AbobotulinumtoxinA (Dysport®) injection doses per muscle in paediatric patients with lower limb spasticity

Nigar Dursun,[1] Ann Tilton,[2] Mark Gormley,[3] Marcin Bonikowski,[4] Juan Carlos Velez,[5] Maria Luisa Rodriguez,[6] Philippe Picaut,[6] Anne-Sophie Grandoulier,[6] Mauricio R. Delgado.[7]

[1]Kocaeli University Medical Faculty, Izmit, Turkey; [2]Louisiana State University Health Center and Children's Hospital New Orleans, New Orleans, LA, USA; [3]Gillette Children's Specialty Healthcare, St Paul, MN, USA; [4]Mazovian Neuropsychiatry Center, LLC Neuro Rehabilitation Department Zagórze n. Warsaw, Poland; [5]Club De Leones Cruz Del Sur Rehabilitation Center, Punta Arenas, Chile; [6]Ipsen Pharma, Les Ulis, France; [7]Texas Scottish Rite Hospital for Children, Dallas, TX, USA

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by Toxins

Introduction

- An important challenge in planning botulinum neurotoxin type A (BoNT-A) treatment for the management of cerebral palsy (CP) treatment is the identification of specific muscles to treat.[1] Depending on the functional problem, the clinician might want to target injections into the gastrocnemius, soleus, hamstrings, tibialis posterior, hip adductors and/or other proximal and distal lower limb muscles.[2]
- Once the goals and target muscles have been decided upon, it is also important to determine the dose of BoNT-A to be injected. Although the efficacy and safety of BoNT-A in managing lower limb spasticity has been well established, there is little guidance on dosing per muscle in this population.
- Here we present the doses per muscle used from a multicentre, repeated-treatment, open-label study of abobotulinumtoxinA (Dysport®) injections for lower limb spasticity due to CP.

Methods

- Phase 3, prospective, multicentre, open-label, repeated-treatment, extension study (NCT01251380) to the double-blind study reported.[3]
- Ambulatory (GMFCS I-III) paediatric patients (2-17 years old) with dynamic equinus foot deformity due to CP who completed double-blind study (≥12 weeks of follow-up) without major protocol deviations or ongoing adverse events (AEs).
- Open-label treatment consisted of a maximum of 4 Treatment Cycles (TC) of abobotulinumtoxinA at doses of 5 to 30 U/kg, occurring at intervals of ≥12 weeks (Figure 1).

