Improvement in poststroke upper limb spasticity patterns after treatment with Botulinum toxin A: A prospective observational study

Josep Valls-Solé[a,*], Fernanda Maria Caballero[b], Maria José Durà[c], Lluis Guirao[d], Montserrat Bernabeu[e], Helena Bascuñana[f], Irene Garcia[g], Pascal Maisonobe[h]

[a]Hospital Clínic i Provincial de Barcelona, Barcelona, Spain; [b]Hospital Parc Taulí, Sabadell, Spain; [c]Hospital Germans Trias i Pujol, Badalona, Spain; [d]Hospital de Mataró, Mataró, Spain; [e]Institut Guttmann, Badalona, Spain; [f]Hospital de Sant Pau, Barcelona; [g]Ipsen Pharma, Medical Department, Barcelona, Spain; [h]Corporate Ipsen Biostatistics, Boulogne-Billancourt, France

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

QR code to be provided by Toxins

Introduction and Objectives

- Botulinum toxin A (BoNT-A) is an effective treatment for poststroke spasticity. However, more data are needed to assess its beneficial effects in real-life clinical settings.
- The aim of this study was to document functional changes in upper limb poststroke spasticity before and after treatment with BoNT-A.

Methods

- Postmarketing, multicenter, prospective, observational, longitudinal, open, single-cohort study was conducted in 37 BoNT-A-naive adults (age ≥18 years), diagnosed with poststroke upper limb spasticity in Catalonia, Spain. The study enrollment period was December 19, 2012 to June 10, 2014.
- All patients received one cycle of BoNT-A injections. Patients were assessed at baseline (pre-BoNT-A treatment) and at 3 to 6 months post-treatment.
- The primary outcome measure, assessed at baseline and post-treatment, was change in spasticity patterns. Additional study variables included the Modified Ashworth Scale (MAS) and Goal Attainment Scale (GAS). The Paired T-test (non-parametric bootstrap) was used to test for differences.

Results

- Thirty-four of the 37 patients (92%) completed the study. Median age was 62 years (range: 33 to 86 years). The majority of subjects (70.6%) were males. At baseline, the most common spasticity patterns were elbow flexion (n=24; 70.6%), shoulder adduction/internal rotation (n=23; 67.6%), and wrist flexion (n=22; 64.7%).

Table 1. Demographic data and baseline characteristics – PAS population (n=34)

Age, years, median (range)	62 (33-86)
Male/Female, n (%)	24 (70.6) / 10 (29.4)
Pattern of spasticity, n (%)	
Elbow in flexion position	24 (70.6)
Shoulder in adduction/internal rotation	23 (67.6)
Wrist in flexion position	22 (64.7)
Claw hand	20 (58.8)
Forearm in prone position	19 (55.9)
Subject autonomy, n (%)	
Lives alone	1 (2.9)
Lives in a residence	5 (14.7)
Lives in his / her own home	11 (32.4)
Lives with a relative as a caregiver	14 (41.2)
Has the assistance of a relative as caregiver	3 (8.8)
Has the assistance of a part-time professional caregiver	3 (8.8)
Other situation	2 (5.9)
Height, cm, mean (SD)	166.0 (10.4)
Weight, Kg, mean (SD)	72.9 (15.3)
BMI, Kg/m², mean (SD)	26.3 (4.6)

PAS = Primary Analysis Set. BMI = Body Mass Index.

- The total number of spasticity patterns decreased from 110 (3.24/patient) at baseline to 102 (3.0/patient) post-treatment. The most frequently injected muscles were the flexor digitorum superficialis (50% of patients) and the flexor carpi radialis (47.1% of patients).
- Overall MAS scores decreased significantly from baseline to final study visit ($P<0.0001$), from a mean (SD) of 2.7 (0.8) at baseline to 2.2 (0.7) post-treatment, with highly significant decreases in claw hand, elbow flexion, forearm prone position, shoulder adduction and wrist flexion (all $P<0.0001$).
- Visual Analog Scale (VAS) pain scores improved significantly ($P<0.0001$) from a mean (SD) of 50.8 (35.7) at baseline to 17.4 (23.7).

