Impact of Early Intervention With OnabotulinumtoxinA Treatment in Adult Patients With Post-Stroke Lower Limb Spasticity

Atul T. Patel,[a] Anthony B. Ward,[b] Carolyn Geis,[c] Chengcheng Liu,[d] Wolfgang H. Jost,[e] Rozalina Dimitrova[f]

[a]Kansas City Bone and Joint Clinic, Overland Park, KS; [b]North Staffordshire Rehabilitation Centre, Staffordshire University, Stoke on Trent, United Kingdom; [c]Department of Rehabilitation Medicine, Halifax Health, Brooks Rehabilitation, Daytona Beach, FL; [d]Biostatistics, Allergan plc, Bridgewater, NJ; [e]University of Freiburg, Department of Neurology, Freiburg, Baden-Württemberg, Germany; [f]Neurology and Pain Clinical Development, Allergan plc, Irvine, CA

Scan to obtain PDF of poster

INTRODUCTION

- The REFLEX Study demonstrated the clinical efficacy and safety of onabotulinumtoxinA in patients with post-stroke lower limb spasticity (PSLLS), leading to its approval for the treatment of PSLLS in adults in the United States[1]
 - Improvements in ankle spasticity, as measured by the Modified Ashworth Scale (MAS), the Clinical Global Impression of Change (CGI), and the Goal Attainment Scale (GAS), were significantly greater for those treated with onabotulinumtoxinA compared to placebo
- Preliminary findings in patients with mixed upper or lower limb spasticity support a benefit of early treatment[2]
 - Little is known regarding the relationship between the timing of onabotulinumtoxinA treatment and clinical outcomes among those with PSLLS

OBJECTIVE

- To evaluate the impact of timing of treatment initiation since stroke on the efficacy of onabotulinumtoxinA in patients with PSLLS in the REFLEX Study

METHODS

Study Design

- Multicenter, global, randomized, double-blind, placebo-controlled Phase 3 study undertaken in 60 global study centers of onabotulinumtoxinA for treatment of PSLLS followed by a repeated-treatment, open-label extension **(Figure 1)**
 - Complete primary methods and results have been published
 - The current analysis focuses on findings from the double-blind phase with respect to the timing of treatment initiation since stroke (i.e., ≤24 months or >24 months)