Figure 1. Study design

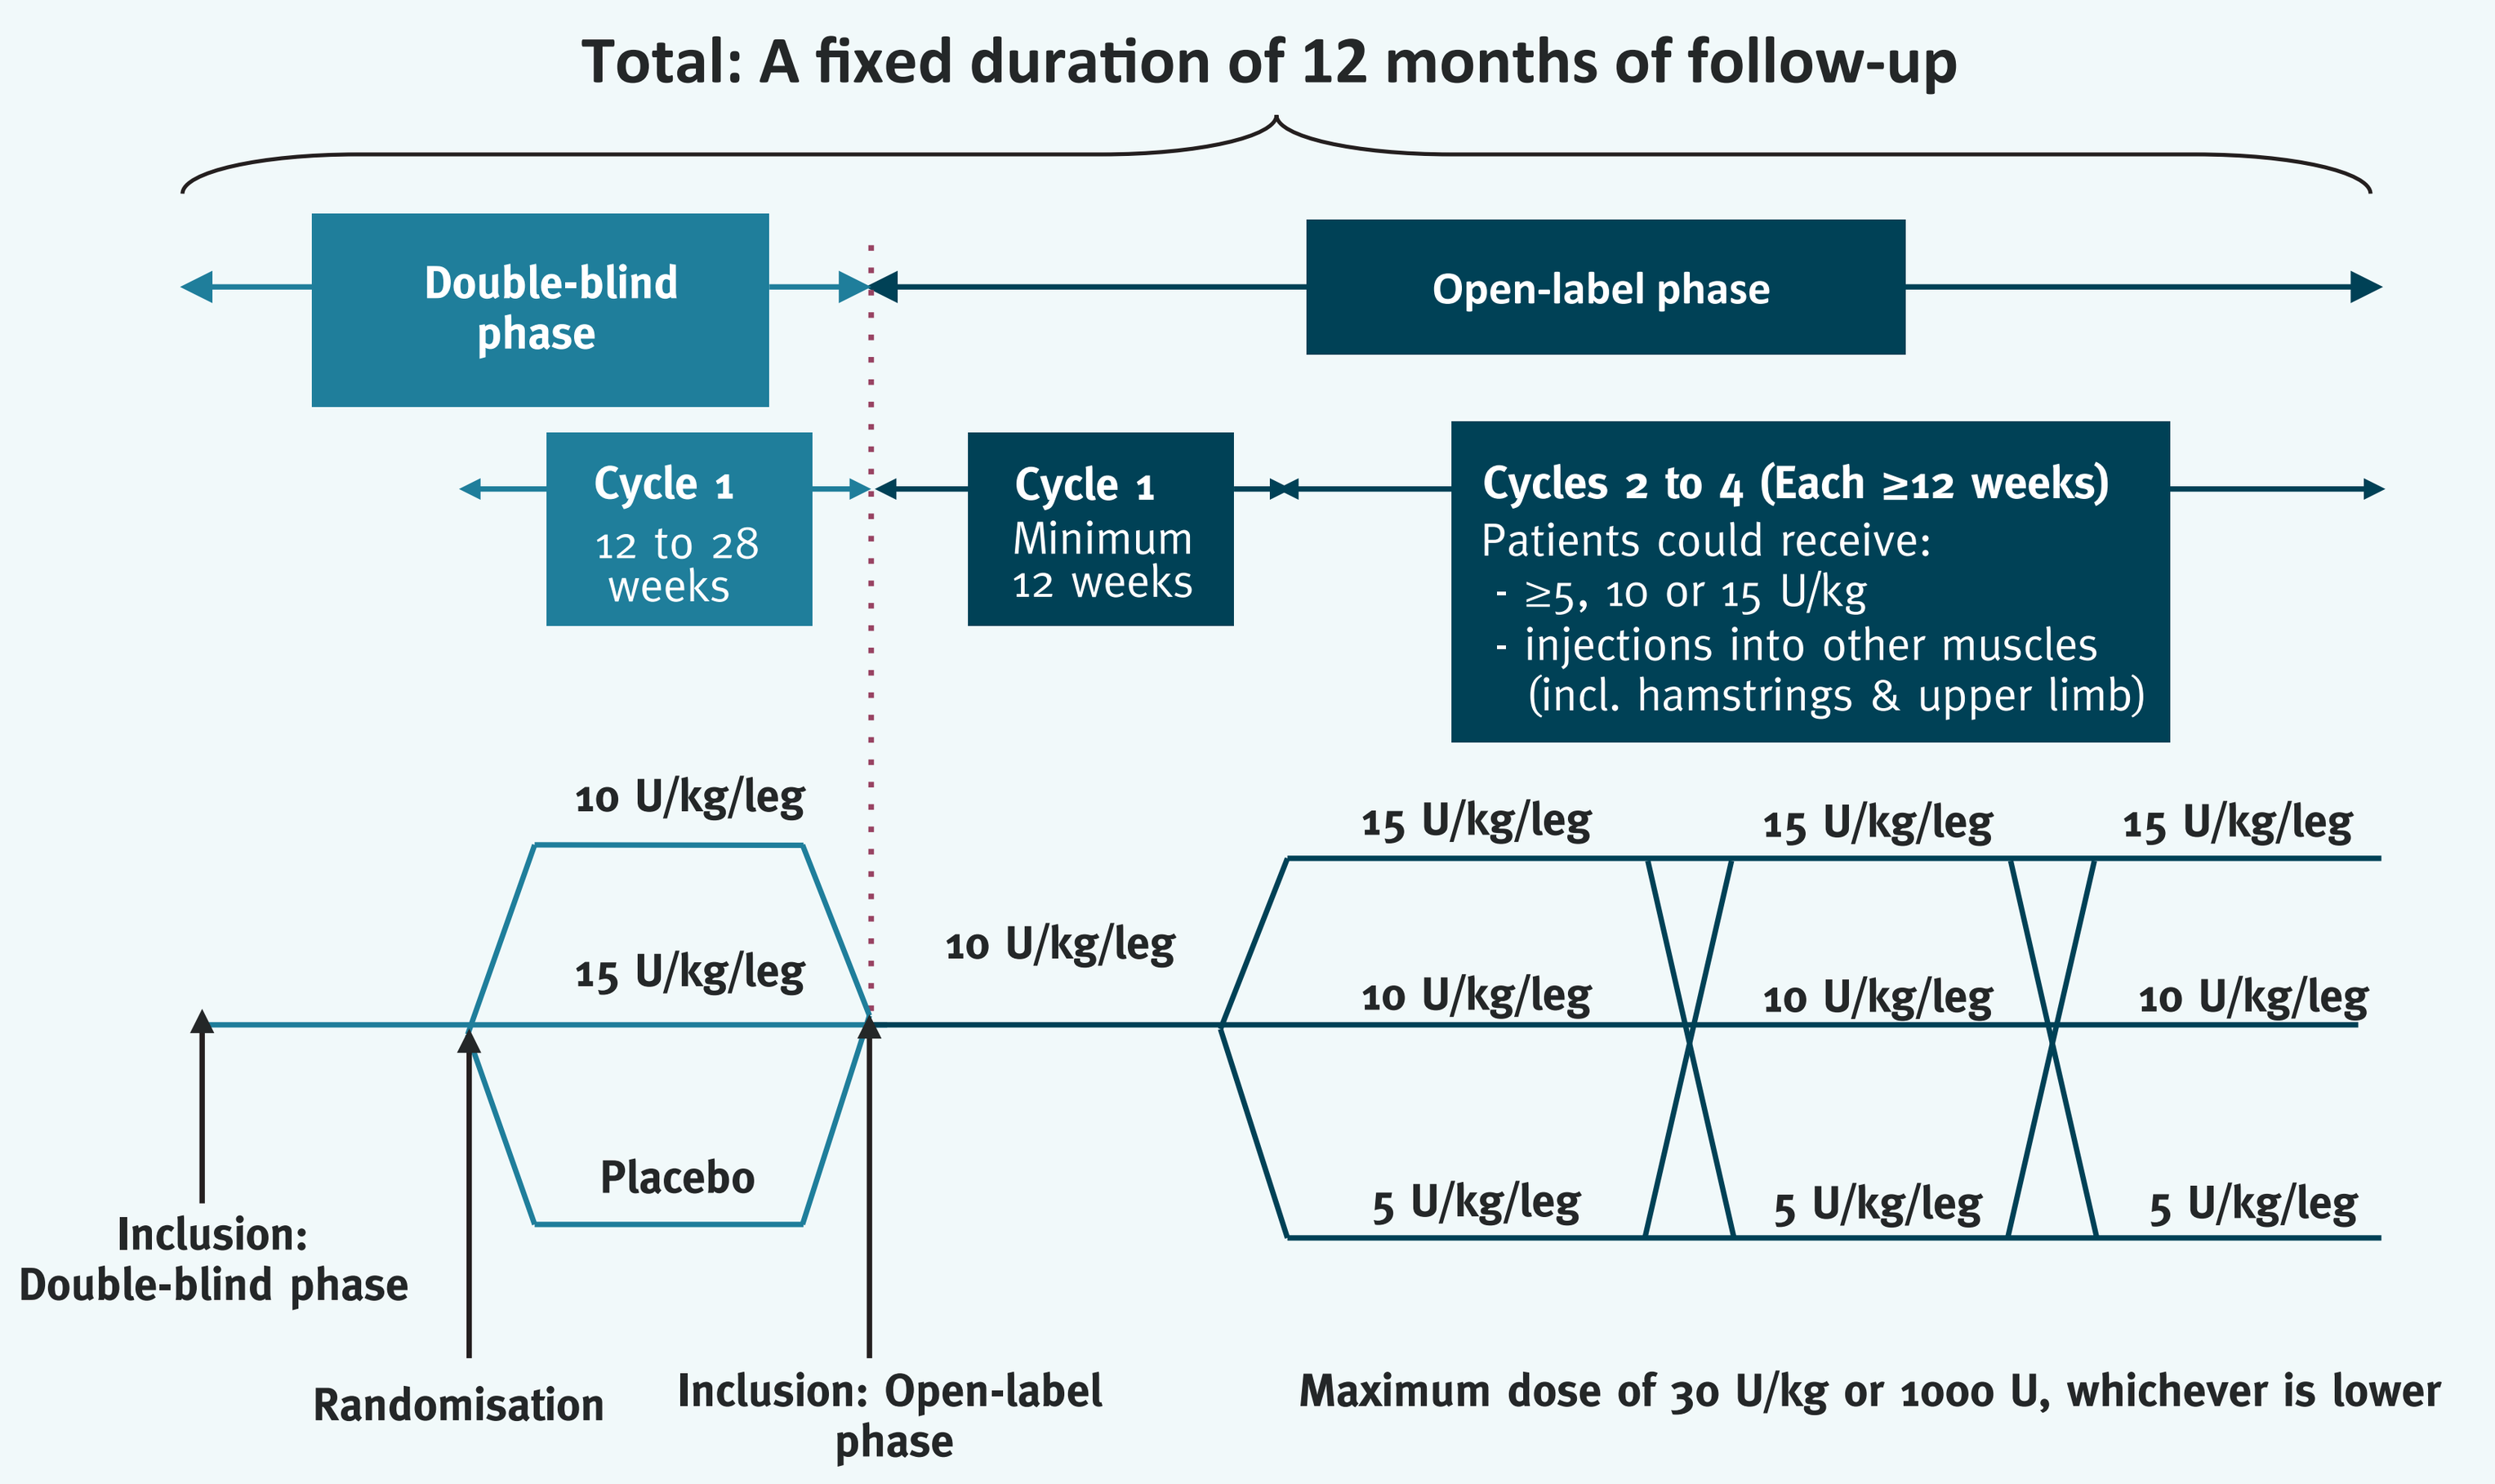
Total: A fixed duration of 12 months of follow-up
Double-blind phase
Open-label phase
Cycle 1
12 to 28 weeks
Cycle 1
Minimum 12 weeks
Cycles 2 to 4 (Each ≥12 weeks)
Patients could receive:
- ≥5, 10 or 15 U/kg
- injections into other muscles (incl. hamstrings & upper limb)
10 U/kg/leg
15 U/kg/leg
Placebo
10 U/kg/leg
15 U/kg/leg
15 U/kg/leg
15 U/kg/leg
10 U/kg/leg
10 U/kg/leg
10 U/kg/leg
5 U/kg/leg
5 U/kg/leg
5 U/kg/leg
Inclusion: Double-blind phase
Randomisation
Inclusion: Open-label phase
Maximum dose of 30 U/kg or 1000 U, whichever is lower