Table 2. Secondary efficacy outcomes – PAS population (n=34)

	Baseline	Final Visit	
Derived MAS score[a], mean (SD)	2.7 (0.8)	2.2 (0.7)	$P<0.0001$
Pain score (VAS), mean (SD)	50.8 (35.7)	17.4 (23.7)	$P<0.0001$
Unweighted GAS T-score, mean (SD)	35.5 (5.7)	55.0 (9.8)	$P<0.0001$

[a]MAS scored on a derived scale where a score of 0 = 0, 1 = 1, +1 = 1.5, 2 = 2, 3 = 3, and 4 = 4.
PAS=Primary Analysis Set

- The majority of patients (28/34; 82.4%) achieved their primary GAS objective, with the most common being pain reduction (achieved in 14/17 pts; 82.4%) and improved self-care (7/8 pts; 87.5%). The mean (SD) unweighted GAS T-score also improved significantly, from 35.5 (5.7) at baseline to 55.0 (9.8) post-treatment ($P<0.0001$).
- One death, unrelated to treatment, was reported.

Figure 1. Goals set at baseline.

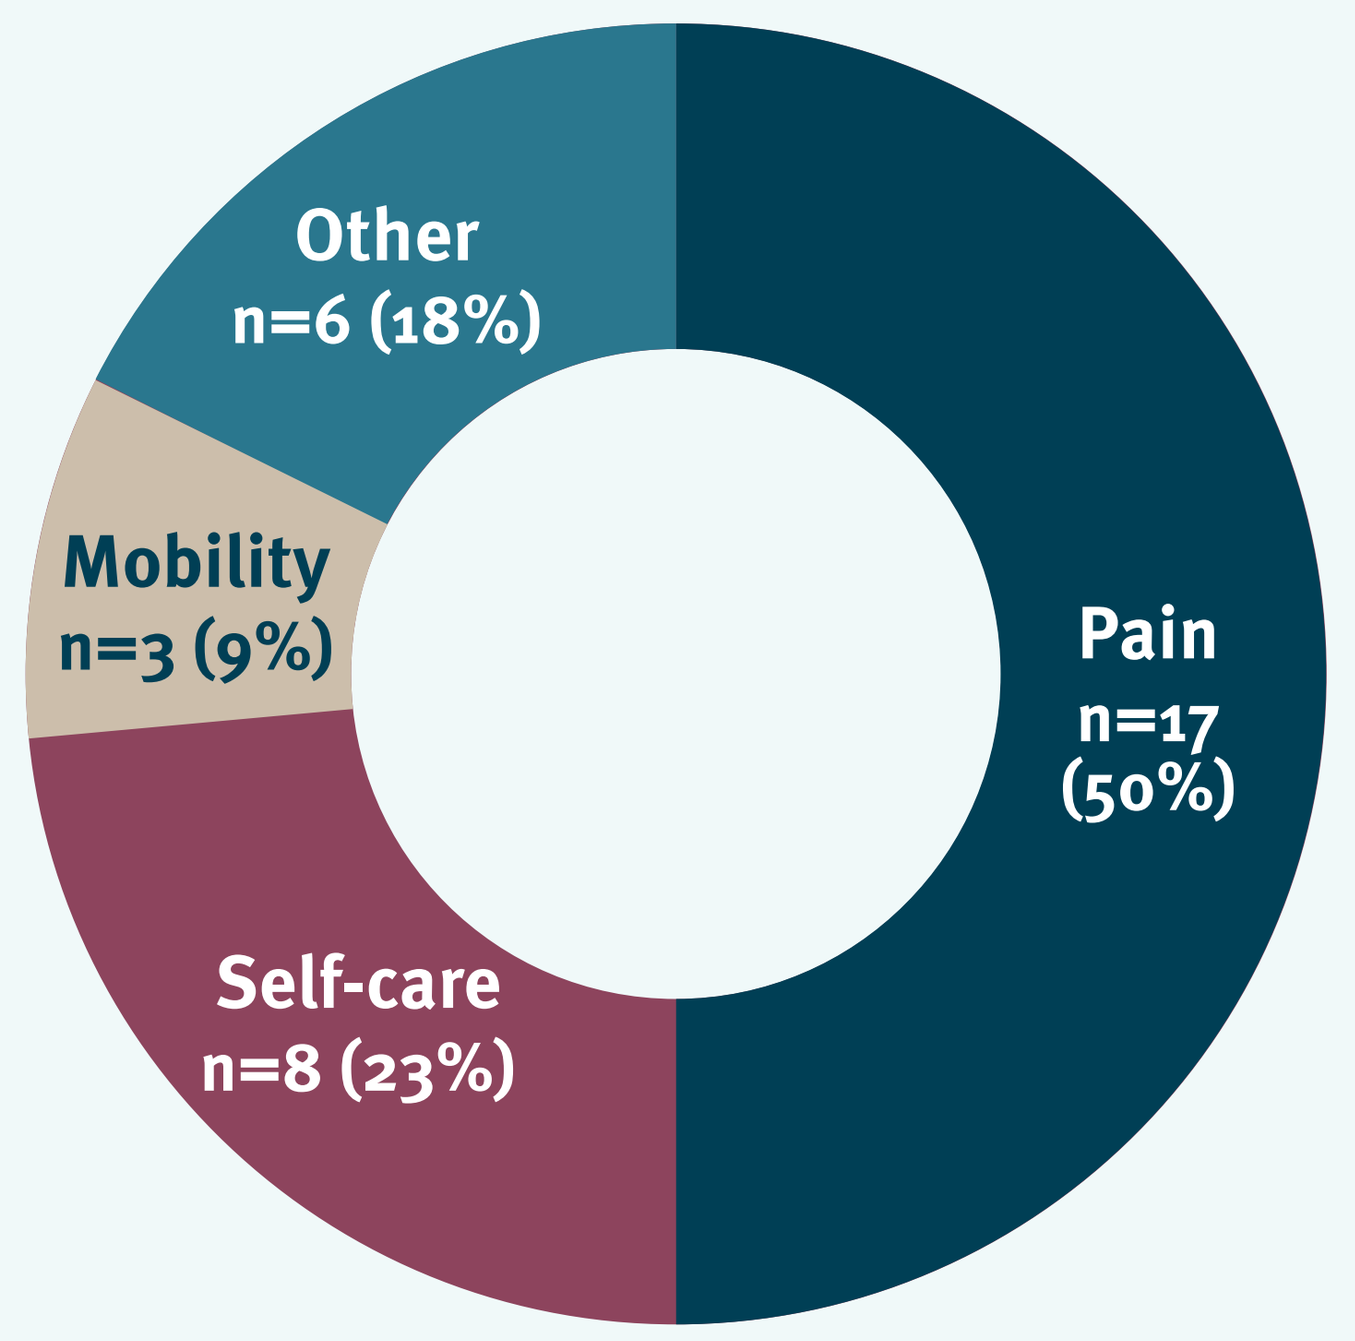
Other
n=6 (18%)
Mobility
n=3 (9%)
Pain
n=17
(50%)
Self-care
n=8 (23%)

Figure 2. Goal achievement after one cycle of BONT-A injection.

