Efficacy and Safety of OnabotulinumtoxinA in an Open-Label Study for the Prophylactic Treatment of Chronic Migraine in Adult Patients: COMPEL

Andrew M. Blumenfeld,[a] Richard Stark,[b] Aubrey Manack Adams,[c] Amelia Orejudos,[d] Sheena K. Aurora[e]

[a]The Neurology Center, Carlsbad, CA, USA; [b]Monash University and Alfred Hospital, Melbourne, Victoria, Australia; [c]Allergan plc, Irvine, CA, USA; [d]Allergan plc, Bridgewater, NJ, USA; [e]Stanford University Medical Center, Stanford, CA, USA

Scan to obtain PDF of poster

INTRODUCTION

- Chronic migraine (CM) affects approximately 2% of adults worldwide and has a substantial quality of life and economic burden[1-5]
- OnabotulinumtoxinA is approved for CM headache prophylaxis and has a favorable safety/efficacy profile; however, data are limited to 56 weeks
- COMPEL (NCT01516892) evaluates the longer-term (108 week) efficacy and safety of onabotulinumtoxinA for CM headache prophylaxis

OBJECTIVES

- To evaluate the longer-term efficacy and safety of onabotulinumtoxinA for the treatment of chronic migraine (CM)

METHODS

- Multi-center, open-label study in adult CM patients
- Treated with 155 U onabotulinumtoxinA every 12 weeks using a fixed-site, fixed-dose injection paradigm
- Primary efficacy measure:
 - Change in frequency of headache days from baseline at 108 weeks (following 9 treatments)
- Secondary/exploratory measures included:
 - 6-Item headache impact test (HIT-6)
 - 30% responder rate
 - Migraine Disability Assessment Questionnaire (MIDAS)
 - Migraine Specific Quality of Life Questionnaire (MSQ)
- Data are summarized using descriptive statistics
 - Missing data were imputed using modified last observation carried forward (mLOCF) method
- Efficacy cohort: patients who received ≥1 onabotulinumtoxinA treatment and ≥1 efficacy assessment
- Safety cohort: patients who received ≥1 onabotulinumtoxinA treatment

RESULTS

Patient Demographics and Clinical Characteristics

- A total of 716 patients were enrolled
 - 52.1% patients (n=373) completed the study
 - 47.9% (n=343) patients discontinued the study
- The most common reasons for study discontinuation were withdrawal of consent (n=92, 12.8%) and loss to follow-up (n=82, 11.5%)
- The analysis population included 715 patients (1 enrolled patient missed Visit 3 and was discontinued from the study)
- Patients were primarily female and white (**Table 1**)

Table 1. Baseline Patient Demographics and Clinical Characteristics

	Overall (N=715)
Age	
n	715
Mean (SD)	43.0 (11.3)
Min, Max	18, 73
Gender, n (%)	
Female	606 (84.8)
Male	109 (15.2)
Ethnicity, n (%)	
Hispanic/Latino	42 (5.9)
Not Hispanic/Latino	673 (94.1)
Race, n (%)	
White	581 (81.3)
Black/African American	41 (5.7)
Asian	89 (12.4)
Native Hawaiian/Other Pacific Islander	3 (0.4)
American Indian/Alaska Native	1 (0.1)

Headache Days

- Patients reported an average 22.0 (SD=22.0) headache days per month at baseline
- 713 patients (99.6%) reported headaches with moderate or severe pain at baseline
 - Pain was primarily characterized as throbbing or pulsing (n=507; 70.8%)
- By 108 weeks, a significant reduction in the number of headache days per month (-10.7 days, *P*<.0001; n=715/715) was observed (**Figure 1**)
 - 237 of 316 patients (75.0%) experienced a ≥30% decrease in headache days from baseline