- In TC1, the total dose for unilateral injection (i.e. for patients with hemiparesis) was 10 U/kg and the total dose for bilateral injection (i.e. patients with diparesis or tetraparesis) was 20 U/kg. Injection of the hamstrings was also permitted in TC1.
- From TC2 onwards, the total dose could be adapted for each individual patient as clinically indicated. Although all patients continued to be injected into the gastrocnemius-soleus complex (GSC), individualised treatment could include injections of other lower limb muscles (hamstrings, tibialis posterior, hip adductors or iliopsoas) or upper limb muscles. The dose per lower limb could have been increased or decreased by no more than 5 U/kg per affected lower limb per TC (between 5-20 U/kg/leg), or remain the same. Upper limb injections had to be concomitant to lower limb injections and could not exceed 10 U/kg.
- The total dose of abobotulinumtoxinA (irrespective of muscles injected) per TC was no more than 30U/kg or 1000U, whichever was the lower value.

Table 1. Baseline demographics

	All patients (n = 216)
Sex, n (%)	
Male	130 (60.2)
Female	86 (39.8)
Age (years); Mean ± SD	5.9 ± 3.3
Aged 2-9; n (%)	183 (84.7)
Aged 10-17; n (%)	33 (15.3)
BoNT status; n (%)	
BoNT naïve	115 (53.2)
Previously BoNT treated	101 (46.8)
Pattern of paresis; n (%)	
Hemiparesis	111 (51.4)
Diparesis	96 (44.5)
Tetraparesis	9 (4.2)
GMFCS level; n (%)	
I	128 (59.3)
II	67 (31.0)
III	21 (9.7)

Table 2. Muscles treated and abobotulinumtoxinA (Dysport®) doses per muscle (Interval between injections and number of injections were left to investigator judgement)

