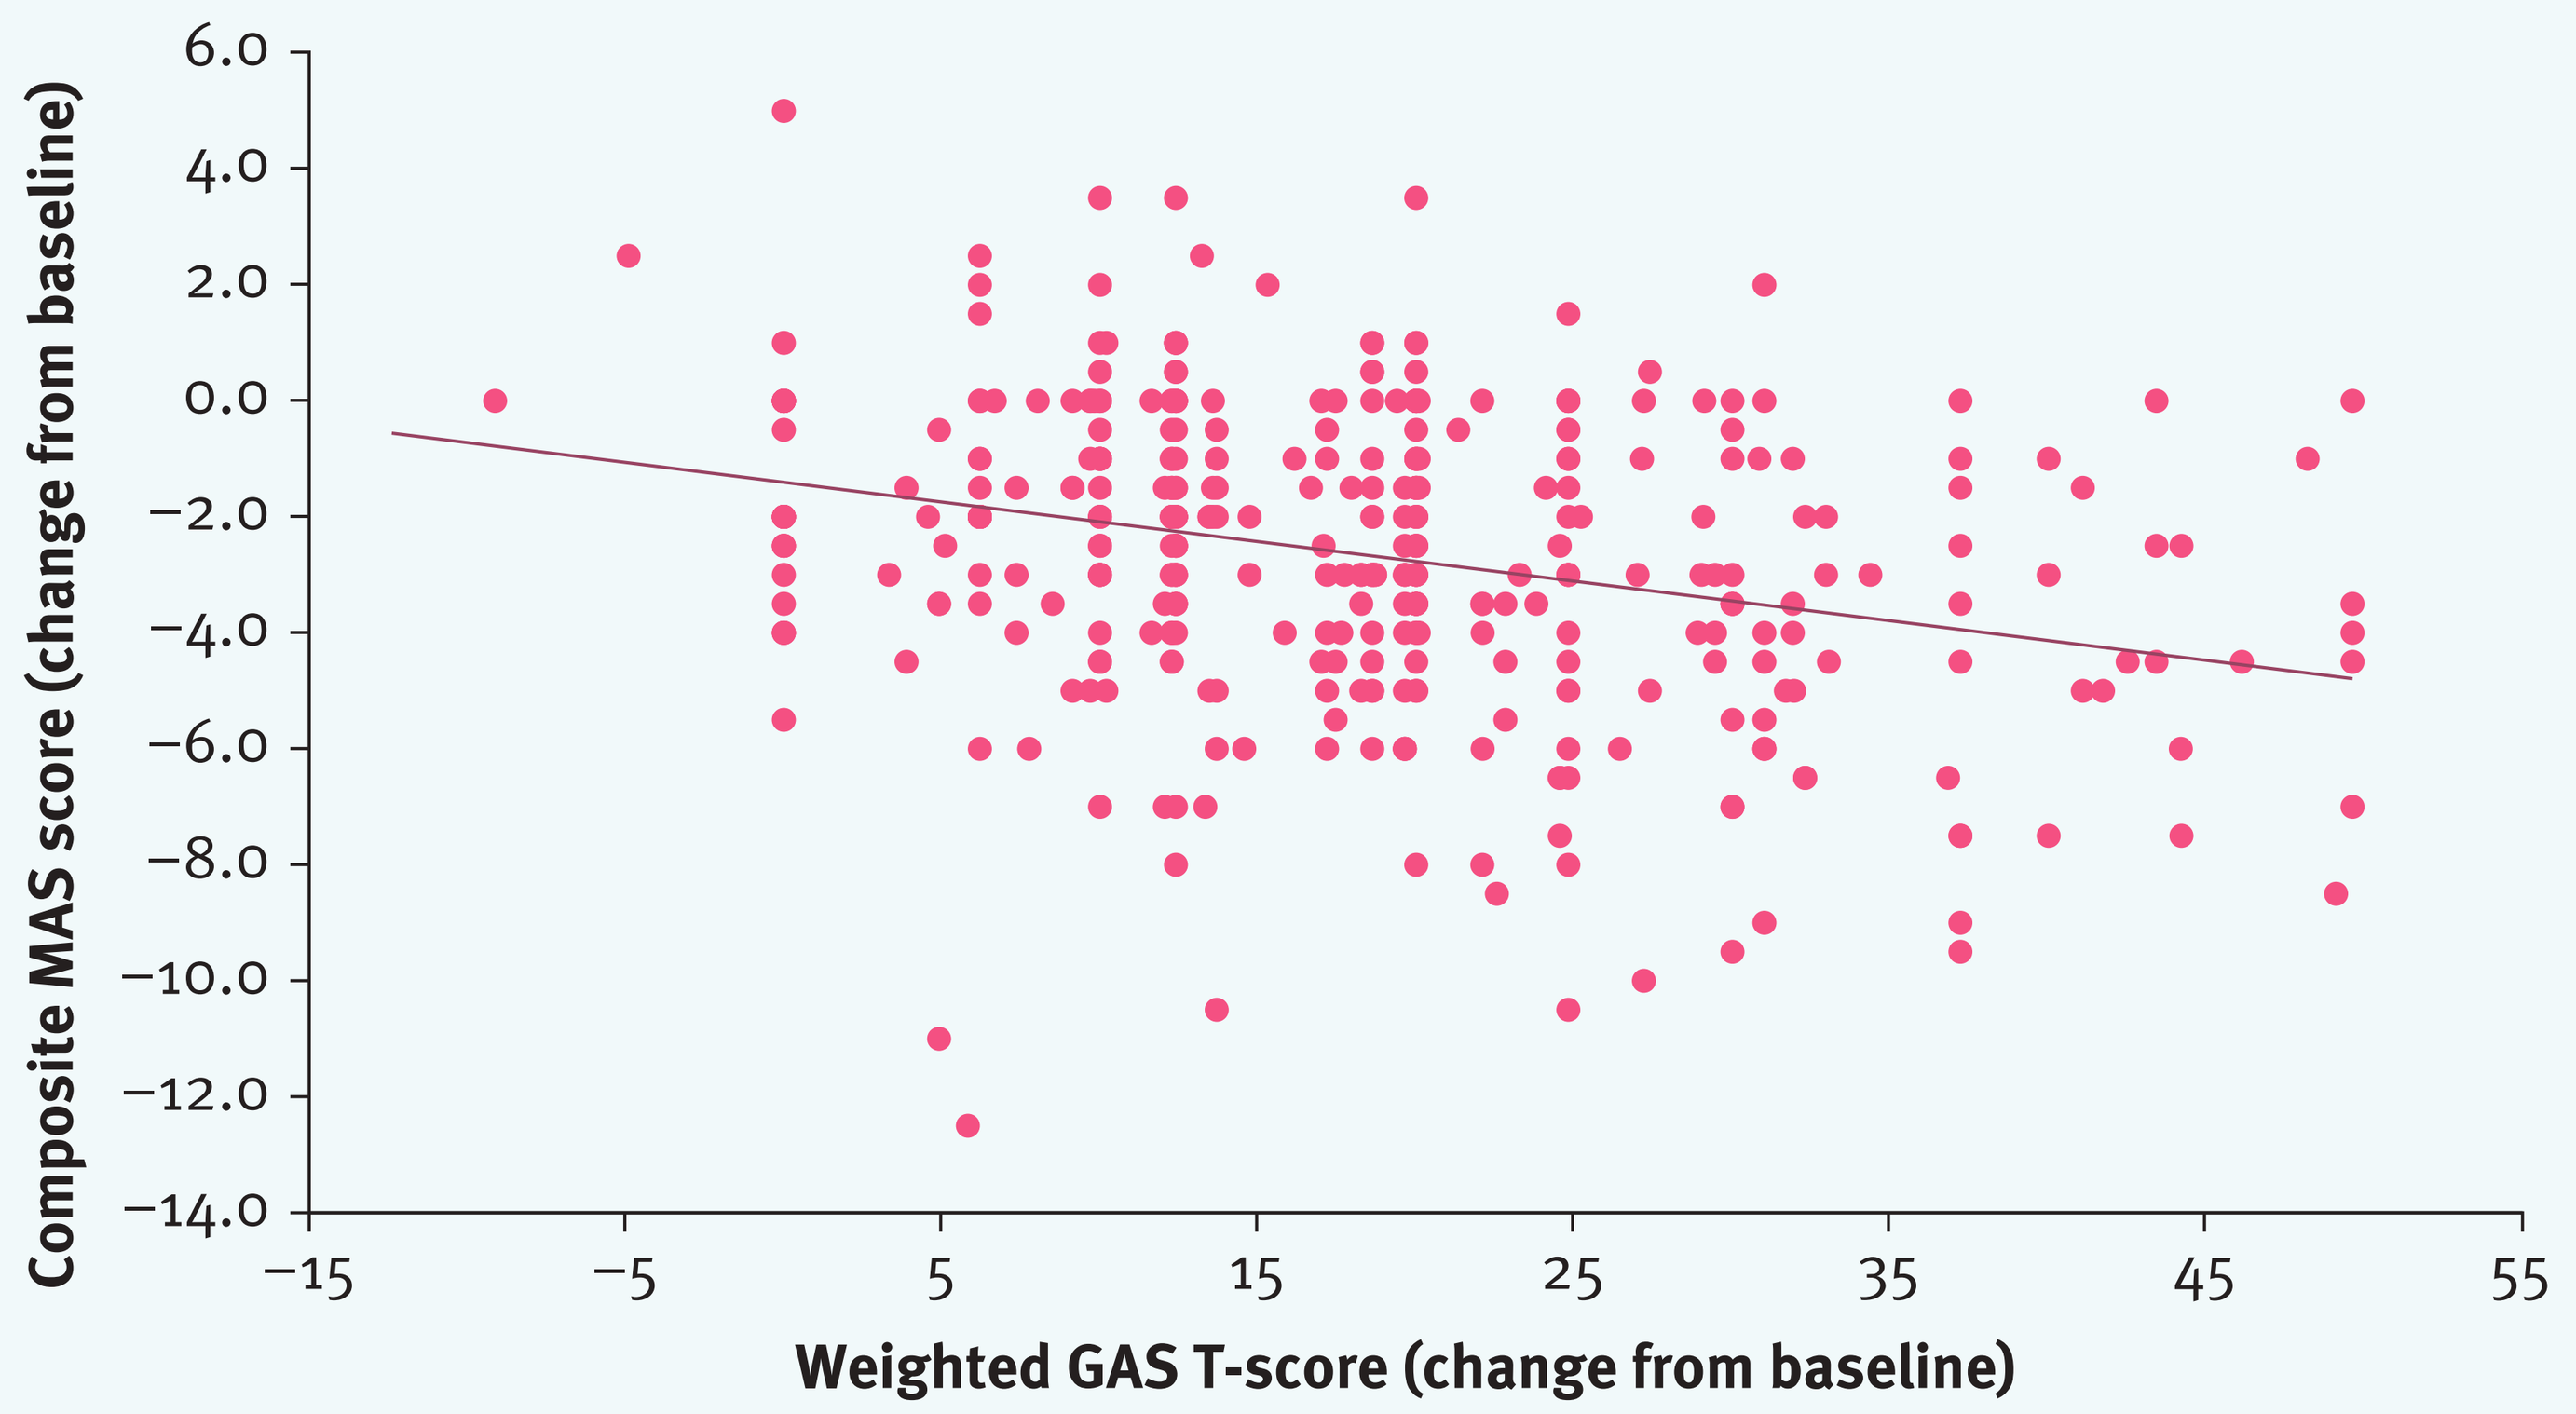
Composite MAS score (change from baseline)
6.0
4.0
2.0
0.0
−2.0
−4.0
−6.0
−8.0
−10.0
−12.0
−14.0
−15
−5
5
15
25
35
45
55
Weighted GAS T-score (change from baseline)

