OnabotulinumtoxinA for Chronic Migraine in Korea: 1-Month Post-Marketing Surveillance and Phase 4 Trial Results

ByungKun Kim,[a] Manho Kim,[b] Chin-Sang Chung,[c] Ronald DeGryse,[d] Karen Lai,[d] Irina Yushmanova,[d] Arlene Lum,[d] Gudarz Davar[d]

[a]Eulji Hospital, Eulji University, Seoul, South Korea; [b]Seoul National University Hospital, Seoul, South Korea;
[c]Samsung Medical Center, Sungkyunkwan University School of Medicine, Seoul, South Korea; [d]Allergan plc, Irvine, CA, USA

Scan to obtain PDF of poster

INTRODUCTION

- Chronic migraine (CM) is a debilitating neurological disorder that affects approximately 1.4%–2.2% of adults worldwide[1]
 - The prevalence of CM in Korea has not been reported; however, the prevalence of chronic daily headache in Korea is approximately 1.8% (two-thirds diagnosed with CM)[2]
 - CM prevalence has been reported in other parts of Asia (i.e., Taiwan) as 1.0%–1.7%[1]
- CM is characterized by ≥15 headache days/month for >3 months, with ≥8 days/month of headache with migraine features[3]
- OnabotulinumtoxinA (BOTOX®, Allergan plc, Dublin, Ireland) demonstrates favorable efficacy and safety profiles[4] and is approved in Korea for headache prophylaxis in patients with CM

OBJECTIVE

- To assess the safety and effectiveness of onabotulinumtoxinA in Korean adults with CM

METHODS

Study Design and Patients

- This is a pooled analysis of post-marketing surveillance (PMS) and open-label phase 4 clinical trial data prospectively collected between March 2011 and March 2015
- Patients ≥15 years of age received 1, 2, or 3 treatments of onabotulinumtoxinA for CM, according to the treating physician's usual practices, at 28 Korean hospital sites (PMS) and 6 Korean study sites (phase 4 trials)

Assessments

- Short-term effectiveness was assessed using the validated Korean 6-item Headache Impact Test (HIT-6; score range, 36–78),[5,6] and defined as ≥1-point improvement in HIT-6 score at week 4
- Safety was assessed based on the rates and types of adverse events (AEs; causality not specified) and adverse drug reactions (ADRs; causality attributed to treatment)
- The possible influence of specific factors on safety (ie, sex, age, prior onabotulinumtoxinA treatment, onabotulinumtoxinA injection condition, past and current medical history, concomitant medication) and effectiveness (ie, sex, age, duration of illness, prior treatments, onabotulinumtoxinA dose, past and current medical history, concomitant medication, elderly status, renal disorders, hepatopathy) was also assessed

Statistical Analysis

Effectiveness

- HIT-6 score change pre- to post-injection was determined for the final effectiveness analysis: change ≥1, "improved"; change = 0, "unchanged"; change <0, "worsened"
 - Effectiveness was defined as "improved," and ineffectiveness as "unchanged" or "worsened"
 - Results were also analyzed by demographic and clinical parameters (ie, sex, age, prior onabotulinumtoxinA treatment, onabotulinumtoxinA dose, past and current medical history, allergy history, concomitant medication, elderly status, renal disorders, hepatopathy)

Safety

- AEs were defined as "expected" if they were reported in the Korean Ministry of Food and Drug Safety label (PMS) or Investigator's Brochure (phase 4 trials); all other AEs were "unexpected"
- AE severity was classified as "mild," "moderate," and "severe"
- AE progression was classified as "resolved," "unresolved," "unknown," and "death"
- Causality between AE and onabotulinumtoxinA was classified as "certain," "probable/likely," "possible," "unlikely," "conditional/unclassified," and "not assessable/classifiable"
- OnabotulinumtoxinA administration after AE incidence was classified as "no change," "permanently discontinued," "temporarily stopped," "dosage reduced," "dosage increased," "regimen changed," and "not applicable"
- Administration of corrective treatment (ie, treatment provided or required for AEs) after AE incidence was classified as "yes" and "no"