Figure 1. Mean Change from Baseline in Headache Days

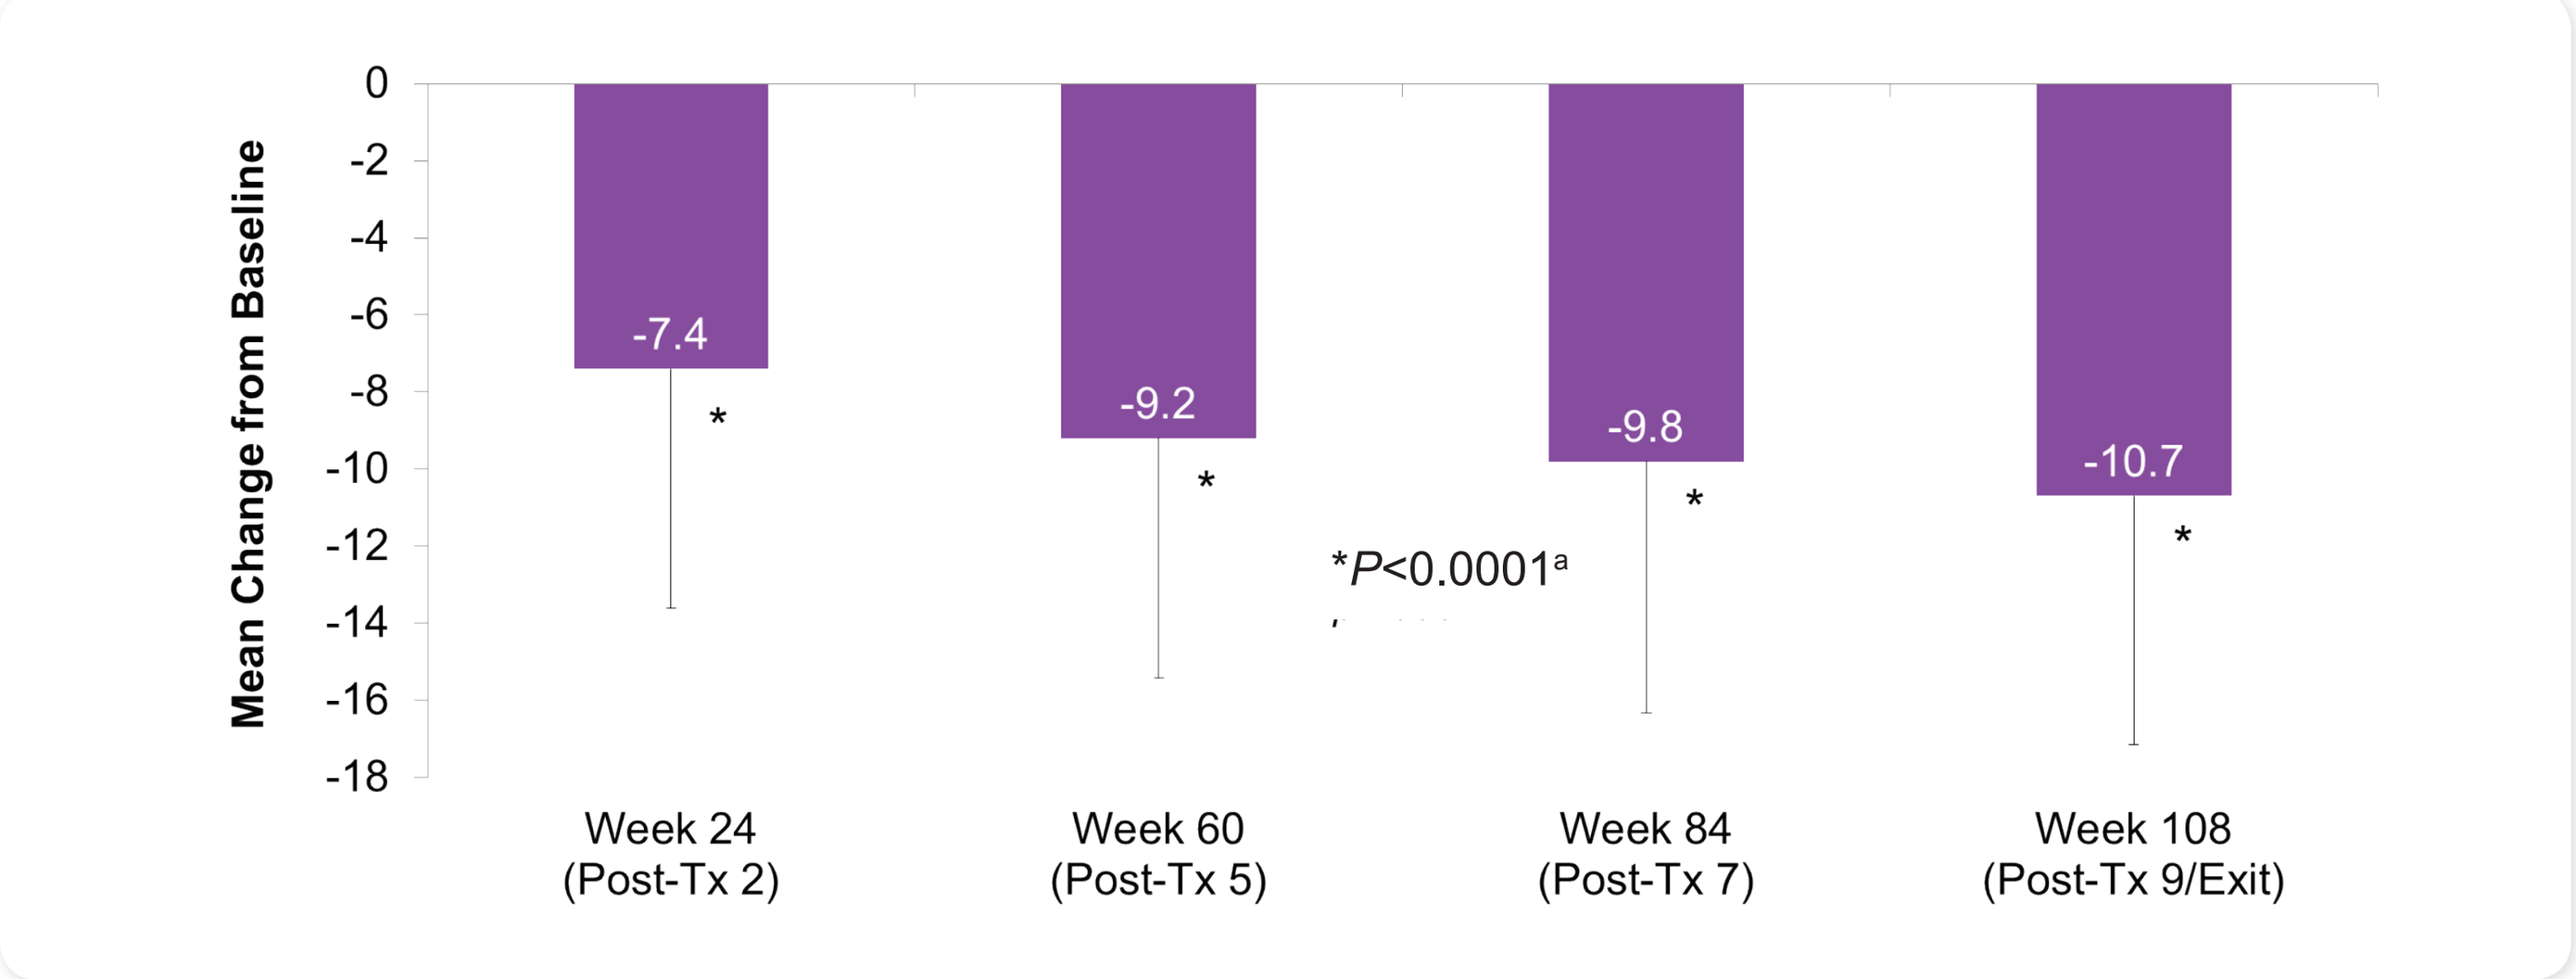

Mean Change from Baseline
0
-2
-4
-6
-8
-10
-12
-14
-16
-18
-7.4
-9.2
-9.8
-10.7
*P<0.0001a
Week 24 (Post-Tx 2)
Week 60 (Post-Tx 5)
Week 84 (Post-Tx 7)
Week 108 (Post-Tx 9/Exit)

NOTE: Missing data were imputed using modified last observation carried forward (mLOCF) method
[a] Two-sided P-value comparing post-baseline visit to baseline from the paired *t*-test; significance level is 0.025

HIT-6 Scores

- Average HIT-6 score at baseline was 64.7 (SD=4.8; n=713/715)
- By 108 weeks, a significant improvement in HIT-6 scores (–7.1 point change, *P*<0.0001; n=713/715) was observed (**Figure 2**)