Muscle group; n (%)	Open-label			
	Cycle 1	Cycle 2	Cycle 3	Cycle 4
Number of patients*	N=117 unilateral N=84 bilateral	N=98 unilateral N=70 bilateral	N=42 unilateral N=38 bilateral	N=3 unilateral N=5 bilateral
Number of legs injected	285	238	118	13
GSC				
Number (%) legs injected	284 (99.6%)	238 (100%)	118 (100%)	13 (100%)
Median [range] dose (U/kg/leg)	10.0 [3.5-16.0]	10.0 [2.5-15.0]	10.0 [3.9-17.0]	10.0 [7.5-15.0]
Hamstrings				
Number (%) legs injected	51 (17.9%)	64 (26.9%)	26 (22.0%)	2 (15.4%)**
Median [range] dose (U/kg/leg)	5.0 [2.0-10.0]	7.5 [2.5-10.0]	5.3 [1.5–10.0]	7.5
Tibialis posterior				
Number (%) legs injected	-	25 (10.5%)	8 (10.0%)	-
Median [range] dose (U/kg/leg)		4.0 [1.2–7.5]	4.1 [2.3–8.3]	
Hip adductors				
Number (%) legs injected	-	6 (2.5%)	2 (1.7%)**	-
Median [range] dose (U/kg/leg)		2.5 [2.3–6.5]	3.8	
Iliopsoas				
Number (%) legs injected	-	1 (0.4%)	2 (1.7%)**	-
Median [range] dose (U/kg/leg)		2.0	1.9	

* Average dosing data is presented per leg and not per patient because doses for bilateral injections could be split unequally between the 2 legs.

**1 patient injected bilaterally into relevant muscle (i.e. 2 legs). GSC=gastrocnemius/soleus complex.

Results

Patient disposition and baseline characteristics

- Of 241 patients randomised in the double-blind study, 216 (90%) patients entered the open-label extension study and 207 patients received at least another abobotulinumtoxinA injection cycle. Only 11 patients required a fourth injection within the open-label study timeframe and entered TC4.
- Baseline demographics and clinical characteristics are summarised in Table 1.

Treatment exposure

- During the open-label study, investigators chose to inject non-GSC muscles in up to 25% of patients, as per the individual pattern of spasticity and treatment goals (Table 2).

Efficacy and safety

- Efficacy of abobotulinumtoxinA in the double-blind phase was maintained across open-label cycles. Improvement (approximately 1 grade on MAS at the ankle joint) was similar across TC 1–3, and comparable to that in the double-blind study.[3] In those patients who received proximal injections, treatment with abobotulinumtoxinA also improved muscle tone in the knee flexors.
- The only treatment-related TEAE reported by >2 patients was injection site pain (n=10). There were no differences in the number of AEs dose. Seven (3.4%) subjects reported serious AE, all considered unrelated to study treatment.

Conclusions

- Flexibility in BoNT-A dosing and injection intervals (74% of patients had an injection interval of at least 16 weeks in this study) is key when tailoring injection schemes to meet the functional goals of each individual child with CP. These data provide dosage ranges for abobotulinumtoxinA injections into proximal and distal leg muscles, based on fixed dosing of 5-15 U/kg/leg in the GSC, that have shown a favourable risk – benefit profile in children with dynamic equinus due to CP with a GMFCS level I to III.
- In this population of patients, the most commonly targeted muscles were the gastrocnemius and soleus (mandatory in this study design) followed by the hamstrings and the tibialis posterior (per investigator judgment). The adductors and iliopsoas were rarely injected in this population.

Acknowledgements

This study was funded by Ipsen Pharma. The authors would like to thank all investigators and patients involved in the studies, and Anita Chadha-Patel (ACP Clinical Communications Ltd) for writing assistance (funded by Ipsen).

Disclosures

All authors (external from Ipsen) have been investigators in Ipsen-sponsored clinical trials (including the present study) and they or their institutions have received payment for participation. No author has received financial remuneration for the preparation of this abstract or poster.

References

1. Strobl W, Toxins (Basel) 2015;7(5):1629-1648; 2. Bar-On L, One 2014;9(3):e91759; 3. Delgado MR, Pediatrics 2016;137(2):1-9.

Presented at TOXINS, 18–21 January, 2017, Madrid, Spain.

This study was sponsored by Ipsen.