RESULTS

Patient Demographics and Treatment Characteristics

- Data were collected from 683 patients (PMS, n=401; phase 4, n=282)
 - Median (range) age was 48 y (15–83 y) and 85.5% were women
 - Most patients received 1 injection of onabotulinumtoxinA, with a median dose of 155 U **(Table 1)**

Table 1. Patient Demographics and Treatment Characteristics

	Patients (N=683)
Age, y, median (min, max)	48 (15, 83)
Women, n (%)	584 (85.5)
BMI*, kg/m², median (min, max)	23 (15, 42)
Examination type†, n (%)	
Inpatient	203 (29.7)
Outpatient	480 (70.3)
Elderly (≥65 y), n (%)	75 (11.0)
Currently pregnant‡, n (%)	0 (0)
OnabotulinumtoxinA injections§, n (%)	
1	663 (97.6)
2	12 (1.8)
3	4 (0.6)
OnabotulinumtoxinA dose§, U, median (min, max)	155 (155, 465)

BMI=body mass index.
*Data available for 529 patients.
†All phase 4 examinations were considered to be outpatient.
‡Data available for 469 women.
§Data available for 679 patients.

Effectiveness

- Effectiveness data were available for 651 patients
- As reflected by a ≥1-point improvement in HIT-6 total score at week 4, the overall effectiveness was 81.7%
- HIT-6 score was unchanged in 112 (17.2%) and worsened in 7 (1.1%) patients
- Statistically significant differences were found in effectiveness for several prespecified subgroups **(Figure 1)**. Although some of these potential effects could be predicted (eg, concomitant medications or illness), interpretation of their clinical meaningfulness was confounded, in part, by within-group differences (eg, imbalances in sample size) and limited efficacy analyses