Figure 1. Study Design

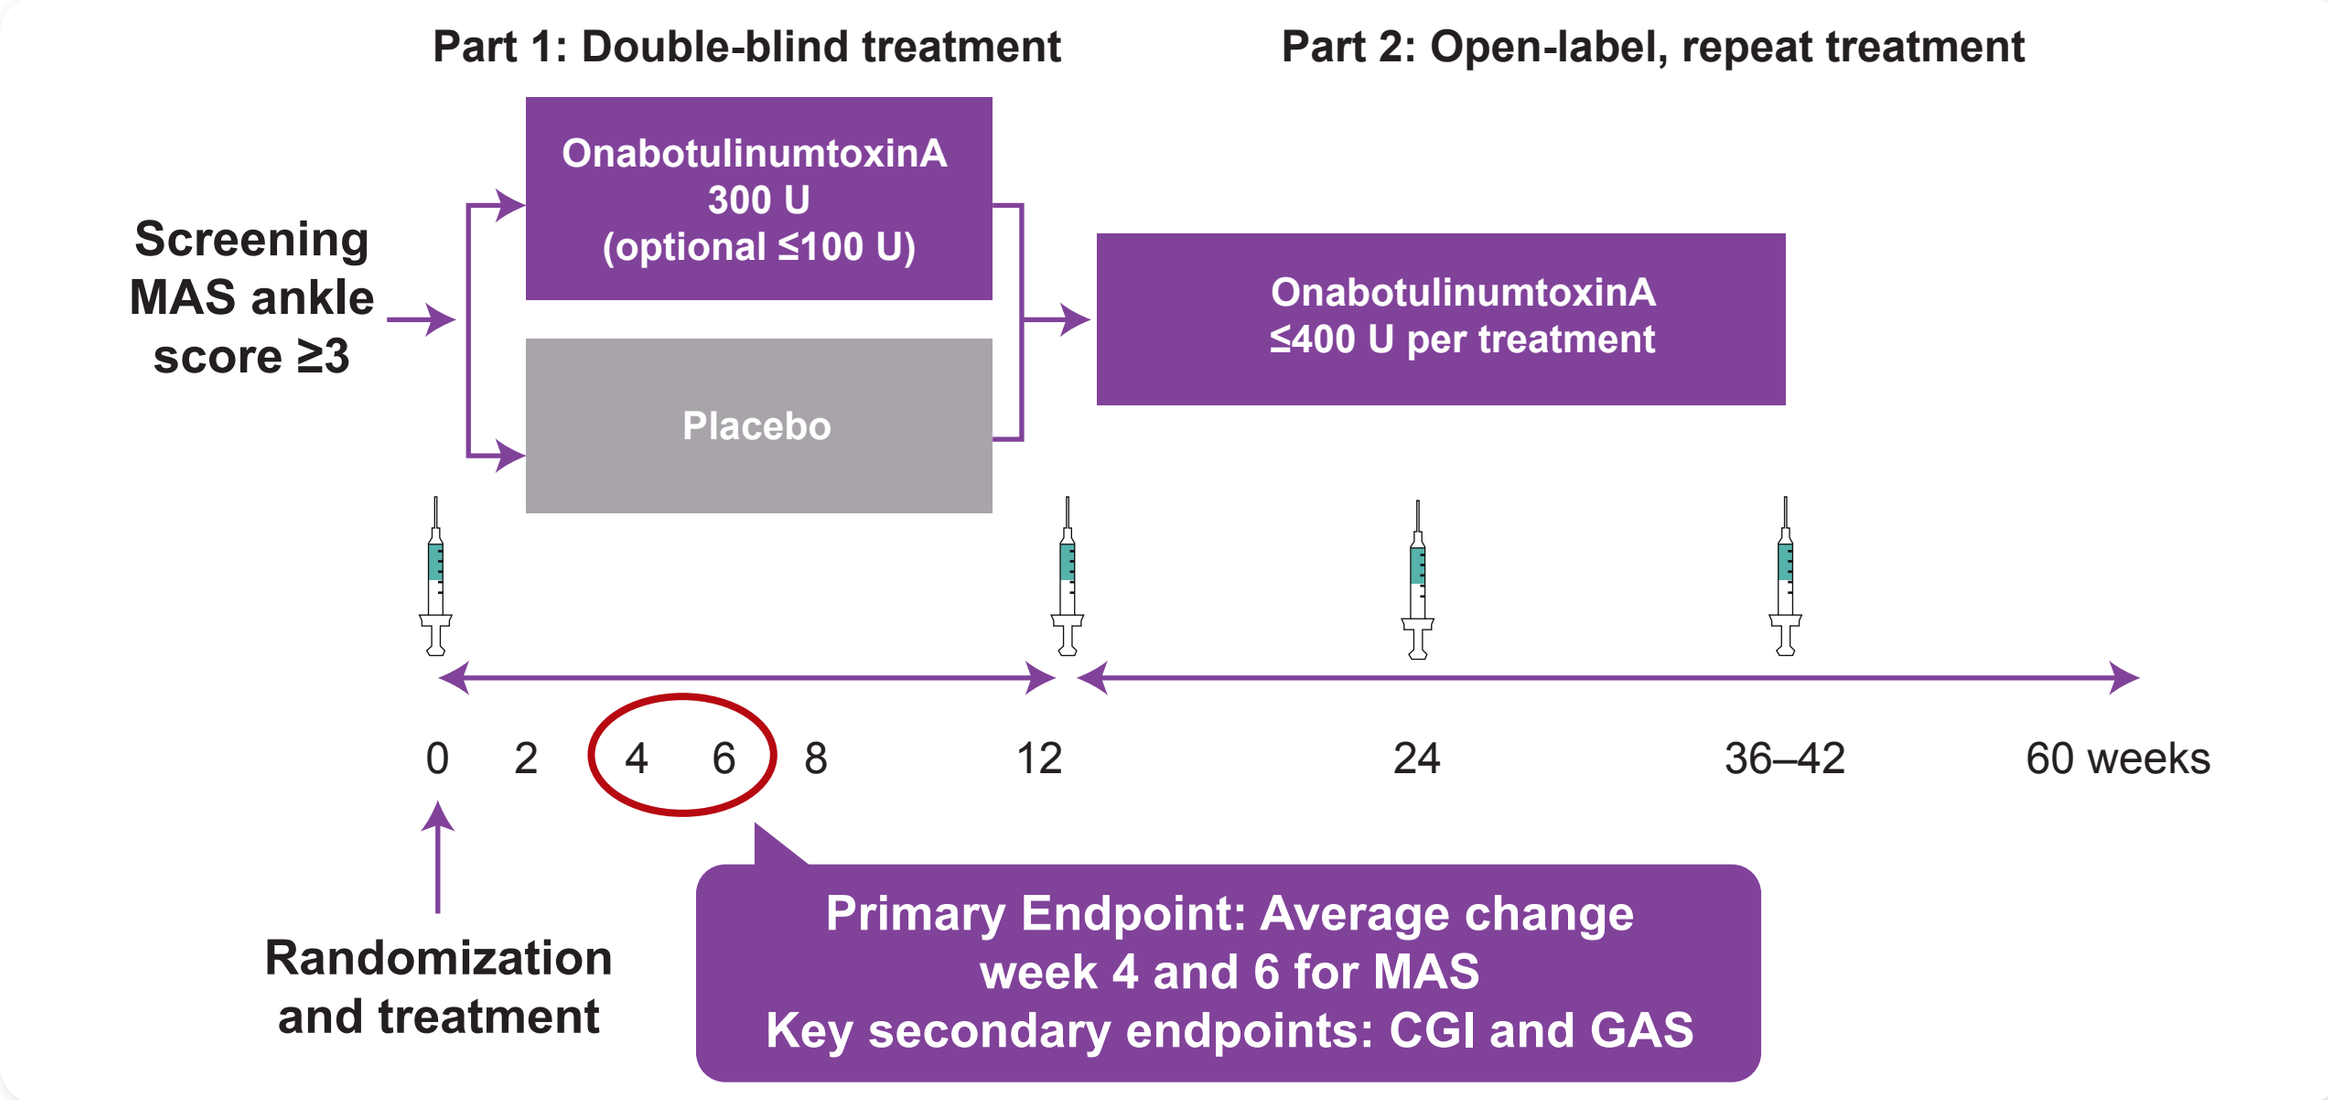

Part 1: Double-blind treatment
Part 2: Open-label, repeat treatment
Screening MAS ankle score ≥3
OnabotulinumtoxinA 300 U (optional ≤100 U)
Placebo
OnabotulinumtoxinA ≤400 U per treatment
0
2
4
6
8
12
24
36–42
60 weeks
Randomization and treatment
Primary Endpoint: Average change week 4 and 6 for MAS
Key secondary endpoints: CGI and GAS

MAS=Modified Ashworth Scale.

- Enrolled patients were men and women aged 18–85 years with diagnosed PSLLS (determined by MAS score ≥3 in ankle plantar flexors), who were naïve to or had not received onabotulinumtoxinA for ≥20 weeks (spasticity indication) or ≥12 weeks (any other indication), with the most recent stroke occurring ≥3 months before screening
- Subjects received intramuscular injections of onabotulinumtoxinA 300 U or placebo into 3 sites each of the gastrocnemius (medial and lateral head), soleus, and tibialis posterior muscles (i.e., mandatory ankle muscles) **(Figure 2)**
 - An optional dose of up to 100 U onabotulinumtoxinA or placebo was injected into the flexor digitorum longus, flexor digitorum brevis, flexor hallucis longus, extensor hallucis, and/or rectus femoris muscles if clinically indicated

Figure 2. Mandatory and Optional Dose and Muscles for Injection

Mandatory Ankle Muscles	Dose
Gastrocnemius medial head	75 U (25 U × 3 sites)
Gastrocnemius lateral head	75 U (25 U × 3 sites)
Soleus	75 U (25 U × 3 sites)
Tibialis posterior	75 U (25 U × 3 sites)
Optional Muscles	**Dose**
Flexor digitorum longus	50 U (25 U × 2 sites)
Flexor digitorum brevis	25 U (1 site)
Flexor hallucis longus	50 U (25 U × 2 sites)
Extensor hallucis	25 U (1 site)
Rectus femoris	100 U (25 U × 4 sites)