Figure 2. Mean Change from Baseline in HIT-6 Scores

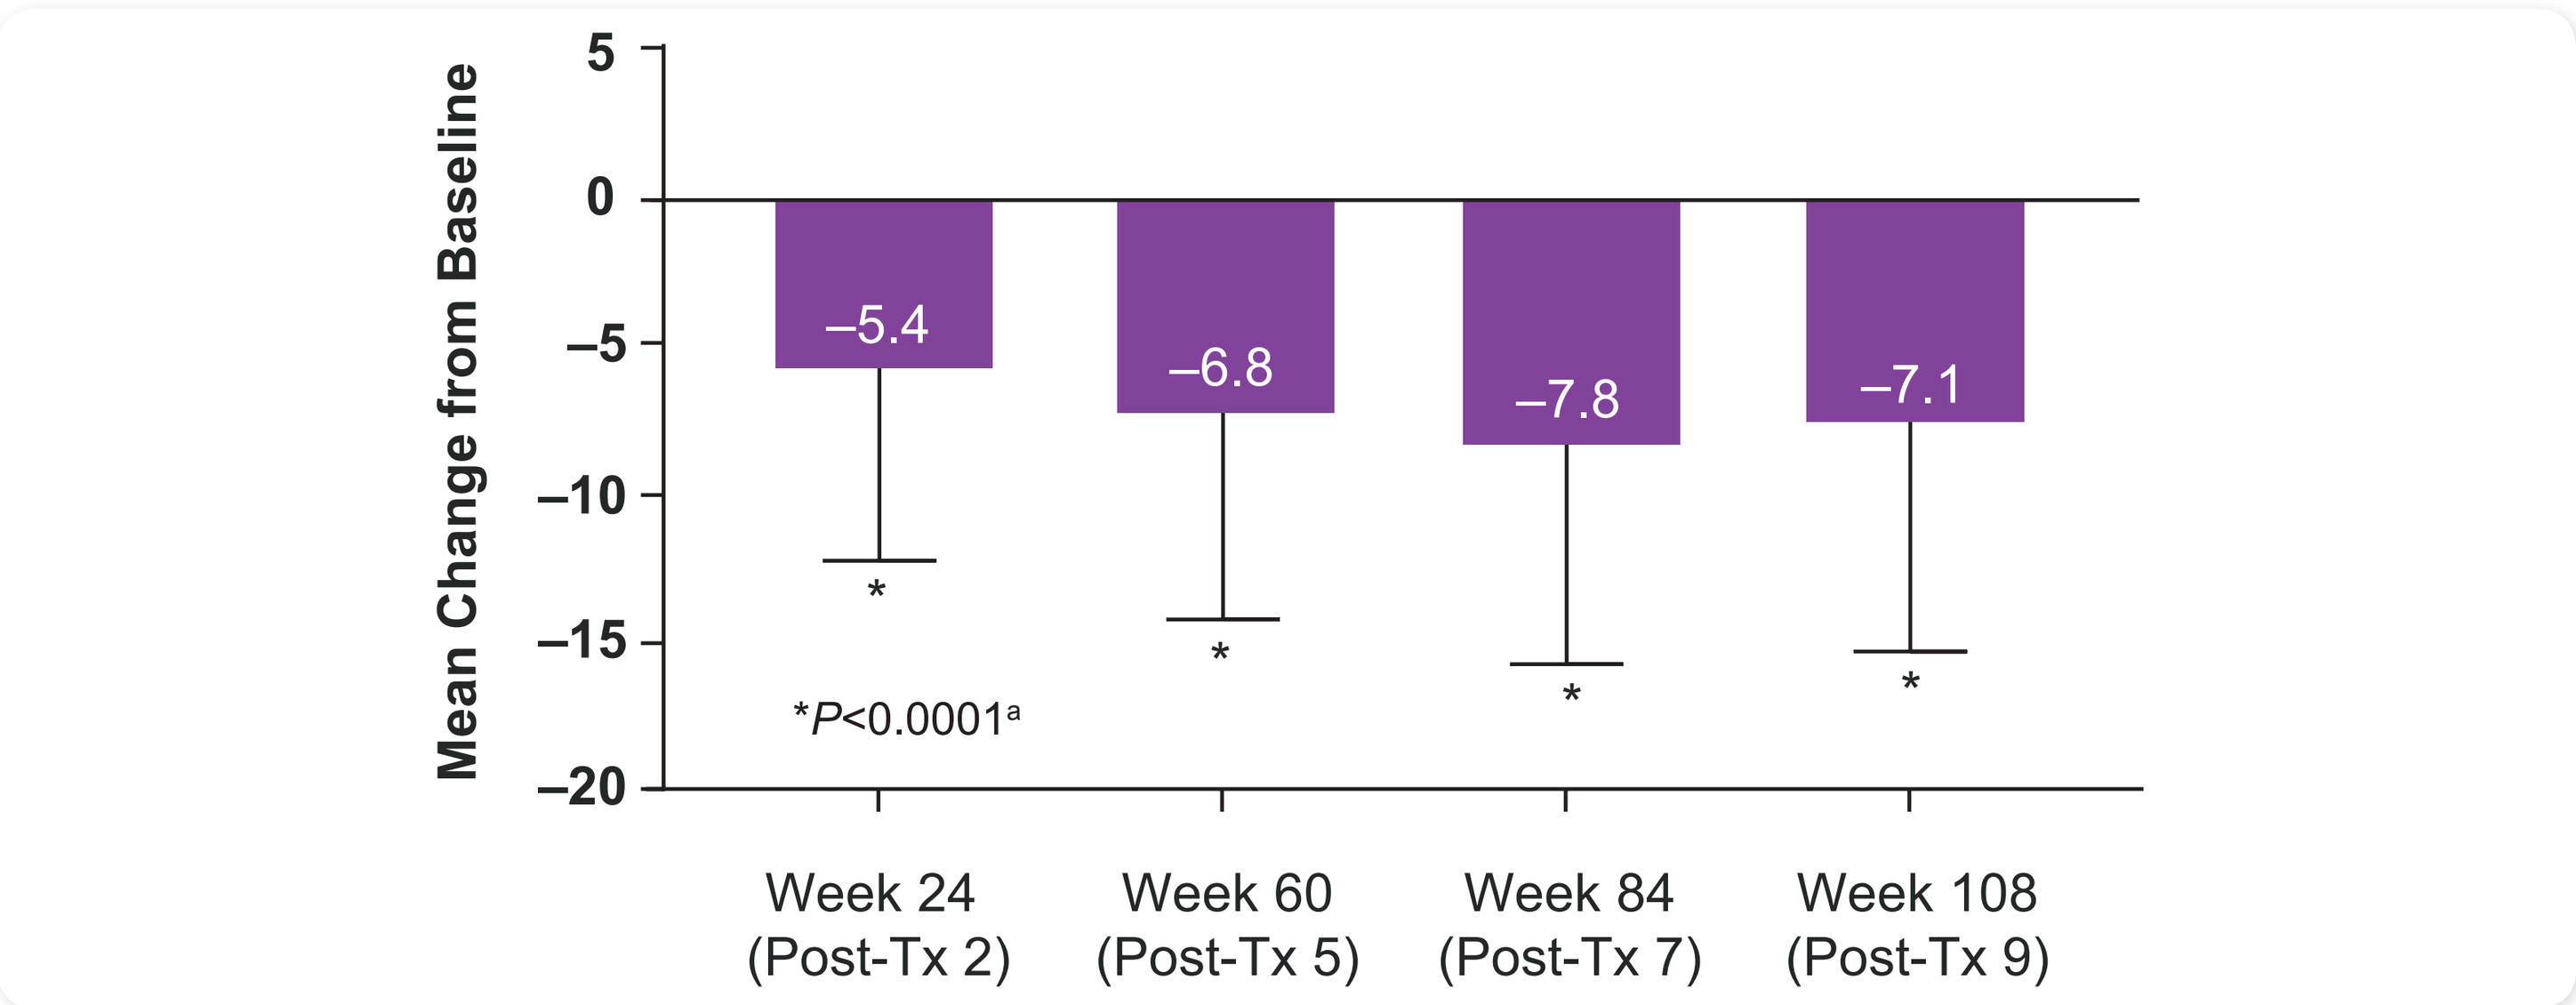

Mean Change from Baseline
5
0
–5
–10
–15
–20
–5.4
–6.8
–7.8
–7.1
*P<0.0001a
Week 24 (Post-Tx 2)
Week 60 (Post-Tx 5)
Week 84 (Post-Tx 7)
Week 108 (Post-Tx 9)

HIT-6=6-item headache impact test
NOTE: Missing data were imputed using modified last observation carried forward (mLOCF) method
[a] Two-sided *P*-value comparing post-baseline visit to baseline from the paired *t*-test; significance level is 0.025