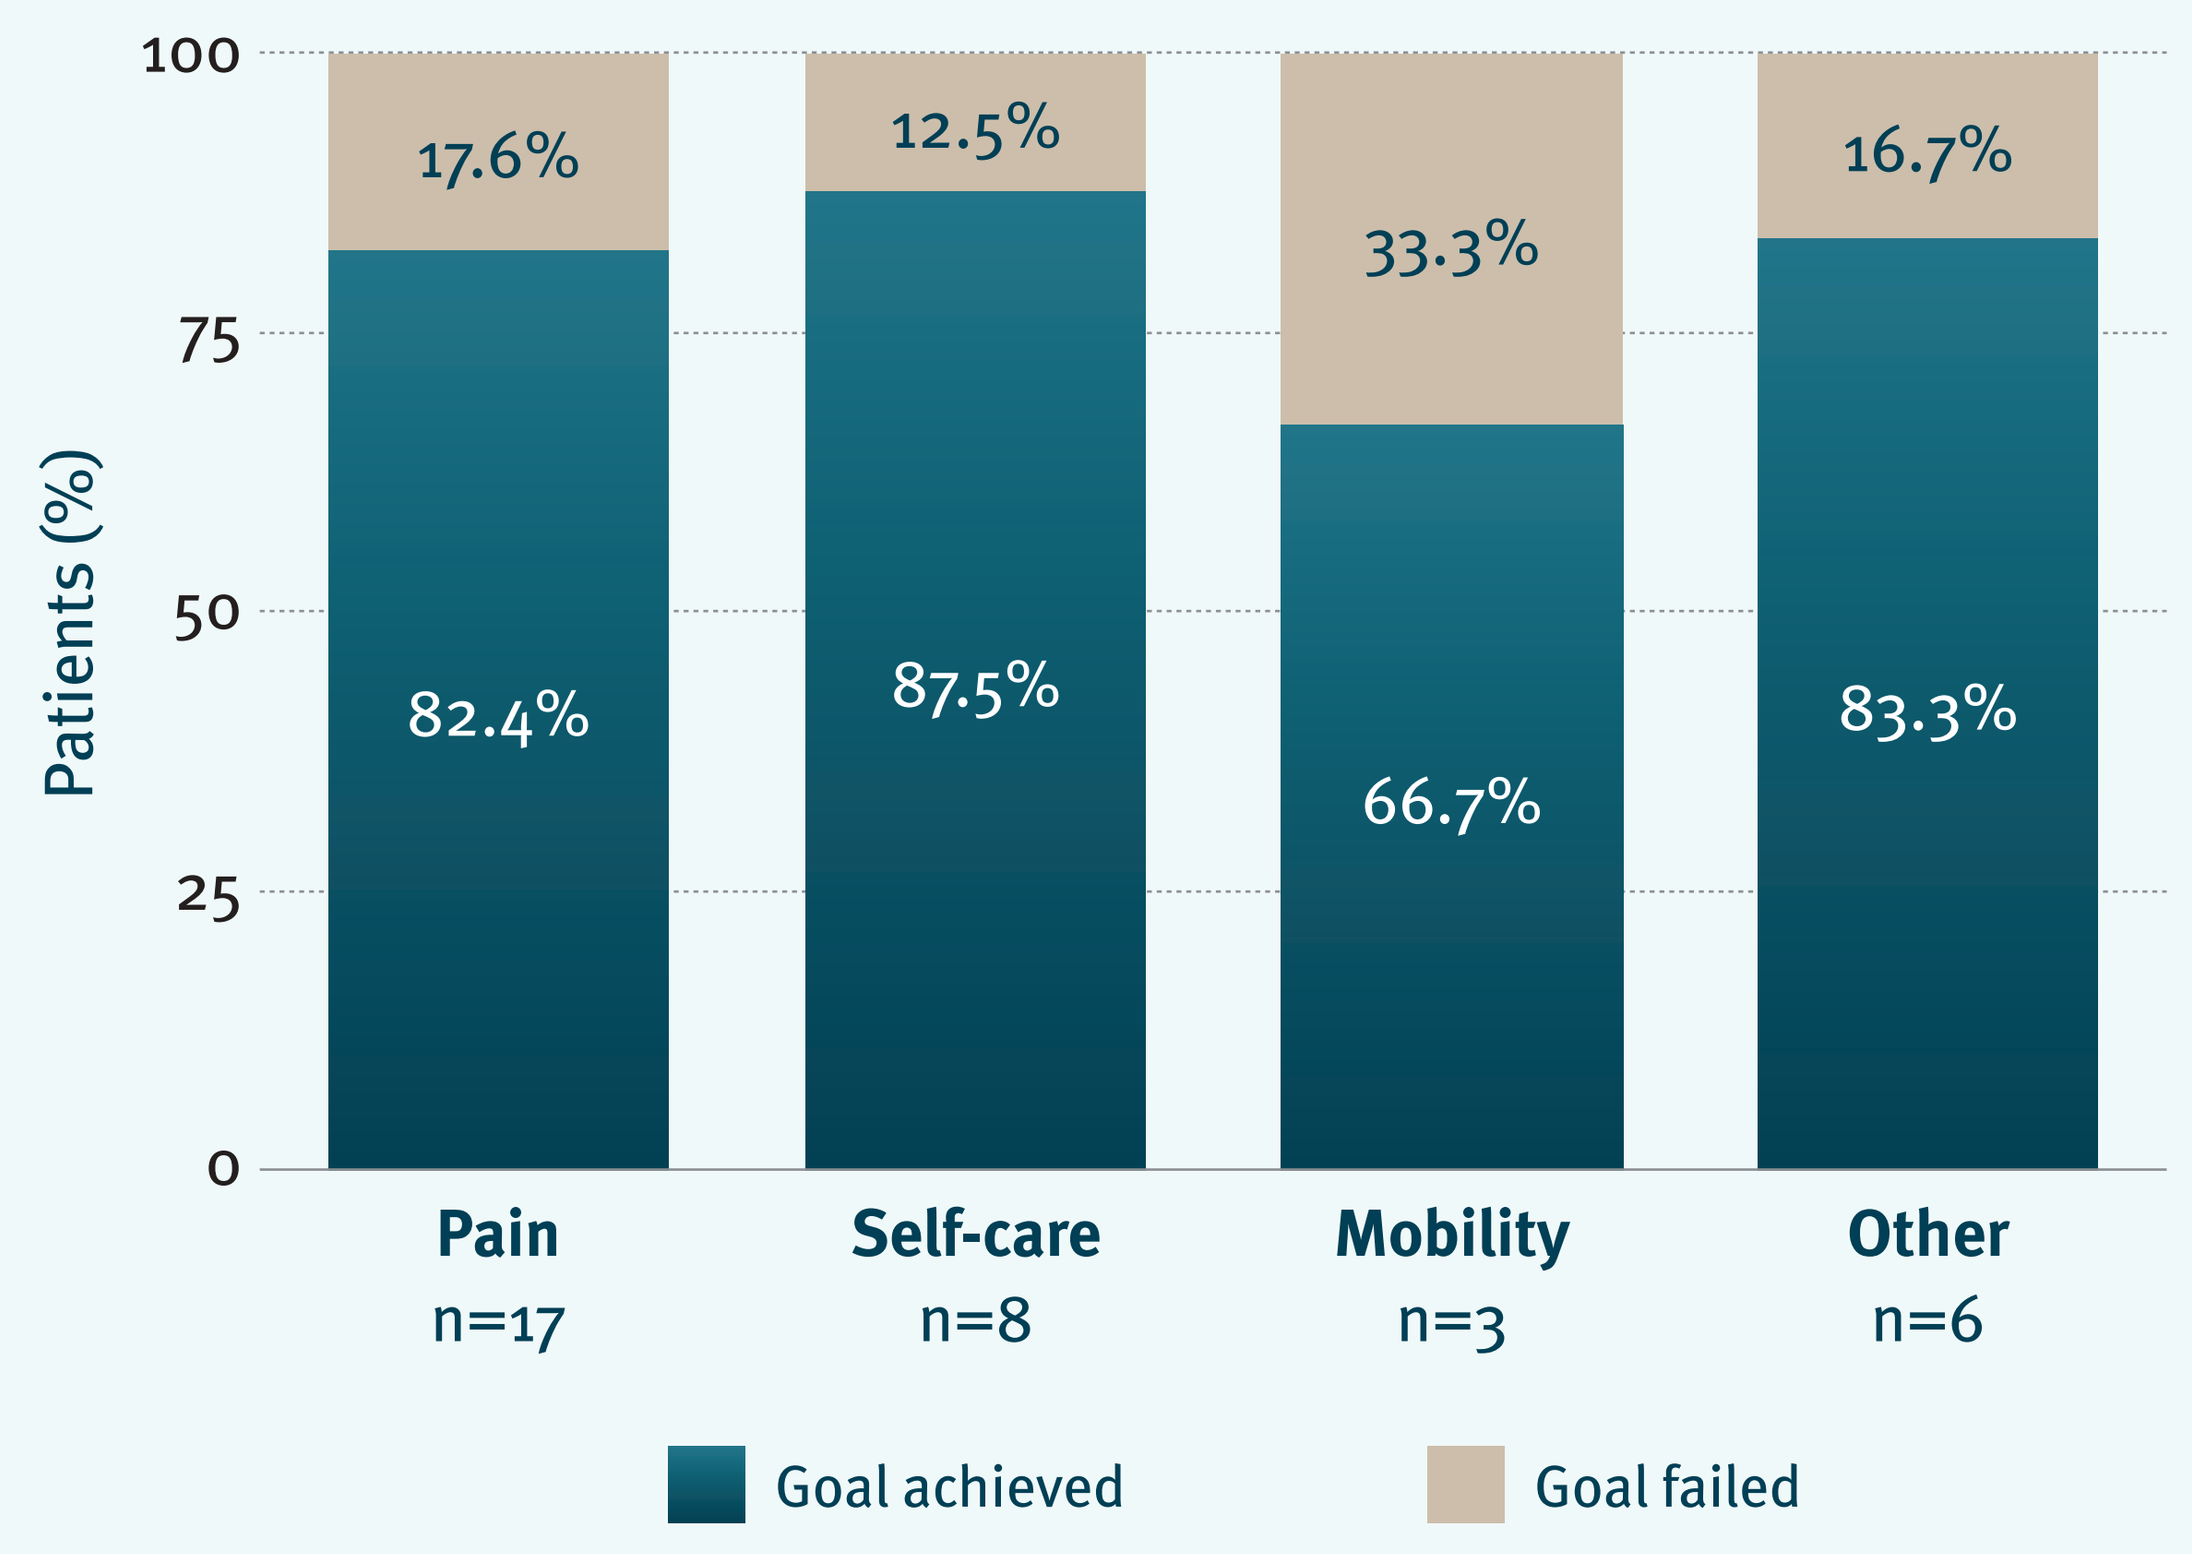
100
75
50
25
0
Patients (%)
17.6%
82.4%
12.5%
87.5%
33.3%
66.7%
16.7%
83.3%
Pain
n=17
Self-care
n=8
Mobility
n=3
Other
n=6
Goal achieved
Goal failed

Conclusions

- In this sample of patients with severe spasticity, the majority of patients benefited from treatment with a single cycle of BoNT-A injections, as evidenced by the significant improvements in pain, MAS, and GAS T-scores.
- These data, obtained in a real-life setting, further support the use of BoNT-A for the treatment of poststroke spasticity.

Acknowledgments:

We thank all the patients who participated in this study and their families. The clinical research organisation responsible for the study was Linical. The poster was written with editorial assistance from Bradley Londres.

Presented at TOXINS, 18–21 January 2017, Madrid, Spain

This study was sponsored by Ipsen