Mandatory Ankle Injection Locations

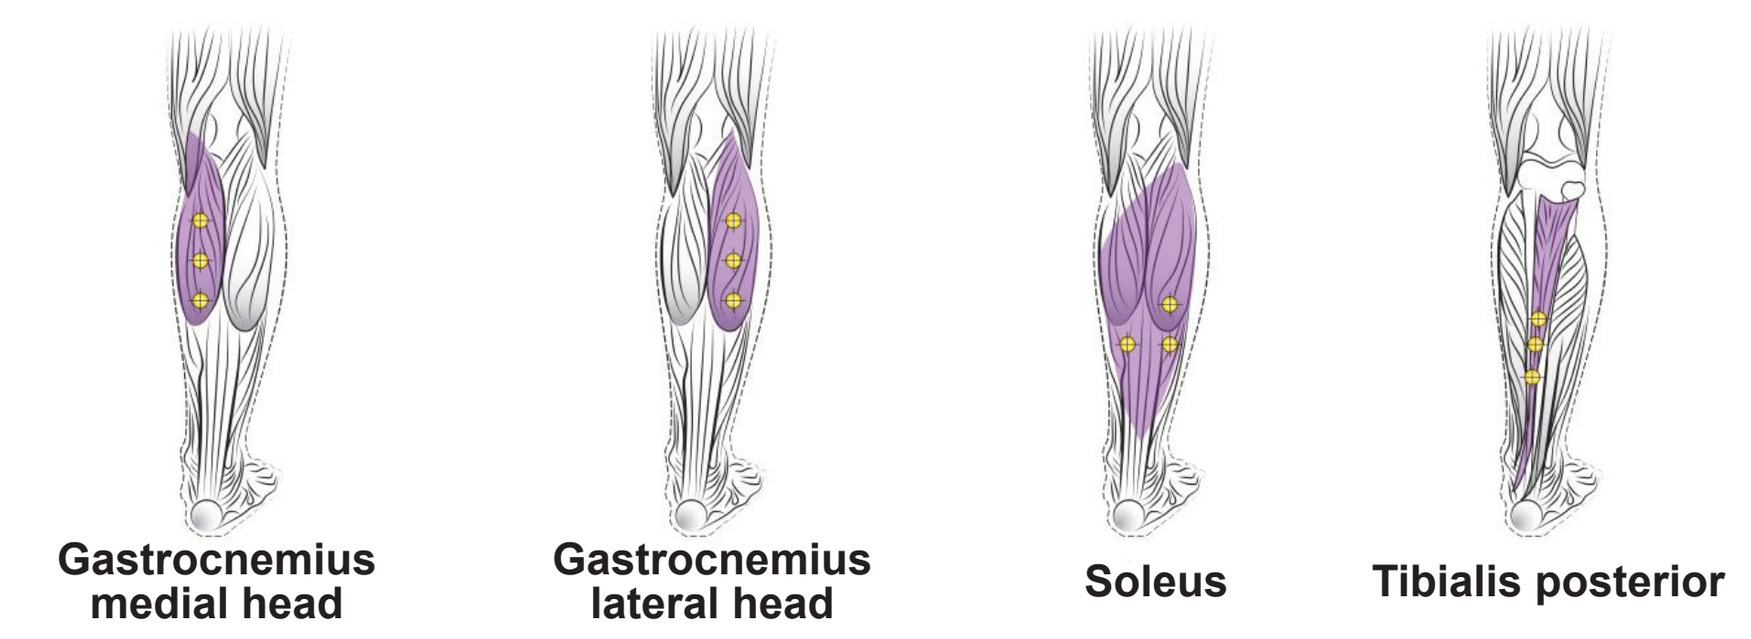

Gastrocnemius medial head
Gastrocnemius lateral head
Soleus
Tibialis posterior

Assessments

- Primary outcome: Ankle MAS change from baseline to average score of weeks 4 and 6
- Secondary outcomes:
 - Clinical Global Impression of Change (CGI) by physician average of weeks 4 and 6
 - Goal Attainment Scale (GAS) by physician and patient (active and passive goals) at weeks 8 and 12
 - Assessment of the GAS scores was defined based on the absolute GAS scores and the proportion of patients who achieved active or passive goals (GAS score of ≥0) and the proportion of patients who made progress toward active or passive goals (GAS score of ≥–1)
- Adverse events (AEs) including severity and relation to study drug were recorded at baseline and at each follow-up visit

Statistical Analysis

- Efficacy was evaluated using the intent-to-treat population; safety was evaluated with all treated patients
- Primary efficacy endpoint and CGI: *P* values and 95% confidence intervals for between-group comparisons were obtained by covariance (ANCOVA) model including treatment and investigational site as factors, with baseline ankle MAS and muscle group injected as covariates
- GAS goal achievement and goal improvement: Between-group comparison *P* values determined by Pearson chi-square testor Fisher exact test
- Treatment differences and 95% confidence intervals for MAS, CGI, and GAS scores in patients treated ≤24 months vs >24 months since stroke

RESULTS

Disposition and Demographics

- 468 patients were randomized to onabotulinumtoxinA (n=233: ≤24 months, n=80; >24 months, n=153) or placebo (n=235: ≤24 months, n=73; >24 months, n=162)
 - 447 (95.5%) patients completed the double-blind phase
- No significant baseline demographic differences were observed between patients treated ≤24 months or >24 months since stroke; however, compared with patients treated ≤24 months since stroke, patients treated >24 months since stroke were approximately 2 years older and were slightly heavier **(Table 1)**

Table 1. Baseline Demographics

	Time since stroke ≤24 months		Time since stroke >24 months	
Characteristic	OnabotulinumtoxinA n=80	Placebo n=73	OnabotulinumtoxinA n=153	Placebo n=162
Mean (SD) age, y	54.3 (11.9)	55.7 (12.0)	56.9 (12.9)	57.5 (11.8)
Male, n (%)	55 (68.8)	52 (71.2)	93 (60.8)	103 (63.6)
Caucasian, n (%)	68 (85.0)	62 (84.9)	116 (75.8)	132 (81.5)
Mean (SD) weight, kg	76.5 (14.8)	77.9 (14.4)	82.1 (18.9)	80.3 (15.9)
Mean (SD) time since stroke, y	1.1 (0.5)	1.1 (0.5)	8.0 (6.5)	7.0 (6.7)
Stroke severity*, n (%)				
Mild	7 (8.8)	10 (13.7)	16 (10.5)	15 (9.3)
Moderate	55 (68.8)	42 (57.5)	105 (68.6)	108 (66.7)
Severe	18 (22.5)	21 (28.8)	32 (20.9)	39 (24.1)

Kg=kilogram; mo=months; SD=standard deviation; y=years.
*Severity score: mild (minor deficit, functionally non-impairing), moderate (moderate deficit significantly interfering with activities of daily living), severe (dependent, requiring chronic care).

Efficacy

- OnabotulinumtoxinA treatment significantly improved MAS (–0.81 vs –0.61; *P*=0.01), CGI (0.86 vs 0.65; *P*=0.01) and passive GAS scores (wk 12, –0.6 vs –0.9; *P*=0.042) compared with placebo
- Mean change in MAS (average weeks 4 and 6) was superior for patients treated ≤24 months vs >24 months since stroke (mean change –0.31 [*P*=0.02 vs placebo] vs –0.17 [*P*=0.09 vs placebo], respectively **[Figure 3]**), representing a mean difference improvement of –0.14 for the earlier, compared to the later-treated subgroup