MIDAS

- Average MIDAS score at baseline was 78.0 (SD=57.7; n=711/715)
- Patients demonstrated significant improvements from baseline at 108 weeks in MIDAS (-34.8 point change/ n=486/715) (**Figure 3**)

Figure 3. Mean Change from Baseline in MIDAS

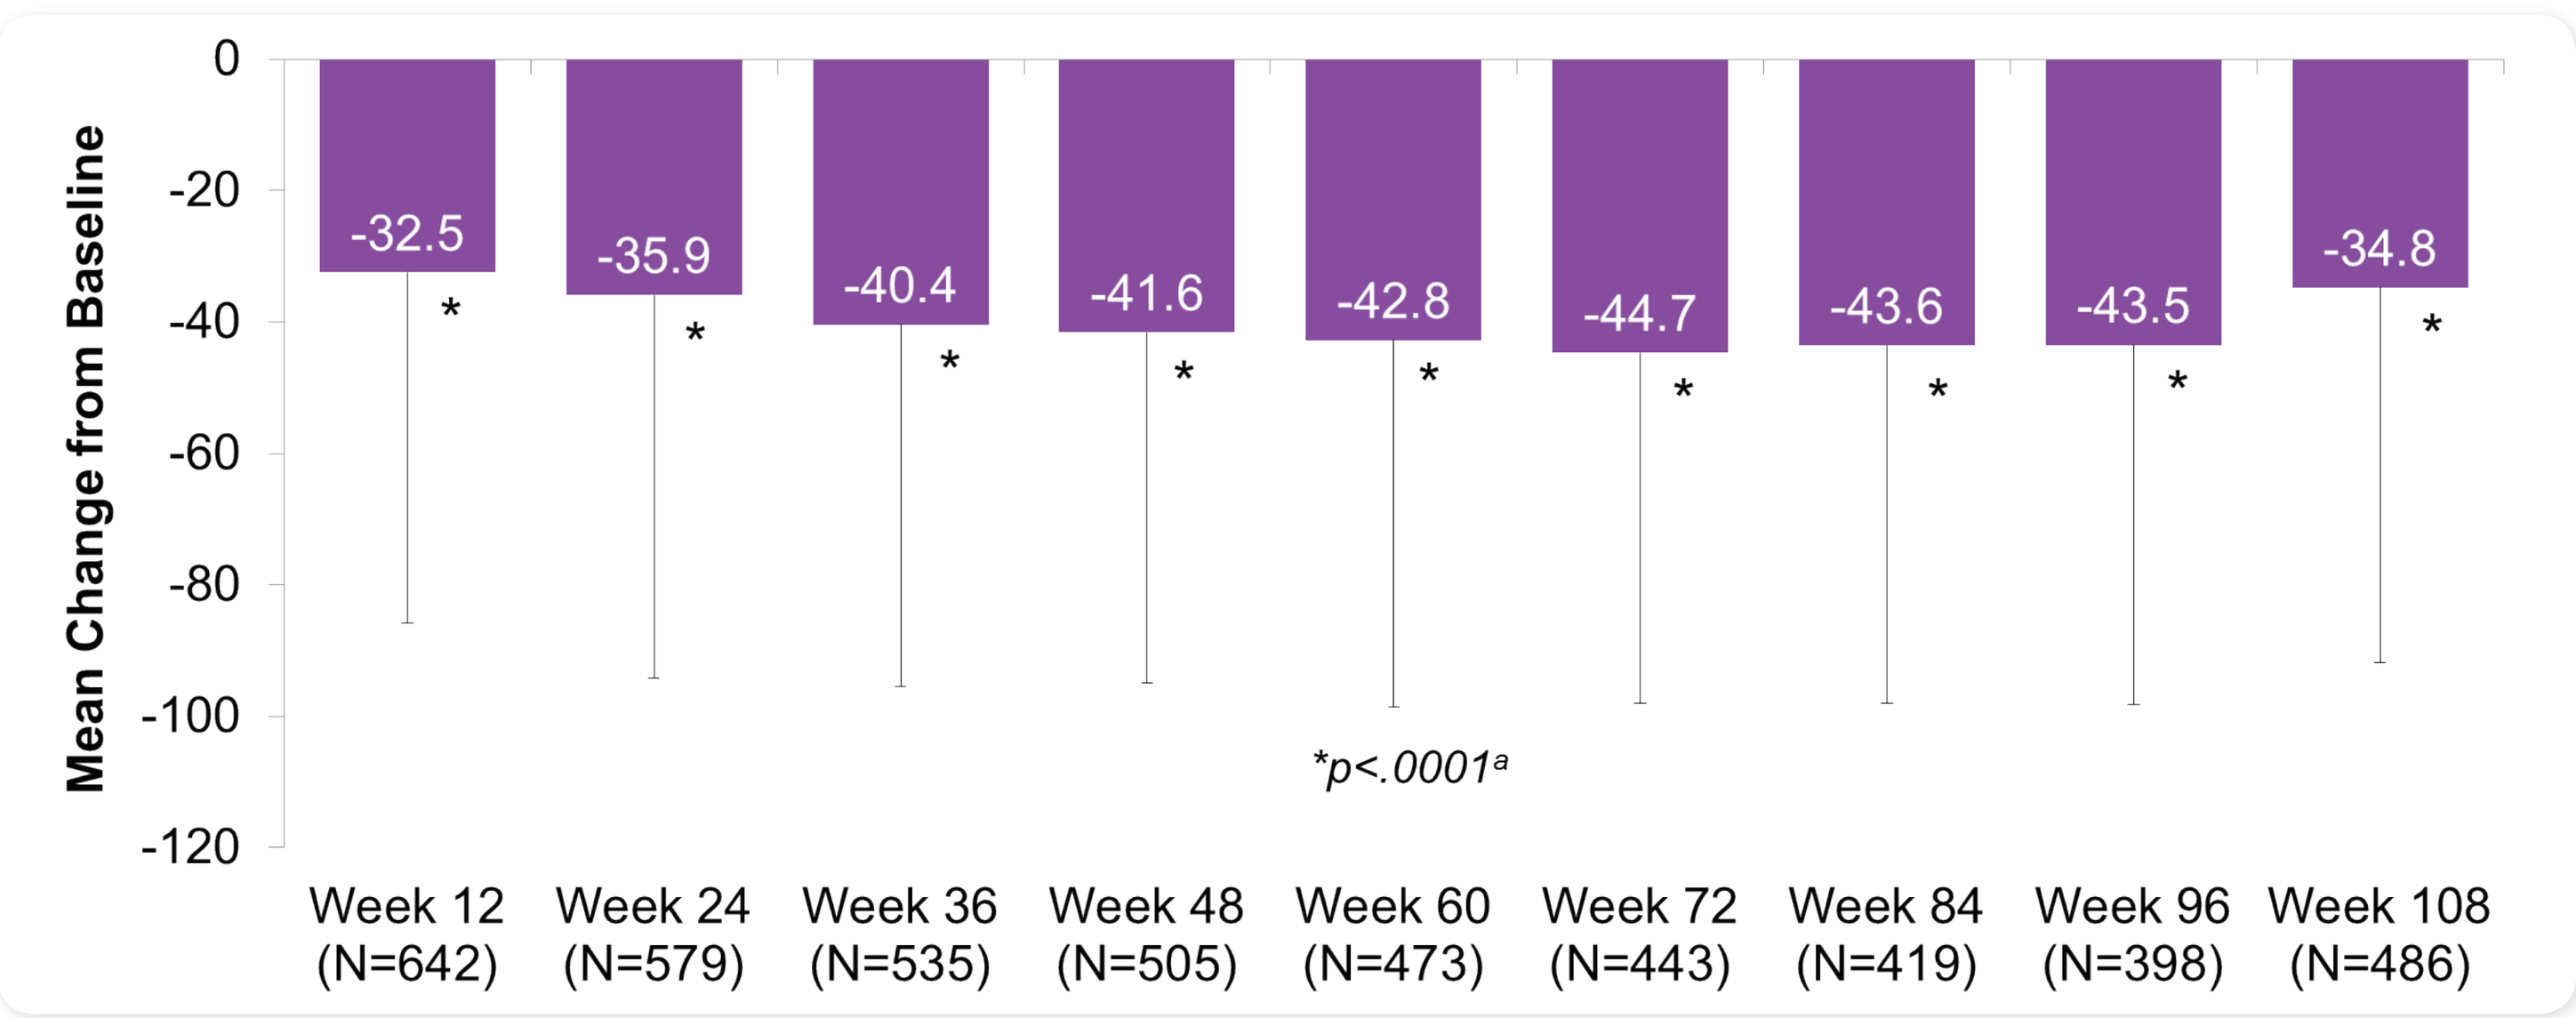

Mean Change from Baseline
0
-20
-40
-60
-80
-100
-120
-32.5
-35.9
-40.4
-41.6
-42.8
-44.7
-43.6
-43.5
-34.8
*p<.0001a
Week 12 (N=642)
Week 24 (N=579)
Week 36 (N=535)
Week 48 (N=505)
Week 60 (N=473)
Week 72 (N=443)
Week 84 (N=419)
Week 96 (N=398)
Week 108 (N=486)

NOTE: Missing post-baseline values were imputed using modified last observation carried forward (mLOCF) method; if 50% or more of the questions are answered, the total score is extrapolated from the mean score across answered questions, otherwise the score is set to missing
[a] Two-sided *P*-value comparing post-baseline visit to baseline from the paired *t*-test; significance level is 0.025

MSQ

- Average MSQ score at baseline (n=713/715)
 - Role Function Preventative = 60.2 (SD=22.4)
 - Role Function Restrictive = 42.6 (SD=19.6)
 - Emotional Function = 50.3 (SD=26.1)
- Significant improvements from baseline in MSQ domain scores were seen up to 108 weeks (**Figure 4**)