GAS, Goal Attainment Scaling; MAS, Modified Ashworth Scale.

Table 2. Correlations between clinical outcomes and change from baseline in weighted GAS T-score

Parameter	Spearman rho	Probability
Reduction in muscle tone		
Change in MAS (n=388)	−0.28	<0.001
Global assessment of benefit		
Investigator assessment (n=456)	0.39	<0.001
Patient assessment (n=456)	0.47	<0.001

GAS, Goal Attainment Scaling; MAS, Modified Ashworth Scale.

- There were weak correlations between the change in MAS composite score and both investigator- and patient-reported benefits (rho=0.28 and 0.20, respectively; *p*<0.001; Table 3).
 - The association was slightly stronger for investigator-reported benefits than for patient-reported benefits.

Table 3. Correlation between change in MAS composite scores and reported benefits

Parameter	Spearman rho	Probability
MAS composite change score		
Investigator's assessment (n=388)	0.28	<0.001
Patient assessment (n=388)	0.20	<0.001

MAS, Modified Ashworth Scale.

Conclusions

- In ULIS-II, in patients receiving botulinum toxin type A treatment, the change in Modified Ashworth Scale composite score was only weakly associated with investigator and patient global assessment of benefit.
- This association was slightly stronger for investigator-reported benefits than for patient-reported benefits, which may suggest that change in Modified Ashworth Scale scores may not necessarily translate into patient benefit.
- A stronger relationship was seen between the goal attainment (change in GAS T-score) and global benefits compared with change in muscle tone.

Acknowledgements

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical, an Ashfield Company, for medical writing support in preparing this poster, funded by Ipsen. Financial support for poster and manuscript preparation was also provided by the Dunhill Medical Trust.

Disclosures

KD has received honorarium and consulting fees from Ipsen, but has no financial interest in any of the materials mentioned in this poster. LTS, SA, JJ and KF have all recieved honoraria and conference attendance fees from Ipsen whilst undertaking this research but have no financial interest in any of the materials mentioned in this poster. PM and JB are employees of Ipsen.

References

1. Royal College of Physicians. Spasticity in Adults: Management Using Botulinum Toxin. National Guidelines. London, 2009.
2. Ashford S, Turner-Stokes L. *Physiother Res Int* 2013;18:178–89.
3. Institute of Medicine CoQoHCi, America. Crossing the quality chasm: A new health system for the 21st Century. Washington, D.C.: National Academy Press; 2001.
4. Holliday RC, *et al. J Neurol Neurosurg Psychiatry* 2007;78:576–80.
5. Brown M, *et al. Disabil Rehabil* 2014;36:1020–6.
6. Turner-Stokes L, *et al. J Rehabil Med* 2010;42:81–9.
7. Kiresuk T, Sherman R. *Community Ment Health J* 1968;4:443–53.
8. Ashford S, Turner-Stokes L. *Physiother Res Int* 2006;11:24–34.
9. Turner-Stokes L, *et al. BMJ Open* 2013;3:e002230.
10. Turner-Stokes L, *et al. BMJ Open* 2013;3:e002771.
11. Turner-Stokes L. *Clin Rehabil* 2009;23:362–70.

Presented at TOXINS, 18–21 January 2017, Madrid, Spain.

This study was sponsored by Ipsen