Figure 3. Ankle MAS Change From Baseline

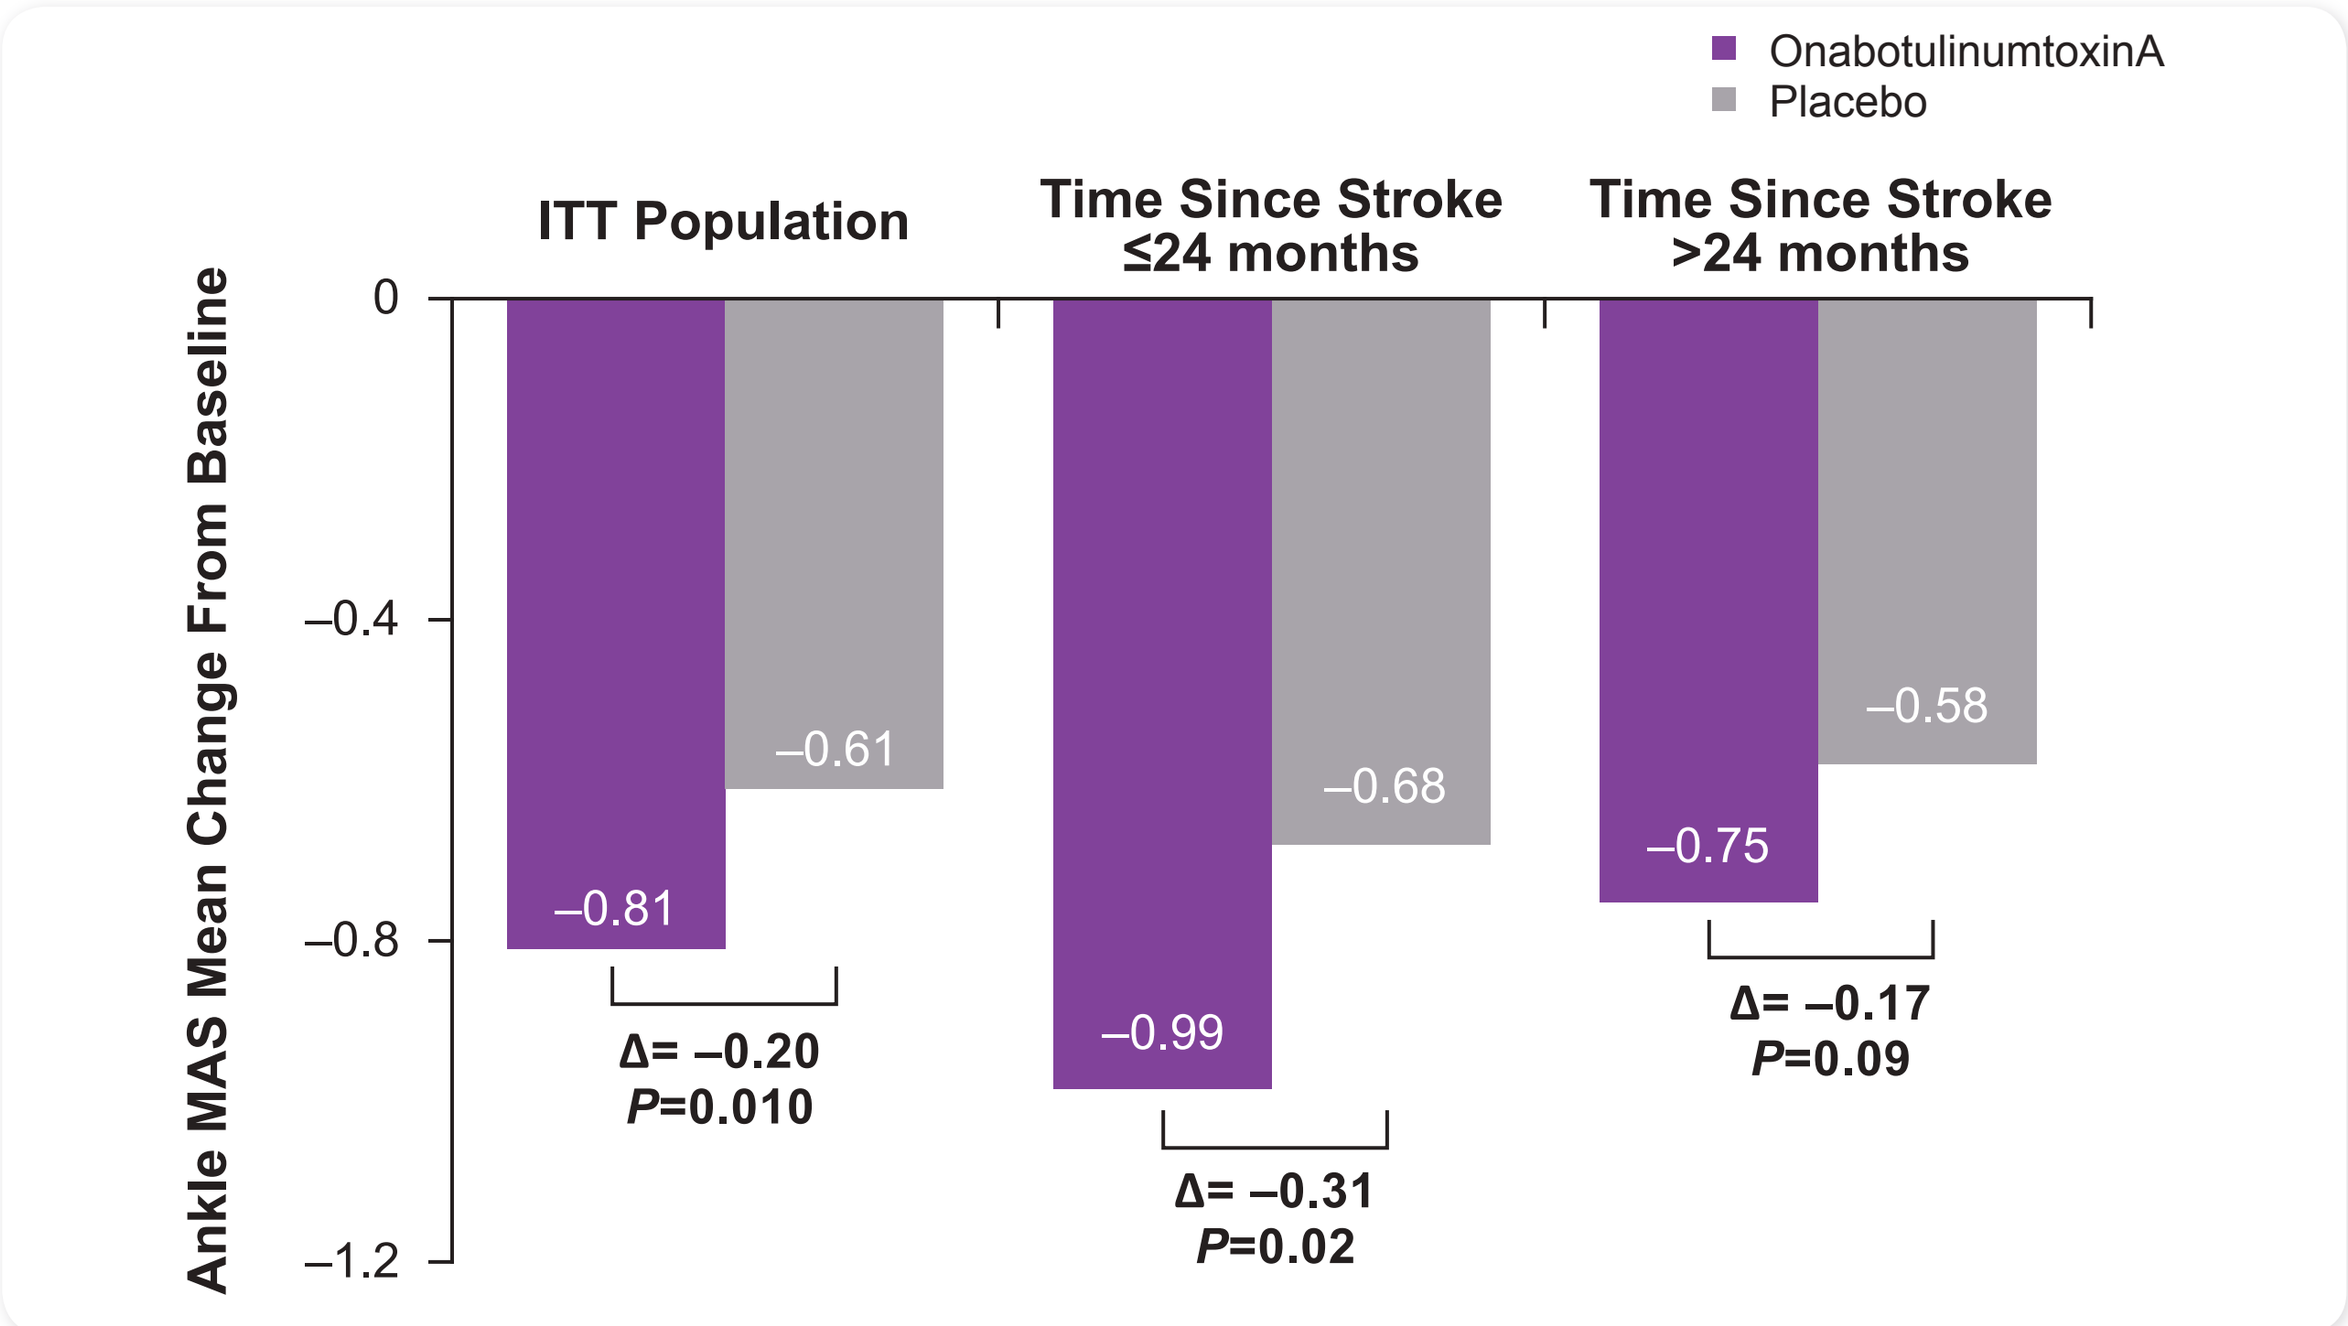

OnabotulinumtoxinA
Placebo
Ankle MAS Mean Change From Baseline
0
–0.4
–0.8
–1.2
ITT Population
Time Since Stroke ≤24 months
Time Since Stroke >24 months
–0.81
–0.61
Δ= –0.20
P=0.010
–0.99
–0.68
Δ= –0.31
P=0.02
–0.75
–0.58
Δ= –0.17
P=0.09

ANCOVA=analysis of covariance; ITT=intent-to-treat; MAS=Modified Ashworth Scale.
Data are least squares mean of weeks 4 and 6 change from baseline.
P-values for between-group comparisons were obtained from ANCOVA using imputation for missing values.

- Mean CGI by physician (average weeks 4 and 6) indicated greater clinical improvement in patients treated ≤24 months vs >24 months since stroke (mean change 0.49 [*P*<0.001 vs placebo] vs 0.12 [*P*=0.25 vs placebo], respectively, **[Figure 4]**), representing nearly a doubling in the mean difference, and thus greater improvement, in the earlier-treated subgroup