Figure 4. Mean Change from Baseline in MSQ

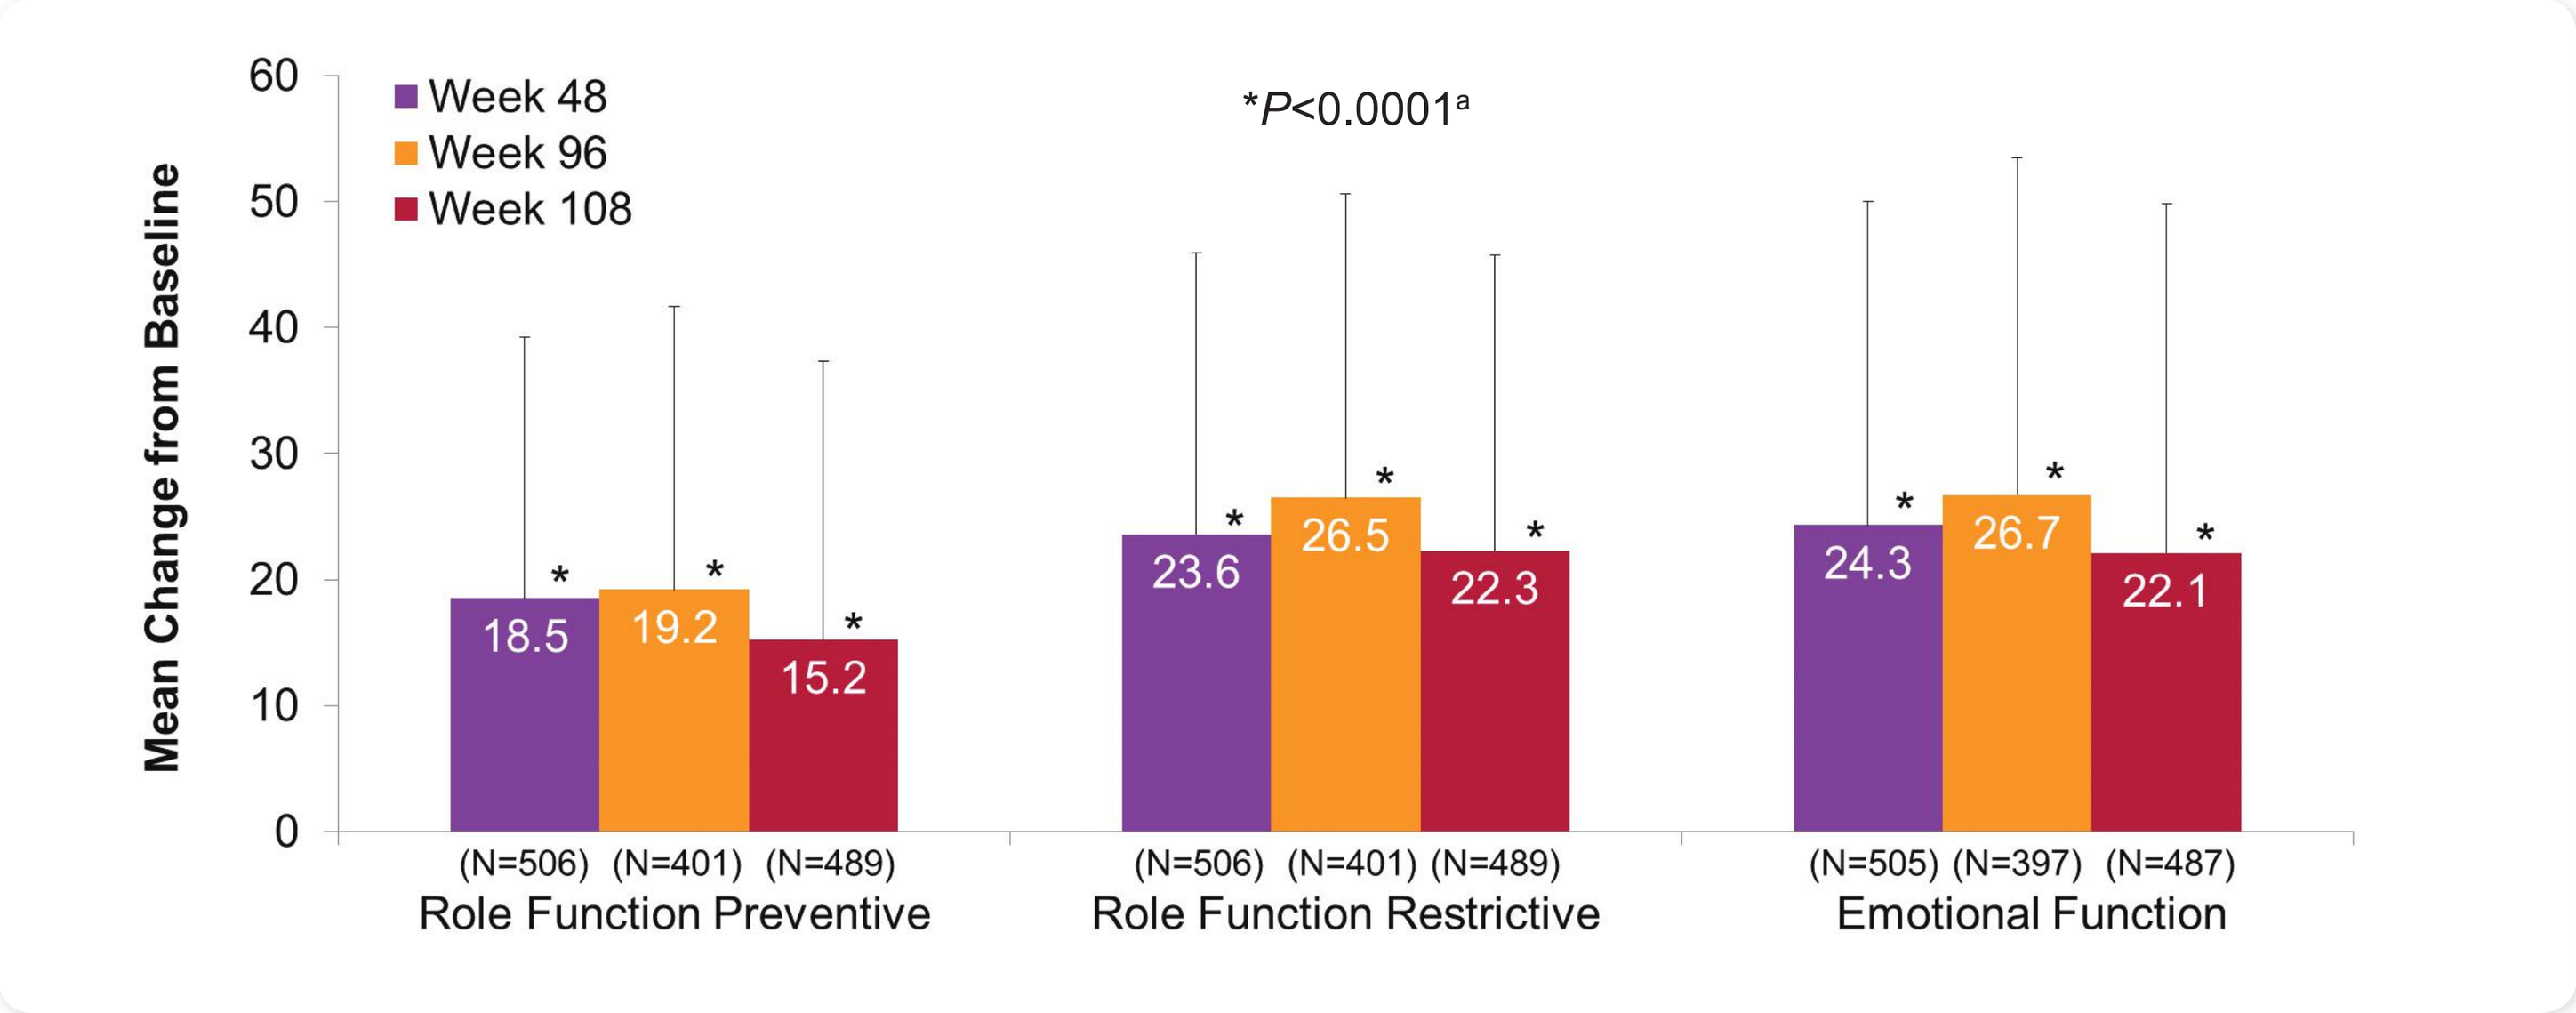

Mean Change from Baseline
60
50
40
30
20
10
0
Week 48
Week 96
Week 108
*P<0.0001a
18.5
19.2
15.2
23.6
26.5
22.3
24.3
26.7
22.1
(N=506) (N=401) (N=489)
Role Function Preventive
(N=506) (N=401) (N=489)
Role Function Restrictive
(N=505) (N=397) (N=487)
Emotional Function

NOTE: Only patients with baseline and visit observed data are included in the change from baseline at each visit
[a] Two-sided *P*-value comparing post-baseline visit to baseline from the paired *t*-test; significance level is 0.025

Safety

- 131 patients (18.3%) reported ≥1 treatment-related adverse event (TRAE)
- Most frequently reported was neck pain (n=29, 4.1%)
- One patient reported a serious TRAE (rash)
- No treatment-related deaths were reported

CONCLUSIONS

- Progressive improvements in efficacy following treatment with onabotulinumtoxinA for CM were observed through 108 weeks
- No new safety signals were identified
- Data from COMPEL supports the efficacy and safety of onabotulinumtoxinA for CM up to 108 weeks (9 treatment cycles)

REFERENCES

1. Blumenfeld AM, et al. Cephalalgia. 2011;31:301-315.
2. Buse DC, et al. Headache. 2012;52(10):1456-1470.
3. Buse DC, et al. J Neurol Neurosurg Psychiatry. 2010;81:428-432.
4. Chen YC, et al. J Headache Pain. 2012;13(4):311-319.
5. Lipton RB, et al. Neurology. 2011;77(15):1465-1472.

DISCLOSURES

Financial arrangements of the authors with companies whose products may be related to the present report are listed below, as declared by the authors.