Figure 1. Effectiveness Proportions by Background Factors

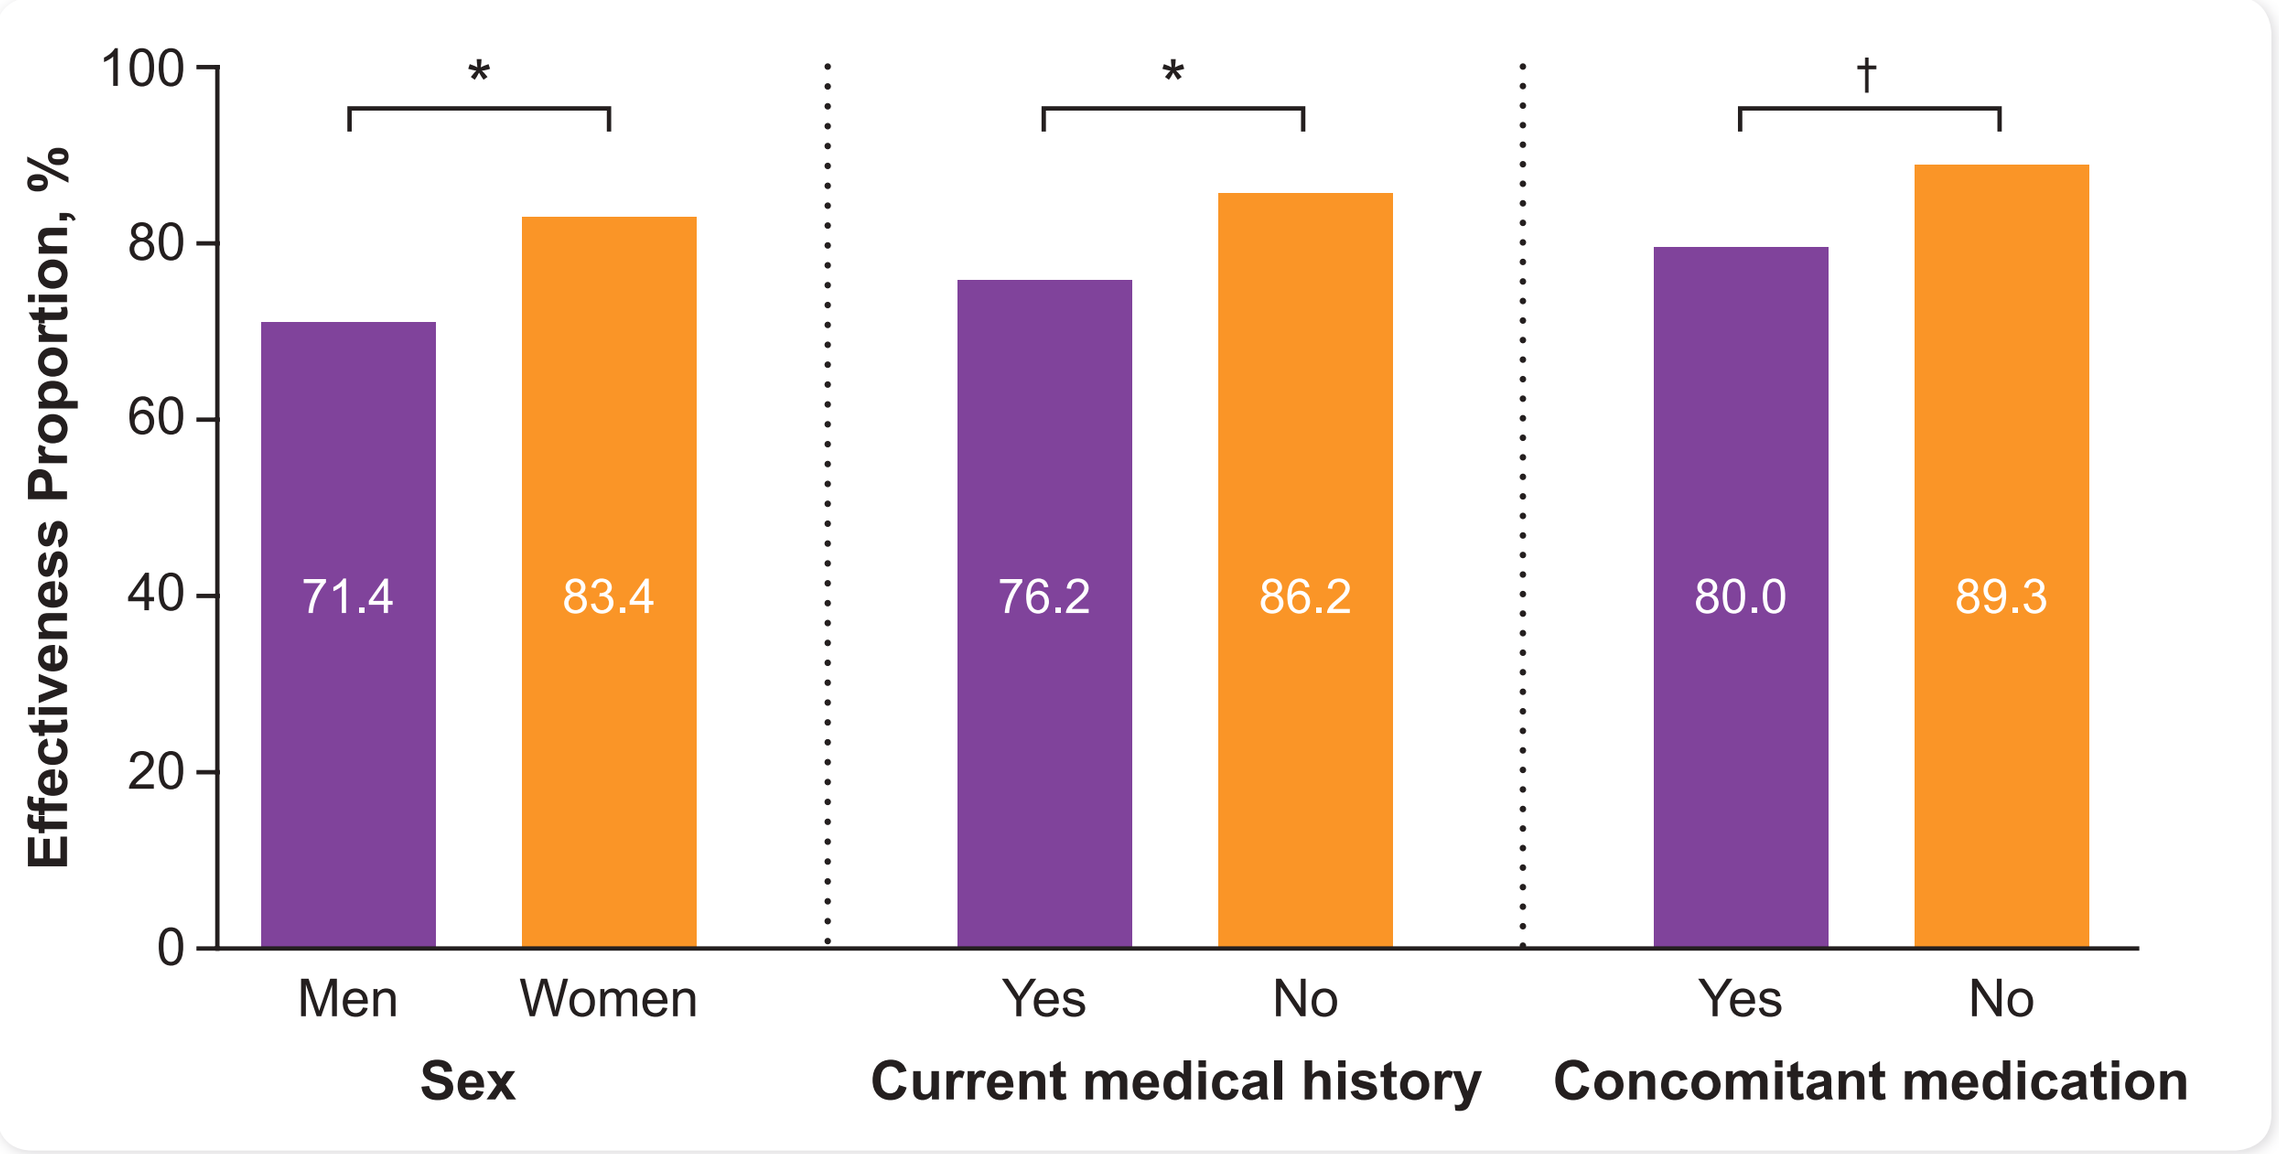

Effectiveness Proportion, %
100
80
60
40
20
0
*
*
†
71.4
83.4
76.2
86.2
80.0
89.3
Men
Women
Yes
No
Yes
No
Sex
Current medical history
Concomitant medication

*P<0.01; †P<0.05.

Safety

- Safety data were available for 672 patients
- 92 AEs and ADRs were reported by 80 (11.9%) patients **(Table 2)**
- Most AEs were expected (90.2% [83/92]) and all were mild (91.3% [84/92]) or moderate (8.7% [8/92]) in severity. Unexpected or serious AEs and ADRs are presented in **Table 3**
- 64.1% (59/92) of AEs were unresolved, 34.8% (32/92) were resolved, and resolution was unknown for 1.1% (1/92) of AEs
- Causality by onabotulinumtoxinA was certain for 37.0% (34/92), probable/likely in 30.4% (28/92), possible in 10.9% (10/92), and unlikely in 20.7% (19/92) of AEs
- OnabotulinumtoxinA administration was unchanged after 98.9% (91/92) of AEs and was permanently discontinued after 1.1% (1/92) of AEs
- Corrective treatment was administered after 18.5% (17/92) of AEs
- No specific factors were identified to have affected the incidence proportion of AEs, but analysis by background factors indicated statistically significant differences in age, current medical history, concomitant medication, and hepatopathy (all P<0.01)
- No new safety concerns were identified