Figure 4. Physician-Assessed CGI Average Score of Weeks 4 and 6

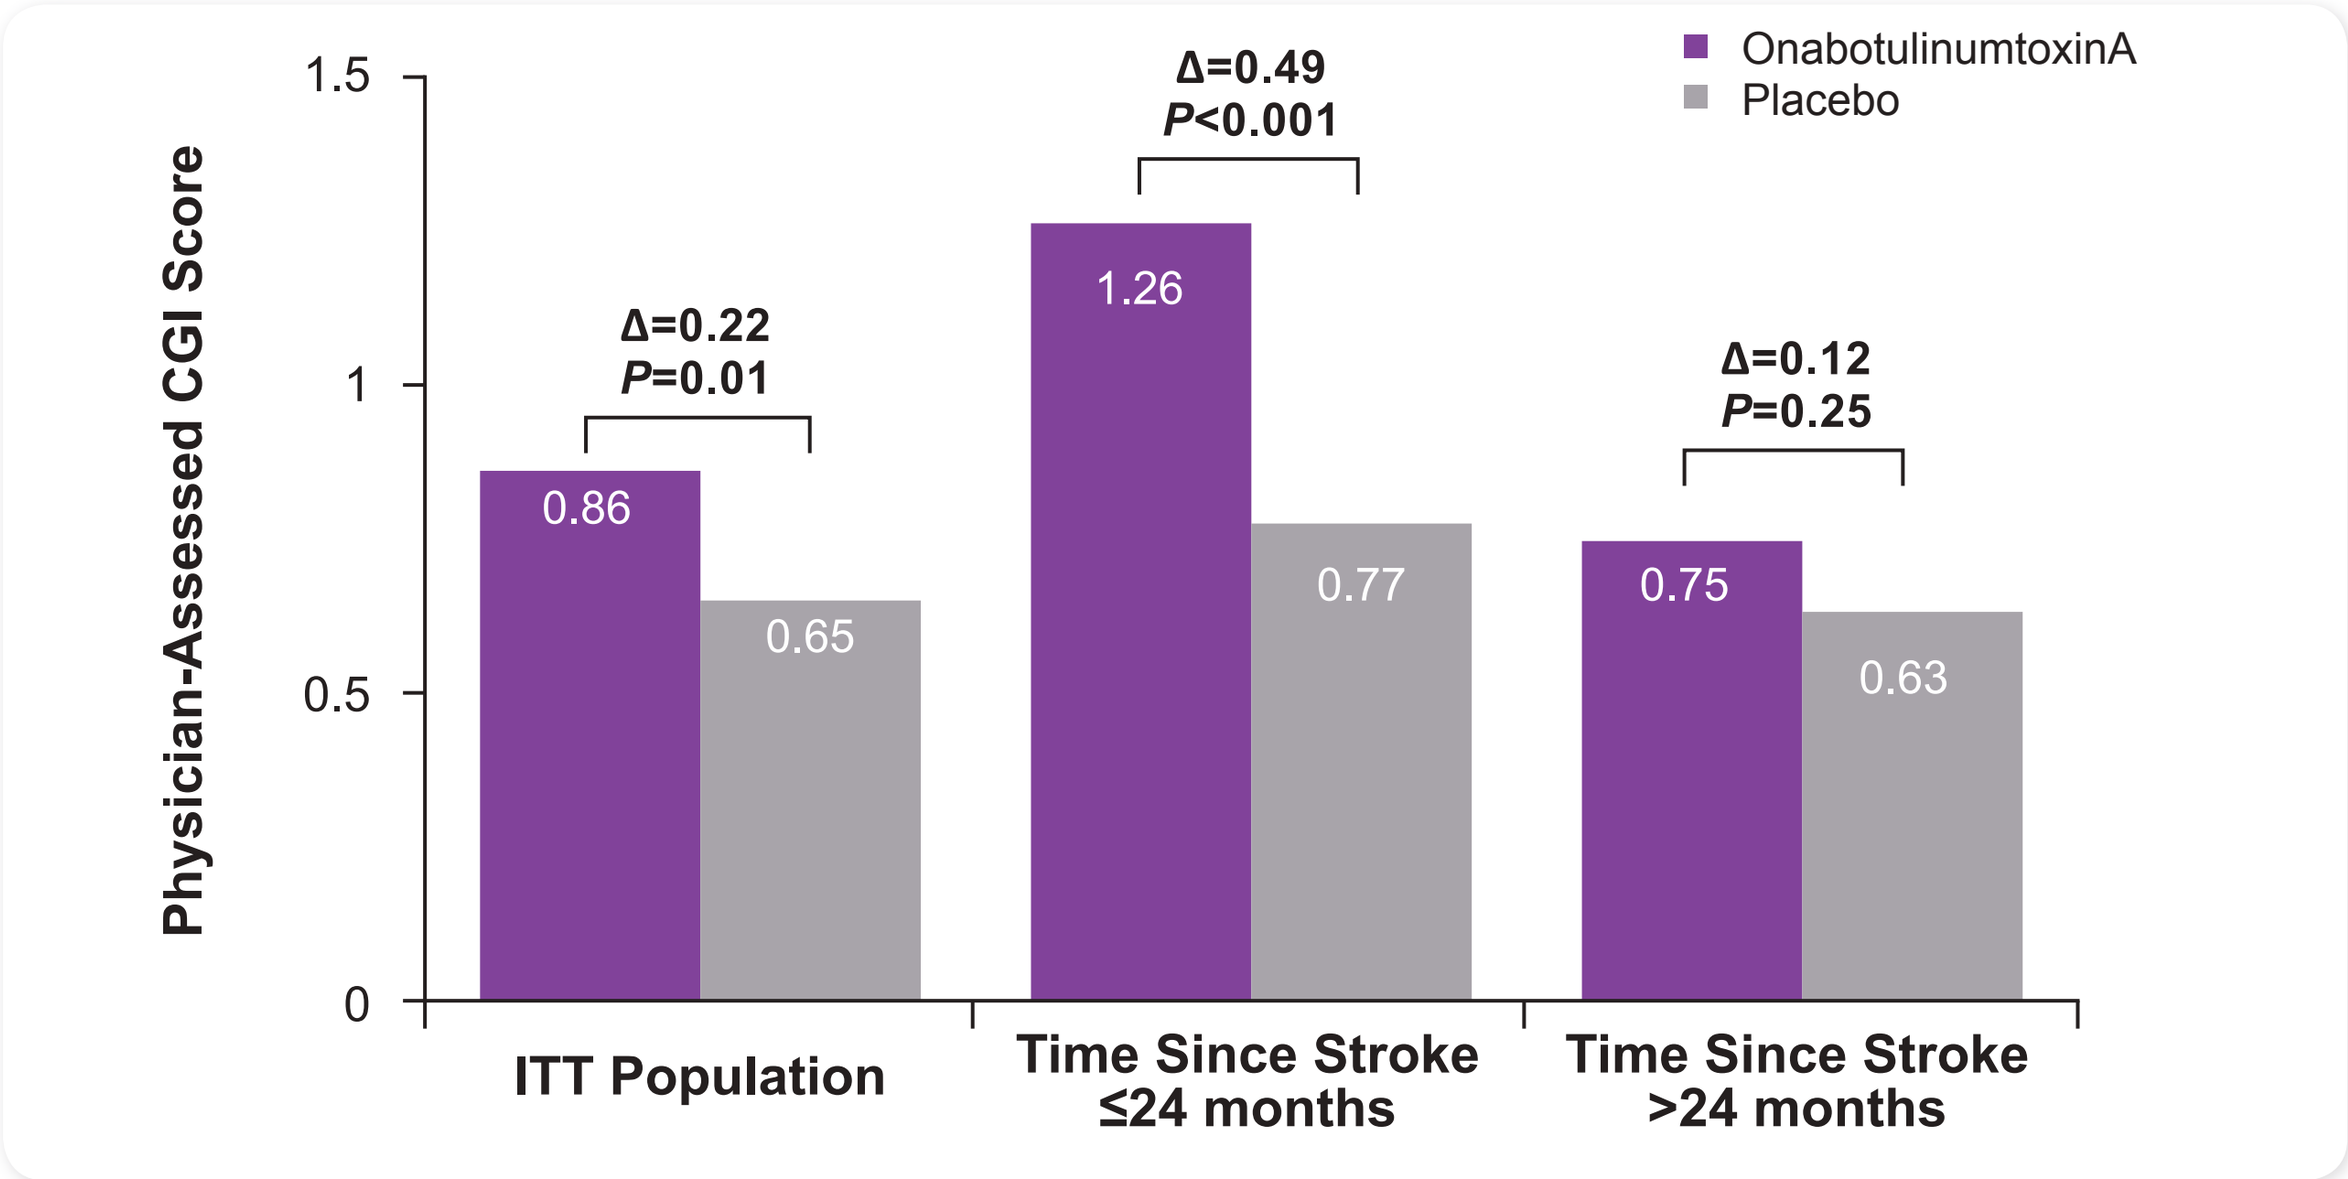

OnabotulinumtoxinA
Placebo
Physician-Assessed CGI Score
1.5
1
0.5
0
ITT Population
Time Since Stroke ≤24 months
Time Since Stroke >24 months
Δ=0.22
P=0.01
0.86
0.65
Δ=0.49
P<0.001
1.26
0.77
Δ=0.12
P=0.25
0.75
0.63

ANCOVA=analysis of covariance; ITT=intent-to-treat; CGI=Clinical Global Impression of Change.
Assessed and scored by the treating physician on a 9-point scale ranging from 'marked worsening'(–4) to 'very marked improvement' (+4).
Data are least squares mean of weeks 4 and 6 change from baseline.
P-values for between-group comparisons were obtained from ANCOVA using imputation for missing values.

- Goal Achievement (GAS≥0): onabotulinumtoxinA treatment ≤24 months since stroke resulted in numerically higher rate of passive goal achievement vs placebo
 - Week 8 (34.7%, onabotulinumtoxinA; 29.0%, placebo; *P*=not significant)
 - Week 12 (38.4%, onabotulinumtoxinA; 23.5%, placebo, *P*=0.058)
- Goal Improvement (GAS ≥ –1): A greater proportion of patients treated with onabotulinumtoxinA ≤24 months since stroke (vs placebo) achieved ≥ –1-point improvement for active goal at week 12 (69.2%, onabotulinumtoxinA; 52.8%, placebo; *P*=0.04) and for passive goal at week 8 (70.8%, onabotulinumtoxinA; 52.2%, placebo; *P*=0.02; **Figure 5**)