Andrew M. Blumenfeld, MD, has consulting agreements with Allergan including intellectual property and promotional activites and in the past 12 months has provided promotional activities and consulted for Avanir, Teva, Pernix, Supernus, Depomed, Dentex, Zosano Pharma, GLG, Guidepoint, and Autodigest; **Richard Stark, MD,** has received lecture and/or consulting fees from Allergan, MSD, and inVivo Communications relating to a Pfizer-sponsored project and holds stock/stock options in CSL; **Aubrey Manack Adams, MD,** and **Amelia Orejudos, MSc,** are full-time employees of Allergan plc; **Sheena K Aurora, MD** has worked as a consultant to Allergan, Dr. Reddy's Laboratory, and Teva and has received researched support from Amgen, Gamma core, Labyrus, Eli Lilly, and Teva and is currently a full-time employee of Eli Lilly.

ACKNOWLEDGMENTS

This study was sponsored by Allergan plc, Dublin, Ireland. Activities related to study set-up, data collection, and monitoring were carried out by a contract research organization, Kantar Health, GmbH, Munich, Germany. Writing and editorial assistance was provided to the authors by Amy Kuang, PhD of Allergan plc, Irvine, CA. All authors met the ICMJE authorship criteria. Neither honoraria nor payments were made for authorship.

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit www.allergancongressposters.com/129481

Charges may apply.
No personal information is stored.

Presented at TOXINS; January 18–21, 2017; Madrid, Spain