Table 2. Summary of all AEs and ADRs*

System organ classification	Patients with AE, n (%)	Patients with ADR, n (%)
Musculoskeletal system disorders	32 (4.8)	31 (4.6)
Central/peripheral NS disorders	22 (3.3)	20 (3.0)
Resistance mechanism disorders	8 (1.2)	7 (1.0)
Gastrointestinal system disorders	6 (0.9)	1 (0.2)
Application site disorders	4 (0.6)	4 (0.6)
General disorders	5 (0.8)	3 (0.5)
Skin and appendages	4 (0.6)	3 (0.5)
Psychiatric disorders	1 (0.2)	0 (0)
Respiratory system disorders	1 (0.2)	0 (0)
Vision disorders	1 (0.2)	1 (0.2)
Other	1 (0.2)	0 (0)

AE=adverse event; ADR=adverse drug reaction.
*AEs do not assume drug causality; ADRs were considered to be conditionally or certainly associated with drug administration.

Table 3. Unexpected or Serious AEs or ADRs*

AE/ADR	Patients with AE, n (%)	Patients with ADR, n (%)
Unexpected		
Upper abdominal pain†	1 (0.2)	0 (0)
Bloating‡	1 (0.2)	0 (0)
Colitis†	1 (0.2)	1 (0.2)
Irritable bowel syndrome†	1 (0.2)	0 (0)
Oral pain‡	1 (0.2)	0 (0)
Facial edema	1 (0.2)	1 (0.2)
Insomnia†	1 (0.2)	0 (0)
Hemoptysis	1 (0.2)	0 (0)
Other	1 (0.2)	0 (0)
Serious		
Migraine‡	1 (0.2)	0 (0)
Common cold	1 (0.2)	0 (0)
Diarrhea	1 (0.2)	0 (0)
Fever	1 (0.2)	0 (0)
Hemoptysis	1 (0.2)	0 (0)

AE=adverse event; ADR=adverse drug reaction.
*AEs do not assume drug causality; ADRs were considered to be conditionally or certainly associated with drug administration
All AEs and ADRs were of mild severity unless otherwise annotated.
†Unresolved.
‡Moderate severity.

CONCLUSIONS

- Our pooled PMS and phase 4 study results show that onabotulinumtoxinA is an effective and safe treatment among Korean adult patients
- No new safety concerns were identified
- Continued worldwide PMS will assess ongoing patient safety

REFERENCES

1. Natoli JL, et al. *Cephalalgia*. 2010;30(5):599-609.
2. Park JW, et al. *J Clin Neurol*. 2014;10(3):236-243.
3. Headache Classification Committee of the International Headache Society (IHS). *Cephalalgia*. 2013;33(9):629-808.
4. Aurora SK, et al. *Headache*. 2011;51(9):1358-1373.
5. Chu MK. *J Korean Neurol Assoc*. 2009;27(1):1-6.
6. Shin HE, et al. *J Clin Neurol*. 2008;4(4):158-163.

DISCLOSURES

Financial arrangements of the authors with companies whose products may be related to the present report are listed below, as declared by the authors.

ByungKun Kim, **MD**, **PhD**, has recieved honoraria from Allergan, Pfizer, Sandoz, Abbott, and Yuyu company for lecture fees. **Manho Kim**, **MD**, **PhD**, and **Chin-Sang Chung**, **MD**, **PhD**, have no conflicts of interest to report. **Ronald DeGryse**, **MS**, **MA**, **Karen Lai**, **PharmD**, **Irina Yushmanova**, **MD**, **Arlene Lum**, **PMP**, and **Gudarz Davar**, **MD**, are employees of Allergan plc and own stock in the company.

ACKNOWLEDGMENTS

This study was sponsored by Allergan plc (Dublin, Ireland). Writing and editorial assistance was provided to the authors by Scarlett Geunes-Boyer, PhD, and Amanda Kelly, MPhil, MSHN, of Complete Healthcare Communications, LLC (Chadds Ford, PA, USA), a CHC Group company, and funded by Allergan plc (Dublin, Ireland). All authors met the ICMJE authorship criteria. Neither honoraria nor payments were made for authorship.

To obtain a PDF of this poster:
- Scan the QR code

 OR
- Visit www.allergancongressposters.com/255705

Charges may apply. No personal information is stored.

Allergan

Presented at TOXINS; January 18–21, 2017; Madrid, Spain