Figure 5. Proportion of Patients Achieving GAS Score ≥ –1

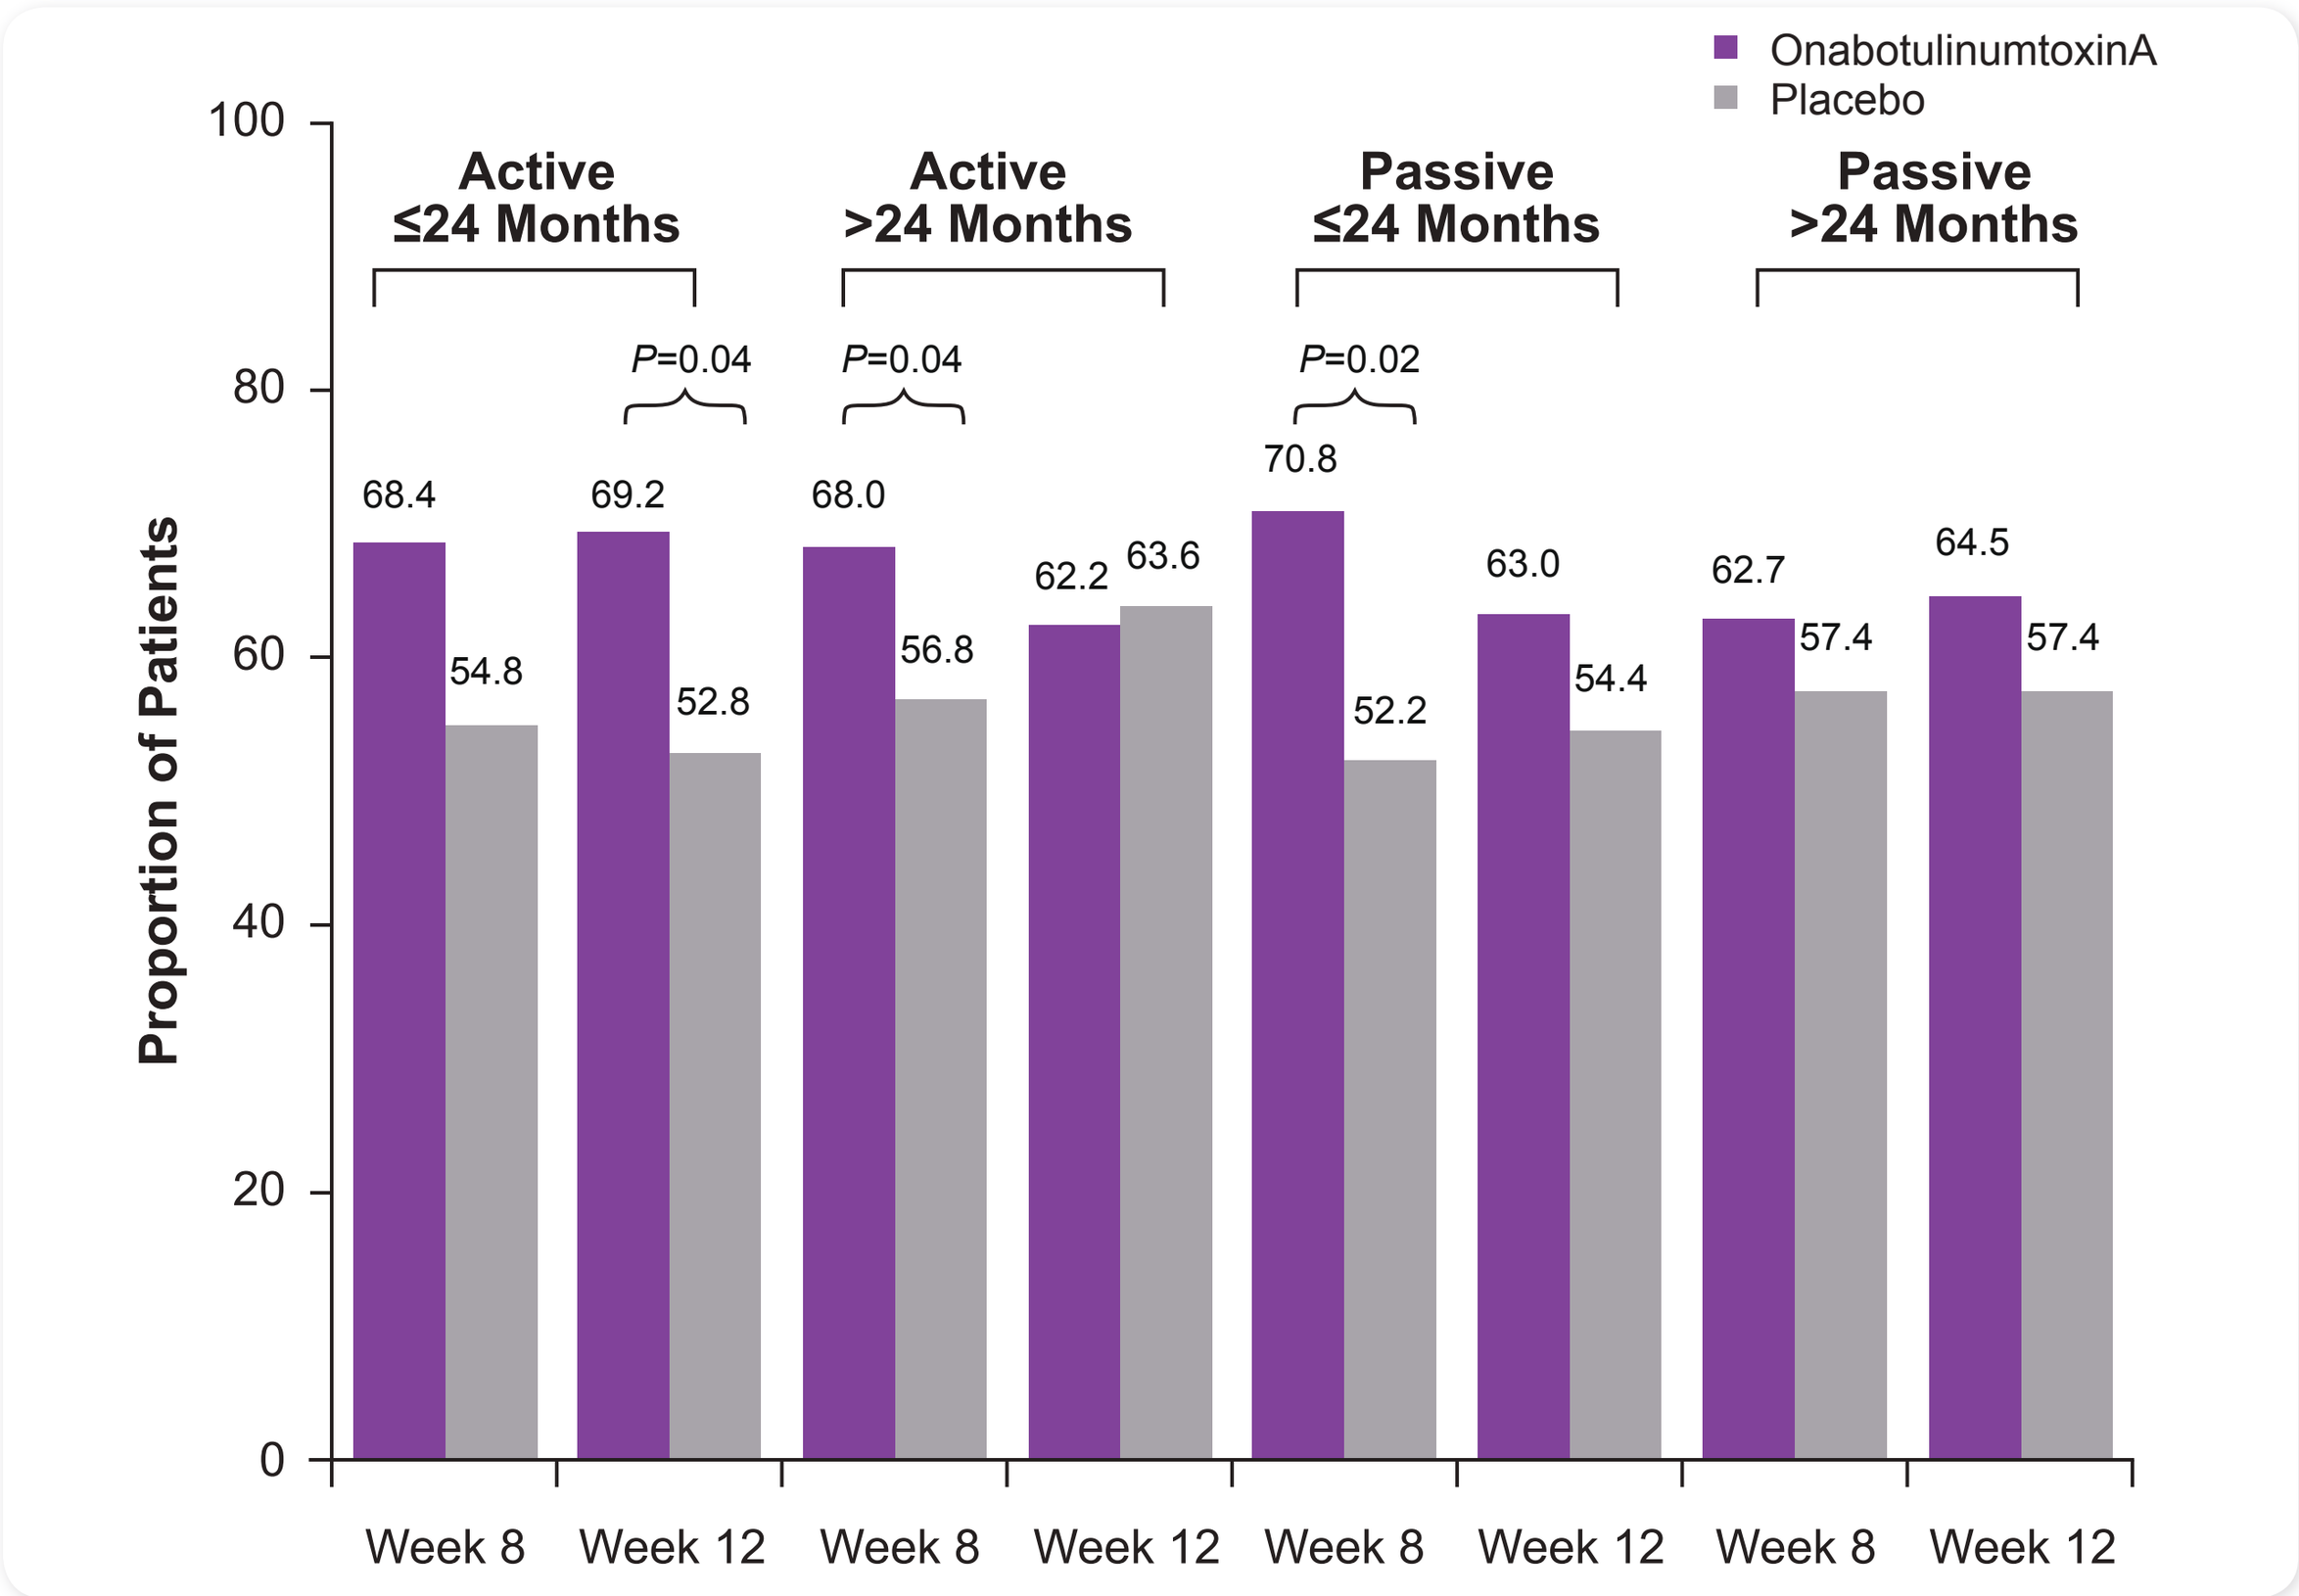

OnabotulinumtoxinA
Placebo
Proportion of Patients
100
80
60
40
20
0
Active ≤24 Months
Active >24 Months
Passive ≤24 Months
Passive >24 Months
P=0.04
P=0.04
P=0.02
68.4
54.8
69.2
52.8
68.0
56.8
62.2
63.6
70.8
52.2
63.0
54.4
62.7
57.4
64.5
57.4
Week 8
Week 12
Week 8
Week 12
Week 8
Week 12
Week 8
Week 12

P-values for between-group comparisons are determined by Pearson's chi-square or Fisher's exact test.

Safety

- Most AEs were mild or moderate and not considered related to study treatment
- No new safety signals were identified and no patients discontinued due to lack of efficacy **(Table 2)**
- Further analyses have shown no significant differences in the incidence of AEs by time since stroke

Table 2. Adverse Event Profile (Safety Population)

Adverse Event, n (%)	OnabotulinumtoxinA (N=231)	Placebo (N=233)
All TEAEs occurring in ≥2% of patients	95 (41.1)	80 (34.3)
Pain in extremity	11 (4.8)	10 (4.3)
Nasopharyngitis	8 (3.5)	6 (2.6)
Fall	6 (2.6)	8 (3.4)
Back pain	6 (2.6)	4 (1.7)
Arthralgia	6 (2.6)	2 (0.9)
Injection site pain	5 (2.2)	2 (0.9)
Treatment-related AEs	13 (5.6)	14 (6.0)
Serious AEs	10 (4.3)	9 (3.9)
Deaths	0	0
Treatment-related serious AEs	0	1 (0.4)
Discontinued because of AEs	5 (2.2)	2 (0.9)

2 patients from each group were not treated and, therefore, were not included in the safety population.
AE=adverse events, TEAE=treatment-emergent adverse event.

CONCLUSIONS

- Improvements in MAS, CGI, and GAS scores indicated that onabotulinumtoxinA 300 U–400 U is effective for the treatment of PSLLS
- Compared with patients who initiated onabotulinumtoxinA treatment >24 months since stroke, those who started treatment ≤24 months since stroke demonstrated greater benefits with further improvements in muscle tone and global functioning as measured by the physician global response scale and goal attainment
- No new safety signals were observed

REFERENCES

1. BOTOX® (onabotulinumtoxinA) for injection, for intramuscular use. Irvine, CA: Allergan, Inc.; 2016.
2. Wissel J, et al. *J Neurol.* 2010;257(7):1067-1072.

DISCLOSURES

Financial arrangements of the authors with companies whose products may be related to the present report are listed below, as declared by the authors.

Atul T. Patel, MD, MHSA, has received research grant support from Allergan plc, Merz and Ipsen and speaking honoraria from Merz and Allergan plc; **Anthony B. Ward, MD, FRCP,** is a speaker and consultant for Allergan plc and Ipsen; **Carolyn Geis, MD,** has received research support from and is a speaker/consultant for Allergan plc; **Wolfgang H. Jost, MD, PhD,** is a speaker and consultant for Allergan plc, Ipsen and Merz; **Chengcheng Liu, PhD,** and **Rozalina Dimitrova, MD, MPH,** are employees of Allergan plc.

ACKNOWLEDGEMENTS

This study was sponsored by Allergan plc, Dublin, Ireland. Writing and editorial assistance was provided to the authors by Scarlett Geunes-Boyer, PhD, at Complete Healthcare Communications, LLC (Chadds Ford, PA), a CHC Group company, and Dana Franznick, PharmD, and funded by Allergan plc, Dublin, Ireland. All authors met the International Conference of Medical Journal Editors authorship criteria. Neither honoraria nor payments were made for authorship.

To obtain a PDF of this poster:

- Scan the QR code
 OR
- Visit www.allergancongressposters.com/501341

Charges may apply. No personal information is stored.

Presented at TOXINS; January 18–21, 2017; Madrid, Spain